IDWeek 2019 | Poster #1445

Antimicrobial Activity of Novel β-Lactamase Inhibitor Combinations Tested against Organisms Causing Complicated Urinary Tract Infections in United States Medical Centers

Helio S. Sader, Cecilia G. Carvalhaes, Dee Shortridge, Michael D. Huband
JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Cefepime-tazobactam (WCK 4282) is under clinical development at 2g/2g, q8 hours dosage as a 90-minute infusion
- Cefepime was initially approved by the United States Food and Drug Administration (US FDA) in 1997
 - The clinical indications in the current US FDA product package insert include treatment for moderate to severe pneumonia, uncomplicated and complicated urinary tract infections (cUTIs), complicated intra-abdominal infections, and uncomplicated skin and skin structure infections, as well as empiric therapy for febrile neutropenic patients
- Cefepime dosages vary from 1g q12 hours to 2g q8 hours administered as a 30-minute infusion, but in WCK 4282 a cefepime dose of 2g is combined with a high tazobactam dose of 2g to be infused for a longer duration (90 minutes)
- The cefepime-tazobactam combination demonstrated activity against several isolates producing extended-spectrum β-lactamases (ESBLs) and AmpC β-lactamases, including those resistant to piperacillin-tazobactam, cefoperazone-sulbactam, and ceftolozane-tazobactam
- We evaluated the potency and spectrum of activity of cefepime-tazobactam, ceftolozane-tazobactam, ceftazidime-avibactam, and comparators against Gram-negative bacilli causing cUTIs in US hospitals

Materials and Methods

- A total of 3,023 Gram-negative organisms, including 2,841 *Enterobacterales*, 165 *Pseudomonas aeruginosa*, and 17 other non-fermentative Gram-negative bacilli (1/patient) were consecutively collected in 2018 as part of a worldwide surveillance program
- Susceptibility testing against cefepime-tazobactam (tazobactam at fixed 8 mg/L), ceftolozane-tazobactam (tazobactam at fixed 4 mg/L), ceftazidime-avibactam (avibactam at fixed 4 mg/L), and comparators was performed by reference broth microdilution method
- Percentages of isolates inhibited at cefepime ≤8 mg/L (Clinical and Laboratory Standards Institute [CLSI], cefepime high-dose breakpoint) and at ≤16 mg/L (pharmacokinetic/pharmacodynamic [PK/PD]-susceptible breakpoint based on high-dose extended infusion) in presence of tazobactam were evaluated
- CLSI breakpoints were applied for comparators and for categorizing the resistant subsets

Results

- Cefepime-tazobactam ($MIC_{50/90}$, 0.03/0.06 mg/L) and ceftazidime-avibactam ($MIC_{50/90}$, 0.12/0.25 mg/L) exhibited potent activity against *Escherichia coli* with 100% susceptibility (Table 1)
- The most active agents against *Klebsiella pneumoniae* were cefepime-tazobactam (99.2% and 99.6% inhibited at ≤8 mg/L and ≤16 mg/L, respectively), ceftazidime-avibactam (100.0% susceptible [S]), and amikacin (99.4%S)
 - MIC_{90} values of cefepime-tazobactam against *K. pneumoniae* were 8 times lower than those of ceftolozane-tazobactam (Table 1)
- All *Proteus mirabilis* isolates were susceptible to cefepime-tazobactam (highest MIC, 0.12 mg/L), ceftolozane-tazobactam, ceftazidime-avibactam, meropenem, piperacillin-tazobactam, and amikacin (data not shown)
- Cefepime-tazobactam was highly active against *Enterobacter cloacae* (n=94; MIC_{90}, 0.5 mg/L; 98.9% inhibited at ≤16 mg/L) and *Citrobacter* spp. (n=91; MIC_{90}, 0.12 mg/L; highest MIC, 0.5 mg/L; data not shown)
- Cefepime-tazobactam activity against *Enterobacterales* was greater than cefepime alone
 - Overall, cefepime-tazobactam (99.8% and 99.9% inhibited at ≤8 mg/L and ≤16 mg/L, respectively), ceftazidime-avibactam (99.9%S) and meropenem (99.5%S) were the most active agents against *Enterobacterales* (data not shown)
- When tested against ESBL-producing *Enterobacterales*, cefepime-tazobactam inhibited 98.8% and 99.6% of isolates at ≤8 mg/L and 16 mg/L, respectively
 - Ceftazidime-avibactam and ceftolozane-tazobactam were active against 99.6% and 87.7% of isolates, respectively (Table 1 and Figure 1)
- Against *P. aeruginosa*, cefepime-tazobactam inhibited 90.9% and 97.6% of isolates at ≤8 mg/L and ≤16 mg/L, respectively (Table 1 and Figure 2)
- The cefepime-tazobactam spectrum of activity ($MIC_{50/90}$, 1/8 mg/L; 90.9% and 97.6% of isolates inhibited at ≤8 mg/L and ≤16 mg/L, respectively) was similar to ceftazidime-avibactam ($MIC_{50/90}$, 2/4 mg/L; 96.4%S), ceftolozane-tazobactam ($MIC_{50/90}$, 1/2 mg/L; 99.4%S), and tobramycin ($MIC_{50/90}$, 0.5/2 mg/L; 93.3%S; Table 1 and Figure 2)
- Cefepime-tazobactam (90.9% and 97.6% of isolates inhibited at ≤8 mg/L and ≤16 mg/L, respectively) exhibited greater anti-pseudomonal spectrum than meropenem ($MIC_{50/90}$, 0.5/8 mg/L; 85.5%S) and piperacillin-tazobactam ($MIC_{50/90}$, 4/32 mg/L; 87.3%S; Table 1 and Figure 2)

Table 1. Activity of cefepime-tazobactam and comparator antimicrobial agents when tested against *Enterobacterales* and *P. aeruginosa* collected from medical centers in the United States during 2018

Antimicrobial agent	mg/L		%S[a]	%R[a]
	MIC_{50}	MIC_{90}		
E. coli (1,814)				
Cefepime-tazobactam	0.03	0.06	[100.0/100.0][b]	
Ceftolozane-tazobactam	0.25	0.5	99.0	0.7
Piperacillin-tazobactam	2	4	97.6	0.6
Ceftazidime-avibactam	0.12	0.25	100.0	0.0
Ampicillin-sulbactam	8	64	55.8	28.3
Cefepime	0.06	2	90.2	7.8
Ceftriaxone	≤0.06	>8	88.6	11.2
Ceftazidime	0.25	4	91.2	7.3
Meropenem	≤0.015	0.03	99.8	0.1
Ertapenem	0.008	0.015	99.5	0.2
Gentamicin	0.5	2	90.5	9.3
Amikacin	2	4	99.8	0.1
Levofloxacin	0.03	16	74.0	25.1
K. pneumoniae (473)				
Cefepime-tazobactam	0.03	0.12	[99.2/99.6][b]	
Ceftolozane-tazobactam	0.25	1	97.6	1.8
Piperacillin-tazobactam	2	16	93.9	3.2
Ceftazidime-avibactam	0.12	0.25	100.0	0.0
Ampicillin-sulbactam	8	64	71.9	18.8
Cefepime	0.06	4	89.9	9.1
Ceftriaxone	≤0.06	>8	88.4	11.0
Ceftazidime	0.25	16	88.6	10.8
Meropenem	0.03	0.03	98.3	1.1
Ertapenem	0.008	0.03	98.3	1.7
Gentamicin	0.25	0.5	93.2	5.7
Amikacin	1	2	99.4	0.2
Levofloxacin	0.06	1	87.7	7.4
Other *Enterobacterales* (554)[c]				
Cefepime-tazobactam	0.06	0.12	[99.5/99.6][b]	
Ceftolozane-tazobactam	0.5	1	93.1	4.8
Piperacillin-tazobactam	2	16	91.7	4.9
Ceftazidime-avibactam	0.12	0.25	99.6	0.4
Ampicillin-sulbactam	16	64	49.9	34.9
Cefepime	0.06	0.5	95.9	2.2
Ceftriaxone	0.12	>8	84.8	13.5
Ceftazidime	0.12	8	89.4	9.7
Meropenem	0.03	0.06	99.6	0.4
Ertapenem	0.015	0.12	97.5	0.7
Gentamicin	0.5	1	95.3	3.4
Amikacin	2	4	99.8	0.2
Levofloxacin	0.06	1	87.5	10.0
ESBL-producing *Enterobacterales* (252)[d]				
Cefepime-tazobactam	0.06	0.5	[98.8/99.6][b]	
Ceftolozane-tazobactam	0.5	4	87.7	7.9
Piperacillin-tazobactam	4	64	82.1	7.9
Ceftazidime-avibactam	0.12	0.5	99.6	0.4
Ampicillin-sulbactam	32	>64	15.9	69.4
Cefepime	64	>256	9.5	74.6
Ceftriaxone	>8	>8	3.2	96.0
Ceftazidime	16	>32	21.0	69.0
Meropenem	0.03	0.06	96.0	2.4
Ertapenem	0.03	0.5	94.0	4.0
Gentamicin	1	>16	61.9	35.7
Amikacin	4	8	98.8	0.4
Levofloxacin	8	32	19.9	75.3
P. aeruginosa (165)				
Cefepime-tazobactam	2	8	[90.9/97.6][b]	
Ceftolozane-tazobactam	1	2	99.4	0.6
Piperacillin-tazobactam	4	32	87.3	5.5
Ceftazidime-avibactam	2	4	96.4	3.6
Cefepime	2	8	90.3	3.0
Ceftazidime	2	8	90.9	5.5
Meropenem	0.5	8	85.5	10.9
Amikacin	4	8	97.6	0.6
Tobramycin	0.5	2	93.3	6.7
Levofloxacin	0.5	32	68.3	22.0

ESBL, extended-spectrum β-lactamase
[a] Criteria as published by CLSI (2019).
[b] Percentage of isolates inhibited at cefepime-tazobactam of ≤8/16 mg/L in brackets for comparison purpose only.
[c] Organisms include: *Citrobacter amalonaticus/farmeri* (3), *C. freundii* species complex (52), *C. koseri* (36), *Enterobacter cloacae* species complex (94), *Hafnia alvei* (4), *Klebsiella aerogenes* (43), *K. oxytoca* (66), *K. variicola* (2), *Morganella morganii* (26), *Proteus hauseri* (1), *P. mirabilis* (165), *P. penneri* (1), *P. vulgaris* group (6), *Providencia alcalifaciens* (1), *P. rettgeri* (13), *P. stuartii* (9), *Raoultella ornithinolytica* (1), *Serratia marcescens* (23), unspeciated *Pantoea* (1), unspeciated *Providencia* (2), unspeciated *Raoultella* (5).
[d] Organisms include: *Citrobacter freundii* species complex (2), *Enterobacter cloacae* species complex (7), *Escherichia coli* (184), *Klebsiella oxytoca* (5), *K. pneumoniae* (53), *Proteus mirabilis* (1).

Figure 1. Antimicrobial susceptibility of 252 ESBL-producing *Enterobacterales* isolates from complicated urinary tract infections collected worldwide in 2018

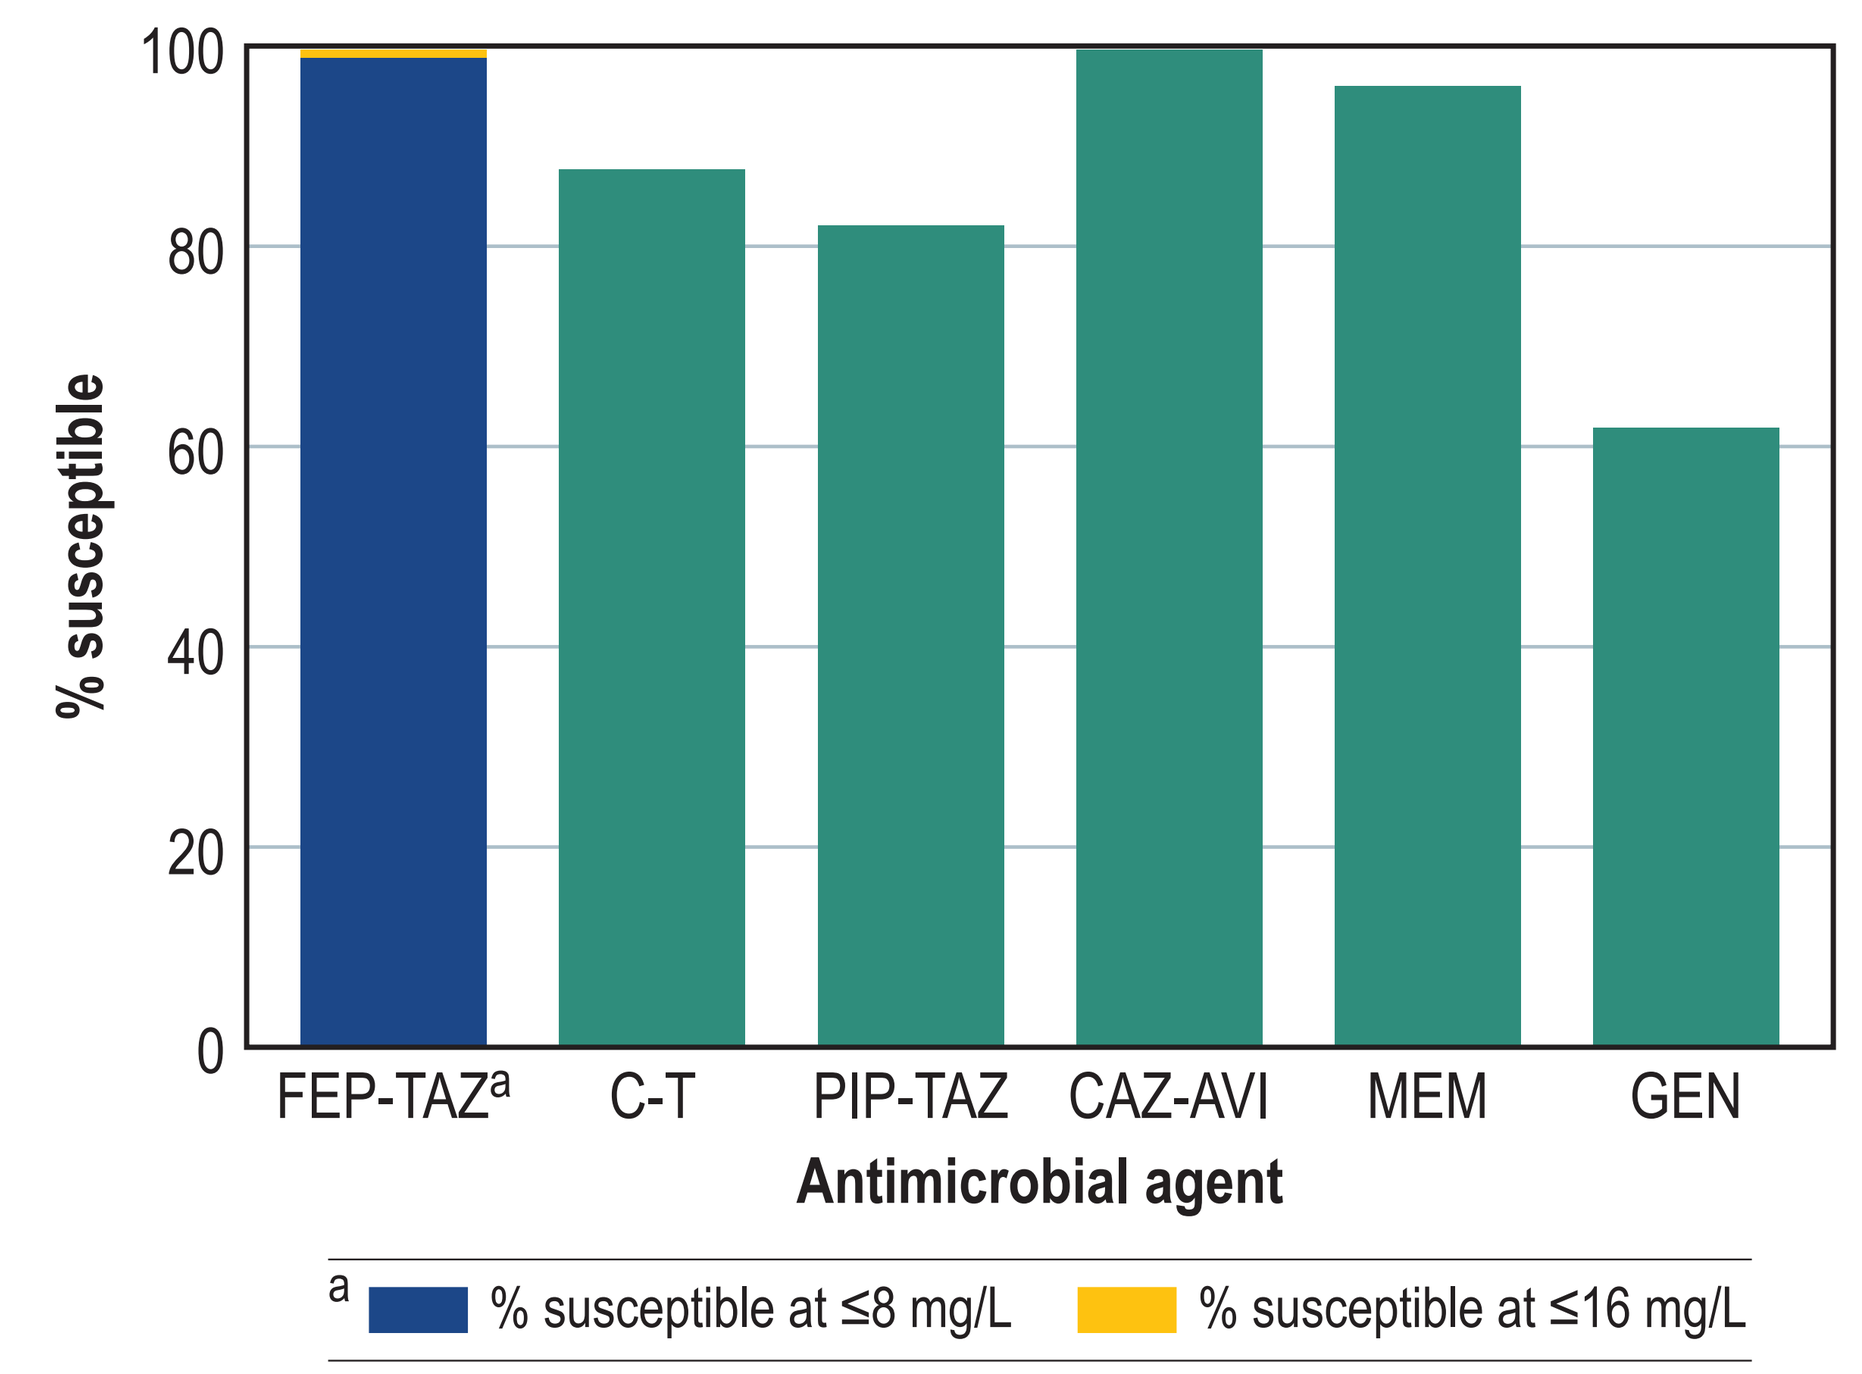
% susceptible
0
20
40
60
80
100
FEP-TAZ[a]
C-T
PIP-TAZ
CAZ-AVI
MEM
GEN
Antimicrobial agent
[a] % susceptible at ≤8 mg/L
% susceptible at ≤16 mg/L

ESBL, extended-spectrum β-lactamase; FEP-TAZ, cefepime-tazobactam; C-T, ceftolozane-tazobactam; PIP-TAZ, piperacillin-tazobactam; CAZ-AVI, ceftazidime-avibactam; MEM, meropenem; GEN, gentamicin.

Figure 2. Antimicrobial susceptibility of 165 *P. aeruginosa* isolates from complicated urinary tract infections collected worldwide in 2018

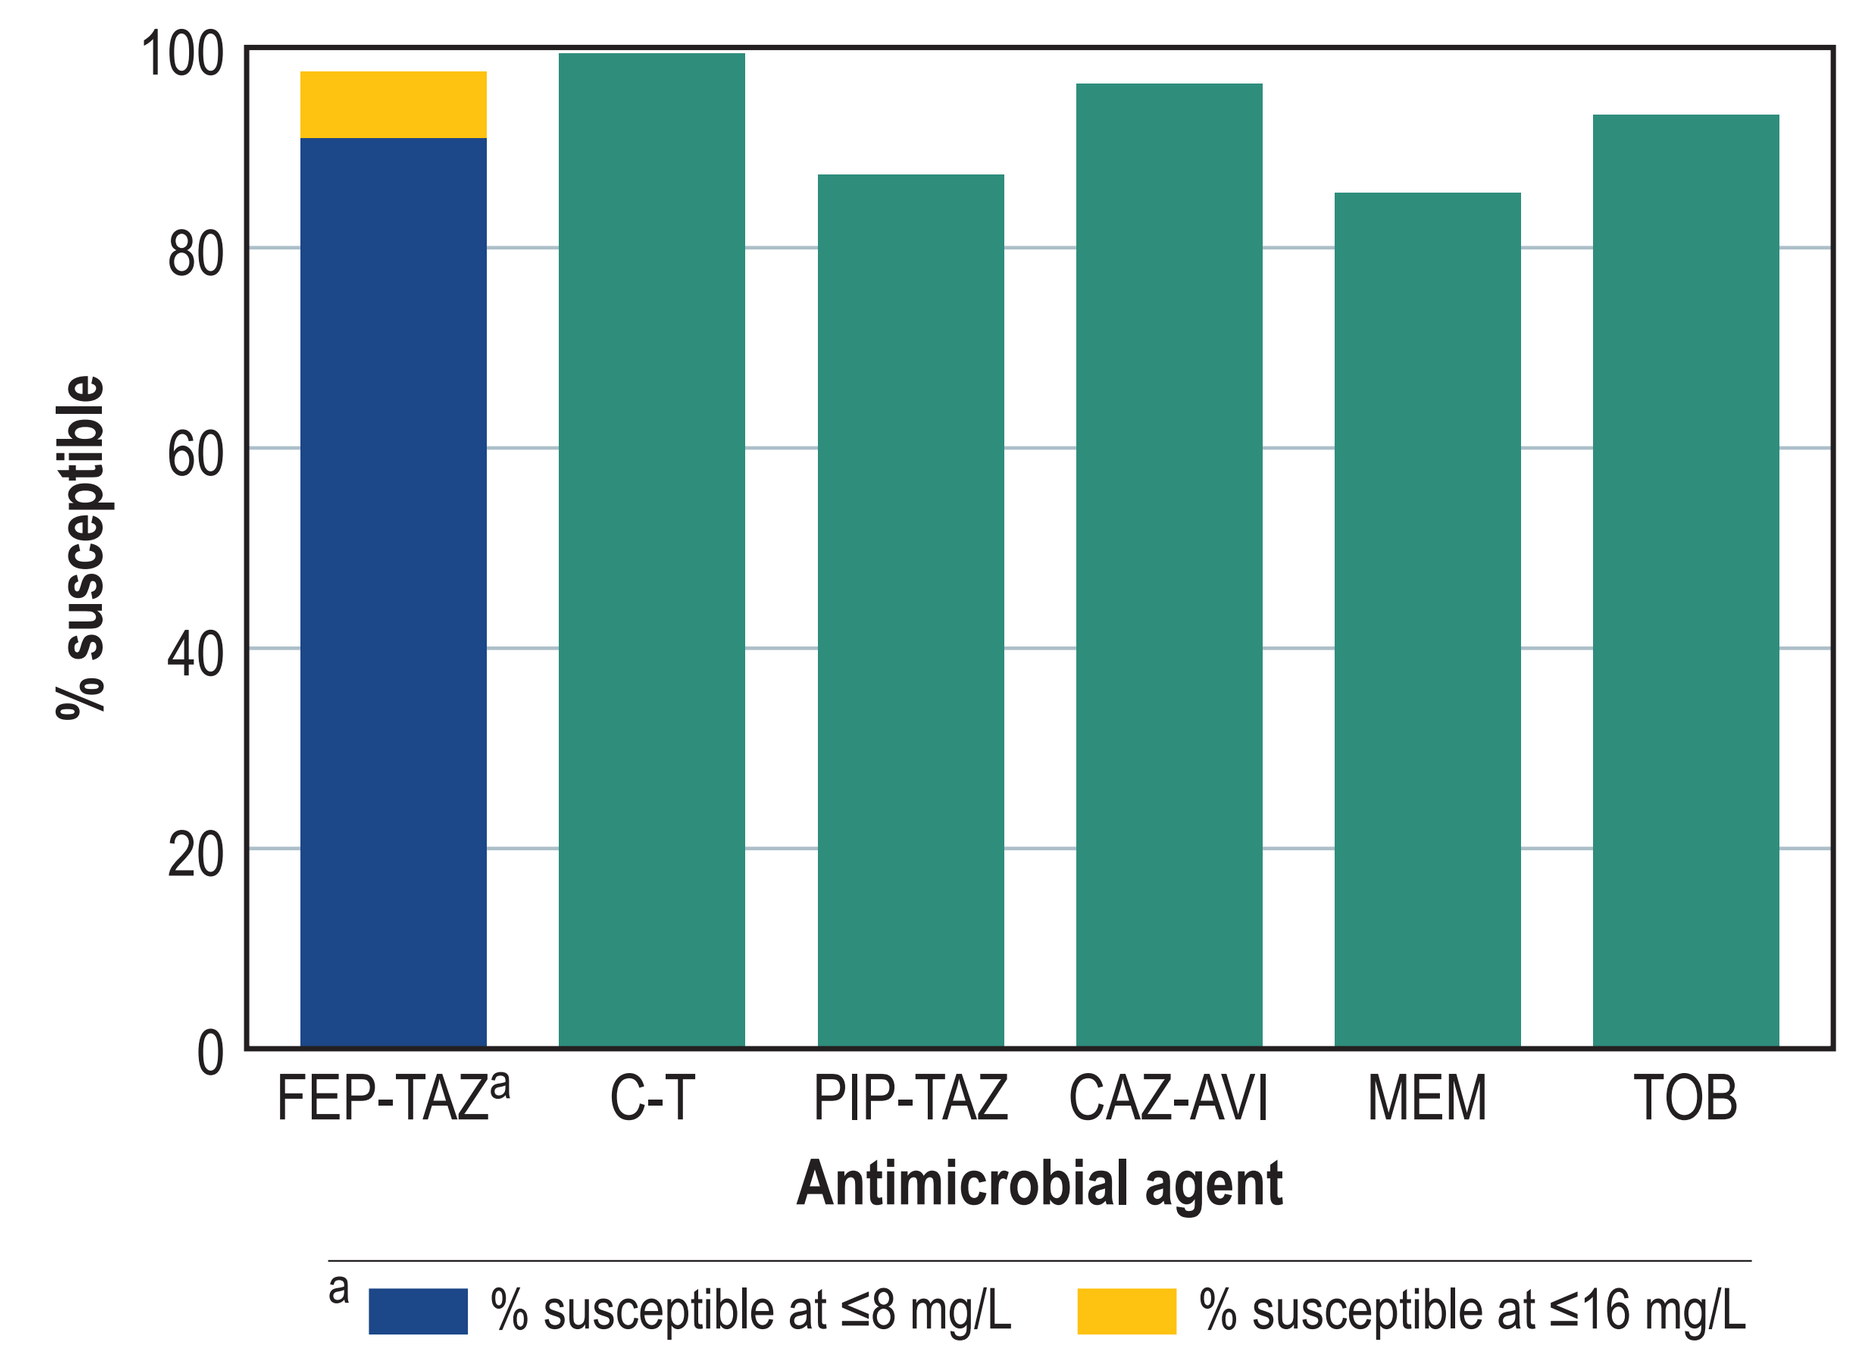
% susceptible
0
20
40
60
80
100
FEP-TAZ[a]
C-T
PIP-TAZ
CAZ-AVI
MEM
TOB
Antimicrobial agent
[a] % susceptible at ≤8 mg/L
% susceptible at ≤16 mg/L

FEP-TAZ, cefepime-tazobactam; C-T, ceftolozane-tazobactam; PIP-TAZ, piperacillin-tazobactam; CAZ-AVI, ceftazidime-avibactam; MEM, meropenem; TOB, tobramycin.

Conclusions

- Cefepime-tazobactam showed potent activity against *Enterobacterales* and *P. aeruginosa* isolated from patients with cUTIs in US hospitals in 2018
- Cefepime-tazobactam's overall spectrum (*Enterobacterales* + *P. aeruginosa*) was similar to ceftazidime-avibactam and greater than ceftolozane-tazobactam, piperacillin-tazobactam, and meropenem when the cefepime-tazobactam proposed PK/PD susceptible breakpoint of ≤16 mg/L was applied
- High-dose extended infusion cefepime-tazobactam may represent a carbapenem- and ceftazidime-avibactam-sparing valuable empiric therapy option, superior to piperacillin-tazobactam and ceftolozane-tazobactam for the treatment of infections caused by *Enterobacterales* and *P. aeruginosa*
- The cefepime-tazobactam combination also provides coverage of certain carbapenem-nonsusceptible organisms

Acknowledgements

This study was supported by Wockhardt Bio AG.

References

Burgess SV, Mabasa VH, Chow I, et al. (2015). Evaluating outcomes of alternative dosing strategies for cefepime: a qualitative systematic review. *Ann Pharmacother* 49: 311–322.

Castanheira M, Duncan LR, Rhomberg PR, et al. (2017). Enhanced activity of cefepime-tazobactam (WCK 4282) against KPC-producing *Enterobacteriaceae* when tested in media supplemented with human serum or sodium chloride. *Diagn Microbiol Infect Dis* 89: 305–309.

Clinical and Laboratory Standards Institute (2019). *M100Ed29E. Performance standards for antimicrobial susceptibility testing: 29th informational supplement*. Wayne, PA: CLSI.

Maxepime Package Insert (2012). Available at http://www.accessdata.fda.gov /drugsatfda_docs/label/2012/050679s036lbl.pdf. Accessed February 18, 2016.

Sader HS, Castanheira M, Mendes RE, et al. (2017). Antimicrobial activity of high-proportion cefepime-tazobactam (WCK 4282) against a large number of Gram-negative isolates collected worldwide in (2014). *Antimicrob Agents Chemother* 61: e02409.

Contact

Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2019-cefepime-tazobactam-urinary-tract-infection.pdf

Charges may apply. No personal information is stored.

IDWeek 2019, October 2–6, 2019, Washington, DC