IDWeek 2017

Poster #1205

Ceftobiprole Activity When Tested against Contemporary Bacteria Causing Bloodstream Infections in the US (2016)

RK FLAMM[1], LR DUNCAN[1], D SHORTRIDGE[1], JI SMART[2], K HAMED[2], RE MENDES[1], AND HS SADER[1]

[1]JMI Laboratories, North Liberty, Iowa, USA; [2]Basilea Pharmaceutica International, Ltd., Basel, Switzerland

Contact Information:
Robert K. Flamm, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: robert-flamm@jmilabs.com

To obtain a PDF of this poster:
- Scan the QR code
 OR
- Visit https://www.jmilabs.com/data/posters/IDWeek2017-ceftobiprole-BSI.pdf

Charges may apply.
No personal information is stored.

Introduction

- Ceftobiprole is an investigational antimicrobial in Phase 3 development in the US, and is approved in a number of countries in Europe and other parts of the world for the treatment of adult patients with either community- or hospital-acquired pneumonia, excluding ventilator-associated pneumonia
- This agent is a parenteral advanced-generation cephalosporin that was rationally designed to bind strongly to and inhibit penicillin-binding protein 2A, and it is active against many Gram-positive and -negative bacteria
- Ceftobiprole has potent activity against *Staphylococcus aureus*, including methicillin-resistant *S. aureus* (MRSA) and penicillin-resistant *Streptococcus pneumoniae*, and has activity against *Enterobacteriaceae* and *Pseudomonas aeruginosa*
- In this study, the activity of ceftobiprole and comparator antimicrobial agents was evaluated against contemporary bacterial pathogens from the SENTRY Antimicrobial Surveillance Program, which were collected from patients in US medical centers and designated as causing bloodstream infections (BSI) during 2016

Materials and Methods

Bacterial isolates

- A total of 2,787 bacterial isolates were collected prospectively from patients at 30 medical centers in the US
- Isolates were collected from BSI
 - Species identification was confirmed by standard biochemical algorithms and matrix-assisted laser desorption ionization-time of flight mass spectrometry for select organisms using the Bruker Daltonics MALDI Biotyper (Billerica, Massachusetts, USA) following manufacturer instructions

Susceptibility testing

- Ceftobiprole, comparators, and quality-control organisms were tested according to Clinical and Laboratory Standards Institute guidelines using broth microdilution panels
 - CLSI (2017) and EUCAST (2017) interpretive criteria were applied
- For *Klebsiella* spp., *Escherichia coli*, and *Proteus mirabilis* in this analysis, an extended-spectrum β-lactamase (ESBL) screen-positive phenotype was defined as an MIC of ≥2 µg/mL for ceftazidime, ceftriaxone, and/or aztreonam
 - Although other β-lactamases, such as *Amp*C and KPC, may also produce an ESBL screening-positive phenotype, these strains were grouped because they usually demonstrate resistance to various broad-spectrum β-lactam compounds

Results

- The distribution of organisms collected from BSI is shown in Figure 1
- $MIC_{50/90}$ for ceftobiprole when tested against *S. aureus* was 0.5/2 µg/mL (99.6% susceptible [EUCAST criteria]; Tables 1 and 2)
 - 41.7% of *S. aureus* isolates were MRSA
 - $MIC_{50/90}$ for MRSA and methicillin-susceptible *S. aureus* (MSSA) isolates were 1/2 µg/mL and 0.5/0.5 µg/mL, respectively (Tables 1 and 2)
 - All MRSA isolates were susceptible to tigecycline, vancomycin, and linezolid; 96.5% of MRSA isolates were susceptible to ceftaroline (CLSI criteria)

Figure 1. Distribution (number) of organisms isolated in bloodstream infections

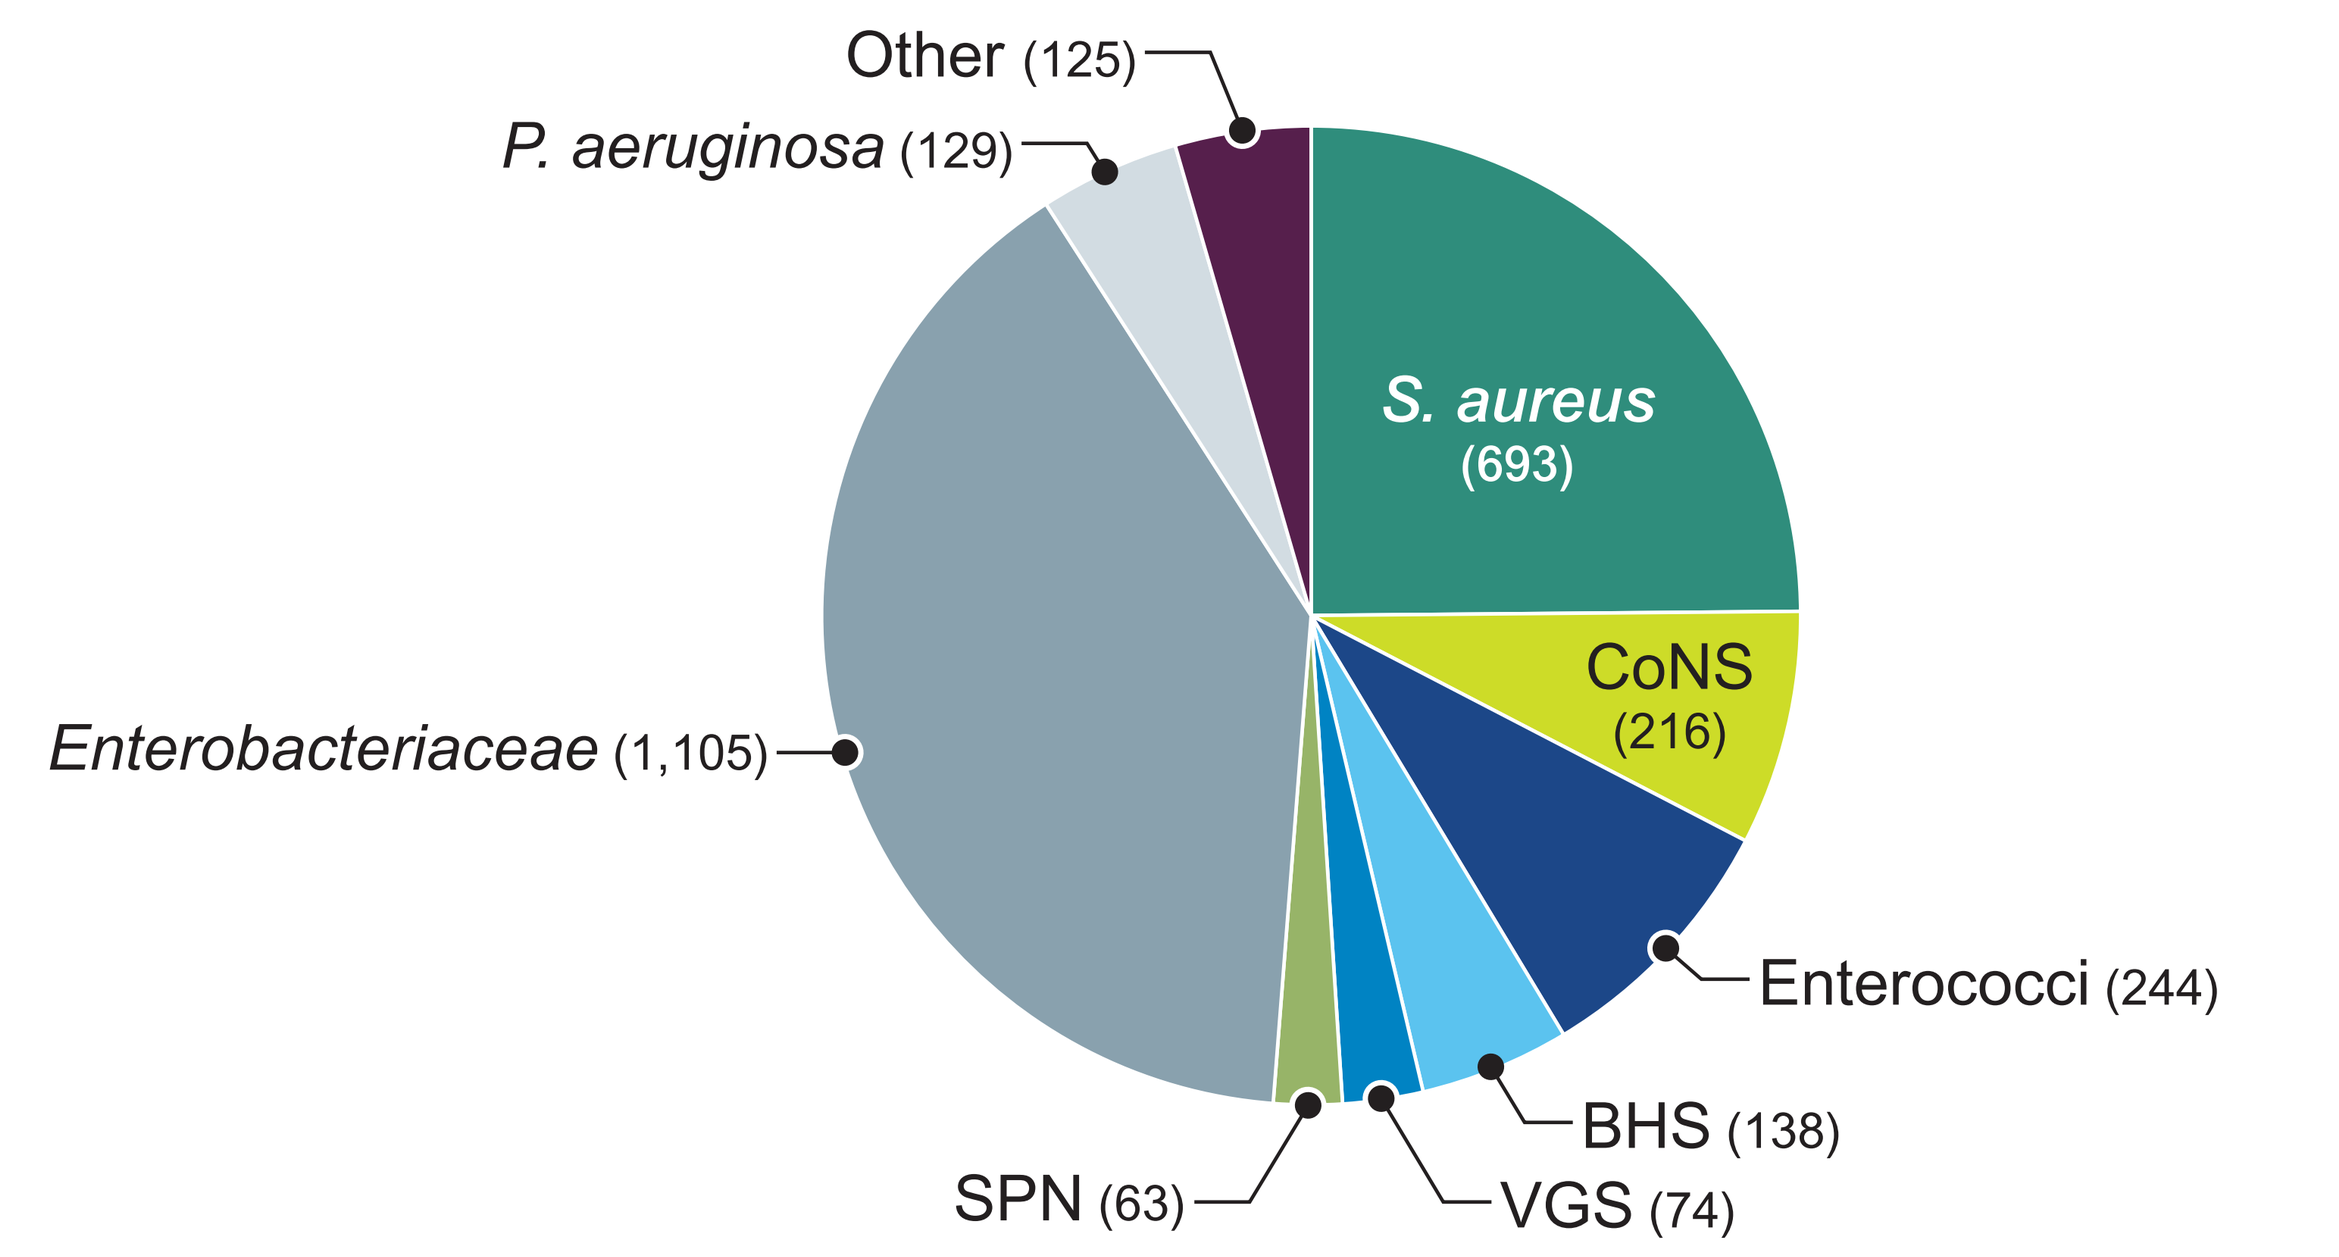

Other (125)
P. aeruginosa (129)
S. aureus (693)
CoNS (216)
Enterobacteriaceae (1,105)
Enterococci (244)
BHS (138)
VGS (74)
SPN (63)

CoNS, coagulase-negative staphylococci; BHS, beta-haemolytic streptococci; VGS, viridans group streptococci; SPN, *Streptococcus pneumoniae*

 - MRSA generally showed higher resistance to most agents compared to MSSA
 - Erythromycin resistance in MRSA was 86.9% (CLSI) and levofloxacin resistance was 73.7% (Table 2)
 - Ceftobiprole and ceftaroline ($MIC_{50/90}$, 0.25/0.25 µg/mL) were the most potent cephalosporins tested when tested against MSSA isolates and were 8- to 16-fold more potent than ceftriaxone (Table 2)
- Against 216 isolates of coagulase-negative staphylococci (CoNS), the $MIC_{50/90}$ values for ceftobiprole were 0.5/1 µg/mL, and all MIC values were ≤4 µg/mL (Table 1)
 - $MIC_{50/90}$ values for methicillin-resistant CoNS and methicillin-susceptible CoNS were 1/1 and 0.12/0.5 µg/mL, respectively (Table 1)
 - Ceftaroline ($MIC_{50/90}$, 0.25/0.5 µg/mL) and ceftobiprole ($MIC_{50/90}$, 0.5/1 µg/mL) were the most potent β-lactam agents tested against CoNS (Table 2)
 - All isolates were susceptible to daptomycin, tigecycline (EUCAST), and vancomycin; 95.4% were susceptible to linezolid (Table 2)
- For the enterococci, ceftobiprole was active against *Enterococcus faecalis* ($MIC_{50/90}$, 0.5/2 µg/mL; n=153) but not against *E. faecium* ($MIC_{50/90}$, >4/>4 µg/mL; n=90, Table 1)
 - Ceftaroline was active against *E. faecalis* ($MIC_{50/90}$, 2/8 µg/ml; 4-fold less active than ceftobiprole) but not against *E. faecium* ($MIC_{50/90}$, >8/>8 µg/mL)
- β-lactam agents exhibiting high levels of activity when tested against *S. pneumoniae* included ceftobiprole ($MIC_{50/90}$, 0.015/0.06 µg/mL; 100.0% susceptible [EUCAST criteria]), ceftaroline ($MIC_{50/90}$, ≤0.008/0.03 µg/mL [100.0% susceptible]), ceftriaxone ($MIC_{50/90}$, 0.03/0.12 µg/mL [100.0% susceptible]), and penicillin ($MIC_{50/90}$, 0.015/0.12 µg/mL [100.0% susceptible; parenteral nonmeningitis breakpoints]; Table 2)
- Ceftobiprole was also very active against β-haemolytic streptococci ($MIC_{50/90}$, 0.015/0.03 µg/mL) and viridans group streptococci ($MIC_{50/90}$, 0.03/0.5 µg/mL)
- Against *Enterobacteriaceae*, ceftobiprole demonstrated a susceptibility profile similar to that of other expanded-spectrum cephalosporins
 - Ceftobiprole ($MIC_{50/90}$, 0.03/>16 µg/mL; 85.0% susceptible [EUCAST criteria]) activity was similar to cefepime ($MIC_{50/90}$, ≤0.12/8 µg/mL; 88.9% susceptible [CLSI criteria]) and ceftazidime ($MIC_{50/90}$, 0.25/16 µg/mL; 87.2% susceptible [CLSI criteria]; ceftaroline susceptibility was 77.9% [$MIC_{50/90}$, 0.03/>16 µg/mL]; Tables 1 and 3)
 - Non-ESBL screen-phenotype *E. coli* and *K. pneumoniae* were highly susceptible to ceftobiprole (99.8% for *E. coli* and 100.0% for *K. pneumoniae* [EUCAST criteria]; Table 3)
- A total of 76.7% of *P. aeruginosa* isolates were inhibited at ≤4 µg/mL and 84.5% at ≤8 µg/mL (Table 1)
 - 85.3% were susceptible to cefepime and 89.1% to ceftazidime (CLSI criteria, Table 3)
- A total of 78.0% of *Acinetobacter* spp. isolates were inhibited at ≤4 µg/mL and 80.5% at ≤8 µg/mL, and cefepime and ceftazidime inhibited 75.6% and 73.2% of the isolates at their susceptibility breakpoints (CLSI), respectively (Table 3)
- Ceftobiprole was inactive against all *Stenotrophomonas maltophilia* isolates tested (MIC, >16 µg/mL; Table 1)

Table 1. Antimicrobial activity of ceftobiprole tested against the main organisms and organism groups of bloodstream infections (µg/mL)

Organism / organism group (no. of isolates)	No. of isolates at MIC (µg/mL; cumulative %) ≤0.002	0.004	0.008	0.015	0.03	0.06	0.12	0.25	0.5	1	2	4	8	16	>	MIC_{50}	MIC_{90}
Staphylococcus aureus (693)						0 0.0	2 0.3	161 23.5	262 61.3	185 88.0	80 99.6	3 100.0				0.5	2
MSSA (404)						0 0.0	2 0.5	160 40.1	242 100.0							0.5	0.5
MRSA (289)							0 0.0	1 0.3	20 7.3	185 71.3	80 99.0	3 100.0				1	2
Coagulase-negative staphylococci (216)					8 3.7	8 7.4	31 21.8	28 34.7	55 60.2	77 95.8	5 98.1	4 100.0				0.5	1
MS-CoNS (74)					8 10.8	8 21.6	30 62.2	21 90.5	7 100.0							0.12	0.25
MR-CoNS (142)						0 0.0	1 0.7	7 5.6	48 39.4	77 93.7	5 97.2	4 100.0				1	1
Enterococcus faecalis (153)					2 1.3	3 3.3	7 7.8	20 20.9	69 66.0	23 81.0	23 96.1	6 100.0				0.5	2
Enterococcus faecium (90)								0 0.0	1 1.1	6 7.8	10 18.9	4 23.3			69 100.0	>4	>4
Streptococcus pneumoniae (63)	0 0.0	1 1.6	20 33.3	27 76.2	8 88.9	1 90.5	1 92.1	0 92.1	5 100.0							0.015	0.06
Viridans group streptococci (74)	0 0.0	2 2.7	6 10.8	16 32.4	14 51.4	10 64.9	13 82.4	4 87.8	6 95.9	1 97.3	1 98.6				1 100.0	0.03	0.5
β-haemolytic streptococci (138)	0 0.0	1 0.7	33 24.6	42 55.1	61 99.3	1 100.0										0.015	0.03
Other β-haemolytic streptococci (32)	0 0.0	1 3.1	9 31.2	21 96.9	1 100.0											0.015	0.015
Enterobacteriaceae (1,105)			0 0.0	42 3.8	610 59.0	201 77.2	62 82.8	24 85.0	11 86.0	11 87.0	7 87.6	7 88.2	4 88.6	4 89.0	122 100.0	0.03	>16
Escherichia coli (529)			0 0.0	14 2.6	339 66.7	79 81.7	10 83.6	6 84.7	0 84.7	2 85.1	0 85.1	0 85.1	0 85.1	1 85.3	78 100.0	0.03	>16
ESBL-phenotype *Escherichia coli* (89)				0 0.0	2 2.2	2 4.5	3 7.9	2 10.1	0 10.1	1 11.2	0 11.2	0 11.2	0 11.2	1 12.4	78 100.0	>16	>16
Non-ESBL-phenotype *Escherichia coli* (440)			0 0.0	14 3.2	337 79.8	77 97.3	7 98.9	4 99.8	0 99.8	1 100.0						0.03	0.06
Klebsiella pneumoniae (268)			0 0.0	19 7.1	171 70.9	35 84.0	6 86.2	2 86.9	0 86.9	3 88.1	3 89.2	0 89.2	0 89.2	0 89.2	29 100.0	0.03	>16
ESBL-phenotype *Klebsiella pneumoniae* (40)				0 0.0	3 7.5	0 7.5	1 10.0	1 12.5	0 12.5	3 20.0	3 27.5	0 27.5	0 27.5	0 27.5	29 100.0	>16	>16
Non-ESBL-phenotype *Klebsiella pneumoniae* (228)			0 0.0	19 8.3	168 82.0	35 97.4	5 99.6	1 100.0								0.03	0.06
Klebsiella oxytoca (45)				0 0.0	6 13.3	4 22.2	16 57.8	12 84.4	2 88.9	0 88.9	1 91.1	0 91.1	0 91.1	0 91.1	4 100.0	0.12	2
Pseudomonas aeruginosa (129)								0 0.0	2 1.6	27 22.5	56 65.9	14 76.7	10 84.5	16 96.9	4 100.0	2	16
Acinetobacter spp. (41)			1 2.4	2 7.3	2 12.2	2 17.1	3 24.4	4 34.1	10 58.5	7 75.6	0 75.6	1 78.0	1 80.5	0 80.5	8 100.0	0.5	>16
Stenotrophomonas maltophilia (30)														0 0.0	30 100.0	>16	>16

Table 2. Activity of ceftobiprole and comparator agents when tested against select Gram-positive bacteria from bloodstream infections (US; 2016)

Antimicrobial agent	MIC_{50}	MIC_{90}	Range	CLSI[a] %S	CLSI[a] %I	CLSI[a] %R	EUCAST[a] %S	EUCAST[a] %I	EUCAST[a] %R
MRSA (289)									
Ceftobiprole	1	2	0.25 – 4				99.0		1.0
Ceftaroline	0.5	1	0.25 – 2	96.5	3.5	0.0	96.5		3.5
Ceftriaxone	>8	>8	8 – >8	0.0		100.0			
Clindamycin	≤0.25	>2	≤0.25 – >2	63.7	0.0	36.3	63.7	0.0	36.3
Daptomycin	0.25	0.5	≤0.12 – 2	99.7			99.7		0.3
Erythromycin	>8	>8	0.12 – >8	9.0	4.2	86.9	10.0	1.0	88.9
Gentamicin	≤1	≤1	≤1 – >8	95.2	0.3	4.5	95.2		4.8
Levofloxacin	4	>4	0.12 – >4	23.2	3.1	73.7	23.2		76.8
Linezolid	1	1	0.25 – 2	100.0		0.0	100.0		0.0
Tetracycline	≤0.5	1	≤0.5 – >8	97.9	0.3	1.7	94.8	2.8	2.4
Tigecycline	0.06	0.12	≤0.015 – 0.25	100.0		[b]	100.0		0.0
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	≤0.5 – >4	95.5		4.5	95.5	0.7	3.8
Vancomycin	0.5	1	0.25 – 2	100.0	0.0	0.0	100.0		0.0
MSSA (404)									
Ceftobiprole	0.5	0.5	0.12 – 0.5				100.0		0.0
Ceftaroline	0.25	0.25	≤0.06 – 0.5	100.0	0.0	0.0	100.0		0.0
Ceftriaxone	4	4	1 – 8	100.0		0.0			
Clindamycin	≤0.25	≤0.25	≤0.25 – >2	96.5	0.0	3.5	96.3	0.2	3.5
Daptomycin	0.25	0.5	≤0.12 – 1	100.0			100.0		0.0
Erythromycin	0.25	>8	≤0.06 – >8	69.3	8.7	22.0	70.0	3.7	26.2
Gentamicin	≤1	≤1	≤1 – >8	99.5	0.2	0.2	98.8		1.2
Levofloxacin	0.12	4	≤0.03 – >4	86.1	0.5	13.4	86.1		13.9
Linezolid	1	1	0.25 – 2	100.0		0.0	100.0		0.0
Tetracycline	≤0.5	≤0.5	≤0.5 – >8	97.3	0.2	2.5	95.5	0.2	4.2
Tigecycline	0.06	0.12	≤0.015 – 0.25	100.0		[b]	100.0		0.0
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	≤0.5 – >4	99.0		1.0	99.0	0.2	0.7
Vancomycin	0.5	1	0.25 – 2	100.0	0.0	0.0	100.0		0.0
CoNS (216)									
Ceftobiprole	0.5	1	≤0.03 – 4						
Ceftaroline	0.25	0.5	≤0.06 – 2						
Ceftriaxone	8	>8	0.5 – >8	34.3		65.7			
Clindamycin	≤0.25	>2	≤0.25 – >2	66.2	0.9	32.9	65.7	0.5	33.8
Daptomycin	0.25	0.5	≤0.12 – 1	100.0			100.0		0.0
Erythromycin	>8	>8	≤0.06 – >8	34.3	5.6	60.2	36.6	1.9	61.6
Gentamicin	≤1	>8	≤1 – >8	70.4	2.8	26.9	67.6		32.4
Levofloxacin	0.5	>4	0.06 – >4	51.9	1.9	46.3	51.9		48.1
Linezolid	0.5	1	≤0.12 – >8	95.4		4.6	95.4		4.6
Oxacillin	>2	>2	≤0.25 – >2	34.3		65.7	34.7		65.3
Tetracycline	≤0.5	>8	≤0.5 – >8	83.3	0.0	16.7	81.5	1.4	17.1
Tigecycline	0.06	0.12	0.03 – 0.5				100.0		0.0
Trimethoprim-sulfamethoxazole	≤0.5	>4	≤0.5 – >4	63.9		36.1	63.9	25.0	11.1
Vancomycin	1	2	0.25 – 2	100.0	0.0	0.0	100.0		0.0
β-haemolytic streptococci (138)									
Ceftobiprole	0.015	0.03	0.004 – 0.06						
Ceftaroline	≤0.008	0.015	≤0.008 – 0.03	100.0			100.0		0.0
Ceftriaxone	0.06	0.06	≤0.015 – 0.12	100.0			100.0		0.0
Clindamycin	≤0.25	>2	≤0.25 – >2	78.3	0.7	21.0	79.0		21.0
Daptomycin	0.12	0.25	≤0.06 – 1	100.0			100.0		0.0
Erythromycin	0.06	>32	≤0.015 – >32	59.1	0.0	40.9	59.1	0.0	40.9
Levofloxacin	0.5	1	0.25 – 2	100.0	0.0	0.0	100.0		0.0
Linezolid	1	1	0.5 – 2	100.0			100.0	0.0	0.0
Meropenem	0.015	0.06	≤0.008 – 0.06	100.0			100.0		0.0
Penicillin	0.03	0.06	≤0.004 – 0.06	100.0			100.0		0.0
Tetracycline	>8	>8	≤0.25 – >8	45.7	2.2	52.2	44.9	0.7	54.3
Vancomycin	0.25	0.5	0.25 – 0.5	100.0			100.0		0.0
***Enterococcus faecalis* (153)**									
Ceftobiprole	0.5	2	≤0.03 – 4						
Ampicillin	1	2	≤0.5 – 4	100.0		0.0	100.0	0.0	0.0
Ceftaroline	2	8	≤0.06 – >8						
Daptomycin	1	1	≤0.25 – 2	100.0					
Levofloxacin	1	>4	0.06 – >4	73.9	0.0	26.1	73.9		26.1[c]
Linezolid	1	2	0.25 – 2	100.0	0.0	0.0	100.0		0.0
Teicoplanin	≤0.5	≤0.5	≤0.5 – >16	95.4	0.0	4.6	95.4		4.6
Tigecycline	0.06	0.12	≤0.015 – 0.12	100.0		[b]	100.0	0.0	0.0
Vancomycin	1	2	0.5 – >16	95.4	0.0	4.6	95.4		4.6
***Enterococcus faecium* (90)**									
Ceftobiprole	>4	>4	0.5 – >4						
Ampicillin	>16	>16	≤0.5 – >16	23.3		76.7	22.2	1.1	76.7
Ceftaroline	>8	>8	0.12 – >8						
Daptomycin	1	2	≤0.25 – >8	97.8					
Levofloxacin	>4	>4	0.5 – >4	14.4	8.9	76.7	23.3		76.7[c]
Linezolid	1	2	0.25 – 8	97.8	1.1	1.1	98.9		1.1
Teicoplanin	>16	>16	≤0.5 – >16	36.7	3.3	60.0	35.6		64.4
Tigecycline	0.03	0.06	≤0.015 – 0.25				100.0	0.0	0.0
Vancomycin	>16	>16	0.25 – >16	35.6	0.0	64.4	35.6		64.4
Viridans group streptococci (74)									
Ceftobiprole	0.03	0.5	0.004 – >2						
Ceftaroline	0.03	0.12	≤0.008 – 0.5						
Ceftriaxone	0.12	1	≤0.015 – >2	91.9	4.1	4.1	86.5		13.5
Clindamycin	≤0.25	>2	≤0.25 – >2	81.1	1.4	17.6	82.4		17.6
Daptomycin	0.25	1	≤0.06 – 1	100.0					
Erythromycin	1	>32	≤0.015 – >32	47.3	1.4	51.4			
Levofloxacin	1	>4	0.25 – >4	89.2	0.0	10.8			
Linezolid	1	1	0.12 – 1	100.0					
Meropenem	0.06	0.5	≤0.008 – >1	93.2			100.0		0.0
Penicillin	0.12	2	0.008 – 8	64.9	31.1	4.1	75.7	20.3	4.1
Tetracycline	1	>8	≤0.25 – >8	56.8	4.1	39.2			
Vancomycin	0.5	0.5	0.25 – 1	100.0			100.0		0.0
***Streptococcus pneumoniae* (63)**									
Ceftobiprole	0.015	0.06	0.004 – 0.5				100.0		0.0
Ceftaroline	≤0.008	0.03	≤0.008 – 0.12	100.0		[d]	100.0		0.0
Ceftriaxone	0.03	0.12	≤0.015 – 1	93.7	6.3	0.0[e]	93.7	6.3	0.0
				100.0	0.0	0.0[d]			
Clindamycin	≤0.25	≤0.25	≤0.25 – >2	95.2	0.0	4.8	95.2		4.8
Erythromycin	0.03	16	≤0.015 – >32	68.3	1.6	30.2	68.3	1.6	30.2
Levofloxacin	1	1	0.5 – 2	100.0	0.0	0.0	100.0		0.0
Linezolid	1	2	0.5 – 2	100.0			100.0	0.0	0.0
Penicillin	0.015	0.12	0.015 – 2	81.0	12.7	6.3[f]	81.0		19.0[e]
				81.0		19.0[g]	81.0	19.0	0.0[d]
				100.0	0.0	0.0[h]			
Tetracycline	≤0.25	0.5	≤0.25 – >8	92.1	0.0	7.9	92.1	0.0	7.9
Trimethoprim-sulfamethoxazole	0.25	1	≤0.12 – >4	84.1	9.5	6.3	90.5	3.2	6.3
Vancomycin	0.25	0.25	0.12 – 0.5	100.0			100.0		0.0

[a] Criteria as published by CLSI [2017] and EUCAST [2017]
[b] Breakpoints from FDA package insert revised 12/2014
[c] Uncomplicated UTI only
[d] Using nonmeningitis breakpoints
[e] Using meningitis breakpoints
[f] Using oral breakpoints
[g] Using parenteral, meningitis breakpoints
[h] Using parenteral, nonmeningitis breakpoints

Table 3. Activity of ceftobiprole and comparator agents when tested against select Gram-negative bacteria from bloodstream infections (US; 2016)

Antimicrobial agent	MIC_{50}	MIC_{90}	Range	CLSI[a] %S	CLSI[a] %I	CLSI[a] %R	EUCAST[a] %S	EUCAST[a] %I	EUCAST[a] %R
***Enterobacteriaceae* (1,105)**									
Ceftobiprole	0.03	>16	0.015 – >16				85.0		15.0
Ampicillin-sulbactam	16	>32	0.5 – >32	48.8	17.6	33.7	48.8		51.2
Aztreonam	0.06	>16	≤0.03 – >16	86.6	1.3	12.1	84.5	2.1	13.4
Cefepime	≤0.12	8	≤0.12 – >16	88.9	1.9	9.2[b]	87.4	2.5	10.0
Ceftaroline	0.12	>32	≤0.015 – >32	77.9	3.9	18.2	77.9		22.1
Ceftazidime	0.25	16	0.03 – >32	87.2	0.9	11.9	84.9	2.4	12.8
Ceftriaxone	≤0.06	>8	≤0.06 – >8	83.8	0.9	15.3	83.8	0.9	15.3
Colistin	0.12	>8	≤0.06 – >8				87.4		12.6
Gentamicin	0.5	8	≤0.06 – >8	89.7	0.6	9.7	89.4	0.3	10.3
Imipenem	≤0.12	0.5	≤0.12 – >8	97.3	2.0	0.7	99.3	0.6	0.1
Levofloxacin	0.06	>4	≤0.03 – >4	80.2	2.1	17.8	76.7	2.3	21.0
Meropenem	≤0.015	0.06	≤0.015 – 16	99.4	0.2	0.5	99.5	0.4	0.1
Piperacillin-tazobactam	2	16	≤0.5 – >64	91.9	4.3	3.8	88.4	3.5	8.1
Tigecycline	0.25	0.5	≤0.06 – 8	98.1	1.7	0.2[c]	95.7	2.4	1.9
Trimethoprim-sulfamethoxazole	≤0.5	>4	≤0.5 – >4	75.1		24.9	75.1	0.8	24.1
***E. coli* (529)**									
Ceftobiprole	0.03	>16	0.015 – >16				84.7		15.3
Ampicillin-sulbactam	16	>32	0.5 – >32	44.2	23.1	32.7	44.2		55.8
Aztreonam	0.12	>16	≤0.03 – >16	85.1	1.7	13.2	83.6	1.5	14.9
Cefepime	≤0.12	>16	≤0.12 – >16	85.4	1.1	13.4[b]	84.7	0.9	14.4
Ceftaroline	0.12	>32	≤0.015 – >32	80.0	2.3	17.8	80.0		20.0
Ceftazidime	0.25	16	0.03 – >32	86.4	1.3	12.3	83.7	2.6	13.6
Ceftriaxone	≤0.06	>8	≤0.06 – >8	83.7	0.2	16.1	83.7	0.2	16.1
Colistin	0.12	0.25	≤0.06 – 8				99.4		0.6
Gentamicin	0.5	>8	0.12 – >8	86.2	0.0	13.8	86.0	0.2	13.8
Imipenem	≤0.12	≤0.12	≤0.12 – 4	99.8	0.0	0.2	99.8	0.2	0.0
Levofloxacin	0.06	>4	≤0.03 – >4	64.8	3.2	31.9	63.3	0.9	35.7
Meropenem	≤0.015	0.03	≤0.015 – 8	99.8	0.0	0.2	99.8	0.2	0.0
Piperacillin-tazobactam	2	8	≤0.5 – >64	94.0	3.0	3.0	91.1	2.8	6.0
Tigecycline	0.12	0.25	≤0.06 – 4	99.8	0.2	0.0[c]	99.8	0.0	0.2
Trimethoprim-sulfamethoxazole	≤0.5	>4	≤0.5 – >4	60.9		39.1	60.9	1.1	38.0
Non-ESBL-phenotype *E. coli* (440)									
Ceftobiprole	0.03	0.06	0.015 – 1				99.8		0.2
Ampicillin-sulbactam	8	>32	0.5 – >32	51.1	24.8	24.1	51.1		48.9
Aztreonam	0.06	0.25	≤0.03 – 1	100.0	0.0	0.0	100.0	0.0	0.0
Cefepime	≤0.12	≤0.12	≤0.12 – 2	100.0	0.0	0.0[b]	99.5	0.5	0.0
Ceftaroline	0.06	0.25	≤0.015 – 8	95.7	2.5	1.8	95.7		4.3
Ceftazidime	0.12	0.25	0.03 – 1	100.0	0.0	0.0	100.0	0.0	0.0
Ceftriaxone	≤0.06	0.12	≤0.06 – 0.5	100.0	0.0	0.0	100.0	0.0	0.0
Colistin	0.12	0.25	≤0.06 – 8				99.8		0.2
Gentamicin	0.5	2	0.12 – >8	91.1	0.0	8.9	90.9	0.2	8.9
Imipenem	≤0.12	≤0.12	≤0.12 – 0.5	100.0	0.0	0.0	100.0	0.0	0.0
Levofloxacin	≤0.03	>4	≤0.03 – >4	75.5	3.6	20.9	74.3	0.9	24.8
Meropenem	≤0.015	≤0.015	≤0.015 – 0.12	100.0	0.0	0.0	100.0	0.0	0.0
Piperacillin-tazobactam	2	4	≤0.5 – >64	97.3	1.4	1.4	96.8	0.5	2.7
Tigecycline	0.12	0.25	≤0.06 – 0.5	100.0	0.0	0.0[c]	100.0	0.0	0.0
Trimethoprim-sulfamethoxazole	≤0.5	>4	≤0.5 – >4	65.9		34.1	65.9	0.7	33.4
***Klebsiella pneumoniae* (268)**									
Ceftobiprole	0.03	>16	0.015 – >16				86.9		13.1
Ampicillin-sulbactam	8	>32	2 – >32	73.9	7.5	18.7	73.9		26.1
Aztreonam	0.06	16	≤0.03 – >16	89.2	0.7	10.1	86.6	2.6	10.8
Cefepime	≤0.12	4	≤0.12 – >16	89.6	2.2	8.2[b]	88.8	1.9	9.3
Ceftaroline	0.12	>32	≤0.015 – >32	82.1	3.4	14.6	82.1		17.9
Ceftazidime	0.12	16	0.06 – >32	88.8	1.1	10.1	86.2	2.6	11.2
Ceftriaxone	≤0.06	>8	≤0.06 – >8	86.9	0.7	12.3	86.9	0.7	12.3
Colistin	0.12	0.25	≤0.06 – 8				98.5		1.5
Gentamicin	0.25	2	0.12 – >8	90.3	1.1	8.6	90.3	0.0	9.7
Imipenem	≤0.12	0.25	≤0.12 – 8	99.6	0.0	0.4	99.6	0.4	0.0
Levofloxacin	0.06	1	≤0.03 – >4	93.6	1.9	4.5	89.1	3.7	7.1
Meropenem	0.03	0.03	≤0.015 – 16	99.6	0.0	0.4	99.6	0.0	0.4
Piperacillin-tazobactam	2	16	≤0.5 – >64	92.9	3.4	3.7	86.9	6.0	7.1
Tigecycline	0.25	1	≤0.06 – 4	98.5	1.5	0.0[c]	96.6	1.9	1.5
Trimethoprim-sulfamethoxazole	≤0.5	>4	≤0.5 – >4	85.8		14.2	85.8	0.0	14.2
Non-ESBL-phenotype *K. pneumoniae* (228)									
Ceftobiprole	0.03	0.06	0.015 – 0.25				100.0		0.0
Ampicillin-sulbactam	8	16	2 – >32	86.0	7.0	7.0	86.0		14.0
Aztreonam	≤0.03	0.12	≤0.03 – 0.5	100.0	0.0	0.0	100.0	0.0	0.0
Cefepime	≤0.12	≤0.12	≤0.12 – 1	100.0	0.0	0.0[b]	100.0	0.0	0.0
Ceftaroline	0.12	0.25	≤0.015 – 2	96.5	3.1	0.4	96.5		3.5
Ceftazidime	0.12	0.5	0.06 – 1	100.0	0.0	0.0	100.0	0.0	0.0
Ceftriaxone	≤0.06	0.12	≤0.06 – 1	100.0	0.0	0.0	100.0	0.0	0.0
Colistin	0.12	0.25	≤0.06 – 8				99.1		0.9
Gentamicin	0.25	0.5	0.12 – >8	99.1	0.0	0.9	99.1	0.0	0.9
Imipenem	≤0.12	0.25	≤0.12 – 1	100.0	0.0	0.0	100.0	0.0	0.0
Levofloxacin	0.06	0.25	≤0.03 – >4	98.7	0.4	0.9	96.5	1.3	2.2
Meropenem	0.03	0.03	≤0.015 – 0.25	100.0	0.0	0.0	100.0	0.0	0.0
Piperacillin-tazobactam	2	8	≤0.5 – >64	97.4	1.8	0.9	93.4	3.9	2.6
Tigecycline	0.25	0.5	0.12 – 4	99.1	0.9	0.0[c]	97.8	1.3	0.9
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	≤0.5 – >4	94.3		5.7	94.3	0.0	5.7
***Pseudomonas aeruginosa* (129)**									
Ceftobiprole	2	16	0.5 – >16						
Amikacin	4	8	0.5 – >32	98.4	0.0	1.6	93.8	4.7	1.6
Ampicillin-sulbactam	>32	>32	32 – >32						
Aztreonam	8	>16	1 – >16	81.4	3.1	15.5	1.6	82.9	15.5
Cefepime	2	16	0.5 – >16	85.3	10.9	3.9	85.3		14.7
Ceftaroline	16	>32	1 – >32						
Ceftazidime	2	16	0.5 – >32	89.1	1.6	9.3	89.1		10.9
Colistin	1	1	0.25 – 2	100.0		0.0	100.0		0.0
Gentamicin	2	8	0.25 – >8	88.4	6.2	5.4	88.4		11.6
Imipenem	1	8	≤0.12 – >8	72.9	6.2	20.9	79.1	13.2	7.8
Levofloxacin	0.5	>4	0.12 – >4	79.1	3.9	17.1	70.5		29.5
Piperacillin-tazobactam	4	32	1 – >64	86.0	6.2	7.8	86.0		14.0
Tigecycline	8	>8	1 – >8						
Trimethoprim-sulfamethoxazole	4	>4	≤0.5 – >4						
***Acinetobacter* spp. (41)**									
Ceftobiprole	0.5	>16	≤0.008 – >16						
Amikacin	1	>32	≤0.25 – >32	82.9	0.0	17.1	82.9	0.0	17.1
Ampicillin-sulbactam	2	>32	≤0.25 – >32	75.6	9.8	14.6			
Aztreonam	>16	>16	4 – >16						
Cefepime	4	>16	≤0.12 – >16	75.6	2.4	22.0			
Ceftaroline	2	>32	0.12 – >32						
Ceftazidime	4	>32	0.5 – >32	73.2	4.9	22.0			
Colistin	0.5	4	≤0.06 – >8	85.4		14.6	85.4		14.6
Gentamicin	0.5	>8	≤0.06 – >8	80.5	0.0	19.5	80.5		19.5
Imipenem	≤0.12	>8	≤0.12 – >8	82.9	0.0	17.1	82.9	0.0	17.1
Levofloxacin	0.12	>4	≤0.03 – >4	80.5	2.4	17.1	80.5	0.0	19.5
Piperacillin-tazobactam	2	>64	≤0.5 – >64	67.5	5.0	27.5			
Tigecycline	0.25	2	≤0.06 – 8						
Trimethoprim-sulfamethoxazole	≤0.5	>4	≤0.5 – >4	75.6		24.4	75.6	4.9	19.5

[a] Criteria as published by CLSI [2017] and EUCAST [2017]
[b] Intermediate interpreted as susceptible-dose dependent
[c] Breakpoints from FDA package insert revised 12/2014

Conclusions

- Ceftobiprole demonstrated potent *in vitro* activity against many Gram-positive and Gram-negative pathogens from BSI, including multidrug-resistant MRSA
- Ceftobiprole *in vitro* activity against Gram-negative pathogens, including *Enterobacteriaceae* and *P. aeruginosa,* was similar to other extended-spectrum cephalosporins, such as ceftazidime and cefepime
- Overall, the *in vitro* results of this 2016 surveillance study evaluating 2,787 bacterial pathogens isolated from patients at 30 medical centers across the US demonstrated the wide-spectrum nature of ceftobiprole activity

Acknowledgements

This study was supported by Basilea Pharmaceutica International Ltd. This project has been funded in whole or in part by federal funds from the Department of Health and Human Services; Office of the Assistant Secretary for Preparedness and Response; Biomedical Advanced Research and Development Authority, under Contract No. HHSO100201600002C with Basilea Pharmaceutica International Ltd.

References

Clinical and Laboratory Standards Institute (2015). *M07-A10. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard—tenth edition*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.

EUCAST (2017). Breakpoint tables for interpretation of MIC's and zone diameters. Version 7.1, March 2017. Available at http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_7.1_Breakpoint_Tables.pdf. Accessed March 2017.

Farrell DJ, Flamm RK, Sader HS, et al. (2014). Ceftobiprole activity against over 60,000 clinical bacterial pathogens isolated in Europe, Turkey, and Israel from 2005 to 2010. *Antimicrob Agents Chemother* 58: 3882-3888.

Farrell DJ, Flamm RK, Sader HS, et al. (2014). Activity of ceftobiprole against methicillin-resistant *Staphylococcus aureus* strains with reduced susceptibility to daptomycin, linezolid or vancomycin, and strains with defined SCC*mec* types. *Int J Antimicrob Agents* 43: 323-327.

Torres A, Mouton JW, Pea F (2016). Pharmacokinetics and dosing of ceftobiprole medocaril for the treatment of hospital- and community-acquired pneumonia in different patient populations. *Clin Pharmacokinet* 55: 1507-1520.