Activity of Ibrexafungerp and Comparator Antifungals Tested Against *Candida* and *Aspergillus* Isolates Collected from Invasive Infections in a Global Surveillance Program in 2023

Marisa L Winkler[1], Paul Rhomberg[1], Abigail L. Klauer[1], Samuel Edeker[1], Sharon Min[2], Mariana Castanheira[1]

[1]Element Iowa City (JMI Laboratories), North Liberty, IA, USA; [2]GSK, Collegeville, PA, USA

Digital poster

SCAN ME

SCAN ME

https://www.jmilabs.com/data/posters/IDWeek2024_24-GSK-05_A1_ibrexa_surveillance.pdf

Introduction

- Ibrexafungerp is a novel triterpenoid antifungal agent approved by the United States Food and Drug Administration for the treatment of vulvovaginal candidiasis.
- There are ongoing clinical trials of ibrexafungerp in the treatment of invasive candidiasis.
- Rising azole resistance rates complicate the treatment of *Candida* infections and ibrexafungerp is the only non-azole antifungal agent with an approved oral formulation.
- We evaluated the *in vitro* activity of ibrexafungerp; echinocandins—micafungin and anidulafungin; azoles—fluconazole (yeast only), itraconazole, voriconazole; and amphotericin B against a global collection of *Candida* spp. and *Aspergillus* spp. isolates causing invasive infections in 2023.

Methods

- A total of 1,119 *Candida* spp. isolates and 203 *Aspergillus* spp. isolates were collected in 2023 as a part of the SENTRY global surveillance program.
- These isolates were tested for susceptibility to ibrexafungerp, micafungin, anidulafungin, fluconazole, itraconazole, voriconazole, amphotericin B (Table 1) and caspofungin and posaconazole (data not included). Isolates included the 6 most common *Candida* spp. including *Candida auris* and the 3 most common *Aspergillus* spp.
- Isolates were from 11 medical centers in North America, 18 medical centers in Europe, 7 medical centers in the Asia-West Pacific region, and 5 medical centers in Latin America (Figure 1).
- Only the first isolate collected per patient was submitted and tested.
- Isolate identification was performed by MALDI-TOF MS and/or molecular methods.
- Susceptibility testing was performed by broth microdilution according to CLSI standards. Minimum inhibitory concentrations (MICs) were determined for yeast with all agents and for molds with azoles and amphotericin B. Minimum effective concentrations (MECs) were determined for molds with ibrexafungerp and echinocandins.
- There are no current breakpoints for ibrexafungerp by CLSI criteria, but CLSI breakpoints were applied for comparator agents where available and CDC tentative breakpoints were used for *C. auris.*

Table 1. MIC/MEC$_{50/90}$ (mg/L) for ibrexafungerp (IBX) and comparator agents against 5 most common *Candida* spp., *C. auris*, and 3 most common *Aspergillus* spp. in 2023 SENTRY Antifungal Surveillance Program

Organism (#)	IBX	MFG	AND	FLC	ITC	VRC	AMB
C. albicans (395)	0.06/0.12	0.015/0.015	0.03/0.06	0.25/0.5	0.06/0.12	0.008/0.015	0.5/1
C. glabrata (246)	0.25/0.5	0.015/0.03	0.12/0.12	4/8	0.5/1	0.06/0.12	1/1
C. parapsilosis (181)	0.25/0.5	1/1	2/2	0.5/32	0.12/0.25	0.008/0.25	1/1
C. tropicalis (134)	0.12/0.25	0.015/0.06	0.03/0.06	0.5/1	0.12/0.25	0.03/0.06	1/1
C. krusei (33)	0.5/1	0.12/0.12	0.06/0.06	32/32	0.5/1	0.25/0.25	1/2
C. auris (25)	0.5/1	0.25/1	0.5/1	64/>128	0.25/1	0.25/1	2/2
A. fumigatus (141)	0.03/0.06	0.004/0.008	0.015/0.03	NT	1/1	0.5/1	1/2
A. section *Nigri* (29)	0.03/0.03	0.004/0.008	0.004/0.008	NT	1/>8	1/4	0.25/0.5
A. section *Flavi* (19)	0.03/0.06	≤0.002/0.008	0.004/0.008	NT	0.5/0.5	0.5/1	1/2

MIC, minimum inhibitory concentration; MEC, minimum effective concentration (for *Aspergillus* and ibrexafungerp or echinocandins); IBX, ibrexafungerp; MFG, micafungin; AND, anidulafungin; FLC, fluconazole; ITC, itraconazole; VRC, voriconazole; AMB, amphotericin B; NT, not tested

Results

Overall

- Of the 1119 *Candida* spp. isolates tested, 20 species were represented (Figure 2).
- The range of ibrexafungerp MICs for all *Candida* spp. was ≤0.004 mg/L to 2 mg/L with an overall MIC$_{50}$ of 0.12 mg/L and MIC$_{90}$ of 0.5 mg/L.
- Of the 203 *Aspergillus* spp. isolates tested, 8 species were represented (Figure 3).
- The range of ibrexafungerp MICs for all *Aspergillus* spp. was 0.008 mg/L to 8 mg/L with an overall MIC$_{50}$ of 0.03 mg/L and MIC$_{90}$ of 0.06 mg/L.
- Species of interest are discussed in more depth below.

***Candida albicans* n=395**

- Ibrexafungerp MICs ranged from 0.015–0.5 mg/L with MIC$_{50}$ of 0.06 mg/L and MIC$_{90}$ of 0.12 mg/L (Figure 4, Table 1).
- The MIC$_{90}$ for ibrexafungerp was 2 to 8x higher than echinocandins, 4x lower than fluconazole, the same as itraconazole, 8x higher than voriconazole, and 8x lower than amphotericin B.
- One isolate was resistant to anidulafungin, two to micafungin, and one to fluconazole.

***Candida glabrata* n=246**

- Ibrexafungerp MICs ranged from ≤0.004–2 mg/L with MIC$_{50}$ of 0.25 mg/L and MIC$_{90}$ of 0.5 mg/L (Figure 5, Table 1).
- The MIC$_{90}$ for ibrexafungerp was 4 to 16x higher than echinocandins, 16x lower than fluconazole, 2x lower than itraconazole, 4x higher than voriconazole, and 2x lower than amphotericin B.
- There were 3 isolates resistant to anidulafungin and micafungin and 8 isolates resistant to fluconazole.

***Candida parapsilosis* n=181**

- Ibrexafungerp MICs ranged from 0.06–1 mg/L with MIC$_{50}$ of 0.25 mg/L and MIC$_{90}$ of 0.5 mg/L (Table 1).
- The MIC$_{90}$ for ibrexafungerp was 2 to 4x lower than echinocandins, 64x lower than fluconazole, 2x higher than itraconazole, 2x higher than voriconazole, and 2x lower than amphotericin B.
- No isolates were resistant to anidulafungin or micafungin; 24 isolates were resistant to fluconazole.

