IDWeek 2023 | Poster #2756

Activity of Aztreonam-avibactam and Ceftazidime-avibactam against United States Isolates Producing β-lactamases (2020–2021)

Mariana Castanheira, Valerie Kantro, Timothy B. Doyle, Rodrigo E. Mendes, Helio S. Sader

JMI Laboratories, North Liberty, Iowa, USA

CONCLUSIONS

ESBLs, mainly CTX-M enzymes, were prevalent among isolates collected in US hospitals.

These isolates display resistance against many β-lactams and comparators, but they are still predominantly susceptible to carbapenems and new β-lactam/β-lactamase inhibitor combinations.

Aztreonam-avibactam was the most active agent against carbapenemase producers.

Avibactam combinations were active against common β-lactamase–producing isolates from US hospitals, including carbapenemase-producing isolates for which therapeutic options are limited.

Contact Information

Mariana Castanheira, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Mariana.Castanheira@element.com

Funding

This study was supported by Abbvie. Authors are employees of JMI Laboratories, which was paid consultant to Abbvie in connection with the development of this poster.

Acknowledgements

The authors thank the participant sites of the INFORM Program for the providing isolates.

References

1. CLSI. M07Ed11. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically: eleventh edition. Wayne, PA, Clinical and Laboratory Standards Institute, 2018.
2. CLSI. M100Ed33. Performance standards for antimicrobial susceptibilty testing: 33nd informational supplement. Wayne, PA, Clinical and Laboratory Standards Institute, 2023.
3. Castanheira M, Simner PJ, Bradford PA. Extended-spectrum β-lactamases: an update on their characteristics, epidemiology and detection. JAC Antimicrob Resist. 2021 Jul 16;3(3).
4. Castanheira M, Kimbrough JH, DeVries S, Mendes RE, Sader HS. Trends of β-Lactamase Occurrence Among *Escherichia coli* and *Klebsiella pneumoniae* in United States Hospitals During a 5-Year Period and Activity of Antimicrobial Agents Against Isolates Stratified by β-Lactamase Type. Open Forum Infect Dis. 2023 Jan 27;10(2).
5. Tamma PD, Aitken SL, Bonomo RA, Mathers AJ, van Duin D, Clancy CJ. Infectious Diseases Society of America 2023 Guidance on the Treatment of Antimicrobial Resistant Gram-Negative Infections. Clin Infect Dis. 2023 Jul 18: Epub ahead of print.

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie ID Week 2023 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2023_22-ALG-04_P2_ATM-AVI_BLs.pdf

SCAN ME

To submit a medical question, please visit www.abbviemedinfo.com

INTRODUCTION

- β-lactams are the most used antimicrobial class for the treatment of a broad range of infections; however, the use of these agents is compromised by isolates producing β-lactamases.
- Unfortunately, isolates producing β-lactamases are widespread.
- The strategies to overcome the presence of β-lactamases include developing new β-lactam agents that are stable in the presence of these enzymes or pairing existing agents with β-lactamase inhibitors that bind to these enzymes, allowing the β-lactam agent to reach its target.
- We evaluated the activity of the β-lactam/β-lactamase inhibitor combinations ceftazidime-avibactam and aztreonam-avibactam as well as their comparator agents against isolates producing common β-lactamases detected in US hospitals during 2020–2021.

Figure 1. Occurrence of β-lactamases among *E. coli*, *K. pneumoniae*, *Citrobacter* spp. and *E. cloacae* species complex isolates showing elevated MIC values to cephalosporin compounds

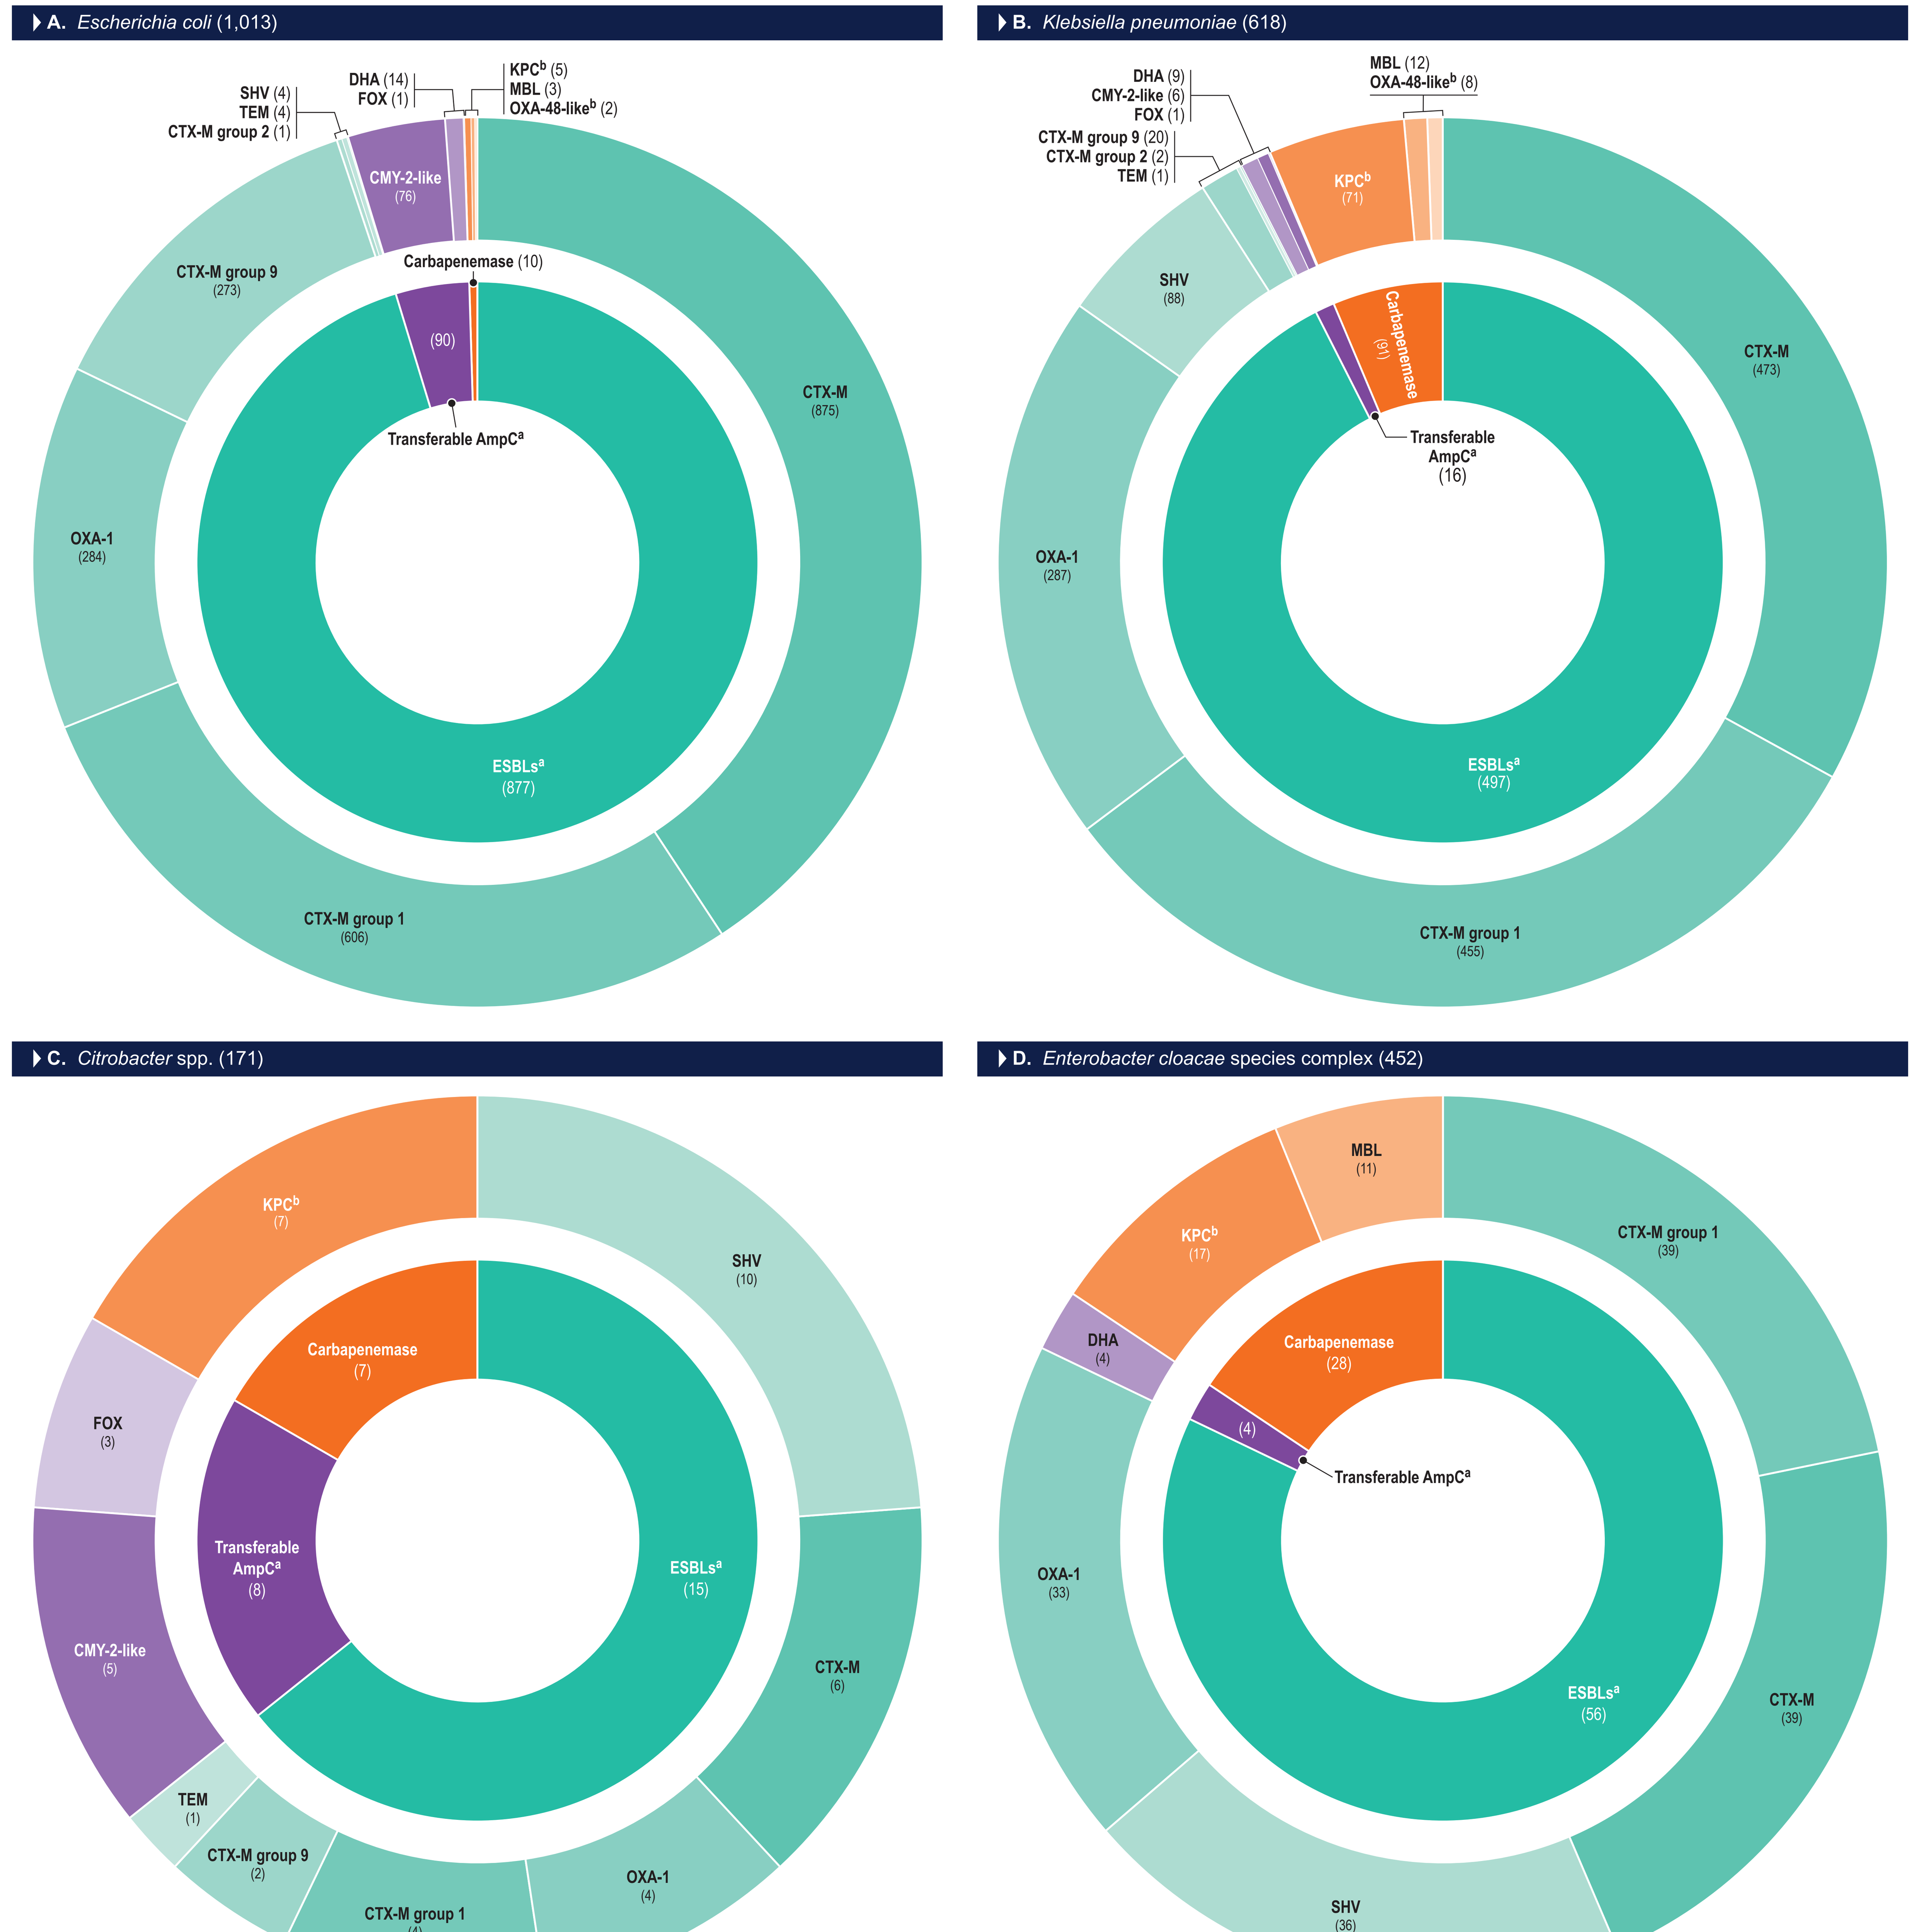
A. *Escherichia coli* (1,013)
KPC[b] (5)
MBL (3)
OXA-48-like[b] (2)
DHA (14)
FOX (1)
SHV (4)
TEM (4)
CTX-M group 2 (1)
CMY-2-like (76)
CTX-M group 9 (273)
Carbapenemase (10)
(90)
Transferable AmpC[a]
OXA-1 (284)
CTX-M (875)
ESBLs[a] (877)
CTX-M group 1 (606)
B. *Klebsiella pneumoniae* (618)
MBL (12)
OXA-48-like[b] (8)
DHA (9)
CMY-2-like (6)
FOX (1)
CTX-M group 9 (20)
CTX-M group 2 (2)
TEM (1)
KPC[b] (71)
SHV (88)
Carbapenemase (91)
CTX-M (473)
Transferable AmpC[a] (16)
OXA-1 (287)
ESBLs[a] (497)
CTX-M group 1 (455)
C. *Citrobacter* spp. (171)
KPC[b] (7)
SHV (10)
Carbapenemase (7)
FOX (3)
Transferable AmpC[a] (8)
ESBLs[a] (15)
CMY-2-like (5)
CTX-M (6)
TEM (1)
CTX-M group 9 (2)
CTX-M group 1 (4)
OXA-1 (4)
D. *Enterobacter cloacae* species complex (452)
MBL (11)
KPC[b] (17)
CTX-M group 1 (39)
DHA (4)
Carbapenemase (28)
(4)
Transferable AmpC[a]
OXA-1 (33)
ESBLs[a] (56)
CTX-M (39)
SHV (36)

