IDWeek 2023 | Poster #2146

Antimicrobial Activity of Aztreonam-avibactam, Ceftazidime-avibactam, and Meropenem-vaborbactam against Enterobacterales causing Bloodstream Infection in US Medical Centers (2020–2022)

Helio S. Sader, Cecilia Carvalhaes, John H. Kimbrough, Rodrigo E. Mendes, Mariana Castanheira

JMI Laboratories, North Liberty, Iowa, USA

CONCLUSIONS

ATM-AVI demonstrated potent activity against a large collection of Enterobacterales isolated from patients with BSI in US hospitals, including CREs and isolates resistant to recently approved β-lactamase inhibitor combinations.

ATM-AVI may represent valuable treatment options for infections caused by MBL-producing Enterobacterales.

The results of this investigation support further clinical development of ATM-AVI.

INTRODUCTION

- As the frequency of Enterobacterales producing metallo-β-lactamase (MBL) and/or OXA-48 is increasing in some US medical centers, effective antimicrobials to treat the infections caused by these organisms are urgently needed.
- Aztreonam-avibactam (ATM-AVI) is under clinical development for treatment of infections caused by Gram-negative bacteria, including MBL producers.
- We evaluated the activities of ATM-AVI, ceftazidime-avibactam (CAZ-AVI), meropenem-vaborbactam (MEM-VAB), and comparators against Enterobacterales isolated from patients with bloodstream infections (BSIs).

Figure 1. ATM-AVI MIC distributions for Enterobacterales and CRE isolates

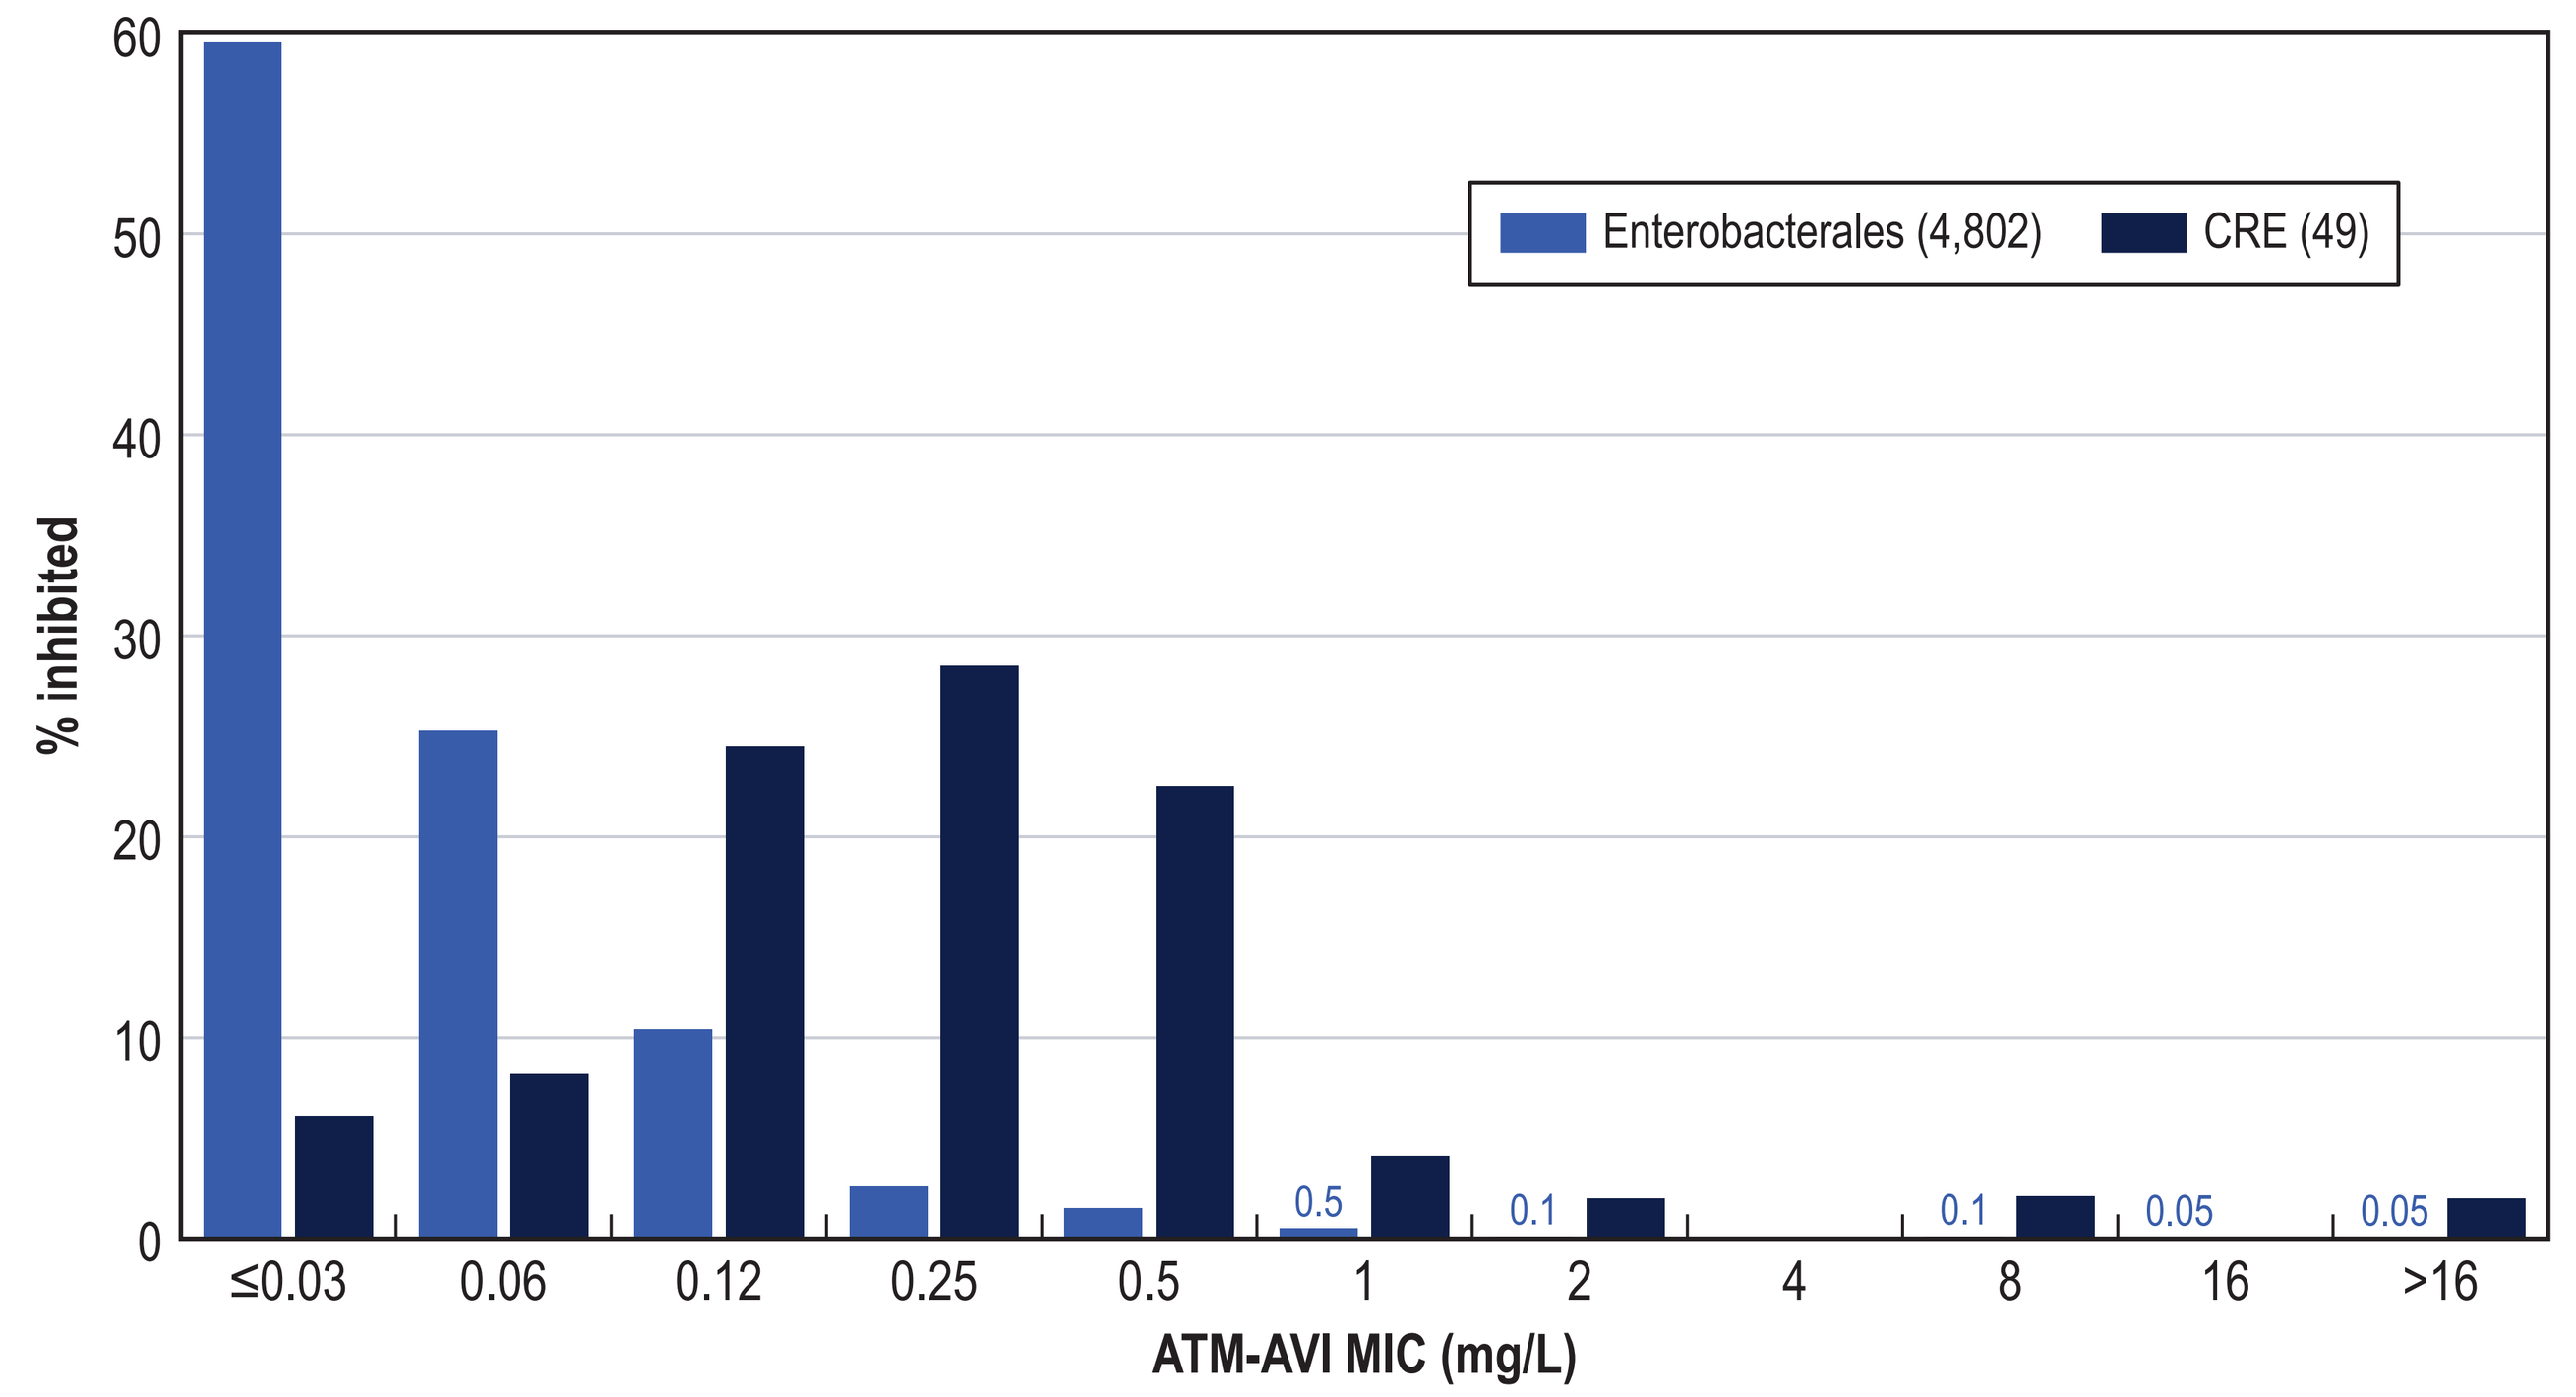
% inhibited
Enterobacterales (4,802)
CRE (49)
0.5
0.1
0.1
0.05
0.05
≤0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
>16
ATM-AVI MIC (mg/L)

Figure 2. Aztreonam-avibactam (ATM-AVI), ceftazidime-avibactam (CAZ-AVI), and meropenem-vaborbactam (MEM-VAB) against Enterobacterales and selected resistant subsets

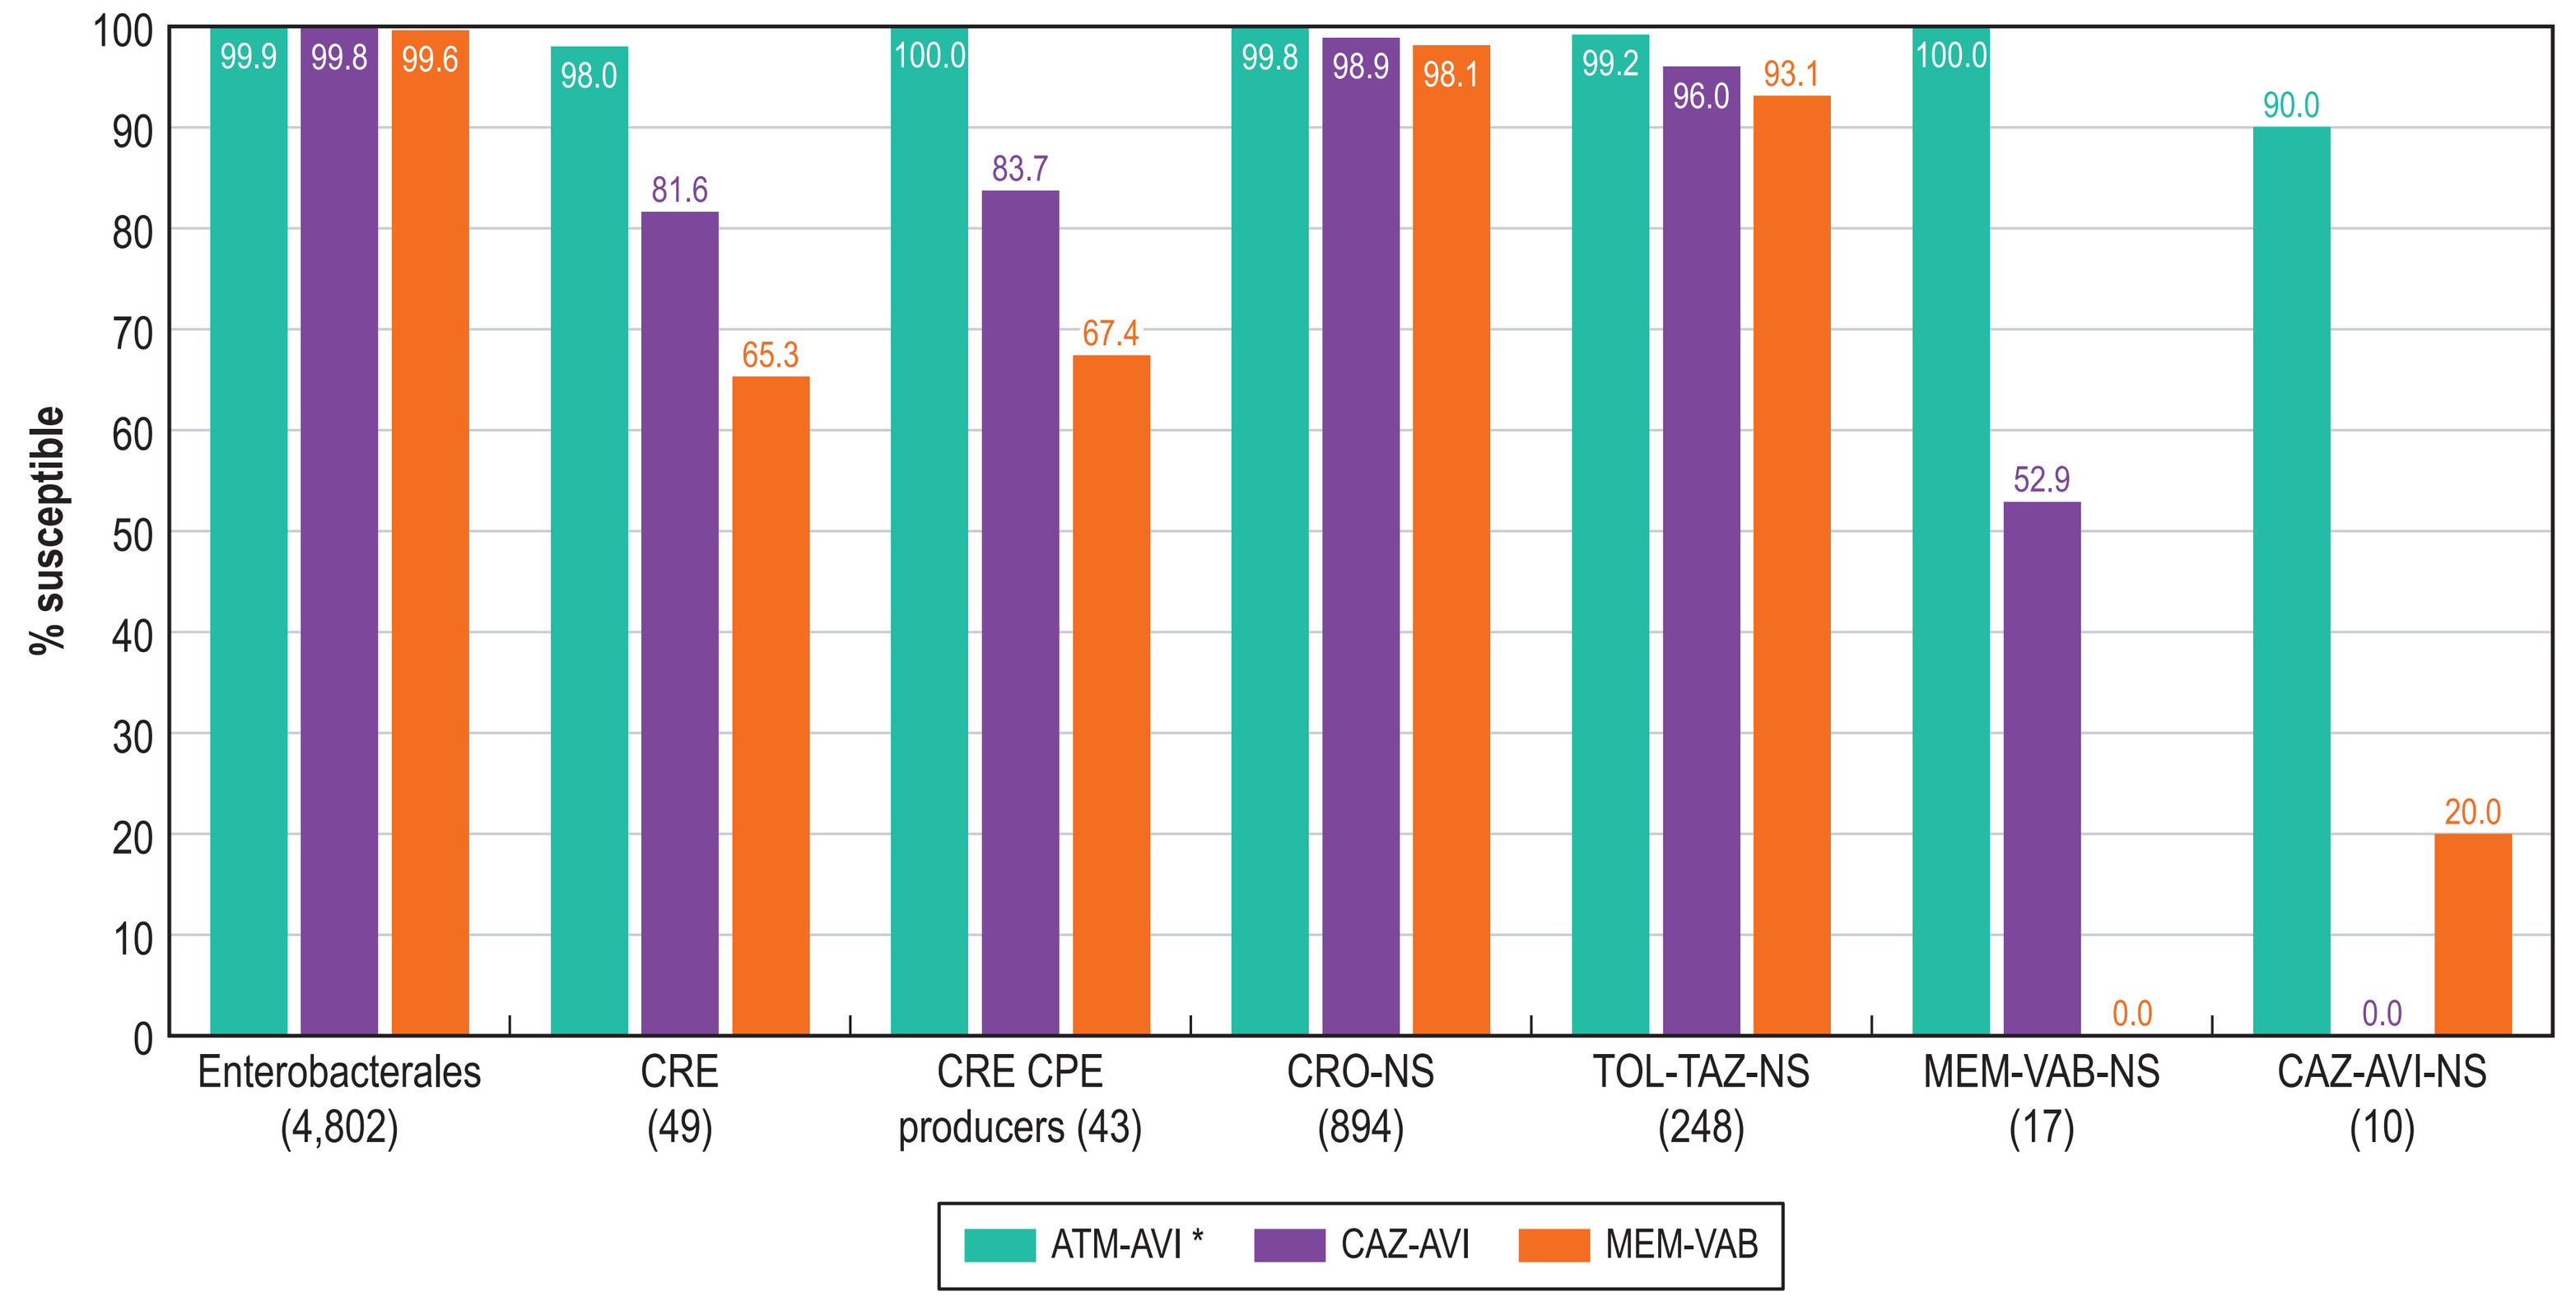
% susceptible
99.9
99.8
99.6
98.0
81.6
65.3
100.0
83.7
67.4
99.8
98.9
98.1
99.2
96.0
93.1
100.0
52.9
0.0
90.0
0.0
20.0
Enterobacterales (4,802)
CRE (49)
CRE CPE producers (43)
CRO-NS (894)
TOL-TAZ-NS (248)
MEM-VAB-NS (17)
CAZ-AVI-NS (10)
ATM-AVI *
CAZ-AVI
MEM-VAB

% inhibited at ≤8 mg/L.
Abbreviations: CRE, carbapenem-resistant Enterobacterales; CPE, carbapenemase; CRO, ceftriaxone; NS, nonsusceptible; and TOL-TAZ, ceftolozane-tazobactam.

Figure 3. Frequency of carbapenemases among CRE isolates from patients hospitalized with bloodstream infections

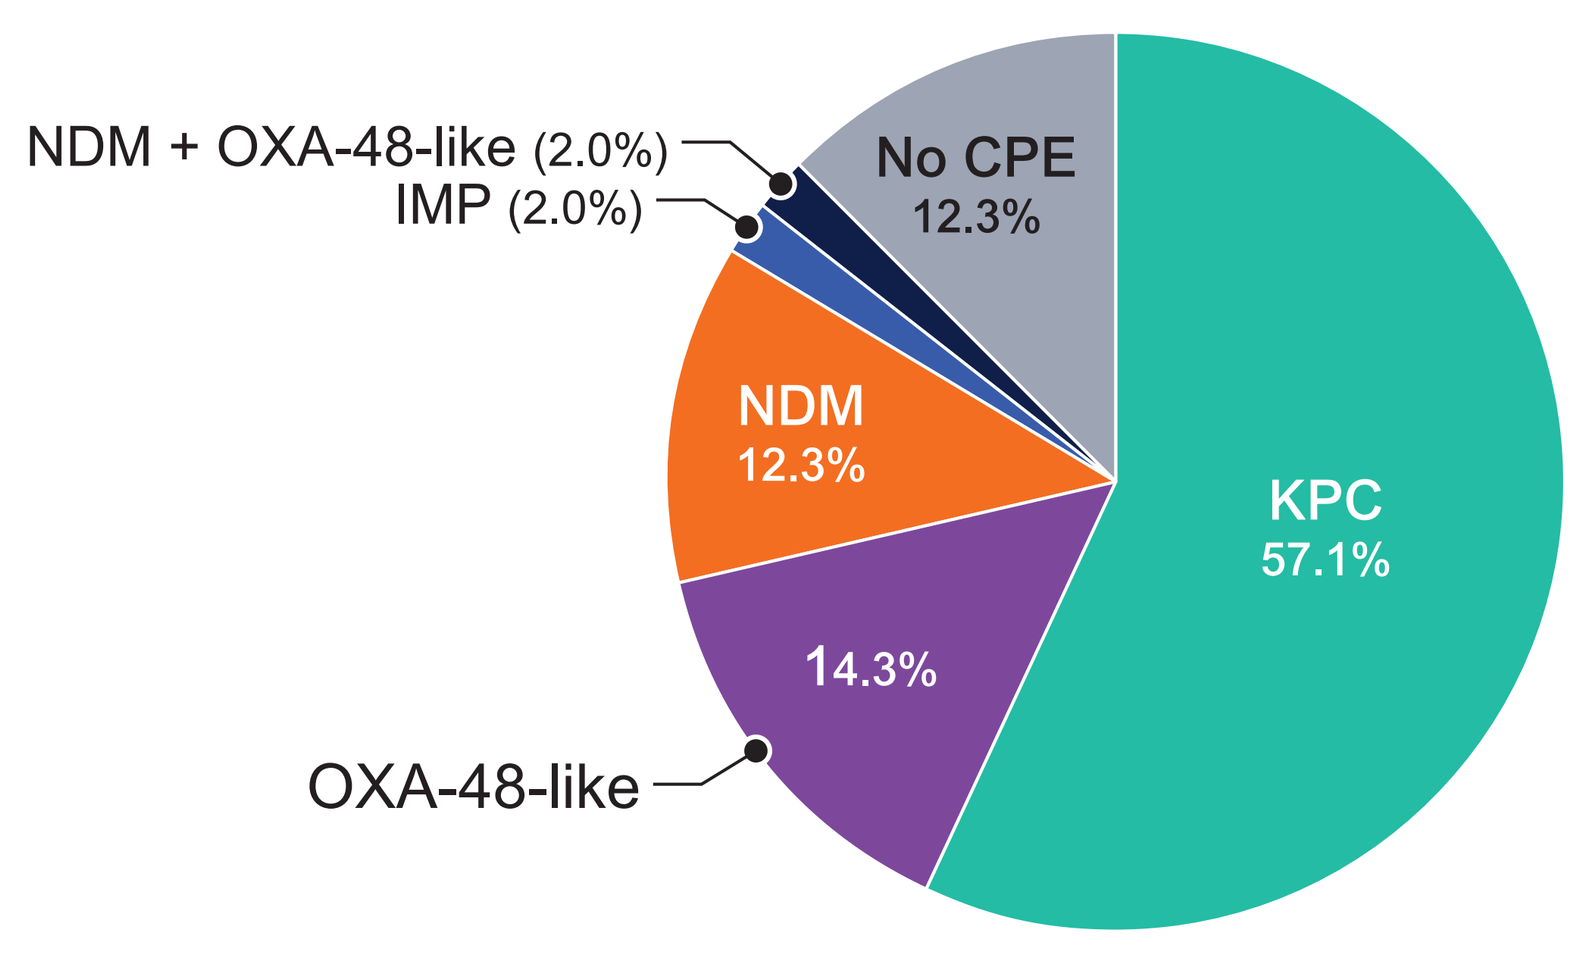
NDM + OXA-48-like (2.0%)
IMP (2.0%)
No CPE
12.3%
NDM
12.3%
KPC
57.1%
14.3%
OXA-48-like

METHODS

- 4,802 Enterobacterales were consecutively collected (1/patient) from 72 US medical centers in 2020–2022 and susceptibility tested by CLSI broth microdilution method.
- ATM-AVI was tested with AVI at a fixed 4 mg/L.
- A pharmacokinetic/pharmacodynamic susceptible breakpoint of ≤8 mg/L was applied to ATM-AVI for comparison.
- Carbapenem-resistant Enterobacterales (CRE) isolates were screened for carbapenemase (CPE) by whole genome sequencing.

RESULTS

- ATM-AVI was highly active against Enterobacterales (Figure 1); only 2 isolates showed ATM-AVI MICs >8 mg/L: 1 *E. coli* (meropenem-susceptible) and 1 *K. aerogenes* (CRE).
- All CPE producers and 98.0% of CREs were inhibited at an ATM-AVI MIC of ≤8 mg/L (Figures 1 and 2).
- CAZ-AVI and MEM-VAB were active against 81.6% and 65.3% of CREs, respectively (Table 1 and Figure 2).
- Ceftolozane-tazobactam (TOL-TAZ) was active against only 72.6% of ceftriaxone (CRO)-nonsusceptible (NS) isolates (Table 1).
- ATM-AVI retained activity (MIC, ≤8 mg/L) against all MEM-VAB-NS and 90.0% of CAZ-AVI-NS isolates (Figure 2).
- The most common CPEs were KPC-2/3 (57.1% of CREs), OXA-48–like (16.3%), and NDM (14.3%; Figure 3).
- A CPE gene was not observed in 12.3% of CREs (Figure 3).
- CAZ-AVI and MEM-VAB were active against 100.0% of KPC producers, but CAZ-AVI showed limited activity against MBL producers and MEM-VAB showed limited activity against OXA-48–like and MBL producers (Figure 4).

Figure 4. Aztreonam-avibactam (ATM-AVI), ceftazidime-avibactam (CAZ-AVI), and meropenem-vaborbactam (MEM-VAB) against CRE isolates stratified by carbapenemase (CPE) type

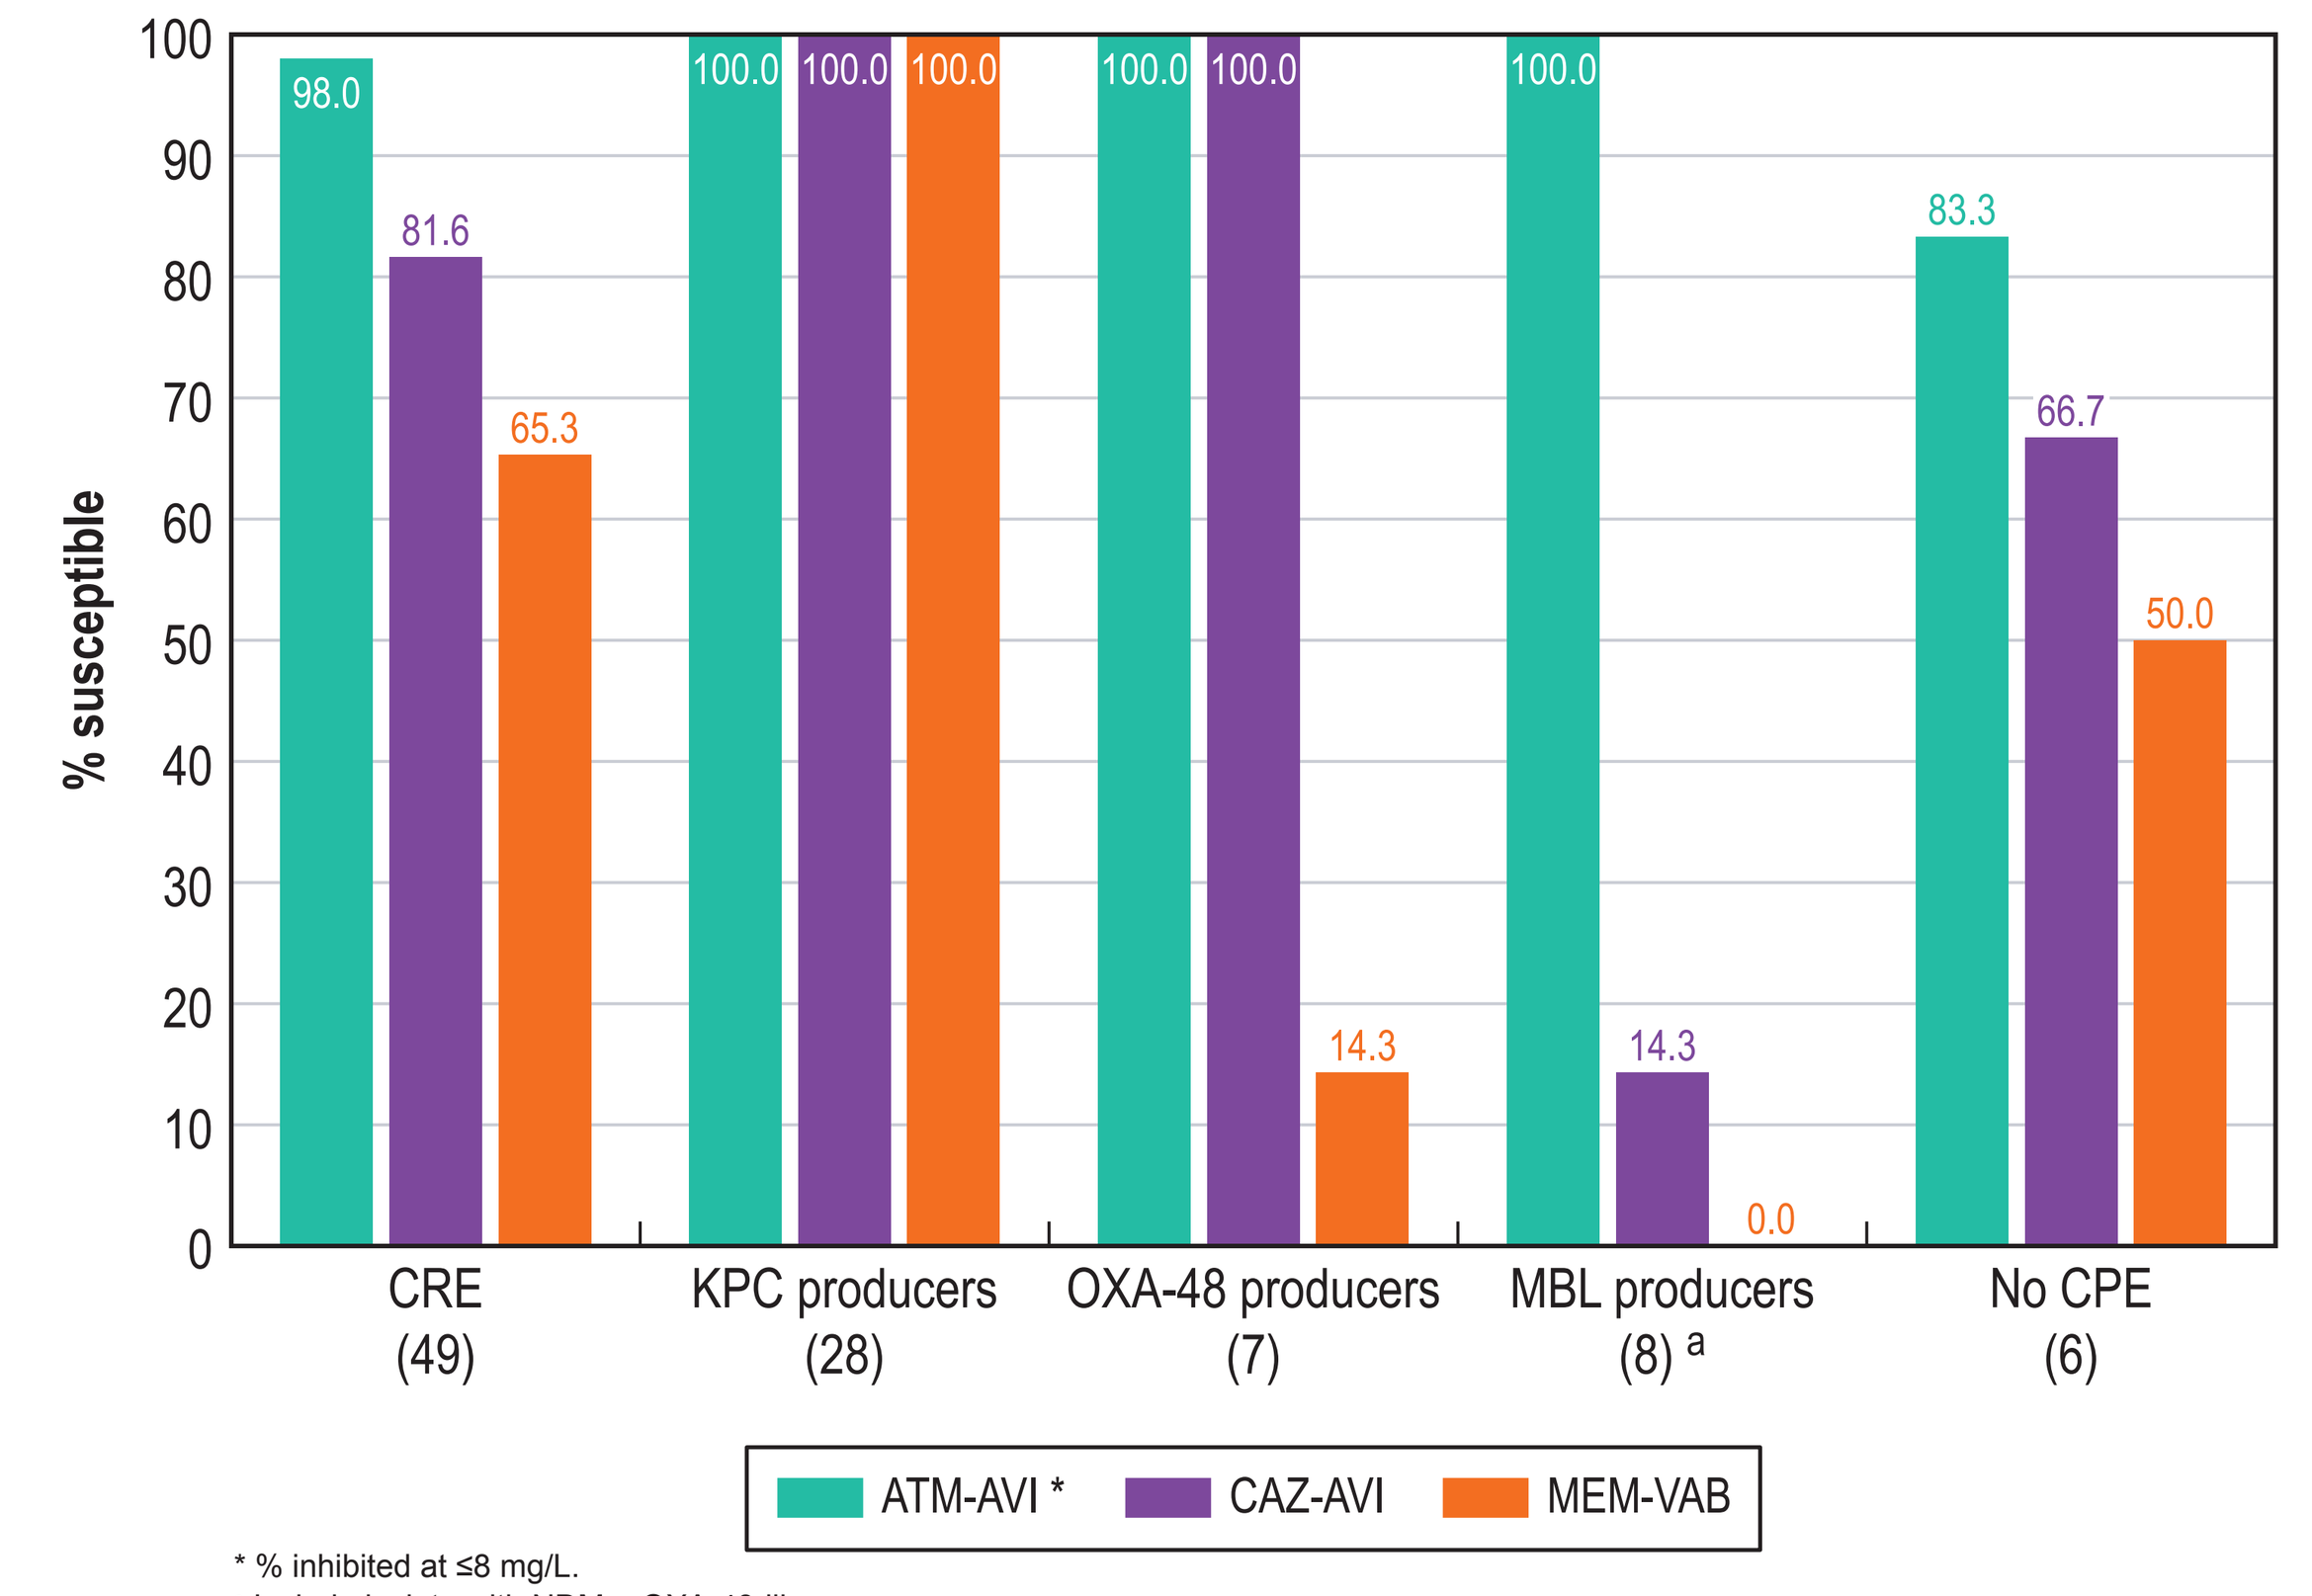
% susceptible
98.0
81.6
65.3
100.0
100.0
100.0
100.0
100.0
14.3
100.0
14.3
0.0
83.3
66.7
50.0
CRE (49)
KPC producers (28)
OXA-48 producers (7)
MBL producers (8) a
No CPE (6)
ATM-AVI *
CAZ-AVI
MEM-VAB

* % inhibited at ≤8 mg/L.
a Include isolate with NDM + OXA-48-like.

Table 1. Antimicrobial susceptibility of organisms from patients with bloodstream infections in US medical centers (2020–2022)

	% Susceptible per CLSI and or FDA criteria (no. of isolates)				
Antimicrobial agent	ENT (4,802)	CRE (49)	CPE (43)	CRO-NS (894)	MDR (437)
Aztreonam-avibactam	>99.9 [a]	98.0 [a]	100.0 [a]	99.8 [a]	99.5 [a]
Ceftazidime-avibactam	99.8	81.6	83.7	98.9	97.9
Meropenem-vaborbactam	99.6	65.3	67.4	98.1	96.3
Imipenem-relebactam	95.0 [b]	61.2 [b]	62.8	94.2 [b]	93.7 [b]
Ceftolozane-tazobactam	94.8	2.0	2.3	72.6	70.0
Piperacillin-tazobactam	88.7	2.0	2.3	53.4	40.4
Ceftriaxone	81.4	0.0	0.0	0.0	3.2
Ceftazidime	84.7	2.0	2.3	20.1	12.4
Cefepime	86.9	2.0	2.3	30.8	12.6
Meropenem	98.9	2.0	2.3	94.1	88.1
Imipenem	92.9 [b]	4.1 [b]	0.0 [b]	89.1 [b]	85.1 [b]
Levofloxacin	79.5	16.3	14.0	42.9	13.3
Gentamicin	90.2	49.0	44.2	66.5	34.0
Amikacin	93.4	44.9	41.9	82.3	63.8
Tigecycline	96.7	95.9	95.3	96.3	95.2

a % inhibited at ≤8 mg/L.
b All Enterobacterales species were included in the analysis, but CLSI excludes *Morganella*, *Proteus*, and *Providencia* species.

Contact Information

Helio S. Sader, MD, Ph.D., FIDSA
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie ID Week 2023 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2023_22-ALG-04_P4_ATM-AVI_BSI.pdf

To submit a medical question, please visit www.abbviemedinfo.com

SCAN ME

Funding

This study was supported by AbbVie. Helio S. Sader, Cecilia Carvalhaes, John H. Kimbrough, Rodrigo E. Mendes, and Mariana Castanheira are employees of JMI Laboratories, which was paid consultant to AbbVie in connection with the development of this poster.

Acknowledgements

The authors thank all the participant centers for their work in providing isolates.

References

1. CLSI. 2023. M100Ed33. Performance standards for antimicrobial susceptibility testing. Clinical and Laboratory Standards Institute, Wayne, PA.
2. Magiorakos AP, Srinivasan A, Carey RB, et al. 2012. Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance. Clin Microbiol Infect 18:268-281.
3. Sader HS, Mendes RE, Carvalhaes CG, Kimbrough JH, Castanheira M. 2023. HYPERLINK "https://pubmed.ncbi.nlm.nih.gov/36846612/" Changing epidemiology of carbapenemases among carbapenem-resistant Enterobacterales from United States hospitals and the activity of aztreonam-avibactam against contemporary Enterobacterales (2019-2021). Open Forum Infect Dis 10(2):ofad046.
4. Tamma PD, Aitken SL, Bonomo RA, Mathers AJ, van Duin D, Clancy CJ. 2022. IDSA guidance on the treatment of AmpC beta-lactamase-producing *Enterobacterales*, carbapenem-resistant *Acinetobacter baumannii*, and *Stenotrophomonas maltophilia* infections. Clin Infect Dis 10: 2089-114.