IDWeek 2017
Poster #1235

Activity of Plazomicin against *Enterobacteriaceae* Isolates Collected in the United States Including Isolates Carrying Aminoglycoside-Modifying Enzymes Detected by Whole Genome Sequencing

M CASTANHEIRA[1], LM DESHPANDE[1], CM HUBLER[1], RE MENDES[1], AW SERIO[2], KM KRAUSE[2], RK FLAMM[1]
[1]JMI Laboratories, North Liberty, Iowa, USA
[2]Achaogen, Inc., South San Francisco, California, USA

Contact Information:
Mariana Castanheira, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: mariana-castanheira@jmilabs.com

To obtain a PDF of this poster:
- Scan the QR code
 OR
- Visit https://www.jmilabs.com/data/posters/IDWeek2017-plazomicin.WGS.pdf

Charges may apply.
No personal information is stored.

Abstract

Background: Plazomicin (PLZ) is a next-generation aminoglycoside (AMG) stable against aminoglycoside-modifying enzymes (AME) that completed Phase 3 studies for complicated urinary tract infections and serious infections due to carbapenem-resistant *Enterobacteriaceae* (ENT). We evaluated the activity of PLZ and AMGs against ENT collected in US hospitals during 2016.

Methods: A total of 2,097 ENT were susceptibility (S) tested by CLSI reference broth microdilution methods. *E. coli*, *Klebsiella* spp., *Enterobacter* spp., and *P. mirabilis* isolates displaying non-S MICs (CLSI criteria) for gentamicin (GEN), amikacin (AMK), and/or tobramycin (TOB) were submitted to WGS, *de novo* assembly and screening for AME genes.

Results: Against ENT, PLZ was more active than all 3 clinically available AMGs. PLZ and AMK activities were stable regardless of the infection type; however, differences were observed for GEN and TOB. Bloodstream isolates displayed higher GEN MICs when compared to the other infection sites. TOB activity varied 4-fold, being higher for bloodstream and pneumonia infections and lower for skin and skin structure tissue and other/unknown specimens. Against 198 isolates carrying 1 or more AME-encoding genes detected among 208 AMG-non-S isolates, PLZ activity was 8- to 16-fold greater when compared to the activity of AMK and at least 16-fold higher than the activity of GEN or TOB.

Conclusions: PLZ was active against ENT isolates from US hospitals regardless of infection type. PLZ displayed activity against isolates carrying AME genes that represent 12.0% of selected species. AME-carrying isolates were considerably more resistant to AMK, GEN, and TOB, highlighting the potential value of PLZ to treat infections caused by these organisms.

This project has been funded under BARDA Contract No. HHSO100201000046C.

Introduction

- Plazomicin is a semi-synthetic aminoglycoside derived from sisomicin and contains structural modifications that allow it to retain activity in the presence of the most prevalent aminoglycoside-modifying enzymes (AMEs)
- Aminoglycoside modification and inactivation mediated by AMEs are the most common resistance mechanisms to aminoglycoside agents in gram-positive and -negative bacteria
- AMEs are grouped according to the type of modification that they catalyze and are aminoglycoside acetyltransferases (AACs), nucleotidyltransferases, or adenyltransferases (ANTs) and phosphotransferases (APHs)
- We evaluated the activity of plazomicin and clinically available aminoglycosides against 2,097 *Enterobacteriaceae* isolates collected in United States (US) hospitals during 2016 and characterized the aminoglycoside resistance mechanisms among these isolates using whole genome sequencing analysis

Materials and Methods

- A total of 2,097 *Enterobacteriaceae* isolates deemed as cause of infection and limited to 1 per patient episode were included in the study
- Isolates collected during 2016 from 30 US hospitals were susceptibility tested using the reference broth microdilution method described by the Clinical and Laboratory Standards Institute (CLSI)
 - Categorical interpretations for all comparator agents were those found in CLSI criteria in M100-S27 (2017), EUCAST breakpoint tables (version 7.0, January 2017), and/or United States Food and Drug Administration (US FDA) package inserts
 - Quality control (QC) was performed according to CLSI guidelines (M07-A9), and all QC MIC results were within acceptable ranges as published in CLSI documents
- A total of 208 *Escherichia coli*, *Klebsiella* spp., *Proteus* spp., and *Enterobacter* spp. isolates displaying nonsusceptible MIC values for gentamicin, amikacin, and/or tobramycin according to CLSI criteria were screened for the presence of AME and 16S rRNA methylase encoding genes
 - Selected isolates were submitted to whole genome sequencing on a MiSeq (Illumina, San Diego, California, US) instrument targeting a 30X coverage
 - Sequences were *de novo* assembled and searched for the presence of AME and 16S rRNA methylase-encoding genes using a curated library and applying criteria of >94% sequencing identity and 40% minimum length coverage

Results

- Plazomicin ($MIC_{50/90}$, 0.5/1 µg/mL) inhibited 97.8% and 99.7% of the 2,097 *Enterobacteriaceae* isolates from US hospitals collected during 2016 at ≤2 µg/mL and ≤4 µg/mL, respectively (Figure 1)
 - Only 7 isolates displayed plazomicin MIC values >4 µg/mL (8–32 µg/mL): 3 *Morganella morganii*, 3 *Providencia stuartii,* and 1 *Providencia rettgeri*
- Applying CLSI/EUCAST breakpoints, gentamicin ($MIC_{50/90}$, 0.5/4 µg/mL) inhibited 90.4/89.9%, amikacin ($MIC_{50/90}$, 2/4 µg/mL) inhibited 99.3/98.0%, and tobramycin ($MIC_{50/90}$, 0.5/4 µg/mL) inhibited 90.1/87.3% of these isolates, respectively (data not shown)
- The activity of plazomicin (MIC_{50} ranges 0.25 to 0.5 µg/mL; MIC_{90} ranges 1 to 2 µg/mL) and amikacin were not affected by infection site; however, differences were observed for gentamicin and tobramycin (Figure 1)
 - Bloodstream isolates displayed a higher gentamicin MIC_{90} value (>8 µg/mL) when compared to the other infection sites (1–4 µg/mL)
 - Bloodstream and pneumonia isolates displayed higher tobramycin MIC_{90} values (8 µg/mL) when compared to skin and skin structure tissue and other/unknown specimens (MIC_{90}; 2 µg/mL)

Figure 1 Plazomicin and comparator aminoglycoside agents tested against *Enterobacteriaceae* isolates collected in US hospitals during 2016

A. All infection sites (2,097 tested)

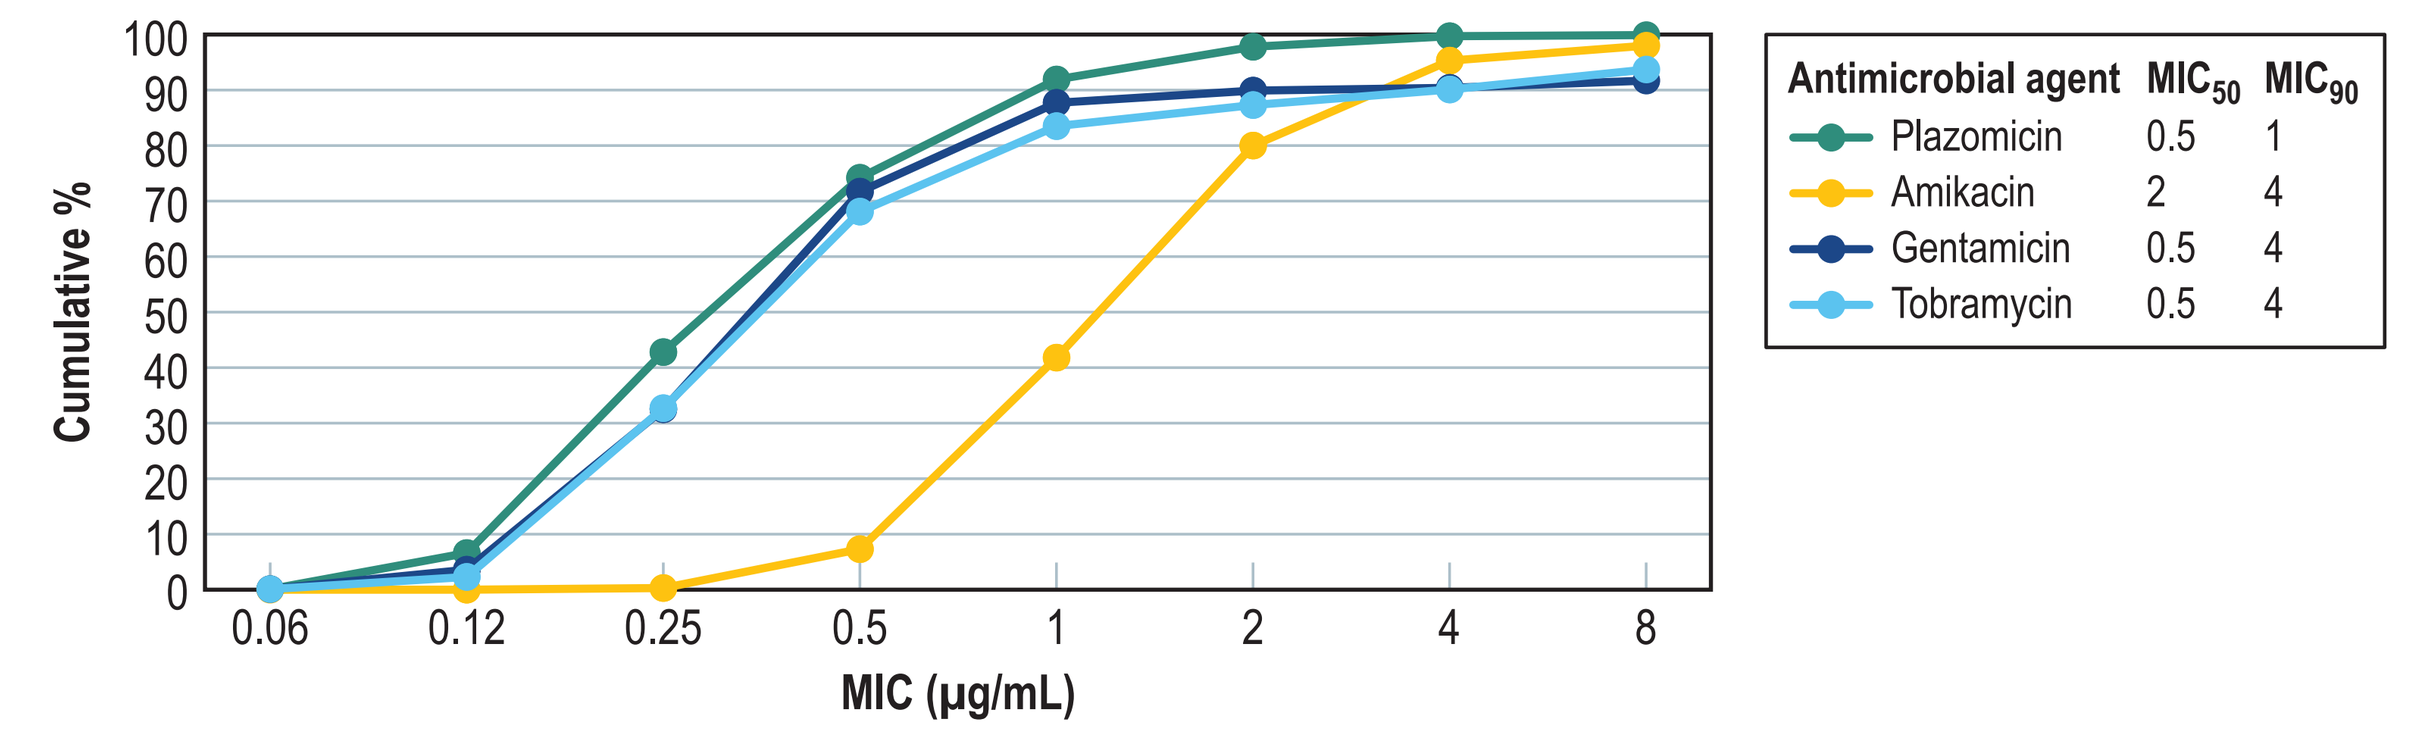
Cumulative %
MIC (µg/mL)
0.06 0.12 0.25 0.5 1 2 4 8
Antimicrobial agent MIC_{50} MIC_{90}
Plazomicin 0.5 1
Amikacin 2 4
Gentamicin 0.5 4
Tobramycin 0.5 4

B. Frequency distributions of plazomicin and comparator agents against *Enterobacteriaceae* isolates

	$MIC_{50/90}$ (µg/mL)			
Organism group/infection type (no. tested)	Plazomicin	Amikacin	Gentamicin	Tobramycin
Enterobacteriaceae (2,097)	0.5/1	2/4	0.5/4	0.5/4
Urinary tract infection (587)	0.5/1	2/4	0.5/2	0.5/4
Bloodstream infection (572)	0.5/1	2/4	0.5/>8	0.5/8
Pneumonia in hospitalized patients (451)	0.25/1	2/4	0.5/4	0.5/8
Skin and skin structure infection (298)	0.5/2	2/4	0.5/1	0.5/2
Intra-abdominal infection (152)	0.5/1	2/4	0.5/4	0.5/4
Other sites (37)	0.25/1	1/2	0.5/1	0.5/2
Isolates carrying AME genes (198)	0.5/1	4/16	>8/>8	>8/>8

Figure 2 AME genes that modify broadly used aminoglycosides detected among *Enterobacteriaceae* isolates collected in US hospitals during 2016

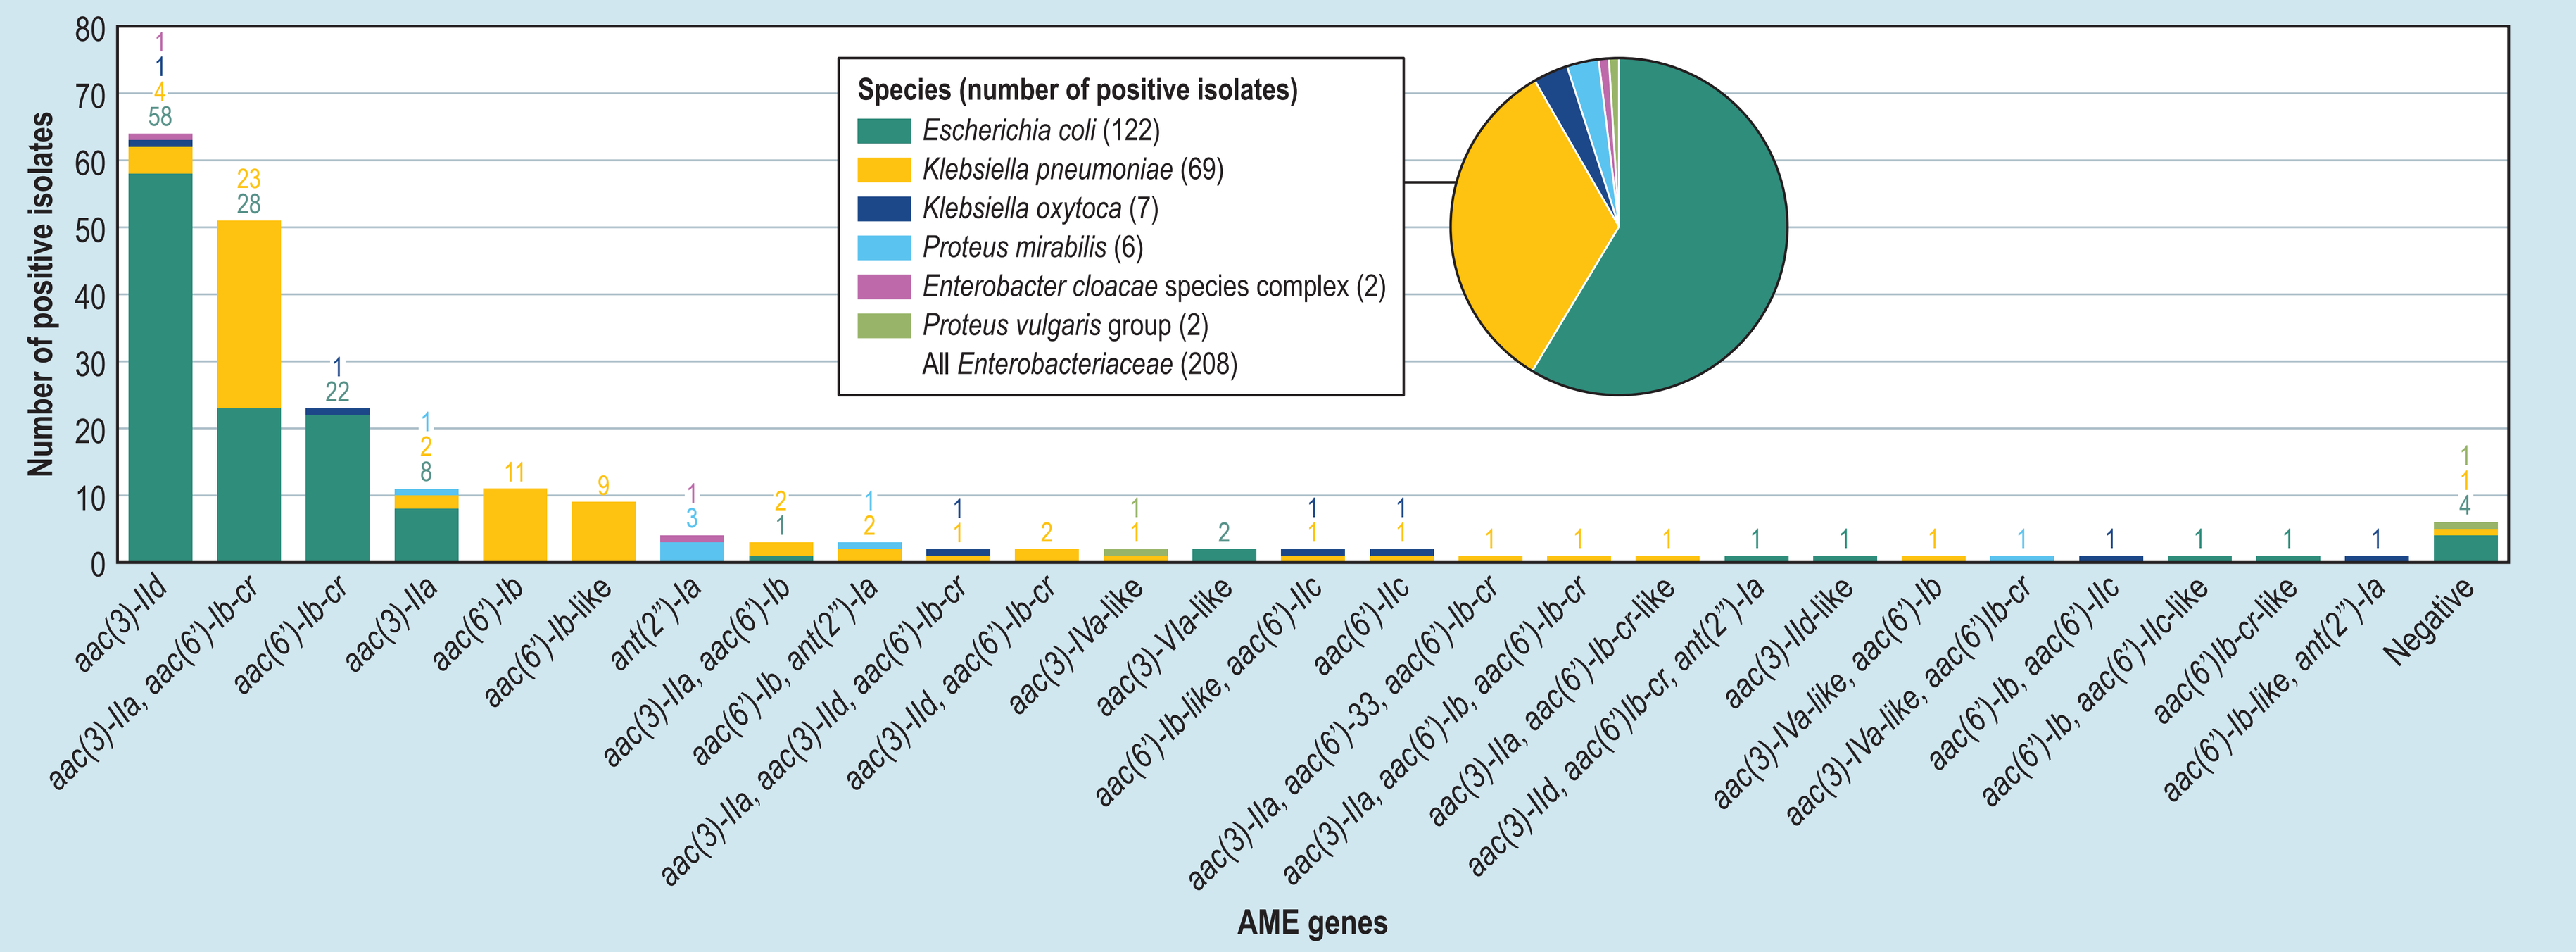
Number of positive isolates
AME genes
Species (number of positive isolates)
Escherichia coli (122)
Klebsiella pneumoniae (69)
Klebsiella oxytoca (7)
Proteus mirabilis (6)
Enterobacter cloacae species complex (2)
Proteus vulgaris group (2)
All Enterobacteriaceae (208)
aac(3)-IId
aac(3)-IIa, aac(6')-Ib-cr
aac(6')-Ib-cr
aac(3)-IIa
aac(6')-Ib
aac(6')-Ib-like
ant(2")-Ia
aac(3)-IIa, aac(6')-Ib
aac(6')-Ib, ant(2")-Ia
aac(3)-IIa, aac(3)-IId, aac(6')-Ib-cr
aac(3)-IId, aac(6')-Ib-cr
aac(3)-IVa-like
aac(3)-VIa-like
aac(6')-Ib-like, aac(6')-IIc
aac(6')-IIc
aac(3)-IIa, aac(6')-33, aac(6')-Ib-cr
aac(3)-IIa, aac(6')-Ib, aac(6')-Ib-cr
aac(3)-IIa, aac(6')-Ib-cr-like
aac(3)-IId, aac(6')Ib-cr, ant(2")-Ia
aac(3)-IId-like
aac(3)-IVa-like, aac(6')-Ib
aac(3)-IVa-like, aac(6')Ib-cr
aac(6')-Ib, aac(6')-IIc
aac(6')-Ib, aac(6')-IIc-like
aac(6')Ib-cr-like
aac(6')-Ib-like, ant(2")-Ia
Negative

- AME genes detected were among all 208 (12.0% of the species tested) isolates tested, and 202 isolates carried genes that are known to encode resistance to 1 or more broadly used aminoglycosides (amikacin, gentamicin, and tobramycin; Figure 2)
 - The most common genes detected were *aac(6')-Ib-cr* (82 isolates), *aac(3)-IIa* (70), *aac(3)-IId* (69), and *aac(6')-Ib* (21)
 - The combination *aac(3)-IIa* plus *aac(6')-Ib-cr* was observed among 51 isolates, and 14 other combinations of 2 or 3 of these genes were also observed (Figure 2)
- Plazomicin was very active against isolates carrying AME genes, and the highest plazomicin MIC against isolates carrying AME genes was 4 µg/mL (Figure 3)
 - Overall, plazomicin ($MIC_{50/90}$, 0.5/1 µg/mL) had 8- to 16-fold more potent activity when compared to amikacin ($MIC_{50/90}$, 4/16 µg/mL) and at least 16-fold more potent activity than gentamicin and tobramycin ($MIC_{50/90}$, >8/>8 µg/mL for both)
- The activity of amikacin, gentamicin, and tobramycin varied against isolates carrying different AME genes alone or in combination (Figures 3B to 3D)
- None of the isolates tested carried genes encoding 16S rRNA methylases

Conclusions

- Plazomicin was active against *Enterobacteriaceae* isolates from US hospitals regardless of infection type
- Plazomicin displayed activity against isolates carrying AME genes, including the genes that encode resistance to amikacin, gentamicin, and tobramycin
- Isolates carrying 16S rRNA methylases were not observed in this collection and remain rare in US hospitals
- AMEs were detected in approximately 10.0% of the overall isolates and 12.0% of the species tested
 - These isolates are common and usually resistant to the currently available aminoglycosides
 - Plazomicin displayed good activity against these isolates regardless of the gene or genes detected

Figure 3 Frequency distributions of plazomicin and comparator aminoglycoside agents tested against AME-carrying isolates

A. All AME-carrying isolates (208 detected)

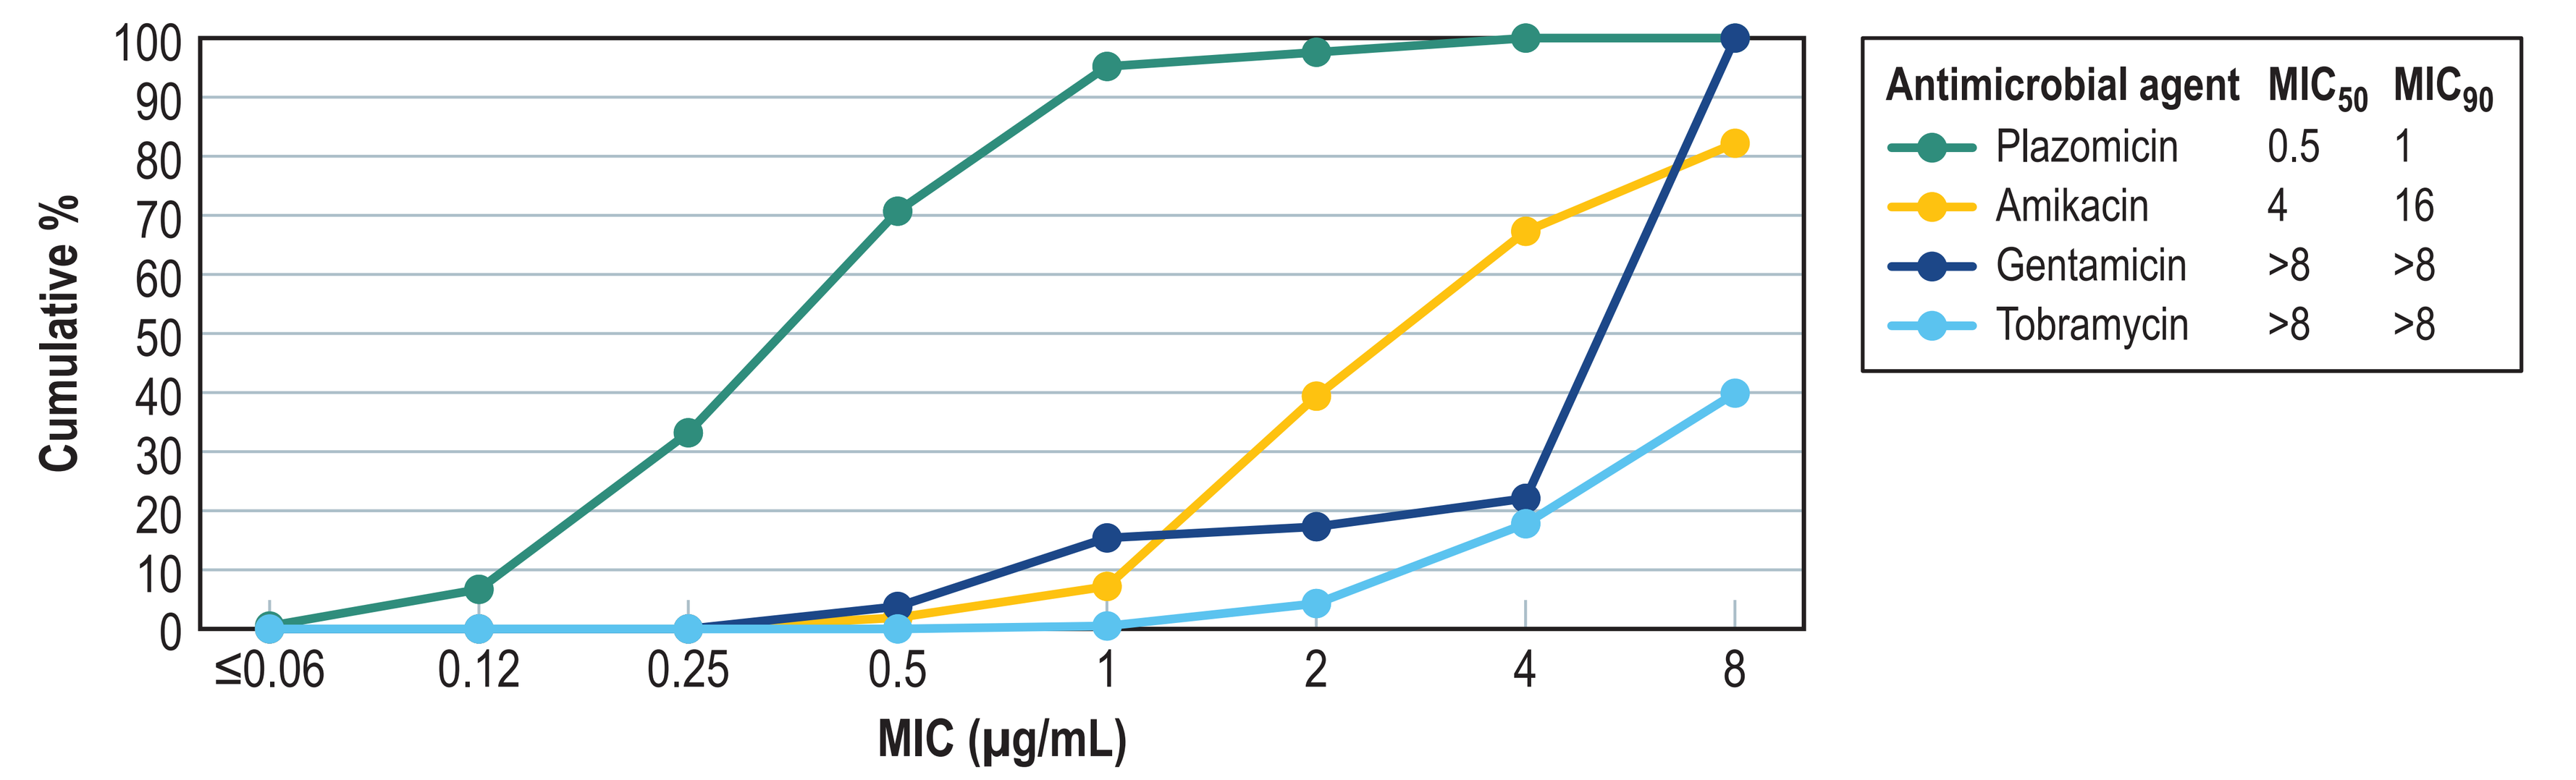
Cumulative %
MIC (µg/mL)
≤0.06 0.12 0.25 0.5 1 2 4 8
Antimicrobial agent MIC_{50} MIC_{90}
Plazomicin 0.5 1
Amikacin 4 16
Gentamicin >8 >8
Tobramycin >8 >8

B. *aac(3)-IId* (64 detected)

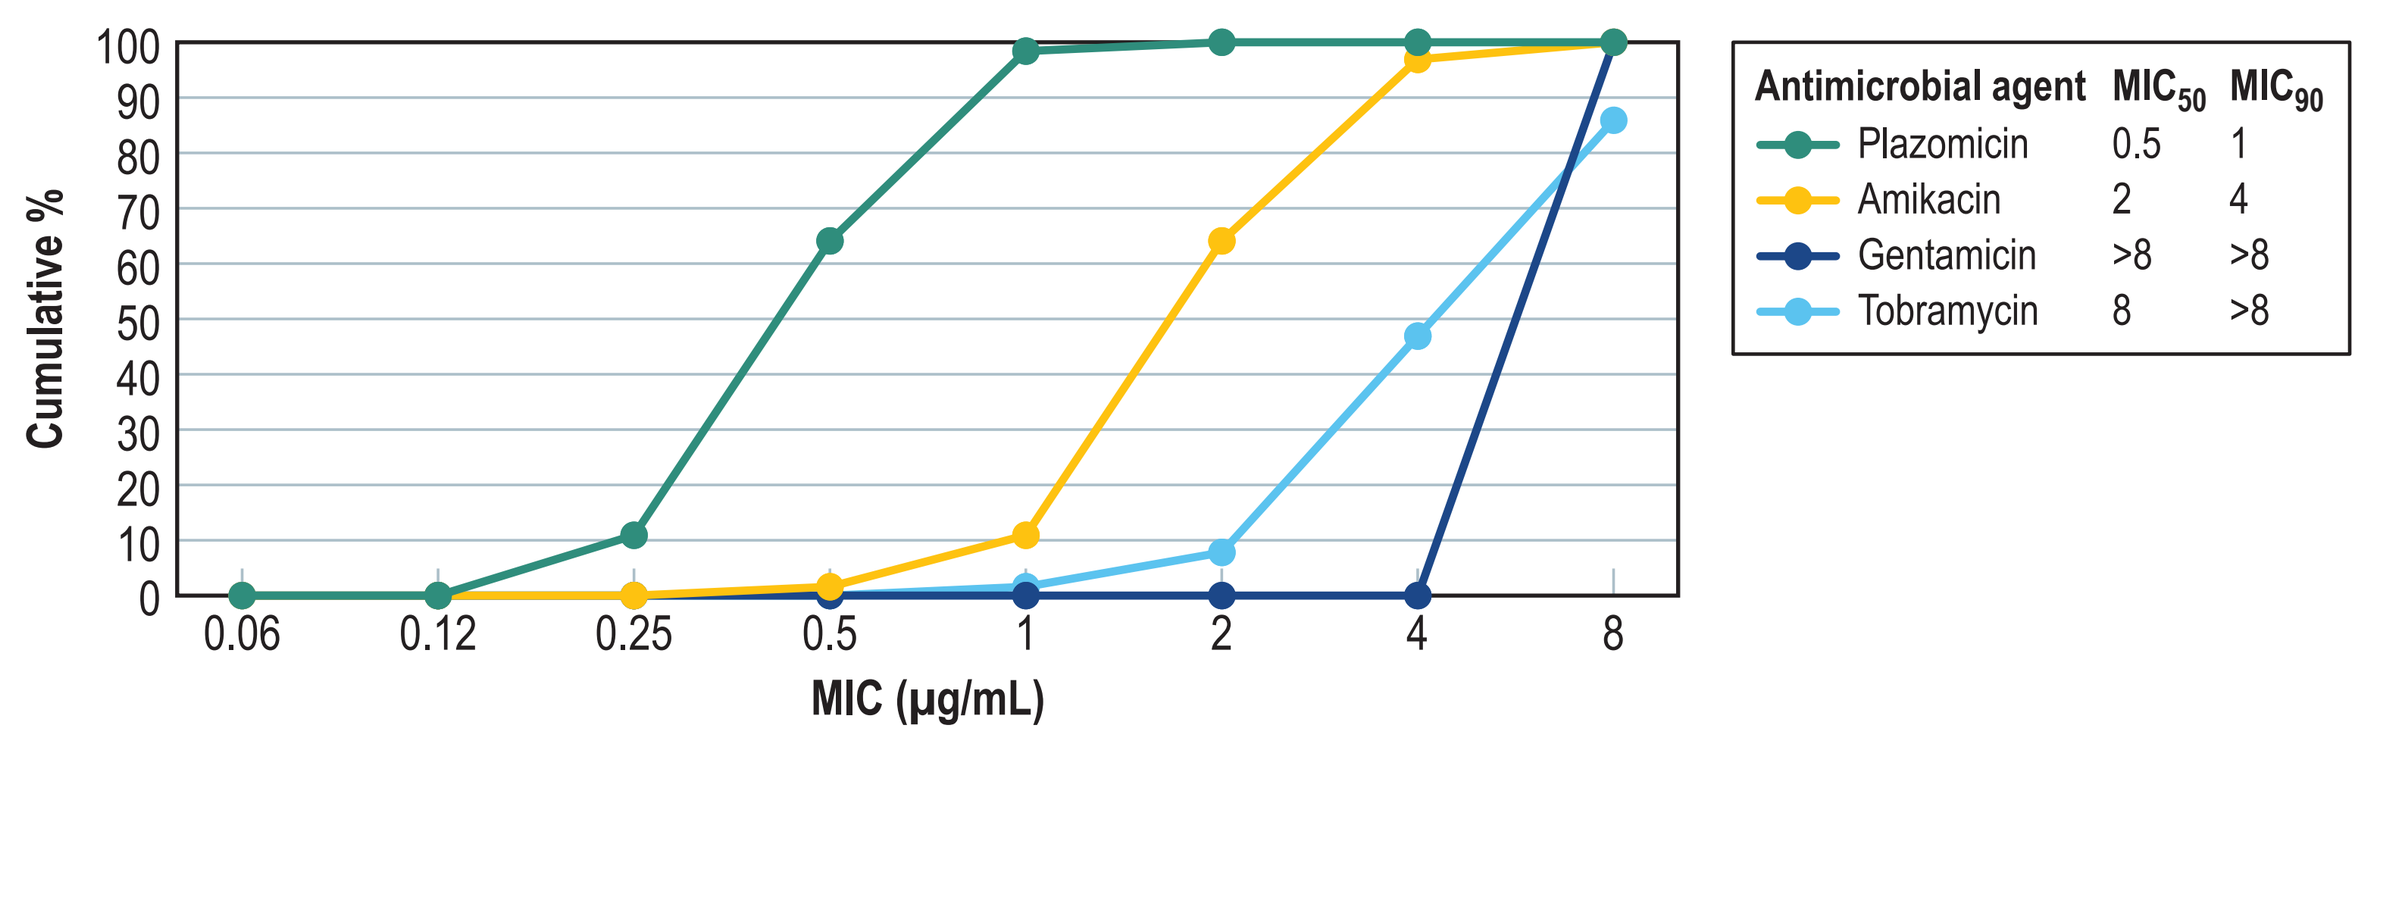
Cumulative %
MIC (µg/mL)
Antimicrobial agent MIC_{50} MIC_{90}
Plazomicin 0.5 1
Amikacin 2 4
Gentamicin >8 >8
Tobramycin 8 >8

C. *aac(3)-IIa, aac(6')-Ib-cr* (51 detected)

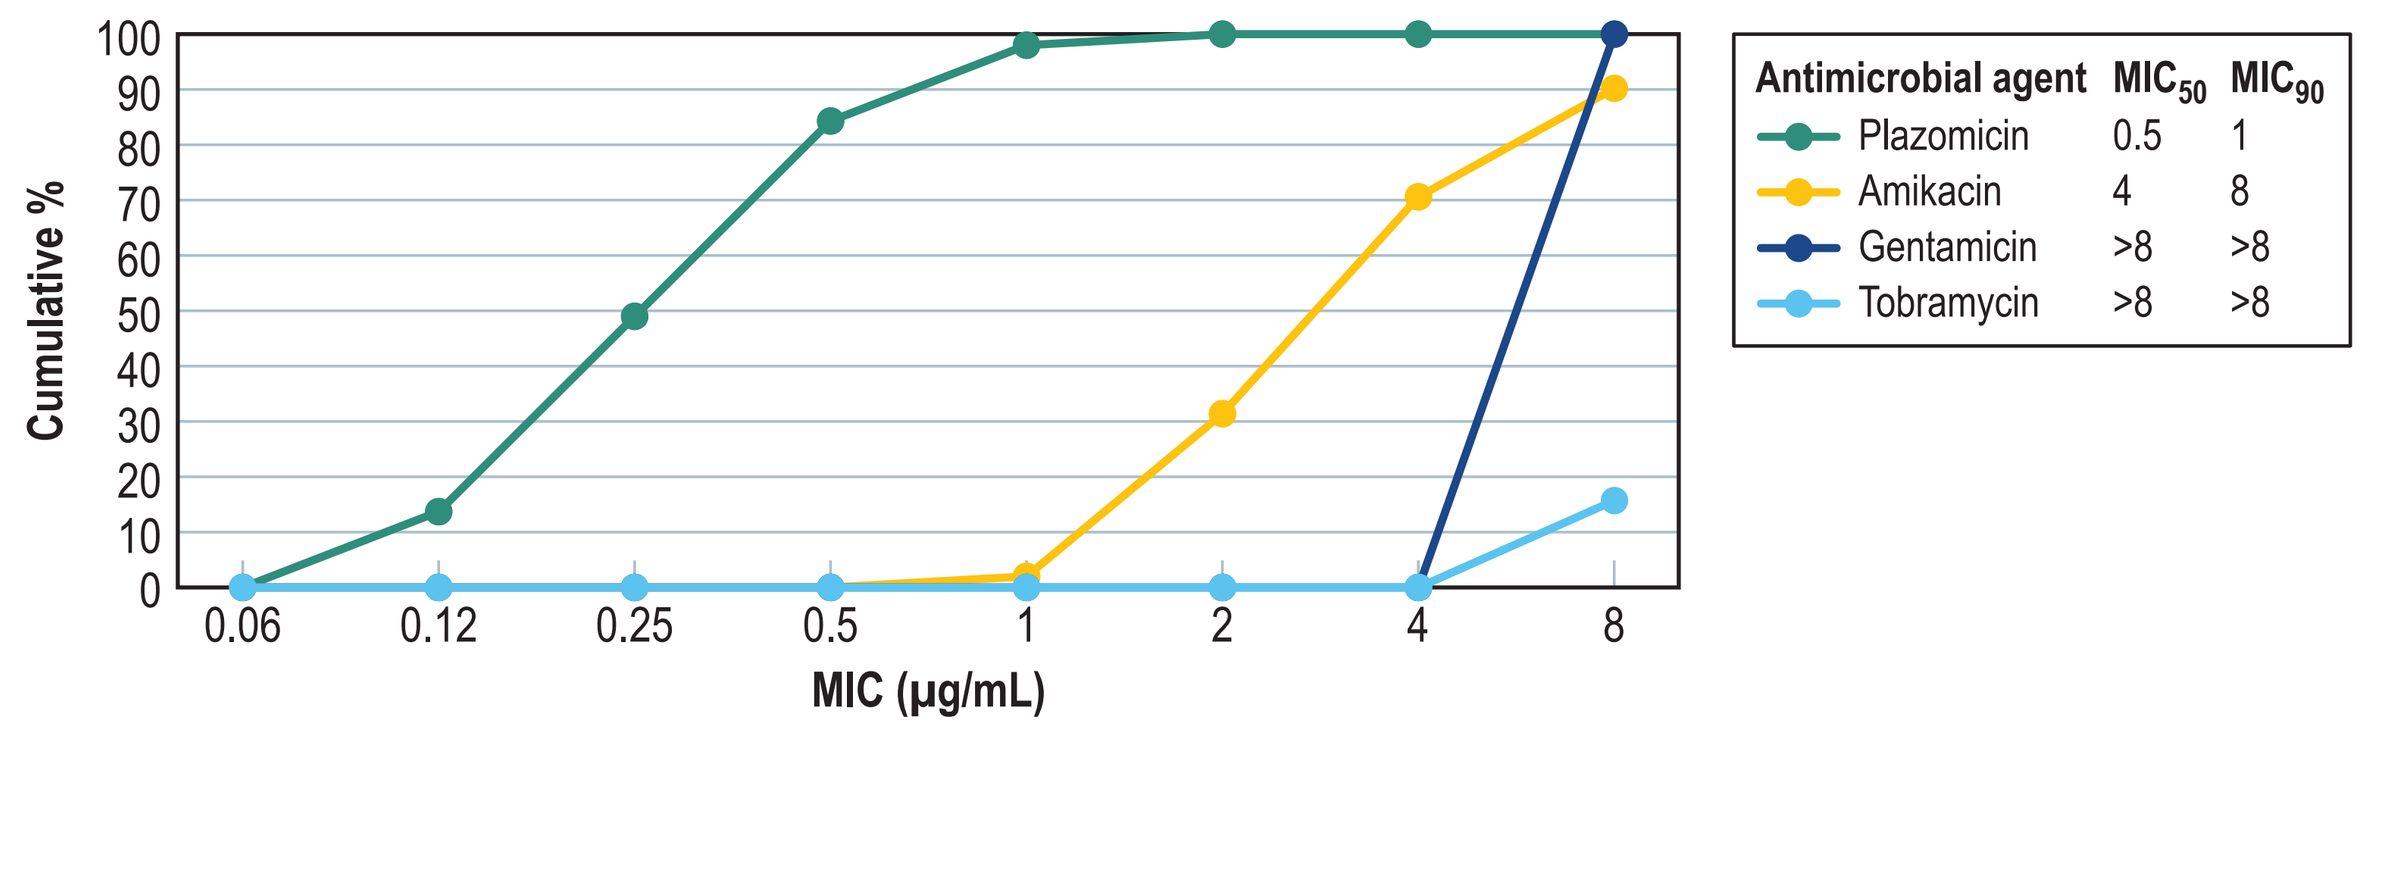
Cumulative %
MIC (µg/mL)
Antimicrobial agent MIC_{50} MIC_{90}
Plazomicin 0.5 1
Amikacin 4 8
Gentamicin >8 >8
Tobramycin >8 >8

D. *aac(6')Ib-cr* (23 detected)

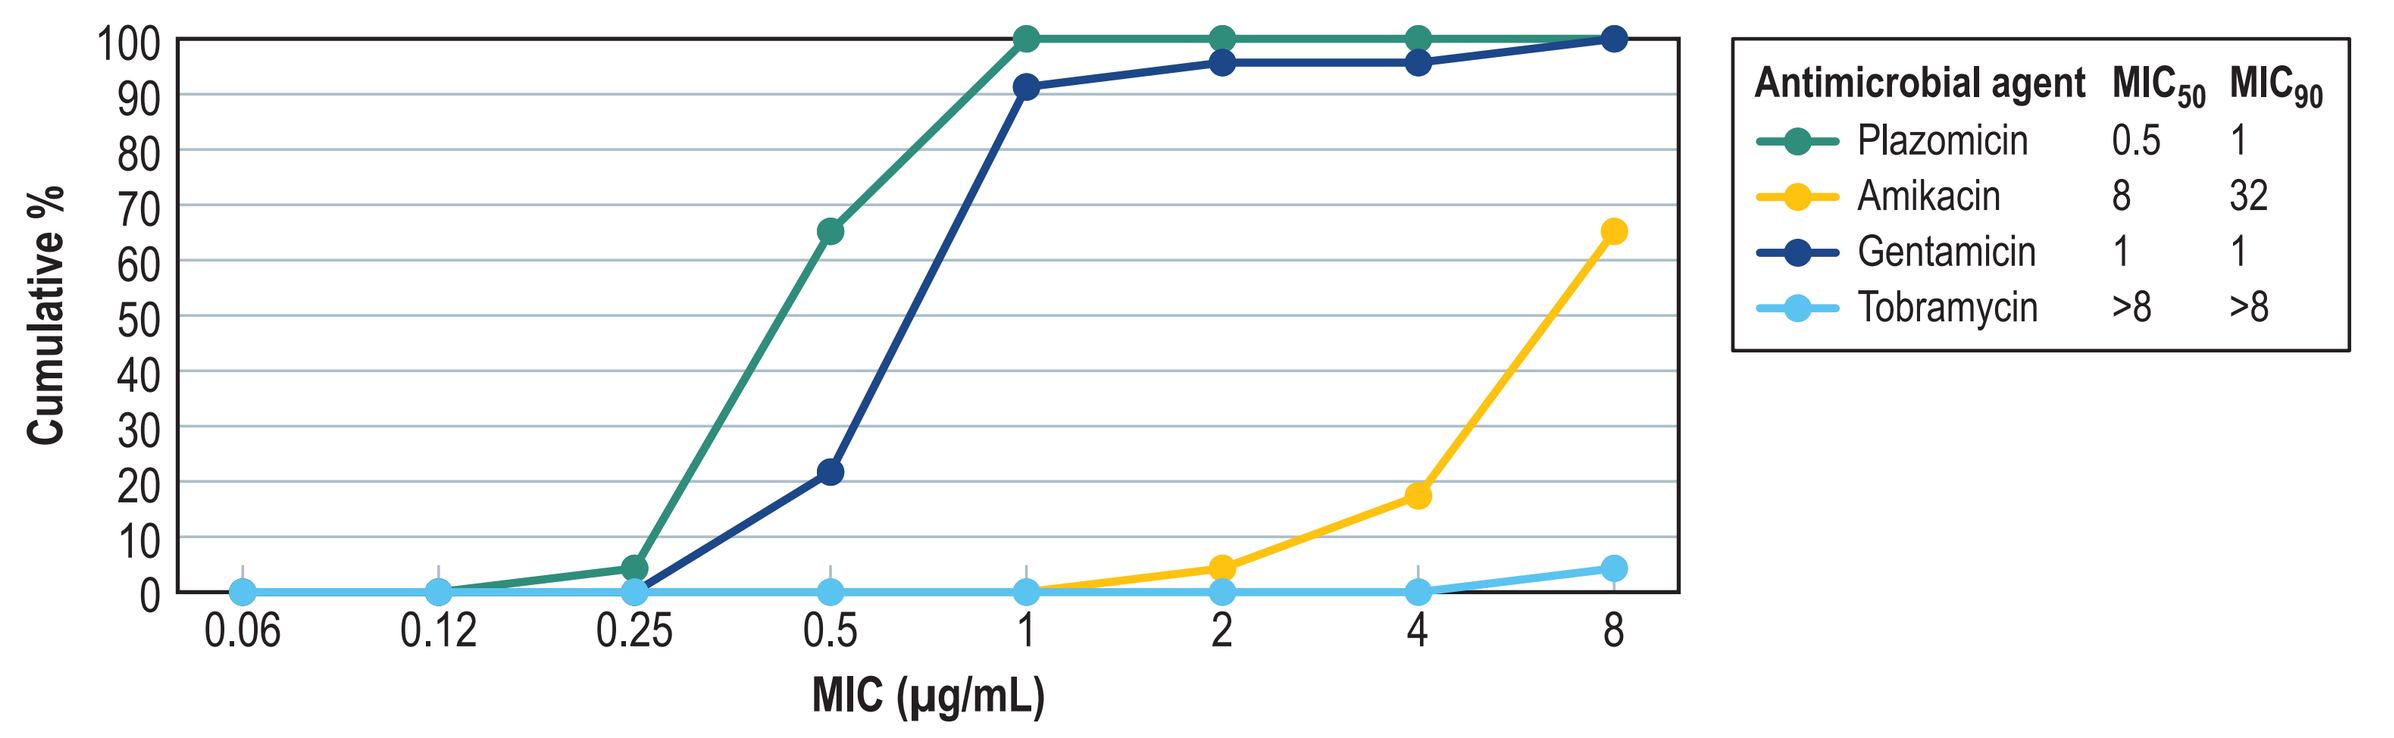
Cumulative %
MIC (µg/mL)
Antimicrobial agent MIC_{50} MIC_{90}
Plazomicin 0.5 1
Amikacin 8 32
Gentamicin 1 1
Tobramycin >8 >8

Acknowledgments

This study was performed by JMI Laboratories and supported by Achaogen, which included funding for services related to preparing this poster.

AW Serio and KM Krause are employees of Achaogen and contributed to the design of the study.

This project has been funded in whole or in part with Federal funds from the Biomedical Advanced Research and Development Authority, Office of the Assistant Secretary for Preparedness and Response, Office of the Secretary, Department of Health and Human Services, under Contract No. HHSO100201000046C.

References

Clinical and Laboratory Standards Institute (2017). *M100-S27. Performance standards for antimicrobial susceptibility testing: 27th informational supplement.* Wayne, PA: CLSI.

Doi Y, Wachino J, Arakawa Y (2016). Aminoglycoside resistance: The emergence of acquired 16S ribosomal RNA methyltransferases. *Infect Dis Clin North Am* 30: 523-537.

EUCAST (2017). Breakpoint tables for interpretation of MIC's and zone diameters. Version 7.1, March 2017. Available at: http://www.eucast.org/fileadmin/src/media/PDFs/EUCAST_files/Breakpoint_tables/v_7.1_Breakpoint_Tables.pdf. Accessed March 2017.

Ramirez MS, Tolmasky ME (2010). Aminoglycoside modifying enzymes. *Drug Resist Updat* 13: 151-171.

Walkty A, Adam H, Baxter M, et al. (2014). In vitro activity of plazomicin against 5,015 gram-negative and gram-positive clinical isolates obtained from patients in Canadian hospitals as part of the CANWARD study, 2011-2012. *Antimicrob Agents Chemother* 58: 2554-2563.