IDWEEK 2025 | Poster #P1697

Performance of the VITEK 2 with Advance Expert System as a Fast Method for Determination of Antimicrobial Susceptibility Against 400 Enterobacterales Isolates from North America and Latin America (2022)

Michael D Huband, Paul R Rhomberg, Allison Fuhrmeister, Christopher Blankers, Mariana Castanheira

Element Iowa City (JMI Laboratories), North Liberty, Iowa, United States

Introduction

- The VITEK 2 with Advanced Expert System (AES) performs fast organism identifications and antimicrobial susceptibility testing.
- AES is based on an extensive, evolving database of MIC distributions and phenotypes, designed to analyze VITEK 2 results for biological validity, infer resistance mechanisms, and provide comments/corrections by labeling them as green (consistent), yellow (consistent with the correction), and red (inconsistent).
- This study evaluated the performance of VITEK 2 instrument with AES enabled and disabled versus reference broth microdilution (BMD) against a collection of 400 Enterobacterales isolates from the SENTRY Antimicrobial Surveillance Program (2022) enhanced for the presence of carbapenem-resistant, ESBL-phenotype, and transferrable AmpC (tAmpC) isolates from North and Latin America (2022).

Materials and Methods

- The 400 Enterobacterales isolates included 56.2% wildtype, 19.0% containing carbapenemases (Figure 1), 21.0% containing ESBLs (Figure 2), and 3.8% with tAmpC genes (Figure 3).
- Organism phenotypes were confirmed by next generation sequencing.
- Isolates were tested by BMD following CLSI M07 (2024) and M100 (2025) guidelines.
- BMD and VITEK 2 MIC results were compared for 30 antimicrobials with AES rules enabled and disabled. Discordant results were repeated by both methods.
- For the 19 β-lactam agents and antibacterial combinations, AES phenotypes were compared to resistant genotypes.
- AES green, yellow, and red categories were compared to reference BMD.

Results

- With the VITEK 2 AES system enabled, carbapenem-resistant, ESBL, and tAmpC phenotypes were correctly reported for 98.7%, 97.6%, and 100.0% of the carbapenemase, ESBL, and tAmpC-containing isolates tested.
- Of the 400 Enterobacterales isolates tested, 77.8% were reported by VITEK 2 with AES system as green (consistent), 15.0% as yellow (consistent with the AES correction), and 7.2% as red (inconsistent with the AES correction).
- Essential agreement (EA) and categorical agreement (CA) rates for 19 β-lactam antimicrobials against 400 Enterobacterales isolates were ≥90.0% for 16/19 and 15/19 of the β-lactam agents tested, respectively, with AES rules enabled, compared to 13/19 and 13/19 with AES rules disabled (Table 1).
- EA and CA rates for the 19 β-lactam antimicrobials were ≥85.5% and ≥81.5% with AES rules enabled compared to ≥84.3% and ≥76.0% with AES rules disabled against the 400 Enterobacterales isolates (Table 1).
- With AES rules enabled, EA and CA rates were ≥90.0% for 12/19 and 15/19 of the β-lactam agents tested against the 76 carbapenem-resistant strains, respectively, and ≥85.5% for 18/19 and 19/19, respectively (Table 2).
- Carbapenem-resistant Enterobacterales isolates consisted primarily of *Klebsiella pneumoniae* (61%), *Enterobacter cloacae* species complex (14%), and *Escherichia coli* (11%) (Figure 1).
- With AES rules enabled, EA and CA rates were ≥90.0% for 14/19 and 12/19 of the β-lactam agents tested against the 84 ESBL-phenotype strains, respectively, and ≥81.0% for 17/19 and 16/19, respectively (Table 2).
- The ESBL-phenotype strains consisted primarily of *E. coli* (42%), *K. pneumoniae* (35%), and *E. cloacae* species complex (15%) (Figure 2).
- With AES rules enabled, EA and CA rates were ≥90.0% for 12/19 and 10/19 of the β-lactam agents tested against the 15 tAmpC strains, respectively, and ≥80.0% for 16/19 and 15/19, respectively (Table 2).
- The tAmpC strains consisted primarily of *E. cloacae* species complex (67%) and *E. coli* (13%) isolates, with *K. pneumoniae* (7%), *Citrobacter koseri* (7%), and *C. freundii* species complex (6%) (Figure 3).
- Among the other antibacterial classes tested, EA and CA rates were ≥90.0% with AES rules enabled for the aminoglycosides (amikacin, gentamicin, and tobramycin), fluoroquinolones (levofloxacin and moxifloxacin), and folate pathway antagonists (trimethoprim-sulfamethoxazole) tested (Table 3).
- EA and CA rates ranged from 95.0%–99.3% and 86.8%–94.8%, respectively, for the tetracycline class agents (doxycycline, minocycline, tetracycline, and tigecycline) with AES rules enabled (Table 3).

Conclusions

- Overall, EA and CA rates with AES rules enabled for 30 antimicrobials tested against the 400 Enterobacterales isolates were ≥90.0% for 27/30 and 24/30 of the antimicrobials tested, respectively, and reduced, 24/30 and 23/30, with AES rules disabled.
- Enabling AES rules improved the percentage of β-lactam antimicrobials with EA and CA rates of ≥90.0% by 15.8% and 10.5%, respectively, when compared to results with AES rules disabled.
- The VITEK 2 automated antimicrobial susceptibility test system with AES enabled accurately reported 98.7%, 97.6%, and 100.0% of the carbapenemase, ESBL, and tAmpC isolates in this study.
- VITEK 2 system with AES provides fast organism identifications and susceptibility results while also improving EA and CA rates when compared to reference BMD and genotype results.

Funding

This study was supported by funding from bioMerieux.

MD Huband, PR Rhomberg, A Fuhrmeister, C Blankers, and M Castanheira are/were employees of Element Iowa City (JMI Laboratories) at the time of this study and were paid consultants to bioMerieux in connection with the development of this abstract/poster.

Table 1. Essential Agreement and Categorical Agreement Rates for 400 Enterobacterales Isolates Against 19 β-lactam Antimicrobials Tested by Broth Microdilution and VITEK 2 Using the Advanced Expert System

Antimicrobial Agent	AES Rules Enabled		AES Rules Disabled	
	EA (%)	CA (%)	EA (%)	CA (%)
Amoxicillin-clavulanate	92.1	90.1	91.8	85.8
Ampicillin	94.4	94.4	88.0	86.3
Ampicillin-sulbactam	92.6	81.5	84.3	76.0
Aztreonam	93.0	91.5	93.0	91.7
Cefazolin	96.5	87.5	96.5	87.3
Cefepime	85.5	90.1	85.2	90.2
Cefotaxime	93.2	97.2	93.2	97.2
Cefoxitin	91.0	88.5	91.0	86.5
Cefpodoxime	92.2	94.3	89.7	92.2
Ceftazidime	88.3	90.3	88.3	89.8
Ceftazidime-avibactam	92.6	99.8	92.6	99.8
Ceftolozane-tazobactam	88.2	93.0	88.2	93.0
Ceftriaxone	94.0	97.0	94.0	97.0
Cefuroxime	95.2	87.7	95.2	91.7
Ertapenem	95.5	97.0	95.5	97.3
Imipenem	96.8	94.1	95.5	92.2
Meropenem	94.0	95.8	94.0	95.8
Meropenem-vaborbactam	97.7	98.2	97.7	98.2
Piperacillin-tazobactam	94.4	92.8	94.5	93.0

Abbreviations: CA, categorical agreement; EA, essential agreement.
Green = EA or CA ≥90.0%, Yellow = EA or CA ≥80.0% to <90.0%, Gray = EA or CA <80.0%

Table 2. Essential Agreement and Categorical Agreement Rates for Carbapenem-resistant, ESBL-phenotype, and Transferrable AmpC Enterobacterales Isolates Against 19 β-lactam Antimicrobials Tested by Broth Microdilution and VITEK 2 Using the Advanced Expert System