***Candida auris* n=25**

- Ibrexafungerp MICs ranged from 0.25–1 mg/L with MIC$_{50}$ of 0.5 mg/L and MIC$_{90}$ of 1 mg/L (Figure 6, Table 1).
- The MIC$_{90}$ for ibrexafungerp was the same as the echinocandins, >128x lower than fluconazole, the same as itraconazole and voriconazole, and 2x lower than amphotericin B.
- Of the 25 isolates tested, 8% (2) were resistant to anidulafungin and micafungin (both isolates from the same hospital), 100% (25) were resistant to fluconazole, and 56% (14) were resistant to amphotericin B.

***Aspergillus fumigatus* n=141**

- Ibrexafungerp MECs ranged from 0.008–0.25 mg/L with MEC$_{50}$ of 0.03 mg/L and MEC$_{90}$ of 0.06 mg/L (Figure 7, Table 1).
- MEC/MIC$_{90}$ for ibrexafungerp was 2 to 8x higher than that of echinocandins and ≥16x lower than that of itraconazole, voriconazole, and amphotericin B.

Figure 1: Geographic distribution of isolates

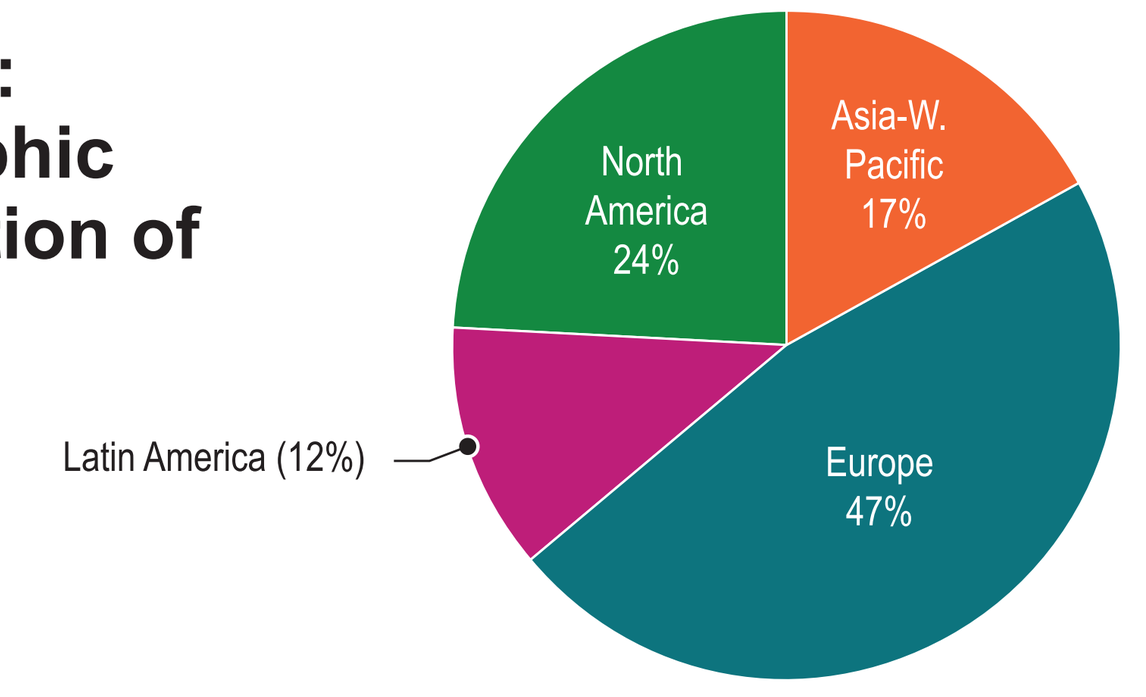
North America 24%
Asia-W. Pacific 17%
Europe 47%
Latin America (12%)

Figure 2: Species of *Candida* isolates included in surveillance (number of isolates, %)

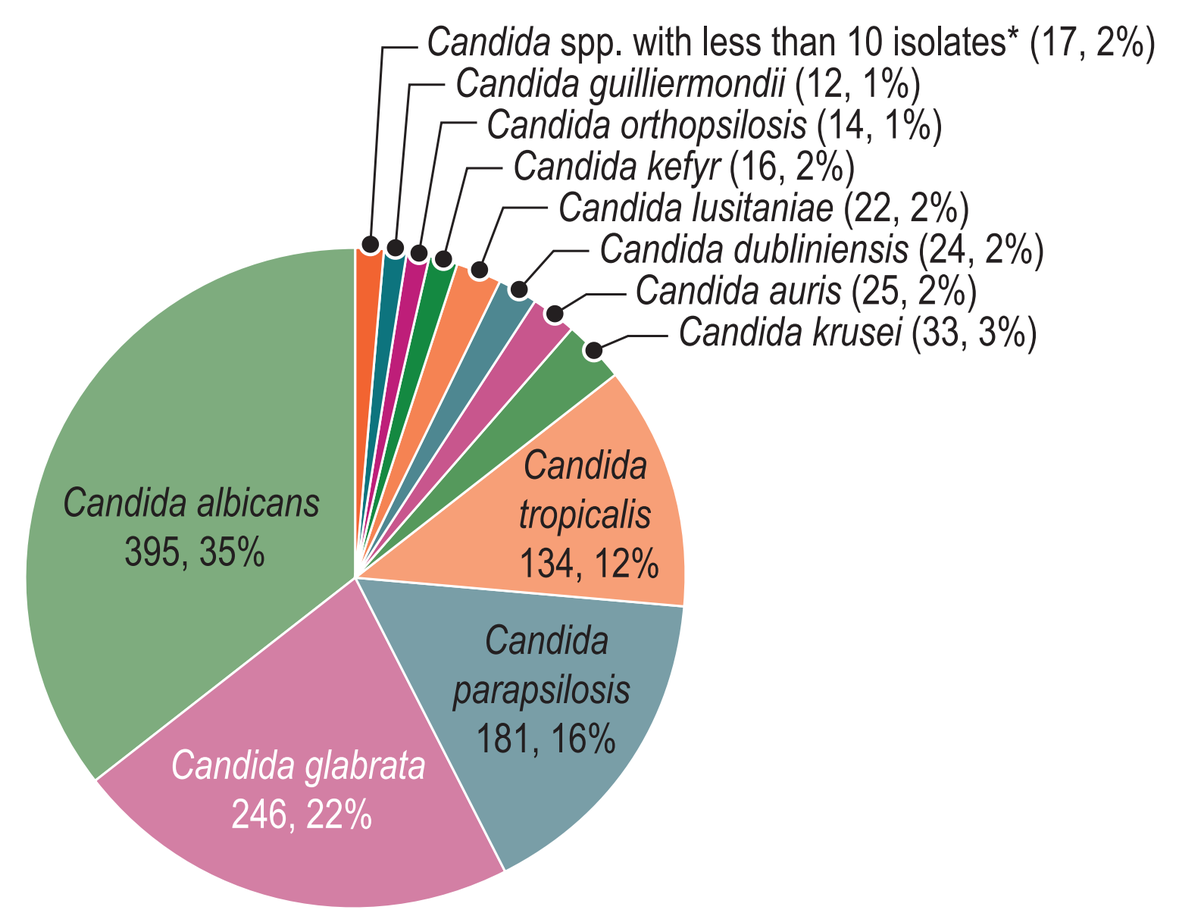
Candida spp. with less than 10 isolates* (17, 2%)
Candida guilliermondii (12, 1%)
Candida orthopsilosis (14, 1%)
Candida kefyr (16, 2%)
Candida lusitaniae (22, 2%)
Candida dubliniensis (24, 2%)
Candida auris (25, 2%)
Candida krusei (33, 3%)
Candida tropicalis 134, 12%
Candida parapsilosis 181, 16%
Candida glabrata 246, 22%
Candida albicans 395, 35%

* Includes *Candida pelliculosa, Candida fabianii,* Unspeciated *Candida, Candida metapsilosis, Candida lipolytica, Candida fermentati, Candida rugosa, Candida pulcherrima, Candida norvegensis*

Figure 3: *Aspergillus* spp. isolates included in surveillance (number of isolates, %)

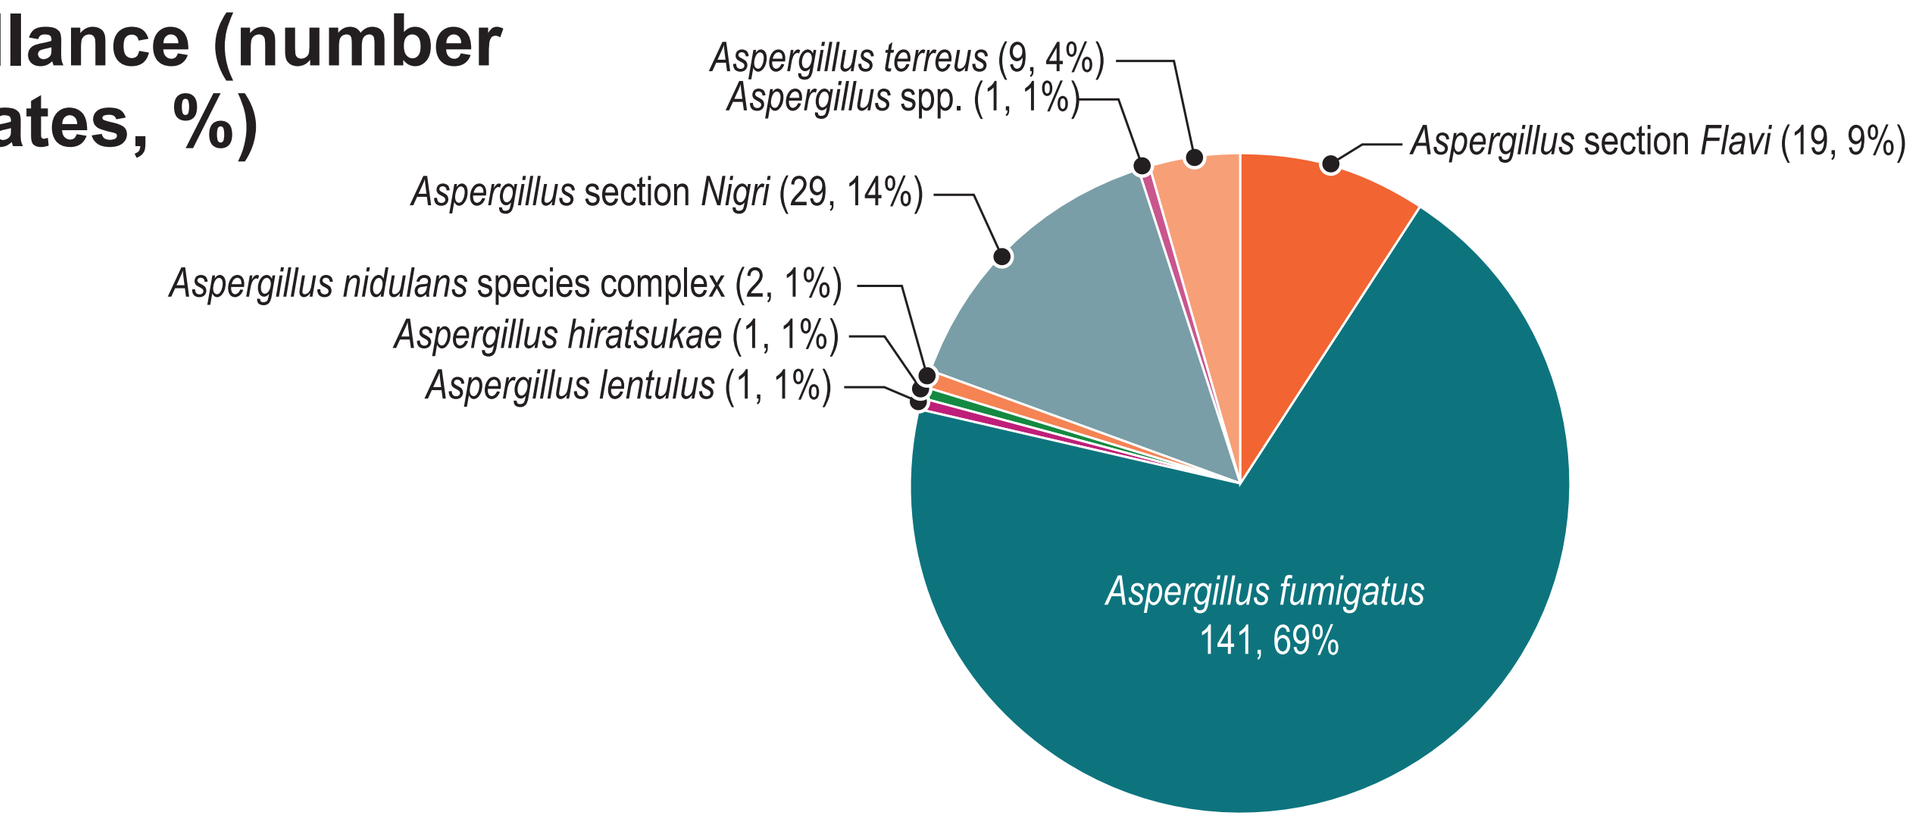
Aspergillus terreus (9, 4%)
Aspergillus spp. (1, 1%)
Aspergillus section Flavi (19, 9%)
Aspergillus section Nigri (29, 14%)
Aspergillus nidulans species complex (2, 1%)
Aspergillus hiratsukae (1, 1%)
Aspergillus lentulus (1, 1%)
Aspergillus fumigatus 141, 69%

