IDWEEK 2025 | Poster #P1205

In vitro activity of EL219 (SF001) and amphotericin B agents against a collection of *Candida* and *Aspergillus* isolates collected in 2024

S.J.R. Arends, P. Rhomberg, M. Castanheira

Element Iowa City (JMI Laboratories), North Liberty, IA, USA

Introduction

- EL219 is a novel polyene antifungal drug (formerly known as SF001) currently in preparation for Phase 2 clinical development (Elion Therapeutics).
- Through synthetic modifications to amphotericin B, EL219 was rationally designed to improve antifungal activity while reducing the potential for toxicity.
- This international surveillance study reports on the *in vitro* activity of EL219 and amphotericin B tested against contemporary *Aspergillus* and *Candida* clinical isolates.

Methods

- A total of 265 *Aspergillus* species (spp.) and 130 *Candida* spp. isolates were collected in 2024 as a part of the SENTRY global surveillance program.
- Isolates were from 25 medical centers in North America, 33 medical centers in Europe, 9 medical centers in the Asia-Pacific region, and 6 medical centers in Latin America (Figure 1).
- Susceptibility to EL219 was evaluated for the 3 most common *Aspergillus* spp. and 4 most common *Candida* spp. (Figure 2).
- Only 1 isolate per patient was included.
- Isolate identification was performed by MALDI-TOF MS and/or molecular methods.
- Susceptibility testing was performed using broth microdilution methods, according to CLSI standards.
- Frozen-form panels used RPMI 1640 broth supplemented with MOPS (morpholinepropane sulfonic acid buffer) and 0.2% glucose and were inoculated with 0.5 to 2.5 X 10^3 cells/mL suspensions for yeasts and 0.4 to 5 X 10^4 cells/mL for moulds.
- EL219 was tested as the micellular drug product formulation in a 1:3 molar ratio with DSG-PEG2000.
- MIC endpoints for both EL219 and amphotericin B were read at 100% inhibition after 24 hours incubation for yeasts and at 48 hours incubation for moulds.
- There are no current breakpoints for EL219 by CLSI criteria, but CLSI breakpoints or epidemiological cutoff values (ECVs) were applied for comparator agents, where available, and CDC tentative breakpoints were used for *C. auris*.

Results

Activity against *Candida* species

- EL219 was also active against all *Candida* species with MIC_{90} values ranging from 0.25–1 μg/mL (Table 1):
 - *C. albicans* (n=36; $MIC_{50/90}$, 0.12/0.25 μg/mL)
 - *C. auris* (n=31; $MIC_{50/90}$, 0.25/1 μg/mL)
 - *C. glabrata* (n=28; $MIC_{50/90}$, 0.12/0.25 μg/mL)
 - *C. parapsilosis* (n=16; $MIC_{50/90}$, 0.12/0.25 μg/mL)
 - Other *Candida* species (n=19; $MIC_{50/90}$, 0.12/0.25 μg/mL).
- Amphotericin B showed activity against all *Candida* species groups whose MIC_{90} values ranged from 1–2 μg/mL.
- On average, EL219 was 4- to 8-fold more active than amphotericin B against *Candida* species including *C. albicans* and *C. auris* (31) (Table 2 and Figure 3).

Activity against *Aspergillus* species

- EL219 was active against all *Aspergillus* species with MIC_{90} values ranging from 0.25–1 μg/mL (Table 1):
 - *A. fumigatus* (n=157; $MIC_{50/90}$, 0.5/1 μg/mL)
 - *A. flavus* species complex (n=32; $MIC_{50/90}$, 0.5/0.5 μg/mL)
 - *A. niger* species complex (n=57; $MIC_{50/90}$, 0.25/0.25 μg/mL)
 - Other *Aspergillus* species (n=19; $MIC_{50/90}$, 0.5/1 μg/mL).
- Amphotericin B showed activity against all *Aspergillus* species groups with MIC_{90} values ranging from 0.5–4 μg/mL (Table 1).
- EL219 was 2- to 4-fold more active against *Aspergillus fumigatus*, *Aspergillus flavus* species complex, and other *Aspergillus* species than amphotericin B (Table 1), whereas amphotericin B and EL219 displayed similar MIC values against *Aspergillus niger* species complex (Table 2 and Figure 3).

Conclusions

- EL219 was active against prevalent *Aspergillus* and *Candida* clinical isolates causing invasive mycoses collected in 2024. EL219 was often 4- to 8-fold more active than amphotericin B when comparing $MIC_{50/90}$ values.
- EL219 showed good activity (all MIC values ≤1 μg/mL) when tested against *Aspergillus* species. This activity was similar for all species including the most prevalent (*A. fumigatus*, *A. flavus*, and *A. niger*) as well as less common species exhibiting reduced polyene susceptibility (*A. lentulus* and *A. terreus*). The activity of EL219 when tested as a micellar formulation in a 1:3 molar ratio with DSG-PEG2000 was 4-fold more active than amphotericin B against *A. fumigatus* and *A. flavus* species complex when compared by MIC_{50}.
- EL219 showed good activity when tested against *Candida* species (all MIC values ≤ 2 mg/L). This activity was similar for all *Candida* species tested with MIC_{50} values ranging from 0.12 to 0.25 mg/L for the most prevalent *Candida* species including *C. albicans*, *C. auris*, *C. glabrata*, *C. parapsilosis*, and *C. tropicalis*. EL219 was 4- to 8-fold more potent by MIC_{50} than amphotericin B when comparing MIC_{50} values.
- These results further support the continued development of EL219 for treatment of invasive fungal infections.

Acknowledgments

This study at Element Iowa City (JMI Laboratories) was supported by Elion Therapeutics, Inc. (Boston, MA). Element Iowa City received compensation fees for services in relation to preparing the poster, which was funded by Elion Therapeutics.

Table 1. Distribution of EL219 and amphotericin B MIC values of *Candida* and *Aspergillus* isolates collected in 2024

Species (No. isolates) / Antifungal agent	≤0.03	0.06	0.12	0.25	0.5	1	2	4	MIC_{50}	MIC_{90}
All *Aspergillus* species (265)										
EL219			15	82	146	22			0.5	0.5
Amphotericin B	2	4	19	27	23	64	121	2	1	2
Aspergillus fumigatus (157)										
EL219			3	25	111	18			0.5	1
Amphotericin B				4	10	54	89		2	2
Aspergillus flavus species complex (32)										
EL219				11	19	2			0.5	0.5
Amphotericin B				1	2	7	22		2	2
Aspergillus niger species complex (57)										
EL219			11	42	4				0.25	0.25
Amphotericin B	2	4	19	22	9	1			0.25	0.5
Other *Aspergillus* species [a] (19)										
EL219			1	4	12	2			0.5	1
Amphotericin B					2	5	10	2	2	4
All *Candida* species (130)										
EL219			73	40	8	8	1		0.12	0.5
Amphotericin B				3	45	57	25		1	2
Candida albicans (36)										
EL219			23	12			1		0.12	0.25
Amphotericin B					28	7	1		0.5	1
Candida auris (31)										
EL219			4	12	7	8			0.25	1
Amphotericin B					1	8	22		2	2
Candida glabrata (28)										
EL219			22	6					0.12	0.25
Amphotericin B					5	23			1	1
Candida parapsilosis (16)										
EL219			9	7					0.12	0.25
Amphotericin B					6	9	1		1	1
Other *Candida* species [b] (19)										
EL219			15	3	1				0.12	0.25
Amphotericin B				3	5	10	1		1	1

EL219 was tested as a micellar formulation in a 1:3 molar ratio with DSG-PEG2000
[a]Included species: *Aspergillus lentulus* (1), *A. nidulans* species complex (5), *A. sclerotiorum* (1), *A. terreus* species complex (11), and unspeciated *Aspergillus* (1)
[b]Included species: *Candida dubliniensis* (3), *C. guilliermondii* (1), *C. intermedia* (1), *C. kefyr* (1), *C. krusei* (1), *C. lusitaniae* (2), *C. metapsilosis* (1), *C. orthopsilosis* (1), *C. tropicalis* (8)

Table 2. Differences between EL219 and amphotericin B MIC values of *Aspergillus* and *Candida* species

Organism/organism group (No. isolates)	No. of isolates (log_2 dilution deviation)[b]								
	Lower EL219 MIC						Higher EL219 MIC		
	≤ −4	−3	−2	−1	0	+1	+2	+3	≥ +4
All *Aspergillus* species (265)		12	119	74	38	16	6		
Aspergillus fumigatus (157)		8	82	55	10	2			
Aspergillus flavus species complex (32)		3	22	6	1				
Aspergillus niger species complex (57)			1	9	27	14	6		
Other *Aspergillus* species [a] (19)		1	14	4					
All *Candida* species (130)	1	44	60	24	1				
Candida albicans (36)		3	24	8	1				
Candida auris (31)		10	13	8					
Candida glabrata (28)		18	9	1					
Candida parapsilosis (16)	1	5	7	3					
Other *Candida* species [b] (19)		8	7	4					

[a]Includes species: *Aspergillus lentulus* (1), *A. nidulans* species complex (5), *A. sclerotiorum* (1), *A. terreus* species complex (11), and unspeciated *Aspergillus* (1)
[b]Includes species: *Candida dubliniensis* (3), *C. guilliermondii* (1), *C. intermedia* (1), *C. kefyr* (1), *C. krusei* (1), *C. lusitaniae* (2), *C. metapsilosis* (1), *C. orthopsilosis* (1), *C. tropicalis* (8)