Antimicrobial Agent	Organism Group (No. isolates)					
	Carbapenem-resistant Enterobacterales (*n*=76)		ESBL-phenotype Enterobacterales (*n*=84)		tAmpC Enterobacterales (*n*=15)	
	EA (%)	CA (%)	EA (%)	CA (%)	EA (%)	CA (%)
Amoxicillin-clavulanate	100.0	96.0	94.0	81.0	93.3	100.0
Ampicillin	100.0	100.0	100.0	100.0	100.0	100.0
Ampicillin-sulbactam	100.0	100.0	92.4	78.8	100.0	66.7
Aztreonam	93.4	96.1	88.1	82.1	66.7	53.3
Cefazolin	98.7	98.7	100.0	100.0	100.0	100.0
Cefepime	88.2	90.8	59.5	73.8	80.0	80.0
Cefotaxime	90.8	98.7	97.6	100.0	93.3	93.3
Cefoxitin	86.8	86.8	92.9	90.5	93.3	93.3
Cefpodoxime	97.2	98.6	100.0	100.0	100.0	100.0
Ceftazidime	82.9	89.5	70.2	71.4	73.3	100.0
Ceftazidime-avibactam	88.2	98.7	95.2	100.0	86.7	100.0
Ceftolozane-tazobactam	88.2	89.5	81.0	90.5	73.3	86.7
Ceftriaxone	98.7	100.0	94.0	100.0	86.7	86.7
Cefuroxime	100.0	100.0	98.8	98.8	93.3	86.7
Ertapenem	85.5	92.1	94.0	97.6	100.0	86.7
Imipenem	95.8	91.6	97.6	89.2	93.3	73.3
Meropenem	77.6	88.2	100.0	94.1	93.3	93.3
Meropenem-vaborbactam	90.8	92.1	97.6	98.8	100.0	100.0
Piperacillin-tazobactam	100.0	95.8	86.7	83.1	80.0	60.0

Abbreviations: CA, categorical agreement; EA, essential agreement; ESBL, extended-spectrum β-lactamase.
Green = EA or CA ≥90.0%, Yellow = EA or CA ≥80.0% to <90.0%, Gray = EA or CA <80.0%

Table 3. Essential Agreement and Categorical Agreement Rates for 400 Enterobacterales Isolates Against Other Drug Classes Tested by Broth Microdilution and VITEK 2 Using the Advanced Expert System

Antimicrobial	Antimicrobial Drug Class	Organism Group (No. isolates)	
		Enterobacterales (*n*=400)	
		EA (%)	CA (%)
Amikacin	Aminoglycoside	99.3	93.3
Doxycycline	Tetracycline	99.3	86.8
Gentamicin	Aminoglycoside	98.8	96.5
Levofloxacin	Fluoroquinolone	98.3	90.8
Minocycline	Tetracycline	99.3	91.8
Moxifloxacin	Fluoroquinolone	98.8	96.3
Nitrofurantoin	Nitrofurans	91.3	68.8
Tetracycline	Tetracycline	97.8	94.8
Tigecycline	Tetracycline	95.0	88.3
Tobramycin	Aminoglycoside	99.5	94.0
Trimethoprim-Sulfamethoxazole	Folate Pathway Antagonists	96.3	96.8

Abbreviations: CA, categorical agreement; EA, essential agreement.
Green = EA or CA ≥90.0%, Yellow = EA or CA ≥80.0% to <90.0%, Gray = EA or CA <80.0%

Figure 1. Composition of 76 Carbapenem-resistant Enterobacterales From Patients in North America and Latin America From the SENTRY Antimicrobial Surveillance Program (2022)

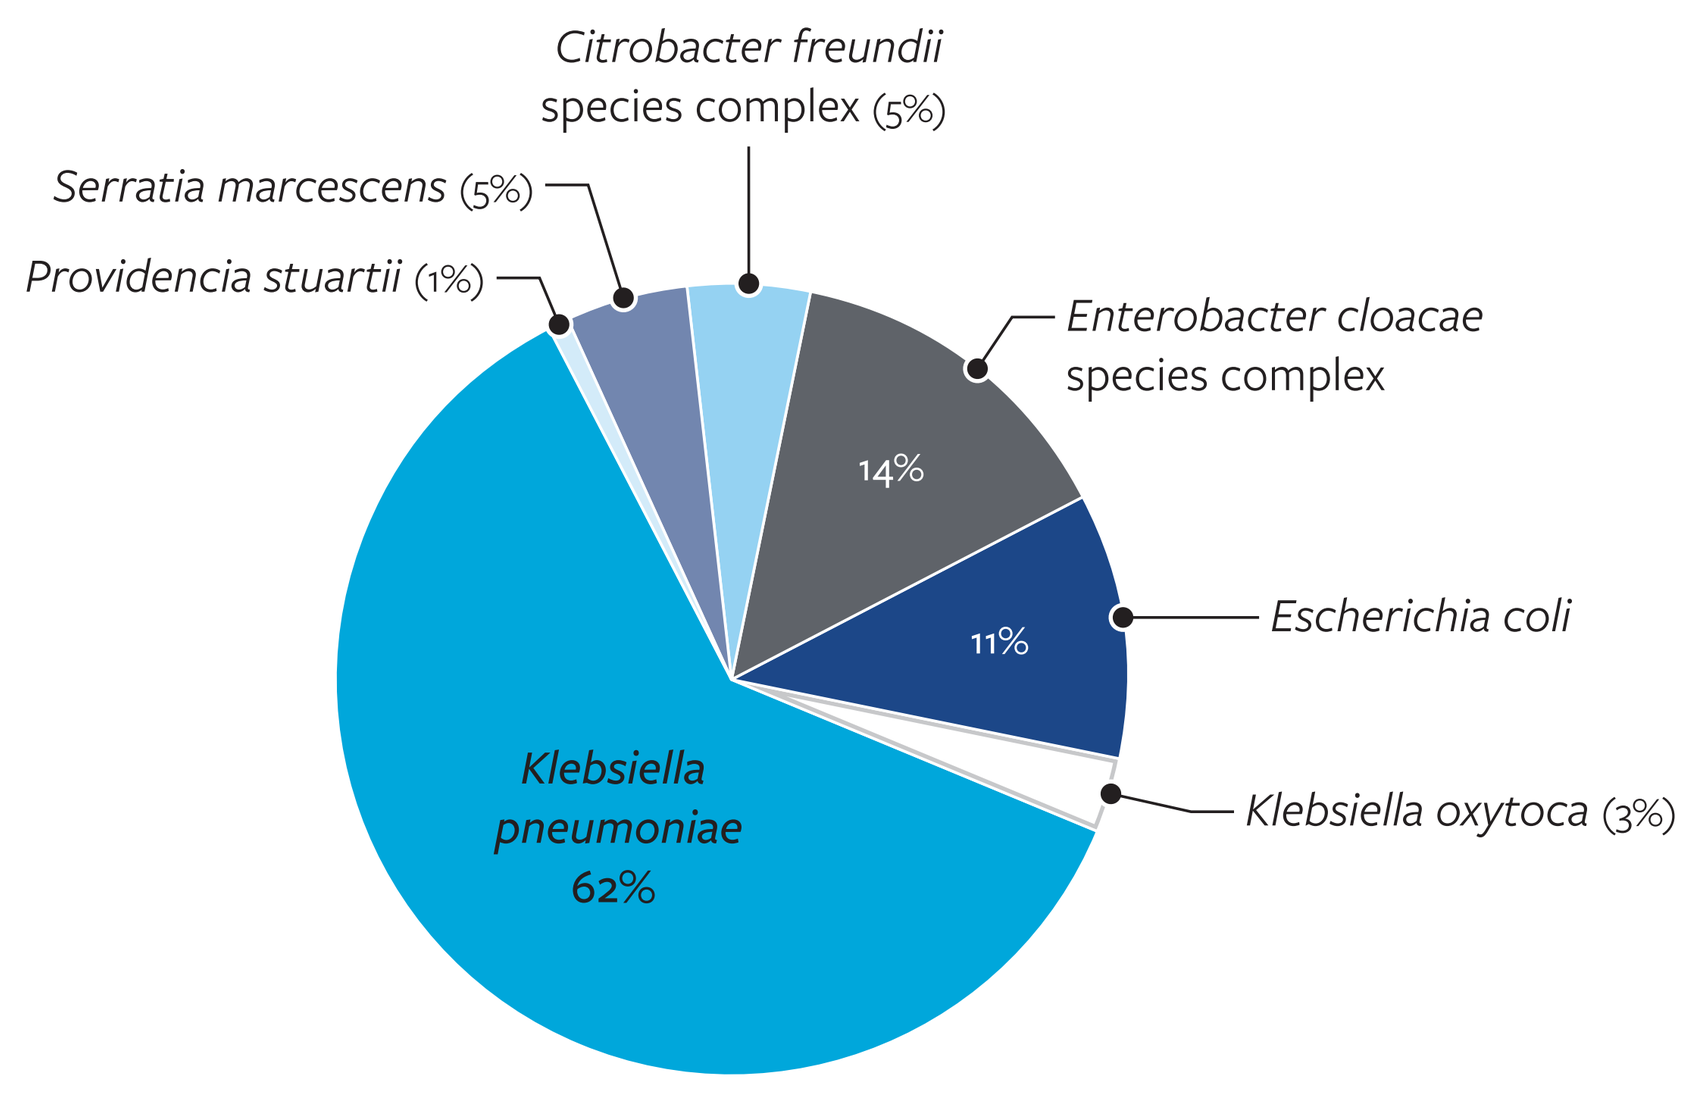

Citrobacter freundii species complex (5%)
Serratia marcescens (5%)
Providencia stuartii (1%)
Enterobacter cloacae species complex
14%
Escherichia coli
11%
Klebsiella oxytoca (3%)
Klebsiella pneumoniae
62%

Figure 2. Composition of 84 ESBL-Phenotype Enterobacterales From Patients in North America and Latin America From the SENTRY Antimicrobial Surveillance Program (2022)

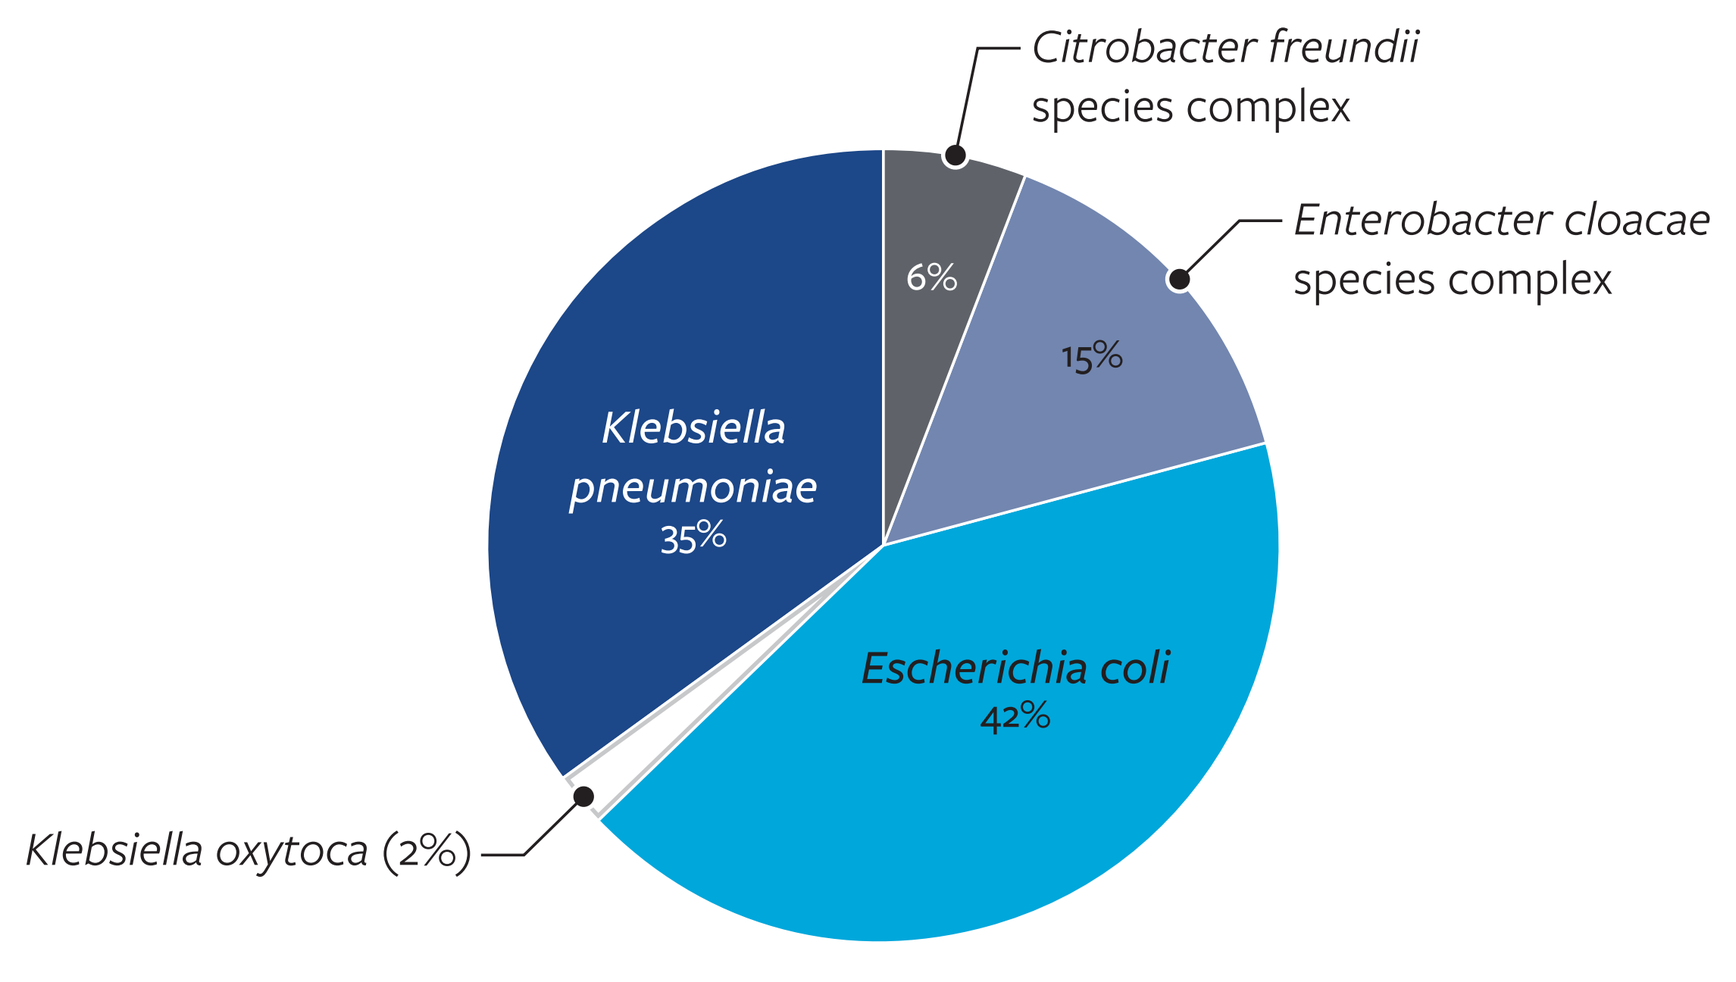

Citrobacter freundii species complex
6%
Enterobacter cloacae species complex
15%
Klebsiella pneumoniae
35%
Escherichia coli
42%
Klebsiella oxytoca (2%)

Figure 3. Composition of 15 tAmpC Enterobacterales From Patients in North America and Latin America From the SENTRY Antimicrobial Surveillance Program (2022)

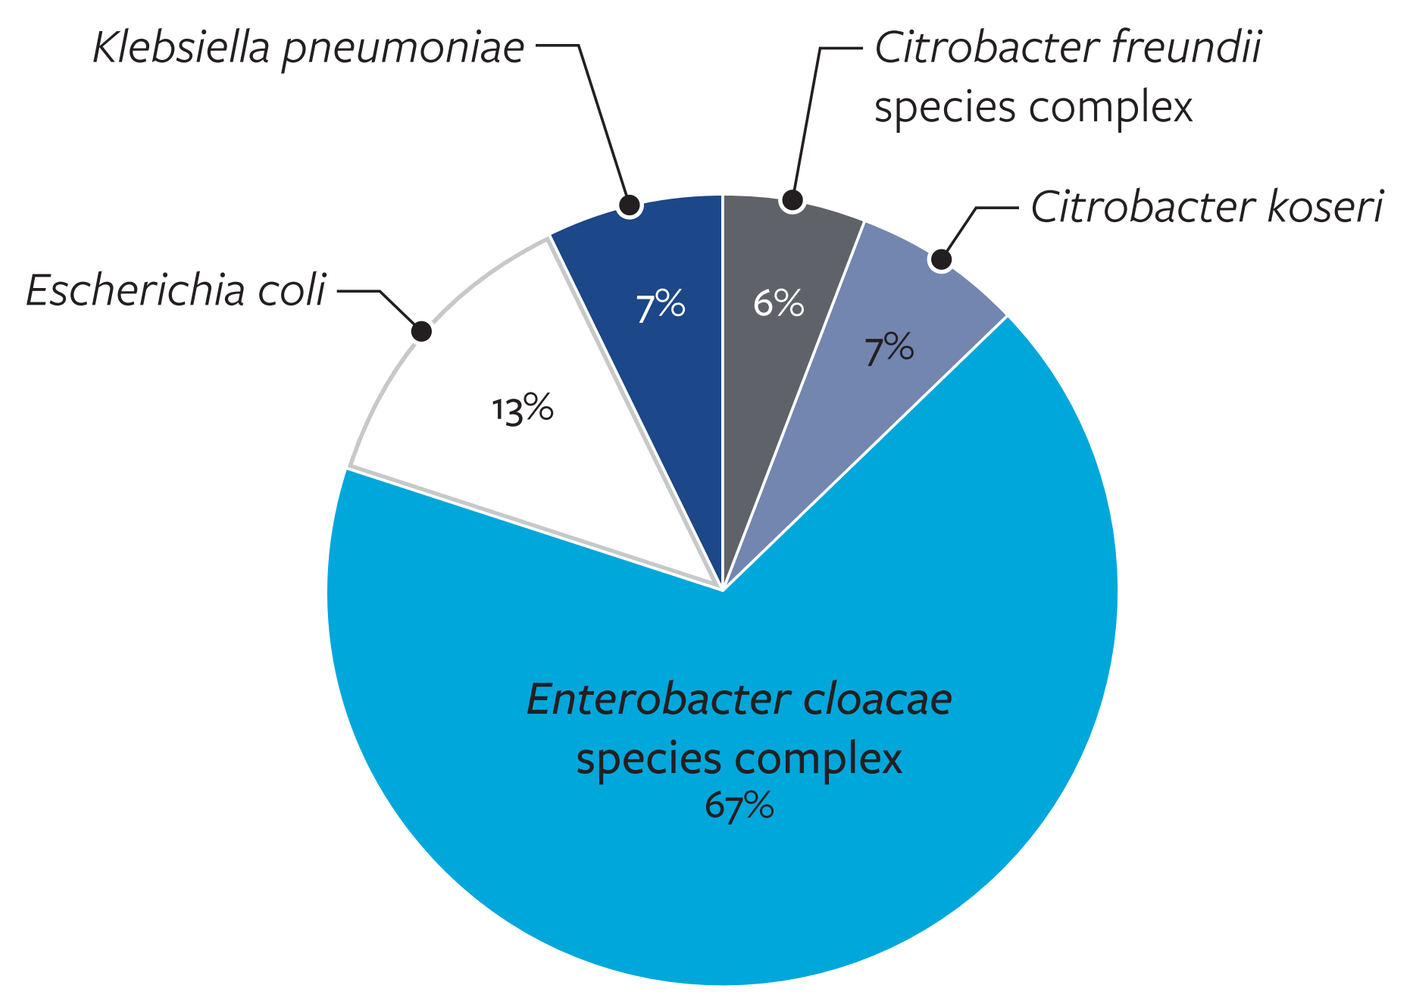

Klebsiella pneumoniae
Citrobacter freundii species complex
Citrobacter koseri
Escherichia coli
7%
6%
7%
13%
Enterobacter cloacae species complex
67%

Acknowledgments

The authors thank the participating medical centers for providing the bacterial isolates.

References

CLSI 2024. M07Ed12. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. Clinical and Laboratory Standards Institute, Wayne, PA.

CLSI. 2025. M100Ed35. Performance standards for antimicrobial susceptibility testing: 35th informational supplement. Clinical and Laboratory Standards Institute, Wayne, PA.

United States Food and Drug Administration (2025). Antibacterial susceptibility test interpretive criteria. Accessed July 2025. https://www.fda.gov/Drugs/Development ApprovalProcess/DevelopmentResources/ucm575163.htm.

Contact

element

Michael D. Huband, Director
Element Iowa City (JMI Laboratories)
335 Beaver Kreek Centre
North Liberty, IA 52317
Phone: (319) 665-3370
Email: michael.huband@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2025_22-BMX-01_P1_VITEK2_AES.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia