IDWeek 2017

Poster #326

Characterization of β-Lactam Resistance Mechanisms among Baseline *Pseudomonas aeruginosa* from 5 Ceftazidime-Avibactam Phase 3 Clinical Trials

RE MENDES[1], M CASTANHEIRA[1], LN WOOSLEY[1], GG STONE[2], R MCLAUGHLIN[3], PA BRADFORD[3], RK FLAMM[1]

[1]JMI Laboratories, North Liberty, Iowa, USA; [2]Pfizer, Groton, Connecticut, USA; [3]Formerly of AstraZeneca Pharmaceuticals LP, Waltham, Massachusetts, USA

Contact Information:
Rodrigo E. Mendes, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo-mendes@jmilabs.com

To obtain a PDF of this poster:
- Scan the QR code
 OR
- Visit https://www.jmilabs.com/data/posters/IDWeek2017-CAZ-AVI-PSA-trials.pdf

Charges may apply.
No personal information is stored.

Introduction

- Ceftazidime-avibactam, a novel β-lactam/β-lactamase inhibitor, was evaluated in Phase 3 trials for the treatment of complicated urinary tract infections (cUTI), complicated intra-abdominal infections (cIAI), and hospital-acquired pneumonia (HAP), including ventilator-associated pneumonia (VAP)
- This combination is approved by the US Food and Drug Administration (FDA) and European Medicines Agency (EMA) for treating cIAI and cUTI
 - In addition, ceftazidime-avibactam is approved by the EMA to treat adults with HAP, including VAP
- The study presented here was carried out to characterize the β-lactam resistance mechanisms in *Pseudomonas aeruginosa* recovered from patients enrolled in 5 adult Phase 3 trials of ceftazidime-avibactam

Materials and Methods

Patients and clinical isolates

- A total of 189 baseline *P. aeruginosa* isolates (1 per patient) met the MIC-based criteria for screening of β-lactam resistance mechanisms and were recovered from patients randomized in 5 Phase 3 trials for ceftazidime-avibactam
- Patients enrolled in this study were hospitalized in medical institutions distributed in 19 countries (Table 1)
- The MIC-based criteria for screening of β-lactam resistance mechanisms were ceftazidime MIC of ≥16 µg/mL and/or carbapenem MIC of ≥8 µg/mL

Antimicrobial susceptibility testing

- Clinical isolate susceptibility testing was centrally performed using the broth microdilution method and following the Clinical Laboratory Standard Institute (CLSI) guidelines
- Ceftazidime was tested in combination with avibactam at a fixed concentration of 4 µg/mL
- Ceftazidime-avibactam (≤8 µg/mL for susceptible and ≥16 µg/mL for resistance) breakpoints published by the FDA and European Committee on Antimicrobial Susceptibility Testing (EUCAST) were applied for *P. aeruginosa*, and MIC values for comparator agents were interpreted using CLSI breakpoints

Table 1 List of countries where *P. aeruginosa* clinical isolates that met the MIC-based screening criteria originated

Countries	Number (percentage) of isolates
Asia-Pacific region	
India	4 (2.1)
Japan	20 (10.6)
Korea	5 (2.6)
China	27 (14.3)
Philippines	1 (0.5)
Vietnam	6 (3.2)
Latin America	
Brazil	8 (4.2)
Mexico	3 (1.6)
Peru	5 (2.6)
Europe	
Bulgaria	8 (4.2)
Czech Republic	28 (14.8)
France	4 (2.1)
Hungary	4 (2.1)
Poland	2 (1.1)
Romania	14 (7.4)
Ukraine	31 (16.4)
United Kingdom	1 (0.5)
Russia	13 (6.9)
Turkey	5 (2.6)
Total	189

Screening of β-lactamase and other β-lactam resistance mechanisms

- Isolates that met the MIC screening criteria were subjected to a microarray-based assay Check-MDR CT101 kit according to the manufacturer's instructions (Check-Points, Wageningen, Netherlands), which is capable of detecting CTX-M groups 1, 2, 8+25, and 9, TEM, SHV, ACC, ACT/MIR, CMY, DHA, FOX, KPC, and NDM encoding genes
- Supplemental multiplex PCR assays were used to detect additional extended-spectrum β-lactamase (ESBL)- (bla_{GES}, bla_{VEB}, bla_{PER}) and oxacillinase enzymes (bla_{OXA-2}-, bla_{OXA-10}-, and bla_{OXA-13}-groups, bla_{OXA-18}, and bla_{OXA-45}) and carbapenemase-encoding genes (bla_{IMP}, bla_{VIM}, bla_{NDM-1}, bla_{OXA-48}, bla_{GES}, bla_{NMC-A}, bla_{SME}, and bla_{IMI})
- All amplicons generated were sequenced on both strands (Sanger method); nucleotide and amino acid sequences were analyzed using the Lasergene software package (DNASTAR, Madison, Wisconsin)
- The transcription levels of the chromosomal *ampC* and efflux pump (*mexA*, *mexC*, *mexE*, and *mexX*) genes were determined using quantitative real-time PCR assays (qRT-PCR)
 - During the quantification process for the target mRNA gene, a normalized expression analysis method was applied and relative comparison to a susceptible control strain was performed
 - A strain was considered to overexpress *ampC* or efflux-pump genes when at least a 5-fold greater difference of transcripts was detected as compared with a wild-type reference control strain
- ESBL-encoding genes were screened in all isolates, but *ampC* expression was not determined in isolates from China (27 isolates)
- Expression analysis of efflux pumps and *oprD* was carried out only in isolates recovered from patients enrolled in the nosocomial pneumonia (NP) Phase 3 clinical trial (NCT01808092)

Results

- Figure 1 shows the susceptibility rates obtained for ceftazidime-avibactam and comparator agents tested against *P. aeruginosa* isolates that met the MIC-based screening criteria
- Isolates showed low susceptibility rates for most comparator agents tested, as follows: ceftazidime (30.7%), cefepime (36.0%), piperacillin-tazobactam (22.8%), imipenem (23.8%), and meropenem (30.2%)
- Ceftazidime-avibactam displayed susceptibility rates of 72.5% and 80.6% against all *P. aeruginosa* or when excluding metallo-β-lactamase (MBL) producers, respectively (Figure 1)
- Among ceftazidime-avibactam-resistant isolates (27.5%; 52/189), 36.5% (19/52) carried MBL enzymes (Table 2)
 - A total of 12 *P. aeruginosa* isolates carried IMP alleles, which were detected in isolates from China (bla_{IMP-25}), Czech Republic (bla_{IMP-7}), Mexico (bla_{IMP-18} and $_{-56}$), Ukraine (bla_{IMP-1}), and Vietnam (bla_{IMP-26}) (data not shown)
 - *P. aeruginosa* isolates carrying bla_{VIM-1} (2) were found in Ukraine and those carrying bla_{VIM-2} (5) were found in Romania and Russia
- Overexpression of the intrinsic *ampC* gene was observed among 60 (37.0%) *P. aeruginosa* (data not shown)
 - bla_{OXA} enzymes (48.8%) were the most prevalent enzymatic β-lactam resistance mechanisms observed, followed by various ESBLs (28.6%) and MBL enzymes (22.6%; Table 2)
- Isolates recovered from patients enrolled in the NP Phase 3 clinical trial showed various arrays of β-lactam resistance mechanisms (Table 3)
 - Lack of or decreased expression of OprD and overexpression of efflux pumps were noted in 47.1% and 22.5% of these pathogens, respectively (Table 3)
 - A total of 50.0% of isolates showed decreased permeability with alterations of OprD expression, overexpression of efflux pumps, or both
 - However, 20.6% of NP isolates did not show any β-lactam resistance mechanisms investigated

Figure 1 Percentage of susceptibility to ceftazidime-avibactam and comparator agents against all *P. aeruginosa* isolates

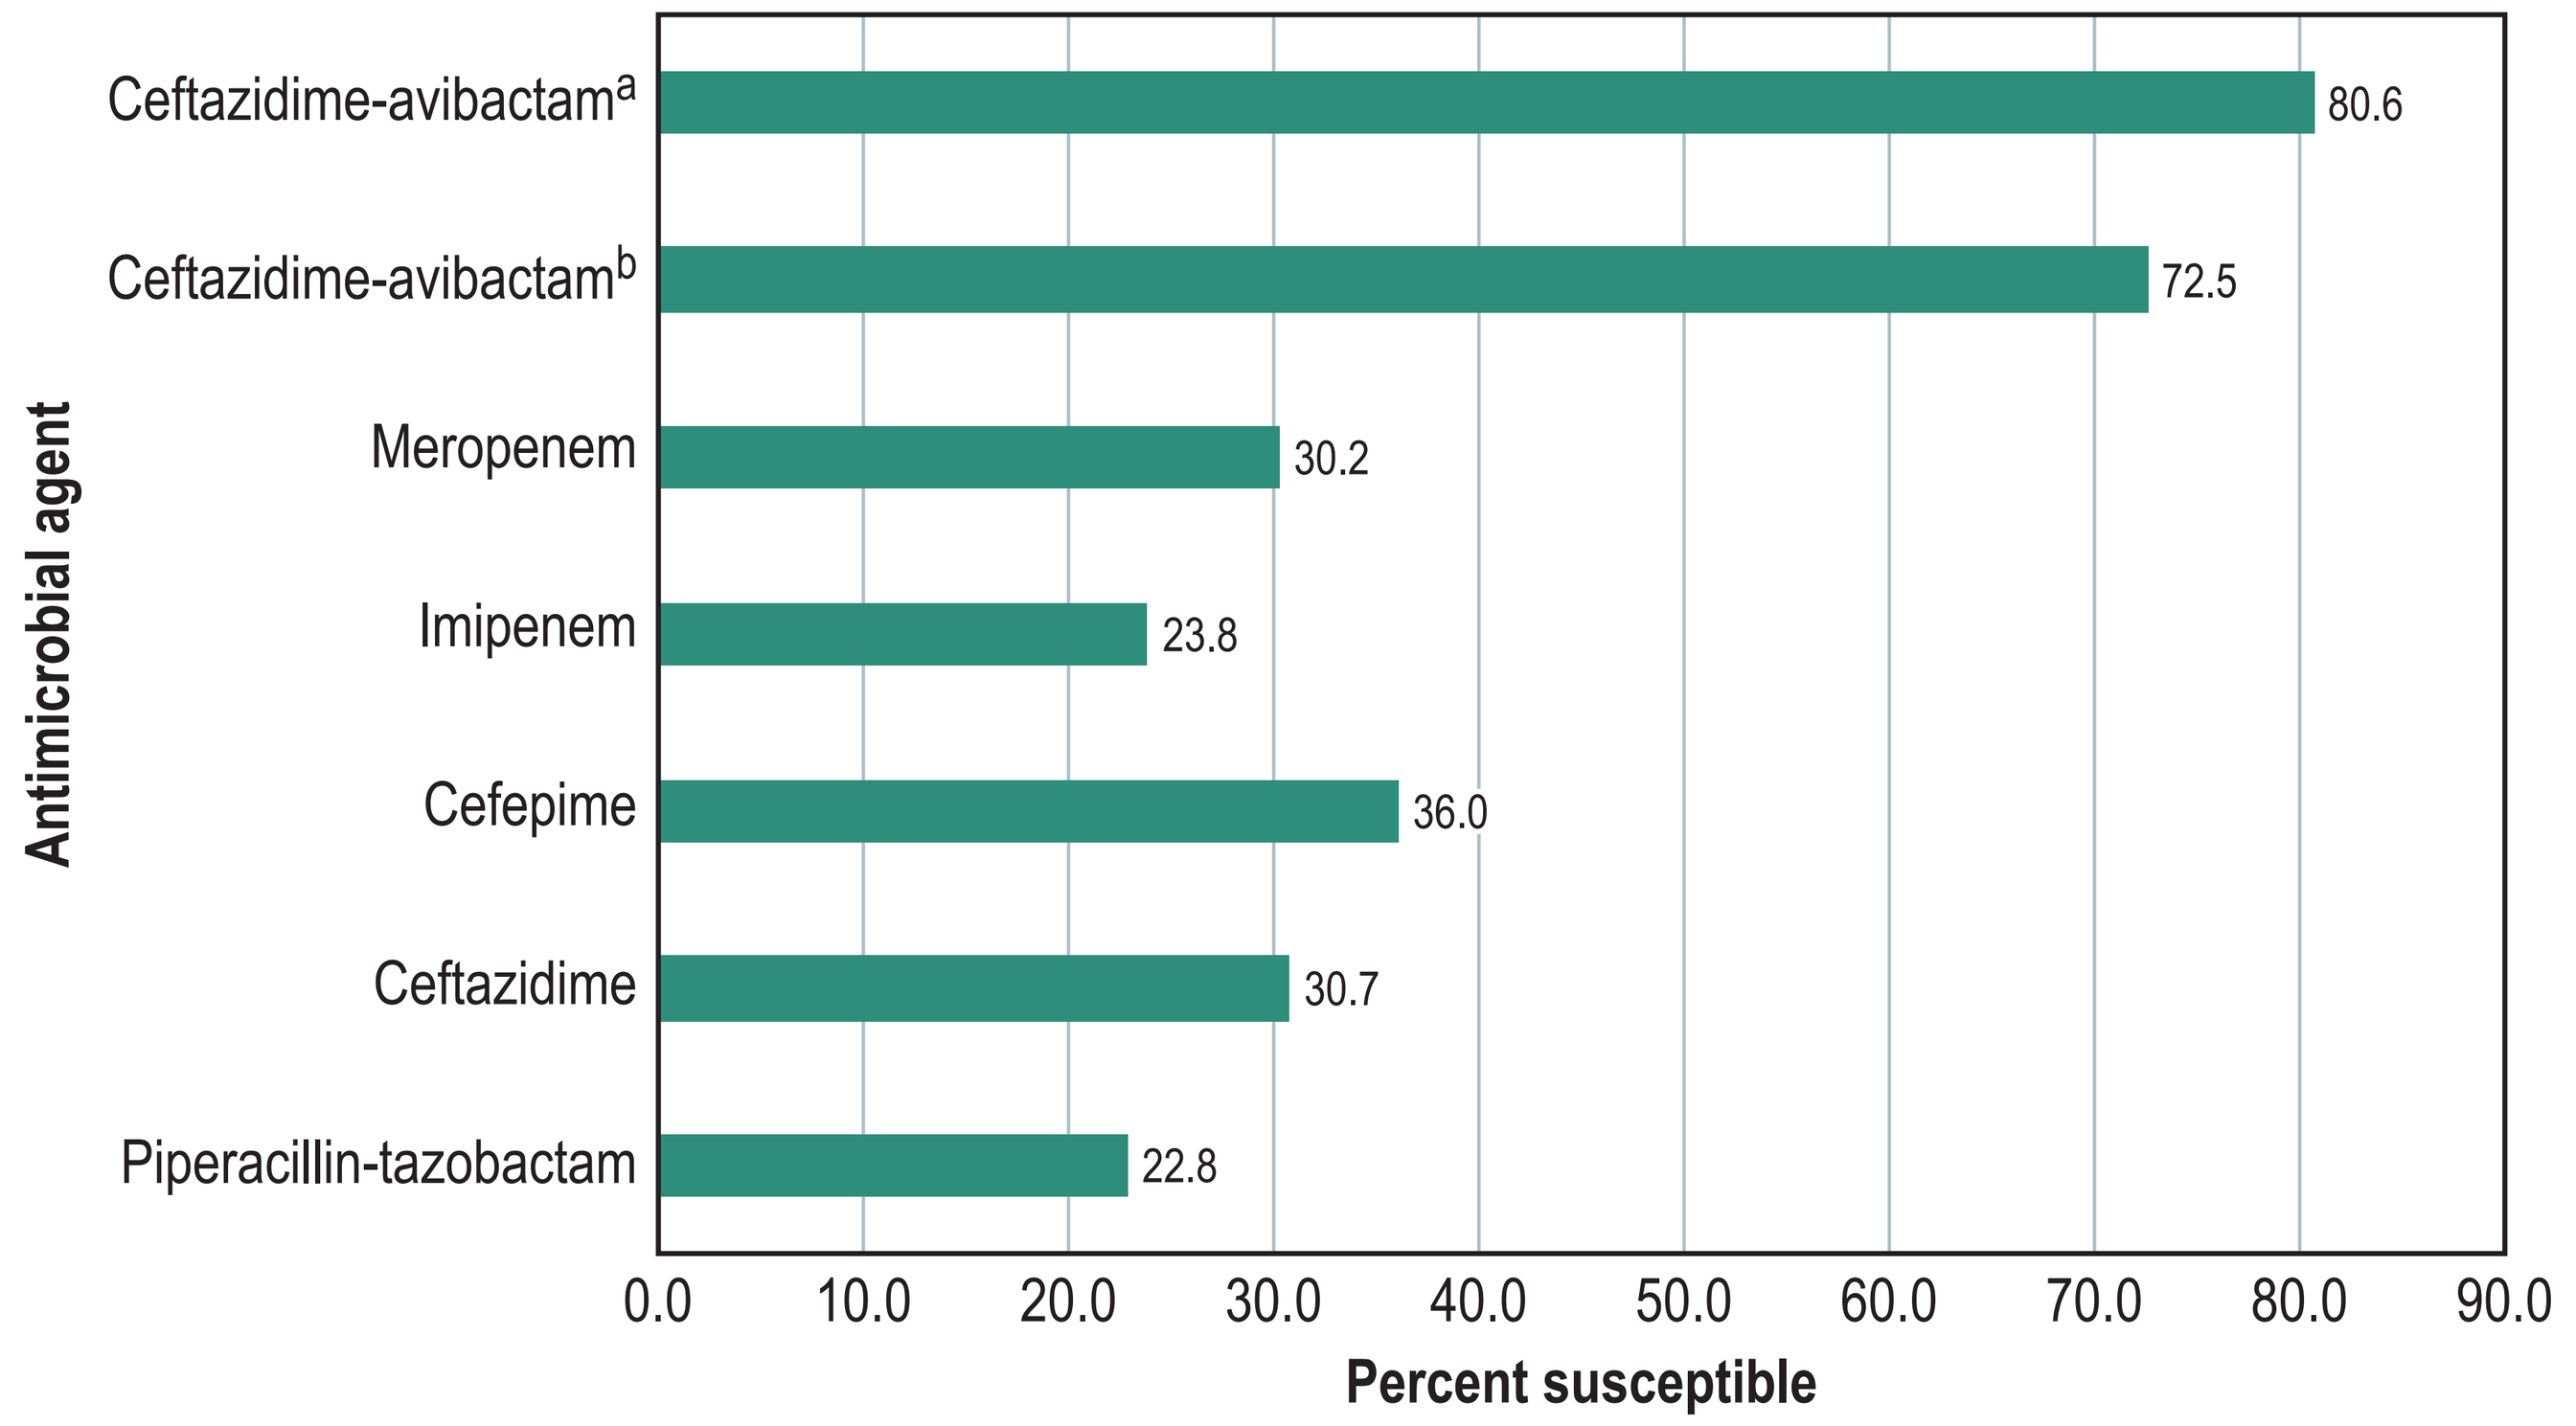

Antimicrobial agent
Ceftazidime-avibactam[a]
Ceftazidime-avibactam[b]
Meropenem
Imipenem
Cefepime
Ceftazidime
Piperacillin-tazobactam
80.6
72.5
30.2
23.8
36.0
30.7
22.8
0.0
10.0
20.0
30.0
40.0
50.0
60.0
70.0
80.0
90.0
Percent susceptible

[a] represents percent susceptible excluding MBL-producing strains; [b] represents percent susceptible against all strains

Table 2 Distribution of acquired β-lactamase-encoding genes detected among *P. aeruginosa*

β-lactamase gene[a]	Number (%)
Metallo-β-lactamase	
IMP-1	3 (3.6)
IMP-7	1 (1.2)
IMP-18	1 (1.2)
IMP-25	2 (2.4)
IMP-26	3 (3.6)
IMP-56	2 (2.4)
VIM-1	2 (2.4)
VIM-2	5 (6.0)
ESBL	
CTX-M-3	1 (1.2)
GES-1	1 (1.2)
PER-1	12 (14.3)
PSE-1	5 (6.0)
VEB-1	1 (1.2)
VEB-9	4 (4.8)
Oxacillinase	
OXA-1	2 (2.4)
OXA-2	23 (27.4)
OXA-4 (OXA-1 variant)	1 (1.2)
OXA-10	8 (9.5)
OXA-17[b] (OXA-10 variant)	1 (1.2)
OXA-74 (OXA-10 variant)	4 (4.8)
OXA-246 (OXA-10 variant)	2 (2.4)
Total	84

[a] Multiple β-lactamase genes were present in some isolates
[b] Enzyme considered to possess an ESBL phenotype

Table 3 Distribution of acquired and intrinsic β-lactamase resistance mechanisms detected among *P. aeruginosa* recovered from patients in nosocomial pneumonia Phase 3 trial[a]

β-lactamase resistance mechanisms[b]	Number (%)
cAmpC	17 (16.7)
cAmpC, OXA-2	2 (2.0)
cAmpC, OXA-2, MexA-MexC	1 (1.0)
cAmpC, PSE-1	2 (2.0)
cAmpC, IMP-56	1 (1.0)
cAmpC, IMP-56, MexA	1 (1.0)
cAmpC, VIM-1	2 (2.0)
cAmpC, VIM-2, OprD	1 (1.0)
cAmpC, MexC-MexX, OprD	2 (2.0)
cAmpC, OprD	5 (4.9)
cAmpC, MexC, OprD	1 (1.0)
cAmpC, MexX, OprD	3 (2.9)
IMP-1	1 (1.0)
IMP-26	1 (1.0)
IMP-7, OXA-2, OprD	1 (1.0)
IMP-1, OXA-10, MexX, OprD	2 (2.0)
IMP-26, OprD	1 (1.0)
IMP-26, MexX, OprD	1 (1.0)
VIM-2	2 (2.0)
PSE-1	2 (2.0)
OprD	20 (19.6)
MexX, OprD	3 (2.9)
MexC, OprD	3 (2.9)
MexA, OprD	2 (2.0)
MexA-MexC, OprD	2 (2.0)
MexA-MexC-MexX, OprD	1 (1.0)
MexE-MexX	1 (1.0)
None detected	21 (20.6)
Total	102

[a] Isolates from China were excluded
[b] cAmpC, MexA, MexC, MexD, and MexX, documented overexpression of respective gene (at least 5-fold higher than the reference wild-type control strain); OprD represents decreased or lack of expression of the OprD porin

Conclusions

- *P. aeruginosa* isolates that met MIC-based screening criteria showed low susceptibility rates (22.8%–36.0%) to otherwise potent clinically utilized anti-pseudomonal agents
- In contrast, ceftazidime-avibactam had a susceptibility rate of 72.5% and as high as 80.6% when excluding isolates carrying MBL, which precludes the drug's activity
- Oxacillin-hydrolyzing class D β-lactamase enzymes were highly prevalent among *P. aeruginosa* clinical trial isolates (48.8%)
 - However, the vast majority of oxacillinases presented here are not considered to confer an ESBL phenotype
 - ESBL and MBL represented 28.6% and 22.6% of enzymes
- Alterations in the bacterial cell permeability were the most common β-lactam resistance mechanisms observed among NP *P. aeruginosa*, especially alterations of OprD expression

Acknowledgements

Funding: This study was sponsored by AstraZeneca (AZ) and Allergan. AZ's rights to ceftazidime-avibactam were acquired by Pfizer in December 2016. The AZ share of the study data, and associated records and documentation, are now wholly owned by Pfizer. JMI Laboratories received financial support from AZ and Pfizer in connection with the study and development of this poster.

Conflict of interest: REM, MC, LNW, and RKF are employees of JMI Laboratories. GGS is an employee of Pfizer and was an employee of and shareholder in AZ at the time of the study. PB and RM were employees of and shareholders in AZ at the time of the study.

References

AVYCAZ® Package Insert (2017). AVYCAZ® (ceftazidime-avibactam), on Allergan USA, Inc. https://www.allergan.com/assets/pdf/avycaz_pi. Accessed February 2017.

Clinical and Laboratory Standards Institute (2015). *M07-A10. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; Approved Standard—Tenth Edition*. Wayne, PA, USA.

Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th Informational Supplement.* Wayne, PA, USA.

EUCAST (2017). Breakpoint tables for interpretation of MIC's and zone diameters. Version 7.1, March 2017. Available at http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_7.1_Breakpoint_Tables.pdf. Accessed March 2017.

Mendes RE, Castanheira M, Gasink L, et al. (2015). β-lactamase characterization of Gram-negative pathogens recovered from patients enrolled in the phase 2 trials for ceftazidime-avibactam: Clinical efficacies analyzed against subsets of molecularly characterized isolates. *Antimicrob Agents Chemother* 60:1328-1335.

Mendes RE, Castanheira M, Woosley LN, et al. (2017). Molecular β-lactamase characterization of aerobic Gram-negative pathogens recovered from patients enrolled in the ceftazidime-avibactam phase 3 trials for complicated intra-abdominal infections: Efficacies analyzed against susceptible and resistant subsets. *Antimicrob Agents Chemother* 61: e02447-16.

Zavicefta Package Insert (2016). Zavicefta® Package Insert, on AstraZeneca AB. Available at http://www.ema.europa.eu/docs/en_GB/document_library/EPAR_-_Product_Information/human/004027/WC500210234.pdf. Accessed September 2016.