In vitro Activity of Gepotidacin Tested Against Molecularly Characterized *Escherichia coli* Isolates Responsible for Urinary Tract Infections in the US (2019–2022)

Rodrigo E. Mendes[1], Danielle Beekman[1], Abigail Scullin[1], Maura Karr[1], Renuka Kapoor[2], Didem Torumkuney[3], S. J. Ryan Arends[1]

[1] Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA; [2] GSK, Collegeville, Pennsylvania, USA; [3] GSK HQ, London

SCAN ME

Digital poster

SCAN ME

Introduction

- Gepotidacin is a novel, bactericidal, first-in-class triazaacenaphthylene antibacterial that inhibits bacterial DNA replication by a unique mechanism of action, distinct binding site[1, 2] and provides well-balanced inhibition of two different type II topoisomerase enzymes in most pathogens causing uncomplicated urinary tract infections (uUTI) and *Neisseria gonorrhoeae*[3, 4].
- Results from two phase 3 clinical trials demonstrated the efficacy of gepotidacin for the treatment of uUTIs[5]. More recently, gepotidacin met its primary efficacy endpoint of non-inferiority in a phase 3 trial comparing gepotidacin with intramuscular ceftriaxone plus oral azithromycin combination for the treatment of urogenital gonorrhea[6].
- This study reports the activity of gepotidacin and other oral antibacterials against *Escherichia coli*, including molecularly characterized isolates carrying ESBL and carbapenemase genes collected from patients with UTI in the United States.

Materials and Methods

Bacterial organisms

- 3,995 *E. coli* collected from 52 US sites located in 9 US Census Regions as part of the gepotidacin uropathogen global surveillance study were included (2019–2022).
- Only consecutive isolates responsible for UTI (1 per patient infection episode) were included.
- Bacterial identification was confirmed by standard algorithms supported by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany).

Susceptibility testing

- Isolates were tested for susceptibility by broth microdilution and agar dilution following Clinical and Laboratory Standards Institute (CLSI) M07 (2018) guidelines[7].
- Frozen-form broth microdilution panels were manufactured by JMI Laboratories (North Liberty, Iowa, USA) and contained cation-adjusted Mueller-Hinton broth. Agar dilution plates were used for testing fosfomycin and included glucose-6-phosphate (25 mg/L)[7, 8].
- Quality assurance was performed by sterility checks, colony counts, and testing CLSI-recommended quality control reference strains[8].
- *E. coli* with MIC results ≥2 µg/mL for aztreonam and/or ceftazidime and/or ceftriaxone were defined as presumptive ESBL producers and subjected to genome sequencing followed by β-lactamase gene screening and epidemiology typing (MLST, O:H, and *fimH*).

Screening of β-lactamase genes

- Selected isolates had total genomic DNA extracted by the fully automated Thermo Scientific KingFisher Flex Magnetic Particle Processor (Cleveland, OH, USA), which was used as input material for library construction.
- DNA libraries were prepared using the Nextera or Illumina DNA Prep library construction protocol (Illumina, San Diego, CA, USA) following the manufacturer's instructions and were sequenced on MiSeq or NextSeq Sequencer platforms at JMI Laboratories.
- FASTQ format sequencing files for each sample set were assembled independently using *de novo* assembler SPAdes 3.15.3. An in-house software was applied to align the assembled sequences against a comprehensive in-house database containing known β-lactamase genes.

Epidemiology typing

- Multilocus sequence typing (MLST) was performed by extracting a defined set of 7 housekeeping gene fragments (~500 bp).
 - Each fragment was compared to known allelic variants for each locus (housekeeping gene) on the MLST website (PubMLST, https://pubmlst.org).
- An allele sharing 100% genetic identity with a known variant received a numeric designation.
 - A 7-number sequence (1 for each housekeeping gene) formed an allelic profile, defined as sequence type (ST).
 - Isolates containing alleles that did not match an existing sequence in the MLST database were submitted/deposited for allele and ST assignments. This information was used to group ST131 and single-locus variants of ST131 into isolates belonging to the clonal complex (CC) 131.
- The O:H serotyping and *fimH* typing were performed using tools available at the Center for Genomic Epidemiology (https://cge.cbs.dtu.dk/services/ResFinder).
- Isolates that met the criteria for the screening of ESBL genes were subjected to MLST and O:H typing. Isolates associated with ST131 were also subjected to *fimH* typing.

Results

- A total of 86.2% (3,444/3,995) *E. coli* isolates did not meet the criteria for screening of β-lactamase genes (presumptive ESBL-negative) (Table 1).
 - Gepotidacin MIC_{50} and MIC_{90} values were 2 µg/mL and 4 µg/mL, respectively, against this group, where cefazolin (97.0% susceptible), fosfomycin (99.7% susceptible), and nitrofurantoin (98.7% susceptible) also had activity among oral comparator agents tested.
 - Among these isolates, 15.3% were not susceptible to ciprofloxacin, whereas 24.1% were not susceptible to trimethoprim-sulfamethoxazole (SXT).
- A presumptive ESBL phenotype was noted in 13.8% (551/3,995) of *E. coli*, with the highest rates observed in the Middle Atlantic (45.8%), followed by East South Central (16.5%), Mountain (16.2%), and Pacific (16.0%) regions in the US. Other regions had rates of 6.1–11.9% (Figure 1).
 - Gepotidacin MIC_{50} and MIC_{90} values of 2 µg/mL and 4 µg/mL, respectively, were obtained against isolates with a presumptive ESBL phenotype (Table 1).
 - The *E. coli* with a presumptive ESBL phenotype were mostly not susceptible to the ciprofloxacin (78.9%), SXT (61.0%), amoxicillin-clavulanate (50.4%), and cefazolin (98.9%) (Table 1).
 - Among comparator agents, only fosfomycin (98.0% susceptible) and nitrofurantoin (92.7% susceptible) showed good activity against these isolates (Table 1).
- Most isolates with a presumptive ESBL phenotype carried CTX-M alleles alone (81.7%; 450/551), whereas 14.0% (77/551) had plasmid AmpC genes (Table 1).
- Approximately half (53.9%; 297/551) of the isolates with a presumptive ESBL phenotype belonged to clonal complex (CC) 131, of which 71.0% (211/297) were O25b:H4 and carried *fimH*30.
- In general, gepotidacin retained activity with MIC_{50} and MIC_{90} values of 2 µg/mL and 4 µg/mL, respectively, against isolates carrying ESBLs, AmpC or carbapenemases, and against CC131 and the O25b:H4 resistant subset (Table 1).

Conclusions

- Gepotidacin showed consistent activity against UTI-causing *E. coli* in the US, including against isolates carrying ESBL, AmpC, and/or carbapenemase genes. In addition, gepotidacin had activity against the multidrug-resistant CC131 *E. coli* clone.
- These data support further clinical development of gepotidacin as a potential treatment option for uUTI caused by *E. coli* including when other oral treatment options are limited due to drug resistance.

Figure 1. Proportions (%) of presumptive ESBL-producing *E. coli* causing UTI in the 9 US Census Regions. The proportion of these isolates belonging to the CC131 clone are also shown.

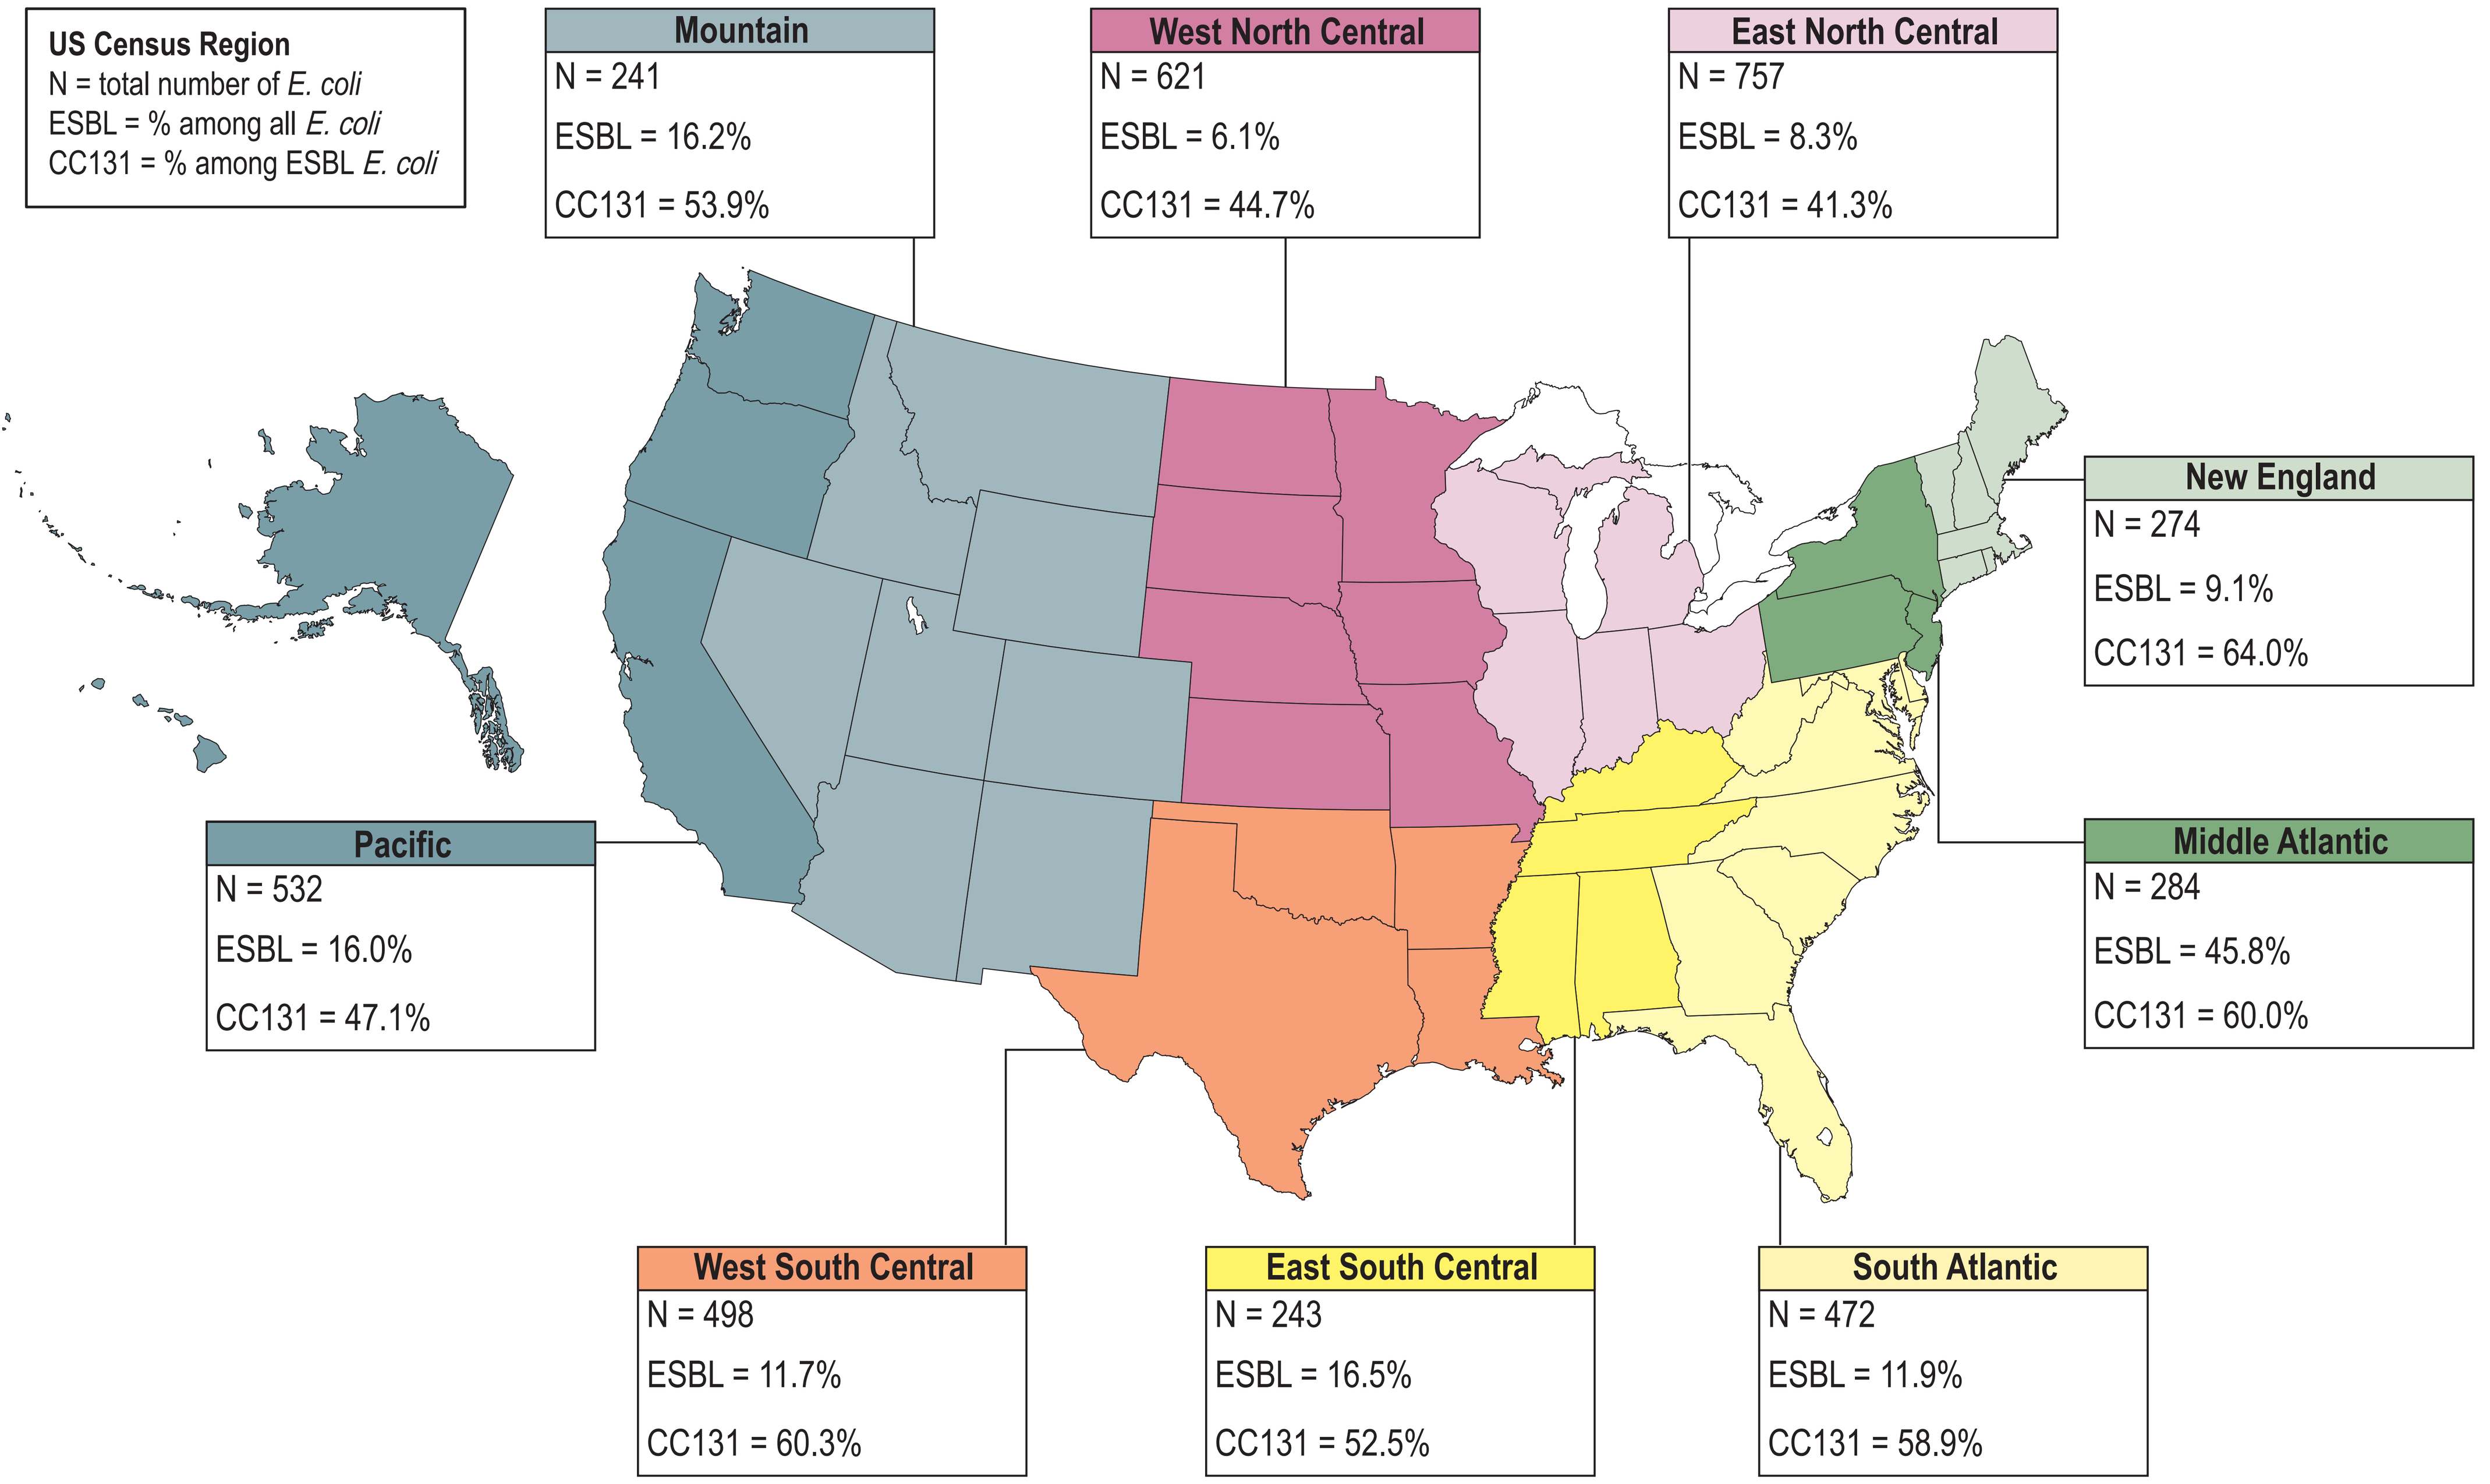
US Census Region
N = total number of E. coli
ESBL = % among all E. coli
CC131 = % among ESBL E. coli
Mountain
N = 241
ESBL = 16.2%
CC131 = 53.9%
West North Central
N = 621
ESBL = 6.1%
CC131 = 44.7%
East North Central
N = 757
ESBL = 8.3%
CC131 = 41.3%
New England
N = 274
ESBL = 9.1%
CC131 = 64.0%
Pacific
N = 532
ESBL = 16.0%
CC131 = 47.1%
Middle Atlantic
N = 284
ESBL = 45.8%
CC131 = 60.0%
West South Central
N = 498
ESBL = 11.7%
CC131 = 60.3%
East South Central
N = 243
ESBL = 16.5%
CC131 = 52.5%
South Atlantic
N = 472
ESBL = 11.9%
CC131 = 58.9%

Table 1. Activity of gepotidacin and oral comparator agents tested against *E. coli* and drug-resistant subsets

Phenotype/genotype (No. isolates)	MIC_{50}/MIC_{90} in µg/mL (% susceptible by CLSI criteria)						
	GEP	AMC	CFZ	CIP	SXT	FOS	NIT
ESBL MIC screen-negative (3,444)	2/4 (—)	4/16 (88.1)	2/8 (97.0)	0.015/>4 (84.7)	≤0.12/>4 (75.9)	0.5/1 (99.7)	16/16 (98.7)
ESBL MIC screen-positive[a] (551)	2/4 (—)	16/32 (49.6)	>32/>32 (1.1)	>4/>4 (21.1)	>4/>4 (39.0)	0.5/2 (98.0)	16/32 (92.7)
FQ-NS (435)	2/4 (—)	8/32 (51.2)	>32/>32 (0.2)	>4/>4 (0.2)	>4/>4 (34.7)	0.5/2 (97.7)	16/32 (90.8)
ESBL/AmpC[b] (541)	2/4 (—)	8/32 (50.2)	>32/>32 (0.9)	>4/>4 (20.2)	>4/>4 (38.3)	0.5/2 (98.0)	16/32 (92.6)
CTX-M (450)	2/4 (—)	8/16 (59.5)	>32/>32 (0.2)	>4/>4 (12.5)	>4/>4 (34.7)	0.5/2 (97.6)	16/32 (92.4)
AmpC (77)	2/4 (—)	32/>32 (0.0)	>32/>32 (5.2)	0.06/>4 (63.6)	0.25/>4 (59.7)	0.5/2 (100)	16/32 (92.2)
Other[c] (14)	2/16 (—)	16/>32 (28.6)	>32/>32 (0.0)	0.5/>4 (28.6)	>4/>4 (35.7)	0.5/2 (100)	16/16 (100)
Negative[d] (10)	1/2 (—)	32/32 (20.0)	32/>32 (10.0)	0.015/>4 (70.0)	≤0.12/>4 (80.0)	0.5/1 (100)	16/16 (100)
CC131[e] (297)	2/4 (—)	8/16 (52.0)	>32/>32 (0.0)	>4/>4 (6.1)	>4/>4 (36.4)	0.5/2 (98.3)	16/32 (91.2)
O25b:H4/fimH30 (211)	2/4 (—)	16/16 (46.2)	>32/>32 (0.0)	>4/>4 (0.5)	>4/>4 (37.4)	0.5/2 (97.6)	16/32 (90.0)
O16:H5/fimH41 (49)	1/2 (—)	8/16 (85.7)	>32/>32 (0.0)	>4/>4 (33.3)	>4/>4 (22.4)	0.5/1 (100)	16/16 (93.9)
Non-CC131[f] (254)	2/4 (—)	16/>32 (46.9)	>32/>32 (2.4)	>4/>4 (38.6)	>4/>4 (42.1)	0.5/2 (97.6)	16/32 (94.5)

MIC, minimal inhibitory concentration; FQ-NS, fluoroquinolone not susceptible; ESBL, extended-spectrum-β-lactamase; GEP, gepotidacin; AMC, amoxicillin-clavulanate; CFZ, cefazolin (used as a surrogate for oral cephalosporins for uUTI); CIP, ciprofloxacin; SXT, trimethoprim-sulfamethoxazole; FOS, fosfomycin; NIT, nitrofurantoin; CLSI breakpoints and interpretive criteria applied; "—" breakpoint not available.

[a] Includes isolates with aztreonam, ceftazidime, ceftriaxone or meropenem MICs of ≥2 µg/mL.

[b] Includes isolates with overexpression of chromosomal *ampC* (27), plasmid AmpC (50), $bla_{CTX\text{-}M}$ (450), $bla_{SHV\text{-}12}$ (3), $bla_{TEM\text{-}29}$ (1), and genotypes described in footnote "c".

[c] Includes 6 $bla_{CTX\text{-}M}$ and bla_{CMY}, 1 bla_{DHA} and bla_{CMY}, 1 bla_{NDM} and $bla_{CTX\text{-}M}$, 1 $bla_{CTX\text{-}M}$ and $bla_{SHV\text{-}12}$, 1 bla_{KPC}, 3 $bla_{SHV\text{-}12}$, and 1 $bla_{TEM\text{-}29}$.

[d] Includes isolates where only narrow-spectrum β-lactamase genes were detected, or overexpression of *ampC*, pAmpC, ESBL or carbapenemase genes were not found.

[e] Includes ST131 (282), ST131-like (7), ST2279 (7), and ST8257 (1).

[f] Includes mostly ST1193 (45), ST38 (31), ST12 (16), ST69 (13), ST648 (12), and ST73 (10). Other STs were comprised of <10 isolates.

Disclosures

REM, AS, JHK, and SRA are employees of Element Iowa City (JMI Laboratories). RK and DT are employees of GSK group of companies. This study at Element was supported by GSK. Element received compensation fees for services in relation to preparing the abstract and poster. This project has been funded in whole or in part with Federal funds from the Office of the Assistant Secretary for Preparedness and Response, Biomedical Advanced Research and Development Authority, under OTA Agreement No. HHSO100201300011C.

References

1. Bax BD et al. (2010). Type IIA topoisomerase inhibition by a new class of antibacterial agents. *Nature* 466: 935–40.

2. Gibson EG et al. (2019). Mechanistic and structural basis for the actions of the antibacterial gepotidacin against *Staphylococcus aureus* gyrase. *ACS infectious diseases* 5: 570–581.

3. Oviatt AA, et al. (2024). Interactions between gepotidacin and *Escherichia coli* gyrase and topoisomerase IV: Genetic and biochemical evidence for well-balanced dual targeting. *ACS Infectious Diseases* 10: 1137–1151.

4. Chan P, et al. (2020). In: IDWeek; Genetic evidence that gepotidacin shows well-balanced dual targeting against DNA gyrase and topoisomerase IV in *Neisseria gonorrhoeae* [poster], October 21–25, 2020; Philadelphia, USA.

5. Wagenlehner F, et al. (2024). Oral gepotidacin versus nitrofurantoin in patients with uncomplicated urinary tract infection (EAGLE-2 and EAGLE-3): Two randomised, controlled, double-blind, double-dummy, phase 3, non-inferiority trials. *Lancet* 403: 741–755.

6. Ross J, et al (2024). In: ECCMID; Oral gepotidacin for the treatment of uncomplicated urogenital gonorrhoea: results of a randomised, multicentre Phase 3 trial (EAGLE-1) [poster], April 27–30, 2024; Barcelona, Spain.

7. Clinical and Laboratory Standards Institute (2024). Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. *CLSI Standard M07 12th Edition*. Wayne, PA, USA.

8. Clinical and Laboratory Standards Institute (2024). Performance standards for antimicrobial susceptibility testing. *CLSI Standard M100 34th Edition*. Wayne, PA, USA.

Contact

Rodrigo E. Mendes, Ph.D., FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo.mendes@element.com

Presentation number P1060
IDWeek 2024 | October 16–19, 2024 | Los Angeles, California