[a] Does not include carbapenemase-producing isolates
[b] Does not include MBL-carrying isolates

MATERIALS AND METHODS

- A total of 21,853 Enterobacterales isolates were collected during 2020–2021 in 71 US hospitals.
 - Isolates were identified as the cause of infection.
 - Isolates were limited to 1 per patient episode.
- Isolates were susceptibility tested against meropenem-vaborbactam, ceftazidime-avibactam, and comparator agents using the reference broth microdilution method as described by the Clinical and Laboratory Standards Institute (CLSI) M07 (2018) and M100 (2022) documents.
 - Avibactam was tested at a fixed concentration of 4 mg/L.
 - Vaborbactam was tested at a fixed concentration of 8 mg/L.
- Quality control (QC) was performed according to the CLSI M100 (2022) criteria.
 - All QC MIC results were within acceptable ranges.
- Categorical interpretations for all comparator agents were those criteria found in the CLSI M100 (2022) or the US Food and Drug Administration (FDA) website.
- Isolates submitted to whole genome sequencing were:
 - *Escherichia coli* (n=1,013) and *Klebsiella pneumoniae* (n=618) displaying MIC values ≥2 mg/L for at least 2 of the following: ceftazidime, ceftriaxone, aztreonam, or cefepime.
 - *Enterobacter cloacae* species complex (*E. cloacae*; n=452) and *Citrobacter* spp. (n=171) displaying MIC values ≥16 mg/L for ceftazidime and/or ≥2 mg/L for cefepime.
 - Enterobacterales (n=240) displaying meropenem and/or imipenem MIC results at >1 mg/L.
- WGS was performed on a MiSeq (Illumina, San Diego, CA) instrument targeting a 30X coverage.
 - Sequences were *de novo* assembled.
 - Analysis of β-lactam resistance mechanisms was performed *in silico.*
 - Genes encoding resistances were searched using a curated library and a criteria of >94% sequencing identity and 40% minimum length coverage was applied.

RESULTS

- A total of 1,446 isolates including 877 *E. coli*, 497 *K. pneumoniae*, 15 *Citrobacter* spp., and 56 *E. cloacae* carried ESBL genes without carbapenemases (Figure 1).
 - CTX-M was the most common ESBL detected (1,349 isolates) and among these enzymes CTX-M Group 1, which includes CTX-M-15, was the dominant type (1,061 isolates).
 - CTX-M Group 9, which includes CTX-M-9, CTX-M-14 and CTX-M-27, was noted among 294 isolates.
 - SHV ESBLs were detected among 101 isolates, mostly *K. pneumoniae.*
- Ceftazidime-avibactam inhibited all ESBL producers while meropenem-vaborbactam inhibited 99.8% to 100% and ceftolozane-tazobactam inhibited 67 to 95.9% of these isolates (Figure 2).
 - Aztreonam-avibactam inhibited ≥99.9% (using a PK/PD breakpoint of 8 mg/L) of the isolates regardless of the ESBL type or organism.
 - Meropenem susceptibility rates against ESBLs ranged from 98 to 100%.
 - Among other classes, amikacin and tigecycline were the most active agents, inhibiting 78.1% and 97.7% of the ESBL-producing isolates.
 - A total of 97.7% of the ESBL-producing isolates had intermediate colistin MIC values.
- A total of 111 isolates harbored genes encoding transferrable AmpCs, including 82 isolates producing CMY-2.
- All isolates carrying transferrable AmpC genes were susceptible to ceftazidime-avibactam and meropenem-vaborbactam.
 - Aztreonam-avibactam and meropenem inhibited 99.1% and 99.1% of these isolates, respectively.
 - Ceftolozane-tazobactam only inhibited 79.1% of the isolates carrying transferrable AmpC enzymes.
- Among *Enterobacterales* screened, 165 carried carbapenemases.
 - Aztreonam-avibactam was the only agent that inhibited all (100.0%) of the isolates carrying carbapenemases
 - KPC-3 and KPC-2 were the most common carbapenemases, followed by NDM-1 (Table 1).
 - Ceftazidime-avibactam and meropenem-vaborbactam susceptibility rates were 81.2% and 80.6%.
 - Ceftolozane-tazobactam and all other β-lactam agents had limited activity against carbapenemase producers (1.8% to 10.3% susceptible; Figure 2).
 - The only comparator displaying activity against these isolates was tigecycline (93.3% susceptible).

Figure 2. Susceptibility patterns of antimicrobial agents against common β-lactamase–producing Enterobacterales isolates from US hospitals

Antimicrobial agent	ESBL producers (1,446)	CTX-M producers (1,349)	CTX-M Group 1 (1,061)	CTX-M Group 9 (294)	SHV ESBLs (101)	Transferable AmpC (111)	CMY-2-producers (82)	Carbapenemase-producers (165)
Ceftazidime-avibactam	100.0	100.0	100.0	100.0	100.0	100.0	100.0	81.2
Aztreonam-avibactam	99.9	99.9	99.9	100.0	100.0	99.1	98.8	100.0
Meropenem-vaborbactam	99.8	99.8	99.8	100.0	100.0	100.0	100.0	80.6
Ceftolozane-tazobactam	87.6	89.4	87.5	95.9	67.0	79.1	80.5	5.5
Piperacillin-tazobactam	66.6	68.2	61.5	92.5	46.0	62.7	65.9	4.2
Aztreonam	6.4	5.9	2.5	17.7	8.9	27.9	15.9	4.2
Ceftriaxone	0.3	0.0	0.0	0.0	2.0	5.4	0.0	1.8
Ceftazidime	15.4	15.8	6.1	50.7	6.9	2.7	2.4	4.2
Cefepime	5.7	3.0	2.5	4.4	37.6	80.2	82.9	6.7
Meropenem	98.9	99.0	98.9	100.0	98.0	99.1	98.8	10.3
Imipenem	99.5	99.6	99.6	100.0	99.0	98.2	97.6	4.8
Levofloxacin	25.0	24.6	26	19.5	33.0	56.8	64.6	20.6
Gentamicin	57.4	58.1	52.4	78.9	45.5	83.8	86.6	47.3
Amikacin	78.1	77.7	73.1	94.2	85.1	92.8	93.9	53.3
Trimethoprim-sulfamethoxazole	29.2	28.2	26.8	33.3	43.0	59.5	68.3	29.7
Tigecycline[b]	97.7	97.8	97.3	99.7	96.0	98.2	100.0	93.3
Colistin[a]	97.7	98.3	97.9	99.3	89.0	98.2	100.0	84.1

0 100
% of susceptibility using CLSI breakpoints
([a] % intermediate for colistin/ [b] US FDA breakpoint for tigecycline)

Table 1. Carbapenemases detected among 165 carbapenem non-susceptible Enterobacterales isolates carrying these enzymes submitted to β-lactamase screening

	No. of isolates by organism									
Carbapenemases	*E. coli*	*K. pneumoniae*	*Citrobacter* spp.	*E. cloacae* species complex	*K. aerogenes*	*K. oxytoca*	*P. mirabilis*	*P. rettgeri*	*S. marcescens*	*Raoultella* spp.
OXA-48–like	5	20				4	1	3		
OXA-181	2	1								
OXA-232		2								
OXA-48		5								
MBL	3	12		11		4	1	3		
IMP-13						1				
IMP-27							1			
IMP-4		1				1				
IMP-4, KPC-3						1				
KPC-3, NDM-5						1				
NDM-1		3		11				3		
NDM-1, OXA-181		1								
NDM-5	2	5								
NDM-5–like	1									
NDM-5, OXA-181		2								
KPC	5	71	7	17	2	8			6	2
KPC-2	3	23	2	6		3			1	
KPC-3	1	48	4	7	2	5			5	2
KPC-4	1		1	3						
KPC-6				1						
Other serine-carbapenemase									3	
SME-2									3	