Antimicrobial Activity of Ceftazidime-Avibactam Tested against Multidrug-Resistant *Enterobacteriaceae* and *Pseudomonas aeruginosa* Isolates from United States Medical Centers (2013–2016)

IDWeek 2017
Poster #1232

Helio S. Sader, Mariana Castanheira, Dee Shortridge, Rodrigo E. Mendes, Robert K. Flamm

JMI Laboratories, North Liberty, Iowa, USA

Contact Information:
Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

ABSTRACT

Background: The *in vitro* activity of ceftazidime-avibactam (CAZ-AVI) and many comparator agents were tested against various resistant subsets of organisms selected among 36,380 *Enterobacteriaceae* and 7,868 *Pseudomonas aeruginosa* isolates.

Methods: Isolates were consecutively collected from 94 US hospitals in 2013-2016 and tested for susceptibility by reference broth microdilution methods in a central monitoring laboratory (JMI Laboratories) as part of the International Network for Optimal Resistance Monitoring (INFORM) program. *Enterobacteriaceae* strains with elevated CAZ-AVI MIC values (≥16 µg/mL) were evaluated for the presence of genes encoding extended-spectrum β-lactamases, KPC, NDM, and transferable AmpC enzymes.

Results: CAZ-AVI inhibited 99.9% of all *Enterobacteriaceae* at the susceptible (S) breakpoint of ≤8 µg/mL and was active against multidrug-resistant (MDR; n=2,953; $MIC_{50/90}$, 0.25/1 µg/mL; 99.2%S), extensively drug-resistant (XDR; n=448; $MIC_{50/90}$, 0.5/2 µg/mL; 97.8%S), and carbapenem-resistant isolates (CRE; n=513; $MIC_{50/90}$, 0.5/2 µg/mL; 97.5%S). Only 82.2% of MDR *Enterobacteriaceae* and 64.2% of ceftriaxone-nonsusceptible (NS) *Klebsiella pneumoniae* (n=1,063) were meropenem-S. Among *Enterobacter cloacae* (n=3,740; 22.2% ceftazidime-NS), 99.8% of isolates, including 99.3% of ceftazidime-NS isolates, were CAZ-AVI-S. Only 23 of 36,380 *Enterobacteriaceae* (0.06%) isolates were CAZ-AVI-NS, including 9 MBL-producers (0.02%) and 2 KPC-producing strains with porin alteration; the remaining 12 strains showed negative results for all β-lactamases tested. CAZ-AVI showed potent activity against *P. aeruginosa* (n=7,868; $MIC_{50/90}$, 2/4 µg/mL; 97.1%S), including meropenem-NS (n=1,471; $MIC_{50/90}$, 4/16 µg/mL; 87.2%S) and MDR (n=1,562; $MIC_{50/90}$, 4/16 µg/mL; 86.5%S) isolates, and inhibited 71.8% of isolates NS to meropenem, piperacillin-tazobactam, and ceftazidime (n=628).

Conclusions: CAZ-AVI demonstrated potent activity against a large US collection (n=44,248) of contemporary gram-negative bacilli, including organisms resistant to most currently available agents, such as CRE and meropenem-NS *P. aeruginosa*.

INTRODUCTION

- The wide dissemination of extended-spectrum β-lactamases (ESBLs; such as CTX-M and SHV) and carbapenemases (such as KPC and OXA-48) has left clinicians and patients with very few treatment options for infections caused by multidrug-resistant (MDR) *Enterobacteriaceae*
- Avibactam effectively inactivates class A (ESBLs and KPC), class C (AmpC), and some class D (such as OXA-48) β-lactamases
- Ceftazidime-avibactam is approved by the United States Food and Drug Administration (US FDA) and by the European Medicine Agency (EMA) to treat complicated intra-abdominal infections (cIAI) in combination with metronidazole, as well as complicated urinary tract infections, including pyelonephritis, in patients with limited or no alternative treatment options
- Ceftazidime-avibactam is also approved to treat nosocomial pneumonia in Europe and has been studied in pediatric patients (NCT01893346)
- As part of the International Network for Optimal Resistance Monitoring (INFORM) surveillance program, we evaluated the activity of ceftazidime-avibactam against a large collection of contemporary (2013–2016) MDR gram-negative organisms causing infections in patients from US medical centers

MATERIALS AND METHODS

Bacterial isolates

- 36,380 *Enterobacteriaceae* and 7,868 *Pseudomonas aeruginosa* isolates were collected from 94 medical centers among 39 states from all 9 US census divisions in 2013-2016 as part of the INFORM program
- These isolates were collected from patients with pneumonia (n=11,666; 26.4%), bloodstream infections (n=6,110; 13.8%), skin and skin structure infections (n=9,728; 22.0%), urinary tract infections (n=12,688; 28.7%), intra-abdominal infections (n=2,038; 4.6%), and other infection types (n=2,018; 4.6%) according to defined protocols
- Only isolates determined to be significant by local criteria as the reported probable cause of infection were included in the program

Resistant subsets

- Carbapenem-resistant *Enterobacteriaceae* (CRE) were defined as isolates displaying imipenem, meropenem, and/or doripenem MIC values at ≥4 µg/mL (CLSI, 2017)
 - Imipenem was not applied to *Proteus mirabilis* and indole-positive Proteeae due to the intrinsically elevated MIC values
- MDR, extensively drug-resistant (XDR), and pan-drug-resistant (PDR) *Enterobacteriaceae* strains were classified according to recommended guidelines (Magiorakos et al., 2012) and were based on the following recommended parameters:
 - MDR = nonsusceptible (NS; CLSI breakpoints) to at least 3 antimicrobial classes
 - XDR = susceptible (S) to 2 or fewer antimicrobial classes
- *Enterobacteriaceae* analysis included the following antimicrobial classes and drug representatives:
 - Broad-spectrum cephalosporins (ceftriaxone, ceftazidime, and cefepime), carbapenems (imipenem, meropenem, and doripenem), broad-spectrum penicillin combined with β-lactamase-inhibitor (piperacillin-tazobactam), fluoroquinolones (ciprofloxacin and levofloxacin), aminoglycosides (gentamicin, tobramycin, and amikacin), glycylcyclines (tigecycline), and the polymyxins (colistin)
- *P. aeruginosa* analysis included the following antimicrobial classes and drug representatives:
 - Antipseudomonal cephalosporins (ceftazidime and cefepime), carbapenems (imipenem, meropenem, and doripenem), broad-spectrum penicillins combined with β-lactamase-inhibitor (piperacillin-tazobactam), fluoroquinolones (ciprofloxacin and levofloxacin), aminoglycosides (gentamicin, tobramycin, and amikacin), and the polymyxins (colistin)

Susceptibility testing

- Broth microdilution test method was conducted according to CLSI, and ceftazidime-avibactam was tested with avibactam at fixed concentration of 4 µg/mL
- CLSI susceptibility interpretive criteria were applied for comparator agents, and the US FDA breakpoint criteria were applied for ceftazidime-avibactam (susceptible at ≤8 µg/mL and resistant at ≥16 µg/mL when testing *Enterobacteriaceae* and *P. aeruginosa*)

RESULTS

- Ceftazidime-avibactam inhibited 99.9% of all *Enterobacteriaceae* isolates (n=36,380) at the susceptible breakpoint of ≤8 µg/mL (Tables 1 and 2 and Figure 1) and was highly active against:
 - MDR: n=2,953; $MIC_{50/90}$, 0.25/1 µg/mL; 99.2%S
 - XDR: n=448; $MIC_{50/90}$, 0.5/2 µg/mL; 97.8%S
 - CRE: n=513; $MIC_{50/90}$, 0.5/2 µg/mL; 97.5%S
- Amikacin (99.2%S), tigecycline (98.0%S [US FDA]), and meropenem (98.5%S) were also very active against the entire collection of *Enterobacteriaceae* isolates, but these antimicrobial agents exhibited limited activity against MDR, XDR, and CRE isolates (Table 2 and Figure 2)
- The most active compound tested against MDR (Figure 2) and XDR *Enterobacteriaceae* isolates was ceftazidime-avibactam, followed by tigecycline, amikacin, and meropenem (Table 2)
- Among CRE, 97.5% of isolates were susceptible to ceftazidime-avibactam, 98.8% of isolates were susceptible (US FDA criteria) to tigecycline, 68.2% were susceptible to amikacin, and 79.1% were susceptible to colistin at ≤2 µg/mL (Table 2)
- Only 23 of 36,380 *Enterobacteriaceae* (0.06%) were ceftazidime-avibactam-nonsusceptible, including:
 - 6 NDM-1-producing strains (3 *Escherichia coli*, 2 *Klebsiella pneumoniae,* and 1 *Enterobacter cloacae* with MIC of >32 µg/mL)
 - 1 VIM-4-producing *K. pneumoniae* (MIC, >32 µg/mL)
 - 1 IMP-64-producing *P. mirabilis* (MIC, >32 µg/mL)
 - 1 IMP-27-producing *Morganella morganii* (MIC, 16 µg/mL)
 - 2 KPC-producing isolates with porin alterations, a *K. pneumoniae* and an *E. cloacae* (MIC, 16 µg/mL)
 - 12 isolates (4 *E. cloacae*, 1 *E. aerogenes*, 5 *Providencia stuartii,* and 2 *Serratia marcescens*) with negative results for all β-lactamases tested
- Ceftazidime-avibactam was also active against colistin-resistant *K. pneumoniae* (99.5%S) and ceftazidime-nonsusceptible *E. cloacae* (99.3%S; Table 1)
- Ceftazidime-avibactam exhibited potent activity against *P. aeruginosa* (97.1%S), including most MDR (86.5%S) and XDR (75.9%S) isolates (Tables 1 and 2 and Figures 1 and 2)

Table 1 Antimicrobial activity of ceftazidime-avibactam tested against antimicrobial resistant *Enterobacteriaceae* and *P. aeruginosa* from US hospitals (2013–2016)

Organism (no. of isolates)	No. of isolates at MIC (µg/mL; cumulative %)													MIC_{50}	MIC_{90}
	≤0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	32	>32		
Enterobacteriaceae (36,380)	1334 3.7	4428 15.8	11139 46.5	12342 80.4	4815 93.6	1631 98.1	448 99.3	142 99.7	63 99.9	15 99.9	13 >99.9	1 >99.9	9 100.0	0.12	0.25
CRE (513)	16 3.1	5 4.1	13 6.6	35 13.5	76 28.3	137 55.0	138 81.9	59 93.4	20 97.3	1 97.5	4 98.2	0 98.2	9 100.0	0.5	2
MDR (2,953)	98 3.3	217 10.7	428 25.2	597 45.4	587 65.3	525 83.0	296 93.1	116 97.0	53 98.8	13 99.2	13 99.7	1 99.7	9 100.0	0.25	1
XDR (448)	16 3.6	9 5.6	20 10.0	25 15.6	53 27.5	126 55.6	117 81.7	44 91.5	23 96.7	5 97.8	4 98.7	0 98.7	6 100.0	0.5	2
Colistin-NS *K. pneumoniae* (205)	9 4.4	4 6.3	22 17.1	49 41.0	30 55.6	36 73.2	32 88.8	13 95.1	9 99.5	0 99.5	0 99.5	0 99.5	1 100.0	0.25	2
CAZ-NS *E. cloacae* (831)	13 1.6	3 1.9	7 2.8	44 8.1	229 35.6	345 77.1	146 94.7	30 98.3	8 99.3	0 99.3	3 99.6	1 99.8	2 100.0	0.5	1
P. aeruginosa (7,868)					141 1.8	388 6.7	2771 41.9	2779 77.3	1137 91.7	426 97.1	134 98.8	43 99.4	49 100.0	2	4
CAZ-NS (1,204)				1 0.1	1 0.2	7 0.7	93 8.5	317 34.8	344 63.4	216 81.3	133 92.4	43 95.9	49 100.0	4	16
MEM-NS (1,471)					4 0.3	8 0.8	121 9.0	382 35.0	485 68.0	282 87.2	107 94.4	37 96.9	45 100.0	4	16
P-T-NS (1,497)				2 0.1	1 0.2	15 1.2	108 8.4	360 32.5	467 63.7	336 86.1	125 94.5	39 97.1	44 100.0	4	16
CAZ, MEM, and P-T-NS (628)							22 3.5	106 20.4	174 48.1	149 71.8	101 87.9	35 93.5	41 100.0	8	32
MDR (1,562)					8 0.5	18 1.7	129 9.9	384 34.5	498 66.4	314 86.5	122 94.3	42 97.0	47 100.0	4	16
XDR (717)				1 0.1	0 0.1	1 0.3	32 4.7	126 22.3	190 48.8	194 75.9	93 88.8	36 93.9	44 100.0	8	32

Abbreviations: CRE, carbapenem-resistant *Enterobacteriaceae*; MDR, multidrug-resistant; XDR, extensively drug-resistant; NS, nonsusceptible; MEM, meropenem; CAZ, ceftazidime; and P-T, piperacillin-tazobactam

- Ceftazidime-avibactam retained *in vitro* activity against *P. aeruginosa* isolates nonsusceptible to meropenem (87.2%S), piperacillin-tazobactam (86.1%S), or ceftazidime (81.3%S), as well as isolates nonsusceptible to meropenem, piperacillin-tazobactam, and ceftazidime (71.8%S; Table 1)
- When tested against *P. aeruginosa*, amikacin was active against 87.1% of MDR; 80.8% of XDR; and 83.0% of isolates nonsusceptible to ceftazidime, meropenem, and piperacillin-tazobactam at the CLSI susceptible breakpoint of ≤16 µg/mL (Table 2 and Figure 2)

Table 2 Activity of ceftazidime-avibactam and comparator antimicrobial agents when tested against *Enterobacteriaceae*, *P. aeruginosa*, and selected resistant subsets

Organism / antimicrobial agent (no. tested)	MIC_{50}	MIC_{90}	CLSI[a] %S	CLSI[a] %R
***Enterobacteriaceae* (36,380)**				
Ceftazidime-avibactam[b]	0.12	0.25	99.9	0.1[b]
Ceftazidime	0.25	8	88.8	9.9
Piperacillin-tazobactam	2	16	92.7	4.1
Meropenem	≤0.06	≤0.06	98.5	1.3
Levofloxacin	≤0.12	>4	82.6	15.6
Gentamicin	≤1	2	91.2	7.6
Amikacin	2	4	99.2	0.2
Tigecycline	0.25	1	98.0	0.1[b]
Colistin[c]	≤0.5	>8	78.2	21.8
MDR (2,953)[d]				
Ceftazidime-avibactam[b]	0.25	1	99.2	0.8[b]
Piperacillin-tazobactam	16	>64	52.7	29.8
Meropenem	≤0.06	8	82.2	15.3
Amikacin	4	16	91.1	2.2
Tigecycline	0.5	4	88.9	0.3[b]
Colistin[c]	≤0.5	>8	60.7	39.3
XDR (448)[e]				
Ceftazidime-avibactam[b]	0.5	2	97.8	2.2[b]
Piperacillin-tazobactam	>64	>64	7.1	83.7
Meropenem	8	>8	21.2	72.5
Amikacin	16	32	60.2	9.6
Tigecycline	0.5	4	90.0	0.2[b]
Colistin[c]	≤0.5	>8	61.3	38.7
CRE (513)[f]				
Ceftazidime-avibactam[b]	0.5	2	97.5	2.5[b]
Piperacillin-tazobactam	>64	>64	3.1	91.2
Meropenem	>8	>8	2.7	89.7
Amikacin	8	32	68.2	7.0
Tigecycline	0.5	1	98.8	0.0[b]
Colistin[c]	≤0.5	>8	79.1	20.9
***P. aeruginosa* (7,868)**				
Ceftazidime-avibactam[b]	2	4	97.1	2.9[b]
Ceftazidime	2	32	84.7	10.9
Piperacillin-tazobactam	4	64	81.0	9.4
Meropenem	0.5	8	81.3	12.8
Levofloxacin	0.5	>4	74.5	18.6
Gentamicin	2	8	87.0	8.4
Amikacin	4	8	96.5	1.9
Colistin	1	2	99.6	0.4
MDR (1,562)				
Ceftazidime-avibactam[b]	4	16	86.5	13.5[b]
Ceftazidime	16	>32	43.6	41.6
Piperacillin-tazobactam	64	>64	31.2	36.2
Meropenem	8	>8	20.0	58.2
Amikacin	8	32	87.1	7.6
Colistin	1	2	99.2	0.8
XDR (717)				
Ceftazidime-avibactam[b]	8	32	75.9	24.1[b]
Ceftazidime	32	>32	21.5	60.0
Piperacillin-tazobactam	>64	>64	7.7	52.6
Meropenem	>8	>8	3.1	79.6
Amikacin	8	>32	80.8	11.6
Colistin	1	2	99.3	0.7
Ceftazidime, meropenem, and piperacillin-tazobactam-nonsusceptible (628)				
Ceftazidime-avibactam[b]	8	32	71.8	28.2[b]
Levofloxacin	>4	>4	21.9	66.2
Gentamicin	4	>8	54.0	37.4
Amikacin	8	>32	83.0	10.7
Colistin	1	2	99.5	0.5

[a] Criteria as published by CLSI
[b] Breakpoints from FDA Package Insert; susceptible at ≤8 µg/mL and resistant at ≥16 µg/mL
[c] EUCAST breakpoints were applied; susceptible at ≤2 µg/mL and resistant at ≥4 µg/mL
[d] Organisms include: *Citrobacter freundii* (59), *C. koseri* (1), *Enterobacter aerogenes* (42), *E. cloacae* (119), *E. cloacae* species complex (153), *Escherichia coli* (797), *Hafnia alvei* (3), *Klebsiella oxytoca* (55), *K. pneumoniae* (793), *Morganella morganii* (174), *Proteus mirabilis* (426), *P. vulgaris* (5), *P. vulgaris* group (1), *Providencia rettgeri* (17), *P. stuartii* (191), *Raoultella ornithinolytica* (1), *Serratia liquefaciens* (1), *S. marcescens* (112), unspeciated *Raoultella* (2), unspeciated *Serratia* (1).
[e] Organisms include: *Citrobacter freundii* (9), *Enterobacter aerogenes* (1), *E. cloacae* (44), *Escherichia coli* (12), *Klebsiella oxytoca* (8), *K. pneumoniae* (297), *Morganella morganii* (17), *Proteus mirabilis* (19), *Providencia stuartii* (18), *Raoultella ornithinolytica* (1), *Serratia marcescens* (21), unspeciated *Raoultella* (1)
[f] Organisms include: *Citrobacter freundii* (6), *Enterobacter aerogenes* (13), *E. cloacae* (5), *E. cloacae* species complex (50), *Escherichia coli* (24), *Klebsiella oxytoca* (16), *K. pneumoniae* (368), *Proteus mirabilis* (4), *Providencia stuartii* (2), *Raoultella ornithinolytica* (1), *Serratia marcescens* (22), unspeciated *Raoultella* (2)

Figure 1. Comparative coverage (susceptibility rates) of ceftazidime-avibactam and meropenem when tested against resistant subsets of *Enterobacteriaceae* and *P. aeruginosa* from US hospitals (2013-2016)

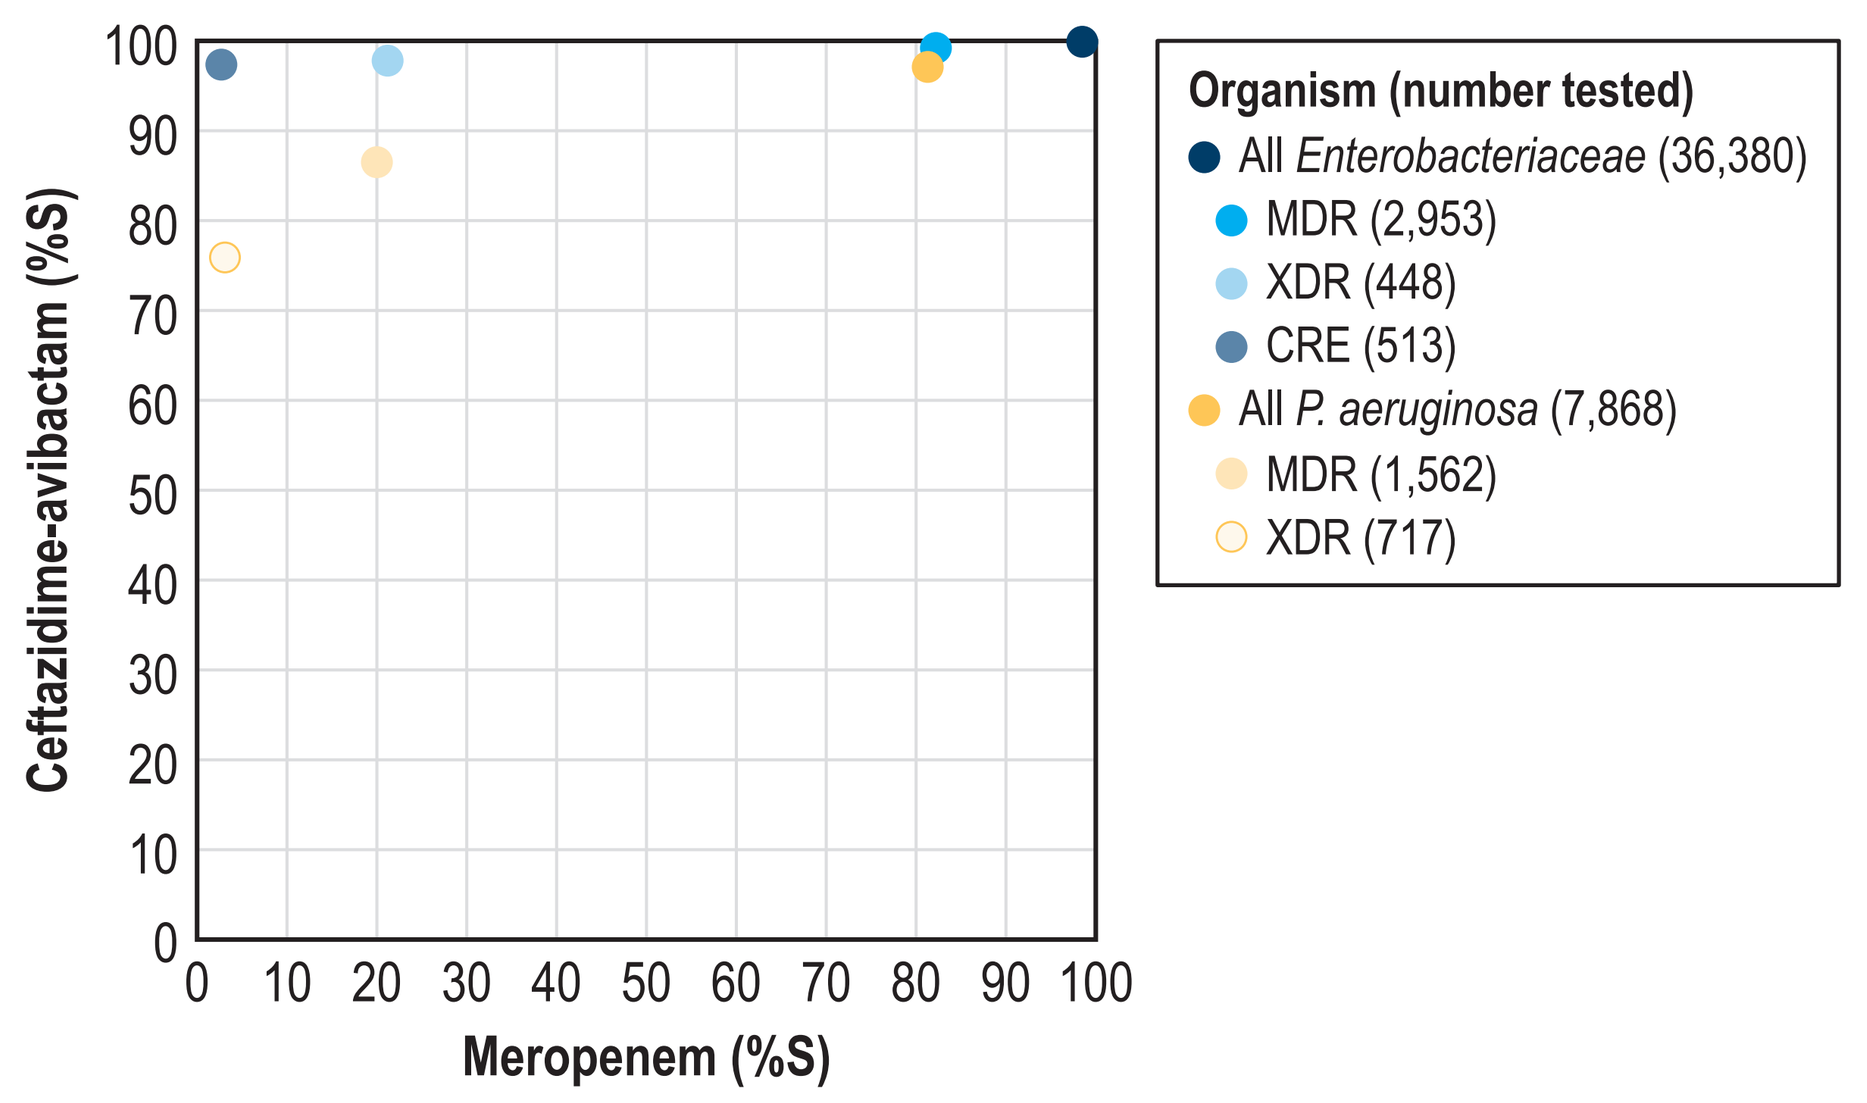

Ceftazidime-avibactam (%S)
Meropenem (%S)
0
10
20
30
40
50
60
70
80
90
100
Organism (number tested)
All Enterobacteriaceae (36,380)
MDR (2,953)
XDR (448)
CRE (513)
All P. aeruginosa (7,868)
MDR (1,562)
XDR (717)

Figure 2. Antimicrobial susceptibility of multidrug-resistant *Entero-bacteriaceae* and multidrug-resistant *P. aeruginosa* from US hospitals

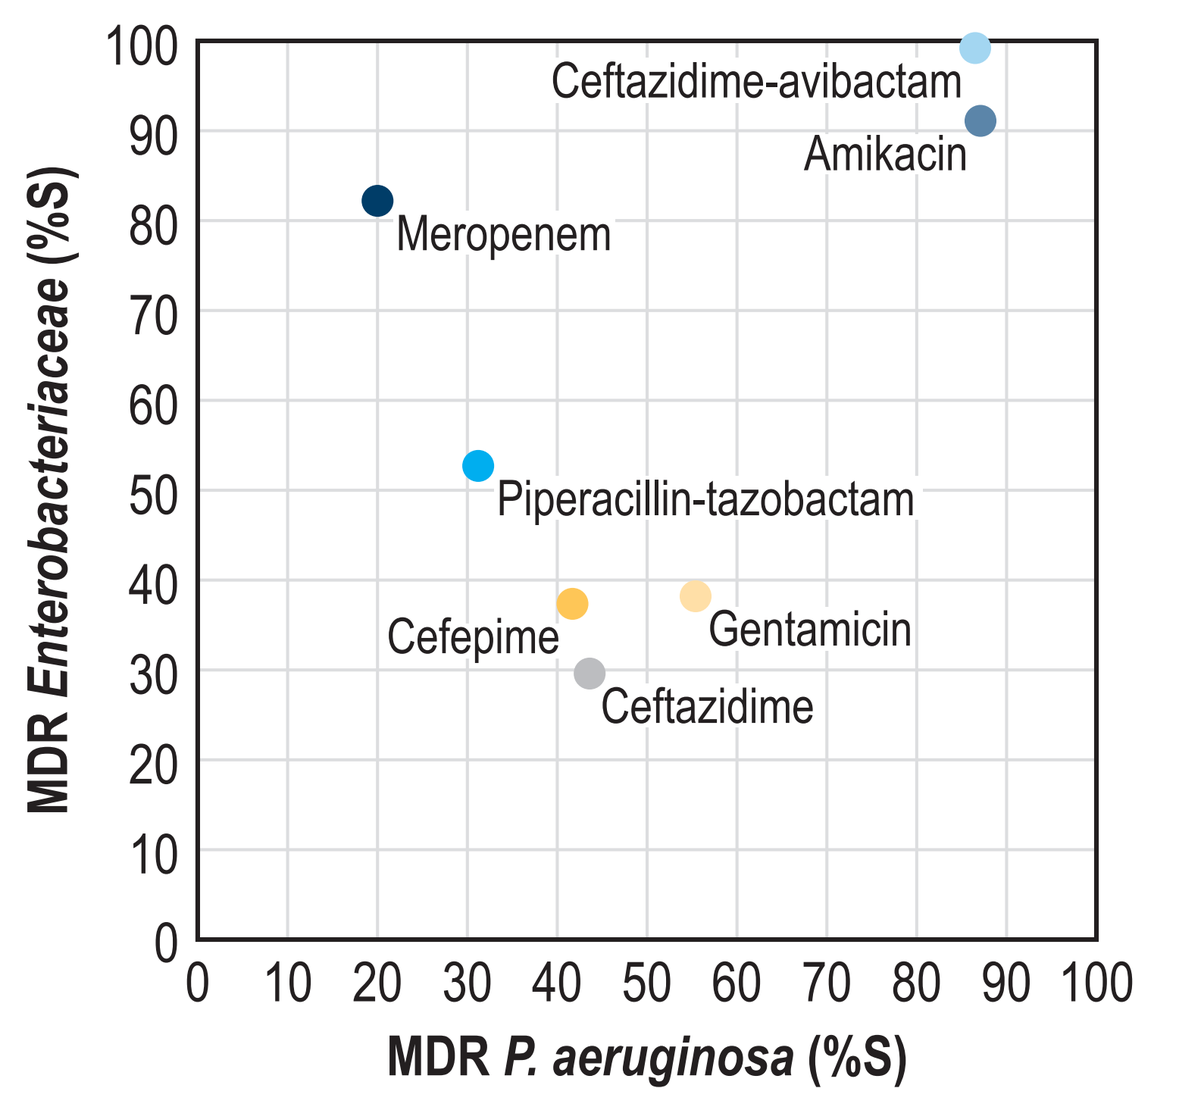

MDR Enterobacteriaceae (%S)
MDR P. aeruginosa (%S)
0
10
20
30
40
50
60
70
80
90
100
Ceftazidime-avibactam
Amikacin
Meropenem
Piperacillin-tazobactam
Cefepime
Gentamicin
Ceftazidime

CONCLUSIONS

- Ceftazidime-avibactam demonstrated potent activity against a large US collection (n=44,248) of contemporary gram-negative bacilli, including organisms resistant to most currently available agents, such as CRE and meropenem-nonsusceptible *P. aeruginosa*
- Only ceftazidime-avibactam and amikacin were active against >85% of MDR *Enterobacteriaceae* and MDR *P. aeruginosa* isolates combined (Figure 2)
- Ceftazidime-avibactam represents a valuable option for treating serious infections caused by antimicrobial resistant gram-negative organisms in US medical centers

ACKNOWLEDGMENTS

The authors would like to thank all participants of the International Network for Optimal Resistance Monitoring (INFORM) program for providing bacterial isolates.

This study was supported by Allergan. Allergan was involved in the design and decision to present these results, and JMI Laboratories received compensation for services in relation to preparing the poster. Allergan had no involvement in the collection, analysis, and interpretation of data.

REFERENCES

AVYCAZ® (2016). AVYCAZ® (ceftazidime-avibactam). Allergan USA, Inc. Available at https://www.allergan.com/assets/pdf/avycaz_pi. Accessed January 2017.

Castanheira M, Mendes RE, Jones RN, Sader HS (2016). Changes in the frequencies of beta-lactamase genes among *Enterobacteriaceae* isolates in U.S. hospitals, 2012 to 2014: Activity of ceftazidime-avibactam tested against beta-lactamase-producing isolates. *Antimicrob Agents Chemother* 60: 4770-4777.

Clinical and Laboratory Standards Institute (2015). *M07-A10. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard, tenth edition.* Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.

Deak D, Outterson K, Powers JH, Kesselheim AS (2016). Progress in the fight against multidrug-resistant bacteria? A review of U.S. Food and Drug Administration-approved antibiotics, 2010-2015. *Ann Intern Med* 165: 363-372.

Falcone M, Paterson D (2016). Spotlight on ceftazidime/avibactam: a new option for MDR Gram-negative infections. *J Antimicrob Chemother* 71: 2713-2722.

Magiorakos AP, Srinivasan A, Carey RB, Carmeli Y, Falagas ME, Giske CG, Harbarth S, Hindler JF, Kahlmeter G, Olsson-Liljequist B, Paterson DL, Rice LB, Stelling J, Struelens MJ, Vatopoulos A, Weber JT, Monnet DL (2012). Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance. *Clin Microbiol Infect* 18: 268-281.

Sader HS, Farrell DJ, Flamm RK, Jones RN (2014). Antimicrobial susceptibility of Gram-negative organisms isolated from patients hospitalized in intensive care units in United States and European hospitals (2009-2011). *Diagn Microbiol Infect Dis* 78: 443-448.

Tangden T, Giske CG (2015). Global dissemination of extensively drug-resistant carbapenemase-producing *Enterobacteriaceae*: clinical perspectives on detection, treatment and infection control. *J Intern Med* 277: 501-512.

Visit http://www.allergan congressposters.com/278027

IDWeek 2017, October 4–8, San Diego, California