Figure 4: *Candida albicans* susceptibility testing (n = 395)

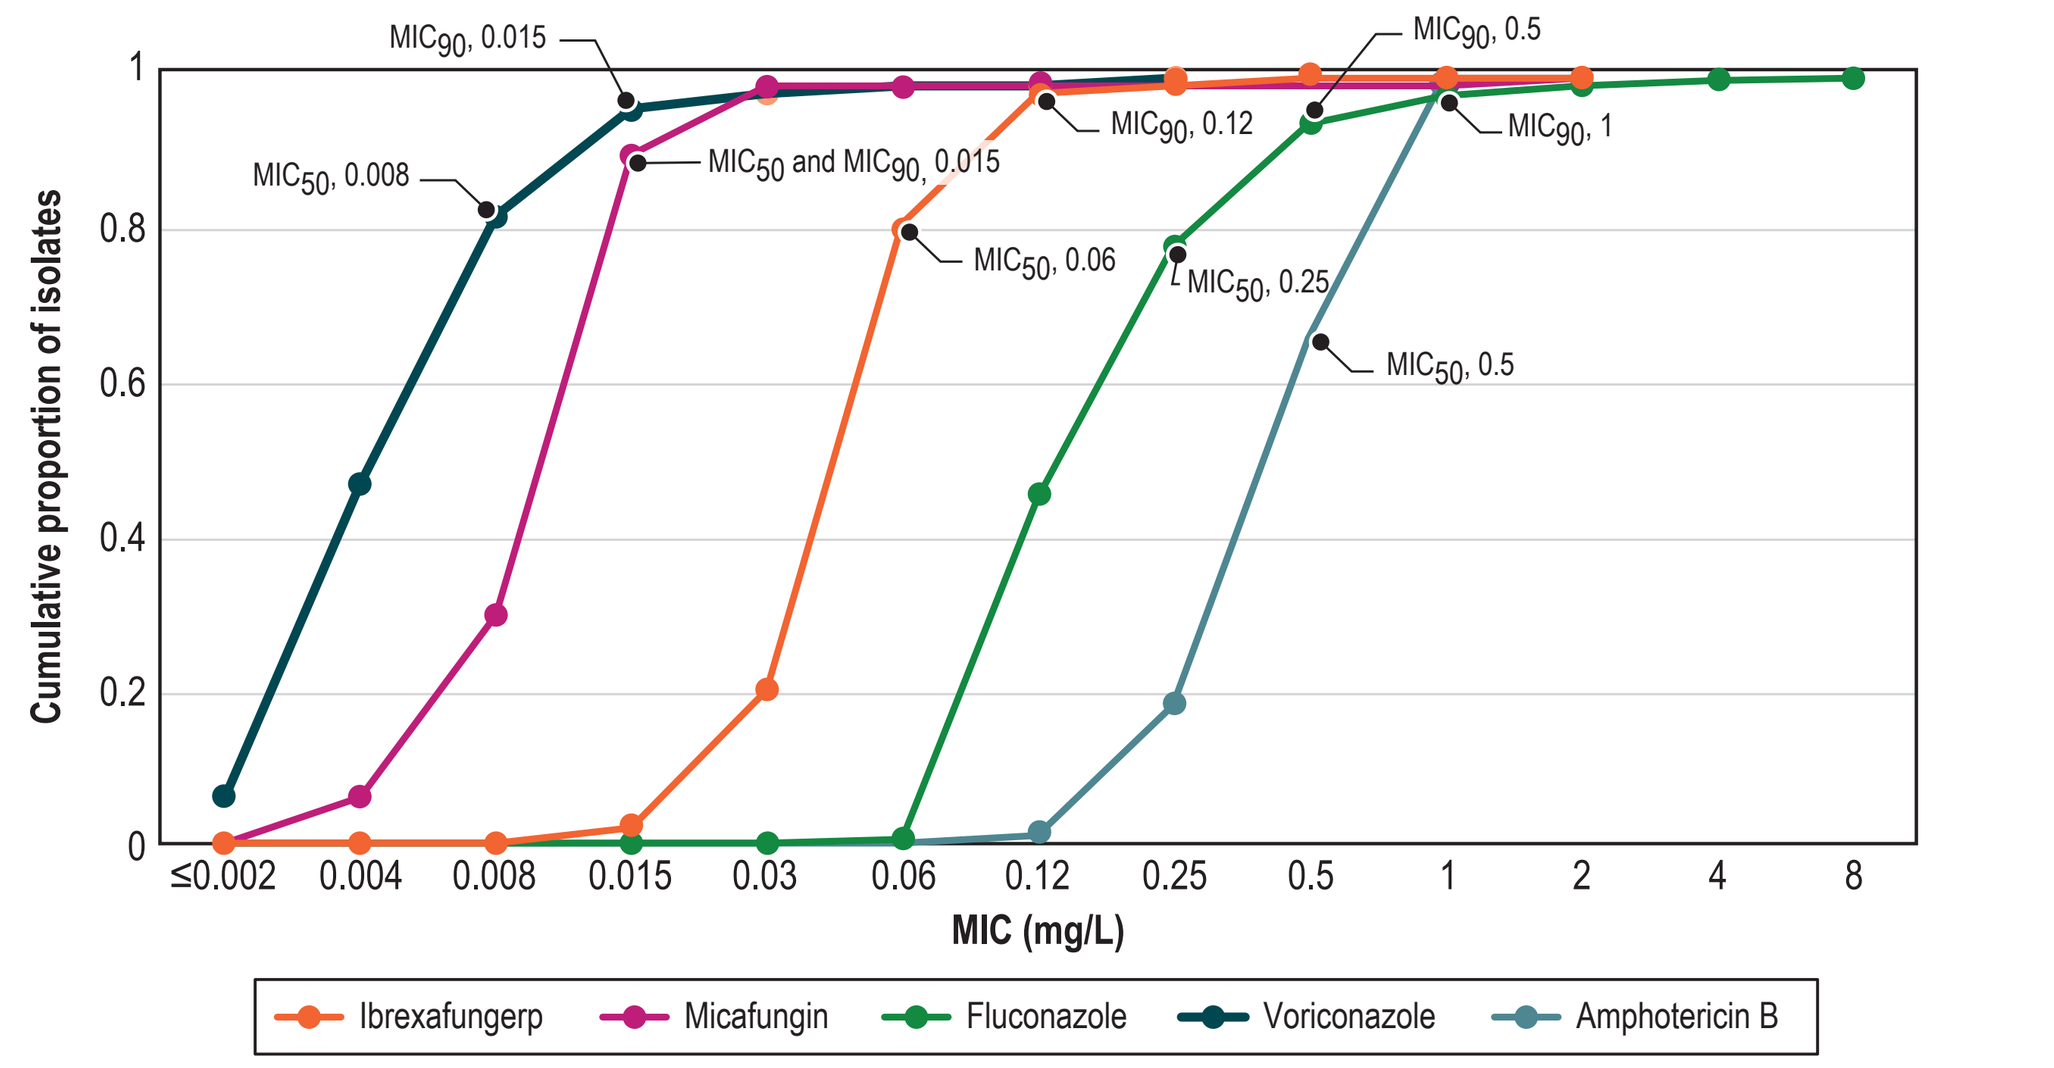
MIC90, 0.015
MIC50, 0.008
MIC50 and MIC90, 0.015
MIC90, 0.12
MIC50, 0.06
MIC50, 0.25
MIC90, 0.5
MIC90, 1
MIC50, 0.5
Cumulative proportion of isolates
MIC (mg/L)
Ibrexafungerp
Micafungin
Fluconazole
Voriconazole
Amphotericin B

Figure 5: *Candida glabrata* susceptibility testing (n = 246)

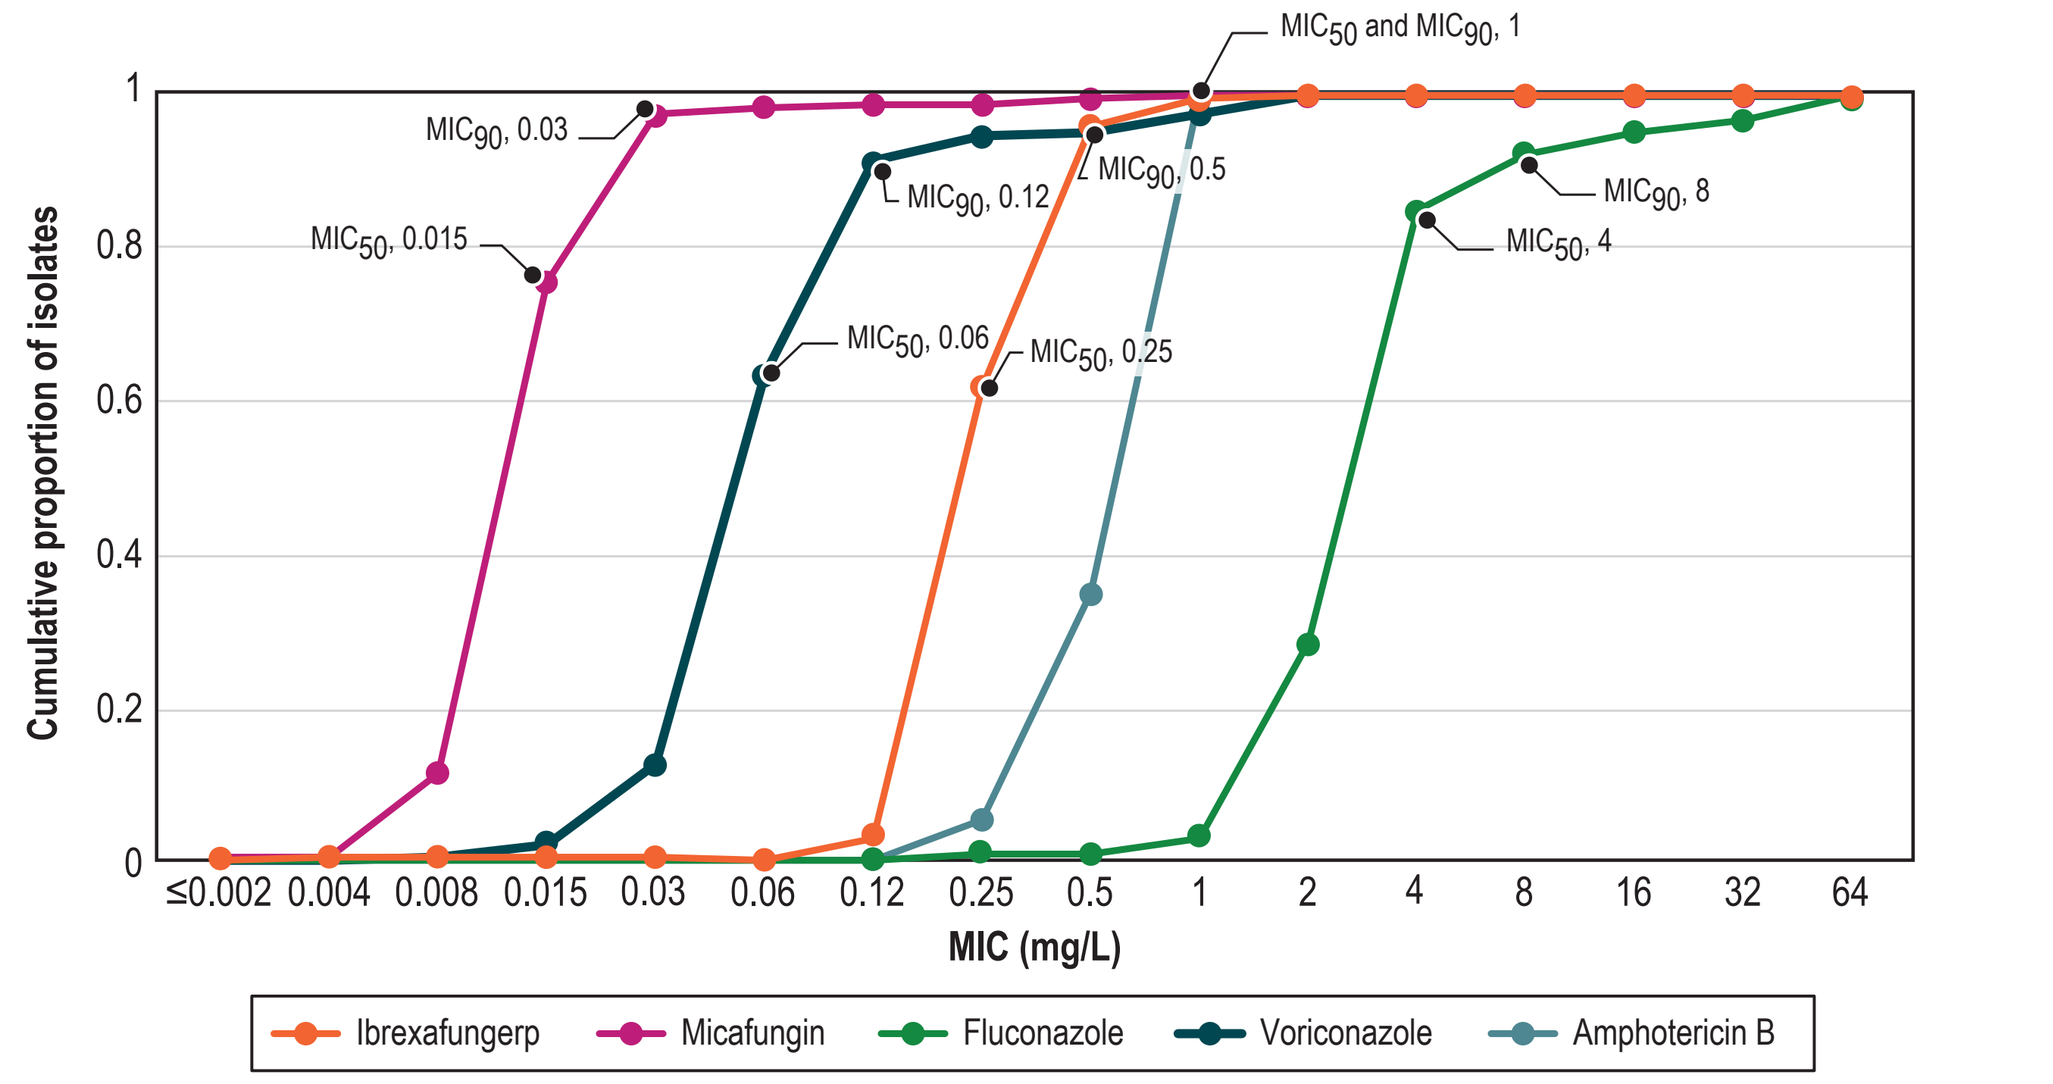
MIC50 and MIC90, 1
MIC90, 0.03
MIC50, 0.015
MIC90, 0.12
MIC90, 0.5
MIC50, 0.06
MIC50, 0.25
MIC90, 8
MIC50, 4
Cumulative proportion of isolates
MIC (mg/L)
Ibrexafungerp
Micafungin
Fluconazole
Voriconazole
Amphotericin B

Figure 6: *Candida auris* susceptibility testing (n = 25)

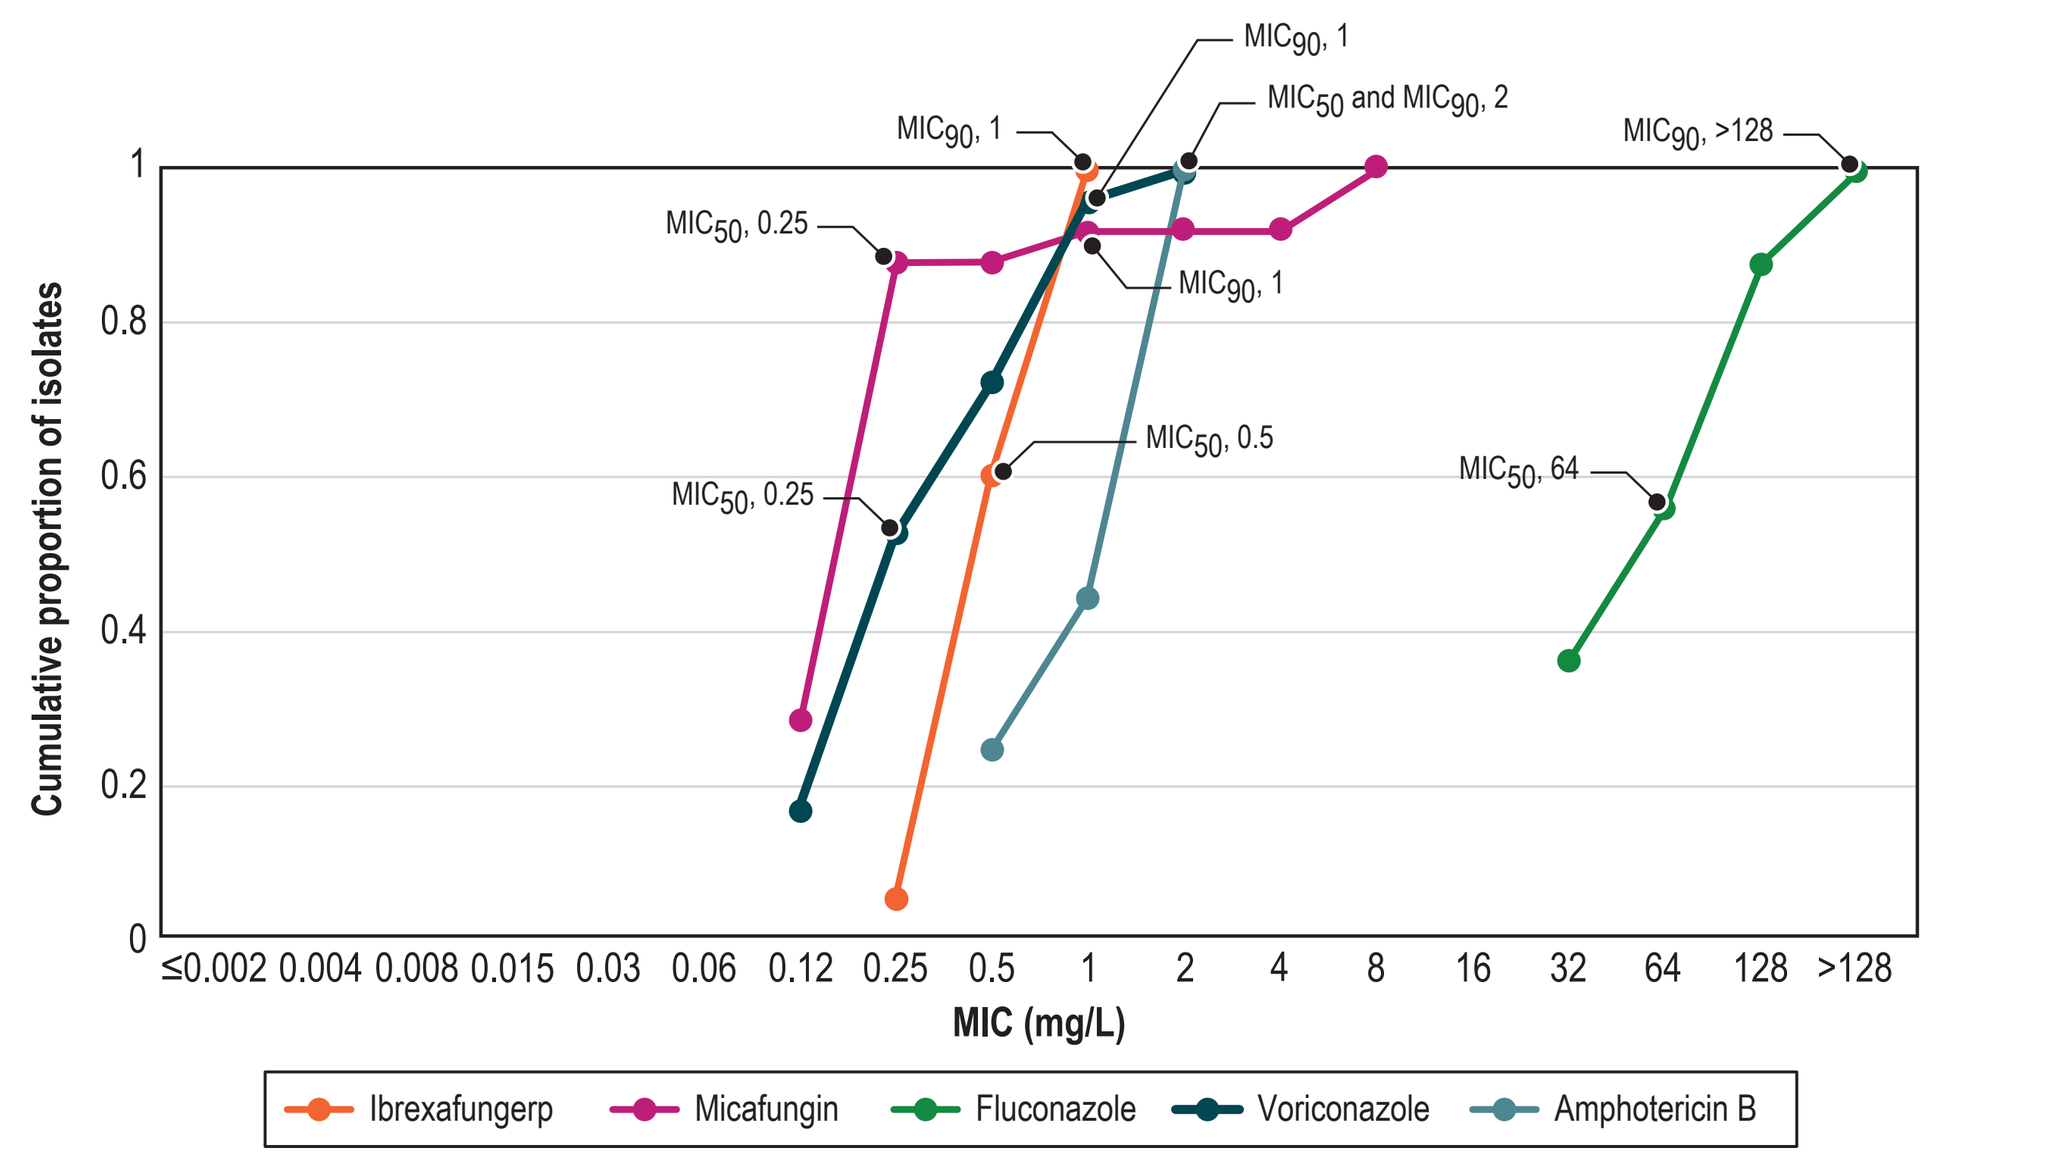
MIC90, 1
MIC50 and MIC90, 2
MIC90, 1
MIC90, >128
MIC50, 0.25
MIC90, 1
MIC50, 0.5
MIC50, 0.25
MIC50, 64
Cumulative proportion of isolates
MIC (mg/L)
Ibrexafungerp
Micafungin
Fluconazole
Voriconazole
Amphotericin B

