IDWeek 2024

Poster #P411

Surveillance of omadacycline in vitro activity against 35,000 bacterial clinical isolates from the United States (2019–2023)

Michael D. Huband[1], Kelley A. Fedler[1], Kelly Wright[2], Rodrigo E. Mendes[1], and Mariana Castanheira[1]

[1] Element Iowa City (JMI Laboratories), North Liberty, Iowa, United States; [2] Paratek Pharmaceuticals, King of Prussia, Pennsylvania, United States

SCAN ME

Scan the QR code or use this link to download this poster: https://www.jmilabs.com/data/posters/IDWeek2024_23-PAR-04_A1_surveillance_summary.pdf

Michael D. Huband, Associate Director
Element Iowa City
345 Beaver Kreek Center
North Liberty, IA 52317
319-665-3370
michael.huband@element.com

INTRODUCTION

- Omadacycline is a tetracycline class (aminomethylcycline) antibacterial with potent activity against bacterial isolates expressing common mechanisms of resistance to tetracyclines, penicillins, fluoroquinolones, macrolides, β-lactams, and vancomycin.
- Omadacycline has previously demonstrated potent *in vitro* activity against Gram-positive (staphylococci, streptococci, and enterococci) and Gram-negative (*Enterobacter cloacae*, *Klebsiella pneumoniae, Escherichia coli,* and *Haemophilus influenzae*) bacterial pathogens commonly associated with acute bacterial skin and skin structure infections (ABSSSI) and community-acquired bacterial pneumonia (CABP), including isolates expressing resistance to tetracycline and other drug classes.
- Omadacycline is also being evaluated in clinical trials for the treatment of diabetic foot infection and acute osteomyelitis (NCT04714411), CABP (NCT04779242), pulmonary disease caused by *Mycobacterium abscessus* complex (NCT04922554), and pharmacokinetics of intravenous and oral omadacycline in children and adolescents with any suspected or confirmed bacterial infections (NCT05217537).
- This study determined the *in vitro* antibacterial activity and susceptibility of omadacycline and tetracycline class comparator agents including doxycycline, minocycline, tetracycline, and tigecycline against 35,000 bacterial isolates collected during the 2019–2023 post-marketing surveillance program (SENTRY and Paratek KEYSTONE JMI MVP (paratek-keystone.com).

MATERIALS AND METHODS

- Bacterial isolates (n=35,000) were recovered from patients (multiple infection types) with documented infections from medical centers in the United States (representing the 9 Census Divisions) using a prevalence mode study design (2019–2023).
- Isolates tested included 9,056 staphylococci, 3,186 streptococci, 1,820 enterococci, 1,352 *Haemophilus* spp., 612 *Moraxella* spp., 5,034 non-fermenters, and 13,939 Enterobacterales.
- Isolates were collected from patients with skin and skin structure infections (7,306 isolates; 20.9%), bloodstream infections (8,721 isolates; 24.9%), CABP (3,060 isolates; 8.7%), intra-abdominal infection (1,643 isolates; 4.7%), pneumonia in hospitalized patients (PIHP) (8,311 isolates; 23.8%), urinary tract infections (5,507 isolates; 15.7%), and other infections (452 isolates; 1.3%).
- Bacterial species identifications were confirmed at Element Iowa City using standard microbiology methods and matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany).
- Broth microdilution susceptibility testing was performed according to Clinical and Laboratory Standards Institute M07 (CLSI; 2024) and M100 (2024) guidelines.

RESULTS

- Omadacycline and tetracycline class comparator agent MIC values and susceptibility data against medically important Gram-positive and Gram-negative bacterial pathogens collected during the 2019–2023 omadacycline post-marketing surveillance program from medical centers in the United States are presented in Tables 1–2.
- Bacterial pathogen occurrence by infection type for omadacycline is presented in Figures 1–2 for ABSSSI and CABP.
- Omadacycline demonstrated potent *in vitro* antibacterial activity against 7,975 *Staphylococcus aureus* (98.2% susceptible [S]) isolates including 3,255 methicillin-resistant *S. aureus* (MRSA) (96.1% S), 4,720 methicillin-susceptible *S. aureus* (MSSA) (99.7% S), and 422 tetracycline-resistant (92.9% S) strains (Table 1).
- Overall, 99.3% of 148 *Staphylococcus lugdunensis* isolates ($MIC_{50/90}$, 0.03/0.06 mg/L) were susceptible to omadacycline (Table 1).
- 99.9% of 1,212 *Enterococcus faecalis* isolates were susceptible to omadacycline including 100.0% of 842 tetracycline-resistant strains and 100% of 30 vancomycin-resistant strains (Table 1).
- 98.5% of 132 *Streptococcus anginosus* group isolates were susceptible to omadacycline ($MIC_{50/90}$, 0.03/0.06 mg/L) including 95.9% of 49 tetracycline-resistant strains (Table 1).
- Omadacycline demonstrated potent *in vitro* antibacterial activity against 1,729 *Streptococcus pneumoniae* (99.9% S) isolates including 193 penicillin-resistant (100.0% S), 775 macrolide-resistant (99.9% S), and 343 tetracycline-resistant (99.7% S) strains (Table 1).
- 99.4% of 520 *Streptococcus pyogenes* isolates were susceptible to omadacycline ($MIC_{50/90}$, 0.06/0.12 mg/L) including 98.5% of 133 macrolide-resistant and 98.1% of 162 tetracycline-resistant strains (Table 1).
- Omadacycline was active against 1,211 *Haemophilus influenzae* isolates (99.9% S) and 141 *H. parainfluenzae* isolates (95.7% S), including 100.0% of the β-lactamase positive isolates (352 and 15 respectively) (Table 2).
- Susceptibility of 1,151 *Enterobacter cloacae* species complex isolates to omadacycline was 92.4% S (Table 2).
- Susceptibility of 2,584 *Klebsiella pneumoniae* isolates to omadacycline was 92.5% S including 81.4% of 485 ESBL-phenotype, 86.0% of 50 carbapenem-resistant, and 71.0% of 548 tetracycline-resistant strains (Table 2).
- Omadacycline inhibited ≥90.0% of *Acinetobacter baumannii* species complex, *Citrobacter freundii* species complex, *Citrobacter koseri*, *E. coli*, *Klebsiella aerogenes*, *Klebsiella oxytoca*, and *Moraxella catarrhalis* isolates at ≤4 μg/mL (Table 2).

Figure 1 Occurrence of acute bacterial skin and skin structure infection pathogens in the omadacycline surveillance program (*n*=7,306; 2019–2023)

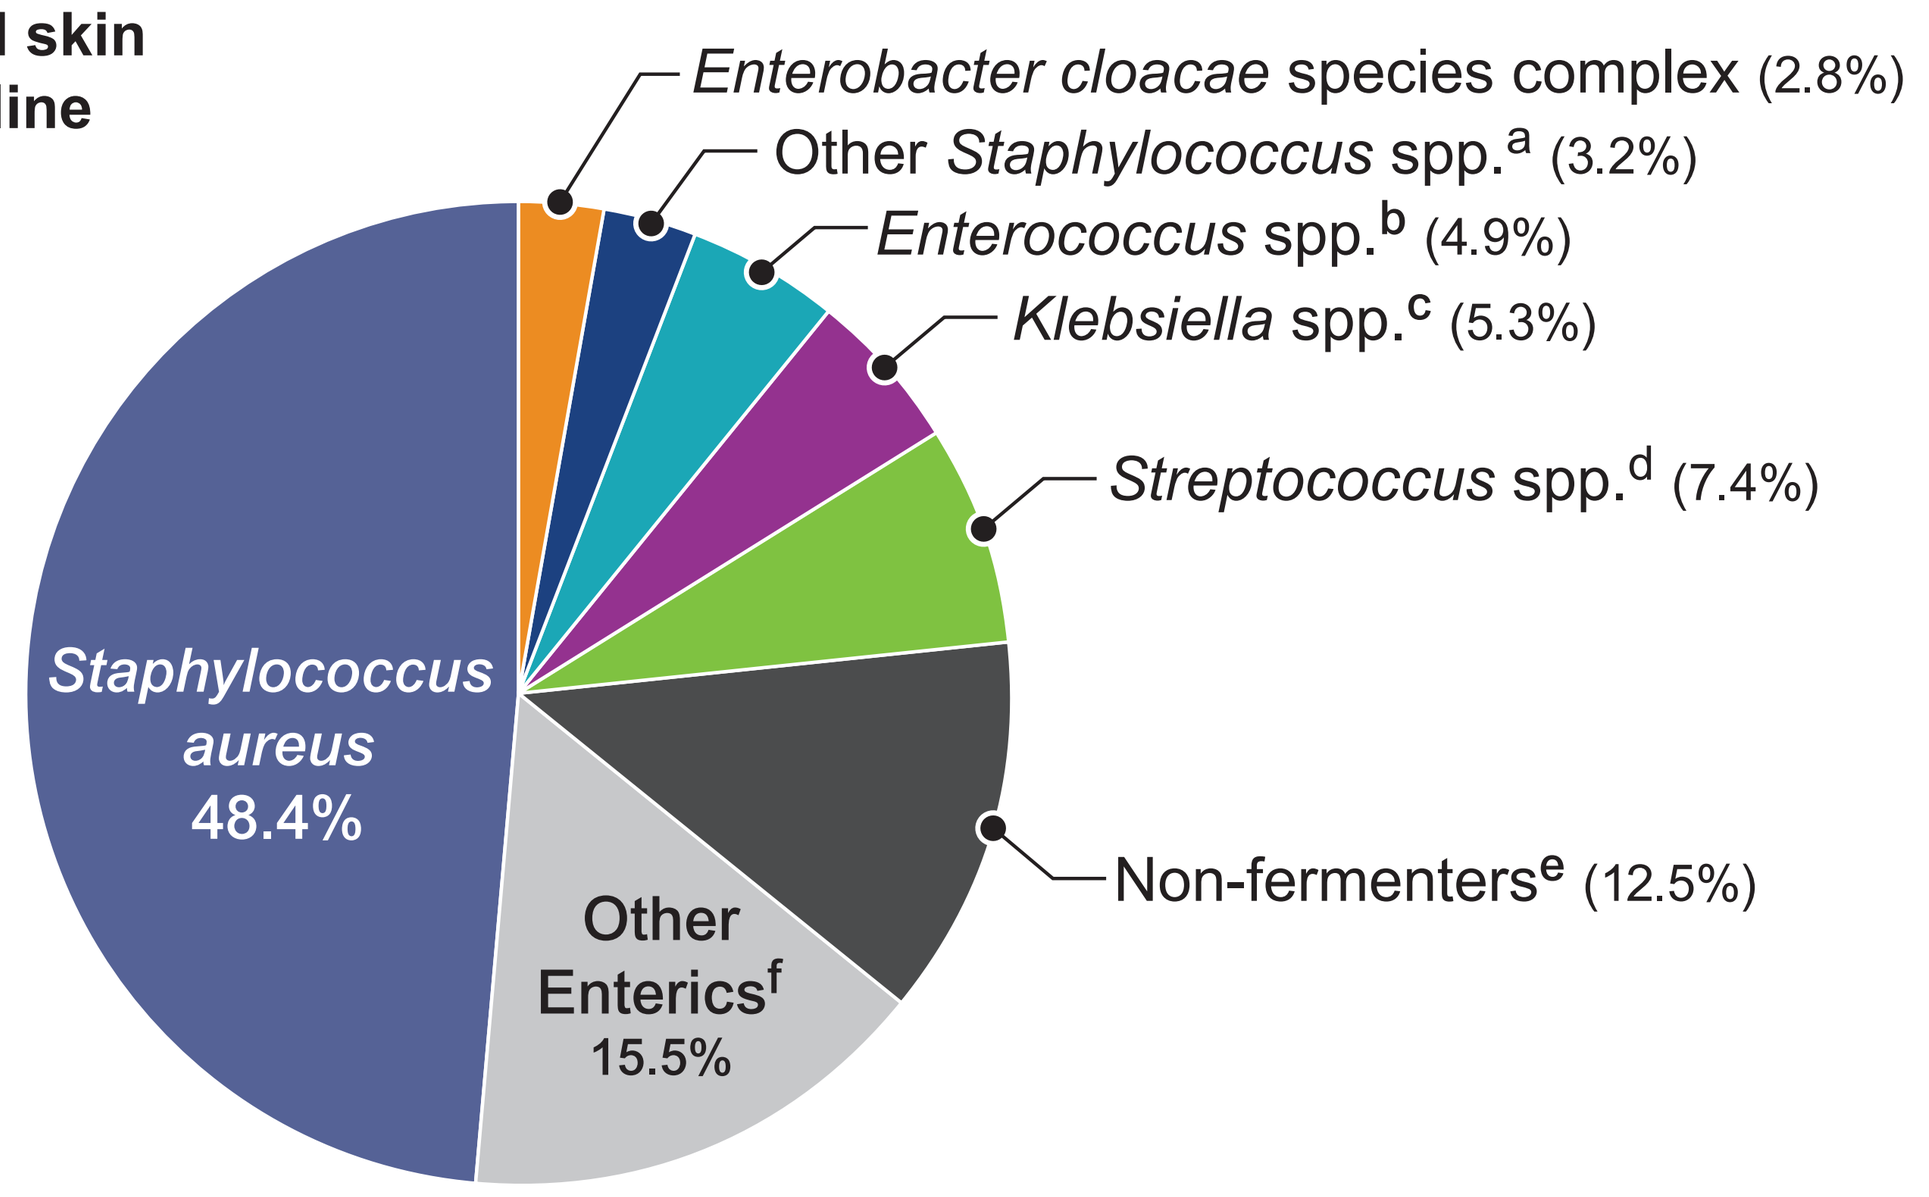

Enterobacter cloacae species complex (2.8%)
Other Staphylococcus spp.[a] (3.2%)
Enterococcus spp.[b] (4.9%)
Klebsiella spp.[c] (5.3%)
Streptococcus spp.[d] (7.4%)
Non-fermenters[e] (12.5%)
Other Enterics[f] 15.5%
Staphylococcus aureus 48.4%

[a] Contains *S. capitis* (0.08%), *S. caprae* (0.07%), *S. epidermidis* (1.4%), *S. haemolyticus* (0.08%), *S. hominis* (0.08%), *S. lugdunensis* (1.3%), *S. saprophyticus* (0.02%), *S. schleiferi* (0.02%), *S. simulans* (0.12%), and *S. warneri* (0.03%).
[b] Contains *Enterococcus avium* (0.1%), *E. casseliflavus* (0.1%), *E. faecalis* (3.6%), *E. faecium* (1.0%), *E. gallinarum* (0.1%), and *E. mundtii* (0.1%).
[c] Contains *K. aerogenes* (0.7%), *K. oxytoca* (1.5%), *K. pneumoniae* (3.05%), and *K. variicola* (0.05%).
[d] Contains *S. agalactiae* (2.1%), *S. anginosus* group (0.7%), *S. canis* (0.05%), *S. dysgalactiae* (0.7%), *S. mitis* group (0.1%), *S. pneumoniae* (0.15%), and *S. pyogenes* (3.6%).
[e] Contains *Acinetobacter* spp. (3.4%), *P. aeruginosa* (8.1%), and *S. maltophilia* (1.0%).
[f] Contains *Citrobacter* spp. (1.3%), *Cronobacter sakazakii* (0.01%), *Enterobacter* spp. (0.05%), *Escherichia coli* (5.6%), *Leclercia adecarboxylata* (0.1%), *Lelliottia amnigena* (0.04%), *Morganella morganii* (1.3%), *Pantoea agglomerans* (0.01%), *Pluralibacter gergoviae* (0.03%), *Proteus hauseri* (0.01%), *P. mirabilis* (3.0%), *P. penneri* (0.1%), *P. vulgaris* (0.3%), *P. vulgaris* group (0.2%), *Providencia rettgeri* (0.2%), *P. stuartii* (0.2%), *Raoultella ornithinolytica* (0.03%), *R. planticola* (0.01%), *Serratia liquefaciens* (0.1%), *S. liquefaciens* complex (0.02%), *Serratia marcescens* (2.8%), *S. plymuthica* (0.02%), unspeciated *Citrobacter* (0.03%), unspeciated *Providencia* (0.02%), and unspeciated *Raoultella* (0.03%).

Table 1 Antimicrobial activity of omadacycline and tetracycline class comparator agents against Gram-positive bacterial isolates collected from medical centers in the United States (2019–2023)

Organism (no. tested)	Omadacycline			Doxycycline			Minocycline			Tetracycline			Tigecycline		
	MIC_{50}	MIC_{90}	% S (FDA)	MIC_{50}	MIC_{90}	% S (CLSI)	MIC_{50}	MIC_{90}	% S (CLSI)	MIC_{50}	MIC_{90}	% S (CLSI)	MIC_{50}	MIC_{90}	% S (FDA)
Staphylococcus aureus (7,975)	0.12	**0.12**	98.2[a]	≤0.06	0.5	98.2	0.06	0.12	98.3	≤0.5	1	93.7	0.06	0.12	99.9
MRSA (3,255)	0.12	**0.25**	96.1[a]	≤0.06	1	96.9	0.06	0.25	96.8	≤0.5	4	90.4	0.06	0.12	99.9
MSSA (4,720)	0.12	**0.12**	99.7[b]	≤0.06	0.12	99.1	0.06	0.12	99.3	≤0.5	≤0.5	95.9	0.06	0.12	100.0
Tetracycline-R (422)	0.12	**0.5**	92.9[a]	4	8	66.1	2	>8	68.9	>8	>8	0.0	0.12	0.25	99.8
Staphylococcus capitis (75)	0.12	**0.25**	—[c]	≤0.06	1	100.0	0.06	0.12	100.0	≤0.5	>8	88.0	0.12	0.25	—
Staphylococcus epidermidis (614)	0.25	**0.5**	—	0.5	4	92.2	0.12	0.5	95.8	1	>8	78.2	0.12	0.25	—
Staphylococcus haemolyticus (41)	0.12	**1**	—	0.5	4	90.2	0.12	0.25	100.0	1	>8	58.5	0.12	0.25	—
Staphylococcus hominis (100)	0.12	**0.25**	—	0.12	4	96.0	0.12	0.25	98.9	≤0.5	>8	67.0	0.06	0.25	—
Staphylococcus lugdunensis (148)	0.03	**0.06**	99.3[a]	≤0.06	≤0.06	100.0	≤0.03	0.06	100.0	≤0.5	≤0.5	—	0.03	0.06	—
Streptococcus agalactiae (413)	0.12	**0.12**	—	>1	>1	—	>1	>1	—	>4	>4	16.0	0.06	0.06	100.0
Macrolide-R (269)	0.12	**0.12**	—	>1	>1	—	>1	>1	—	>4	>4	10.4	0.06	0.06	100.0
Tetracycline-R (344)	0.12	**0.12**	—	>1	>1	—	>1	>1	—	>4	>4	0.0	0.06	0.06	100.0
Streptococcus anginosus group (132)	0.03	**0.06**	98.5[a]	0.25	>1	—	0.06	>1	—	0.5	>4	58.3	0.03	0.06	100.0
Tetracycline-R (49)	0.06	**0.12**	95.9[a]	>1	>1	—	>1	>1	—	>4	>4	0.0	0.03	0.06	100.0
Streptococcus dysgalactiae (135)	0.12	**0.25**	—	0.25	>1	—	0.12	>1	—	0.25	>4	62.7	0.06	0.12	100.0
Streptococcus mitis group (167)	0.06	**0.12**	—	0.25	>1	—	0.12	>1	—	1	>4	56.0	0.03	0.12	100.0
Streptococcus pneumoniae (1,729)[d]	0.06	**0.06**	99.9[b]	0.12	>1	79.9	0.12	>1	—	0.25	>4	79.7	0.03	0.06	98.7
Penicillin-R (193)	0.06	**0.06**	100.0[b]	0.25	>1	54.7	0.12	>1	—	0.5	>4	56.0	0.03	0.06	99.0
Macrolide-R (775)	0.06	**0.06**	99.9[b]	0.12	>1	58.3	0.12	>1	—	0.25	>4	58.9	0.03	0.06	99.0
Tetracycline-R (343)	0.06	**0.12**	99.7[b]	>1	>1	0.4	>1	>1	—	>4	>4	0.0	0.03	0.06	98.8
S. pyogenes (520)	0.06	**0.12**	99.4[a]	0.12	>1	—	0.12	>1	—	0.25	>4	68.7	0.06	0.06	100.0
Macrolide-R (133)	0.06	**0.12**	98.5[a]	>1	>1	—	>1	>1	—	>4	>4	13.5	0.06	0.06	100.0
Tetracycline-R (162)	0.06	**0.12**	98.1[a]	>1	>1	—	>1	>1	—	>4	>4	0.0	0.06	0.06	100.0
Enterococcus faecalis (1,212)	0.06	**0.12**	99.9[a]	4	8	61.7	8	>8	40.0	>16	>16	30.0	0.06	0.12	99.8[a]
Tetracycline-R (842)	0.06	**0.25**	100.0[a]	8	>8	45.0	8	>8	14.0	>16	>16	0.0	0.06	0.12	99.8[a]
Vancomycin-R (30)	0.12	**0.12**	100.0[a]	8	8	34.8	8	>8	16.7	>16	>16	0.0	0.06	0.12	96.7[a]
E. faecium (532)	0.06	**0.12**	—	4	8	60.8	4	>8	52.8	>16	>16	26.0	0.06	0.12	—[e]
Tetracycline-R (380)	0.06	**0.12**	—	8	16	45.9	8	>8	33.9	>16	>16	0.0	0.06	0.12	—[e]
Vancomycin-R (344)	0.06	**0.12**	—	4	8	56.9	8	>8	48.8	>16	>16	15.5	0.06	0.12	—[e]

CLSI, Clinical and Laboratory Standards Institute; ESBL, extended-spectrum β-lactamase; FDA, United States Food and Drug Administration; MRSA, methicillin-resistant *S. aureus*; MSSA, methicillin-susceptible *S. aureus*; R, resistant.

[a] ABSSSI breakpoint applied.
[b] CABP breakpoint applied.
[c] Breakpoint interpretive criteria were unavailable.
[d] Oral dosing (penicillin MIC, ≥2 mg/L).
[e] FDA breakpoints for vancomycin-susceptible isolates were applied to all isolates.

Bold, omadacycline MIC_{90} values.
Green, susceptible according to CLSI or FDA breakpoint interpretive criteria.
Yellow, intermediate according to CLSI or FDA breakpoint interpretive criteria.
Orange, resistant according to CLSI or FDA breakpoint interpretive criteria.

Table 2 Antimicrobial activity of omadacycline and tetracycline class comparator agents against Gram-negative bacterial isolates collected from medical centers in the United States (2019–2023)

Organism (no. tested)	Omadacycline			Doxycycline			Minocycline			Tetracycline			Tigecycline		
	MIC_{50}	MIC_{90}	%S (FDA)	MIC_{50}	MIC_{90}	%S (CLSI)	MIC_{50}	MIC_{90}	%S (CLSI)	MIC_{50}	MIC_{90}	%S (CLSI)	MIC_{50}	MIC_{90}	%S (FDA)
Acinetobacter baumannii-calcoaceticus species complex (792)	0.25	**4**	—	0.25	>8	80.4	0.12	8	88.4	2	>16	64.0	0.25	2	—
Citrobacter freundii species complex (328)	1	**4**	—	2	>8	81.7	1	8	86.3	1	>16	82.9	0.25	0.5	100.0
C. koseri (262)	0.5	**1**	—	0.5	2	98.9	0.5	2	99.6	1	2	98.5	0.25	0.5	100.0
Enterobacter cloacae species complex (1,151)	2	**4**	92.4[a]	2	8	87.3	2	8	88.3	2	16	86.5	0.5	1	97.1
Escherichia coli (5,798)	0.5	**1**	—	1	>8	76.5	0.5	4	90.7	2	>16	71.7	0.12	0.25	99.9
ESBL-phenotype (1,088)	1	**2**	—	4	>8	53.5	1	8	85.6	>16	>16	41.8	0.25	0.25	100.0
Carbapenem-R (11)	1	**4**	—	4	>8	63.6	2	4	90.9	2	>16	63.6	0.25	0.5	100.0
Haemophilus influenzae (1,211)	1	**2**	99.9[b]	—	—	—	—	—	—	0.5	0.5	99.0	0.25	0.25	90.8
β-lactamase positive (352)	0.5	**1**	100.0[b]	—	—	—	—	—	—	0.5	0.5	97.7	0.25	0.5	89.8
Haemophilus parainfluenzae (141)	1	**2**	95.7[b]	—	—	—	—	—	—	0.5	1	90.8	0.25	0.5	—
β-lactamase positive (15)	1	**2**	100.0[b]	—	—	—	—	—	—	0.5	8	86.7	0.25	0.5	—
Klebsiella aerogenes (472)	1	**2**	—	1	4	92.4	1	4	94.3	1	4	92.4	0.25	0.5	98.5
Klebsiella oxytoca (619)	1	**2**	—	0.5	4	93.2	1	2	95.1	≤0.5	4	92.2	0.25	0.5	99.8
Klebsiella pneumoniae (2,584)	1	**4**	92.5[a,b]	1	>8	76.5	1	16	84.1	1	>16	79.7	0.5	1	97.5
ESBL-phenotype (485)	2	**16**	81.4[a,b]	8	>8	36.7	4	>32	67.2	>16	>16	34.9	0.5	2	93.6
Carbapenem-R (50)	2	**8**	86.0[a,b]	4	>8	64.0	4	32	66.0	4	>16	58.0	0.5	2	98.0
Tetracycline-R (548)	4	**16**	71.0[a,b]	>8	>8	2.6	16	>32	38.7	>16	>16	0.0	0.5	4	88.8
Moraxella catarrhalis (604)	≤0.12	**0.25**	—	—	—	—	—	—	—	0.25	0.25	99.2	0.06	0.06	—
Morganella morganii (278)	4	**16**	—	4	>8	54.7	4	32	58.3	2	>16	52.5	1	2	96.0
Serratia marcescens (928)	4	**8**	—	4	8	61.5	2	4	91.9	>16	>16	4.8	1	1	97.6
Stenotrophomonas maltophilia (753)	2	**8**	—	2	4	—	0.5	1	92.1	16	>16	—	1	4	—

CLSI, Clinical and Laboratory Standards Institute; ESBL, extended-spectrum β-lactamase; FDA, United States Food and Drug Administration; R, resistant.
[a] ABSSSI breakpoint applied.
[b] CABP breakpoint applied.

Bold, MIC_{90} values.
Green, susceptible according to CLSI or FDA breakpoint interpretive criteria.
Yellow, intermediate according to CLSI or FDA breakpoint interpretive criteria.
Orange, resistant according to CLSI or FDA breakpoint interpretive criteria.

Figure 2 Occurrence of community-acquired bacterial pneumonia pathogens in the omadacycline surveillance program (*n*=3,060; 2019–2023)

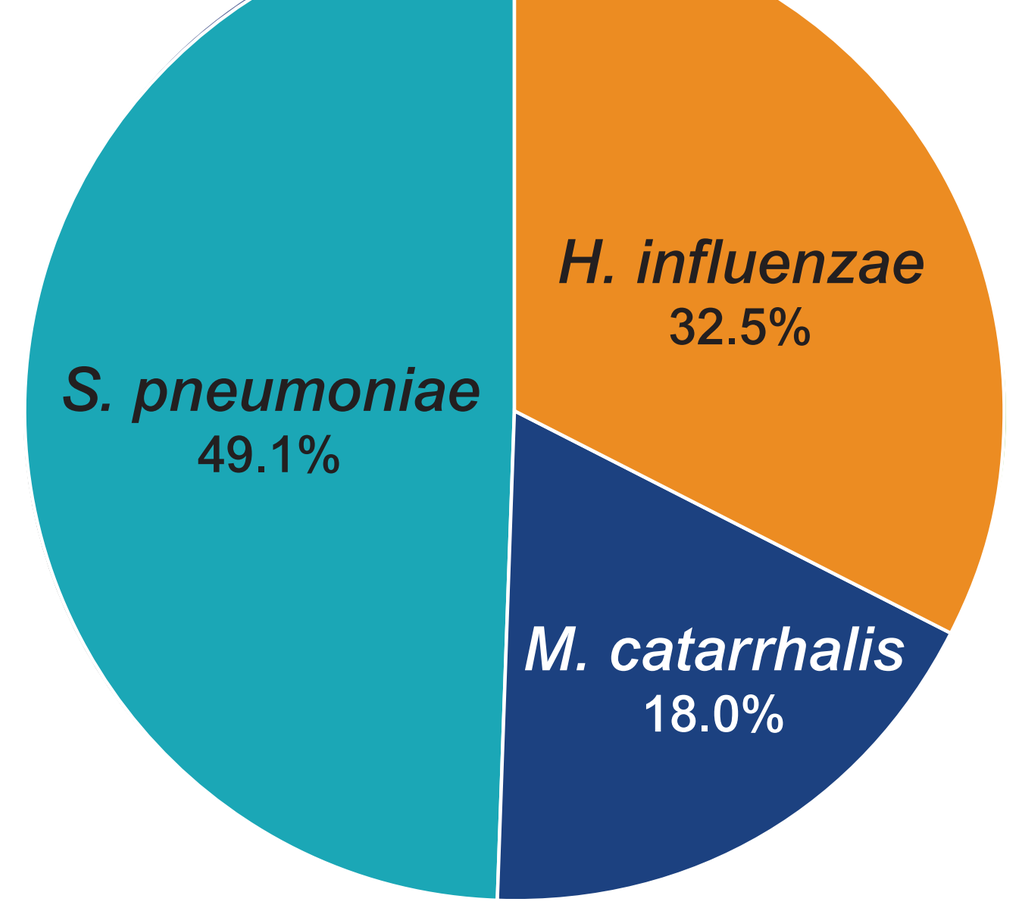

H. influenzae
32.5%
S. pneumoniae
49.1%
M. catarrhalis
18.0%

CONCLUSIONS

- Omadacycline demonstrated potent *in vitro* antibacterial activity with longitudinal consistency over the 5-year period after FDA approval.
- There was no evidence of increasing omadacycline MICs over time against medically important Gram-positive and -negative bacterial pathogens including strains expressing resistance to macrolides, oxacillin/penicillin, vancomycin, tetracycline, and β-lactams.
- With a few exceptions, the MIC_{50} and MIC_{90} values of the reported pathogens were the same or within a two-fold dilution of the resistant phenotypes of interest, further demonstrating omadacycline maintains potency regardless of tetracycline resistance and resistance to other antibiotic classes.
- The results of this surveillance study support the continued use of omadacycline, especially in ABSSSI and CABP infections where resistant pathogens are likely to be encountered and warrant further investigation in disease states where omadacycline is not an FDA-approved treatment.

ACKNOWLEDGEMENTS

This study has been funded in whole or in part by federal funds from the U.S. Department of Health and Human Services, Administration for Strategic Preparedness and Response, Biomedical Advanced Research and Development Authority (BARDA), under Contract No. 75A50120C00001.

This study and poster presentation were also funded, in part, by Paratek Pharmaceuticals, Inc.

REFERENCES

Clinical and Laboratory Standards Institute (2024). M07-A12. *Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard—eleventh edition*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2024). M100-S34. *Performance standards for antimicrobial susceptibility testing: 34th informational supplement.* Wayne, PA: CLSI.

EUCAST (2024). Breakpoint tables for interpretation of MICs and zone diameters. Version 14.0, January 2024. Available at: http://www.eucast.org/clinical_breakpoints/. Accessed July 2024.

US Food and Drug Administration: https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria. Accessed July 2024.