IDWEEK 2025 | Poster #P1195

Activity of manogepix against surveillance yeast isolates collected from the United States in 2023

Marisa L. Winkler, Samuel Edeker, Abby L. Klauer, Paul R. Rhomberg, Mariana Castanheira
Element Materials Technology (JMI Laboratories), North Liberty, Iowa, USA

Introduction

- Manogepix targets the fungal Gwt1 enzyme which is a different target and mechanism of action relative to current azoles, echinocandins, and amphotericin B.
- Fosmanogepix is a prodrug of manogepix undergoing phase 3 clinical trials for the treatment of invasive candidiasis and mould infections.
- There is little cross-resistance among manogepix and other antifungal agents.
- The *in vitro* activity of manogepix and comparator antifungal agents was evaluated against yeast isolates from invasive infections collected in the United States in 2023.

Methods

- A total of 298 yeast isolates were collected from 24 different hospitals in the United States (US). These represented all 9 census regions and 18 different states (Figure 1).
- Only 1 isolate per patient episode was included.
- All isolates were identified by MALDI-TOF MS and/or DNA sequencing.
- Isolates were tested by CLSI reference broth microdilution method (M27) for anidulafungin, fluconazole, amphotericin B, and manogepix.
- CLSI breakpoints (M27M44S) were applied for comparator agents; no breakpoints or epidemiological cutoff values are available for manogepix. Resistant-only CDC breakpoints were used for *C. auris*.
- Isolates that were non-wildtype to echinocandins by CLSI M27S criteria were analyzed for alterations in FKS1 and FKS2.

Results

- 19 different species of yeast were represented (Figure 2) with *Candida glabrata* most common (81/298, 27.2%), followed by *C. albicans* (66/298, 22.1%), *C. parapsilosis* (59/298, 19.8%), and *C. tropicalis* (25/298, 8.4%). There were 12 *C. auris* (4.0%).
- Isolates were from many different infection sources (Figure 3).
 - The most common was bloodstream infection (53.7%), followed by skin and skin structure infection (11.4%) and respiratory tract infection (9.7%).
- Manogepix $MIC_{50/90}$ values against *Candida* spp. were 0.015/0.06 mg/L (Table 1).
- There were 28 *Candida* isolates nonsusceptible (NS) to fluconazole by CLSI breakpoints; MICs for manogepix among these isolates ranged from 0.008 – 0.06 mg/L (Table 2).
 - This included 12 *C. auris* (manogepix MICs range, 0.008 – 0.06 mg/L).
 - It also included 7 *C. glabrata*, 2 *C. albicans*, 6 *C. parapsilosis*, and 1 *C. tropicalis*.
- There were 11 *Candida* isolates NS to anidulafungin (3 *C. glabrata*, 1 *C. albicans*, 2 *C. auris*, and 5 *C. parapsilosis*) (Table 2). 7 isolates had alterations in FKS (Table 3).
 - Manogepix MICs in these organisms were 0.004 – 0.06 mg/L.
- Elevated manogepix MICs were seen among *C. krusei* (MIC_{50} > 4 mg/L) and 2 *Cryptococcus neoformans* (MICs 0.5 mg/L, data not shown).

Conclusions

- Manogepix has potent *in vitro* activity among diverse yeast species collected from clinical infections in the United States as part of a worldwide surveillance program.
 - This includes organisms of critical concern like *C. auris* and organisms with alterations in FKS genes.
- Likely due to its unique mechanism of action, manogepix retained activity against fluconazole-NS and anidulafungin-NS isolates with MICs ≤0.06 mg/L.
 - Activity was also retained against the 4 *C. auris* isolates that were NS to amphotericin B with MICs of 0.015 – 0.06 mg/L (Table 2).
- Elevated MICs were seen for manogepix against *C. krusei* and *Cryptococcus neoformans*.
- Manogepix is a promising novel antifungal agent for the treatment of infections due to yeast.

Funding

This study was supported by Basilea and this project has been funded in part with federal funds from the U.S. Department of Health and Human Services (HHS); Administration of Strategic Preparedness and Response (ASPR); and Biomedical Advanced Research and Development Authority (BARDA), under OT number: 75A50124C00033. The contract and federal funding are not an endorsement of the study results, product, or company.

ML Winkler, S Edeker, AL Klauer, PR Rhomberg, and M Castanheira were employees of Element Materials Technology (JMI Laboratories) at the time of this study, which was a paid consultant to Basilea in connection with the development of this poster.

Acknowledgments

The authors thank all of the SENTRY Program participants for providing the isolates used in this study.

References

Pappas, P.G., Kauffman, C.A., Andes, D.R., Clancy, C.J., Marr, K.A., Ostrosky-Zeichner, L., et al., *Clinical Practice Guideline for the Management of Candidiasis: 2016 Update by the Infectious Diseases Society of America.* Clin Infect Dis, 2016. 62(4): e1-50.

Centers for Disease Control. *Antifungal susceptibility testing for C. auris*. April 23, 2024. https://www.cdc.gov/candida-auris/hcp/laboratories/antifungal-susceptibility-testing.html.

CLSI, *M27M44S Ed3. Performance standards for antifungal susceptibility testing of yeasts*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M57S Ed4. Epidemiological cutoff values for antifungal susceptibility testing*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M27 Ed4. Reference method for broth dilution antifungal susceptibility testing of yeasts*. 2017, Clinical and Laboratory Standards Institute: Wayne, PA.

Figure 1. Map of US census regions and states providing yeast isolates (starred)

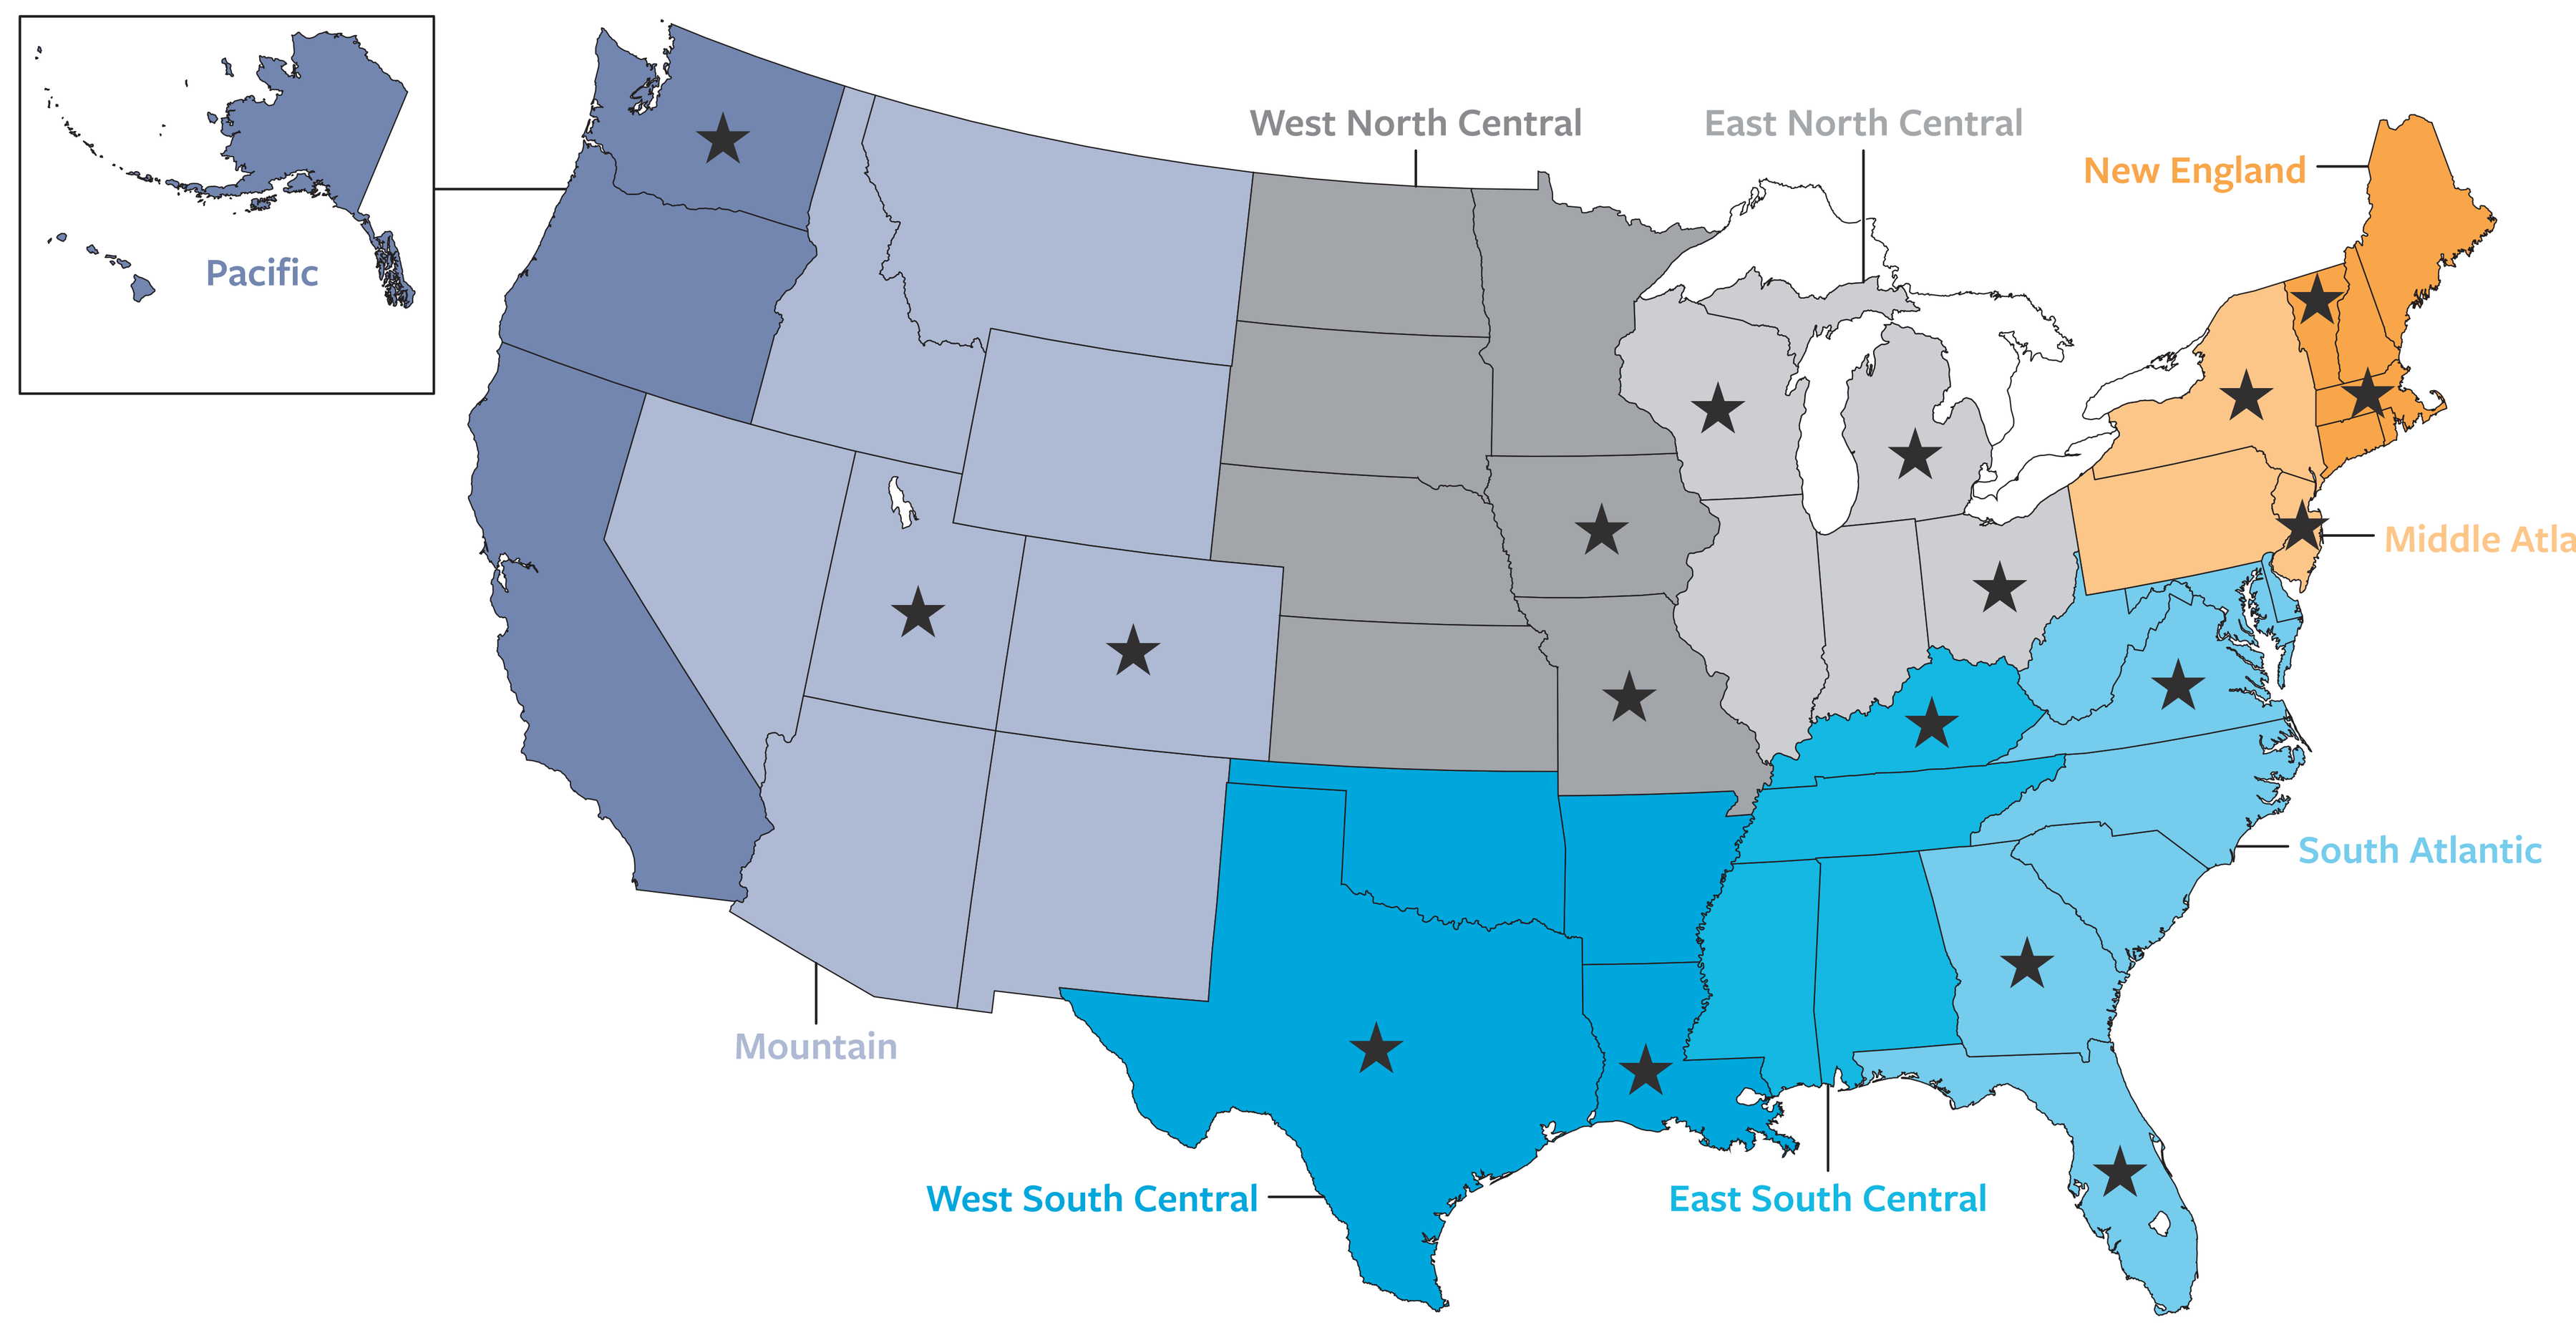
Pacific
West North Central
East North Central
New England
Middle Atlantic
South Atlantic
Mountain
West South Central
East South Central

Figure 2. Yeast species tested as part of the US SENTRY surveillance program

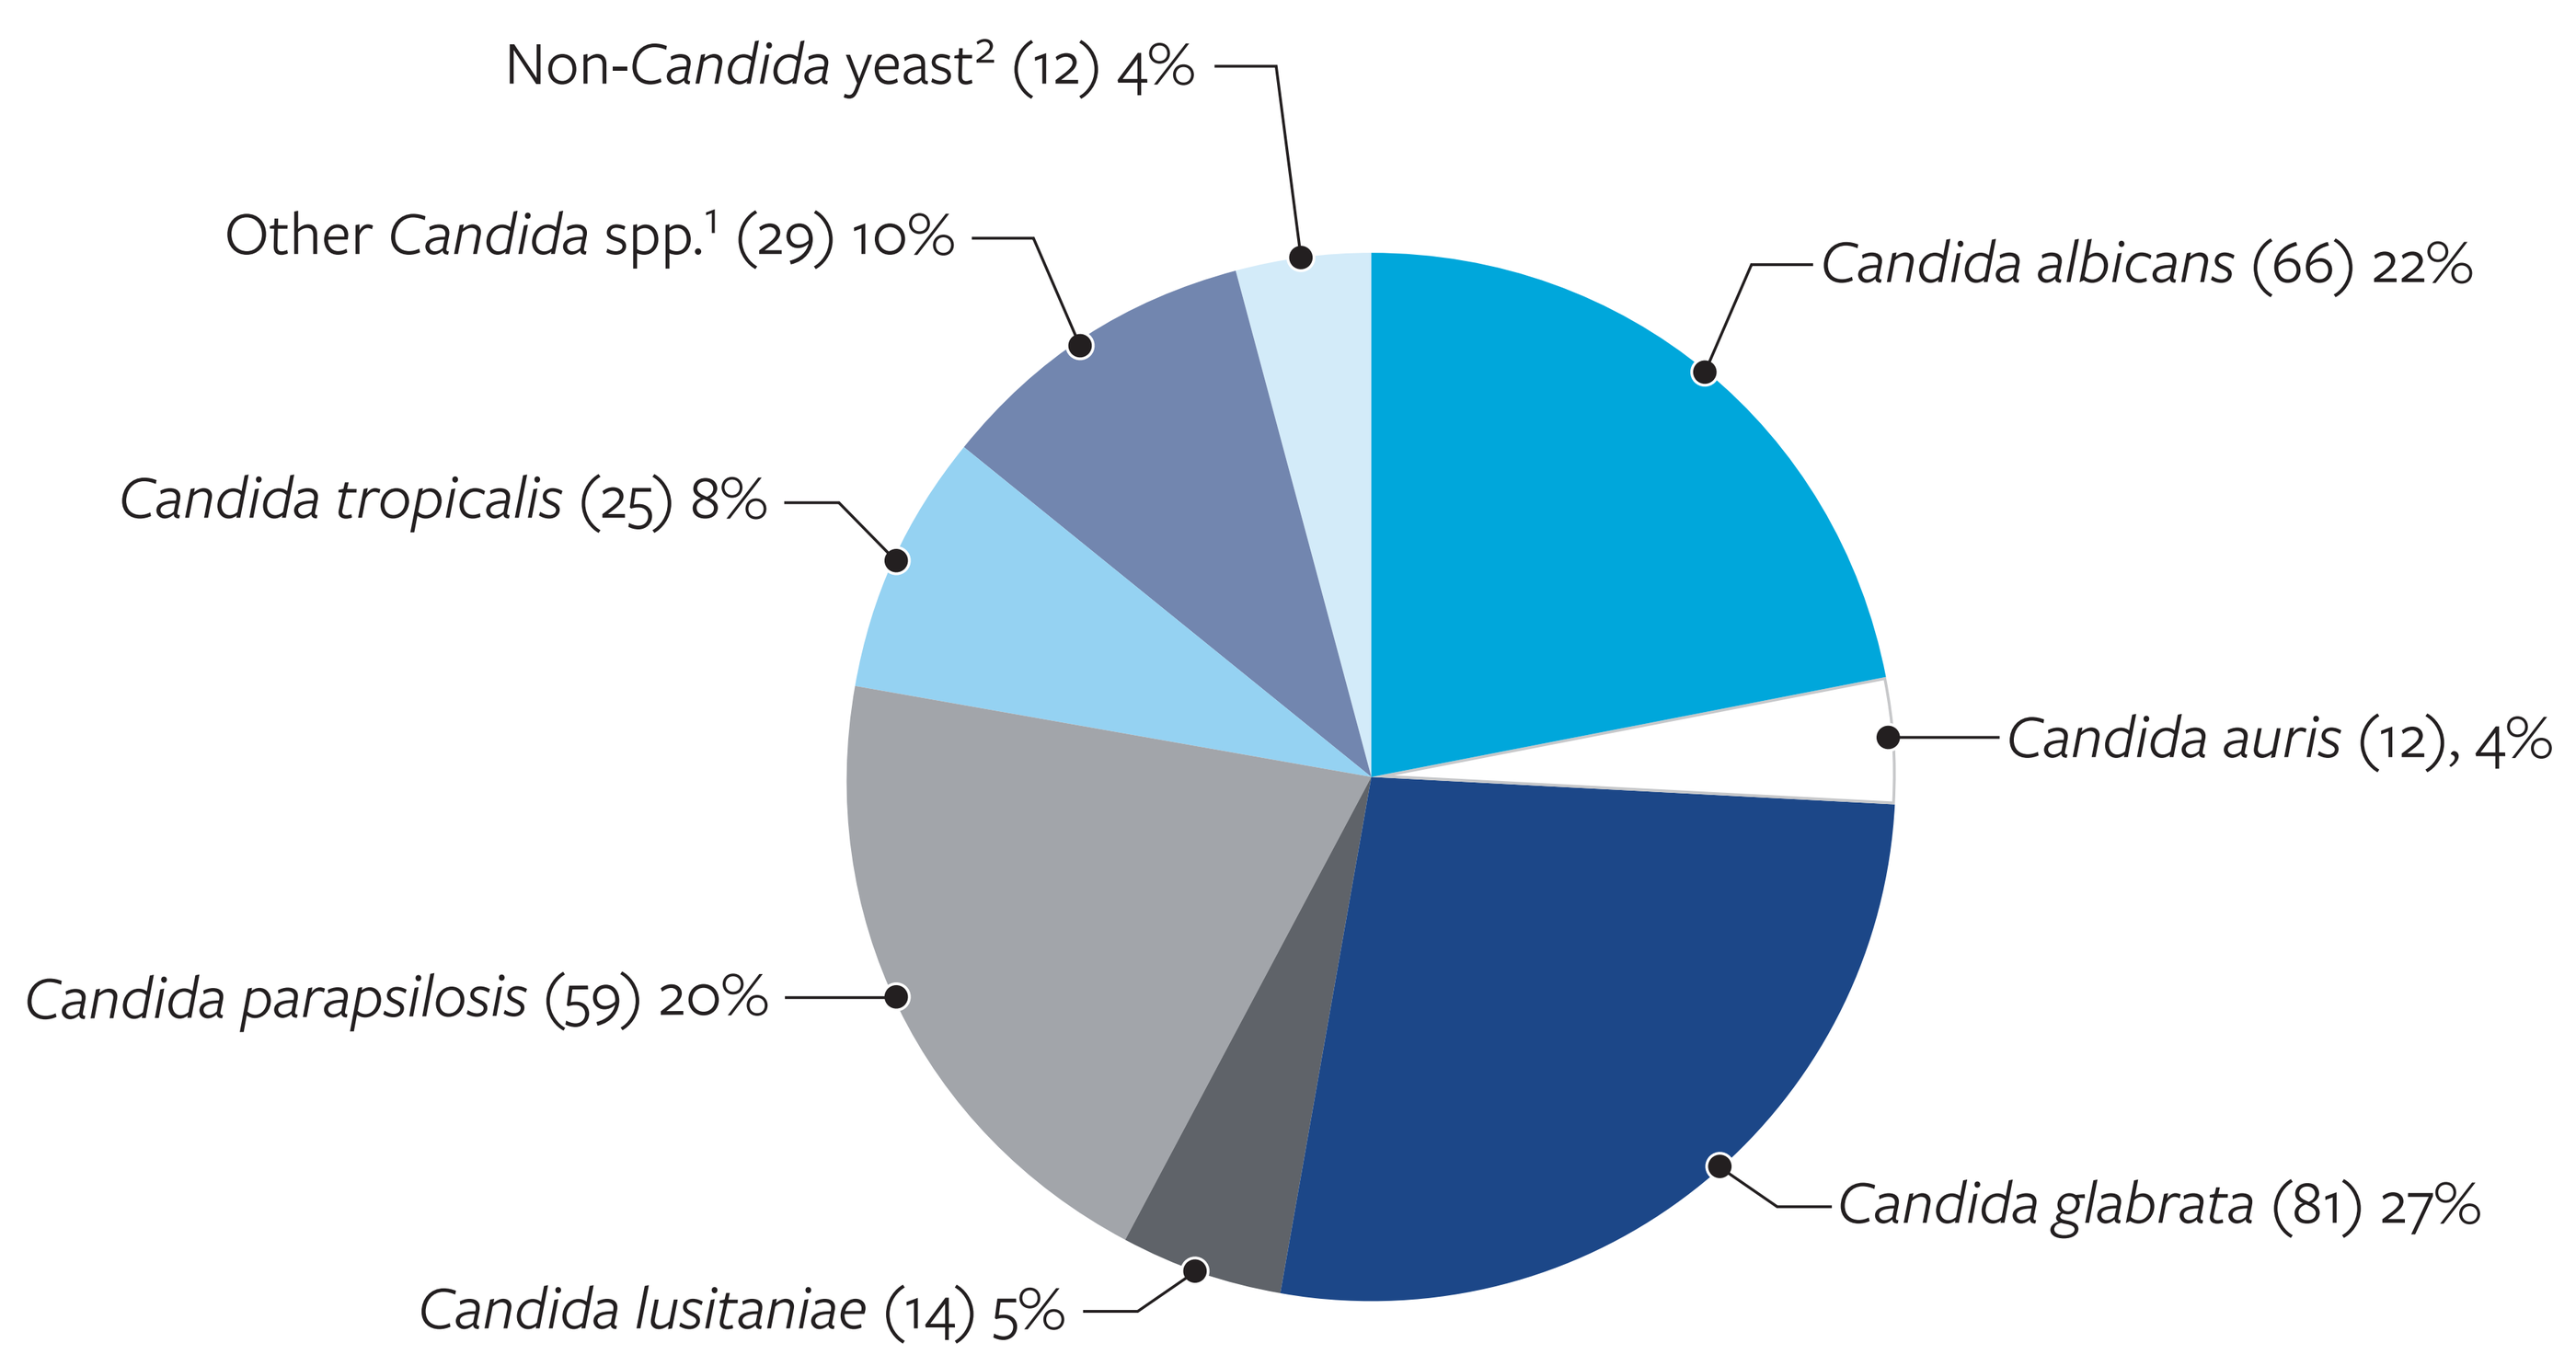
Non-*Candida* yeast² (12) 4%
Other *Candida* spp.¹ (29) 10%
Candida albicans (66) 22%
Candida tropicalis (25) 8%
Candida auris (12), 4%
Candida parapsilosis (59) 20%
Candida glabrata (81) 27%
Candida lusitaniae (14) 5%

[1] Includes *C. cacao* (n=1), *C. dubliniensis* (n=9), *C. fermentati* (n=1), *C. guilliermondii* (n=1), *C. keyfr* (n=1), *C. krusei* (n=9), *C. metapsilosis* (n=1), *C. nivariensis* (n=2), *C. orthopsilosis* (n=3), *C. rugosa* (n=1)
[2] Includes *Cryptococcus neoformans* (n=7), *Rhodotorula mucilaginosa* (n=3), *Saccharomyces cerevisiae* (n=2)

Table 1. Antimicrobial activity of manogepix and comparator agents against yeasts with more than 10 species represented

Organism name (n)	manogepix		anidulafungin		fluconazole		amphotericin B	
	MIC range (mg/L)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L, %S)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L, %S)	$MIC_{50/90}$ (mg/L)	MIC range (mg/L, %S)	$MIC_{50/90}$ (mg/L)
All organisms (298)	0.001 – >4	0.015/0.06	0.004 – >4	0.06/2	0.06 – >128	0.5/32	0.06 – 2	1/1
All *Candida* spp. (286)	0.001 – >4	0.015/0.06	0.004 – >4	0.06/2	0.06 – >128	0.5/32	0.06 – 2	1/1
C. albicans (66)	0.002 – 0.015	0.004/0.008	0.004 – 0.5 (98.5)[1]	0.03/0.06	0.06 – 32 (97.0)[1]	0.25/0.5	0.12 – 1	0.5/1
C. auris (12)	0.008 – 0.06	0.015/0.03	0.12 – >4 (83.3)[2]	0.5/>4	64 – >128 (0)[2]	128/>128	0.5 – 2 (66.7)[2]	0.5/2
C. glabrata (81)	0.004 – 0.12	0.03/0.06	0.03 – 1 (96.3)[1]	0.06/0.12	0.25 – 128 (91.4)[1]	4/32	0.25 – 1	1/1
C. lusitaniae (14)	0.004 – 0.12	0.015/0.12	0.03 – 0.5	0.25/0.5	0.12 – 32	0.5/8	0.12 – 1	0.5/1
C. parapsilosis (59)	0.004 – 0.03	0.008/0.015	0.5 – 4 (91.5)[1]	2/2	0.25 – 128 (89.8)[1]	0.5/4	0.5 – 1	1/1
C. tropicalis (25)	0.004 – 0.03	0.015/0.03	0.008 – 0.06 (100.0)[1]	0.03/0.06	0.12 – 64 (96.0)[1]	0.5/1	0.25 – 1	1/1

[1] Based on breakpoints from CLSI M27M44S 3rd Ed (2022)
[2] Based on CDC resistant-only breakpoints (https://www.cdc.gov/candida-auris/hcp/laboratories/antifungal-susceptibility-testing.html)

Table 2. MICs for anidulafungin, fluconazole, and amphotericin B nonsusceptible (NS) subsets of *Candida* spp.

Organism name (n)	manogepix MIC range (mg/L)	anidulafungin MIC range (mg/L, %S)	fluconazole MIC range (mg/L, %S)	amphotericin B MIC range (mg/L, %S)
Anidulafungin-NS (11)				
Candida albicans (1)	0.008	0.5 (0)	0.25 (100)	1
Candida glabrata (3)	0.03 – 0.06	0.25 – 1 (0)	2 – 64 (66.7)	0.5 – 1
Candida auris (2)	0.008	>4 (0)	64 (0)	0.5 (100)
Candida parapsilosis (5)	0.004 – 0.008	4 (0)	0.25 – 0.5 (100)	1
Fluconazole-NS (28)				
Candida albicans (2)	0.008	0.03 – 0.06 (100)	4 – 32 (0)	0.5 – 1
Candida glabrata (7)	0.03 – 0.06	0.06 – 0.25 (85.7)	64 – 128 (0)	0.5 – 1
Candida auris (12)	0.008 – 0.06	0.12 – >4 (83.3)	64 – >128 (0)	0.5 – 2 (66.7)
Candida parapsilosis (6)	0.008 – 0.03	1 – 2 (100)	4 – 128 (0)	0.5 – 1
Candida tropicalis (1)	0.015	0.015 (100)	64 (0)	1
Amphotericin B-NS (4)				
C. auris (4)	0.015 – 0.06	0.12 – 2 (100)	64 – >128 (0)	2 (0)

Table 3. Antifungal activity and FKS analysis of echinocandin NS *Candida* spp.

Organism name	manogepix MIC (mg/L)	anidulafungin MIC range (mg/L)	fluconazole MIC range (mg/L)	1,3-β-D-glucan synthase mutations[a]: *fks1* HS1	*fks1* HS2	*fks2* HS1	*fks2* HS2
Candida glabrata	0.06	1	2	S629F	Wild type	S663P	Wild type
Candida auris	0.008	>4	64	S639F	Wild type	NT	NT
Candida glabrata	0.03	1	16	Wild type	Wild type	F659del	Wild type
Candida auris	0.008	>4	64	S639F	Wild type	NT	NT
Candida auris	0.015	1	64	D642Y	Wild type	NT	NT
Candida auris	0.015	2	64	D642Y	Wild type	NT	NT
Candida albicans	0.008	0.5	0.25	S645P	Wild type	NT	NT
Candida glabrata	0.06	0.25	64	Wild type	Wild type	Wild type	Wild type
Candida parapsilosis	0.004	4	0.25	NT	NT	NT	NT
Candida parapsilosis	0.008	4	0.25	NT	NT	NT	NT
Candida parapsilosis	0.008	4	0.25	NT	NT	NT	NT
Candida parapsilosis	0.008	4	0.5	NT	NT	NT	NT
Candida parapsilosis	0.008	4	0.5	NT	NT	NT	NT

[a] NT=not tested

Figure 3. Infectious source of collected yeast isolates

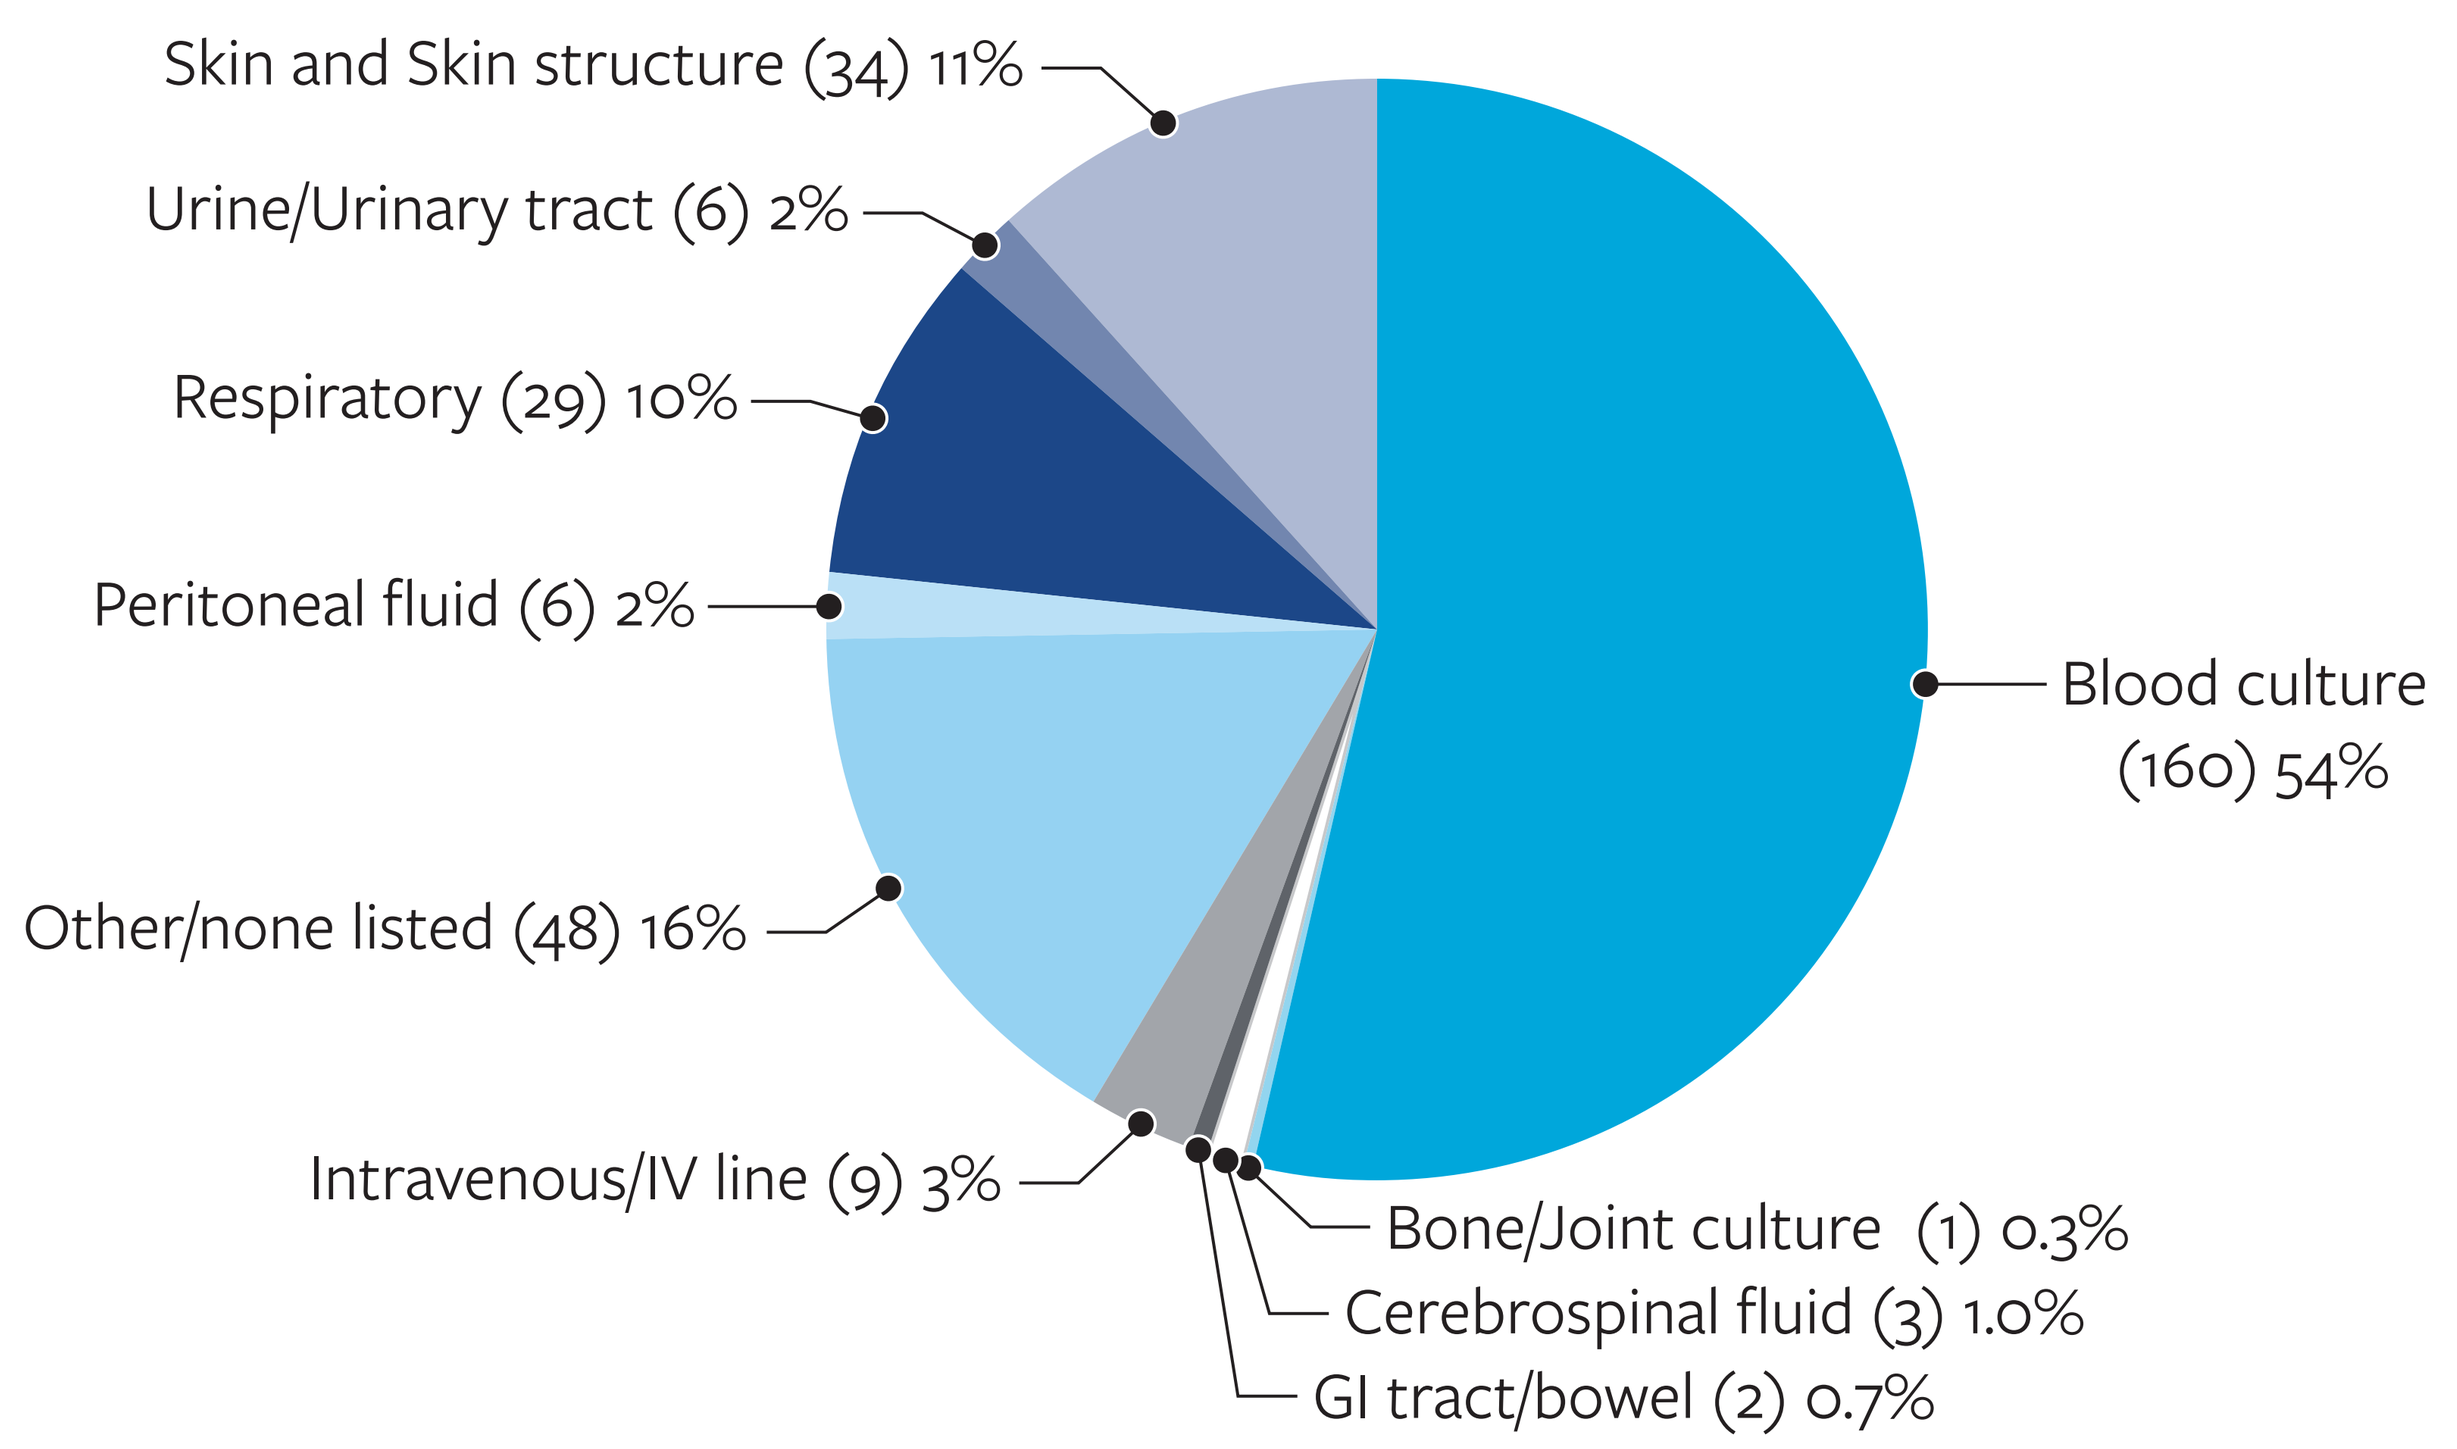
Skin and Skin structure (34) 11%
Urine/Urinary tract (6) 2%
Respiratory (29) 10%
Peritoneal fluid (6) 2%
Other/none listed (48) 16%
Intravenous/IV line (9) 3%
Blood culture (160) 54%
Bone/Joint culture (1) 0.3%
Cerebrospinal fluid (3) 1.0%
GI tract/bowel (2) 0.7%

Contact

element

Marisa Winkler, MD, PhD
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: marisa.winkler@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2025_23-BAS-12_P4_US_Yeast.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia