IDWeek 2019 | Poster #1583

Eight Years of Sustained Potency and Activity of Oritavancin against Gram-Positive Isolates Causing Bacteremia and Endocarditis in the USA, Including Enterococcal Infections Requiring an Optimized Dosing Strategy for Daptomycin

Cecilia G. Carvalhaes, Helio S. Sader, Jennifer M. Streit, Robert K. Flamm, Rodrigo E. Mendes
JMI Laboratories, North Liberty, Iowa, USA

Introduction

- Bloodstream infections (BSIs) have challenged clinicians and infectious disease practitioners due to increased antimicrobial resistance rates
- Oritavancin is a potent lipoglycopeptide with desirable pharmacokinetic/pharmacodynamic parameters for treating serious gram-positive infections
 - Its concentration-dependent activity and prolonged half-life allow for single-dose treatment (against acute bacterial skin and skin structure infections)
 - This agent has multiple mechanisms of action and rapid concentration-dependent bactericidal activity
 - Oritavancin inhibits cell wall synthesis via two mechanisms and impairs membrane barrier function
- In this study, the longitudinal *in vitro* activity of oritavancin was assessed against *Staphylococcus aureus*, *Enterococcus faecalis*, and *Enterococcus faecium* causing BSI, including infective endocarditis (IE), enterococcal infections caused by vancomycin-resistant (VRE) isolates, and infections requiring an optimized dosing strategy for daptomycin

Materials and Methods

Bacterial isolates

- A total of 5,469 *S. aureus*, 1,158 *E. faecalis*, and 721 *E. faecium* were recovered from BSI (1 isolate/patient) in 35 US medical centers from 2011–2018
- Subsets of 84 *S. aureus* isolates causing IE and 233 *E. faecium* with VRE phenotype and elevated daptomycin MIC (≥2 mg/L) were evaluated
- Only isolates determined to be significant by local criteria as the reported probable cause of infection were included in the program
- Bacterial identification was performed by matrix-assisted laser desorption ionization–time of flight mass spectrometry

Susceptibility testing

- Isolates were tested for susceptibility by broth microdilution following guidelines in the CLSI M07 (2018) document
- Frozen-form broth microdilution panels in cation-adjusted Mueller-Hinton broth (CAMHB) were manufactured by JMI Laboratories
 - Polysorbate-80 (0.002%) was included in the CAMHB when testing oritavancin, while calcium (Ca^{2+}) supplementation (50 mg/L) was used for testing daptomycin
- Quality assurance was performed by concurrently testing the CLSI-recommended quality control reference strains *S. aureus* ATCC 25913 and *E. faecalis* ATCC 29212
- Breakpoint criteria for oritavancin and comparator agents were those published in the CLSI M100 (2019) document

Results

- Overall, oritavancin showed similar activity (MIC_{50}/MIC_{90}, 0.03/0.06 mg/L) against methicillin-resistant *S. aureus* (MRSA), methicillin-susceptible *S. aureus* (MSSA), and the MRSA subset causing IE (41.7% of all MRSA; Table 1)
- Similar findings were noted for oritavancin tested against *E. faecalis* isolates (MIC_{50}/MIC_{90}, 0.015/0.06 mg/L) and those displaying daptomycin MIC values ≥2 mg/L (MIC_{50}/MIC_{90}, 0.015/0.06 mg/L; Table 1)
- Oritavancin MIC values against the overall collection of *E. faecium* isolates (MIC_{50}/MIC_{90}, 0.015/0.06 mg/L) were 2- to 4-fold lower than those observed with the collection of VRE isolates displaying daptomycin MIC values ≥2 mg/L ($MIC_{50}/MIC_{90}/MIC_{100}$, 0.06/0.12/0.25 mg/L; 32.3% of all *E. faecium*; Table 1)
- All *E. faecium* were inhibited by oritavancin at MIC values of ≤0.25 mg/L, and 98.5% were inhibited by oritavancin at MIC values of ≤0.12 mg/L (CLSI published susceptible breakpoint for vancomycin-susceptible *E. faecalis*; Table 1)
- The frequencies of *S. aureus* as a cause of BSI in US medical centers ranged from 26.2% (2012) to 22.9% (2015; data not shown), and MRSA from 11.9% (2011) to 9.6% (2017; Figure 1)
- During the study period oritavancin MIC_{50} results varied from 0.03 to 0.12 mg/L and MIC_{90} results varied from 0.015 to 0.06 mg/L (Figure 2)
- Among *E. faecalis* isolates with elevated daptomycin MIC (≥2 mg/L) oritavancin MIC_{50} results varied from 0.008 to 0.03 mg/L and MIC_{90} values ranged from 0.015 to 0.12 mg/L during this period (Table 1)
- Oritavancin inhibited 93.1% (2011) to 100% (2018) of vancomycin-resistant *E. faecium* isolates displaying daptomycin MIC values of ≥2 mg/L at MIC values of ≤0.12 mg/L (Table 2)
- Oritavancin MIC_{50} values varied from 0.03 to 0.06 mg/L and MIC_{90} results varied from 0.06 to 0.12 mg/L among vancomycin-resistant *E. faecium* isolates displaying daptomycin MIC values of ≥2 mg/L (Figure 2)

Table 1 Antimicrobial activity of oritavancin tested against *S. aureus, E. faecalis, E. faecium*, and resistant phenotypes isolated from bloodstream infection, including patients with endocarditis (2011–2018)

Organism/Phenotype	No. tested	Cumulative % inhibited at MIC (mg/L) of: ≤0.008	0.015	0.03	0.06	0.12	0.25	0.5	MIC_{50}	MIC_{90}
MSSA	3,148	4.2	40.1	79.0	95.9	100.0			0.03	0.06
MRSA	2,321	3.6	39.7	77.4	95.0	99.9	100.0		0.03	0.06
MRSA-IE	35	0.0	34.3	77.1	94.3	100.0			0.03	0.06
E. faecalis	1,158	27.6	67.9	88.1	94.3	97.8	99.7	100.0	0.015	0.06
E. faecalis - DAP ≥2 mg/L	83	28.9	61.4	77.1	92.8	98.8	100.0		0.015	0.06
E. faecium	721	34.0	50.1	72.5	90.0	98.5	100.0		0.015	0.06
E. faecium - DAP ≥2 mg/L	348	33.6	44.8	66.4	87.9	98.0	100.0		0.03	0.12
VR *E. faecium* - DAP ≥2 mg/L	233	4.3	18.5	49.8	82.0	97.0	100.0		0.06	0.12

MSSA, methicillin-susceptible *S. aureus*; MRSA, methicillin-resistant *S. aureus*; IE, infective endocarditis; DAP, daptomycin; VRE, vancomycin-resistant

Figure 1 Frequency of MRSA and VRE isolates among bacterial isolates causing bloodstream infections in US medical centers (2011–2018)

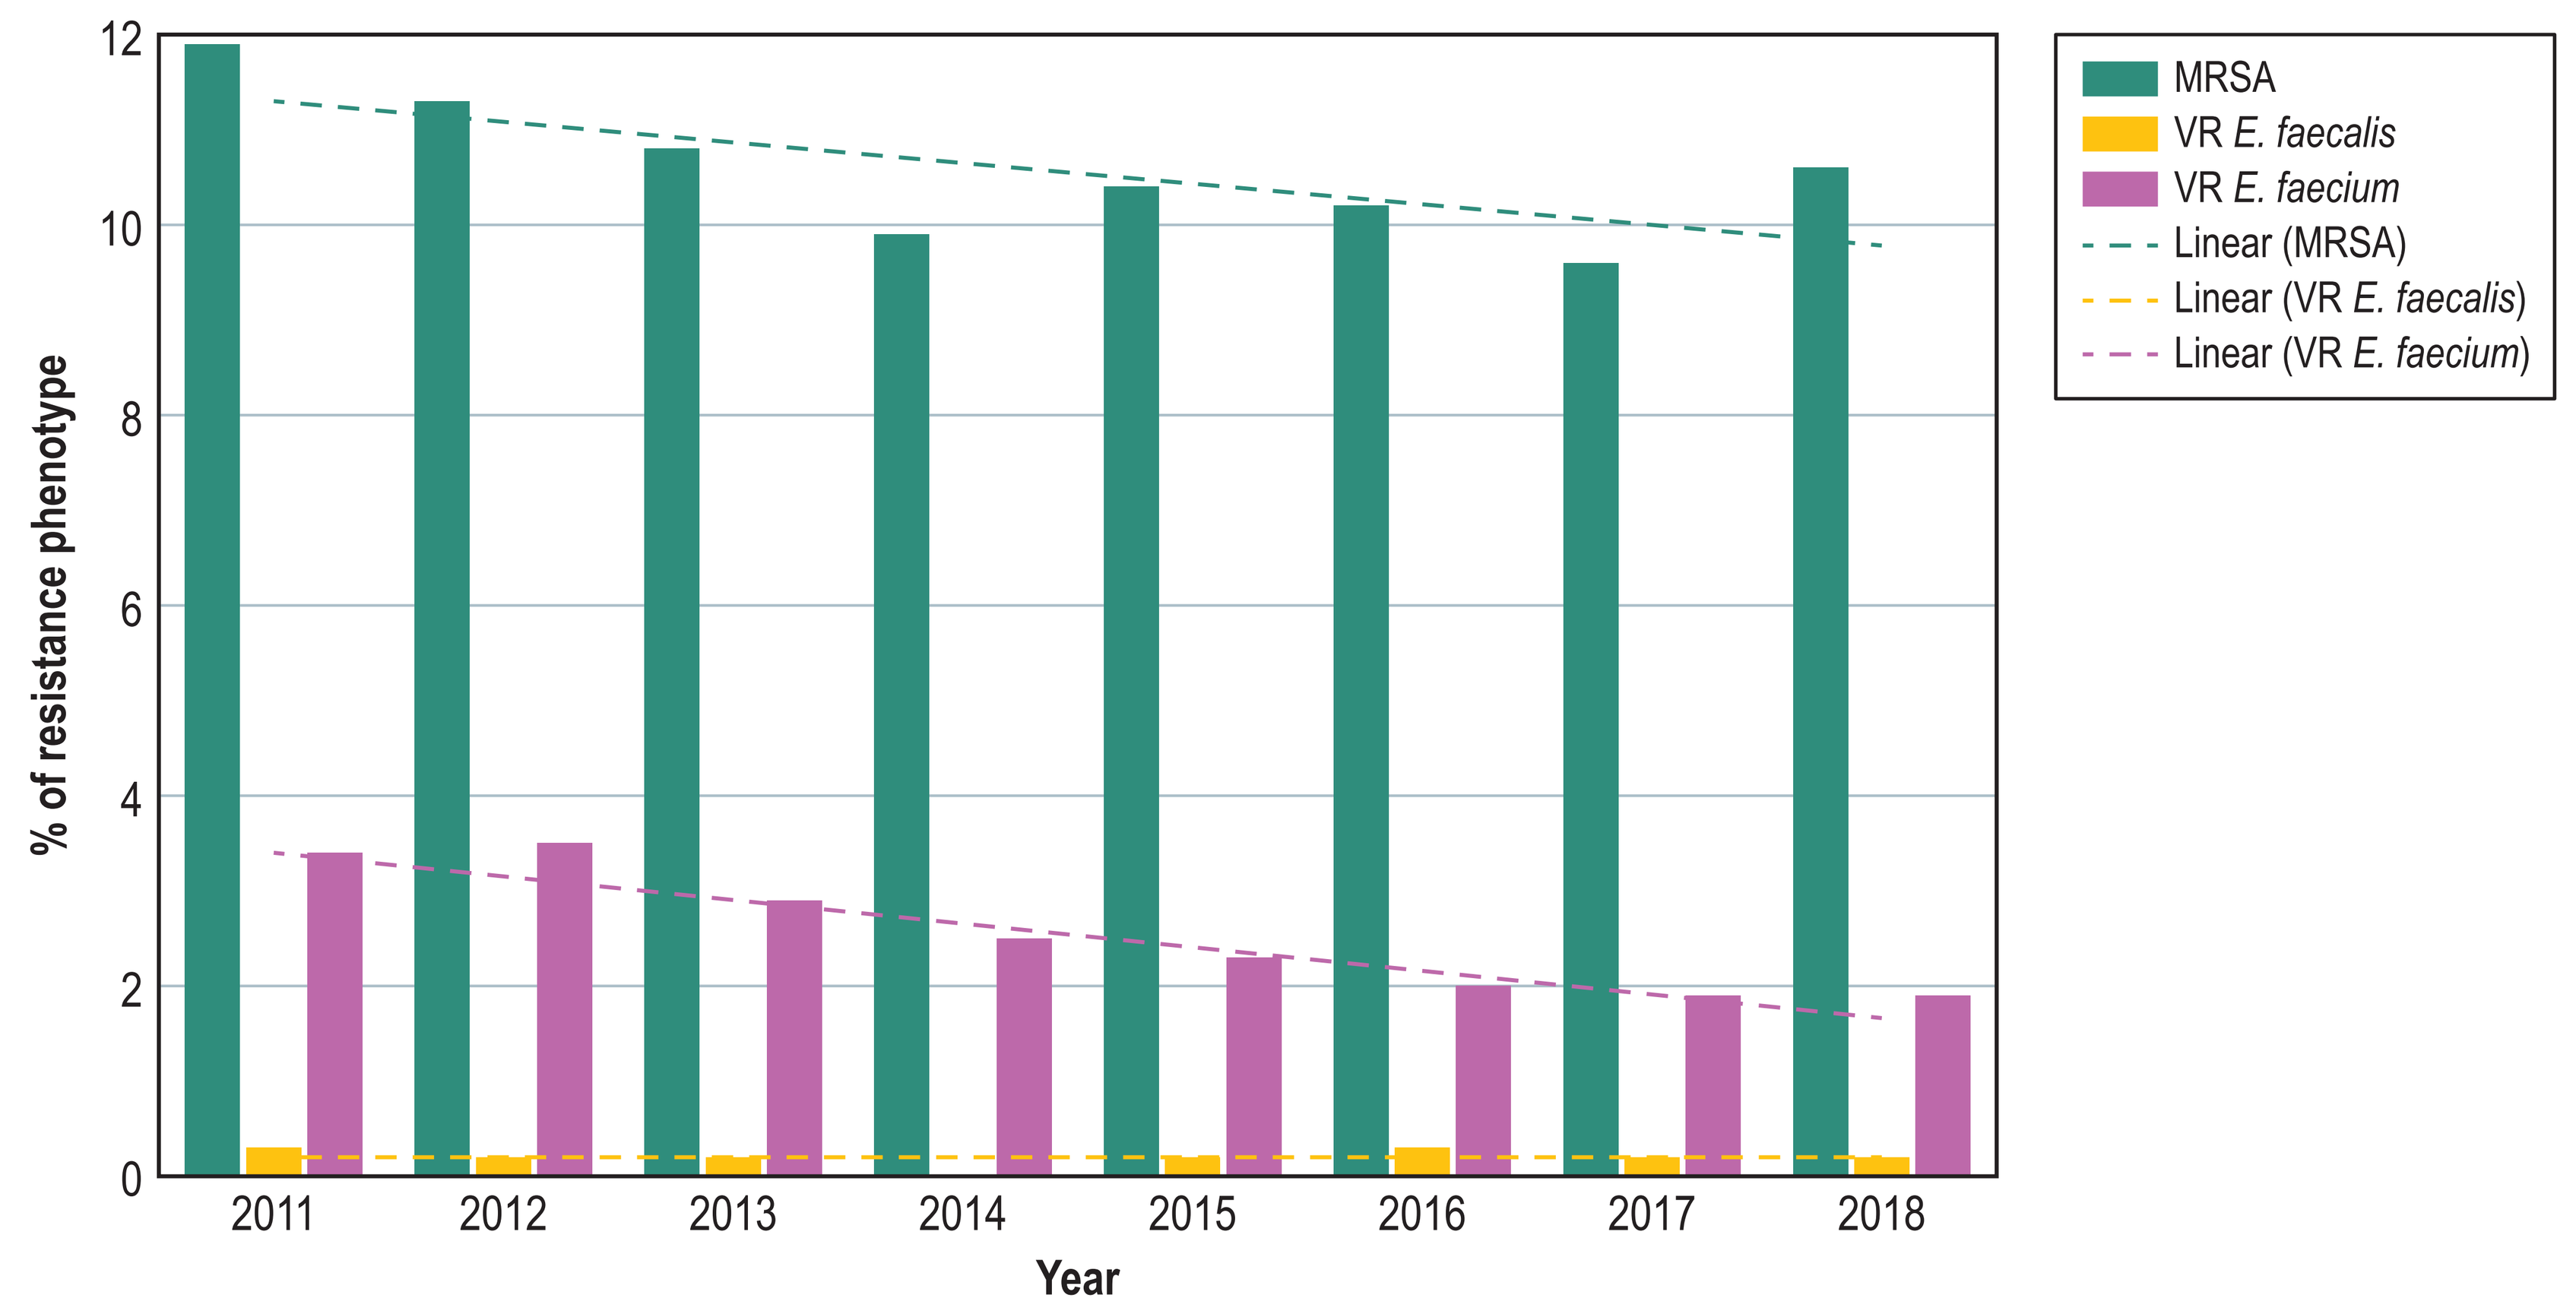
% of resistance phenotype
0
2
4
6
8
10
12
2011
2012
2013
2014
2015
2016
2017
2018
Year
MRSA
VR E. faecalis
VR E. faecium
Linear (MRSA)
Linear (VR E. faecalis)
Linear (VR E. faecium)

Abbreviations: MRSA, methicillin-resistant *S. aureus*; VRE, vancomycin-resistant *Enterococcus*

Figure 2 Temporal activity of oritavancin against isolates causing bloodstream infections in US medical centers (2011–2018)

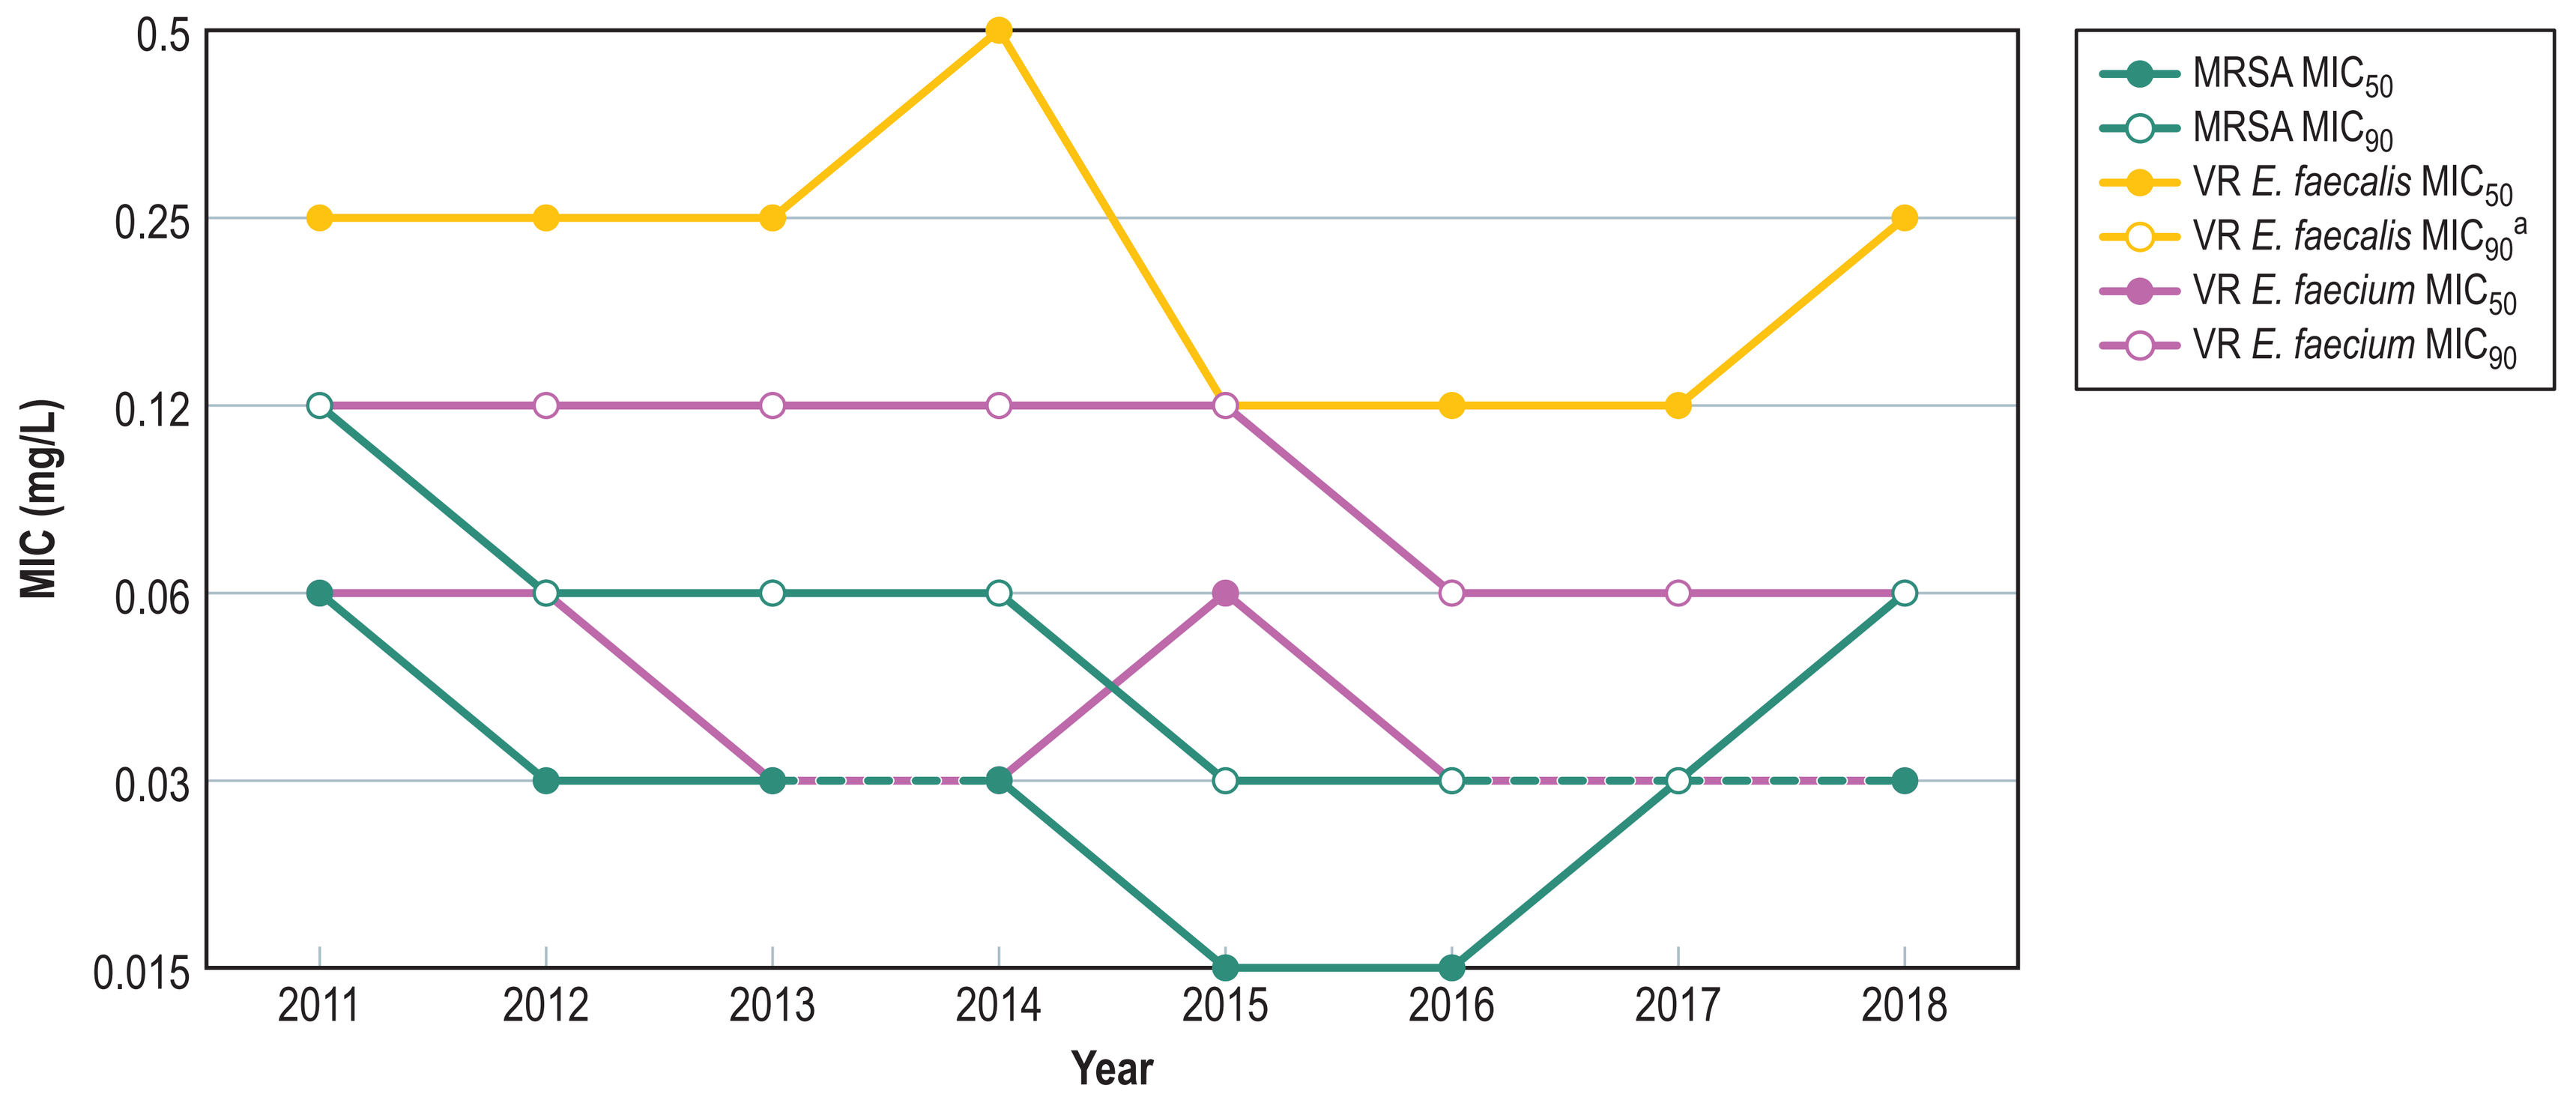
MIC (mg/L)
0.5
0.25
0.12
0.06
0.03
0.015
2011
2012
2013
2014
2015
2016
2017
2018
Year
MRSA MIC50
MRSA MIC90
VR E. faecalis MIC50
VR E. faecalis MIC90 a
VR E. faecium MIC50
VR E. faecium MIC90

[a] MIC_{90} was not calculated due to limited number of isolates (<10).
Abbreviations: MRSA, methicillin-resistant *S. aureus*; VR, vancomycin-resistant

Table 2 Longitudinal activity of oritavancin against *S. aureus* and *Enterococcus* spp. subsets causing bloodstream infections in US medical centers (2011–2018)

Organism/Phenotype (no. tested)	% of oritavancin-susceptible isolates: 2011	2012	2013	2014	2015	2016	2017	2018
MSSA (3,148)	100.0	100.0	100.0	100.0	100.0	100.0	100.0	100.0
MRSA (2,321)	100.0	100.0	100.0	100.0	99.6	99.3	100.0	100.0
E. faecalis (1,158)[a]	97.0	97.8	97.0	98.9	97.9	98.1	100.0	96.6
E. faecium VSE-DAP ≤1 mg/L (97)[b]	100.0	100.0	100.0	100.0	100.0	100.0	100.0	100.0
E. faecium VRE-DAP ≥2 mg/L (233)[b]	93.1	100.0	96.0	94.1	100.0	100.0	100.0	100.0

Abbreviations: MSSA, methicillin-susceptible *S. aureus*; MRSA, methicillin-resistant *S. aureus*; VSE, vancomycin-susceptible enterococci; VRE, vancomycin-resistant enterococci; DAP, daptomycin
[a] CLSI breakpoint established against vancomycin-susceptible *E. faecalis*, applied for all *E. faecalis* isolates for comparison purposes.
[b] CLSI breakpoint established against vancomycin-susceptible *E. faecalis*, applied for *E. faecium* isolates for comparison purposes.

Conclusions

- Oritavancin showed potent activity against this US collection of isolates causing BSI and IE, including isolates with elevated daptomycin MIC values that require higher dosage regimens when treating serious infections
- In addition, oritavancin maintained stable potencies (±1 dilution) throughout the 8-year surveillance study period, which includes at least 3 years post-FDA approval
- Oritavancin inhibited 100% of the challenge subset of VR *E. faecium* displaying daptomycin MIC values of ≥2 mg/L in the last 4 years of the study (2015–2018)
- Oritavancin was approved by regulatory authorities for the adult treatment of skin and skin structure infections; however, its clinical role for treating invasive enterococcal and staphylococcal infections, mainly those caused by highly resistant *E. faecium* or MRSA, warrants further investigations

Acknowledgements

This study was supported by Melinta Therapeutics. Melinta was involved in the design and decision to present these results, and JMI Laboratories received compensation for services related to preparing the poster. Melinta had no involvement in the collection, analysis, or interpretation of data.

References

Baddour LM, Wilson WR, Bayer AS, Fowler Jr VG, Tleyjeh IM, Rybak MJ, Barsic B, Lockhart PB, Gewitz MH, Levison ME, Bolger AF, Steckelberg JM, Baltimore RS, Fink AM, O'Gara P, Taubert KA, American Heart Association Committee on Rheumatic Fever E, Kawasaki Disease of the Council on Cardiovascular Disease in the Young CoCC, Council on Cardiovascular Surgery and Anesthesia aSC (2015). Infective endocarditis in adults: Diagnosis, antimicrobial therapy, and management of complications: A scientific statement for healthcare professionals from the American Heart Association. *Circulation* 132:1435–1486.

ORBACTIV® (Oritavancin; 2014). Allergan USA, Inc. Available at https://www.accessdata.fda.gov/drugsatfda_docs/label/2016/206334s003lbl.pdf. Accessed August 2019.

Clinical and Laboratory Standards Institute (2019). *M100Ed29E. Performance standards for antimicrobial susceptibility testing: 29th informational supplement.* Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: eleventh edition*. Wayne, PA: CLSI.

Food and Drug Administration. Antibacterial Susceptibility Test Interpretive Criteria. Tigecycline. Available at https://www.fda.gov/drugs/development-resources/tigecycline-injection-products

Munita JM, Mishra NN, Alvarez D, Tran TT, Diaz L, Panesso D, Reyes J, Murray BE, Adachi JA, Bayer AS, Arias CA (2014). Failure of high-dose daptomycin for bacteremia caused by daptomycin-susceptible Enterococcus faecium harboring LiaSR substitutions. *Clin Infect Dis* 59:1277–1280.

Contact

Cecilia Carvalhaes, MD, PhD, D(ABMM)
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: cecilia-carvalhaes@jmilabs.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2019-oritavancin.pdf

Charges may apply. No personal information is stored.

IDWeek 2019, October 2–6, 2019, Washington, DC