Figure 7: *Aspergillus fumigatus* susceptibility testing (n = 141)

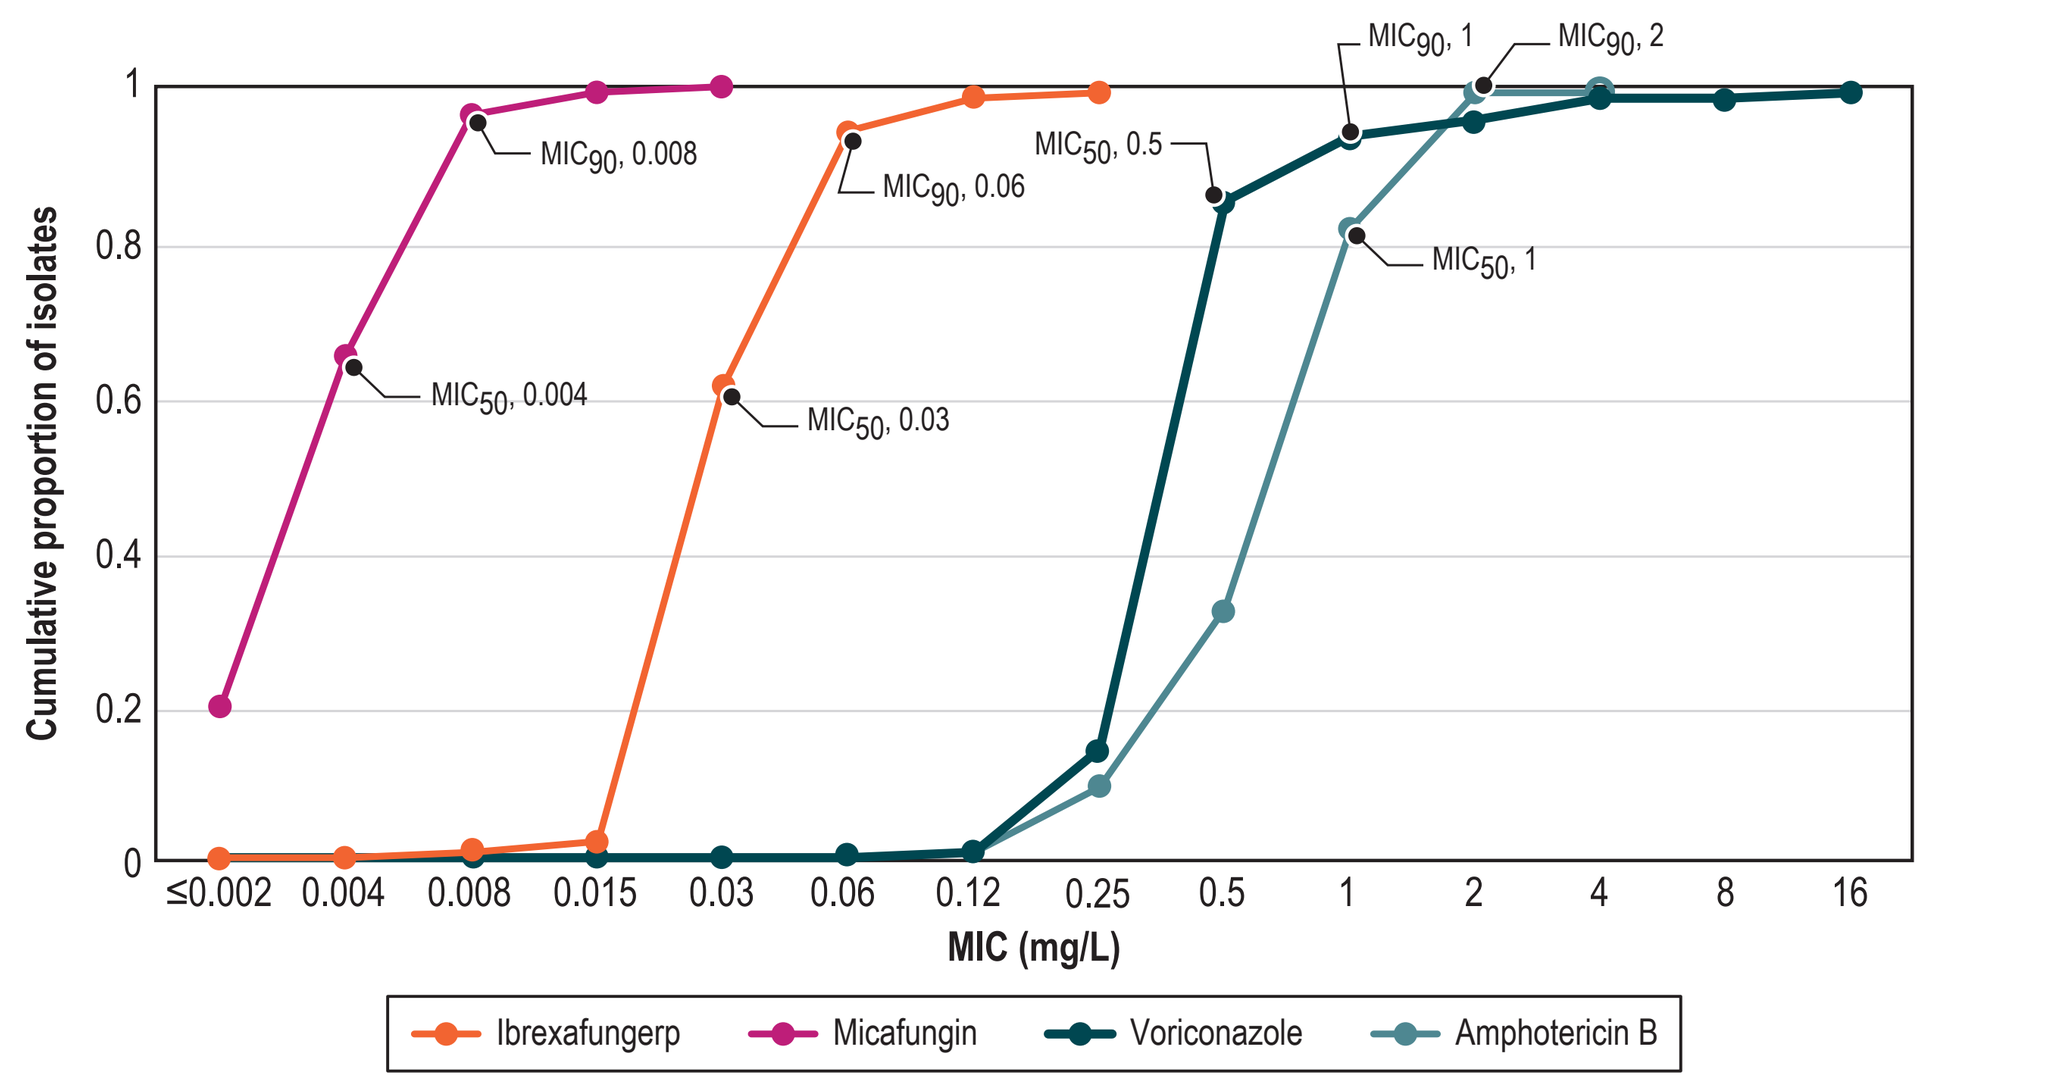
MIC90, 1
MIC90, 2
MIC90, 0.008
MIC90, 0.06
MIC50, 0.5
MIC50, 1
MIC50, 0.004
MIC50, 0.03
Cumulative proportion of isolates
MIC (mg/L)
Ibrexafungerp
Micafungin
Voriconazole
Amphotericin B

Acknowledgements

This study at Element Iowa City (JMI Laboratories) was supported by GSK (Collegeville, PA). Element Iowa City received compensation fees for services in relation to preparing the poster, which was funded by GSK.

References

CLSI. M27Ed4E. Reference method for broth dilution antifungal susceptibility testing of yeasts. Wayne, PA, Clinical and Laboratory Standards Institute, 2017.

CLSI. M27M44SEd3E. Performance Standards for Antifungal Susceptibility Testing of Yeasts. Wayne, PA, Clinical and Laboratory Standards Institute, 2022.

CLSI. MM18Ed2E. Interpretive criteria for identification of bacteria and fungi by targeted DNA sequencing. Wayne, PA, Clinical and Laboratory Standards Institute, 2018.

CLSI. M38Ed3E. Reference method for broth dilution antifungal susceptibility testing of filamentous fungi, third edition. Wayne, PA, Clinical and Laboratory Standards Institute, 2018.

CLSI. M57SEd4E. Epidemiological Cutoff Values for Antifungal Susceptibility Testing, 4th Edition. Wayne, PA, Clinical and Laboratory Standards Institute, 2022.

CLSI. M38M51SEd3E. Performance Standards for Antifungal Susceptibility Testing of Filamentous Fungi, 3rd Edition. Wayne, PA, Clinical and Laboratory Standards Institute, 2022.

Conclusions

- Ibrexafungerp is an oral non-azole agent with low MIC/MEC$_{50/90}$ values against the predominant *Candida* and *Aspergillus* species from a worldwide contemporary surveillance collection.
- MICs were low even for organisms of critical concern like *Candida auris* where the highest observed MIC for ibrexafungerp was 1 mg/L.
- Ibrexafungerp may represent an oral option for the management of difficult-to-treat invasive fungal infections from pathogens such as *C. glabrata*, *C. auris*, and *Aspergillus* spp. in addition to its current indication for vulvovaginal candidiasis.

Contact

Marisa Winkler, MD, Ph.D.
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317

Phone: (319) 665-3370
Fax: (319) 665-3371
Email: marisa.winkler@element.com

Presentation number P1096
IDWeek 2024 | October 16–19, 2024 | Los Angeles, California