IDWeek 2017
Poster #1221

Antimicrobial Activity of Dalbavancin and Comparator Agents Tested against Gram-Positive Clinical Isolates Causing Bone and Joint Infections in United States Medical Centers (2011–2016)

Helio S. Sader, Rodrigo E. Mendes, Robert K. Flamm, Michael A. Pfaller

JMI Laboratories, North Liberty, Iowa, USA

Contact Information:
Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

To obtain a PDF of this poster:
- Scan the QR code
OR
- Visit www.allergancongressposters.com/168310

Charges may apply.
No personal information is stored.

INTRODUCTION

- Community-associated (CA) methicillin-resistant *Staphylococcus aureus* (CA-MRSA) and health care-associated (HA) MRSA as well as macrolide resistance among β-haemolytic streptococci (BHS) complicate therapy of osteomyelitis and other forms of bone and joint infections (BJI) in children and adults
- Dalbavancin belongs to the lipoglycopeptide class of antimicrobial agents that act by interrupting bacterial cell wall synthesis resulting in bacterial death, and it was approved in the United States (2014) and Europe (2015) to treat adults with acute bacterial skin and skin structure infections (ABSSSI) caused by susceptible isolates of *S. aureus*, including MRSA and methicillin-susceptible *S. aureus* (MSSA), *Streptococcus pyogenes*, *Streptococcus agalactiae*, *Streptococcus dysgalactiae*, *Streptococcus anginosus* group, and vancomycin-susceptible *Enterococcus faecalis*
- Dalbavancin allows for very convenient parenteral administration, which can be a single dose of 1500 mg or a dose of 1000 mg followed by 500 mg a week later for treating ABSSSI. Dalbavancin is not licensed for use in pediatric patients or for treating BJI, but is potentially important in treating infections due to highly resistant gram-positive cocci (GPC)
- The findings describe dalbavancin *in vitro* activity and potency when tested against a contemporary (2011–2016) collection of GPC isolates responsible for BJI, including osteomyelitis, recovered from adult and pediatric patients in United States (US) medical centers

MATERIALS AND METHODS

Bacterial isolates

- A total of 744 organisms were evaluated, including 463 *S. aureus*, 88 coagulase-negative staphylococci (CoNS), 104 BHS, 60 *E. faecalis*, and 29 viridans group streptococci (VGS)
- Isolates were collected from 55 US medical centers in 2011–2016
- Isolates were determined to be clinically significant based on local guidelines and submitted to a central monitoring laboratory (JMI Laboratories, North Liberty, Iowa, USA) as part of the International Dalbavancin Evaluation of Activity (IDEA) surveillance program
- Isolates were initially identified by the participating laboratory and bacterial identifications were confirmed by the reference monitoring laboratory by standard algorithms and supported by MALDI–TOF–MS (Bruker Daltonics, Bremen, Germany)

Antimicrobial susceptibility testing

- Isolates were tested for susceptibility by broth microdilution following guidelines in the CLSI M07–A10 document with testing performed using reference 96-well panels manufactured by JMI Laboratories
- Quality assurance was performed by concurrently testing CLSI–recommended QC reference strains (*S. aureus* ATCC 29213, *E. faecalis* ATCC 29212, and *Streptococcus pneumoniae* ATCC 49619), and all QC results were within published acceptable ranges
- The dalbavancin breakpoints approved by the Food and Drug Administration (FDA) for indicated species were applied (ie, ≤0.25 µg/mL), and breakpoint criteria for comparator agents were those from CLSI (M100-S27)

RESULTS

- *S. aureus* (62.2%) was the most common pathogen associated with BJI, followed by BHS (14.0%) and CoNS (11.8%; Figure 1)
- Dalbavancin inhibited the vast majority of organisms (94.1% of total) at ≤0.06 µg/mL (Figure 2)
- All *S. aureus* (41.5% MRSA) isolates were susceptible to dalbavancin, linezolid, teicoplanin, and vancomycin, while daptomycin and clindamycin showed susceptibility rates of 99.8% and 87.7% (CLSI), respectively (Table 1)
- Dalbavancin MIC results ($MIC_{50/90}$, ≤0.03/0.06 µg/mL) were ≥8-fold lower compared to daptomycin ($MIC_{50/90}$, 0.25/0.5 µg/mL) against all *S. aureus* (Table 1)
- Among CoNS (61.4% oxacillin-resistant), dalbavancin ($MIC_{50/90}$, ≤0.03/0.06 µg/mL) was the most potent agent, followed by daptomycin ($MIC_{50/90}$, 0.25/0.5 µg/mL), linezolid ($MIC_{50/90}$, 0.5/1 µg/mL), and vancomycin ($MIC_{50/90}$, 1/2 µg/mL; Table 1)

Figure 1. Frequency of occurrence of gram-positive organisms isolated from patients with bone and joint infections in US medical centers

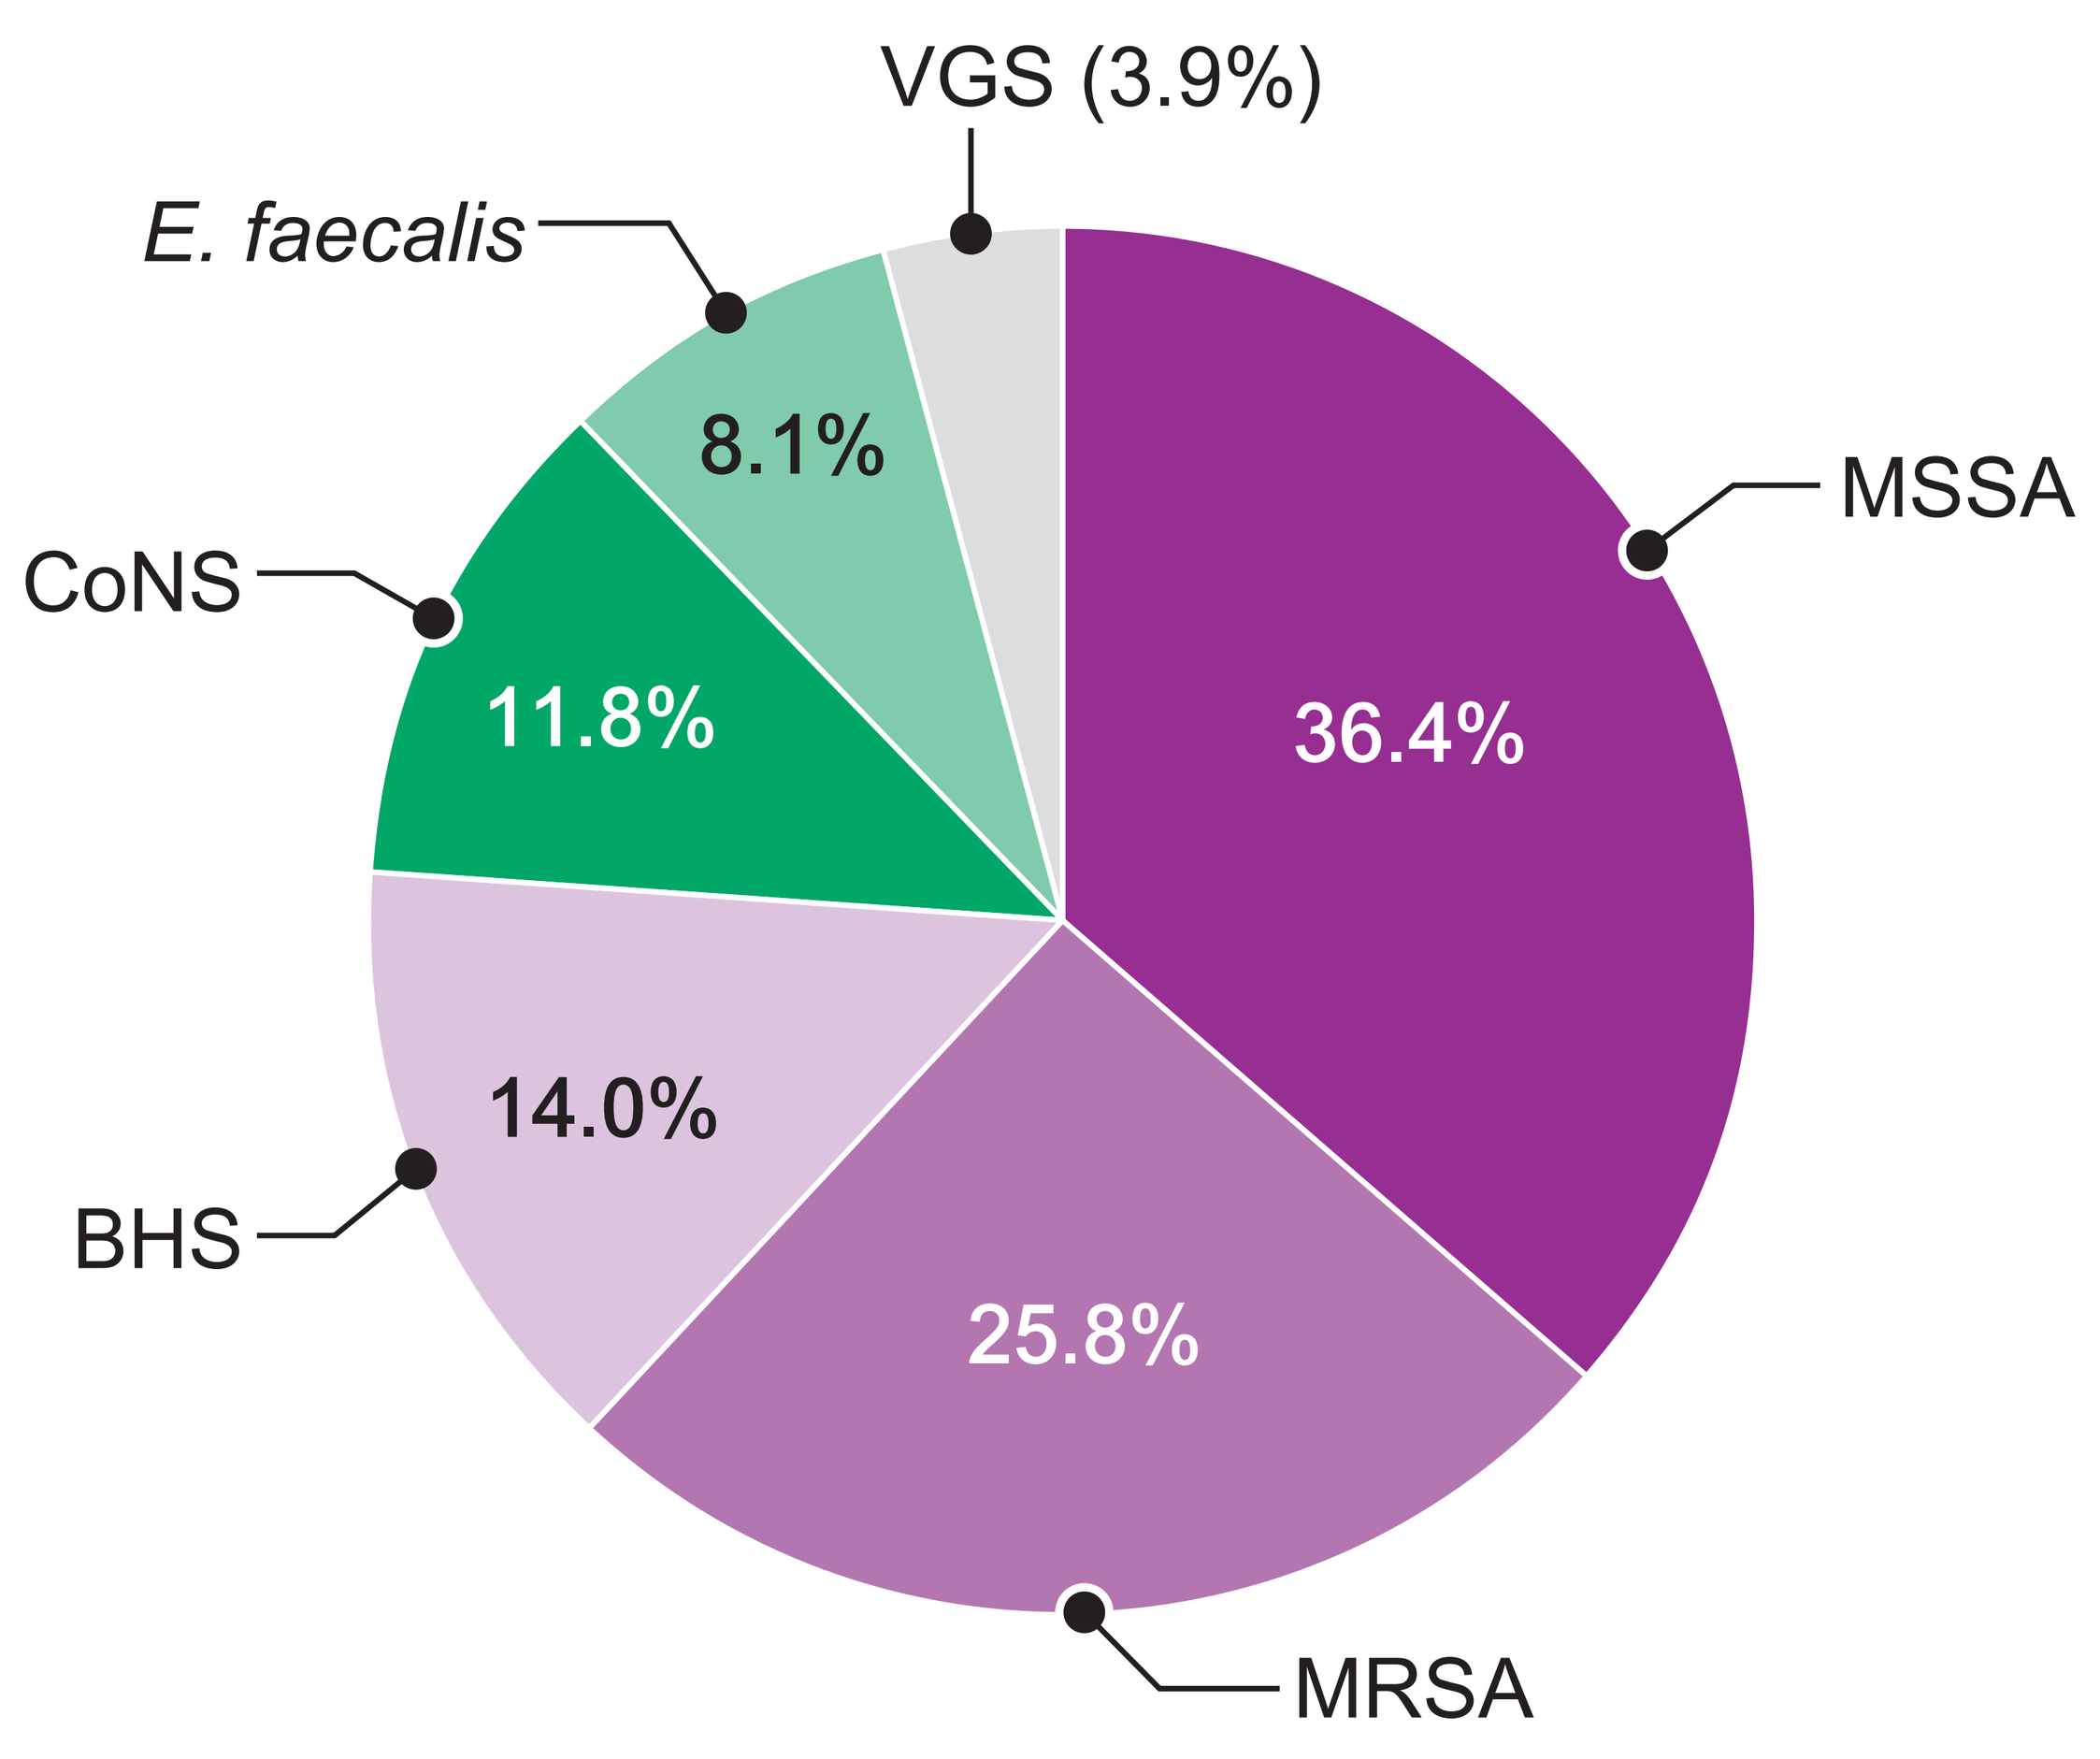

VGS (3.9%)
E. faecalis
8.1%
CoNS
11.8%
36.4%
MSSA
14.0%
BHS
25.8%
MRSA

Abbreviations: MSSA, methicillin-susceptible *S. aureus*; MRSA, methicillin-resistant *S. aureus*; BHS, β-haemolytic streptococci; CoNS, coagulase-negative staphylococci; and VGS, viridans group streptococci

Figure 2. Summary of dalbavancin activity against gram-positive organisms isolated from patients with bone and joint infections from US medical centers

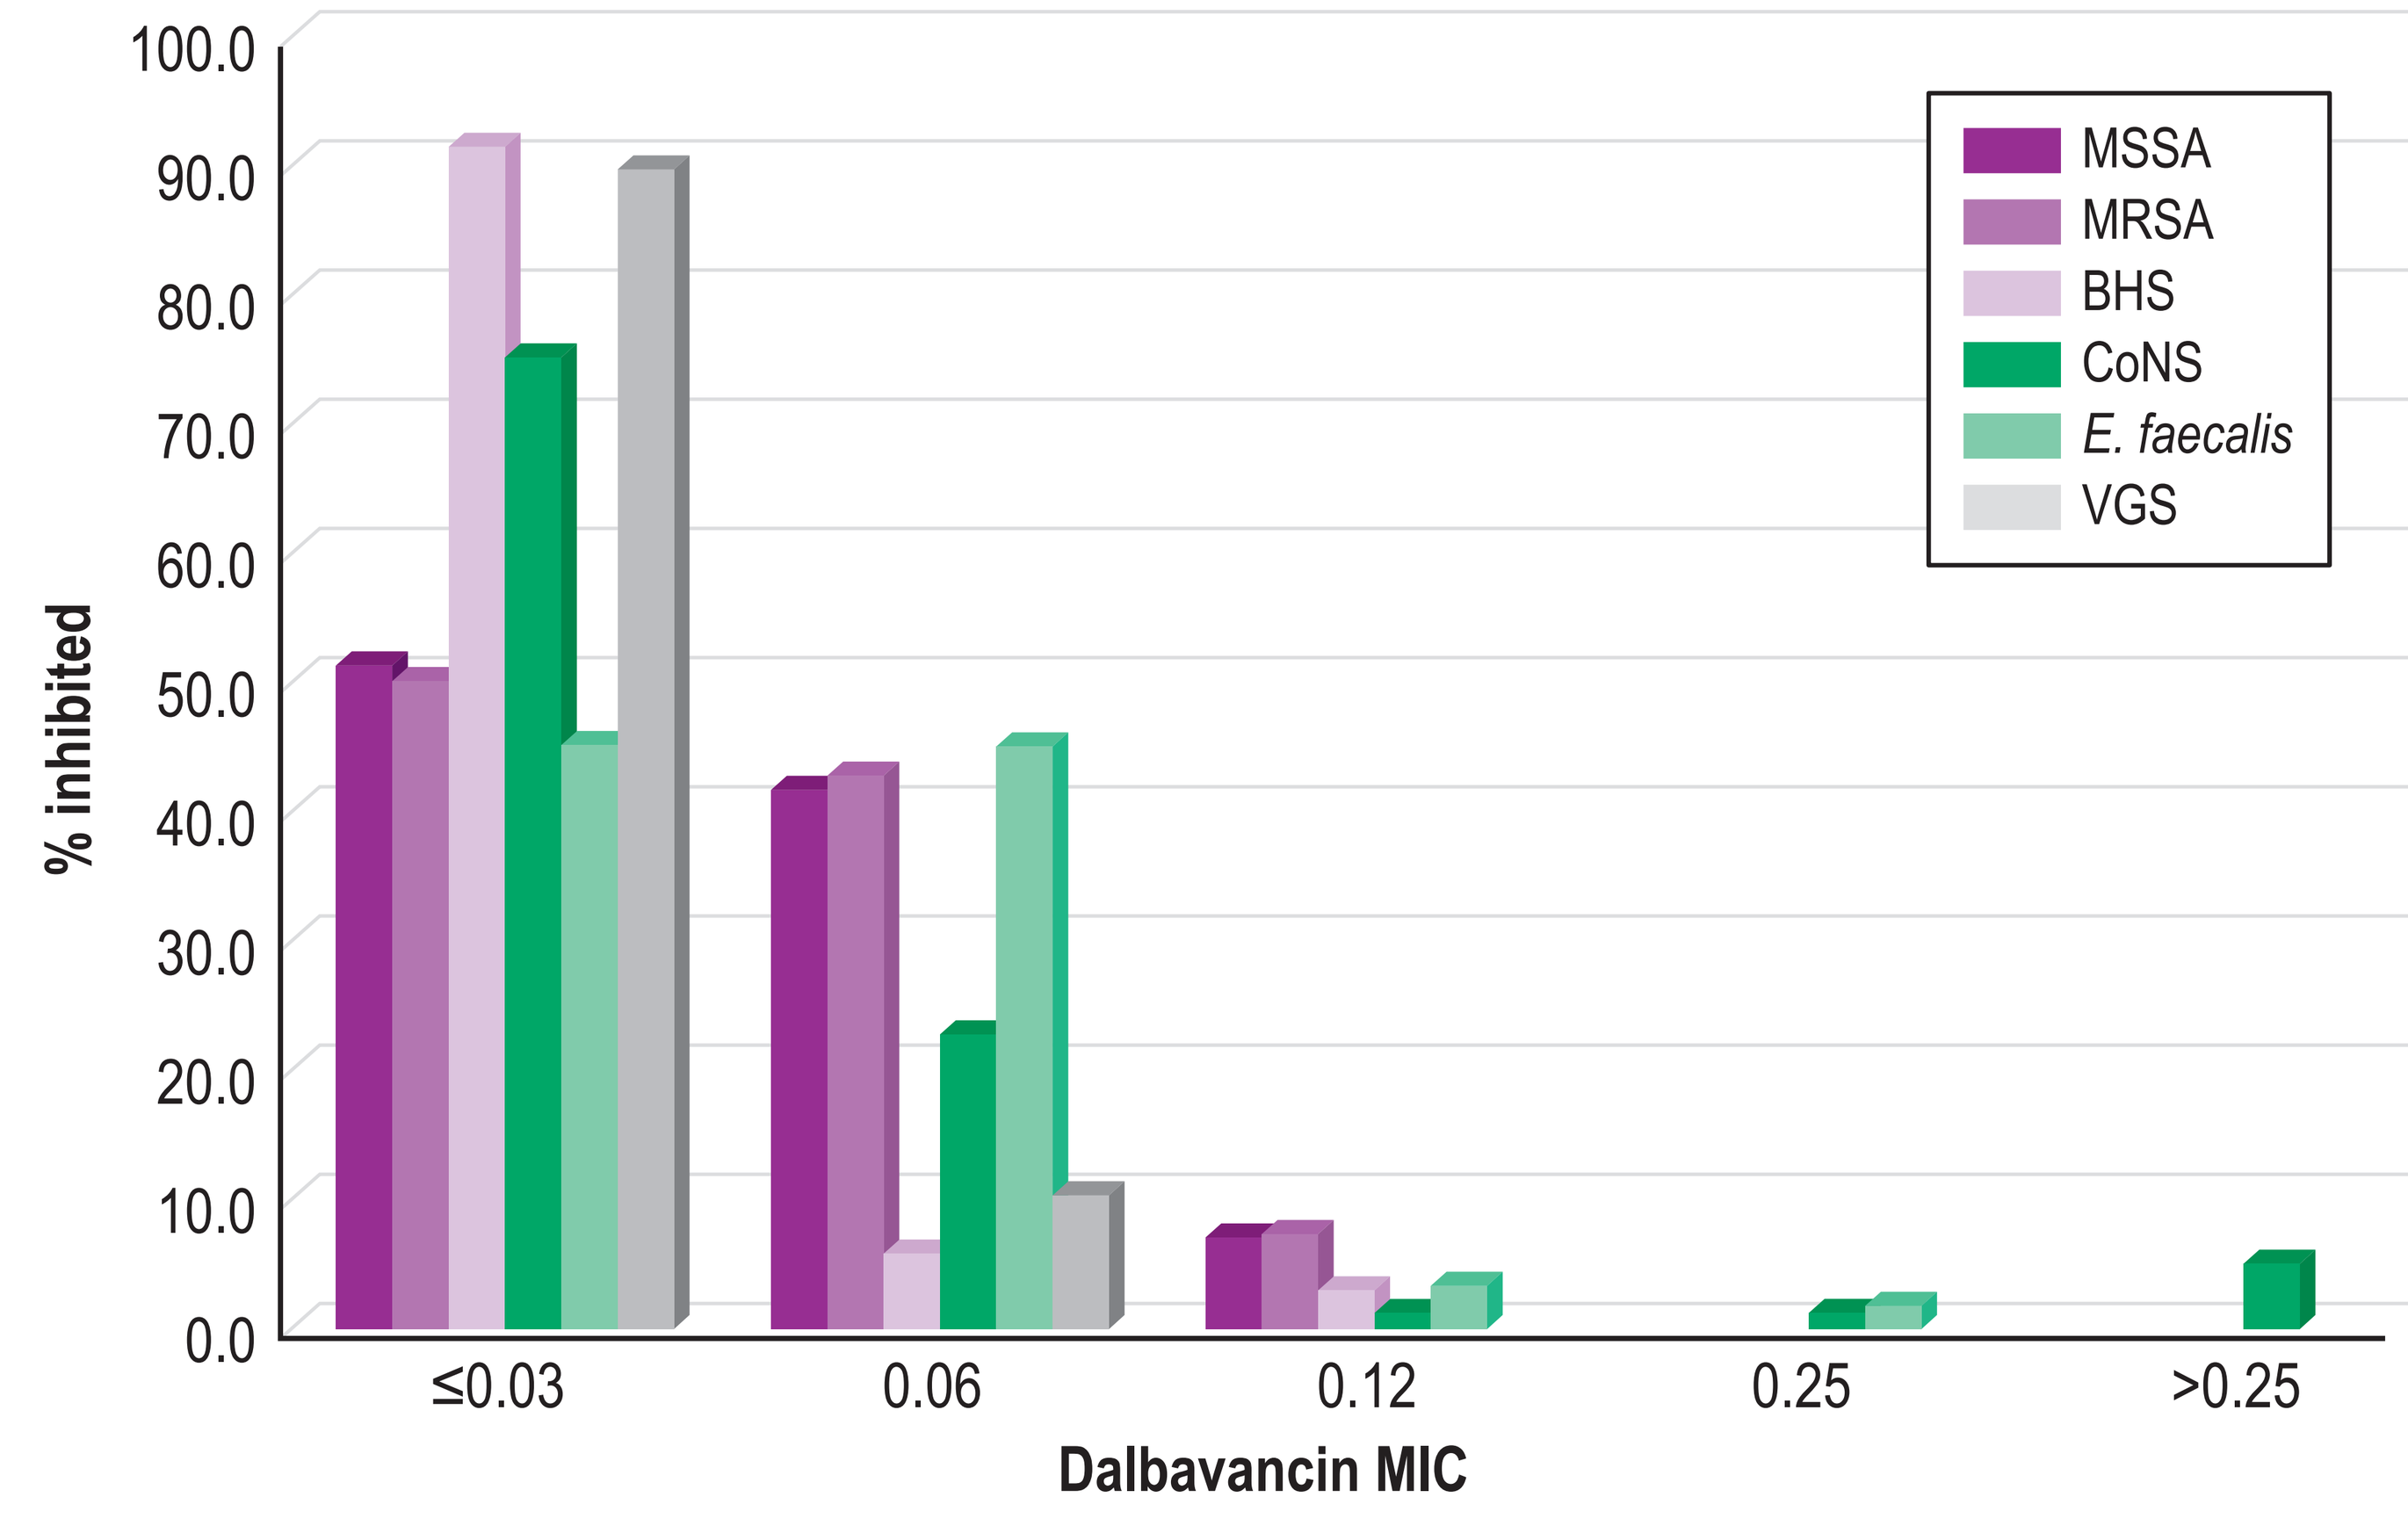

100.0
90.0
80.0
70.0
60.0
50.0
40.0
30.0
20.0
10.0
0.0
% inhibited
≤0.03
0.06
0.12
0.25
>0.25
Dalbavancin MIC
MSSA
MRSA
BHS
CoNS
E. faecalis
VGS

Abbreviations: MSSA, methicillin-susceptible *S. aureus*; MRSA, methicillin-resistant *S. aureus*; BHS, β-haemolytic streptococci; CoNS, coagulase-negative staphylococci; and VGS, viridans group streptococci

Table 1. Activity of dalbavancin and comparator antimicrobial agents when tested against gram-positive organisms isolated from patients with bone and joint infections from US medical centers

Antimicrobial agent	MIC_{50}	MIC_{90}	CLSI[a]	
			%S	%R
S. aureus (463)				
Dalbavancin	≤0.03	0.06	100.0[b]	
Daptomycin	0.25	0.5	99.8	
Teicoplanin	≤2	≤2	100.0	0.0
Vancomycin	1	1	100.0	0.0
Linezolid	1	1	100.0	0.0
Oxacillin	0.5	>2	58.5	41.5
Clindamycin	≤0.25	>2	87.7	12.3
Levofloxacin	0.25	>4	66.7	32.6
Tetracycline	≤0.5	≤0.5	95.2	4.3
TMP-SMX[c]	≤0.5	≤0.5	97.2	2.8
CoNS (88)[d]				
Dalbavancin	≤0.03	0.06		
Daptomycin	0.25	0.5	100.0	
Teicoplanin	≤2	4	100.0	0.0
Vancomycin	1	2	100.0	0.0
Linezolid	0.5	1	100.0	0.0
Oxacillin	2	>2	38.6	61.4
Clindamycin	≤0.25	>2	70.5	28.4
Levofloxacin	0.25	>4	59.1	36.4
Tetracycline	≤0.5	>8	84.1	14.8
TMP-SMX[c]	≤0.5	>4	69.3	30.7
E. faecalis (60)				
Dalbavancin	0.06	0.06	95.0[b]	
Daptomycin	1	1	100.0	
Teicoplanin	≤2	≤2	93.3	6.7
Vancomycin	1	2	93.3	6.7
Linezolid	1	1	100.0	0.0
Ampicillin	1	1	98.3	1.7
Levofloxacin	1	>4	71.7	28.3
Tetracycline	>8	>8	25.0	73.3
β-haemolytic streptococci (104)[e]				
Dalbavancin	≤0.03	≤0.03	100.0[b]	
Daptomycin	0.12	0.25	100.0	
Vancomycin	0.25	0.5	100.0	
Linezolid	1	1	100.0	
Penicillin	≤0.06	≤0.06	100.0	
Ceftriaxone	≤0.06	0.12	100.0	
Clindamycin	≤0.25	>2	76.7	23.3
Levofloxacin	0.5	1	98.1	1.9
Tetracycline	>8	>8	41.3	55.8
Viridans group streptococci (29)				
Dalbavancin	≤0.03	0.06	100.0[b]	
Daptomycin	0.5	1	100.0	
Vancomycin	0.5	1	100.0	
Linezolid	1	1	100.0	
Penicillin	≤0.06	2	79.3	3.4
Ceftriaxone	0.12	1	93.1	0.0
Clindamycin	≤0.25	>2	82.8	17.2
Levofloxacin	1	2	93.1	3.4
Tetracycline	1	>8	58.6	37.9

[a] Criteria as published by CLSI [2017].
[b] Breakpoints from FDA Package Insert applied for all *E. faecalis*, but approved for vancomycin-susceptible isolates only
[c] TMP-SMX, trimethoprim-sulfamethoxazole
[d] Organisms include *Staphylococcus capitis* (1), *S. caprae* (5), *S. cohnii* (1), *S. epidermidis* (49), *S. haemolyticus* (3), *S. hominis* (4), *S. lugdunensis* (16), *S. pettenkoferi* (1), *S. pseudintermedius* (1), *S. simulans* (2), *S. warneri* (3), unspeciated coagulase-negative staphylococci (2)
[e] Organisms include *Streptococcus agalactiae* (60), *S. dysgalactiae* (18), *S. pyogenes* (26)

- Dalbavancin inhibited 95.0% of *E. faecalis* isolates at ≤0.25 µg/mL (FDA susceptible breakpoint); only 3 vancomycin-resistant (VanA) isolates exhibited elevated dalbavancin MIC values with all 3 at >2 µg/mL (Figure 2 and Table 1)
- High susceptibility rates for ampicillin (98.3%; CLSI), daptomycin (100.0%), linezolid (100.0%), teicoplanin (93.3%), and vancomycin (93.3%) were obtained against *E. faecalis* (Table 1)
- Dalbavancin, daptomycin, linezolid, ceftriaxone, penicillin, and vancomycin were active against all BHS (100.0% susceptible; Table 1)
- Dalbavancin ($MIC_{50/90}$, ≤0.03/0.06 µg/mL; 100.0% susceptible) was the most active agent against VGS, inhibiting all isolates at ≤0.06 µg/mL (Table 1)
- Ceftriaxone, levofloxacin, linezolid, daptomycin, and vancomycin were also active against VGS (93.1%–100.0% susceptible per CLSI), whereas clindamycin (82.8% susceptible) had marginal activity (Table 1)

CONCLUSIONS

- *S. aureus* isolates were the most frequent pathogens responsible for BJI in this study population; a total of 41.5% of these isolates were methicillin-resistant, which precludes using both commonly used antimicrobial therapies, cefazolin and oxacillin, for such cases
- Dalbavancin demonstrated potent *in vitro* activity against common gram-positive isolates causing BJI in the US (2011–2016)
- This *in vitro* characteristic along with prolonged half-life and convenient administration make dalbavancin a promising candidate for treating BJI, including osteomyelitis caused by gram-positive cocci

ACKNOWLEDGEMENTS

This study was supported by Allergan. Allergan was involved in the design and decision to present these results, and JMI Laboratories received compensation for services in relation to preparing this presentation. Allergan had no involvement in the collection, analysis, or interpretation of data.

REFERENCES

1. Barnea Y, Lerner A, Aizic A, et al. (2016). Efficacy of dalbavancin in the treatment of MRSA rat sternal osteomyelitis with mediastinitis. *J Antimicrob Chemother* 71: 460-463.
2. Boucher HW, Wilcox M, Talbot GH, et al. (2014). Once-weekly dalbavancin versus daily conventional therapy for skin infection. *N Engl J Med* 370: 2169-2179.
3. Chiappini E, Mastrangelo G, Lazzeri S (2016). A case of acute osteomyelitis: An update on diagnosis and treatment. *Int J Environ Res Public Health* 13: E539.
4. Clinical and Laboratory Standards Institute (2015). *M07-A10. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard—tenth edition*. Wayne, PA: CLSI.
5. Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.
6. Dalvance™ Package Insert (2016). Available at http://www.allergan.com/assets/pdf/dalvance_pi. Accessed March 2017.
7. EUCAST (2017). Breakpoint tables for interpretation of MIC's and zone diameters. Version 7.1, March 2017. Available at http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_7.1_Breakpoint_Tables.pdf. Accessed March 2017.
8. Garnock-Jones KP (2017). Single-dose dalbavancin: A review in acute bacterial skin and skin structure infections. *Drugs* 77: 75-83.
9. Maffulli N, Papalia R, Zampogna B, et al. (2016). The management of osteomyelitis in the adult. *Surgeon* 14: 345-360.
10. Mendes RE, Castanheira M, Farrell DJ, et al. (2016). Update on dalbavancin activity tested against Gram-positive clinical isolates responsible for documented skin and skin-structure infections in US and European hospitals (2011-13). *J Antimicrob Chemother* 71: 276-278.

Allergan

IDWeek 2017, October 4–8, San Diego, California