IDWEEK 2025 | Poster #P1194

Activity of manogepix against mould isolates from the US collected in 2023

Marisa L. Winkler, Samuel Edeker, Abby L. Klauer, Paul R. Rhomberg, Mariana Castanheira
Element Materials Technology (JMI Laboratories), North Liberty, Iowa, USA

Introduction

- Manogepix targets the fungal Gwt1 enzyme which is a different target and mechanism of action relative to current azoles, echinocandins, and amphotericin B.
- Fosmanogepix is a prodrug of manogepix undergoing phase 3 clinical trials for the treatment of invasive candidiasis and mould infections.
- There is little cross-resistance among manogepix and other antifungal agents.
- Manogepix has shown *in vitro* and *in vivo* efficacy against mould isolates with no other treatment options such as *Fusarium* spp. and *Scedosporium*/*Lomentospora* spp.
- The *in vitro* activity of manogepix and comparator antifungal agents was evaluated against mould isolates from invasive infections collected in the United States (US) in 2023.

Methods

- A total of 94 mould isolates were collected from 12 different hospitals in the United States. These represented 8 census regions and 9 different states (Figure 1).
- Only 1 isolate per patient episode was included.
- All isolates were identified by MALDI-TOF MS and/or DNA sequencing.
- Isolates were tested by CLSI reference broth microdilution method (M38) for manogepix, voriconazole, posaconazole, isavuconazole, and amphotericin B.
 - Minimum effective concentration (MEC) was read for manogepix; minimum inhibitory concentration (MIC) was read for all comparators.
- CLSI breakpoints (M38M51S) or epidemiological cutoff values (ECVs, M57S) were applied for comparator agents as available; no breakpoints or epidemiological cutoff values are available for manogepix.

Results

- 27 mould species and 13 genera were represented (Figure 2).
- Isolates were from varied infection sources (Figure 3).
 - The most common was respiratory tract infection (65%) followed by skin and skin structure infection (15%) and sinus infection (10%).
- Manogepix $MEC_{50/90}$ values against all mould isolates were 0.015/0.25 mg/L (Table 1).
 - Among *Aspergillus* spp. manogepix $MEC_{50/90}$ was 0.015/0.03 mg/L with no isolates with MICs above 0.25 mg/L; the MIC_{90} for posaconazole, voriconazole, isavuconazole, and amphotericin B was 16, 32, 32, and 64x higher, respectively (Table 1).
 - There were 5 *Aspergillus fumigatus* isolates nonsusceptible to voriconazole; manogepix MECs among these were 0.008 – 0.03 mg/L; two of these isolates had CYP51A alterations (Table 2).
 - Among 8 *Fusarium* spp., manogepix MEC range was 0.004 – 0.03 mg/L; MEC_{50} for manogepix was 250 – >1000x less than MIC_{50} for amphotericin B, voriconazole, isavuconazole, and posaconazole against these organisms (Table 1).
 - Among 8 Zygomycota, manogepix MEC range was 0.25 – 4 mg/L; MECs of 4 mg/L were seen for 1 *Lichtheimia* spp., 2 *Rhizopus oryzae*, and 1 *Rhizopus microsporus*. 1 *Rhizopus microsporus* had an MEC of 2 mg/L. In contrast, 1 *Mucor circinelloides*, 1 unspeciated *Mucor*, and 1 unspeciated *Rhizopus* had MECs of 0.25 mg/L.
 - 5 *Lomentospora prolificans* and *Scedosporium apiospermum/boydii* had a manogepix MEC range of 0.008 – 0.03 mg/L; the MEC_{50} was >256x, 64x, 512x, and >512x less than that of amphotericin B, voriconazole, posaconazole, and isavuconazole, respectively, against these organisms.

Conclusions

- Manogepix has potent *in vitro* activity among diverse mould species collected from clinical infections in the United States as part of a worldwide surveillance program.
 - This includes organisms like *Fusarium* spp. and *Lomentospora prolificans* and *Scedosporium apiospermum/boydii* which display elevated *in vitro* MICs to amphotericin B and azole comparators.
- Likely due to its unique mechanism of action, manogepix retained activity against azole-nonsusceptible *Aspergillus fumigatus* isolates with MECs ≤0.03 mg/L.
 - This included isolates with CYP51 alterations.
- Elevated MECs were seen for manogepix against *Rhizopus oryzae*, *Rhizopus microsporus*, and *Lichtheimia* spp.
 - Due to the small number of isolates tested (5 in total), conclusions are difficult to draw about overall activity against these organisms.
- Manogepix is a promising novel antifungal agent for the treatment of infections due to moulds including organisms with limited other options for treatment.

Funding

This study was supported by Basilea and this project has been funded in part with federal funds from the U.S. Department of Health and Human Services (HHS); Administration of Strategic Preparedness and Response (ASPR); and Biomedical Advanced Research and Development Authority (BARDA), under OT number: 75A50124C00033. The contract and federal funding are not an endorsement of the study results, product, or company.

ML Winkler, S Edeker, AL Klauer, PR Rhomberg, and M Castanheira were employees of Element Materials Technology (JMI Laboratories) at the time of this study, which was a paid consultant to Basilea in connection with the development of this poster.

Acknowledgments

The authors thank all of the SENTRY Program participants for providing the isolates used in this study.

References

CLSI, *M38M51S Ed3. Performance standards for antifungal susceptibility testing of molds*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M57S Ed4. Epidemiological cutoff values for antifungal susceptibility testing*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M38 Ed3. Reference method for broth dilution antifungal susceptibility testing of filamentous fungi*. 2017, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *FR01. Voriconazole Breakpoints for Aspergillus fumigatus*. 2024, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *FR02. Isavuconazole Breakpoints for Aspergillus fumigatus*. 2025, Clinical and Laboratory Standards Institute: Wayne, PA.

Figure 1. Map of US census regions and states providing mould isolates (starred)

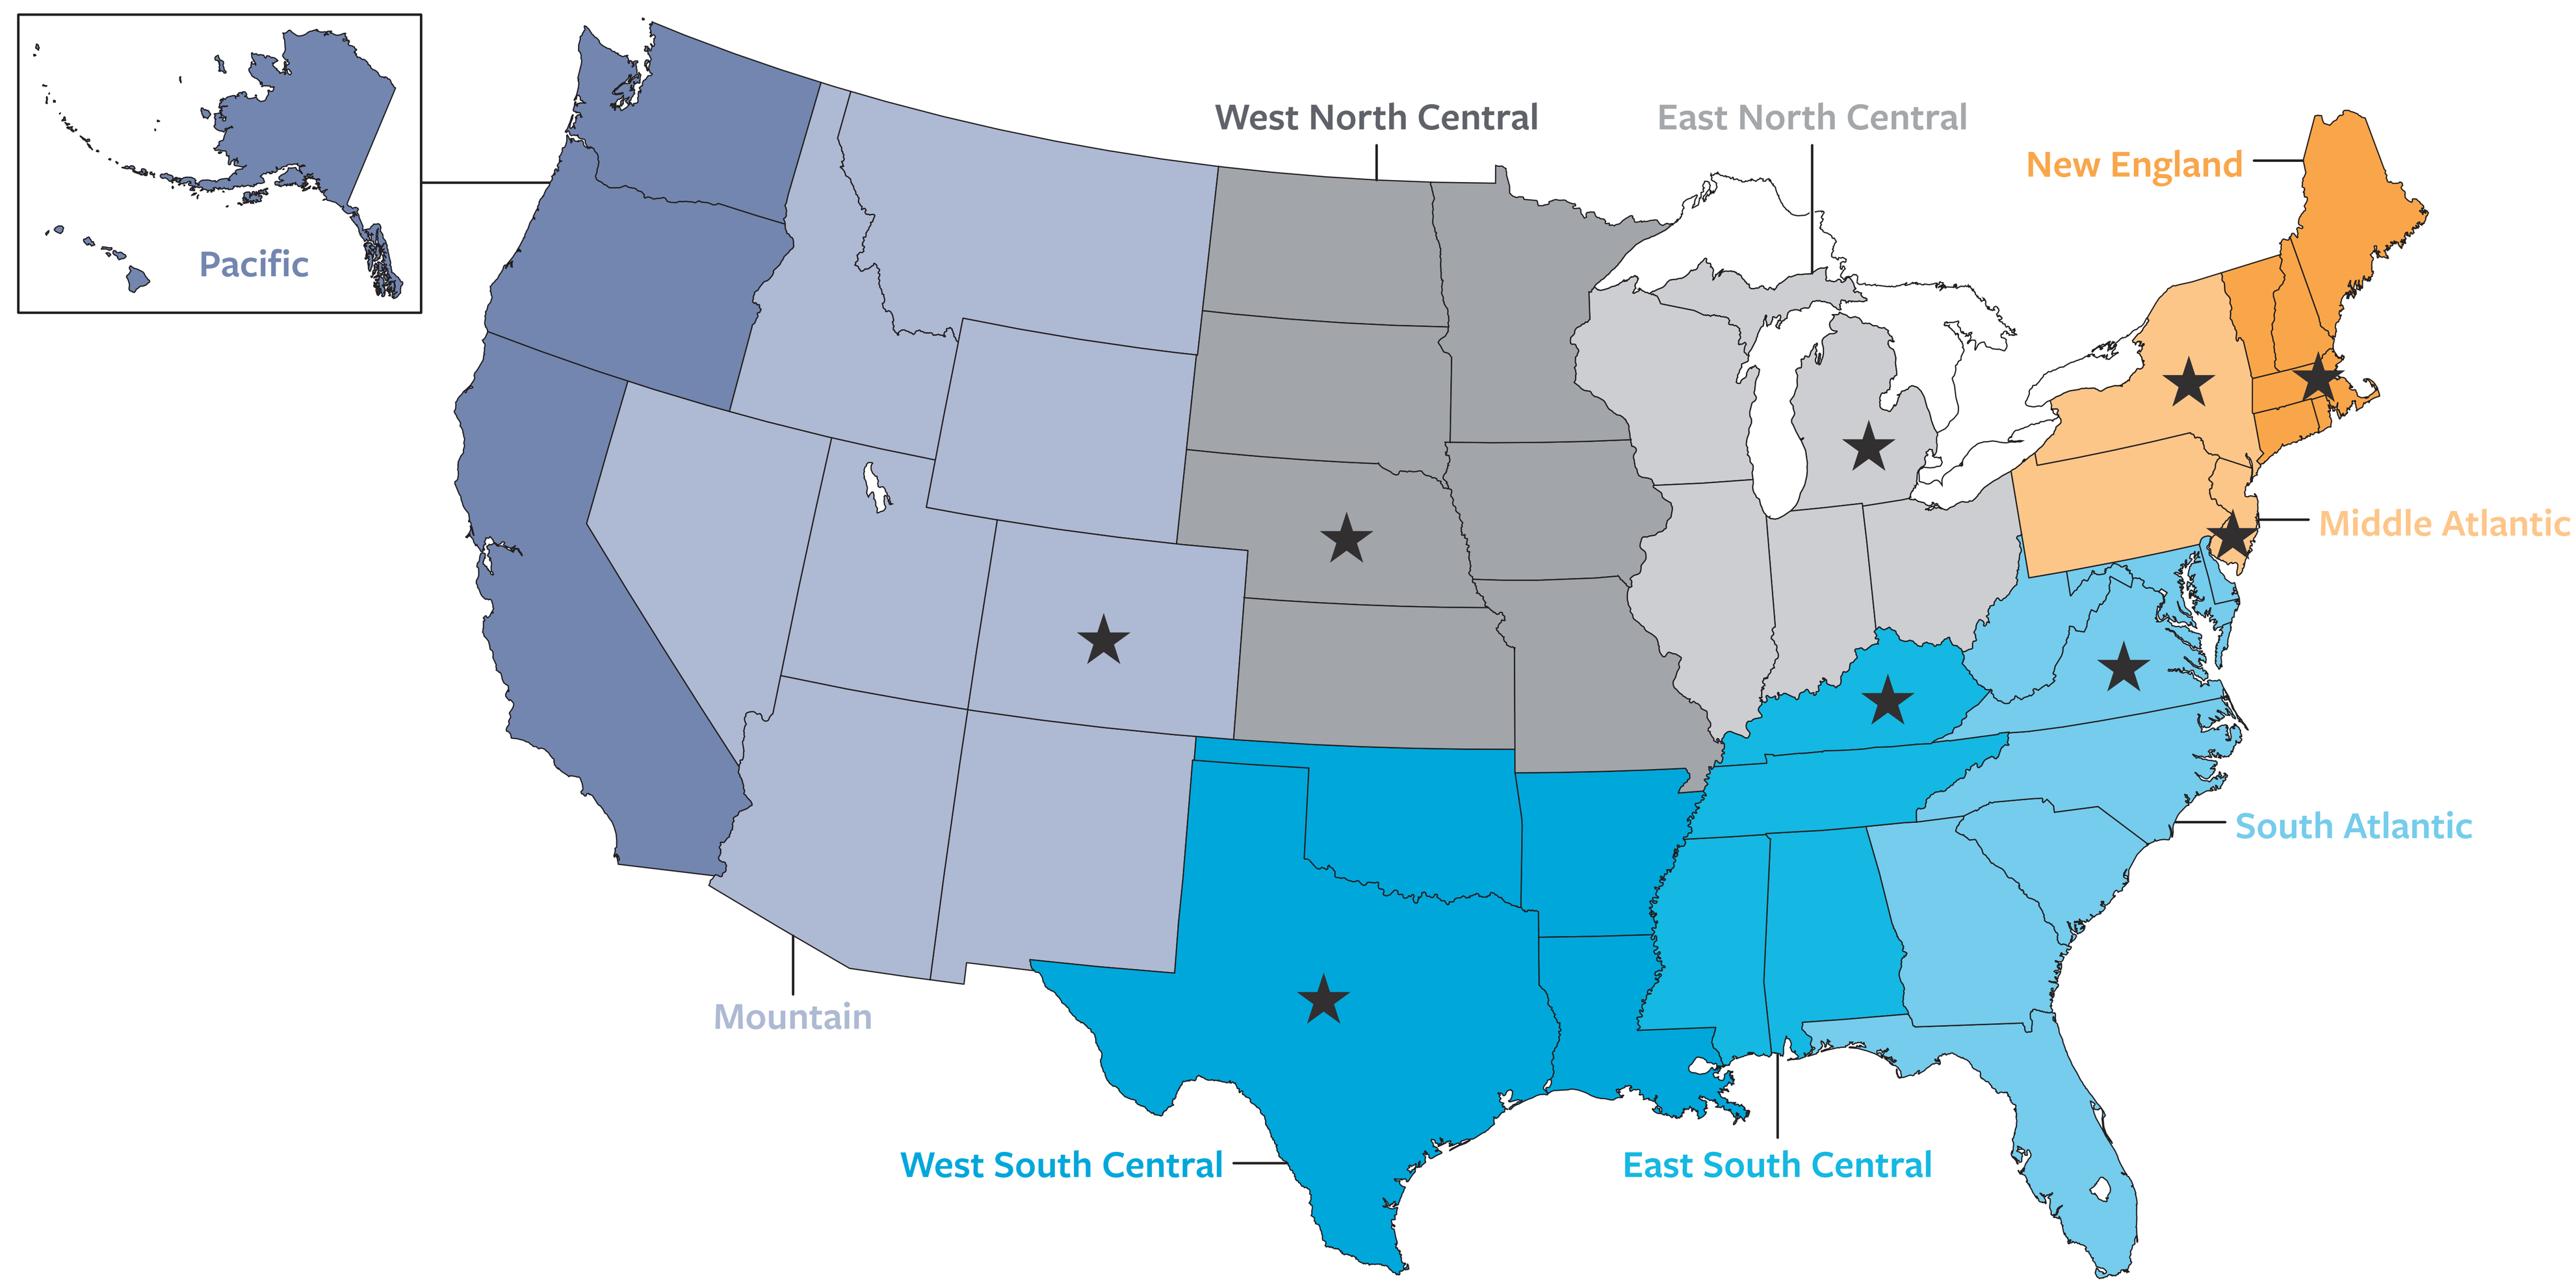
Pacific
West North Central
East North Central
New England
Middle Atlantic
South Atlantic
Mountain
West South Central
East South Central

Figure 2. Mould species tested as part of the US SENTRY surveillance program

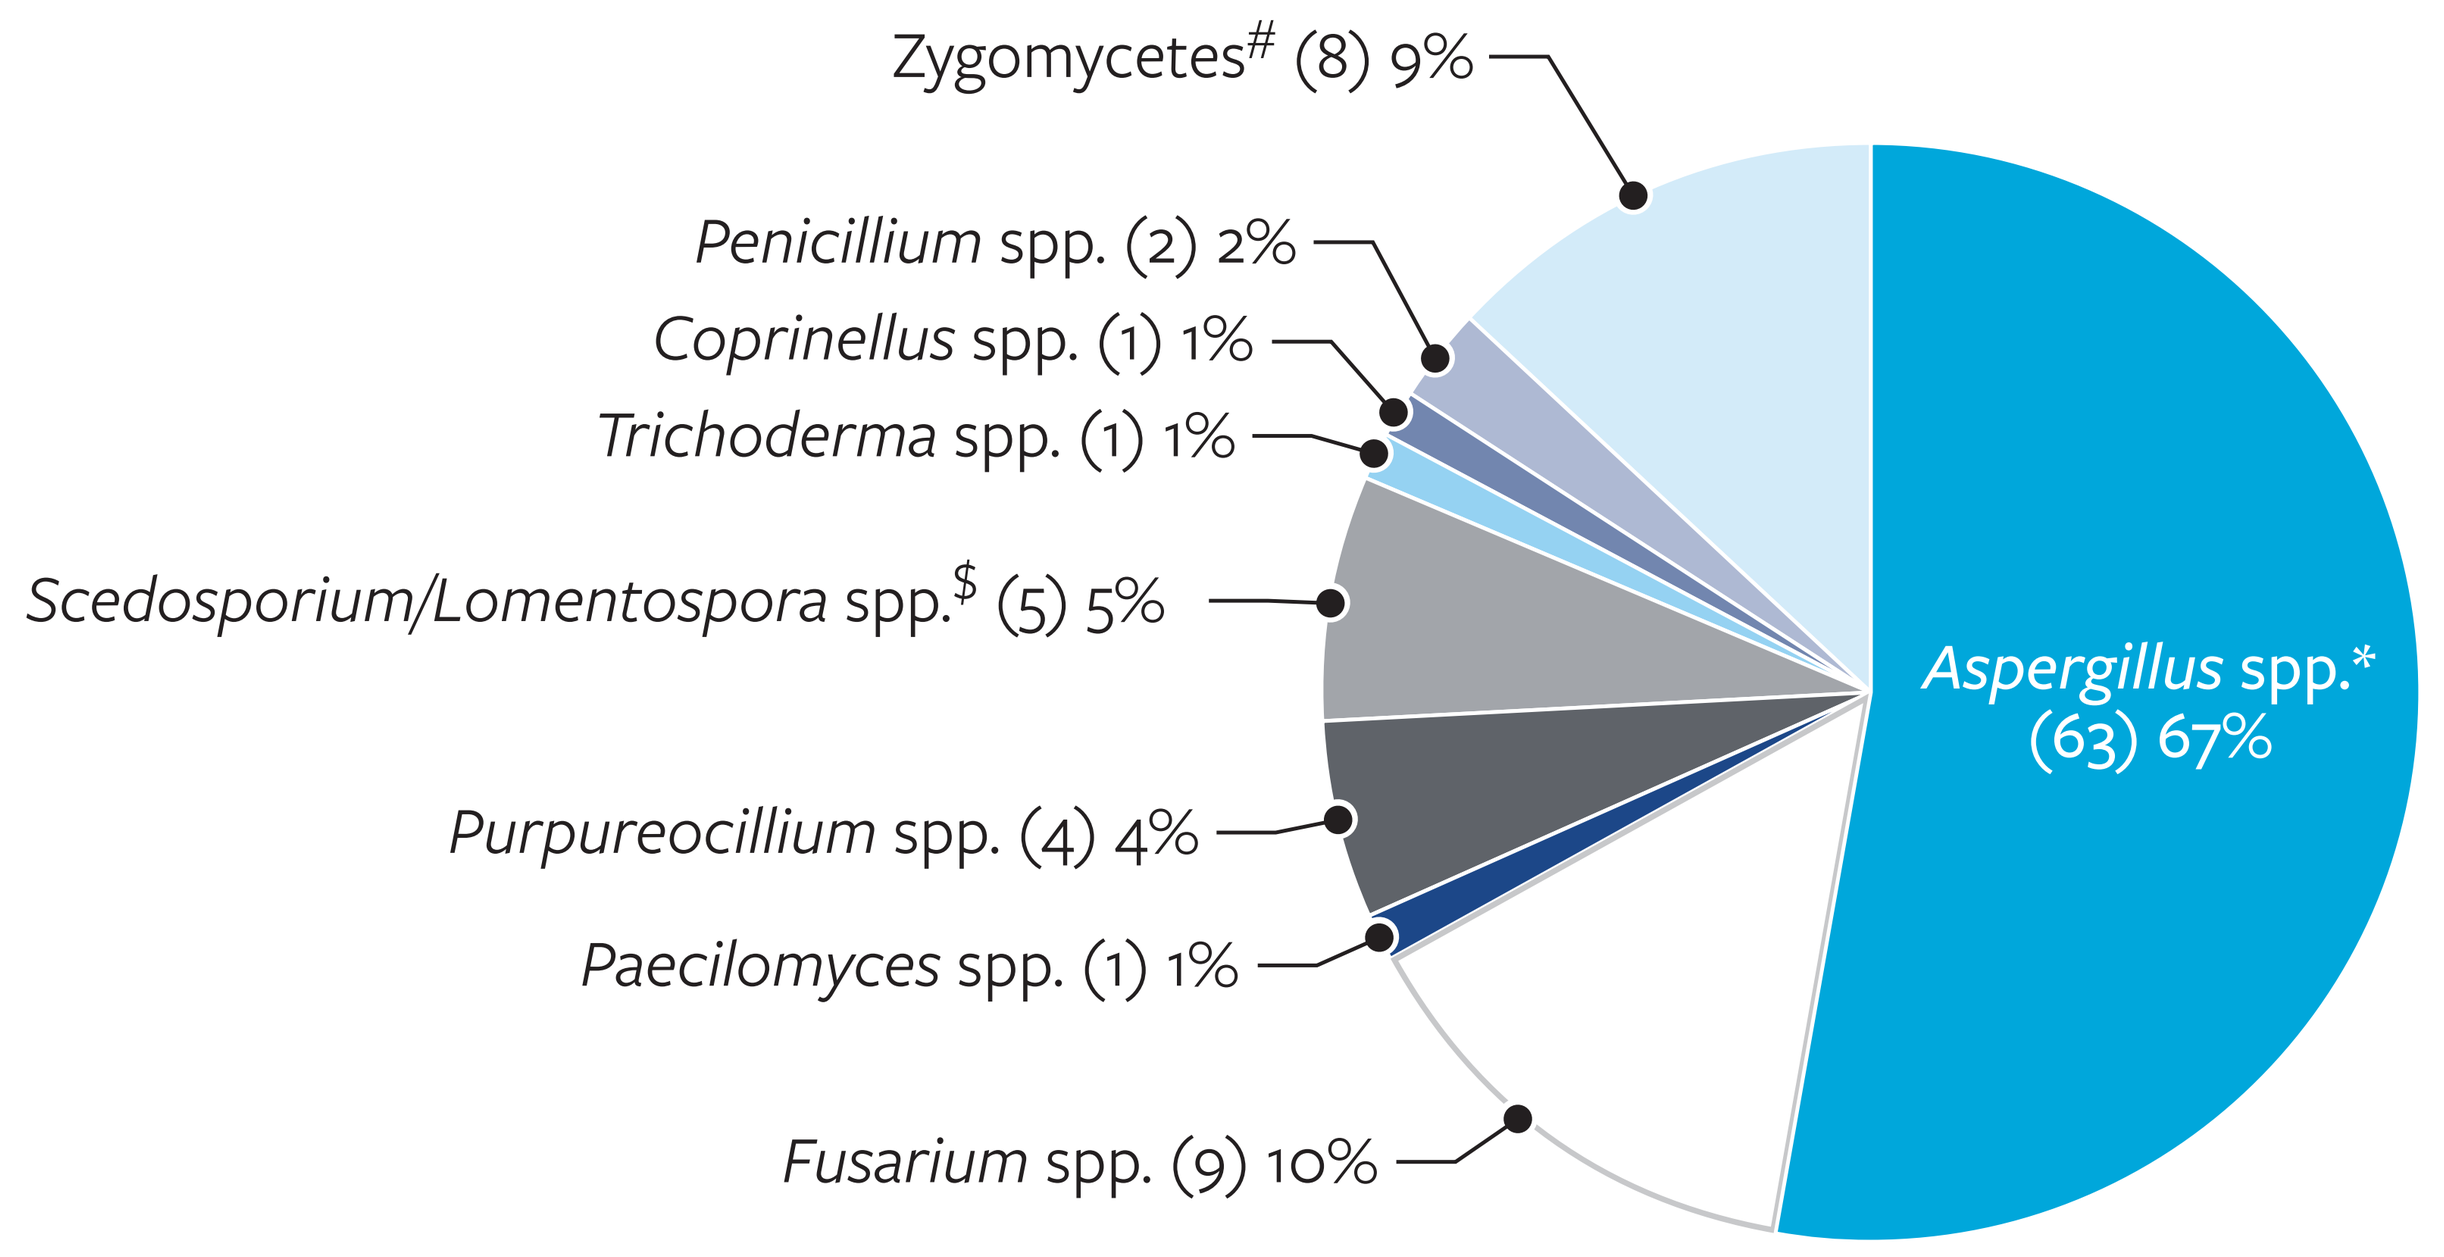
Zygomycetes# (8) 9%
Penicillium spp. (2) 2%
Coprinellus spp. (1) 1%
Trichoderma spp. (1) 1%
Scedosporium/Lomentospora spp.$ (5) 5%
Purpureocillium spp. (4) 4%
Paecilomyces spp. (1) 1%
Fusarium spp. (9) 10%
Aspergillus spp.* (63) 67%

*Includes 1 *Aspergillus caelatus*, 4 *Aspergillus flavus* species complex, 44 *Aspergillus fumigatus*, 9 *Aspergillus niger* species complex, 1 *Aspergillus sydowii*, 3 *Aspergillus terreus* species complex, 1 *Aspergillus versicolor*
Includes 1 *Lichtheimia* spp., 1 *Mucor circinelloides*, 1 *Mucor* spp., 2 *Rhizopus microsporus*, 2 *Rhizopus oryzae*, 1 *Rhizopus* spp.
$ Includes 1 *Lomentospora prolificans* and 4 *Scedosporium apiospermum/boydii*

Figure 3. Infectious source of collected mould isolates (GI, gastrointestinal; CSF, cerebral spinal fluid; SSSI, skin and skin structure infection)

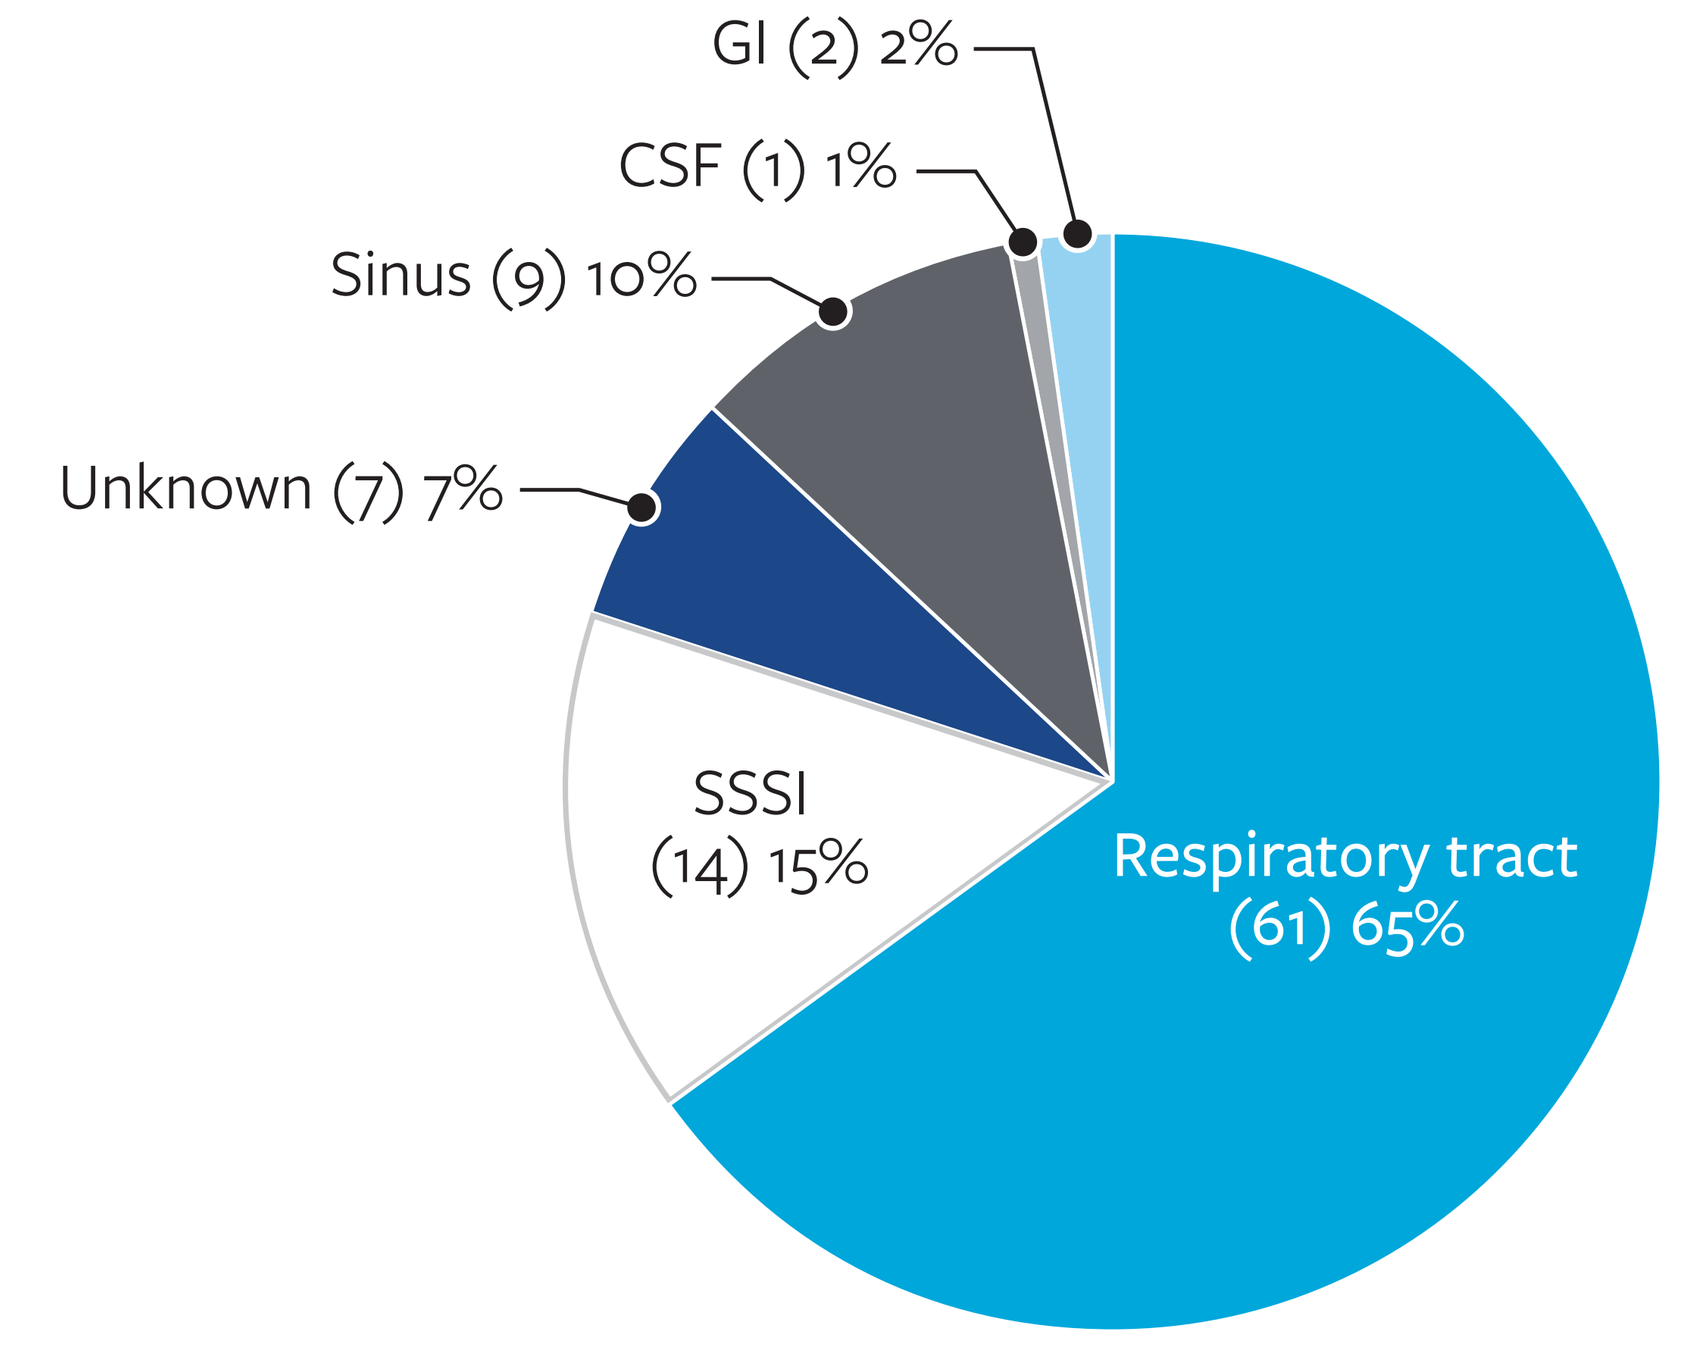
GI (2) 2%
CSF (1) 1%
Sinus (9) 10%
Unknown (7) 7%
SSSI (14) 15%
Respiratory tract (61) 65%

Table 1. MIC/MEC range (mg/L), $MIC/MEC_{50/90}$ (mg/L), and % organisms susceptible for manogepix and comparator agents against tested mould isolates

Organism name (n)	manogepix		voriconazole		posaconazole		isavuconazole		amphotericin B	
	MEC range (mg/L)	$MEC_{50/90}$ (mg/L)	MIC range (mg/L, %S/WT)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L, %S/WT)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L, %S/WT)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L, %S/WT)	$MIC_{50/90}$ (mg/L)
All organisms (94)	0.004 – 4	0.015/0.25	0.03 – >8	0.5/>8	0.008 – >8	0.25/8	0.008 – >8	0.5/>8	≤0.03 – >4	1/2
All *Aspergillus* spp. (63)	0.004 – 0.25	0.015/0.03	0.12 – 2	0.5/1	0.03 – 1	0.25/0.5	0.12 – 2	0.5/1	0.12 – 4	1/2
Aspergillus fumigatus (44)	0.008 – 0.12	0.015/0.03	0.12 – 2 (88.6)[1]	0.03/0.06	0.03 – 1	0.25/0.5	0.12 – 2 (93.2)[2]	0.5/1	0.25 – 2 (100)[3]	1/2
A. section *Nigri* (9)	0.004 – 0.015	0.004/*	0.5 – 2 (100)[3]	1/*	0.25 – 0.5 (100)[3]	0.25/*	0.5 – 2 (100)[3]	1/*	0.12 – 0.25 (100)[3]	0.25/*
Fusarium spp. (8)	0.004 – 0.03	0.008/*	2 – >8	4/*	0.5 – >8	>8/*	1 – >8	4/*	0.5 – 2	2/*
Zygomycota (8)	0.25 – 4	2/*	2 – >8	>8/*	0.12 – 1	0.5/*	0.25 – 8	1/*	0.06 – 0.5	0.12/*
Lomentospora/Scedosporium (5)	0.008 – 0.03	0.015/*	0.5 – >8	1/*	1 – >8	8/*	2 – >8	>8/*	2 – >4	>4/*

[1] Based on breakpoint in M38M51S
[2] Based on breakpoints approved by CLSI but not yet in M38M51S
[3] Based on ECV in CLSI M57S
* No MEC/MIC_{90} able to be calculated in the setting of <10 isolates tested

Table 2. MEC/MICs for 3 voriconazole-nonsusceptible *Aspergillus fumigatus* with CYP51 analysis

Organism	manogepix	isavuconazole	voriconazole	CYP51A	CYP51B
	MEC (mg/L)	MIC (mg/L)	MIC (mg/L)		
Aspergillus fumigatus	0.008	2	1	Wildtype	Wildtype
Aspergillus fumigatus	0.03	2	2	F46Y,M172V,N248T,D255E,E427K	Wildtype
Aspergillus fumigatus	0.03	2	1	F46Y,M172V,N248T,D255E,E427K	Wildtype

Contact

Marisa Winkler, MD, PhD
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: marisa.winkler@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2025_23-BAS-12_P3_US_Mould.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia