IDWEEK 2025 | Poster #1196

Geographic distribution, MIC testing, and susceptibility rates of Candida auris isolates collected in the United States since 2014

Marisa L. Winkler, John H. Kimbrough, Kelley Fedler, Samuel Edeker, Abby Klauer, Paul Rhomberg, Mariana Castanheira

Element Materials Technology (JMI Laboratories), North Liberty, Iowa, USA

Introduction

- *Candida auris* is an emerging infectious threat due to rising infection rates, eradication difficulty, and multidrug resistance.
- Susceptible (S)-only breakpoint for *C. auris* from the Clinical and Laboratory Standards Institute (CLSI) exists for rezafungin at ≤0.5 mg/L.
- The US Centers for Disease Control (CDC) has tentative resistant (R)-only breakpoints against *C. auris* for fluconazole, amphotericin B, anidulafungin, caspofungin, and micafungin with the caveat that the correlation between the values and clinical outcomes is unknown and these values are only based on the modal MIC distributions of approximately 100 isolates.
- The CLSI S-only breakpoint for rezafungin is several dilutions lower than the R CDC breakpoints for other echinocandins.
- Given the emergence and rapid spread of *C. auris* and its high rate of multidrug resistance, it is essential to understand the geographic distribution, susceptibility patterns, and clonality of *C. auris*.

Methods

- A total of 79 *C. auris* isolates were collected from 2015–2024 in the United States.
 - These represent 12 hospitals, 9 states, and 5 census divisions (Figure 1).
 - There were no isolates from the Mountain, Pacific, New England, or West North Central census divisions though fungal isolates were collected by prevalence in these regions.
- Only 1 isolate per patient episode was included.
- All isolates were identified by MALDI-TOF MS and/or DNA sequencing.
- Isolates were tested by CLSI reference broth microdilution method (M27) for rezafungin, anidulafungin, caspofungin, micafungin, fluconazole, voriconazole, and amphotericin B.
- CLSI breakpoints (M27M44S) were applied for rezafungin and CDC breakpoints were applied for comparator agents.
- Non-susceptible (NS) by CLSI definition was defined as an MIC of ≥1 mg/L for rezafungin; for comparators, resistant (R) by the CDC definition was defined as ≥4, ≥2, ≥4, ≥32, and ≥2 mg/L for anidulafungin, caspofungin, micafungin, fluconazole, and amphotericin B, respectively.
- All isolates were whole genome sequenced (WGS) and resulting FASTQ format files were quality trimmed and filtered with Sickle; publicly available references for Clades I–VI were also included.
 - Variant calling was performed with Snippy using B8441 (Clade I) as the reference. Results of the variant calling were used to create a core SNP alignment, and recombination filtering was performed with Gubbins.
 - Phylogenetic reconstruction was performed with FastTree utilizing the generalized time-reversible model and isolates were assigned to clades based on their clustering within the phylogeny; genetic alterations were determined with respect to the intraclade reference strain for FKS1, ERG11, TAC1B, UPC2, MDR1, and CDR1.

Results

- The 79 *C. auris* isolates were from 7 different infection sources.
 - The majority of isolates (55.0%) were from bloodstream infections with 21.0% from tissue infections and 6% from the respiratory tract.
- Prevalence of isolates has exponentially increased since 2014 (Figure 2).
- Rezafungin susceptibility rate was 86.1% among all isolates, 85.7% in fluconazole-resistant isolates, and 88.6% in isolates resistant to fluconazole and amphotericin B (Table 1).
 - Non-resistant rates ranged from 90.9–100.0% for other echinocandins.
 - Fluconazole non-resistant rates were low at 2.5%; amphotericin B were 55.7%.
- Based on WGS, all isolates fell into clade I (50.7%), clade III (43.0%), or clade IV (6.3%) (Figure 3).
 - Clade III isolates were predominately contributed by medical centers in the West South Central census division (91.2%) whereas clade I and IV isolates displayed a more variable geographic representation.
 - Echinocandin-R and/or rezafungin-NS isolates were present in all clades.
 - Almost all (97.5%) isolates across clades were fluconazole-R. In contrast, clade I isolates were 75.0% amphotericin B-R while clade III was 8.8% R and clade IV was 40.0% R.
 - Voriconazole MICs showed significant variability across isolates within the different clades.
- Many isolates in clades I and IV displayed variability in ERG11, TAC1B, and CDR1 compared to the B8441 and B11245 reference strains, respectively.
 - Few isolates in clade III showed variability in the analyzed genes compared to the B11221 reference strain.
 - Only 1 and 3 isolates across clades had alterations in UPC2 or MDR1, respectively, compared to the clade reference.
- Three and four isolates in clades I and III, respectively, had FKS1 alterations and 85.7% of these isolates displayed elevated echinocandin MIC values.
 - There were more isolates with elevated echinocandin MICs in clades III and IV (7.5%, 20.6%, and 20.0% in clades I, III, and IV, respectively).
 - Four isolates in clade III had elevated MICs to one or more echinocandins without identified alterations.

Conclusions

- *C. auris* infections are a growing problem in the United States, though their presence remains geographically limited with low prevalence in some areas.
 - Our data is limited by lack of collection sites in Nevada and California, which have a high prevalence of *C. auris* isolates.
- Resistance rates are high for fluconazole (97.5%) and amphotericin B (44.3%).
 - In contrast, echinocandin resistance remains rare (<10% for agents with CDC R-only breakpoint); 13.9% of isolates were NS to rezafungin reflecting the CLSI S-only breakpoint at 1–2 doubling dilutions below the R-only breakpoint for other echinocandins.
 - 36.4% of isolates with elevated echinocandin MICs did not have genetic changes identified by WGS and warrant further evaluation to understand the mechanistic basis of their MIC pattern.
- Echinocandins remain the most active agent against *C. auris* irrespective of clade type or geographic origin.

Funding

ML Winkler, K Fedler, A Klauer, S Edeker, P Rhomberg, JH Kimbrough, and M Castanheira were employees of Element Materials Technology (JMI Laboratories) at the time of this study. JMI Laboratories was a paid consultant to Melinta in connection with the development of this poster.

Acknowledgments

The authors thank all of the SENTRY Program participants for providing the isolates used in this study.

Figure 1. Map of geography of collected isolates

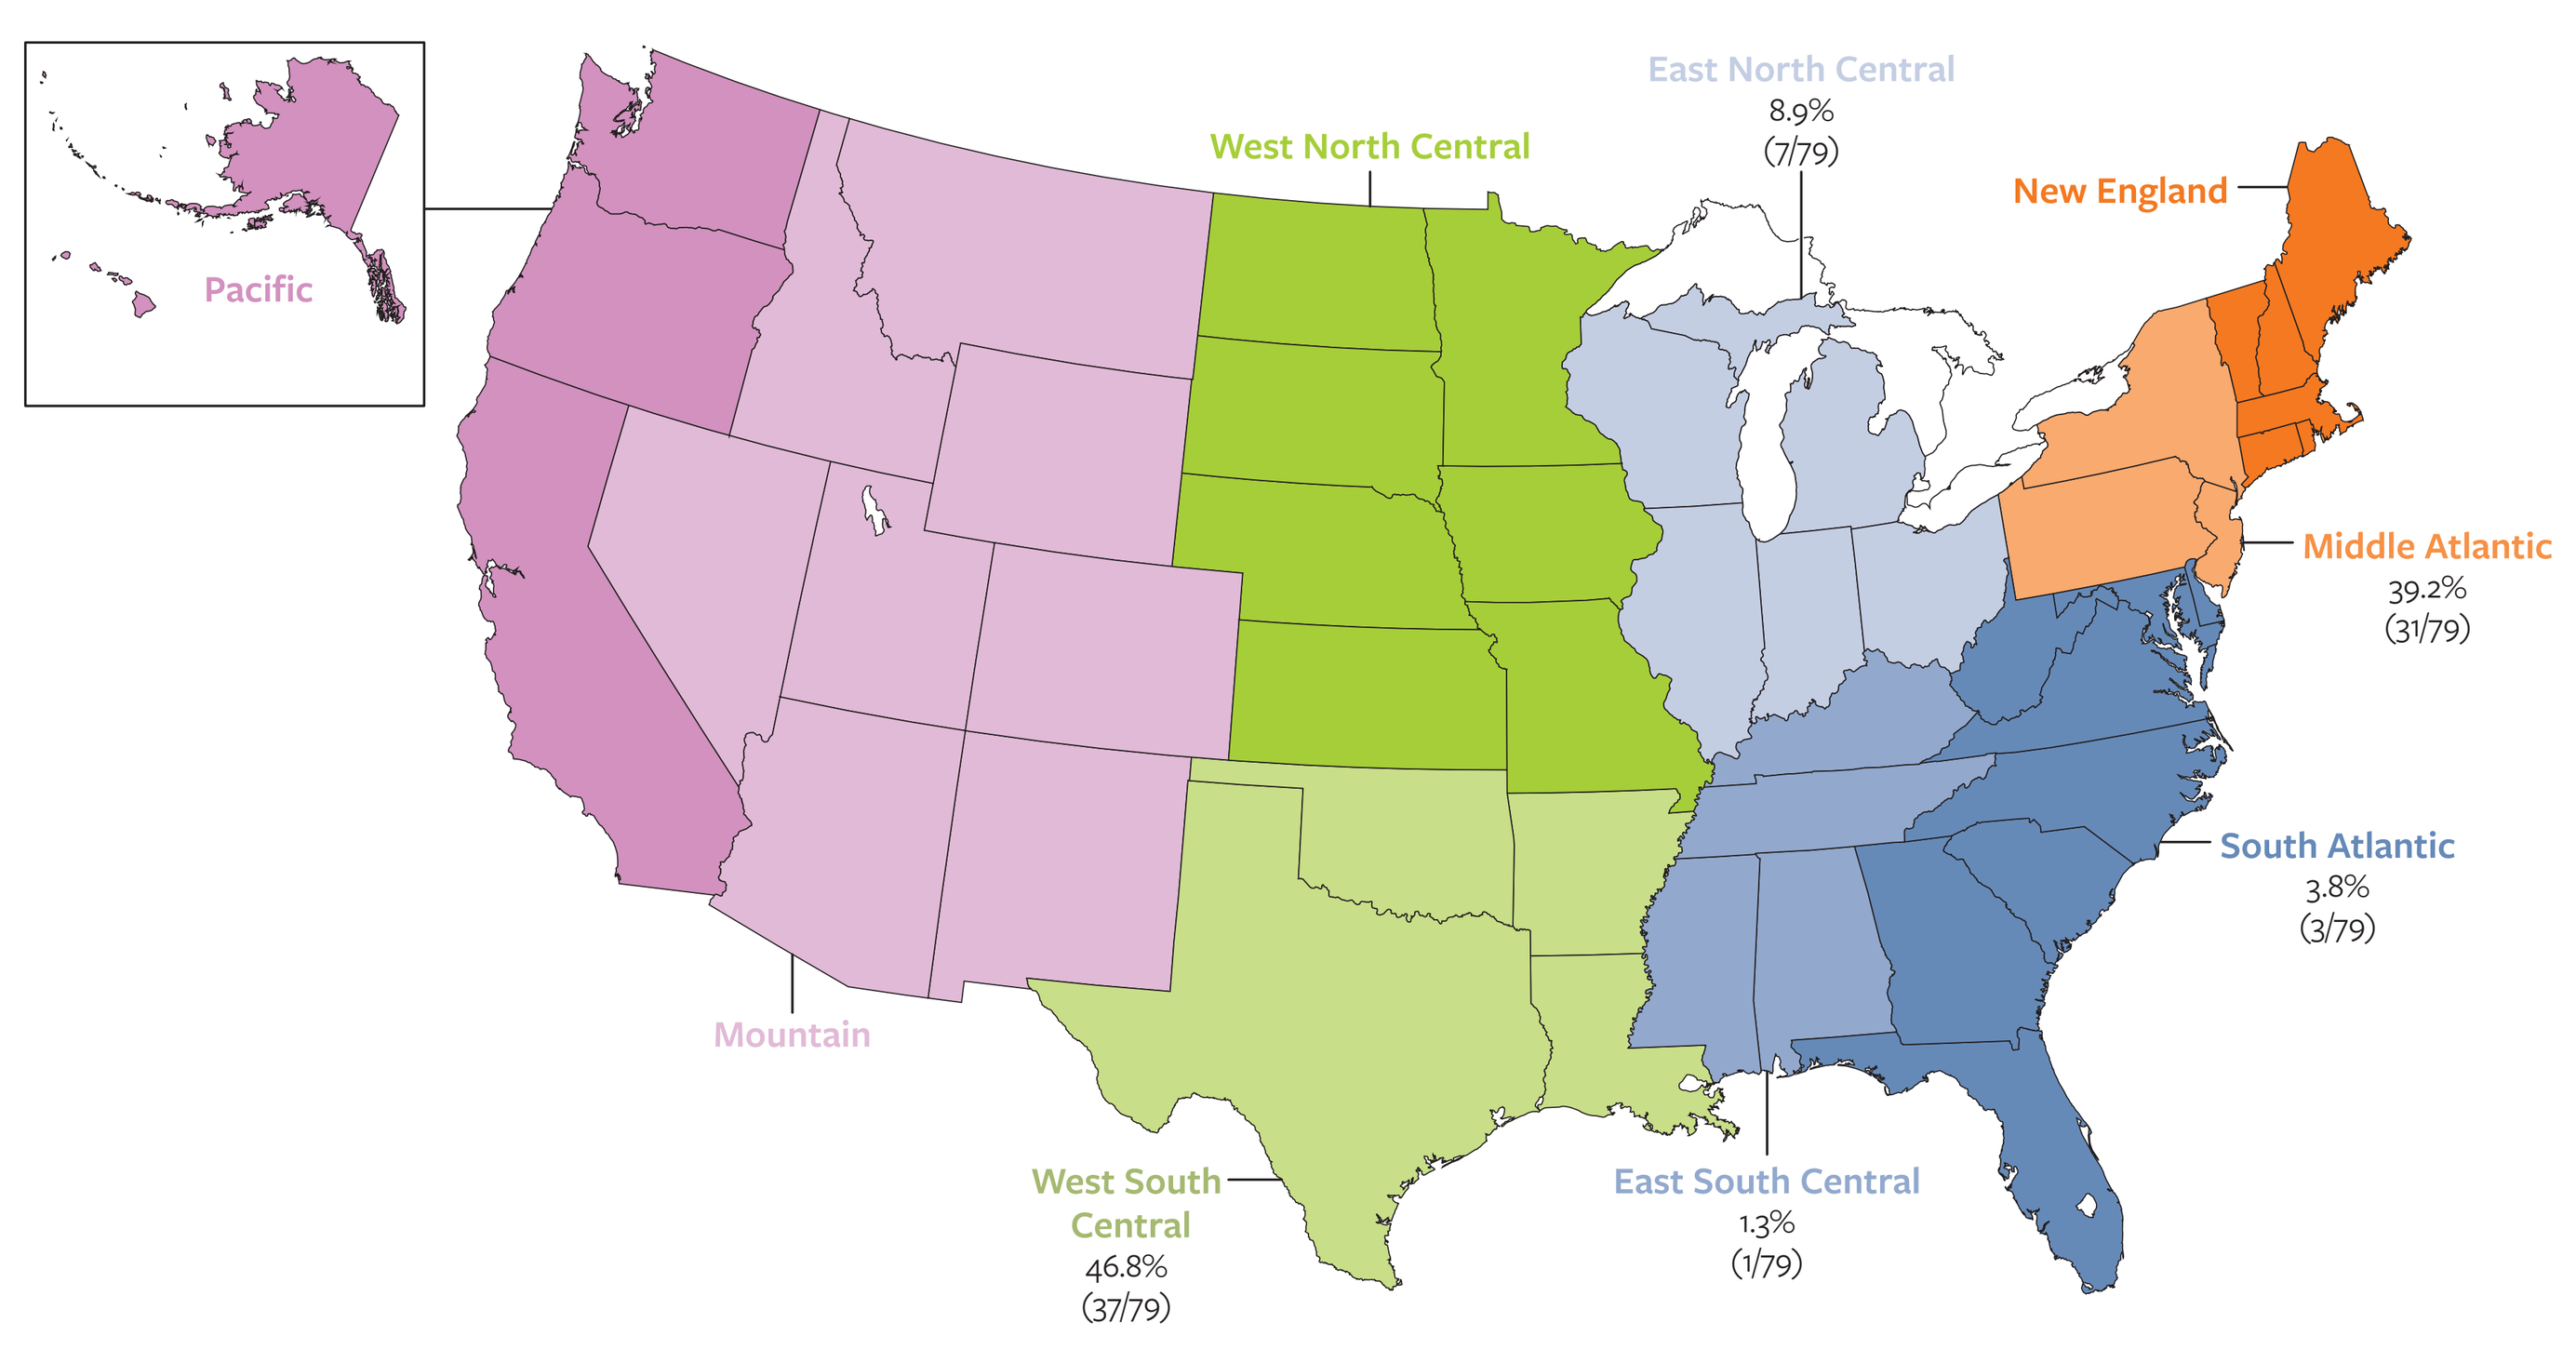

Pacific
Mountain
West North Central
East North Central
8.9%
(7/79)
New England
Middle Atlantic
39.2%
(31/79)
South Atlantic
3.8%
(3/79)
East South Central
1.3%
(1/79)
West South Central
46.8%
(37/79)

Figure 2. Number of *C. auris* isolates collected per year since 2014

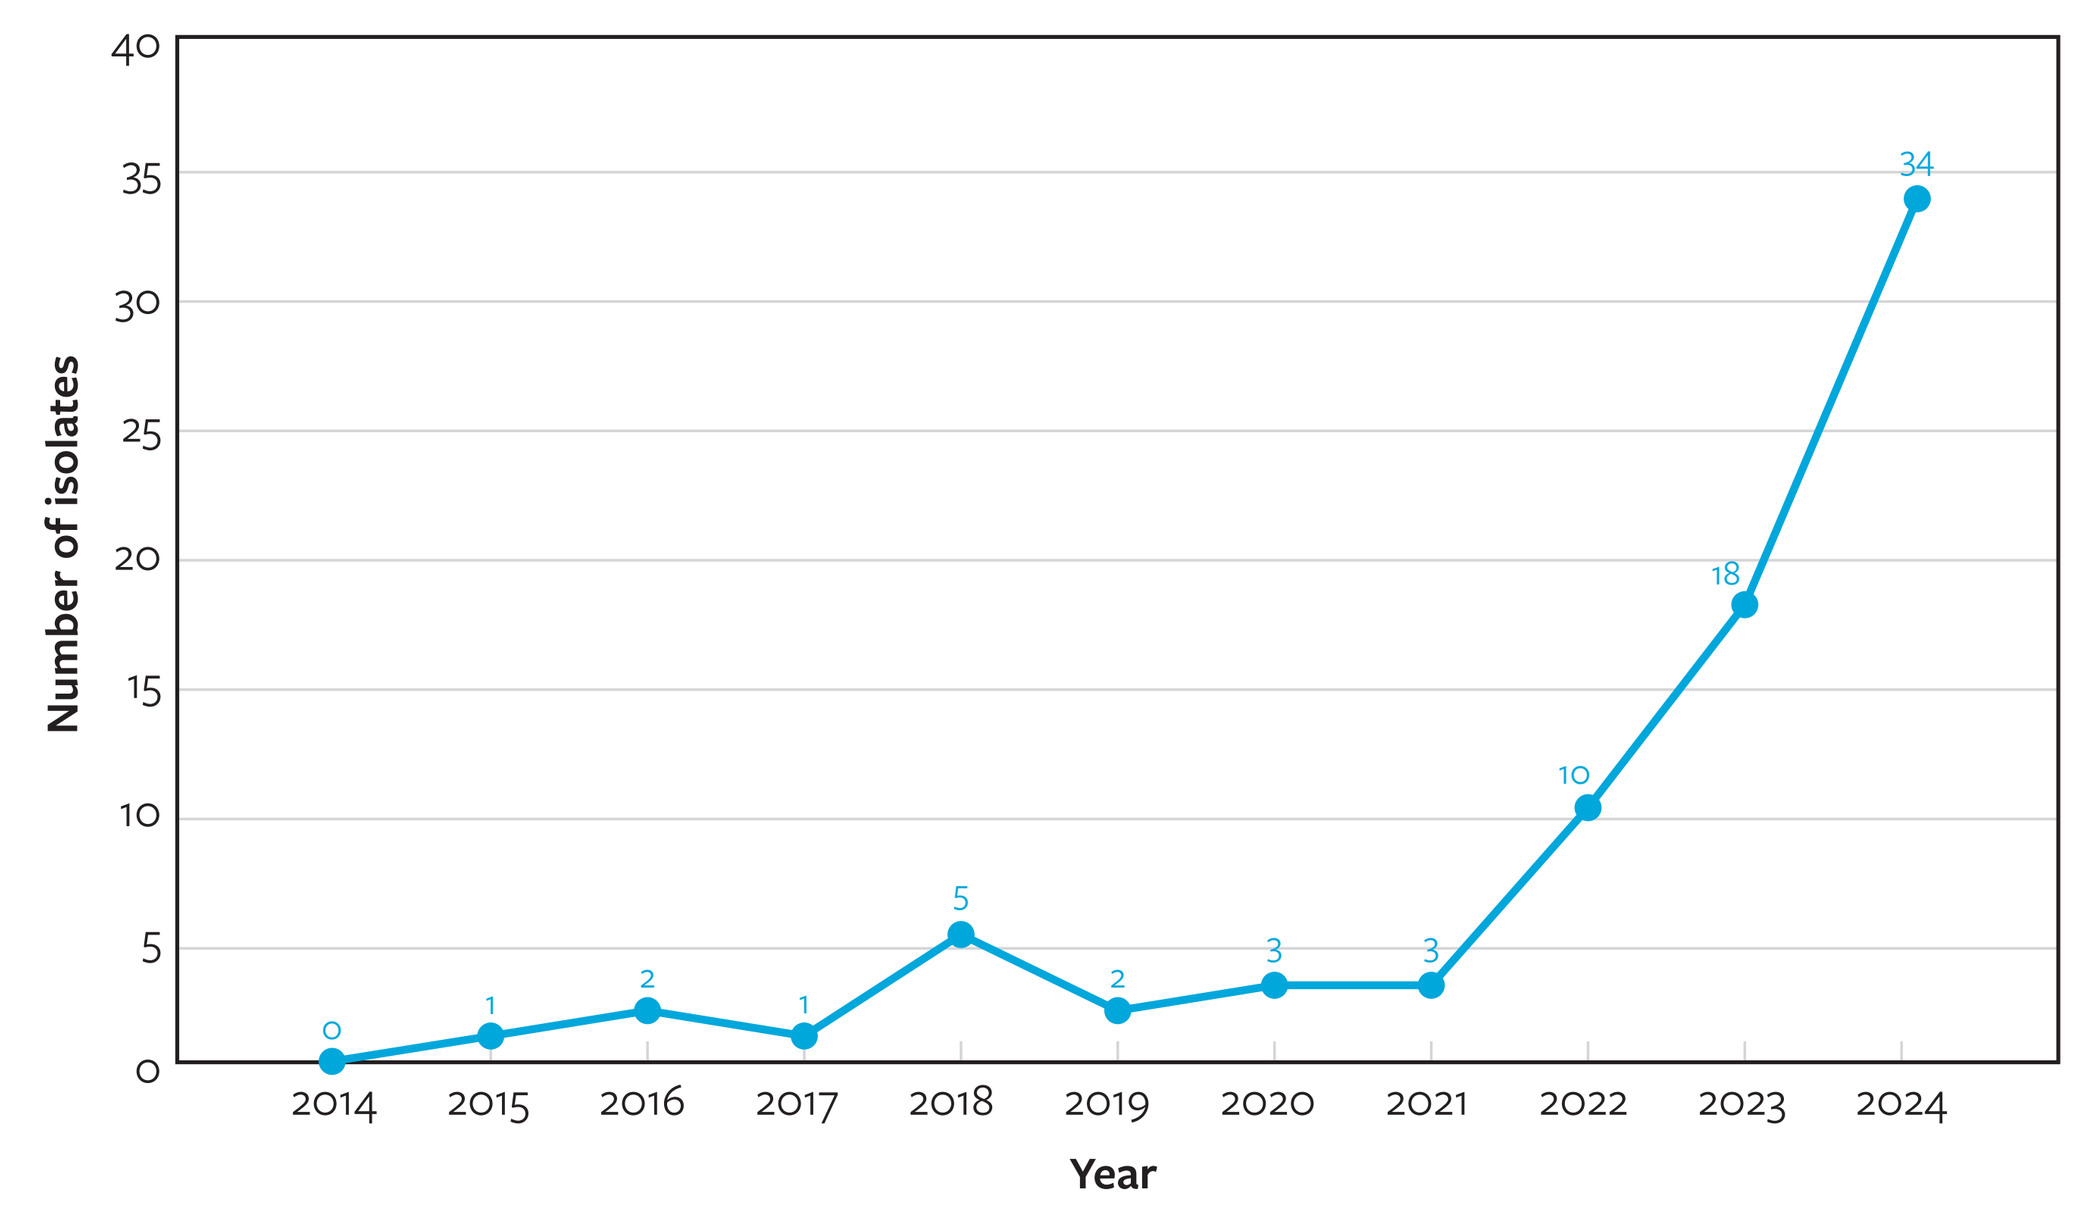

Number of isolates
Year
0
5
10
15
20
25
30
35
40
2014
2015
2016
2017
2018
2019
2020
2021
2022
2023
2024
0
1
2
1
5
2
3
3
10
18
34

Figure 3. Phylogeny of U.S. *C. auris* isolates. Core SNPs were determined for each isolate using Snippy compared to B8441 and used to generate the phylogenetic tree using FastTree's generalized time-reversible model. Clade assignment for each isolate was determined using the core SNPs relative to the reference isolates for each clade. Pairwise comparisons for each protein were performed using the intra-clade reference strain. For each clinical isolate, deidentified with the year of collection-U.S. state postal code-isolate number within the year (e.g., 2024-NJ-16), are (from top to bottom): clade assignment, U.S. census division, CLSI-interpreted rezafungin MIC, CDC-interpreted MIC (echinocandins, amphotericin B, and fluconazole), voriconazole MIC, and alterations within selected proteins previously demonstrated to affect resistance/non-susceptibility to at least one of the tested antifungal agents. Abbreviations: RZF, rezafungin; AND, anidulafungin; CSF, caspofungin; MCF, micafungin; AmB, amphotericin B; FLC, fluconazole; VRC, voriconazole.

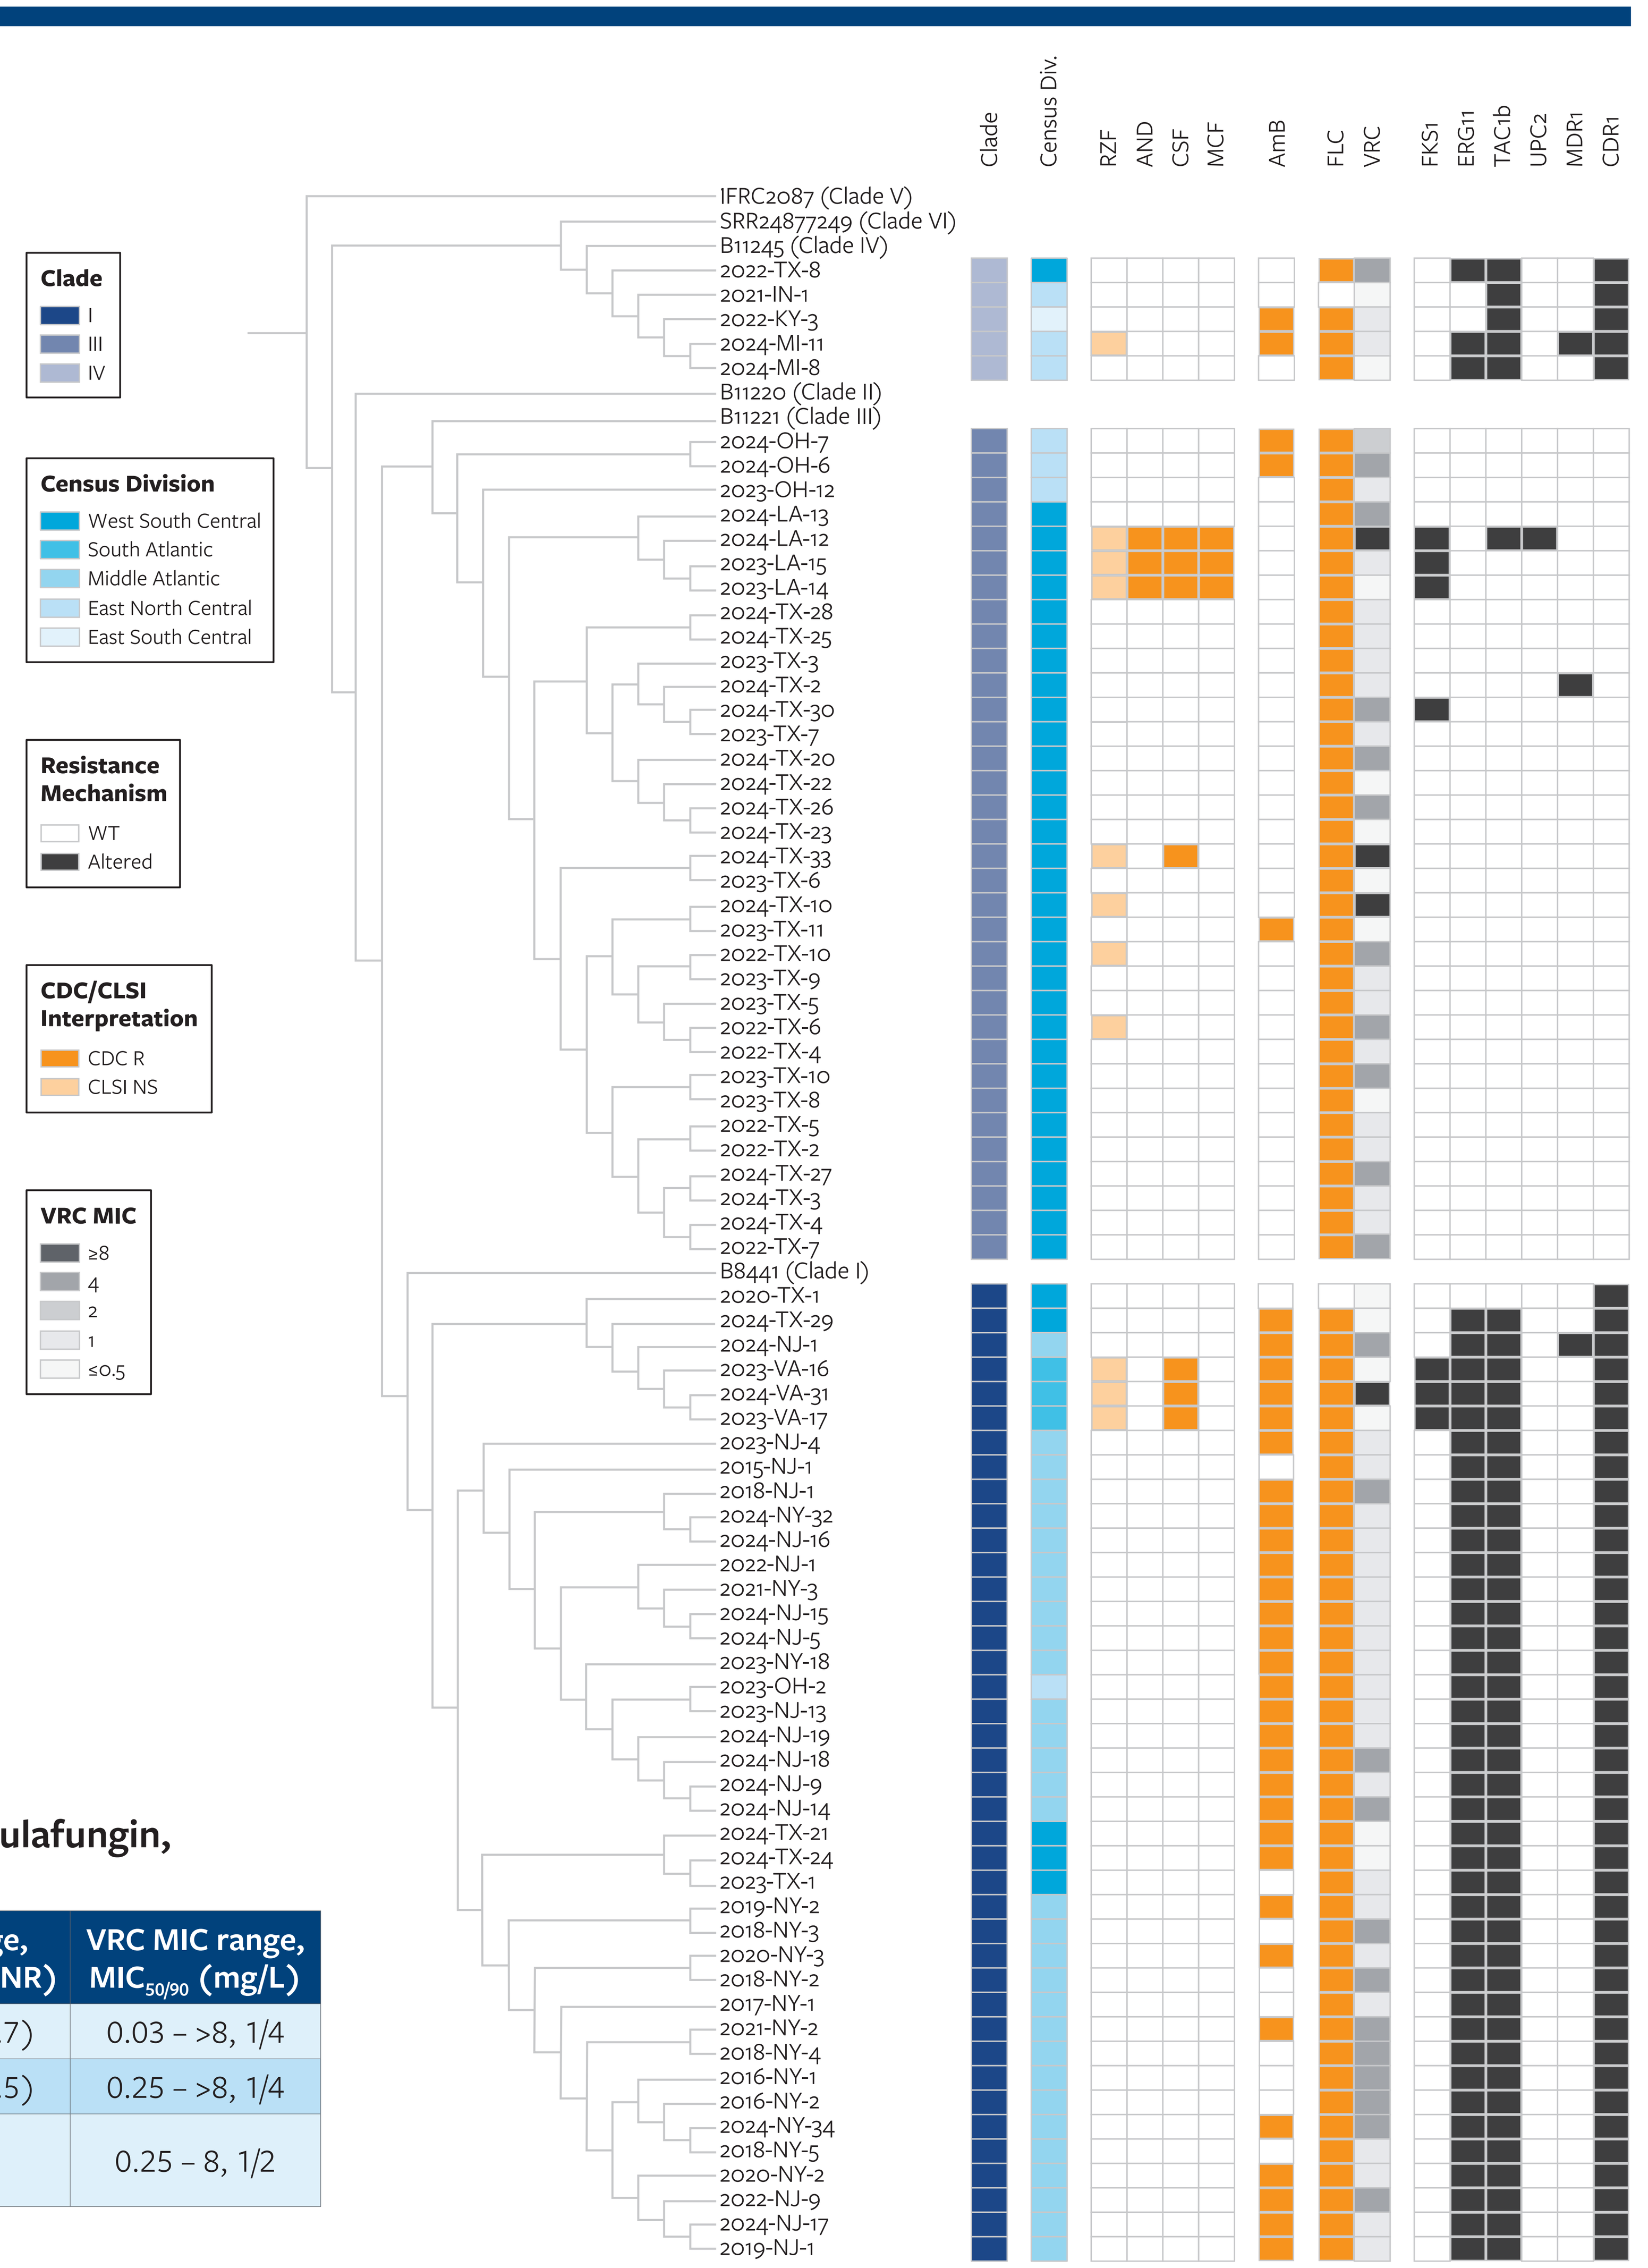

Clade
I
III
IV
Census Division
West South Central
South Atlantic
Middle Atlantic
East North Central
East South Central
Resistance Mechanism
WT
Altered
CDC/CLSI Interpretation
CDC R
CLSI NS
VRC MIC
≥8
4
2
1
≤0.5
Clade
Census Div.
RZF
AND
CSF
MCF
AmB
FLC
VRC
FKS1
ERG11
TAC1b
UPC2
MDR1
CDR1
IFRC2087 (Clade V)
SRR24877249 (Clade VI)
B11245 (Clade IV)
2022-TX-8
2021-IN-1
2022-KY-3
2024-MI-11
2024-MI-8
B11220 (Clade II)
B11221 (Clade III)
2024-OH-7
2024-OH-6
2023-OH-12
2024-LA-13
2024-LA-12
2023-LA-15
2023-LA-14
2024-TX-28
2024-TX-25
2023-TX-3
2024-TX-2
2024-TX-30
2023-TX-7
2024-TX-20
2024-TX-22
2024-TX-26
2024-TX-23
2024-TX-33
2023-TX-6
2024-TX-10
2023-TX-11
2022-TX-10
2023-TX-9
2023-TX-5
2022-TX-6
2022-TX-4
2023-TX-10
2023-TX-8
2022-TX-5
2022-TX-2
2024-TX-27
2024-TX-3
2024-TX-4
2022-TX-7
B8441 (Clade I)
2020-TX-1
2024-TX-29
2024-NJ-1
2023-VA-16
2024-VA-31
2023-VA-17
2023-NJ-4
2015-NJ-1
2018-NJ-1
2024-NY-32
2024-NJ-16
2022-NJ-1
2021-NY-3
2024-NJ-15
2024-NJ-5
2023-NY-18
2023-OH-2
2023-NJ-13
2024-NJ-19
2024-NJ-18
2024-NJ-9
2024-NJ-14
2024-TX-21
2024-TX-24
2023-TX-1
2019-NY-2
2018-NY-3
2020-NY-3
2018-NY-2
2017-NY-1
2021-NY-2
2018-NY-4
2016-NY-1
2016-NY-2
2024-NY-34
2018-NY-5
2020-NY-2
2022-NJ-9
2024-NJ-17
2019-NJ-1

Table 1. MIC range and $MIC_{50/90}$ of *C. auris* isolates with susceptible (rezafungin, by CLSI) or non-resistant (NR) rates (micafungin, anidulafungin, caspofungin, fluconazole, amphotericin B) including in resistant subsets of isolates

Organism group (n)	RZF MIC range, $MIC_{50/90}$ mg/L (%S)	CSF MIC range, $MIC_{50/90}$ mg/L (%NR)	AND MIC range, $MIC_{50/90}$ mg/L (%NR)	MCF MIC range, $MIC_{50/90}$ mg/L (%NR)	FLC MIC range, $MIC_{50/90}$ mg/L (%NR)	AmB MIC range, $MIC_{50/90}$ mg/L (%NR)	VRC MIC range, $MIC_{50/90}$ (mg/L)
All US *C. auris* (79)	0.03 – >4, 0.5/1 (86.1)	0.03 – >4, 0.12/1 (91.1)	0.06 – >4, 0.5/1 (96.2)	0.06 – >4, 0.25/0.5 (96.2)	4 – >128, >128/>128 (2.5)	0.5 – 2, 1/2 (55.7)	0.03 – >8, 1/4
FLC-R *C. auris* (77)	0.03 – >4, 0.5/1 (85.7)	0.03 – >4, 0.12/1 (90.9)	0.06 – >4, 0.5/1 (96.1)	0.06 – >4, 0.25/0.5 (96.1)	64 – >128, >128/>128 (0)	0.5 – 2, 1/2 (54.5)	0.25 – >8, 1/4
AmB-R and FLC-R *C. auris* (35)	0.03 – 2, 0.25/1 (88.6)	0.03 – >4, 0.12/0.5 (91.4)	0.06 – 2, 0.5/0.5 (100.0)	0.06 – 1, 0.25/0.5 (100.0)	64 – >128, >128/>128 (0)	2, 2/2 (0)	0.25 – 8, 1/2

n, number; RZF, rezafungin; MIC, minimum inhibitory concentration; S, susceptible; CSF, caspofungin; NR, non-resistant; AND, anidulafungin; MCF, micafungin; FLC, fluconazole; AmB, amphotericin B; VRC, voriconazole

References

Pappas, P.G., Kauffman, C.A., Andes, D.R., Clancy, C.J., Marr, K.A., Ostrosky-Zeichner, L., et al., *Clinical Practice Guideline for the Management of Candidiasis: 2016 Update by the Infectious Diseases Society of America.* Clin Infect Dis, 2016. 62(4): e1-50.

CLSI, *M27 M44S Ed3. Performance standards for antifungal susceptibility testing of yeasts*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M27 Ed4. Reference method for broth dilution antifungal susceptibility testing of yeasts*. 2017, Clinical and Laboratory Standards Institute: Wayne, PA.

Antifungal Susceptibility Testing for *C. auris*. https://www.cdc.gov/candida-auris/hcp/laboratories/antifungal-susceptibility-testing.html.

Joshi NA, Fass JN. (2011). Sickle: A sliding-window, adaptive, quality-based trimming tool for FastQ files (Version 1.33). Available at https://github.com/najoshi/sickle.

Seemann T (2015) snippy: fast bacterial variant calling from NGS reads (Version 4.6.0 Available at https://github.com/tseemann/snippy).

Price, Morgan N., Paramvir S. Dehal, and Adam P. Arkin. "FastTree: computing large minimum evolution trees with profiles instead of a distance matrix." *Molecular biology and evolution* 26.7 (2009): 1641–1650.

Croucher, Nicholas J., et al. "Rapid phylogenetic analysis of large samples of recombinant bacterial whole genome sequences using Gubbins." Nucleic acids research 43.3 (2015): e15-e15 (Version 2.3.4).

Contact

element

Marisa Winkler, MD, PhD
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: marisa.winkler@element.com

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2025_24-MEL-03_P1_C_auris_US.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia