IDWeek 2022 | Poster #P1684

Antimicrobial Activity of Aztreonam-Avibactam Against a Large Collection of *Stenotrophomonas maltophilia* and *Burkholderia cepacia* Species Complex Causing Infections in United States (US) Medical Centers (2016–2021)

HS Sader, D Shortridge, SJ Ryan Arends, CG Carvalhaes, RE Mendes, M Castanheira

JMI Laboratories, North Liberty, Iowa, USA

CONCLUSIONS

Aztreonam-avibactam demonstrated potent *in vitro* activity against *S. maltophilia* and *B. cepacia* from US hospitals and may represent a valuable option to treat infections caused by these organisms.

Clinical studies are urgently warranted to evaluate the efficacy of aztreonam-avibactam against infection caused by these organisms.

Susceptibility breakpoints for *S. maltophilia* and *B. cepacia* should be re-evaluated for antimicrobials currently used to treat infections caused by these organisms.

INTRODUCTION

- *S. maltophilia* has become a major cause of hospital-associated pneumonia in US medical centers.
- Aztreonam is a monobactam stable to hydrolysis by metallo-β-lactamases (MBLs), including those intrinsically produced by *S. maltophilia*.
- Avibactam is a non–β-lactam β-lactamase inhibitor that inhibits most clinically relevant serine β-lactamases, such as ESBLs, KPCs, AmpCs, and some OXAs.
- Aztreonam-avibactam is being developed for treatment of serious infections caused by Gram-negative bacteria, including MBL producers.
- We evaluated the activity of aztreonam-avibactam against *S. maltophilia* and *B. cepacia* from US hospitals.

METHODS

- 1,565 *S. maltophilia* and 219 *B. cepacia* were consecutively collected (1/patient) in 77 US medical centers in 2016–2021.
- This isolate collection was recovered mainly from patients with pneumonia and bloodstream infection (BSI; Figure 1).
- Only isolates determined to be the probable cause of infection were included.
- Susceptibility testing was performed by the CLSI broth microdilution method.
- CLSI/US FDA breakpoints were applied when available.

Figure 1. Distributions of *S. maltophilia* (1A) and *B. cepacia* (1B) isolates by infection type

A. *S. maltophilia* **B.** *B. cepacia*

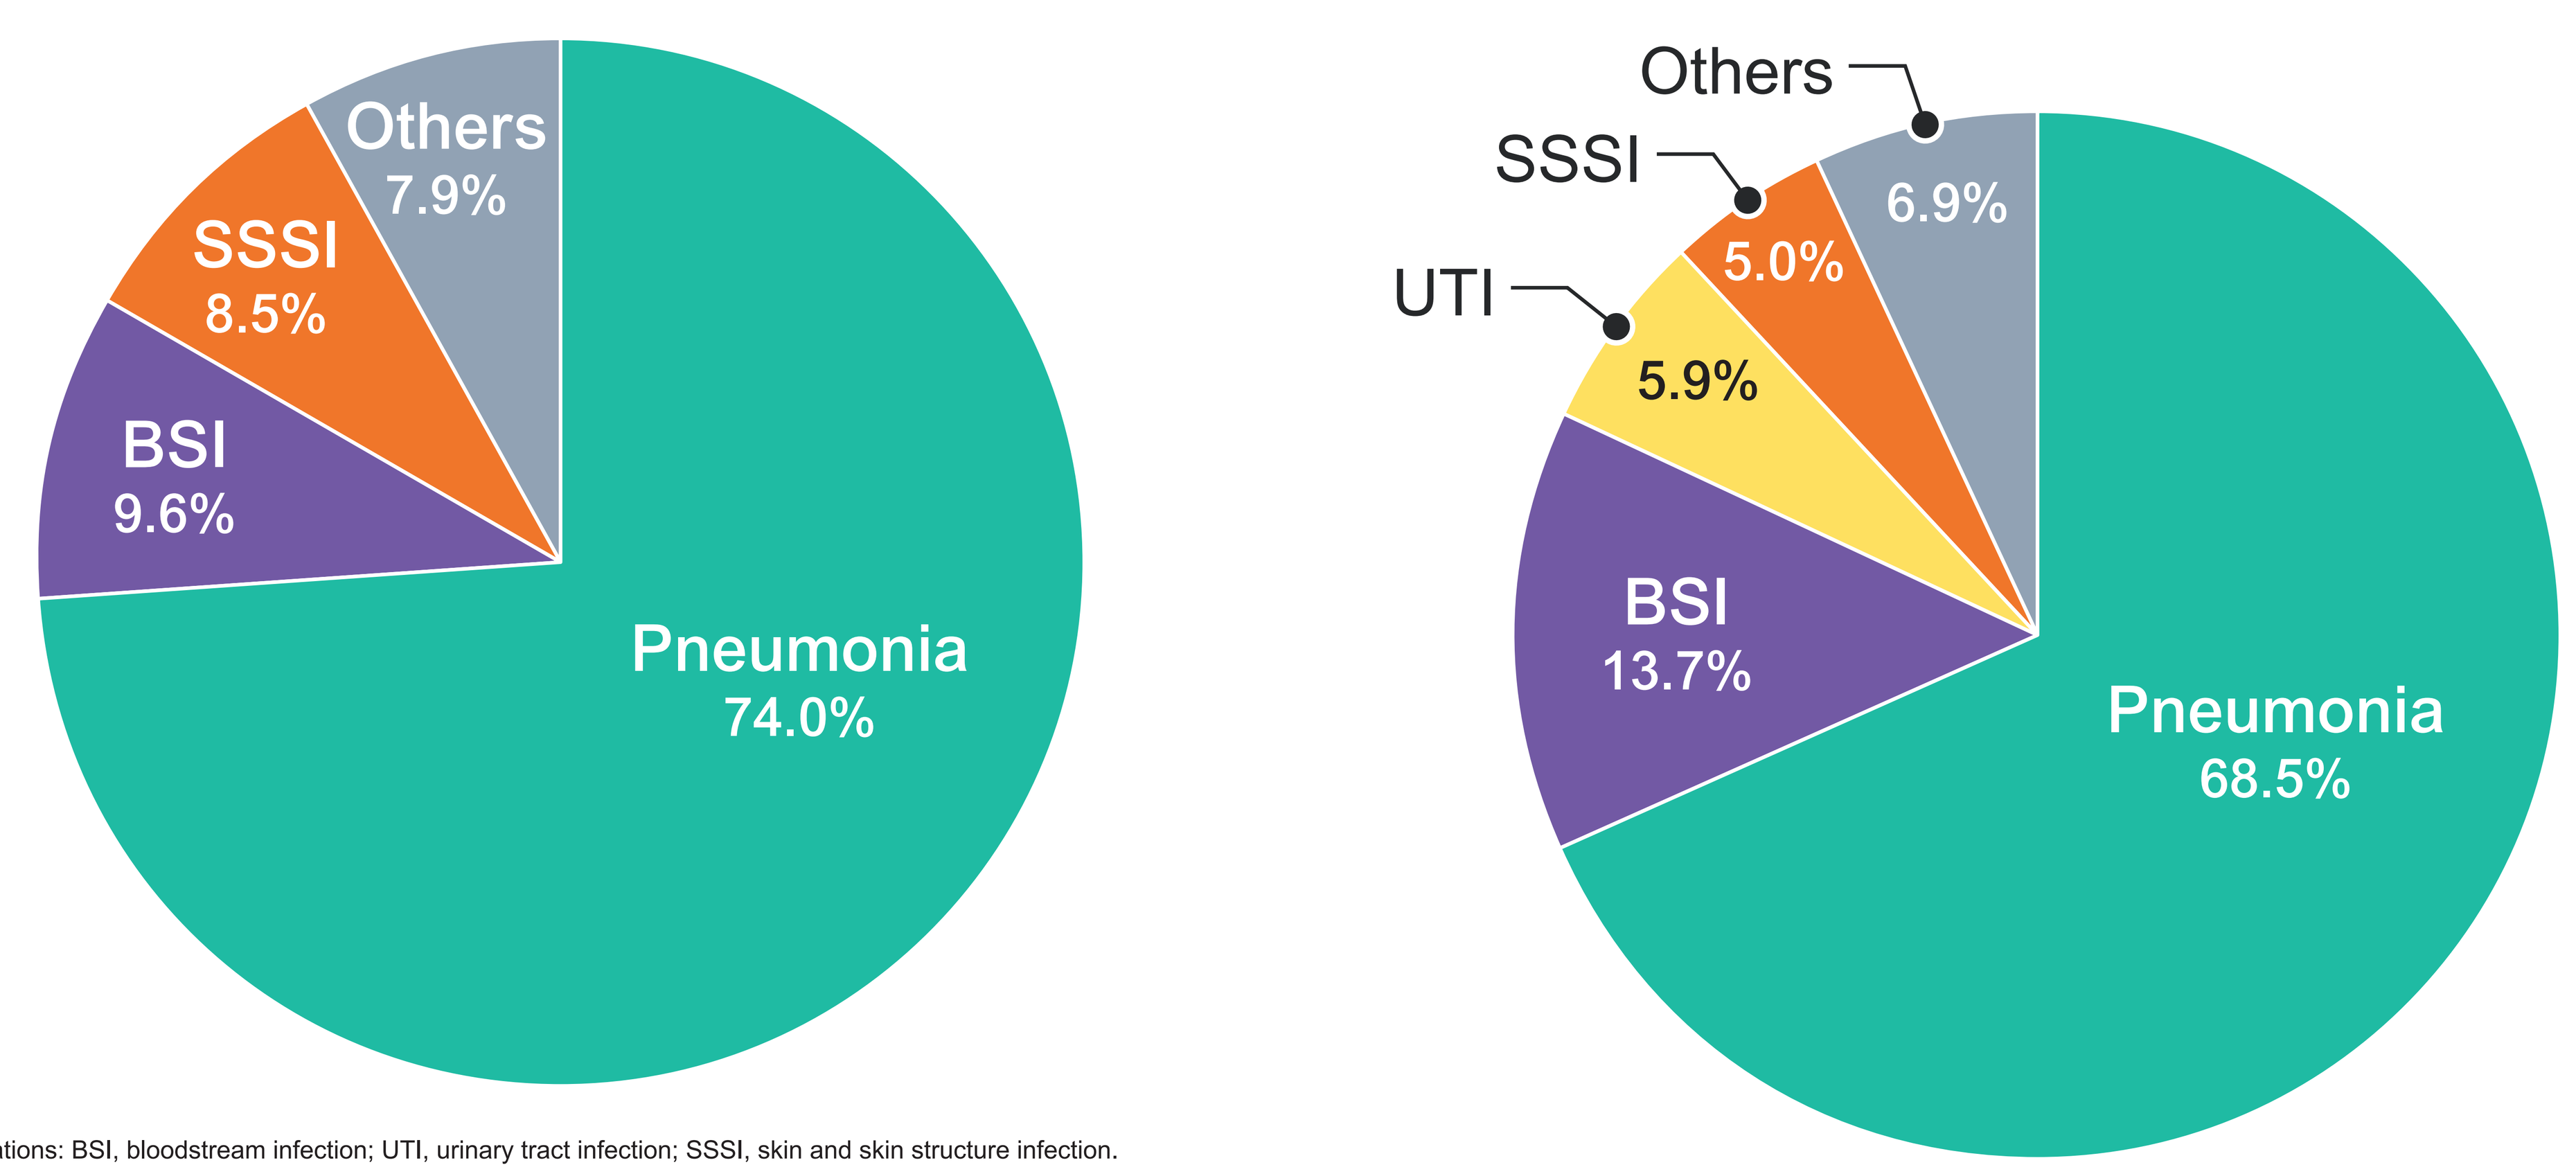
Others
7.9%
SSSI
8.5%
BSI
9.6%
Pneumonia
74.0%
Others
SSSI
UTI
6.9%
5.0%
5.9%
BSI
13.7%
Pneumonia
68.5%

Abbreviations: BSI, bloodstream infection; UTI, urinary tract infection; SSSI, skin and skin structure infection.

RESULTS

- Aztreonam-avibactam was very active against *S. maltophilia*, with $MIC_{50/90}$ values of 2/4 mg/L.
- Trimethoprim-sulfamethoxazole (TMP-SMX; $MIC_{50/90}$, ≤0.5/1 mg/L; 95.7% susceptible [S]) and minocycline ($MIC_{50/90}$, 0.5/2 mg/L; 99.2%S) also were very active against *S. maltophilia* (Figure 2).
- Ceftazidime and levofloxacin were active against 21.3% and 76.2% of *S. maltophilia* isolates per CLSI criteria, respectively (Figure 2).
- Aztreonam-avibactam was also very active against *B. cepacia*, with $MIC_{50/90}$ values of 4/16 mg/L and 88.1% inhibited at ≤8 mg/L (Figure 3), including 93.3% and 87.3% of isolates from BSI and pneumonia, respectively.
- The most active comparators tested against *B. cepacia* were ceftazidime (84.9%S), meropenem (84.5%S), TMP-SMX (83.5%S), and minocycline (80.0%S; Figure 3).

Figure 2. Antimicrobial susceptibility of *S. maltophilia* (*n*=1,565) from US medical centers (2016–2021)

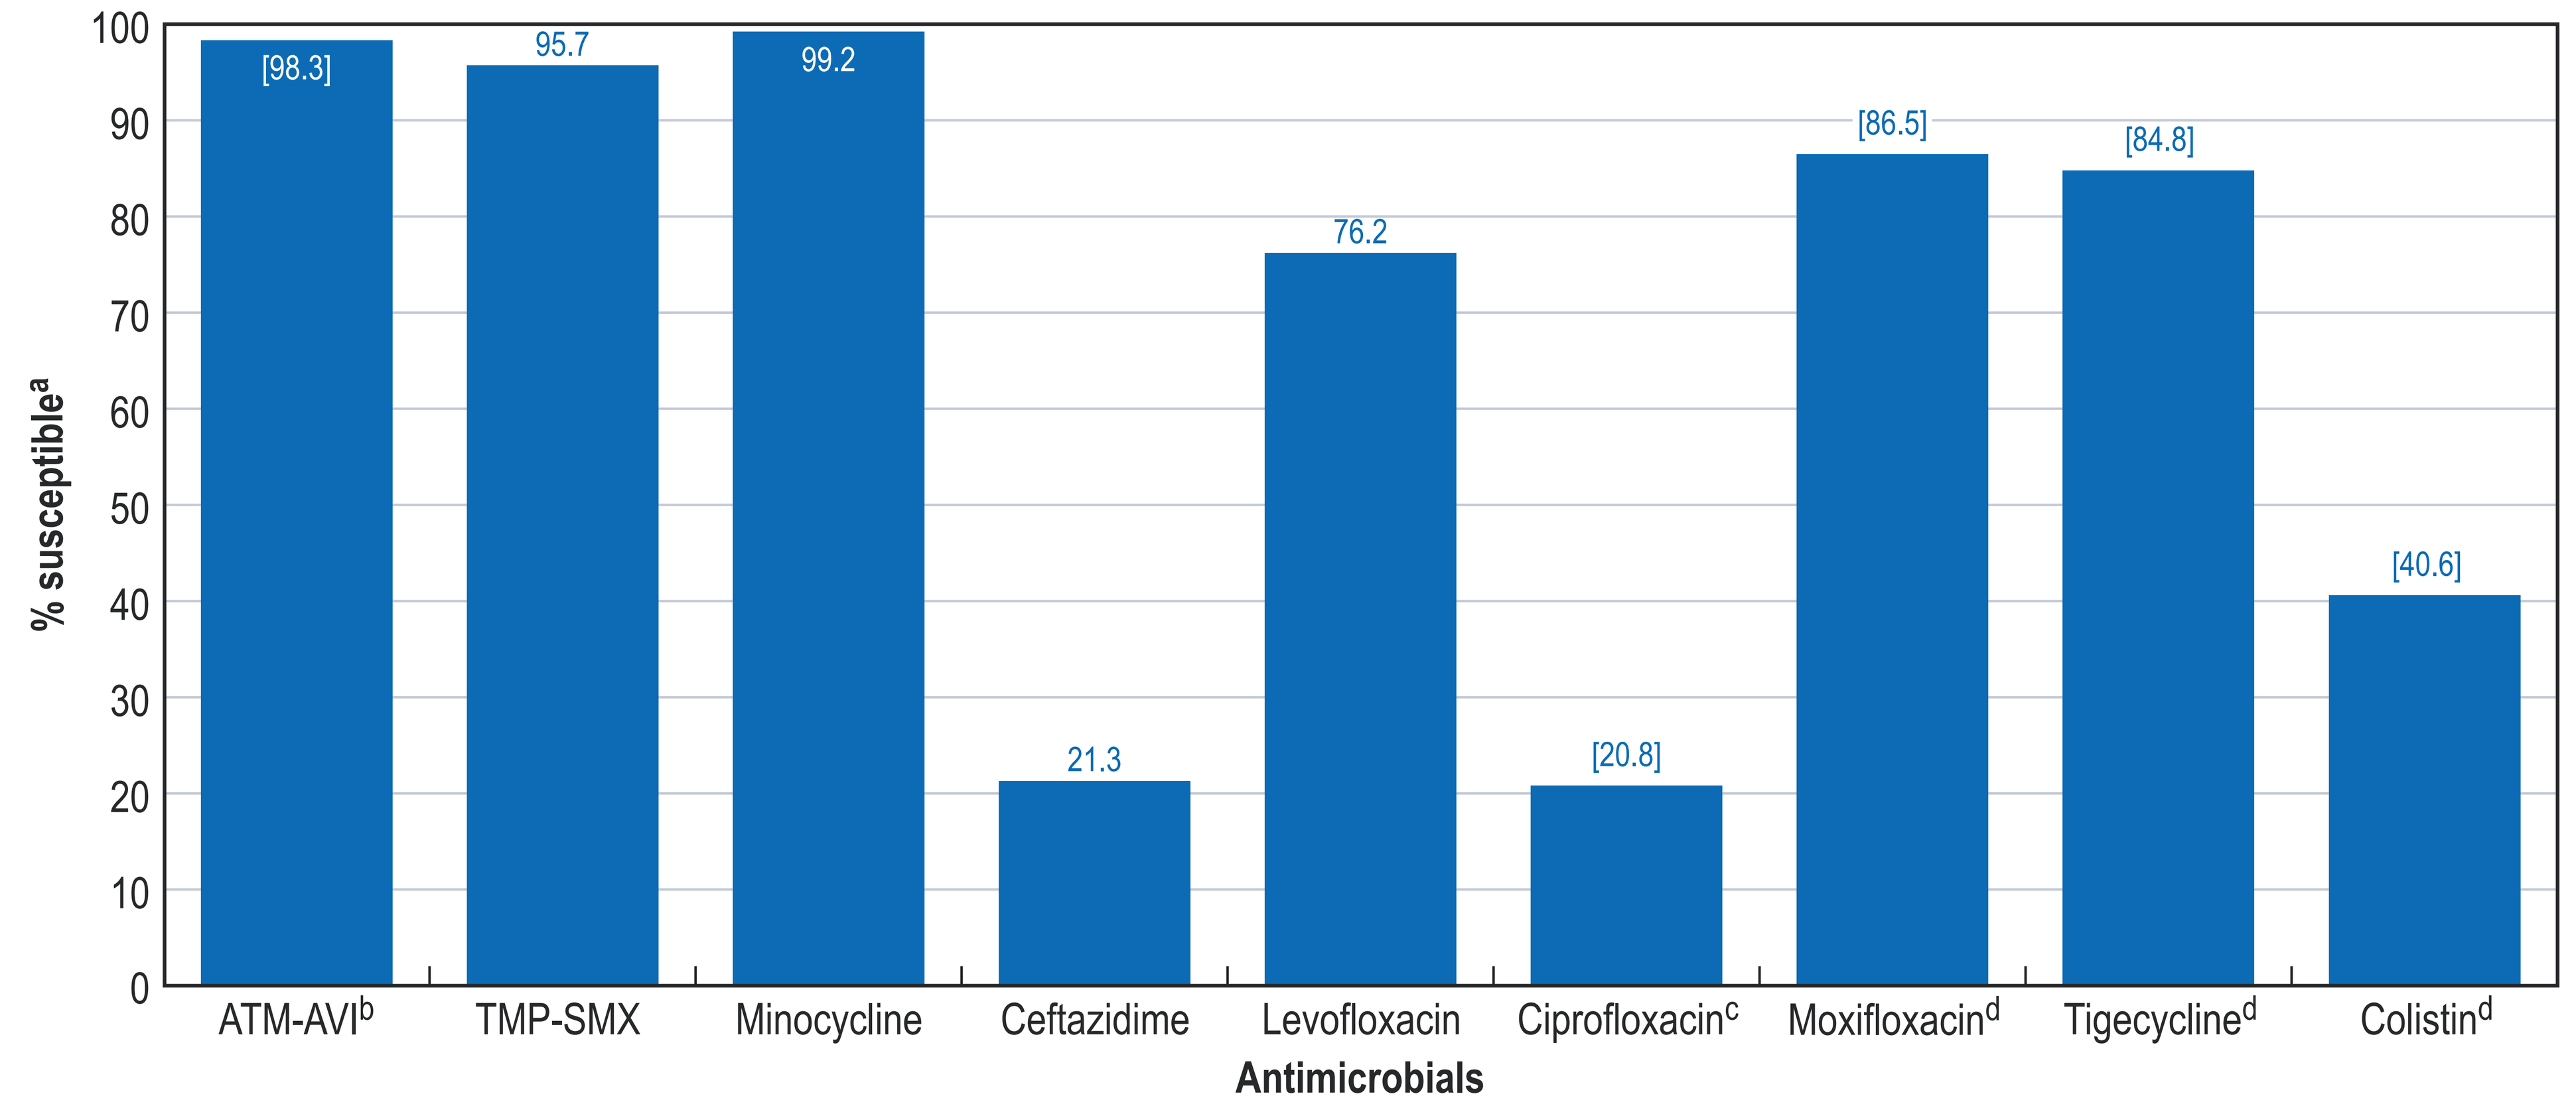
100
90
80
70
60
50
40
30
20
10
0
% susceptible[a]
[98.3]
95.7
99.2
21.3
76.2
[20.8]
[86.5]
[84.8]
[40.6]
ATM-AVI[b]
TMP-SMX
Minocycline
Ceftazidime
Levofloxacin
Ciprofloxacin[c]
Moxifloxacin[d]
Tigecycline[d]
Colistin[d]
Antimicrobials

[a] Criteria as published by CLSI (2022) and US FDA (2022) unless noted.
[b, c, d] Values in brackets indicate % inhibited at: ≤8 mg/L [(b)], ≤1 mg/L [(c)], and ≤2 mg/L [(d)] for comparison.
Abbreviations: ATM-AVI, aztreonam-avibactam; TMP-SMX, trimethoprim-sulfamethoxazole.

Figure 3. Antimicrobial susceptibility of *B. cepacia* species complex (*n*=219) from US medical centers (2016–2021)

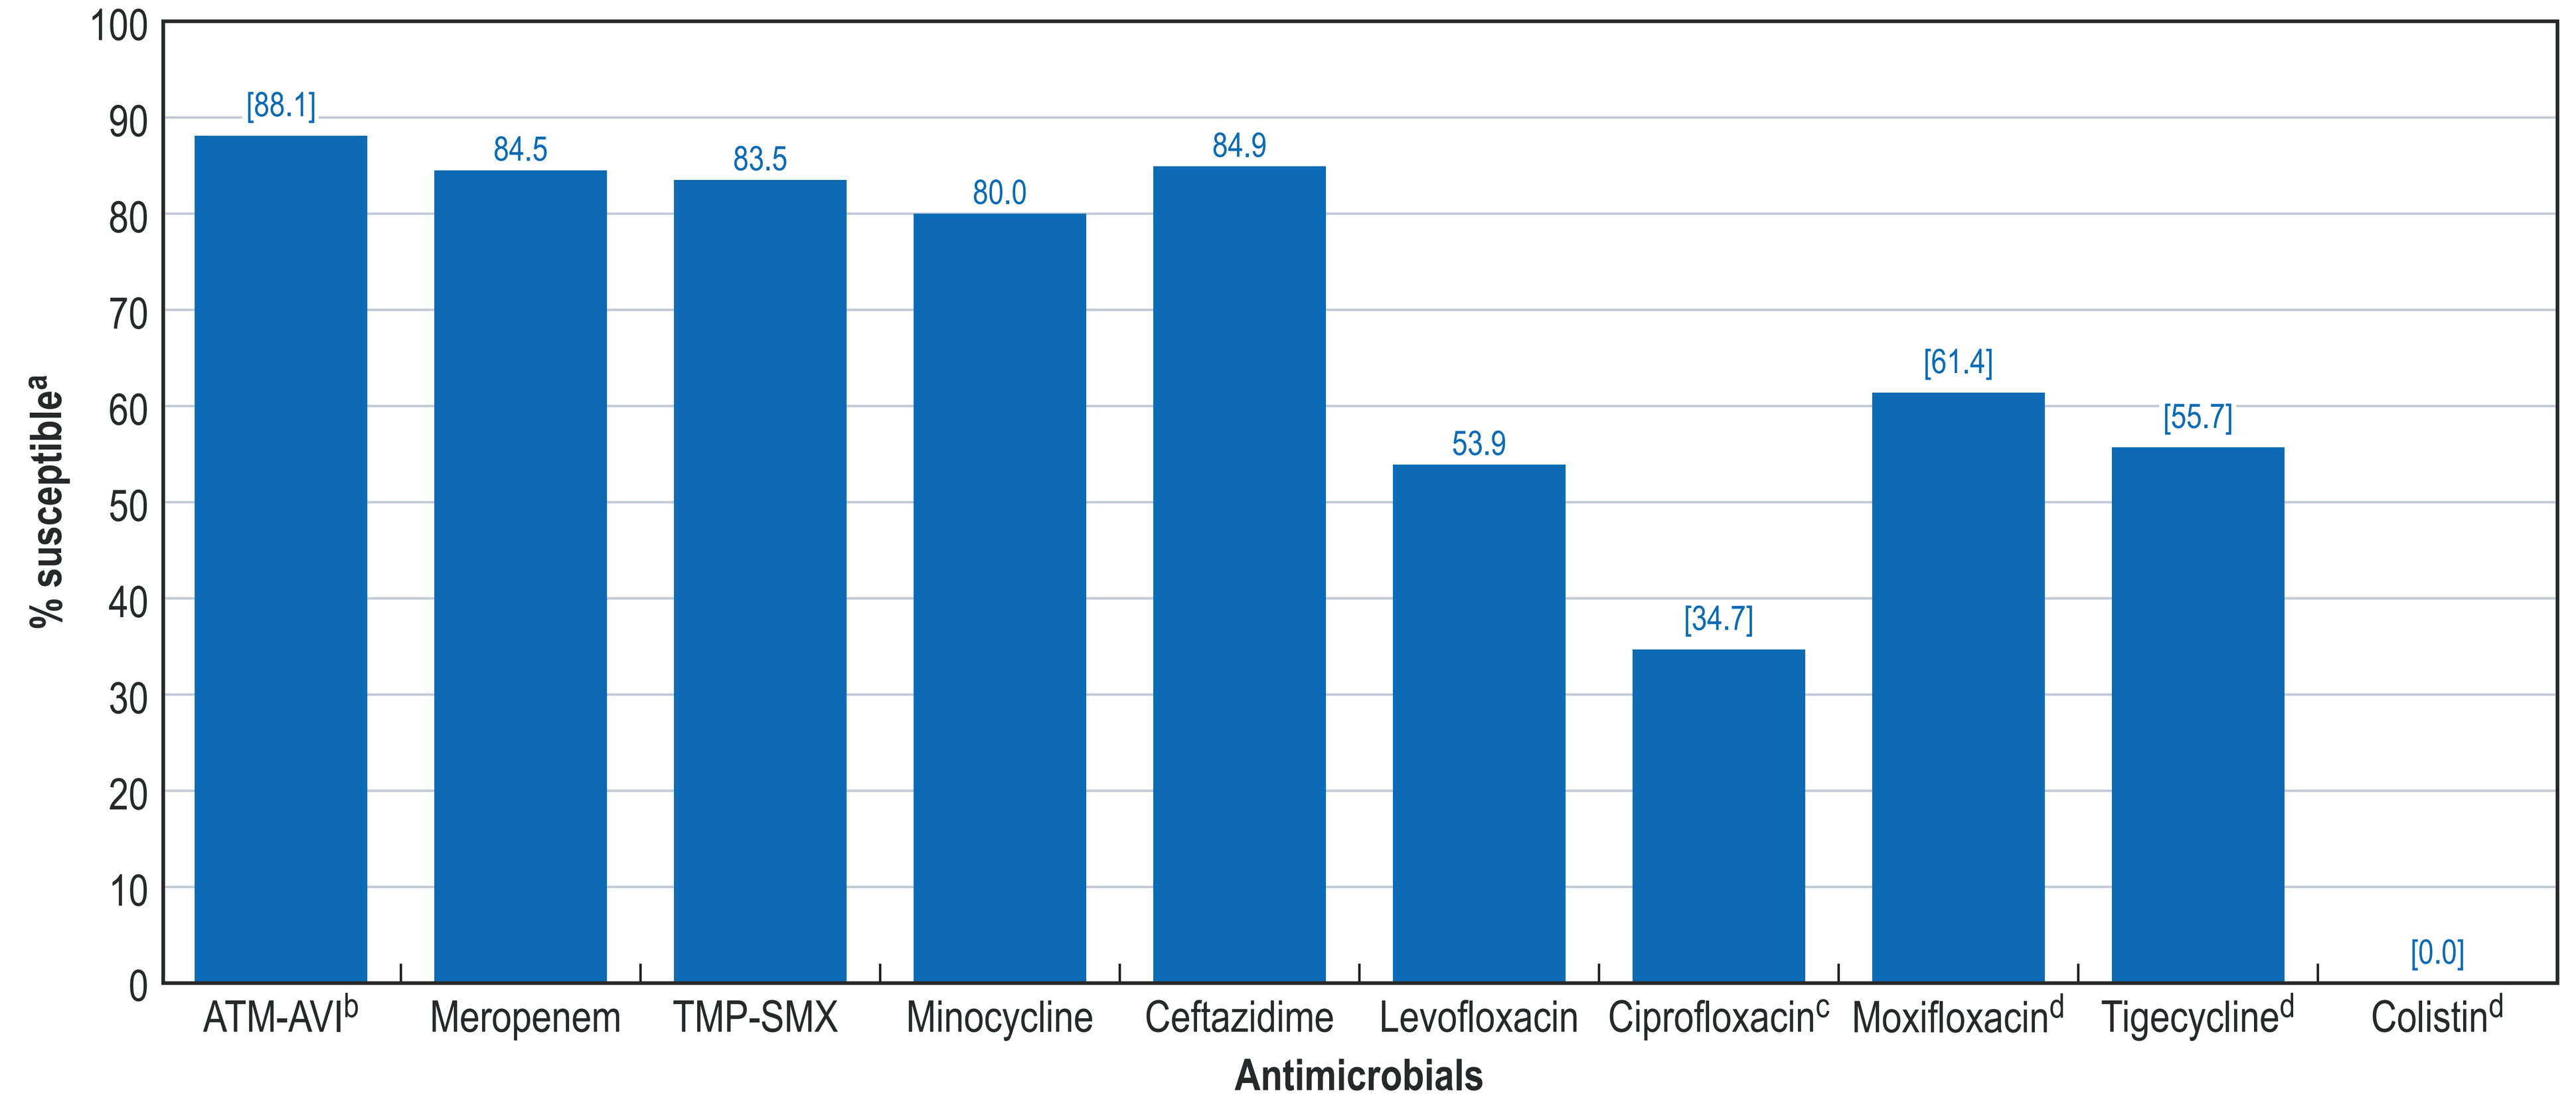
100
90
80
70
60
50
40
30
20
10
0
% susceptible[a]
[88.1]
84.5
83.5
80.0
84.9
53.9
[34.7]
[61.4]
[55.7]
[0.0]
ATM-AVI[b]
Meropenem
TMP-SMX
Minocycline
Ceftazidime
Levofloxacin
Ciprofloxacin[c]
Moxifloxacin[d]
Tigecycline[d]
Colistin[d]
Antimicrobials

[a] Criteria as published by CLSI (2022) and US FDA (2022) unless noted.
[b, c, d] Values in brackets indicate % inhibited at: ≤8 mg/L [(b)], ≤1 mg/L [(c)], and ≤2 mg/L [(d)] for comparison.
Abbreviations: ATM-AVI, aztreonam-avibactam; TMP-SMX, trimethoprim-sulfamethoxazole.

https://abbvie1.outsystemsenterprise.com/GMAEventPublications/Assets.aspx?ConferenceId=440

SCAN ME

https://www.jmilabs.com/data/posters/IDWeek2022_AztAviVsSMaltBcepacia.pdf

Contact Information

Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

Funding

This study was supported by AbbVie. HS Sader, D Shortridge, SJ Ryan Arends, CG Carvalhaes, RE Mendes, and M Castanheira are employees of JMI Laboratories, which was paid consultant to AbbVie in connection with the development of this poster.

Acknowledgments

The authors thank all the participant centers for their work in providing isolates.

References

1. CLSI. 2022. M100Ed32. Performance standards for antimicrobial susceptibility testing: 32nd informational supplement. Clinical and Laboratory Standards Institute, Wayne, PA.
2. Gibb J, Wong DW. 2021. Antimicrobial Treatment Strategies for *Stenotrophomonas maltophilia*: A Focus on Novel Therapies. Antibiotics (Basel) 10(10):1226.
3. Sfeir MM. 2018. *Burkholderia cepacia* complex infections: More complex than the bacterium name suggest. J Infect 18: 77(3):166-170.
4. Tamma PD, Aitken SL, Bonomo RA, Mathers AJ, van Duin D, Clancy CJ. 2022. IDSA guidance on the treatment of AmpC beta-lactamase-producing *Enterobacterales*, carbapenem-resistant *Acinetobacter baumannii*, and *Stenotrophomonas maltophilia* infections. Clin. Infect. Dis. 10: 2089-114.

abbvie