Figure 1. Geographic distribution of isolates

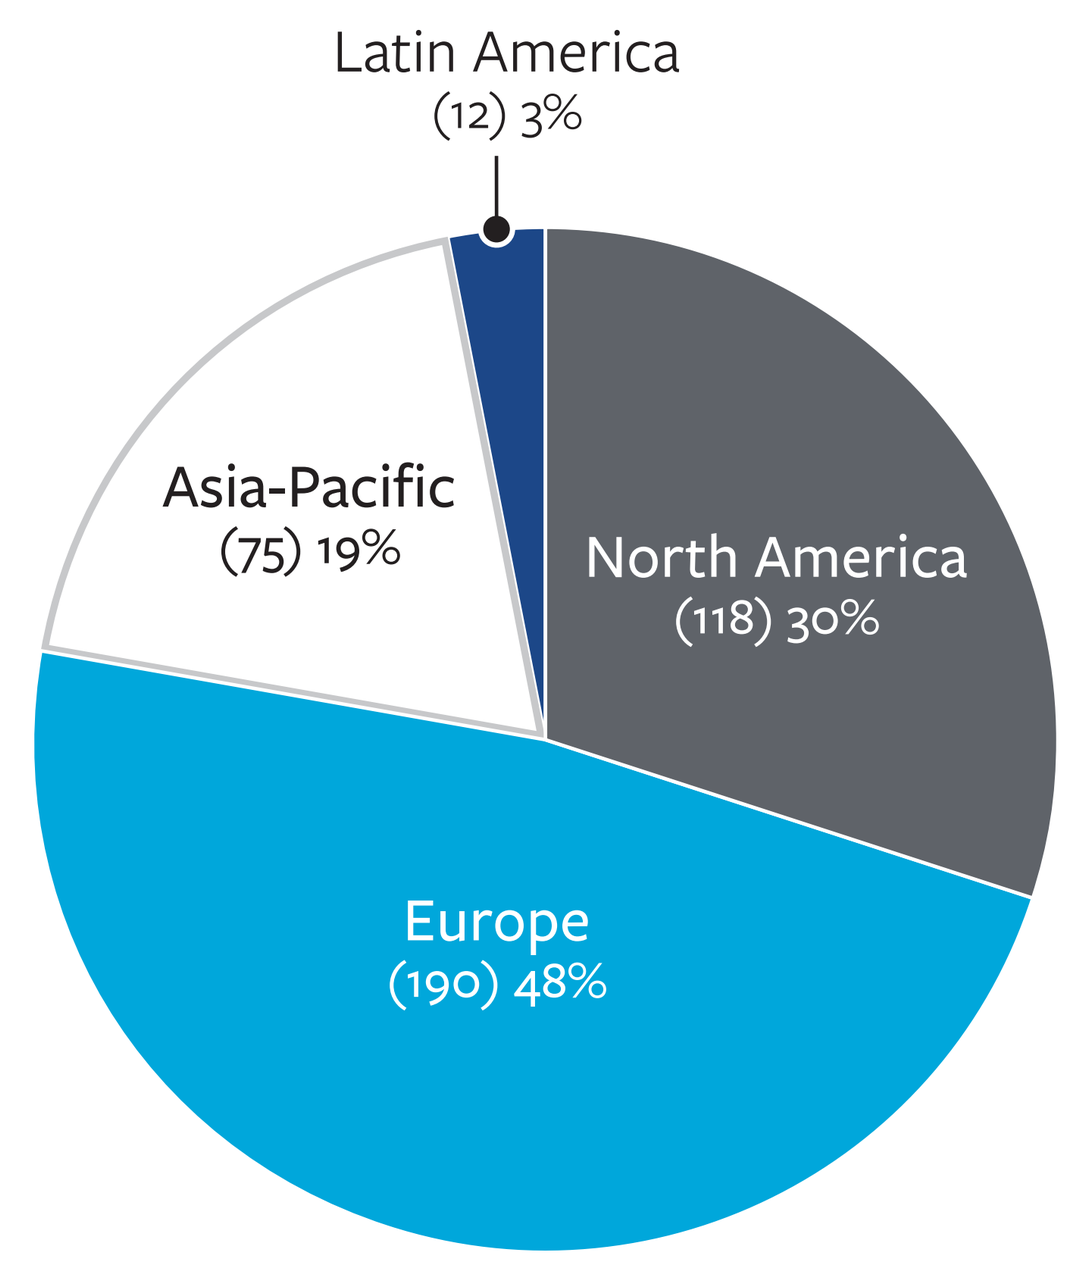

Latin America (12) 3%
Asia-Pacific (75) 19%
North America (118) 30%
Europe (190) 48%

Figure 2. Species of fungal isolates included in surveillance

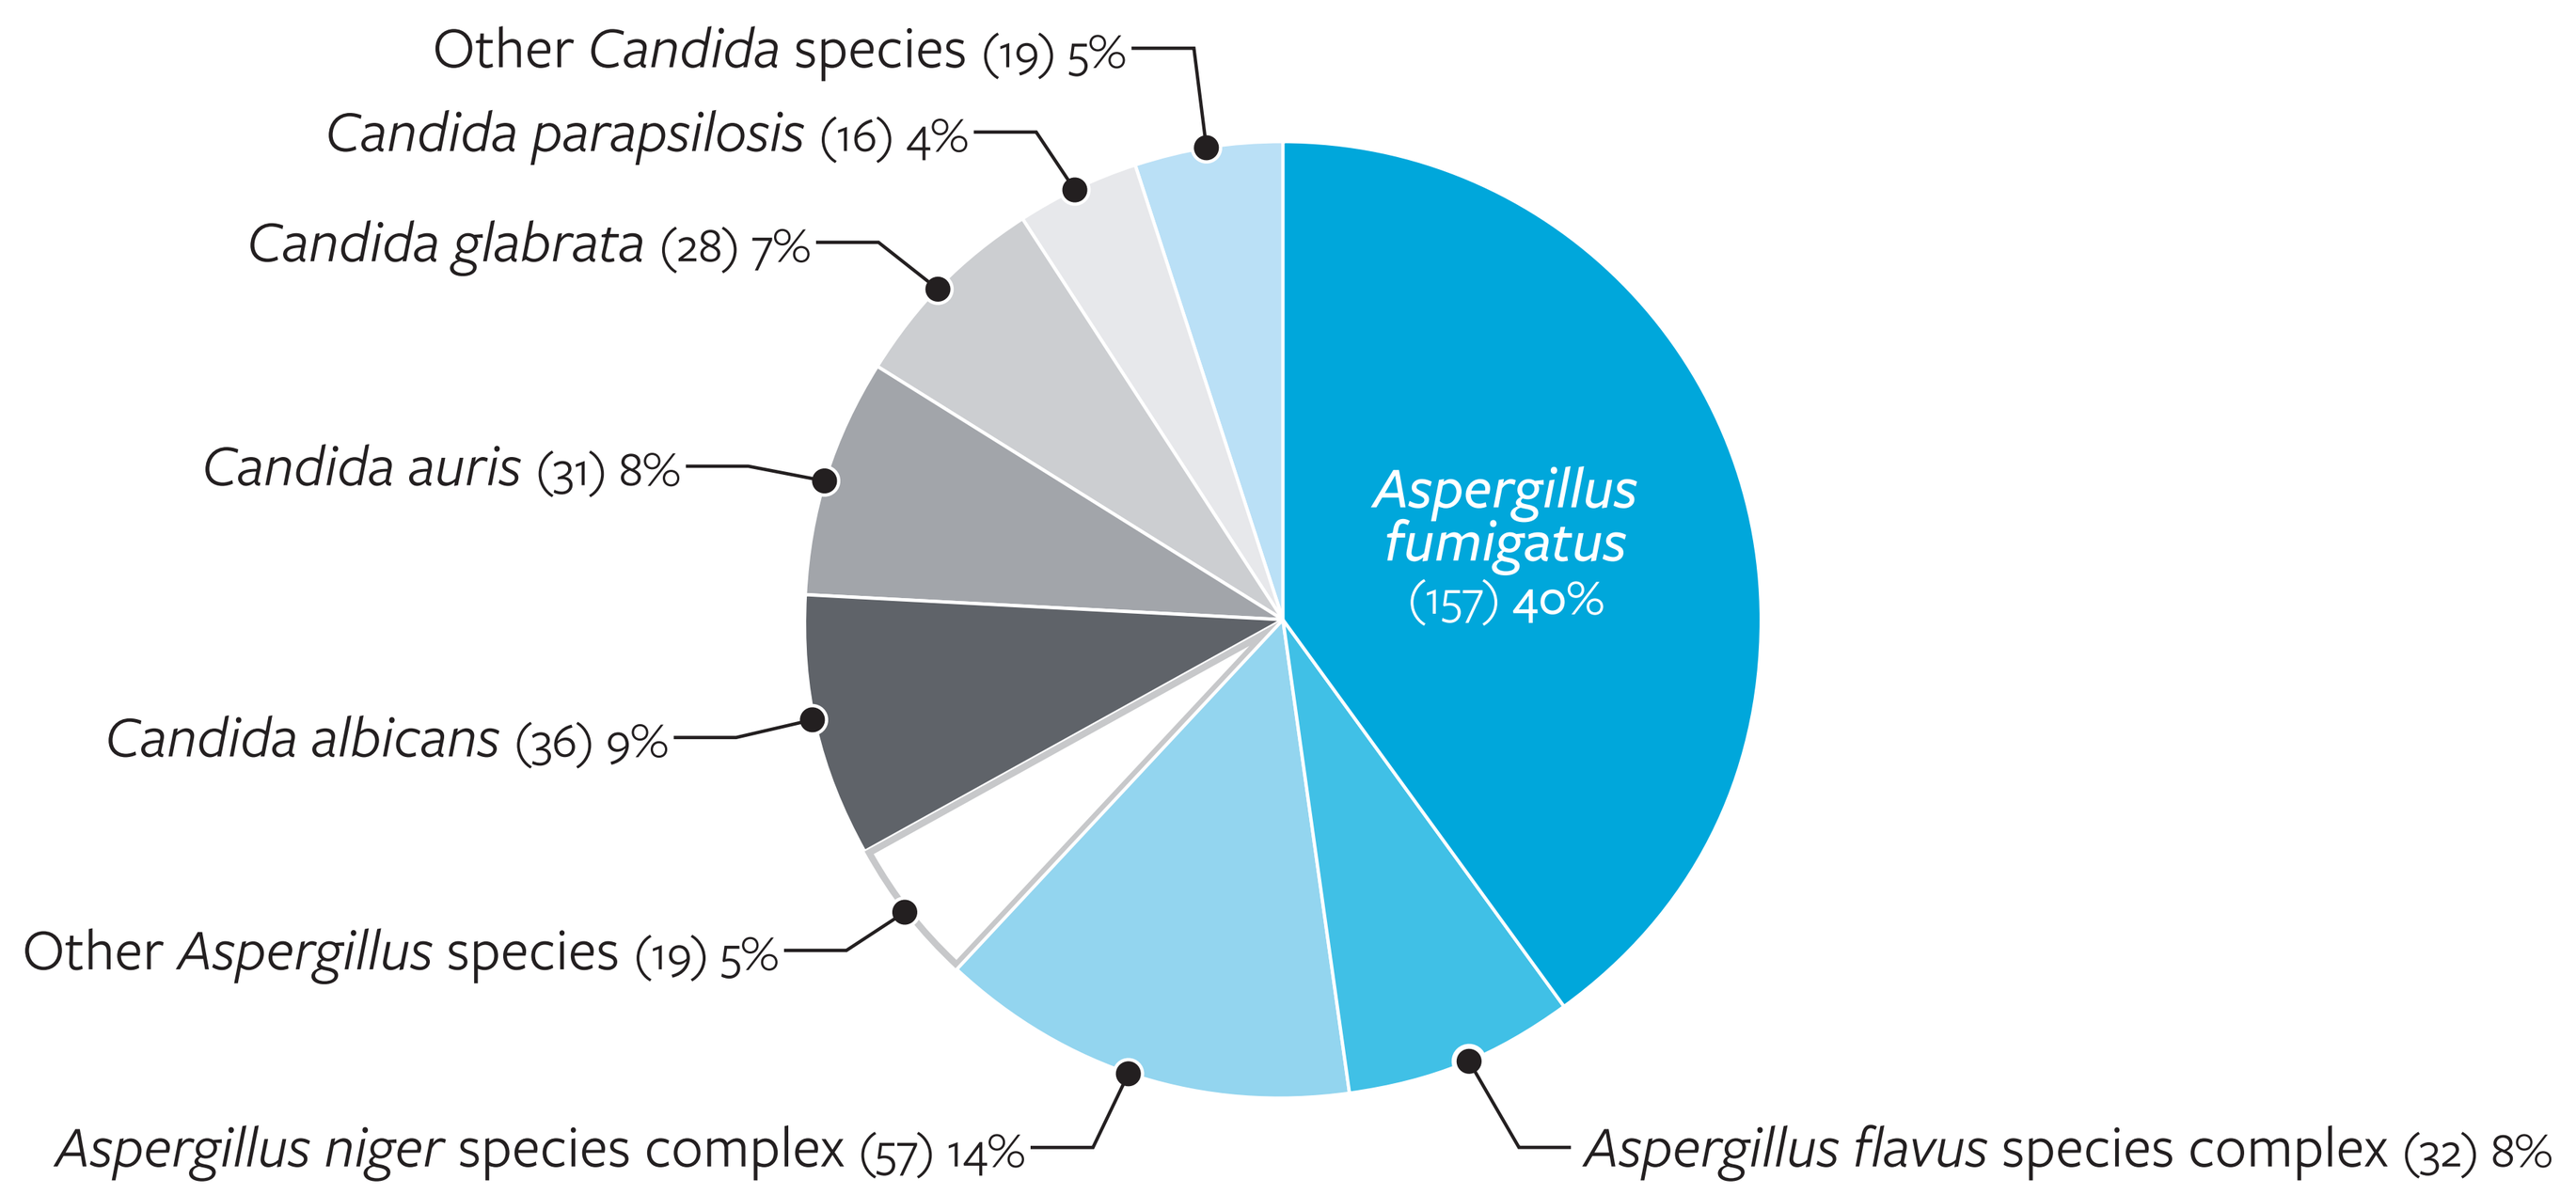

Other Candida species (19) 5%
Candida parapsilosis (16) 4%
Candida glabrata (28) 7%
Candida auris (31) 8%
Candida albicans (36) 9%
Other Aspergillus species (19) 5%
Aspergillus niger species complex (57) 14%
Aspergillus fumigatus (157) 40%
Aspergillus flavus species complex (32) 8%

Figure 3. Scatterplot of EL219 and amphotericin B MIC values of *Aspergillus* and *Candida* species

***Aspergillus* Isolates (n = 265)**

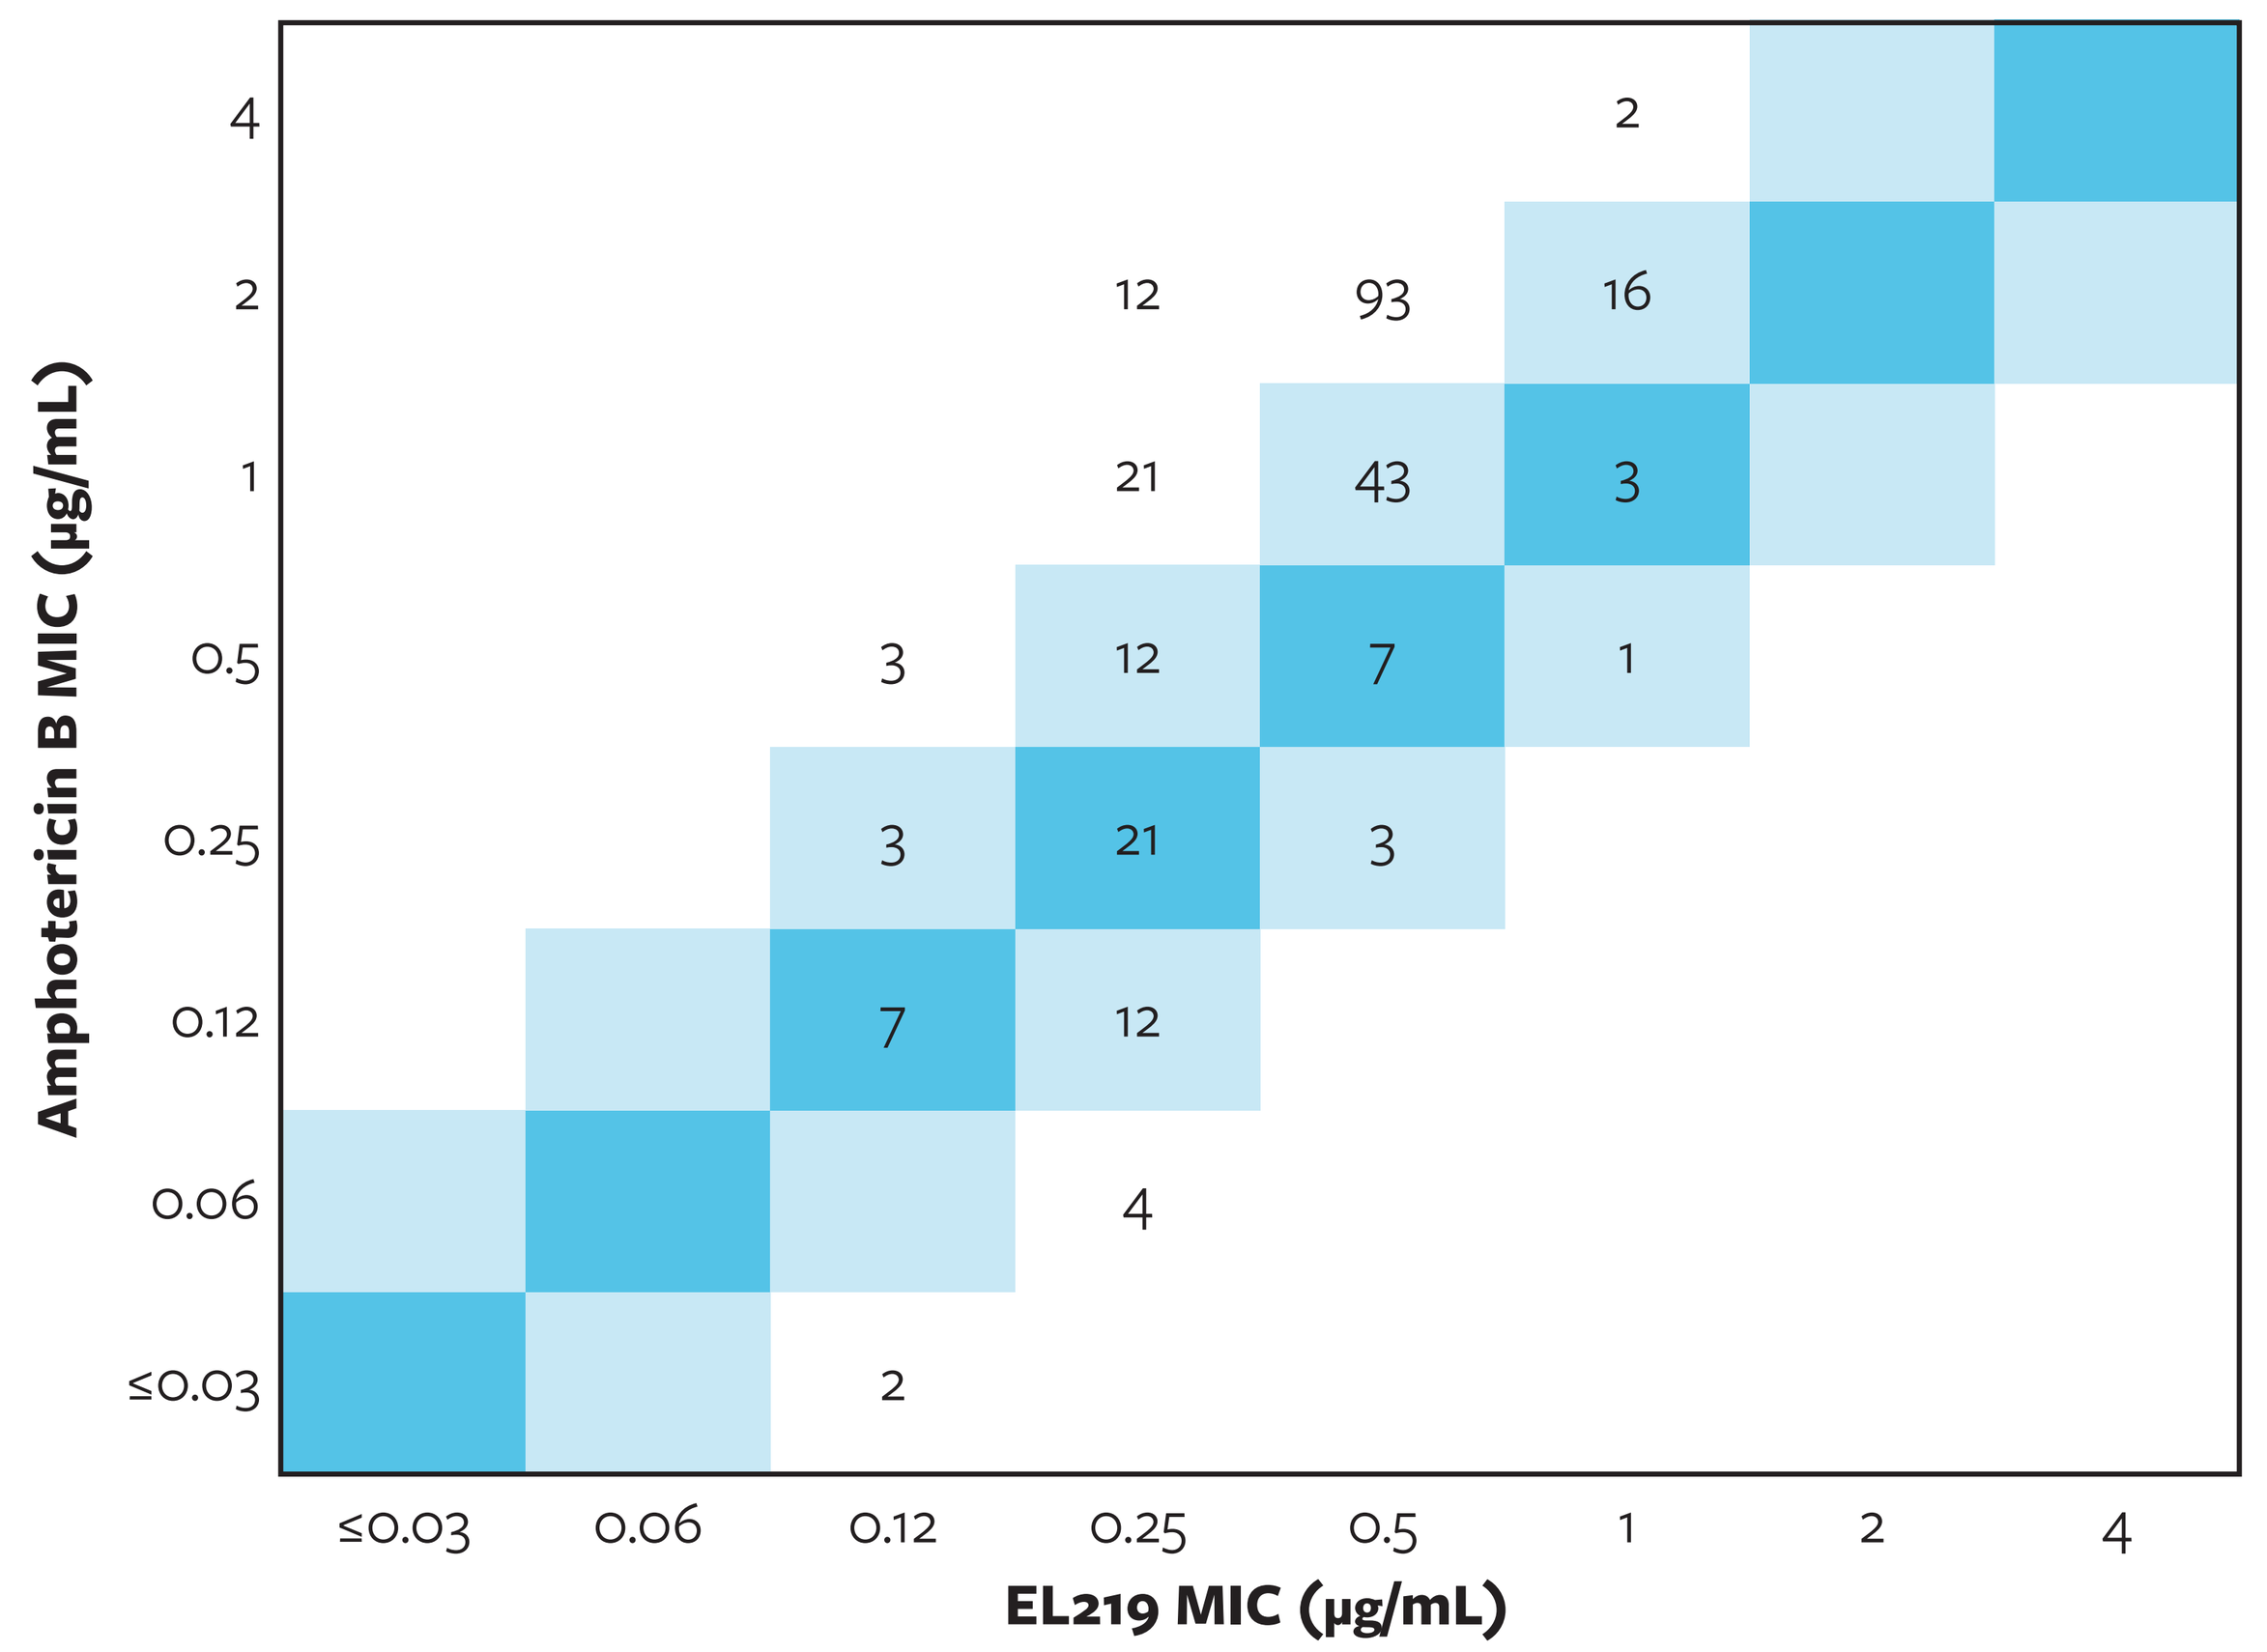

Amphotericin B MIC (μg/mL)
4
2
1
0.5
0.25
0.12
0.06
≤0.03
2
12 93 16
21 43 3
3 12 7 1
3 21 3
7 12
4
2
≤0.03 0.06 0.12 0.25 0.5 1 2 4
EL219 MIC (μg/mL)

***Candida* Isolates (n = 130)**

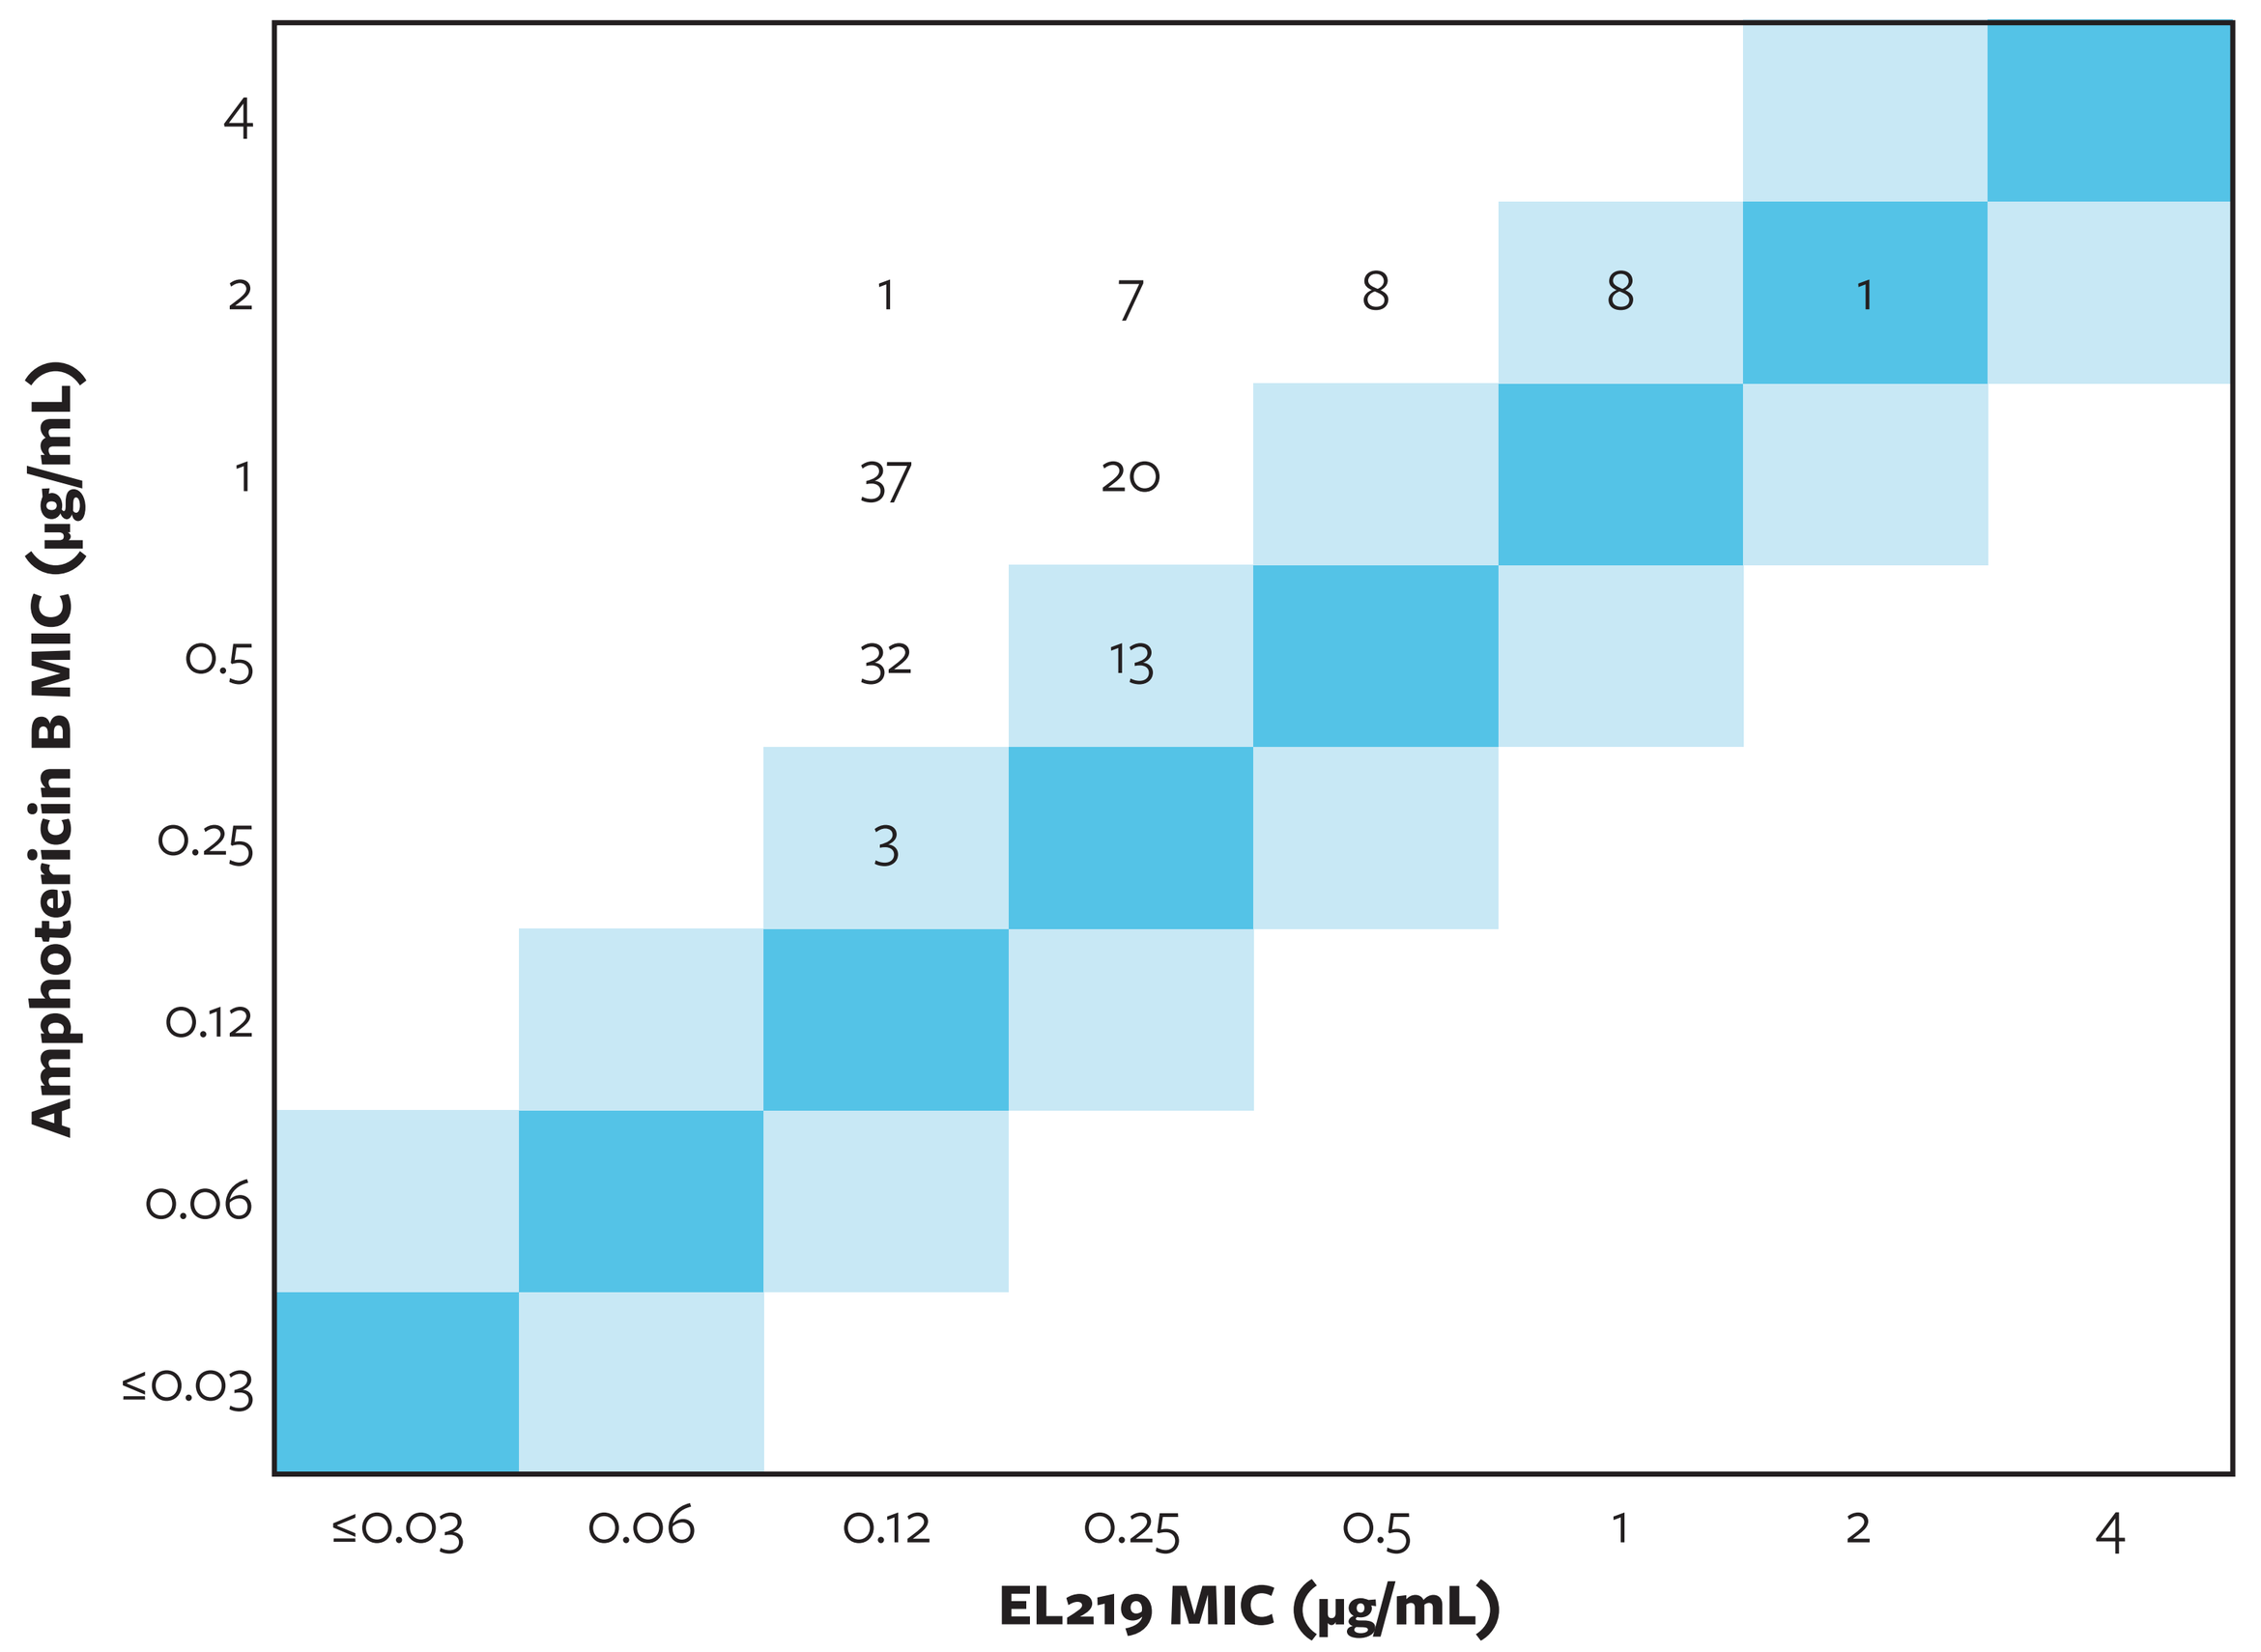

Amphotericin B MIC (μg/mL)
4
2
1
0.5
0.25
0.12
0.06
≤0.03
1 7 8 8 1
37 20
32 13
3
≤0.03 0.06 0.12 0.25 0.5 1 2 4
EL219 MIC (μg/mL)

References

CLSI. M27Ed4E. Reference method for broth dilution antifungal susceptibility testing of yeasts. Wayne, PA, Clinical and Laboratory Standards Institute, 2017.

CLSI. M27M44SEd3E Performance Standards for Antifungal Susceptibility Testing of Yeasts. Wayne, PA, Clinical and Laboratory Standards Institute, 2022.

CLSI. MM18Ed2E. Interpretive criteria for identification of bacteria and fungi by targeted DNA sequencing. Wayne, PA, Clinical and Laboratory Standards Institute, 2018.

CLSI. M38Ed3E. Reference method for broth dilution antifungal susceptibility testing of filamentous fungi, third edition. Wayne, PA, Clinical and Laboratory Standards Institute, 2018.

CLSI. M38M51SEd3E. Reference method for broth dilution antifungal susceptibility testing of filamentous fungi, third edition. Wayne, PA, Clinical and Laboratory Standards Institute, 2022.

CLSI. M57SEd4E Epidemiological Cutoff Values for Antifungal Susceptibility Testing, 4th Edition. Wayne, PA, Clinical and Laboratory Standards Institute, 2022.

Contact

element

Ryan Arends, Ph.D.
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: ryan.arends@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2025_24-ELN-01_P1_SF001.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia