IDWeek 2023 | Poster #P2114

Evaluation of Isavuconazole Activity Against Non-*fumigatus Aspergillus* and Cryptic *Aspergillus* Species Causing Invasive Infections Worldwide

Cecilia G. Carvalhaes, Paul R. Rhomberg, Valerie Kantro, Beth A. Hatch, Mariana Castanheira
Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA

Introduction

- While *Aspergillus fumigatus* is most commonly the cause of invasive aspergillosis (IA), cryptic and non-*fumigatus Aspergillus* spp. are a rising clinical concern.
- Isavuconazole, a second-generation broad-spectrum triazole, is a first-line therapy for the treatment of IA with favorable pharmacokinetic and safety profiles and few drug–drug interactions.
- There is limited data on antifungal susceptibility testing of clinically relevant non-*fumigatus* and cryptic species of *Aspergillus* causing IA.
- The activity of isavuconazole and other azoles against non-*fumigatus* and cryptic species of *Aspergillus* causing IA worldwide was evaluated.

Methods

- A total of 390 non-*fumigatus Aspergillus* were collected in 2017–2021 from 41 medical centers located in Europe (EU; *n*=161; 17 centers), North America (NA; *n*=137; 16 centers), Asia-Pacific (AP; *n*=85; 7 centers), and Latin America (LA, *n*=7; 1 center).
- Only 1 isolate per patient was included.
- Isolates were identified by MALDI-TOF MS and/or ITS and β-tubulin sequencing and tested by CLSI broth microdilution.
- CLSI epidemiological cut-off values (ECV) for *A. fumigatus*, *A. flavus*, *A. niger*, and *A. terreus* were applied to the respective section, where available.
- Posaconazole ECVs of 0.5 mg/L was used against *A.* section *Fumigati*.

Results

Identification of non-*fumigatus Aspergillus*

- The distribution of isolates per *Aspergillus* section is displayed in Figure 1.
- 63 (16.2%) of non-*fumigatus Aspergillus* were considered cryptic *Aspergillus* isolates (Figure 2).
- A total of 222 isolates (56.9%) were identified to the species complex level only (Figure 2).
- *A. terreus* and *A. niger* corresponded to 19.7% (77 isolates) and 6.9% (27 isolates) of the non-*fumigatus Aspergillus* species, respectively (Figure 2).

Azole activity against non-*fumigatus Aspergillus* split by *Aspergillus* sections (Table 1)

- Isavuconazole showed activity against *A.* sections Flavi (n=122; 98.4% WT), Terrei (n=57; 98.2% WT), Nidulans (n=34; $MIC_{50/90}$, 0.12/0.25 mg/L), Versicolores (n=7; MIC_{50}, 1 mg/L), and Circumdati (n=2; MIC range, 0.12–2 mg/L; Table 1).
- Similar activity was displayed by other azoles against those *Aspergillus* sections (Table 1; Figure 3).
- Most of the isolates from *A.* section Fumigati (n=9), Nigri (n=146), and Usti (n=12) exhibited elevated MIC values to isavuconazole ($MIC_{50/90}$, 2/-, 2/4, and 2/8 mg/L), voriconazole ($MIC_{50/90}$, 2/-, 1/2, and 4/8 mg/L), and itraconazole ($MIC_{50/90}$, 2/-, 2/4, and 8/>8 mg/L), respectively (Table 1).
- Posaconazole exhibited lower MIC values than other azoles against *A.* section Fumigati ($MIC_{50/90}$, 0.5/- mg/L) and *A.* section Nigri ($MIC_{50/90}$, 0.5/1), but high MIC values against *A.* section Usti ($MIC_{50/90}$, >8/>8 mg/L; Table 1).

Azole activity against cryptic *Aspergillus* species (Table 2)

- Isavuconazole was active (MIC values, ≤1 mg/L) against *A. parasiticus, A. tamarii, A. nomius, A. nidulans, A. unguis, A. alabamensis,* and *A. hortai,* while isavuconazole MIC values between 2–8 mg/L were observed against cryptic *Aspergillus* species from *A.* section Fumigati (Table 2).
- Isavuconazole inhibited 80.0% of *A. tubingensis* at ≤4 mg/L, the CLSI WT cut-off value for *A. niger* (Table 2).
- Voriconazole, itraconazole, and posaconazole showed similar activity to isavuconazole against most cryptic *Aspergillus* species (Table 2).

Azole activity against other non-*fumigatus Aspergillus* (*A. flavus* species complex, *A. terreus*, and *A. niger*; Table 3)

- Isavuconazole was active against *A. flavus* species complex (98.3% WT), *A.* niger (96.1% WT), and *A.* terreus (100% WT; Table 3).
- Voriconazole, itraconazole, and posaconazole showed similar activity to isavuconazole against *A. flavus* species complex, *A. terreus*, and *A. niger* isolates (Table 3).

Conclusions

- Isavuconazole exhibited potent *in vitro* activity against most non-*fumigatus Aspergillus*, including cryptic *Aspergillus* species.
- However, the activity of isavuconazole and other azoles vary among cryptic *Aspergillus* species: *A. lentulus*, *A. thermomutatus*, and *A. ustus* showed elevated azole MIC values overall.
- Species identification of non-*fumigatus Aspergillus* isolates and susceptibility testing is critical to guide treatment of cryptic *Aspergillus* species causing IA.

Table 1. Activity of isavuconazole and other azoles against non-*fumigatus Aspergillus* split by sections

Aspergillus section	*Aspergillus* species included	No. of isolates	$MIC_{50/90}$[a] (MIC range; %WT[b]); mg/L			
			Isavuconazole	Voriconazole	Itraconazole	Posaconazole
Circumdati	*A. sclerotiorum*	2	0.12 / - (0.12-2; NA)	0.12 / - (0.12-0.5; NA)	1 / - (1; NA)	0.25 / - (0.25-0.5; NA)
Flavi	*A. parasiticus, A. tamarii, A. nomius, A. flavus* species complex	122	0.5 / 1 (0.12-2; 98.4%)	0.5 / 1 (0.25-2; 100%)	0.5 / 1 (0.12-1; 100%)	0.5 / 0.5 (0.12-1; 99.2%)
Fumigati	*A. lentulus, A. thermomutatus, A. udagawae*	9	2 / - (2-8; 0%)	2 / - (2-8; 0%)	2 / - (1-4; 33.3%)	0.5 / - (0.5-1; 66.7%)
Nidulantes	*A. nidulans, A. unguis, A. nidulans* species complex	34	0.12 / 0.25 (0.015-0.5; NA)	0.12 / 0.25 (0.03-0.5; NA)	0.5 / 1 (0.12-4; NA)	0.25 / 0.5 (0.06-1; NA)
Nigri	*A. niger, A. tubingensis, A. niger* species complex	146	2 / 4 (0.06->8; 95.2%)	1 / 2 (0.06-4; 97.9%)	2 / 4 (0.12->8; 92.4%)	0.5 / 1 (0.12-2; 100%)
Terrei	*A. alabamensis, A. hortai, A. terreus, A. terreus* species complex	57	0.5 / 0.5 (0.06-2; 98.2%)	0.25 / 0.5 (0.06-1; 100%)	0.5 / 1 (0.12-1; 100%)	0.25 / 0.5 (0.12-0.5; 100%)
Usti	*A. ustus, A. ustus* species complex	12	2 / 8 (0.12-8; NA)	4 / 8 (0.25->8; NA)	8 / >8 (0.25->8; NA)	>8 / >8 (0.5->8; NA)
Versicolores	*A. versicolor, A. sydowii*	7	1 / - (0.5-2; NA)	1 / - (0.25-1; NA)	1 / - (0.5-2; NA)	1 / - (0.25-1; NA)

Abbreviations: WT, wildtype; NA, not available
[a] MIC90 was not calculated for Aspergillus sections with <10 isolates.
[b] CLSI M57 (2022) ECV criteria for *A. fumigatus*, *A. flavus*, *A. niger*, and *A. terreus* applied to corresponding *Aspergillus* section isolates.

Table 2. Activity of isavuconazole and other azoles against cryptic *Aspergillus* species

Aspergillus section	*Aspergillus* species	No. of isolates	MIC range (%WT[a]) mg/L			
			Isavuconazole	Voriconazole	Itraconazole	Posaconazole
Circumdati	*A. sclerotiorum*	2	0.12-2 (NA)	0.12-0.5 (NA)	1 (NA)	0.25-0.5 (NA)
Flavi	*A. parasiticus*	2	0.5 (100%)	0.5-1 (100%)	0.5-1 (100%)	0.25-0.5(100%)
	A. tamarii	2	0.12-0.25 (100%)	0.25-0.5 (100%)	0.25-0.5 (100%)	0.12-0.25 (100%)
	A. nomius	1	0.5 (100%)	0.5 (100%)	0.5 (100%)	0.5 (100%)
Fumigati	*A. lentulus*	6	2-4 (0.0%)	2 (0.0%)	1-2 (50.0%)	0.5 (100.0%)
	A. thermomutatus	2	2-8 (0.0%)	4-8 (0.0%)	4 (0.0%)	1 (0.0%)
	A. udagawae	1	2 (0.0%)	2 (0.0%)	2 (0.0%)	1 (0.0%)
Nidulantes	*A. nidulans*	24	0.015-0.25 (NA)	0.03-0.5 (NA)	0.12-1 (NA)	0.06-0.5 (NA)
	A. unguis	3	0.25-0.5 (NA)	0.06-0.25 (NA)	0.5-4 (NA)	0.25-1 (NA)
Nigri	*A. tubingensis*	5	2-8 (80.0%)	1-4 (80.0%)	2->8 (80.0%)	0.5-2 (100.0%)
Terrei	*A. alabamensis*	2	0.25 (100%)	0.25-0.5 (100%)	0.5 (100%)	0.25 (100%)
	A. hortai	1	0.25 (100%)	0.12 (100%)	0.5 (100%)	0.12 (100%)
Usti	*A. ustus*	5	2-8 (NA)	4->8 (NA)	4-8 (NA)	4->8 (NA)
Versicolores	*A. versicolor*	6	0.5-2 (NA)	0.25-1 (NA)	0.5-2 (NA)	0.25-1 (NA)
	A. sydowii	1	2 (NA)	1 (NA)	2 (NA)	1 (NA)

Abbreviations: WT, wildtype; NA, not available
[a] CLSI M57 (2022) ECV criteria for *A. fumigatus*, *A. flavus*, *A. niger*, and *A. terreus* applied to cryptic species from the same *Aspergillus* section.

Table 3. Activity of isavuconazole and other azoles against worldwide *A. flavus* species complex, *A. terreus*, and *A. niger* from 2017–2021

Aspergillus section	*Aspergillus* species	No. of isolates	$MIC_{50/90}$ (MIC range; %WT[a]); mg/L			
			Isavuconazole	Voriconazole	Itraconazole	Posaconazole
Flavi	*A. flavus* species complex	117	0.5 / 1 (0.12-2; 98.3%)	0.5 / 1 (0.25-2; 100%)	0.5 / 1 (0.12-1; 100%)	0.5 / 0.5 (0.12-1; 99.1%)
Terrei	*A. terreus*	27	0.5 / 0.5 (0.06-1; 100%)	0.25 / 0.5 (0.12-0.5; 100%)	0.5 / 1 (0.25-1; 100%)	0.25 / 0.5 (0.12-0.5; 100%)
Nigri	*A. niger*	77	2 / 4 (0.5->8; 96.1%)	1 / 1 (0.25-4; 98.7%)	2 / 4 (0.25->8; 97.4%)	0.5 / 4 (0.25-1; 100%)

Abbreviations: WT, wildtype
[a] Using CLSI M57 (2022) ECV criteria.

Figure 1. Distribution of non-*fumigatus Aspergillus* collected from 2017–2021 worldwide by *Aspergillus* section

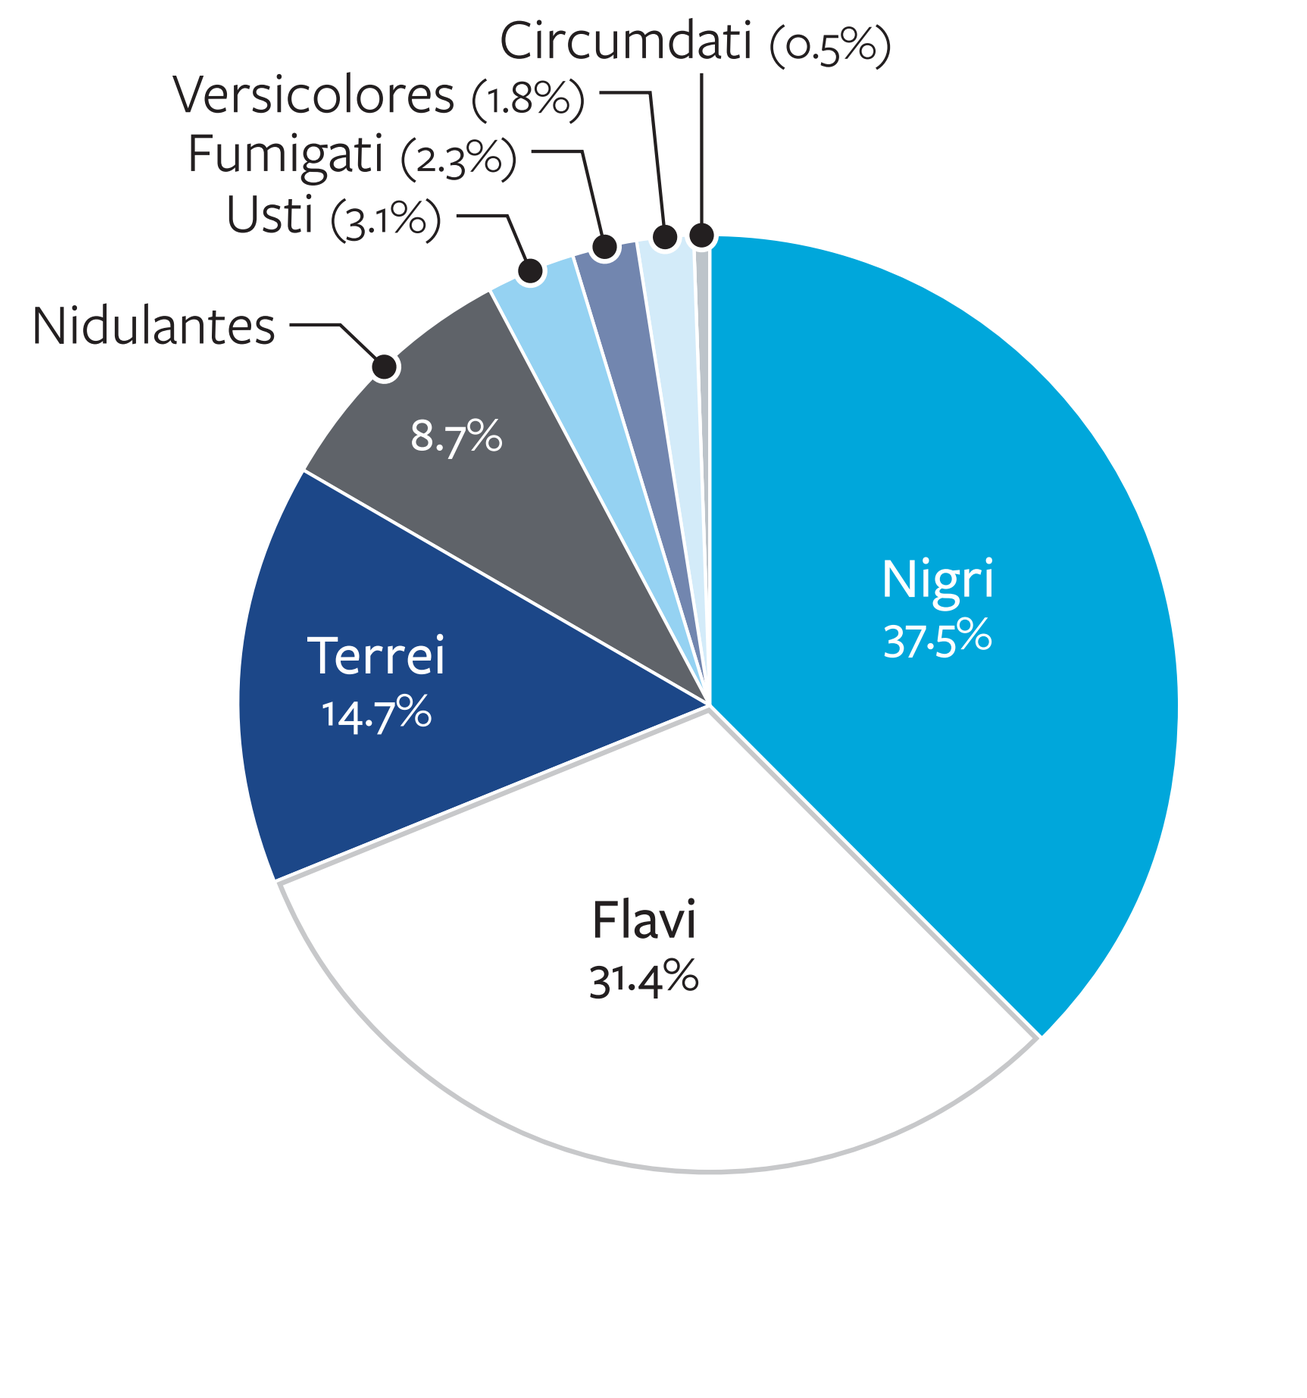
Circumdati (0.5%)
Versicolores (1.8%)
Fumigati (2.3%)
Usti (3.1%)
Nidulantes
8.7%
Terrei
14.7%
Flavi
31.4%
Nigri
37.5%

Figure 2. Distribution of non-*fumigatus Aspergillus* collected from 2017–2021 worldwide by identification group

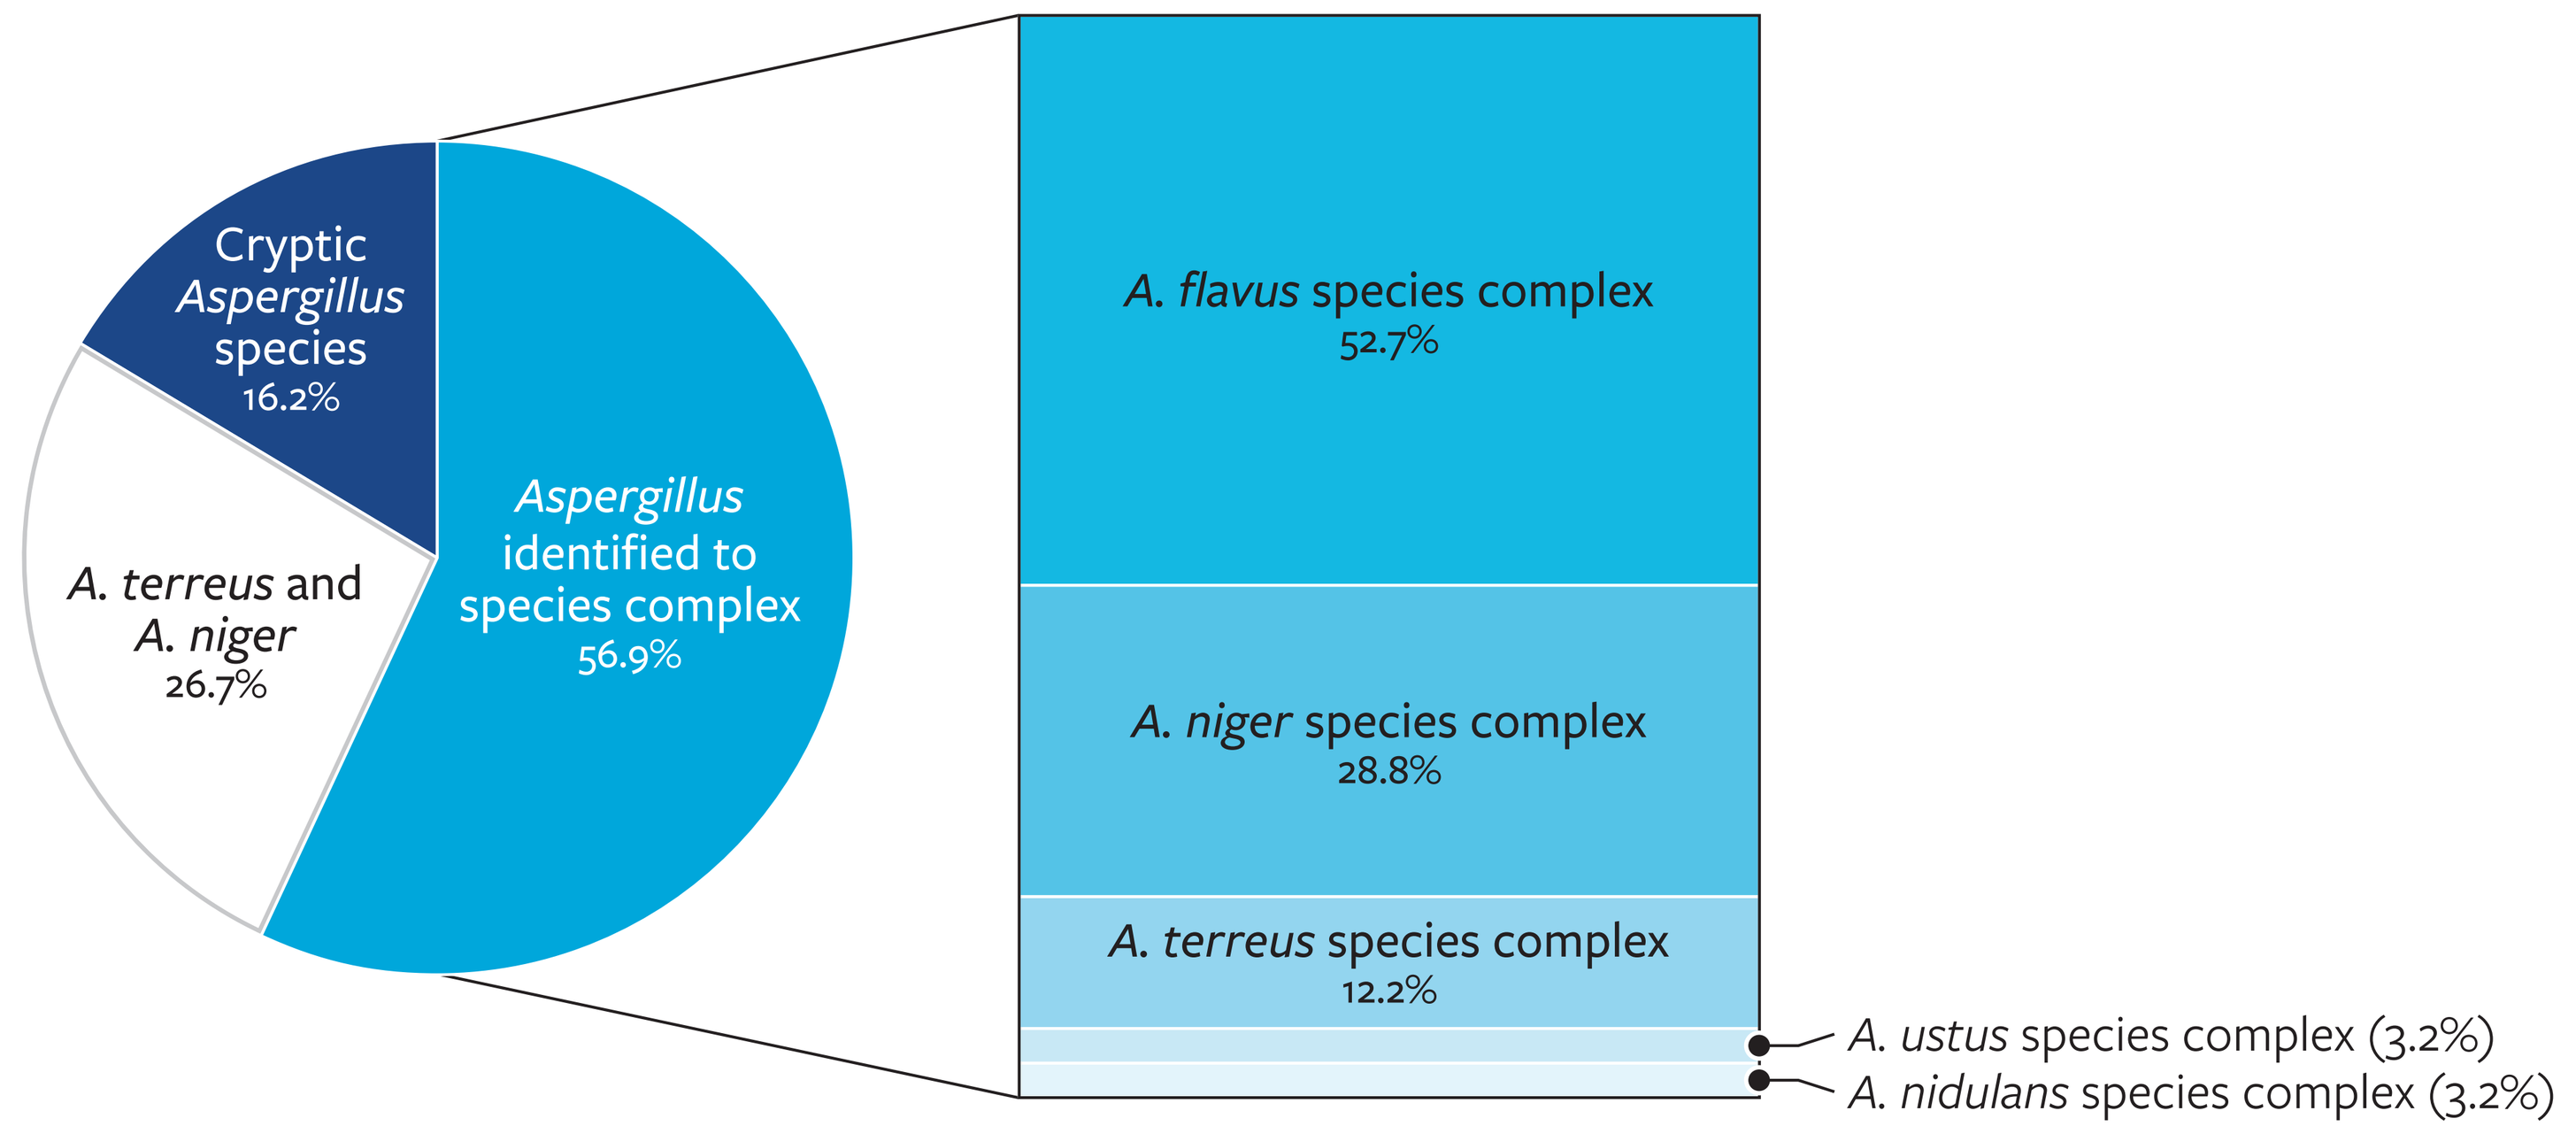
Cryptic Aspergillus species
16.2%
A. terreus and A. niger
26.7%
Aspergillus identified to species complex
56.9%
A. flavus species complex
52.7%
A. niger species complex
28.8%
A. terreus species complex
12.2%
A. ustus species complex (3.2%)
A. nidulans species complex (3.2%)

Figure 3. Activity of isavuconazole and other azoles against non-*fumigatus Aspergillus* isolates split by *Aspergillus* sections (2017–2021)

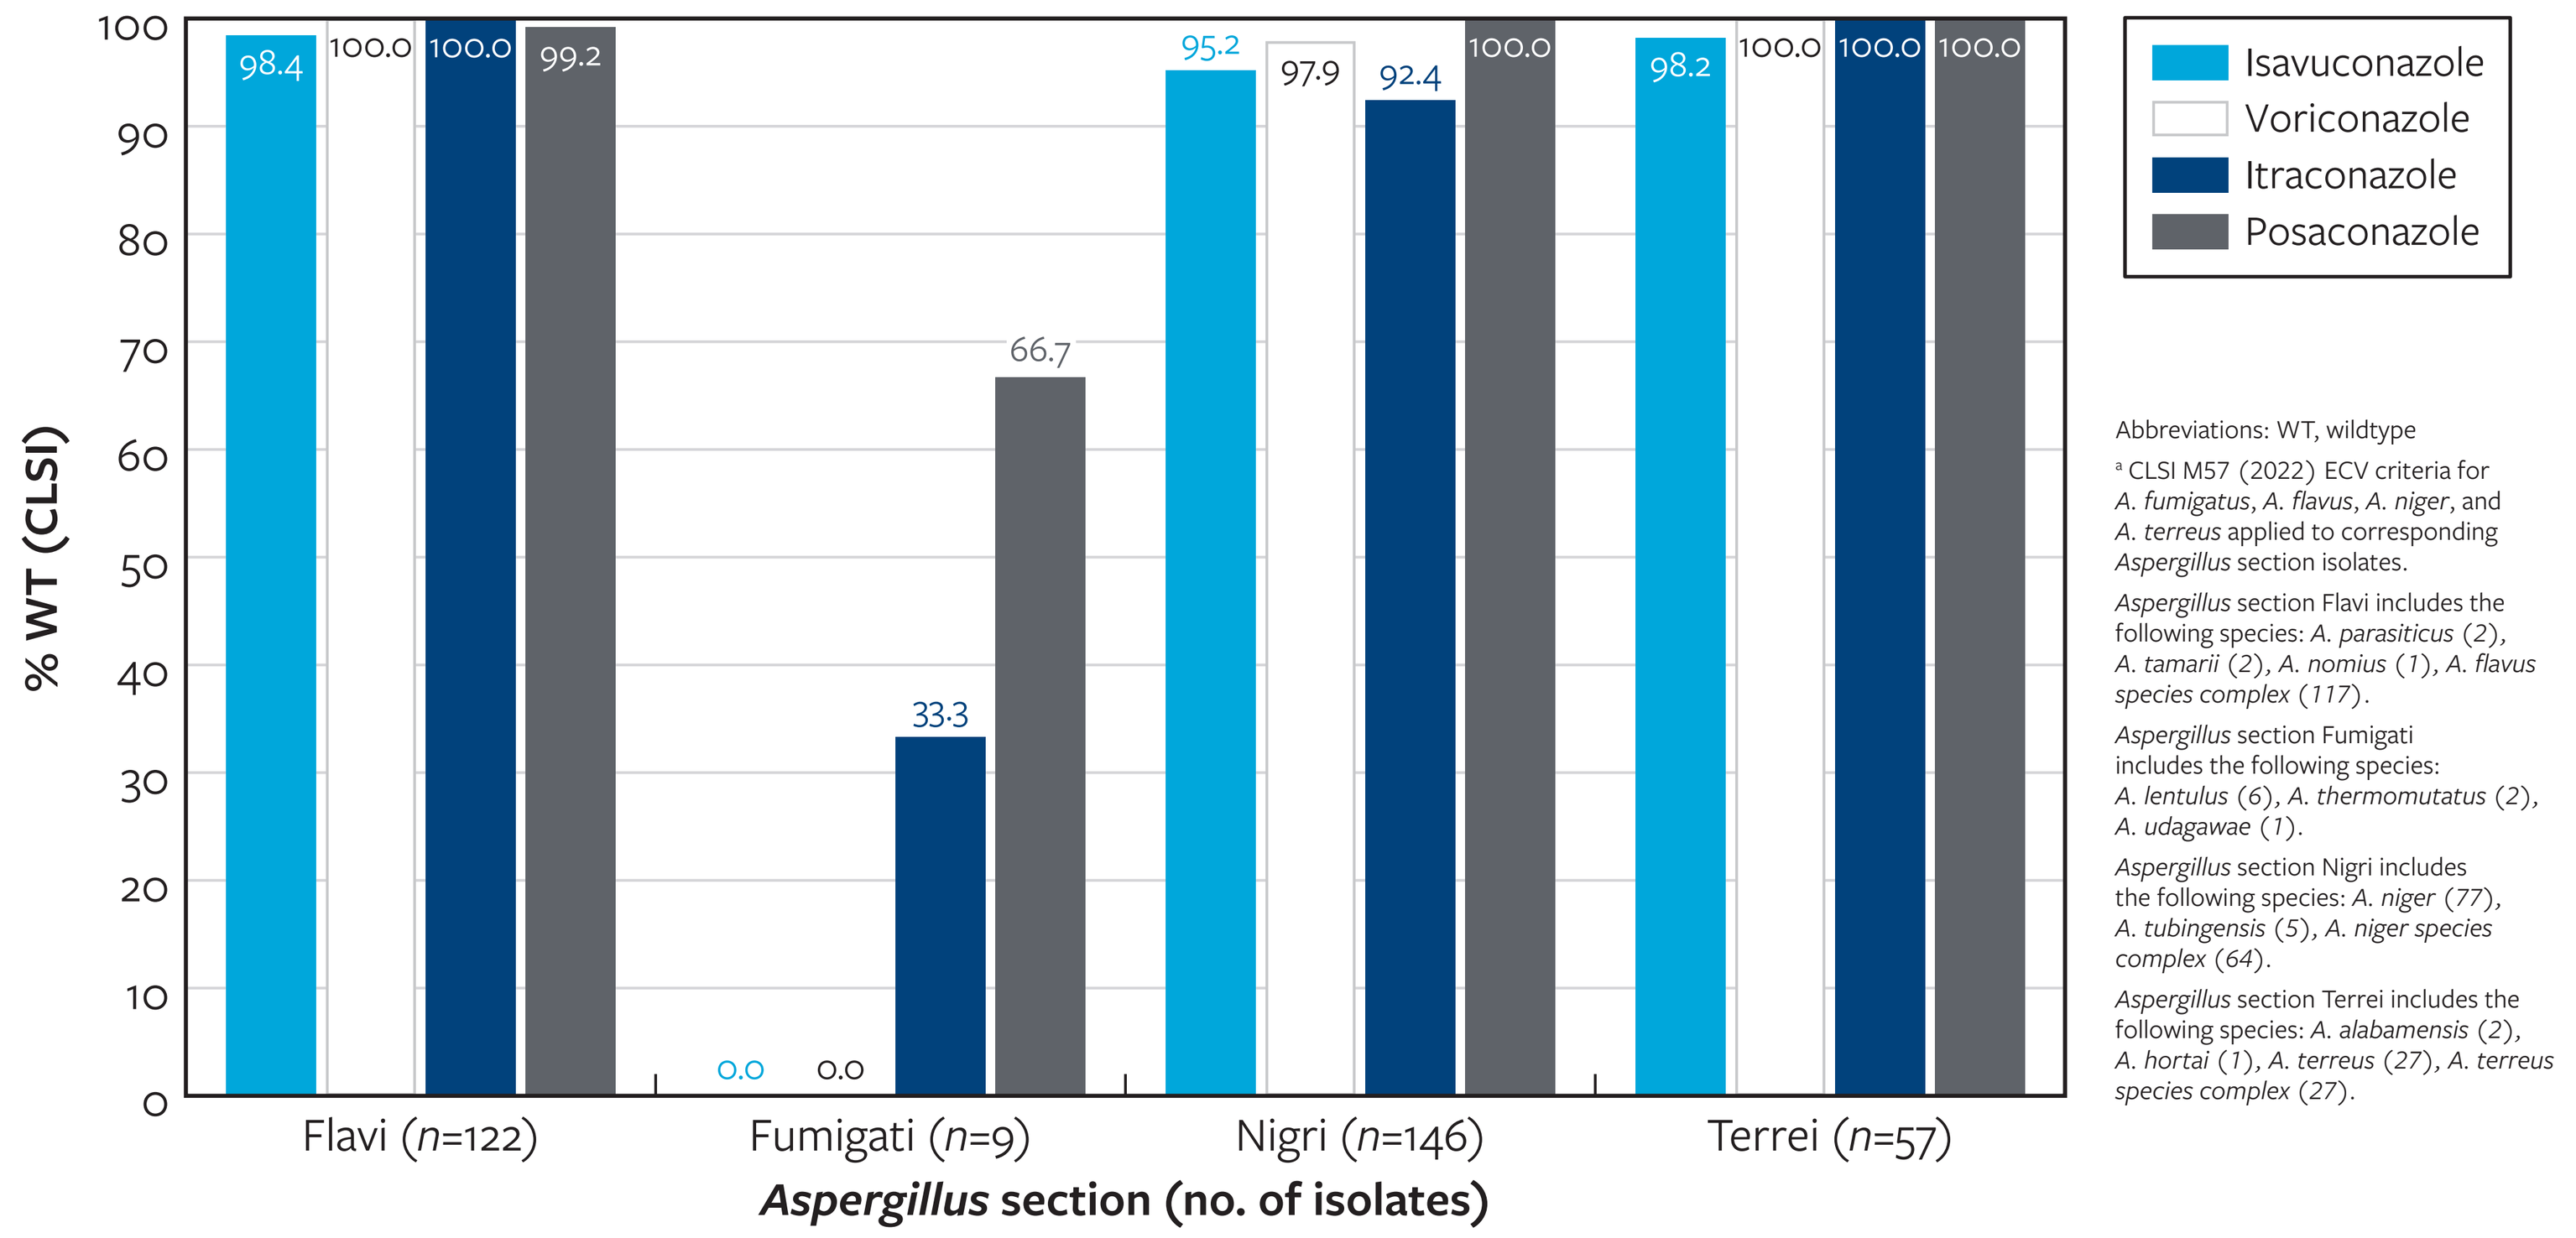
% WT (CLSI)
0
10
20
30
40
50
60
70
80
90
100
98.4
100.0
100.0
99.2
0.0
0.0
33.3
66.7
95.2
97.9
92.4
100.0
98.2
100.0
100.0
100.0
Flavi (n=122)
Fumigati (n=9)
Nigri (n=146)
Terrei (n=57)
Aspergillus section (no. of isolates)
Isavuconazole
Voriconazole
Itraconazole
Posaconazole

Abbreviations: WT, wildtype
[a] CLSI M57 (2022) ECV criteria for *A. fumigatus*, *A. flavus*, *A. niger*, and *A. terreus* applied to corresponding *Aspergillus* section isolates.
Aspergillus section Flavi includes the following species: *A. parasiticus* (2), *A. tamarii* (2), *A. nomius* (1), *A. flavus species complex* (117).
Aspergillus section Fumigati includes the following species: *A. lentulus* (6), *A. thermomutatus* (2), *A. udagawae* (1).
Aspergillus section Nigri includes the following species: *A. niger* (77), *A. tubingensis* (5), *A. niger species complex* (64).
Aspergillus section Terrei includes the following species: *A. alabamensis* (2), *A. hortai* (1), *A. terreus* (27), *A. terreus species complex* (27).

Funding

This study was supported by Pfizer Inc. (New York, NY). CG Carvalhaes, PR Rhomberg, V Kantro, BA Hatch, and M Castanheira are employees of Element Materials Technology (JMI Laboratories) at the time of this study, which was paid consultant to Pfizer in connection with the development of this poster.

References

Bosetti D, Neofytos D. Invasive Aspergillosis and the Impact of Azole-resistance. Curr Fungal Infect Rep. 2023 Mar 18:1-10.

Jenks JD, Salzer HJ, Prattes J, Krause R, Buchheidt D, Hoenigl M. Spotlight on isavuconazole in the treatment of invasive aspergillosis and mucormycosis: design, development, and place in therapy. Drug Des Devel Ther. 2018 Apr 30;12:1033-1044.

CLSI (2017). M38Ed3. Reference method for broth dilution antifungal susceptibility testing of Filamentous Fungi. Wayne, PA.

CLSI (2022). M57SEd4. Epidemiological cutoff values for antifungal susceptibility testing. Wayne, PA.

CLSI (2022). M38M51SEd3. Performance standards for antifungal susceptibility testing of filamentous fungi. Wayne, PA.

Acknowledgments

The authors thank all the participant centers for their work in providing isolates.

Contact

element

Cecilia Carvalhaes, MD, Ph.D., D(ABMM)
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: cecilia.carvalhaes@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2023_23-PZR-06_P2_Cryptic_Aspergillus.pdf

Charges may apply. No personal information is stored.

IDWeek 2023, October 11–15, 2023, Boston, Massachusetts