IDWeek 2023 | Poster #P2113

Evaluation of Isavuconazole Activity against *Aspergillus fumigatus* Causing Invasive Infections Worldwide Using the New CLSI Clinical Breakpoints

Cecilia G. Carvalhaes, Paul R. Rhomberg, Abby L. Klauer, Beth A. Hatch, Mariana Castanheira

Element Materials Technology (JMI Laboratories), North Liberty, Iowa, USA

Introduction

- Isavuconazole was approved by the US FDA in 2015 and is utilized as a first-line antifungal therapy for the treatment of invasive aspergillosis (IA).
- IA remains a serious, life-threatening infection, with *Aspergillus fumigatus* as the most frequently isolated species.
- The CLSI Subcommittee for Antifungal Susceptibility Testing recently approved clinical breakpoints for isavuconazole against *A. fumigatus*.
- The study objective was to evaluate the *in vitro* activity of isavuconazole and comparator antifungal agents by applying the new CLSI clinical breakpoints against a worldwide collection of *A. fumigatus* causing invasive infections.

Methods

- A total of 846 *A. fumigatus* isolates were collected as part of the 2017–2021 SENTRY Antifungal Surveillance Program from 44 medical centers located in North America (NA; *n*=282; 18 centers), Europe (EU; *n*=449; 17 centers), Asia-Pacific (APAC; *n*=102; 8 centers), and Latin America (LA, *n*=13; 1 center; Figure 1).
- Only 1 isolate per patient was included.
- Isolates were identified by MALDI-TOF MS and/or ITS and β-tubulin sequencing and tested by CLSI broth microdilution.
- CLSI interpretative criteria and epidemiological cutoff criteria (ECV) were applied, including the recently approved isavuconazole breakpoints against *A. fumigatus* (≤1 mg/L, susceptible; 2 mg/L, intermediate; and ≥4 mg/L, resistant; January 2023 meeting, CLSI).
- Posaconazole ECVs of 0.5 mg/L was used against *A. fumigatus*.
- *A. fumigatus* isolates showing non-wildtype MIC values for any azole were submitted to *cyp51* analysis by whole genome sequencing.

Results

A. fumigatus

- Overall, isavuconazole ($MIC_{50/90}$, 0.5/1 mg/L) showed similar activity to other azoles against *A. fumigatus* (Table 1), inhibiting 93.4% at ≤1 mg/L (CLSI-approved susceptible clinical breakpoint), regardless of the region (Figure 2).
- Voriconazole ($MIC_{50/90}$, 0.5/0.5 mg/L) inhibited 91.6% at its susceptible clinical breakpoint (Table 1).
- Itraconazole ($MIC_{50/90}$, 1/1 mg/L; 92.0% WT) and posaconazole ($MIC_{50/90}$, 0.25/0.5 mg/L; 97.2% WT) were also active against all *A. fumigatus* at their respective ECV criteria (Table 1).

Azole-NWT *A. fumigatus*

- Figure 3 displays the distribution of azole-NWT *A. fumigatus* isolates per region and surveillance year.
 - NA showed an increase in the azole-NWT rate over the years, while EU and APAC azole-NWT rates trended lower.
- The azole-NWT phenotype was detected in 88 *A. fumigatus* (10.4%).
 - NA showed the highest frequency of azole-NWT isolates (31; 11.0%), followed by EU (48; 10.7%) and APAC (9; 8.8%; Figure 3).
 - All *A. fumigatus* from LA were WT to azoles.
- Azole activity varied against azole-NWT isolates with and without CYP51 alterations (Table 1 and Figure 4).
- Applying the new isavuconazole CLSI breakpoints, 43.9% of the azole-NWT *A. fumigatus* without CYP51 alteration remained susceptible to isavuconazole and 73.2% were susceptible to voriconazole (Figure 4).
- Isavuconazole and voriconazole inhibited 46.7% and 43.3%, respectively, of azole-NWT *A. fumigatus* isolates displaying CYP51 alterations other than L98H/TR34 at the respective breakpoint (Figure 4).
- *A. fumigatus* isolates carrying L98H/TR34 (17 occurrences) in the CYP51A sequence displayed elevated MIC ranges for all azoles: isavuconazole, 2–>8 mg/L; voriconazole, 1–>8 mg/L; itraconazole, 2–>8 mg/L; and posaconazole, 0.5–4 mg/L.

Conclusions

- Isavuconazole exhibited potent *in vitro* activity against *A. fumigatus*, regardless of the region, when the new CLSI clinical breakpoints were applied.
- Azole-NWT *A. fumigatus* rates increased in NA but slightly decreased in EU and APAC regions.
- Azole activity varied against azole-NWT isolates with and without CYP51 alterations.
- Isavuconazole remained active against >40% of azole-NWT *A. fumigatus* isolates displaying wildtype CYP51 sequences or carrying CYP51 alterations other than L98H/TR34.

Funding

This study was supported by Pfizer Inc. (New York, NY). CG Carvalhaes, PR Rhomberg, AL Klauer, B Hatch, and M Castanheira were employees of Element Materials Technology (JMI Laboratories) at the time of this study, which was paid consultant to Pfizer in connection with the development of this poster.

Acknowledgments

The authors thank all the participant centers for their work in providing isolates.

Table 1. Activity of isavuconazole and comparator antifungal agents tested against 846 *A. fumigatus* isolates

Organism group (n)	ISC		VRC		ITC		PSC	
	$MIC_{50/90}$	%S[a]	$MIC_{50/90}$	%S[b]	$MIC_{50/90}$	%WT[c]	$MIC_{50/90}$	%WT[d]
A. fumigatus (846)	0.5/1	93.4	0.5/0.5	91.6	1/1	92.0	0.25/0.5	97.2
AZ-NWT (88)	2/8	36.4	1/2	48.9	2/8	22.7	0.5/1	77.0
WT *CYP51* (41)	2/2	43.9	0.5/1	73.2	2/2	36.6	0.5/0.5	90.0
NWT *CYP51* (47)	2/>8	29.8	1/4	27.7	2/>8	10.6	0.5/1	66.0
CYP51 alteration other than L98H/TR34 (30)	2/>8	46.7	1/4	43.3	2/8	16.7	0.5/1	83.3
North America (282)	0.5/1	94.0	0.5/0.5	90.8	1/1	91.8	0.25/0.5	97.5
Europe (449)	0.5/1	92.7	0.5/0.5	91.1	1/1	90.9	0.25/0.5	96.4
Asia-Pacific (102)	0.5/1	94.1	0.5/0.5	95.1	1/1	96.1	0.25/0.5	99.0
Latin America (13)	0.5/1	100.0	0.5/0.5	100.0	0.5/1	100.0	0.25/0.5	100.0

Abbreviations: AZ, azole; NWT, non-wildtype; ISC, isavuconazole; VRC, voriconazole; ITC, itraconazole; PSC, posaconazole.
[a] Isavuconazole breakpoint criteria was approved in the CLSI Jan 2023 meeting but should not be used for clinical laboratories while not published in the M38M51S document.
[b] CLSI (M38M51S, 2022) breakpoints applied.
[c] CLSI (M57S, 2022) epidemiological cut-off value applied.
[d] Posaconazole epidemiological cut-off value of 0.5 mg/L was applied as published by Pfaller et al., 2021.

Figure 1. Distribution *A. fumigatus* collected from 2017–2021 SENTRY surveillance program split by region

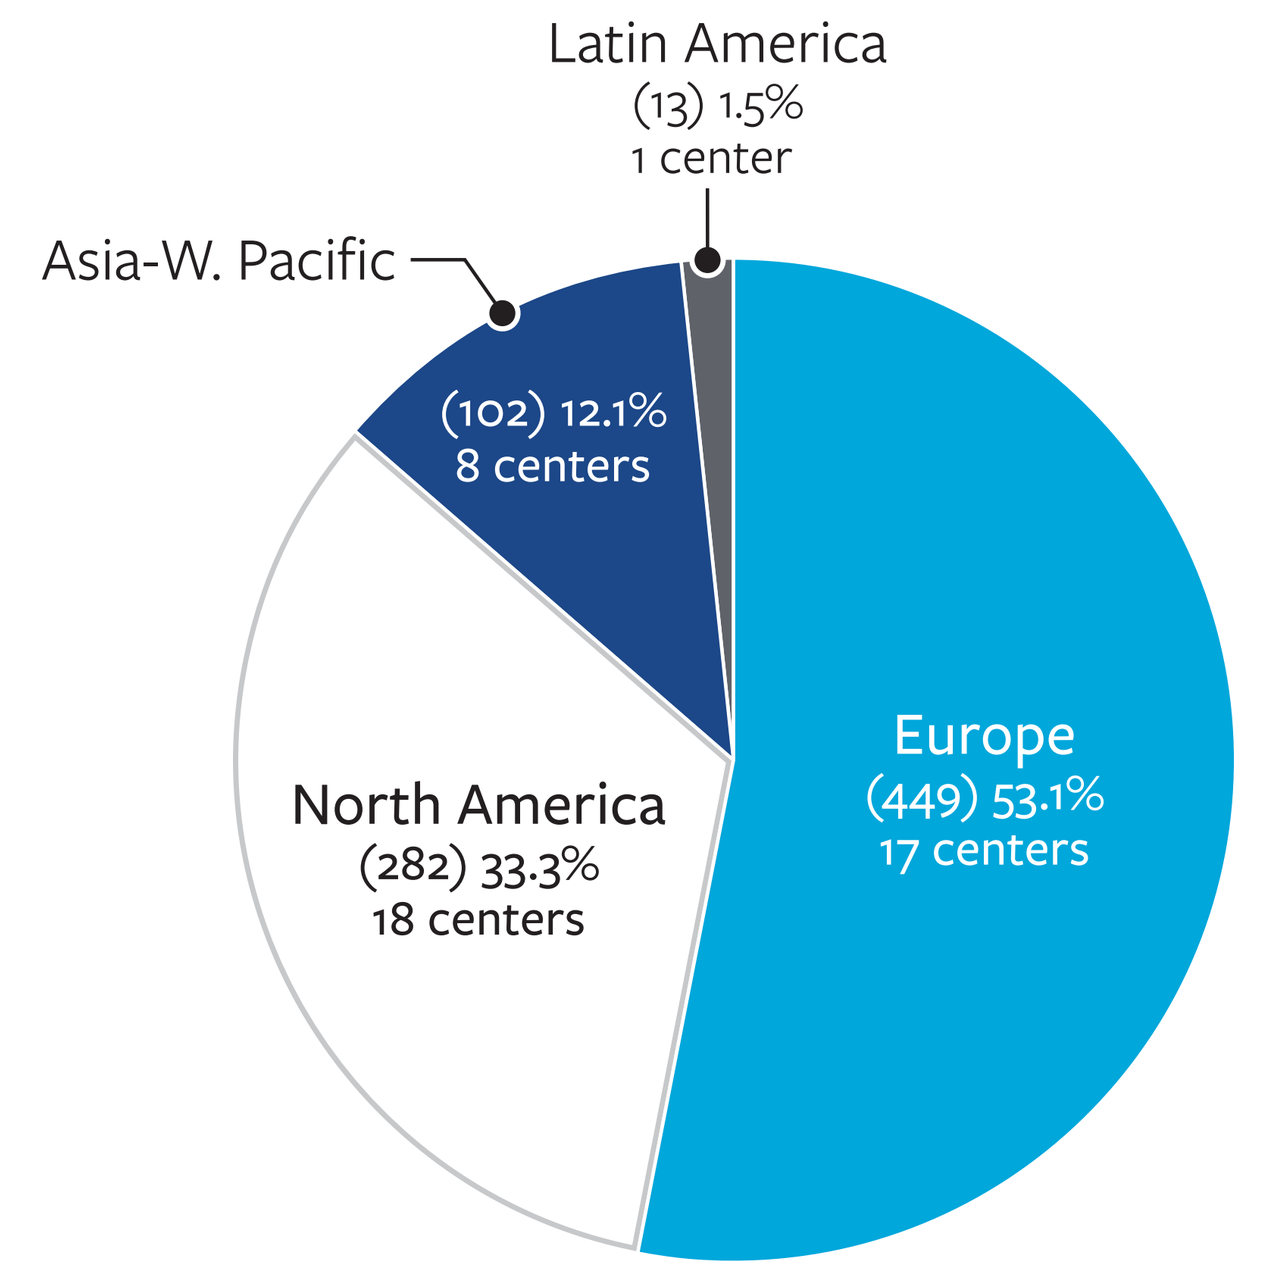

Latin America
(13) 1.5%
1 center
Asia-W. Pacific
(102) 12.1%
8 centers
North America
(282) 33.3%
18 centers
Europe
(449) 53.1%
17 centers

Figure 2. Activity of isavuconazole and other azoles against 846 *A. fumigatus* isolates split by region

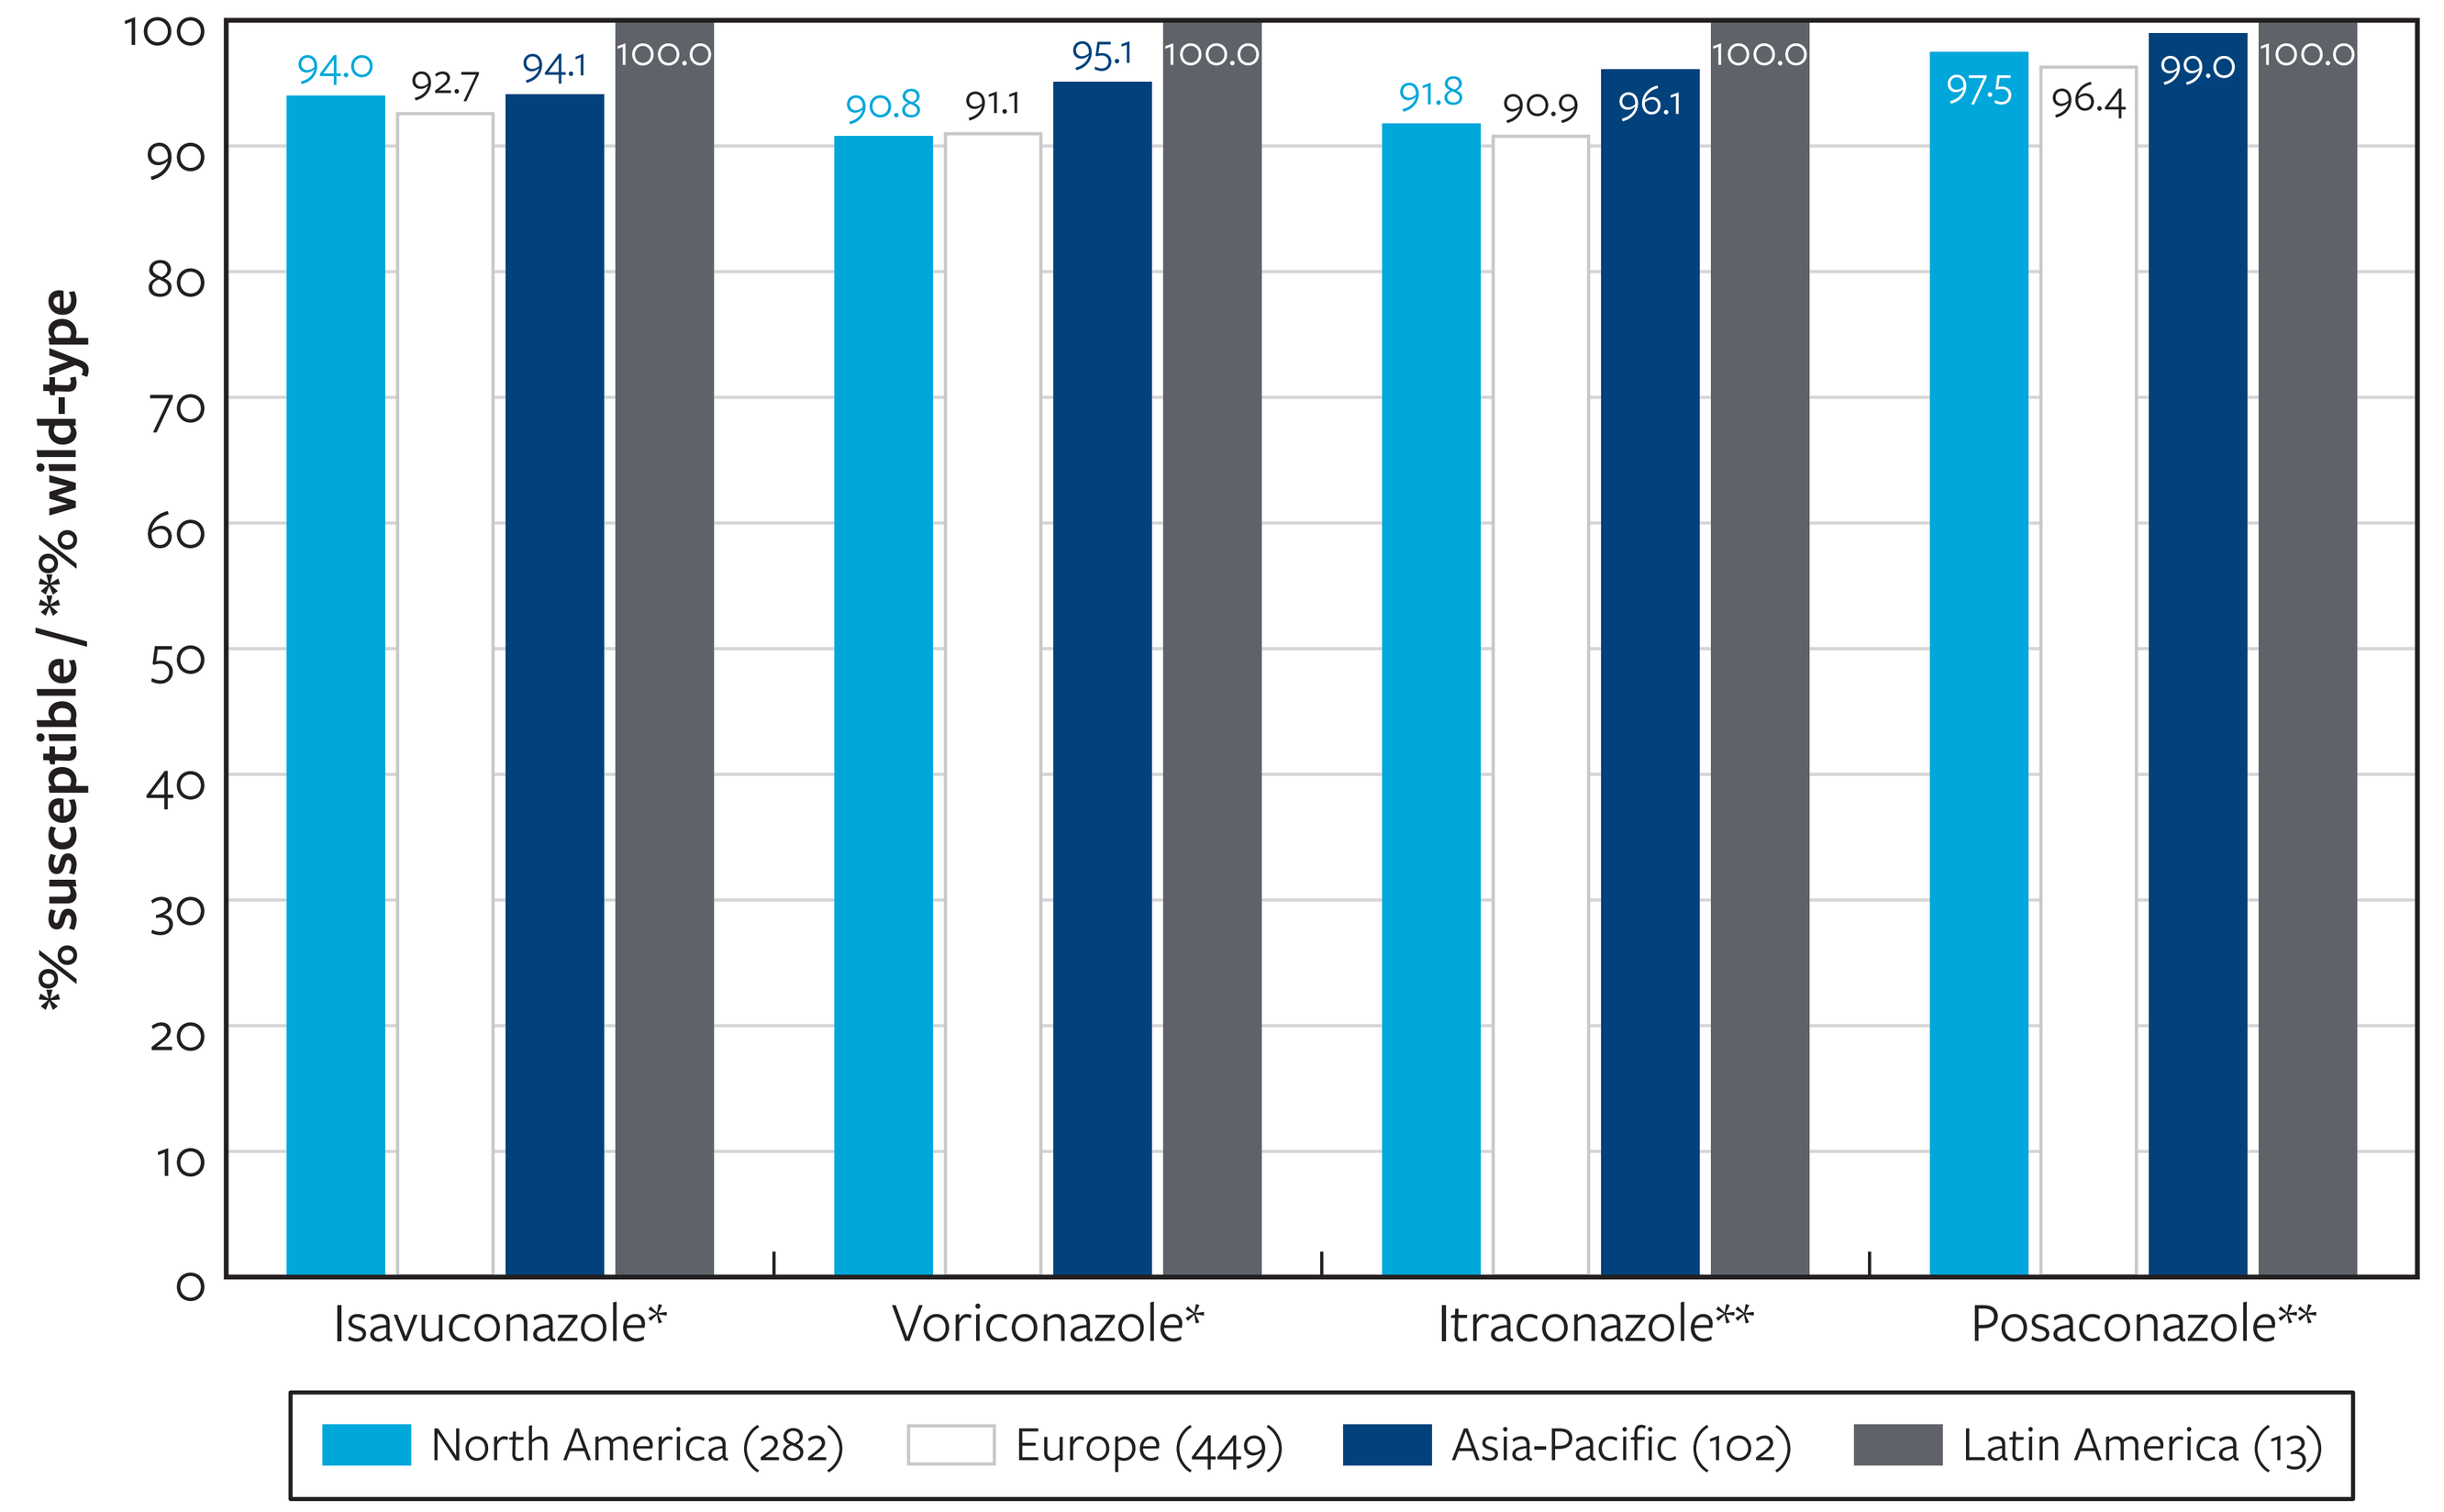

*% susceptible / **% wild-type
94.0
92.7
94.1
100.0
90.8
91.1
95.1
100.0
91.8
90.9
96.1
100.0
97.5
96.4
99.0
100.0
Isavuconazole*
Voriconazole*
Itraconazole**
Posaconazole**
North America (282)
Europe (449)
Asia-Pacific (102)
Latin America (13)

* Using CLSI M38M51S (2022) clinical breakpoint criteria.
** Using CLSI M57S (2022) epidemiological cutoff value criteria.

Figure 3. Trend of azole-NWT *A. fumigatus* isolates evaluated by region and surveillance year

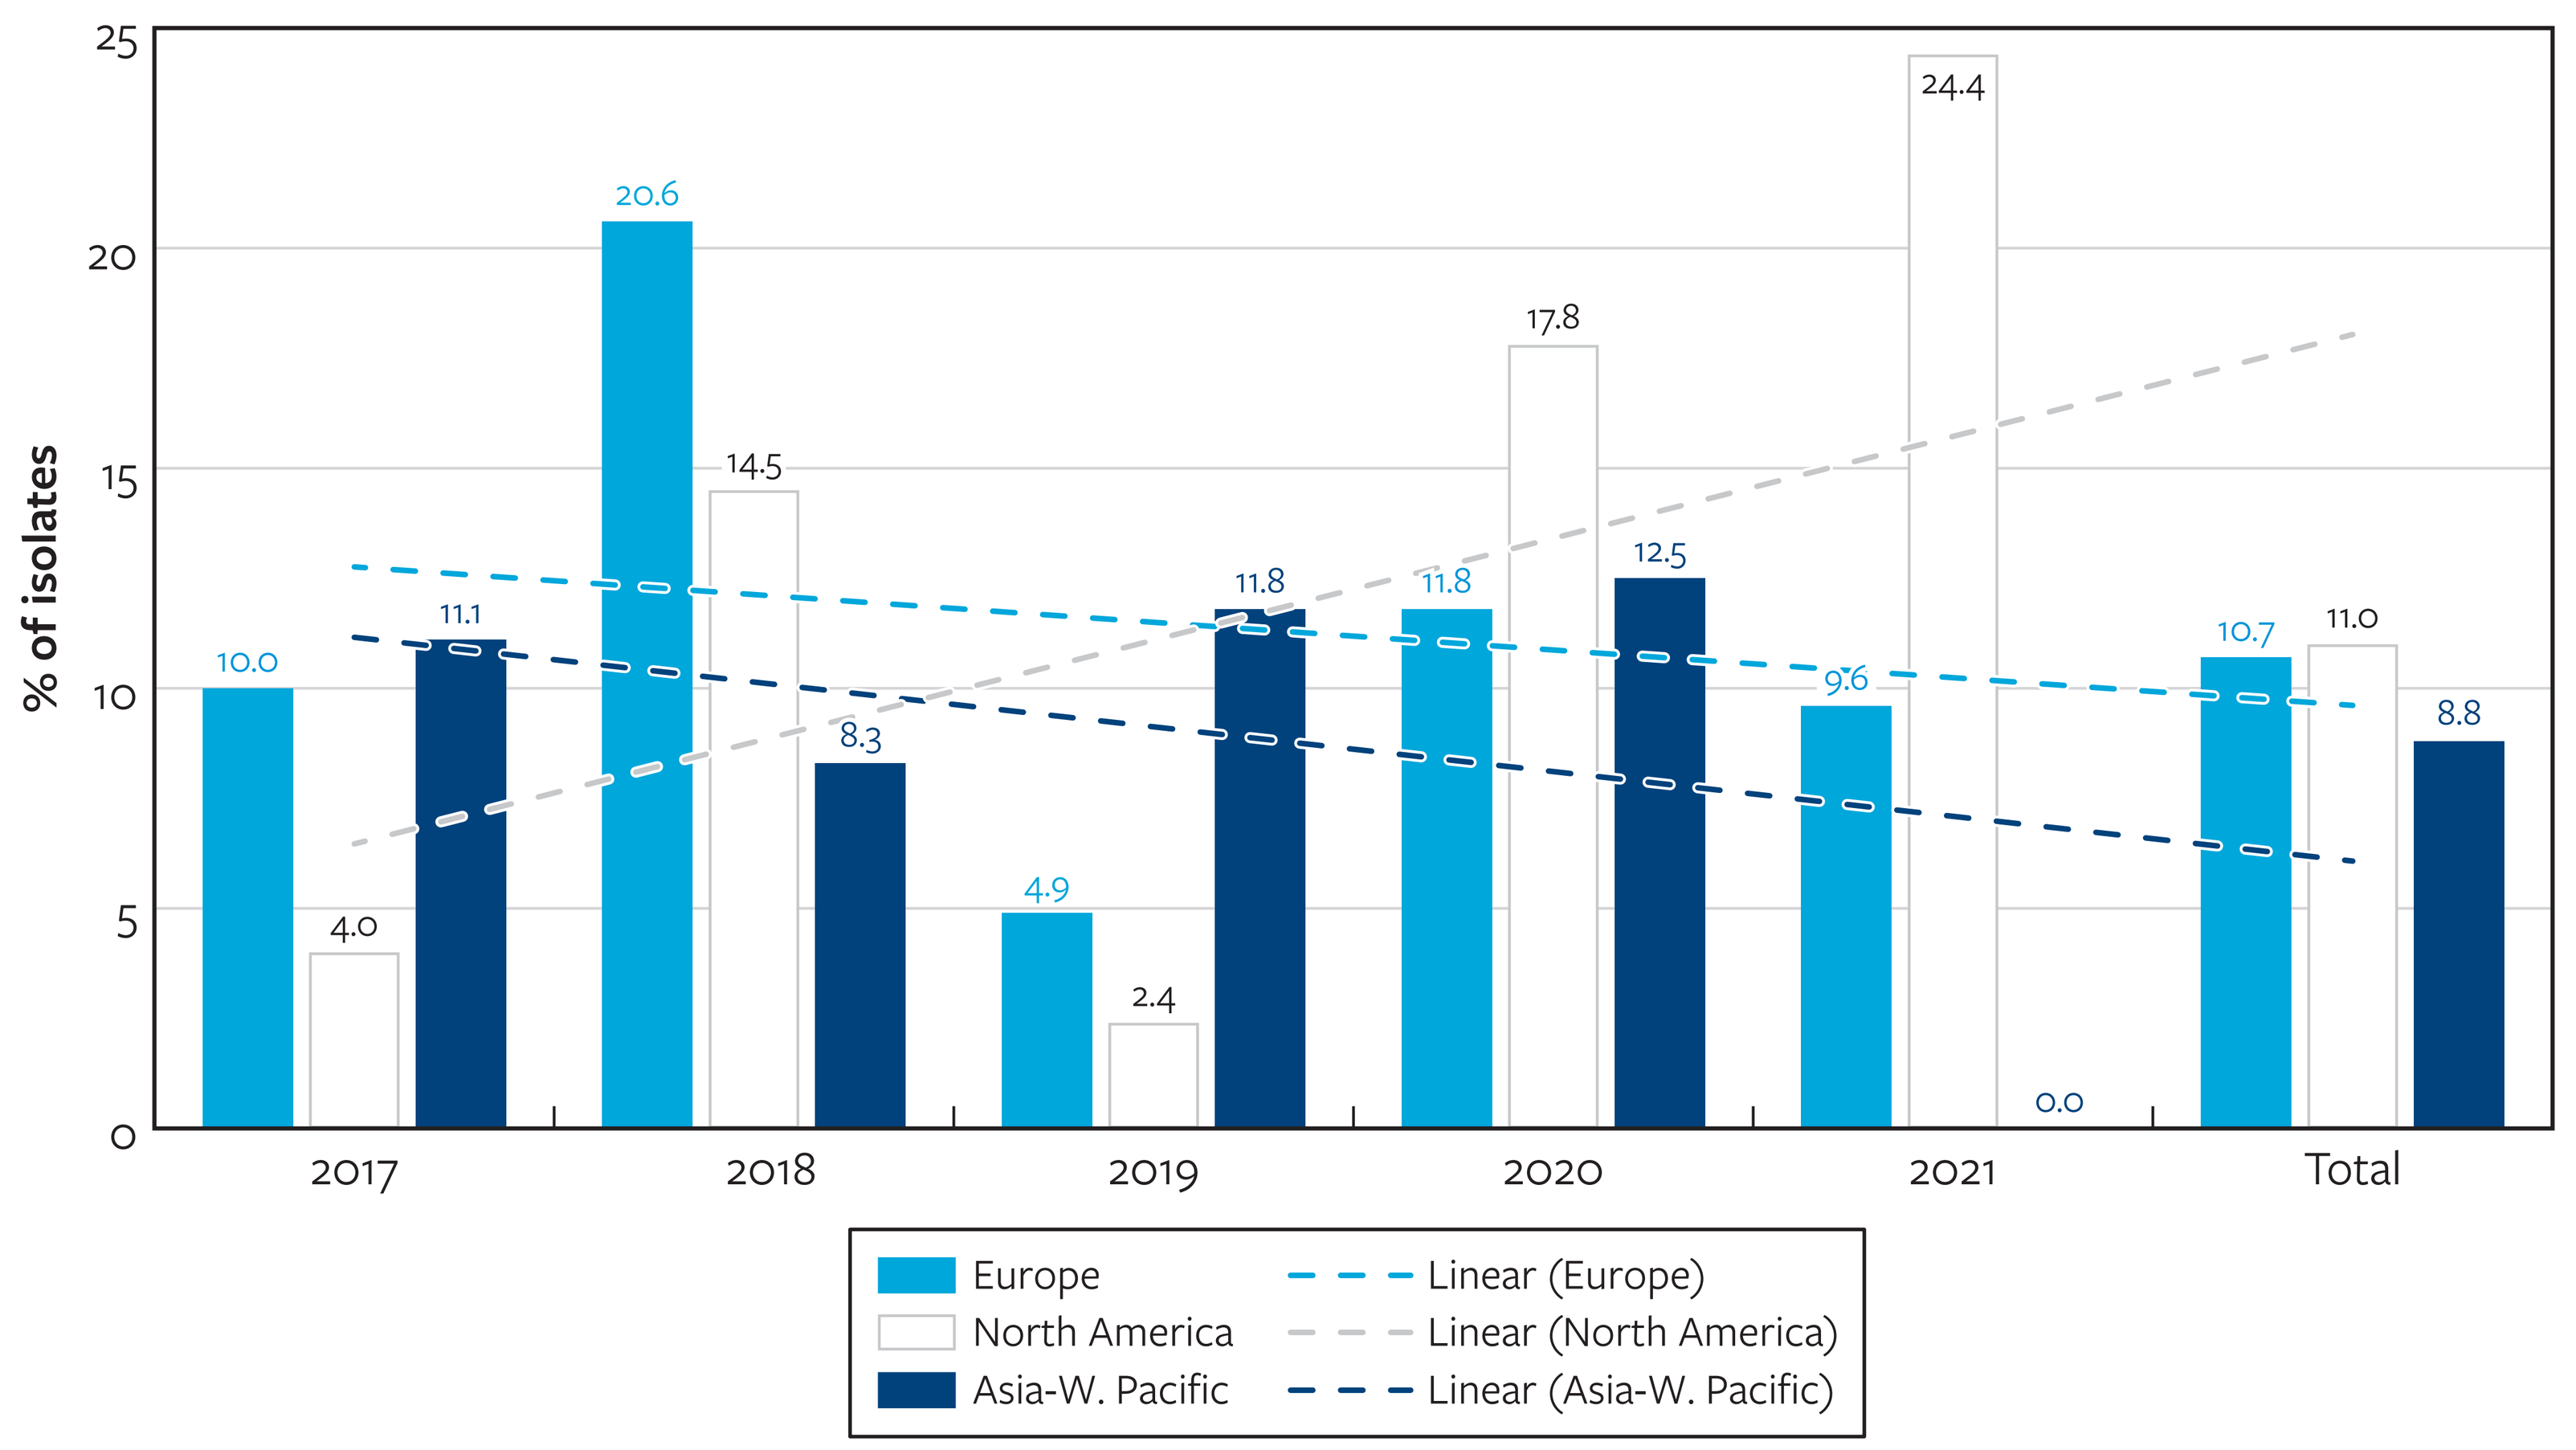

% of isolates
10.0
4.0
11.1
20.6
14.5
8.3
4.9
2.4
11.8
11.8
17.8
12.5
9.6
24.4
0.0
10.7
11.0
8.8
2017
2018
2019
2020
2021
Total
Europe
North America
Asia-W. Pacific
Linear (Europe)
Linear (North America)
Linear (Asia-W. Pacific)

Figure 4. Activity of isavuconazole and other azoles against azole-NWT *A. fumigatus* subgroups

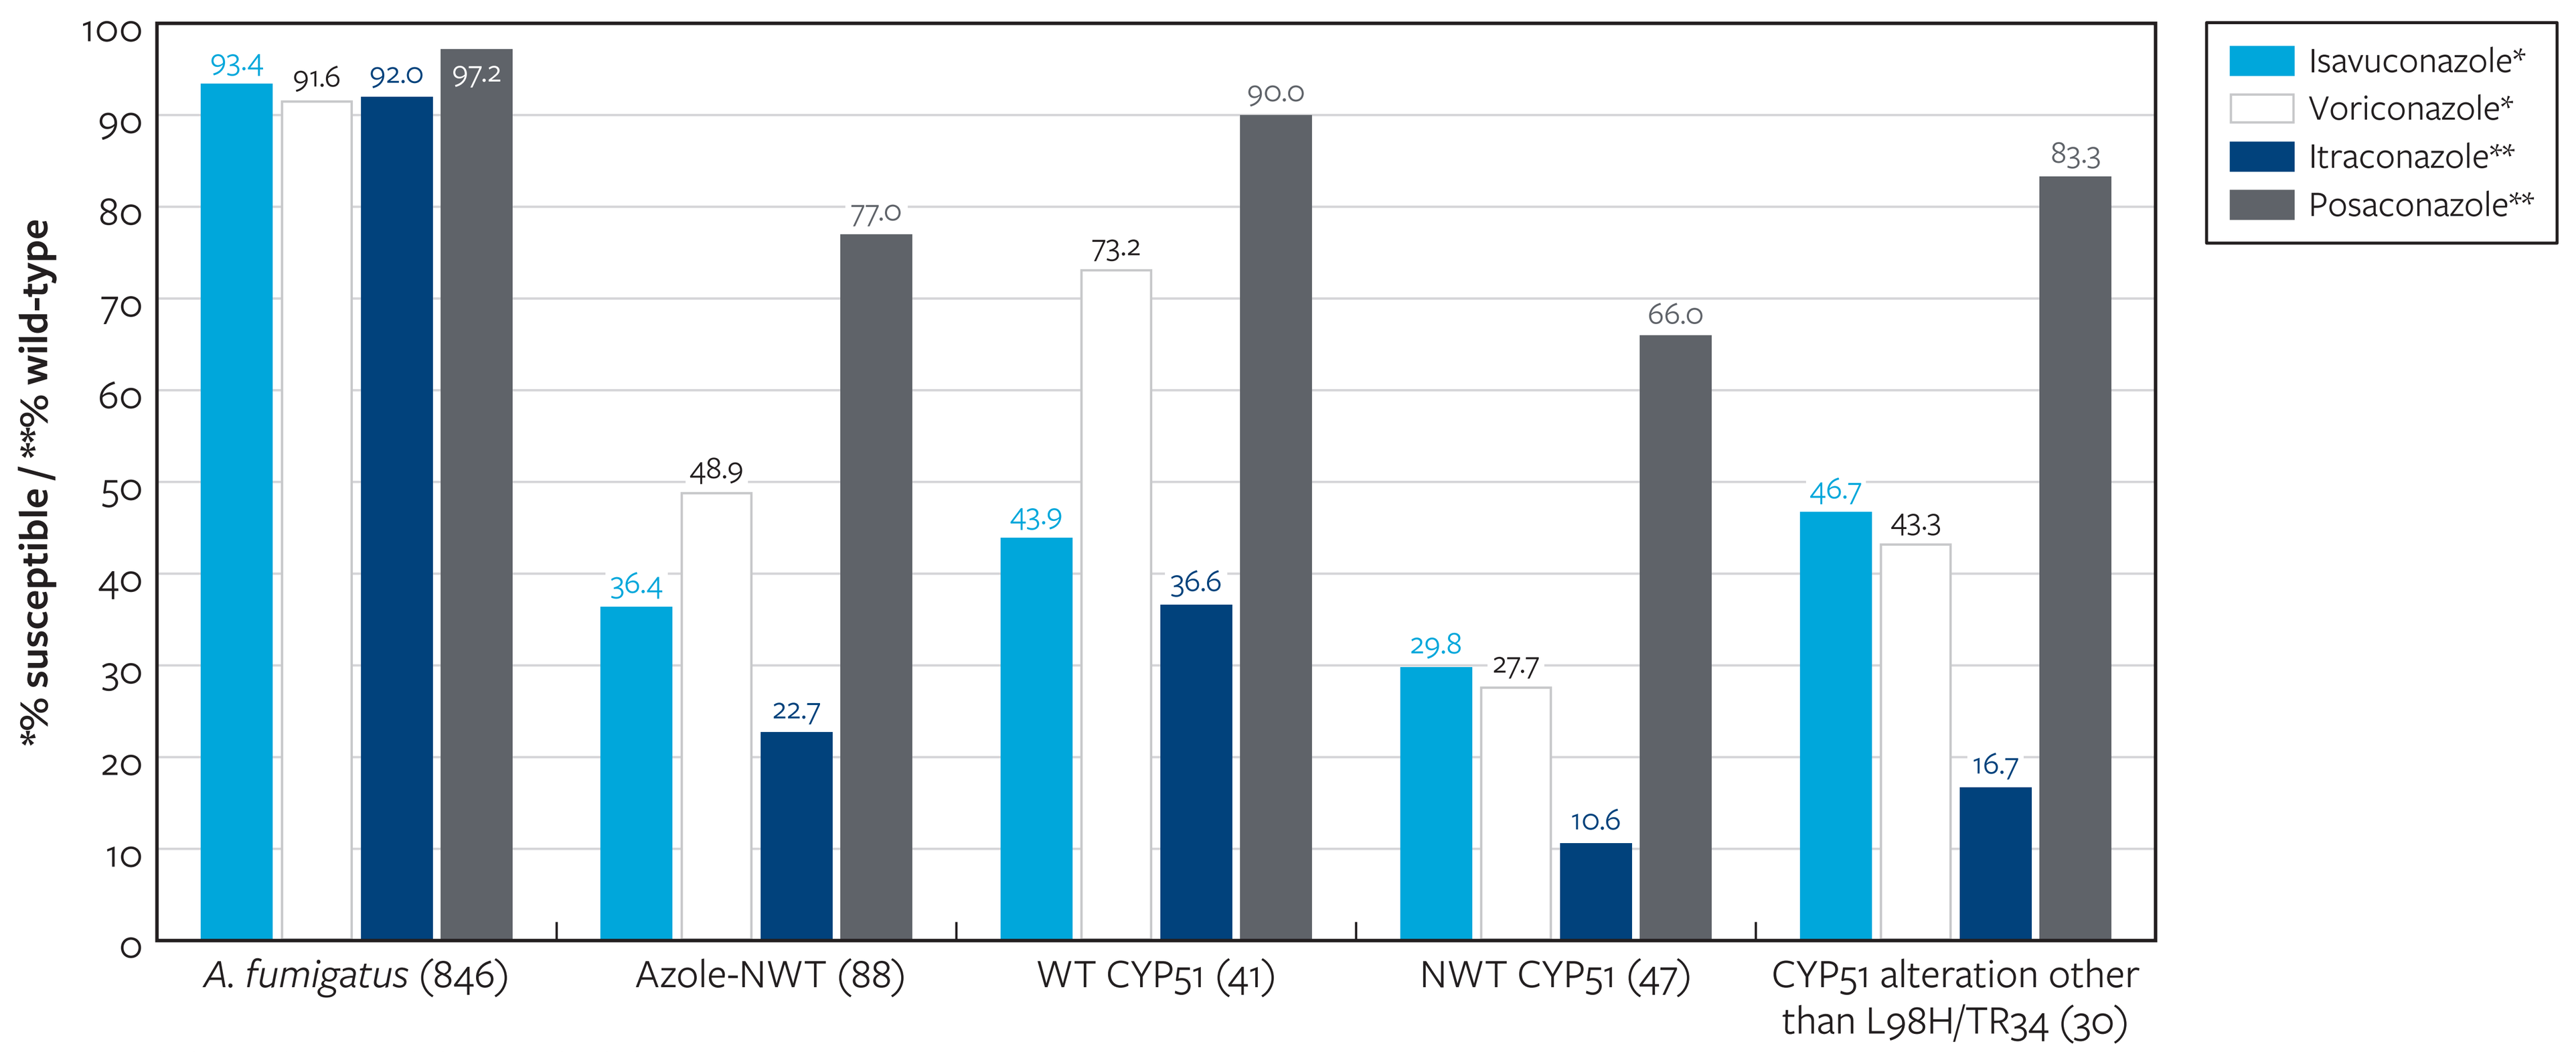

*% susceptible / **% wild-type
93.4
91.6
92.0
97.2
36.4
48.9
22.7
77.0
43.9
73.2
36.6
90.0
29.8
27.7
10.6
66.0
46.7
43.3
16.7
83.3
A. fumigatus (846)
Azole-NWT (88)
WT CYP51 (41)
NWT CYP51 (47)
CYP51 alteration other than L98H/TR34 (30)
Isavuconazole*
Voriconazole*
Itraconazole**
Posaconazole**

* Using CLSI M38M51S (2022) clinical breakpoint criteria.
** Using CLSI M57S (2022) epidemiological cutoff value criteria.

References

Bosetti D, Neofytos D. Invasive Aspergillosis and the Impact of Azole-resistance. Curr Fungal Infect Rep. 2023 Mar 18:1-10.

Jenks JD, Salzer HJ, Prattes J, Krause R, Buchheidt D, Hoenigl M. Spotlight on isavuconazole in the treatment of invasive aspergillosis and mucormycosis: design, development, and place in therapy. Drug Des Devel Ther. 2018 Apr 30;12:1033-1044.

CLSI (2017). M38Ed3. Reference method for broth dilution antifungal susceptibility testing of Filamentous Fungi. Wayne, PA.

CLSI (2022). M57SEd4. Epidemiological cutoff values for antifungal susceptibility testing. Wayne, PA.

CLSI (2022). M38M51SEd3. Performance standards for antifungal susceptibility testing of filamentous fungi. Wayne, PA.

Contact

Cecilia Carvalhaes, MD, Ph.D., D(ABMM)
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: cecilia.carvalhaes@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2023_23-PZR-06_P1_AFM_Isavu.pdf

Charges may apply. No personal information is stored.

IDWeek 2023, October 11–15, 2023, Boston, Massachusetts