IDWEEK 2025 | Poster # P1318

Antimicrobial Activity of Aztreonam-Avibactam and Comparator Agents against *Stenotrophomonas maltophilia* Isolates from Patients Hospitalized with Pneumonia in Europe, Latin America, and Asia (2020–2024)

Helio S. Sader[1], Mariana Castanheira[1], Gregory Stone[2], Katherine Perez[2], Marisa L. Winkler[1], Rodrigo E. Mendes[1]
[1] Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA; [2] Pfizer Inc., New York, New York, USA

Introduction

- Aztreonam-avibactam (ATM-AVI) was recently approved by the US FDA for treatment of complicated IAI and by the EMA in the European Union for treatment of adults with complicated IAI, complicated urinary tract infection (cUTI), hospital-acquired pneumonia, including ventilator-associated pneumonia, and infections due to aerobic Gram-negative bacteria in adults with limited treatment options.
- Antimicrobial treatment selection for *S. maltophilia* infection is hindered by the number of resistance mechanisms carried by *S. maltophilia*, including intrinsic L1 metallo-β-lactamase (MBL) and L2 serine β-lactamase that inactivate most β-lactams.
- Moreover, *S. maltophilia* displays intrinsic resistance to aminoglycosides and can express multidrug efflux transporters that decrease the activity of fluoroquinolones, tetracyclines, and trimethoprim-sulfamethoxazole.
- We evaluated the activity of ATM-AVI against *S. maltophilia* isolated from patients with pneumonia.

Methods

- A total of 784 clinical isolates were collected as part of the SENTRY Antimicrobial Surveillance Program from patients with pneumonia in 60 medical centers located in:
 - Western Europe (W-EU): 457 isolates from 25 centers in 10 countries
 - Eastern Europe (E-EU): 179 isolates from 16 centers in 9 countries
 - The Asia-Pacific region (APAC): 89 isolates from 10 centers in 5 countries
 - Latin America (LATAM): 59 isolates from 9 centers in 6 countries
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of pneumonia were included in the study.
- Isolates were susceptibility tested by CLSI M07 broth microdilution method.
- The ATM-AVI susceptible breakpoint of ≤4 mg/L, which was established by the US FDA and EMA for Enterobacterales, was applied for comparison.
- CLSI breakpoints were applied to comparators when available.

Results

- ATM-AVI demonstrated potent activity against isolates from all geographic regions with overall $MIC_{50/90}$ of 4/4 mg/L and 94.0% of isolates inhibited at ≤4 mg/L (Table 1 and Figure 1).
- The percentage inhibited at ≤4 mg/L ranged from 91.5% in LATAM to 94.4% in E-EU and APAC (Table 1 and Figure 1).
- ATM-AVI activity remained stable during the study period; overall percentage inhibited at ≤4 mg/L varied from 91.0% in 2023 to 97.4% in 2024 (Table 1 and Figure 2).
- Susceptibility to trimethoprim-sulfamethoxazole (TMP-SMX; $MIC_{50/90}$, ≤0.12/0.5 mg/L) ranged from 91.5% in LATAM to 96.5% in W-EU (95.4% overall; Figure 1).
- Susceptibility to minocycline ($MIC_{50/90}$, 0.5/1 mg/L) varied from 90.2% in LATAM to 95.0% in E-EU (94.5% overall; Figure 1).
- Levofloxacin inhibited 84.8% of isolates at the old (2024) CLSI susceptible breakpoint of ≤2 mg/L, ranging from 76.3% in LATAM to 88.8% in APAC (Figure 1).
- Ceftazidime ($MIC_{50/90}$, >32/>32 mg/L; 13.5% inhibited at ≤8 mg/L) and colistin ($MIC_{50/90}$, 4/>8 mg/L; 38.8% inhibited at ≤2 mg/L) showed limited activity (data not shown).
- ATM-AVI retained potent activity against isolates not susceptible to TMP-SMX (83.3% susceptible), minocycline (82.9% susceptible), and/or levofloxacin (87.4% susceptible; Figure 3).

Conclusions

- Our results indicated that ATM-AVI may represent a valuable option to treat *S. maltophilia* infections, addressing a major unmet medical need.
- *S. maltophilia* susceptibility rates for comparator agents should be interpreted with caution since breakpoints were established in the 1980s; i.e. before the knowledge of PK/PD parameters that are currently used to establish breakpoints.

Table 1. Aztreonam-avibactam (ATM-AVI) activity stratified by region and year

Region (no. isolates)	ATM-AVI MIC_{50}/MIC_{90} (% inhibited at ≤4 mg/L) by year (no. of isolates)					
	2020 (116)	2021 (112)	2022 (161)	2023 (167)	2024 (228)	All (784)
All (784)	4/4 (93.1)	4/4 (94.6)	2/4 (92.5)	4/4 (91.0)	4/4 (97.4)	4/4 (94.0)
W-EU (457)	4/4 (95.0)	4/4 (95.4)	2/4 (92.1)	4/8 (89.2)	4/4 (97.9)	4/4 (94.1)
E-EU (179)	4/8 (88.2)	4/4 (90.5)	4/4 (97.4)	4/4 (93.9)	2/4 (98.1)	4/4 (94.4)
APAC (89)	4/4 (92.9)	4/4 (100.0)	4/8 (87.0)	2/4 (95.8)	4/4 (100.0)	4/4 (94.4)
LATAM (59)	2/— (100.0)	2/4 (92.9)	4/4 (90.9)	2/— (87.5)	4/8 (88.9)	4/4 (91.5)

Abbreviations: W-EU, Western Europe; E-EU, Eastern Europe; APAC, Asia-Pacific region; LATAM, Latin America.

Figure 1. Antimicrobial activity of aztreonam-avibactam (ATM-AVI) and comparators against *S. maltophilia* isolates from patients with pneumonia stratified by region

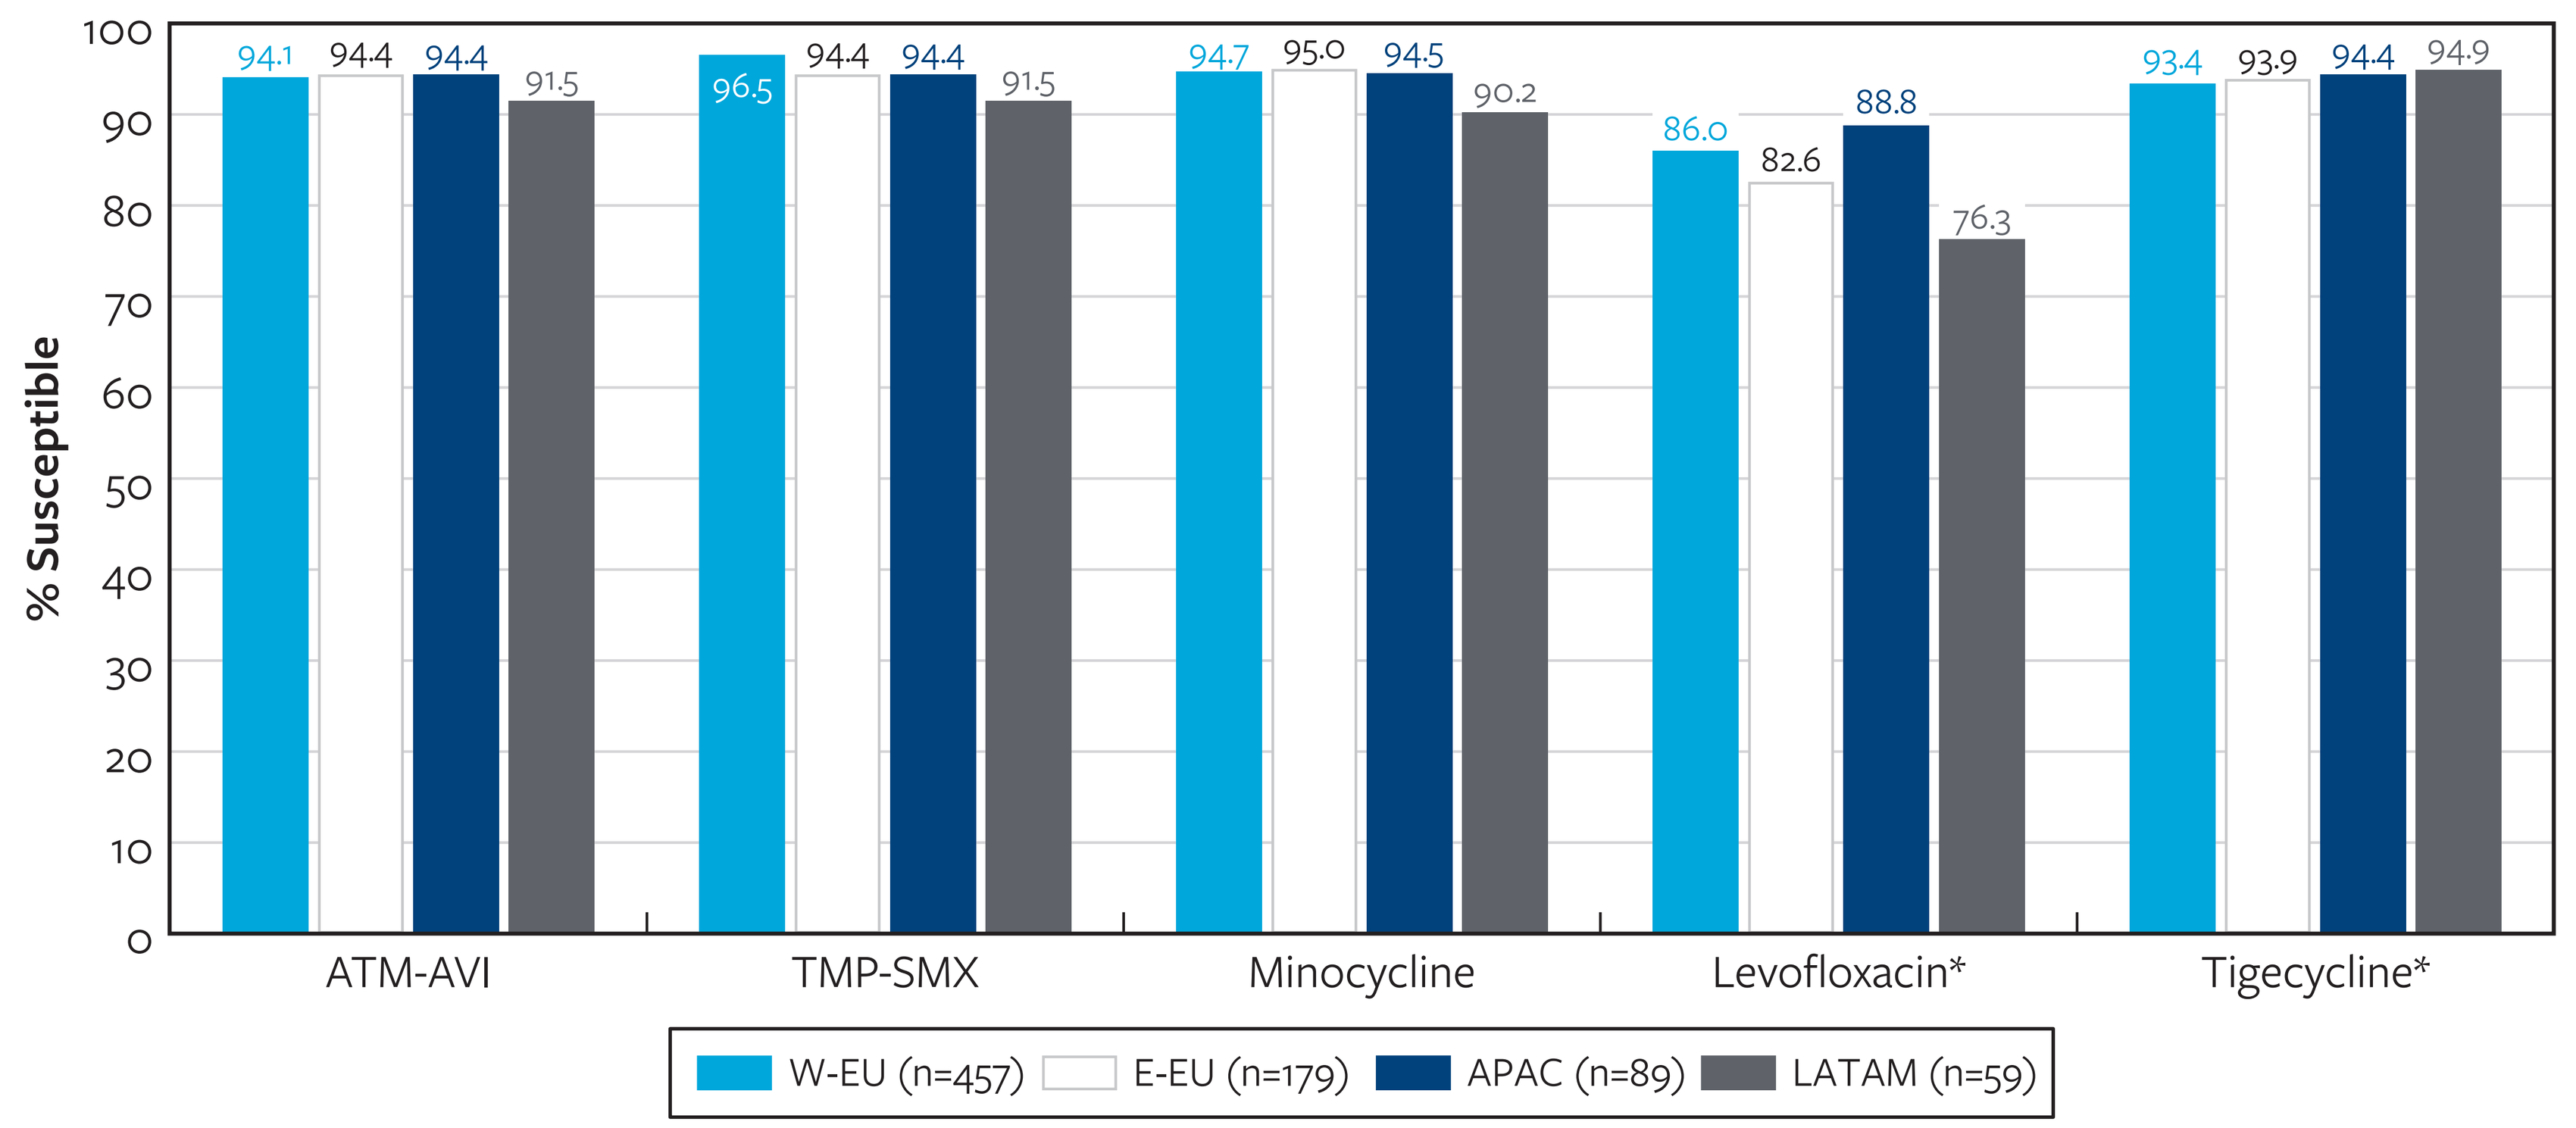
% Susceptible
100
90
80
70
60
50
40
30
20
10
0
94.1
94.4
94.4
91.5
96.5
94.4
94.4
91.5
94.7
95.0
94.5
90.2
86.0
82.6
88.8
76.3
93.4
93.9
94.4
94.9
ATM-AVI
TMP-SMX
Minocycline
Levofloxacin*
Tigecycline*
W-EU (n=457)
E-EU (n=179)
APAC (n=89)
LATAM (n=59)

* % inhibited at ≤2 mg/L.
Abbreviations: ATM-AVI, aztreonam-avibactam; TMP-SMX, trimethoprim-sulfamethoxazole; W-EU, Western Europe; E-EU, Eastern Europe; APAC, Asia-Pacific region; LATAM; Latin America.

Figure 2. Antimicrobial activity of aztreonam-avibactam (ATM-AVI) and comparators against *S. maltophilia* isolates from patients with pneumonia stratified by year

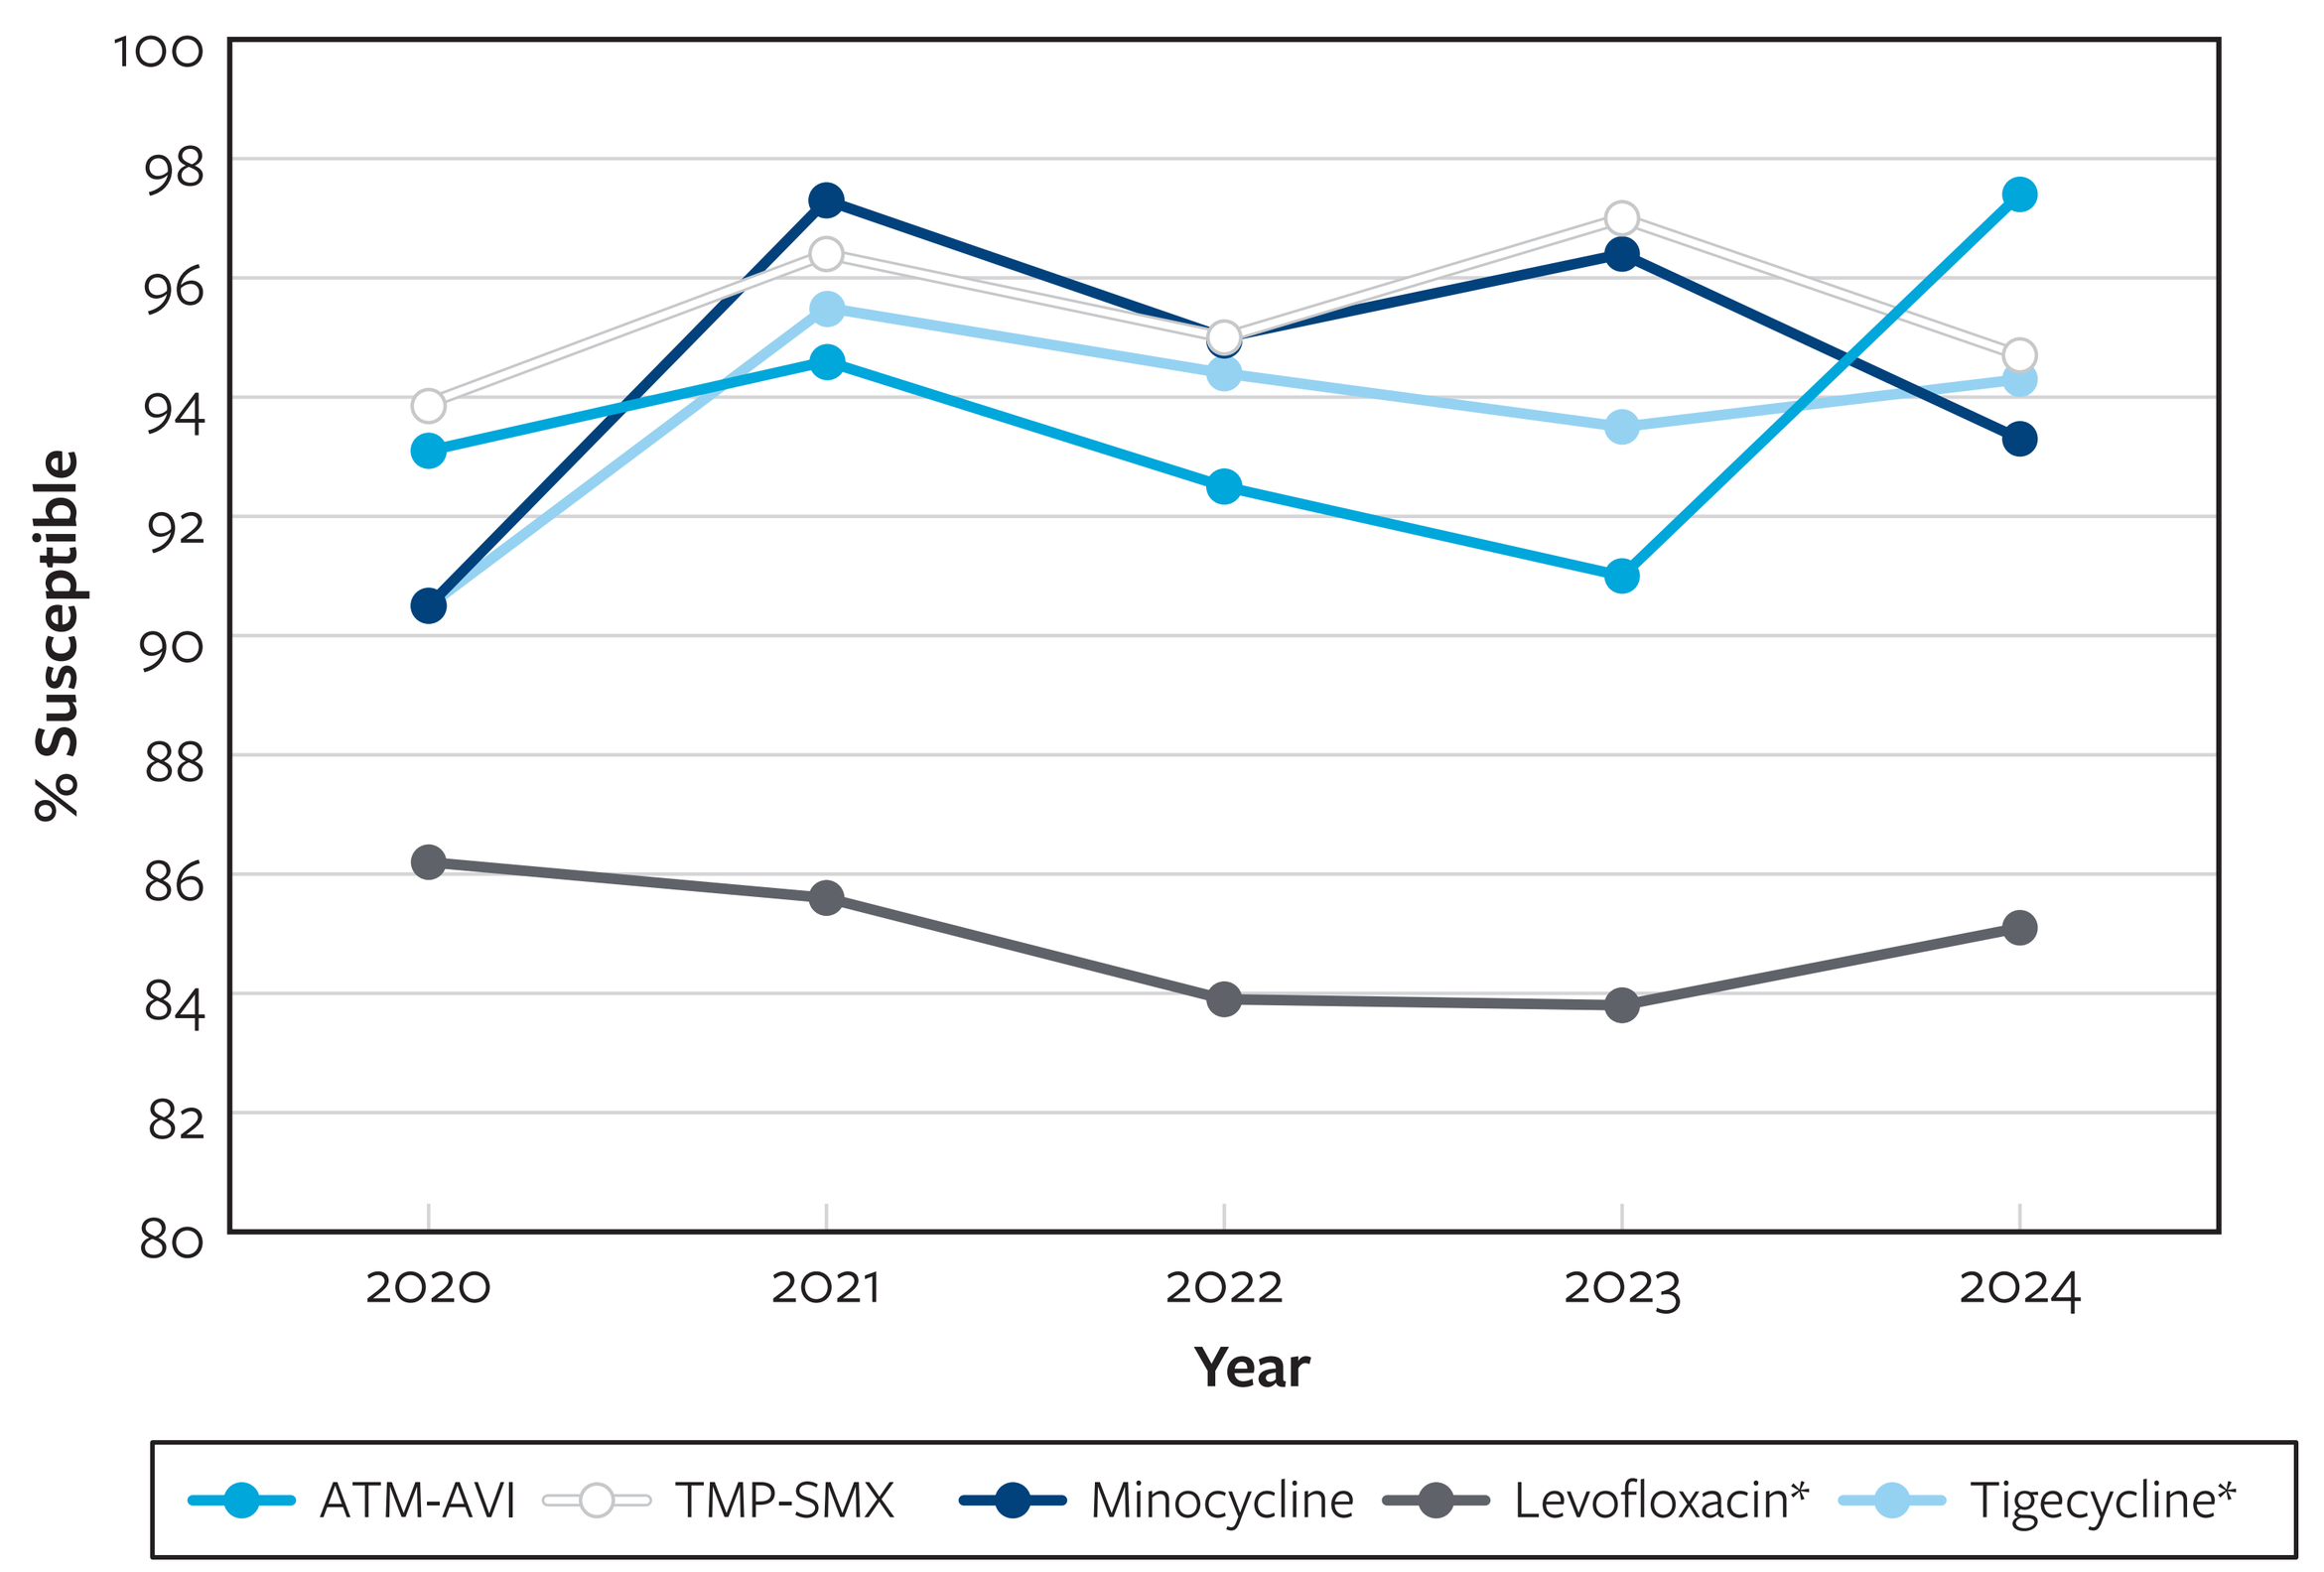
% Susceptible
100
98
96
94
92
90
88
86
84
82
80
2020
2021
2022
2023
2024
Year
ATM-AVI
TMP-SMX
Minocycline
Levofloxacin*
Tigecycline*

* % inhibited at ≤2 mg/L.
Abbreviations: ATM-AVI, aztreonam-avibactam; TMP-SMX, trimethoprim-sulfamethoxazole.

Figure 3. Antimicrobial activity of aztreonam-avibactam (ATM-AVI) and comparators against resistant subsets

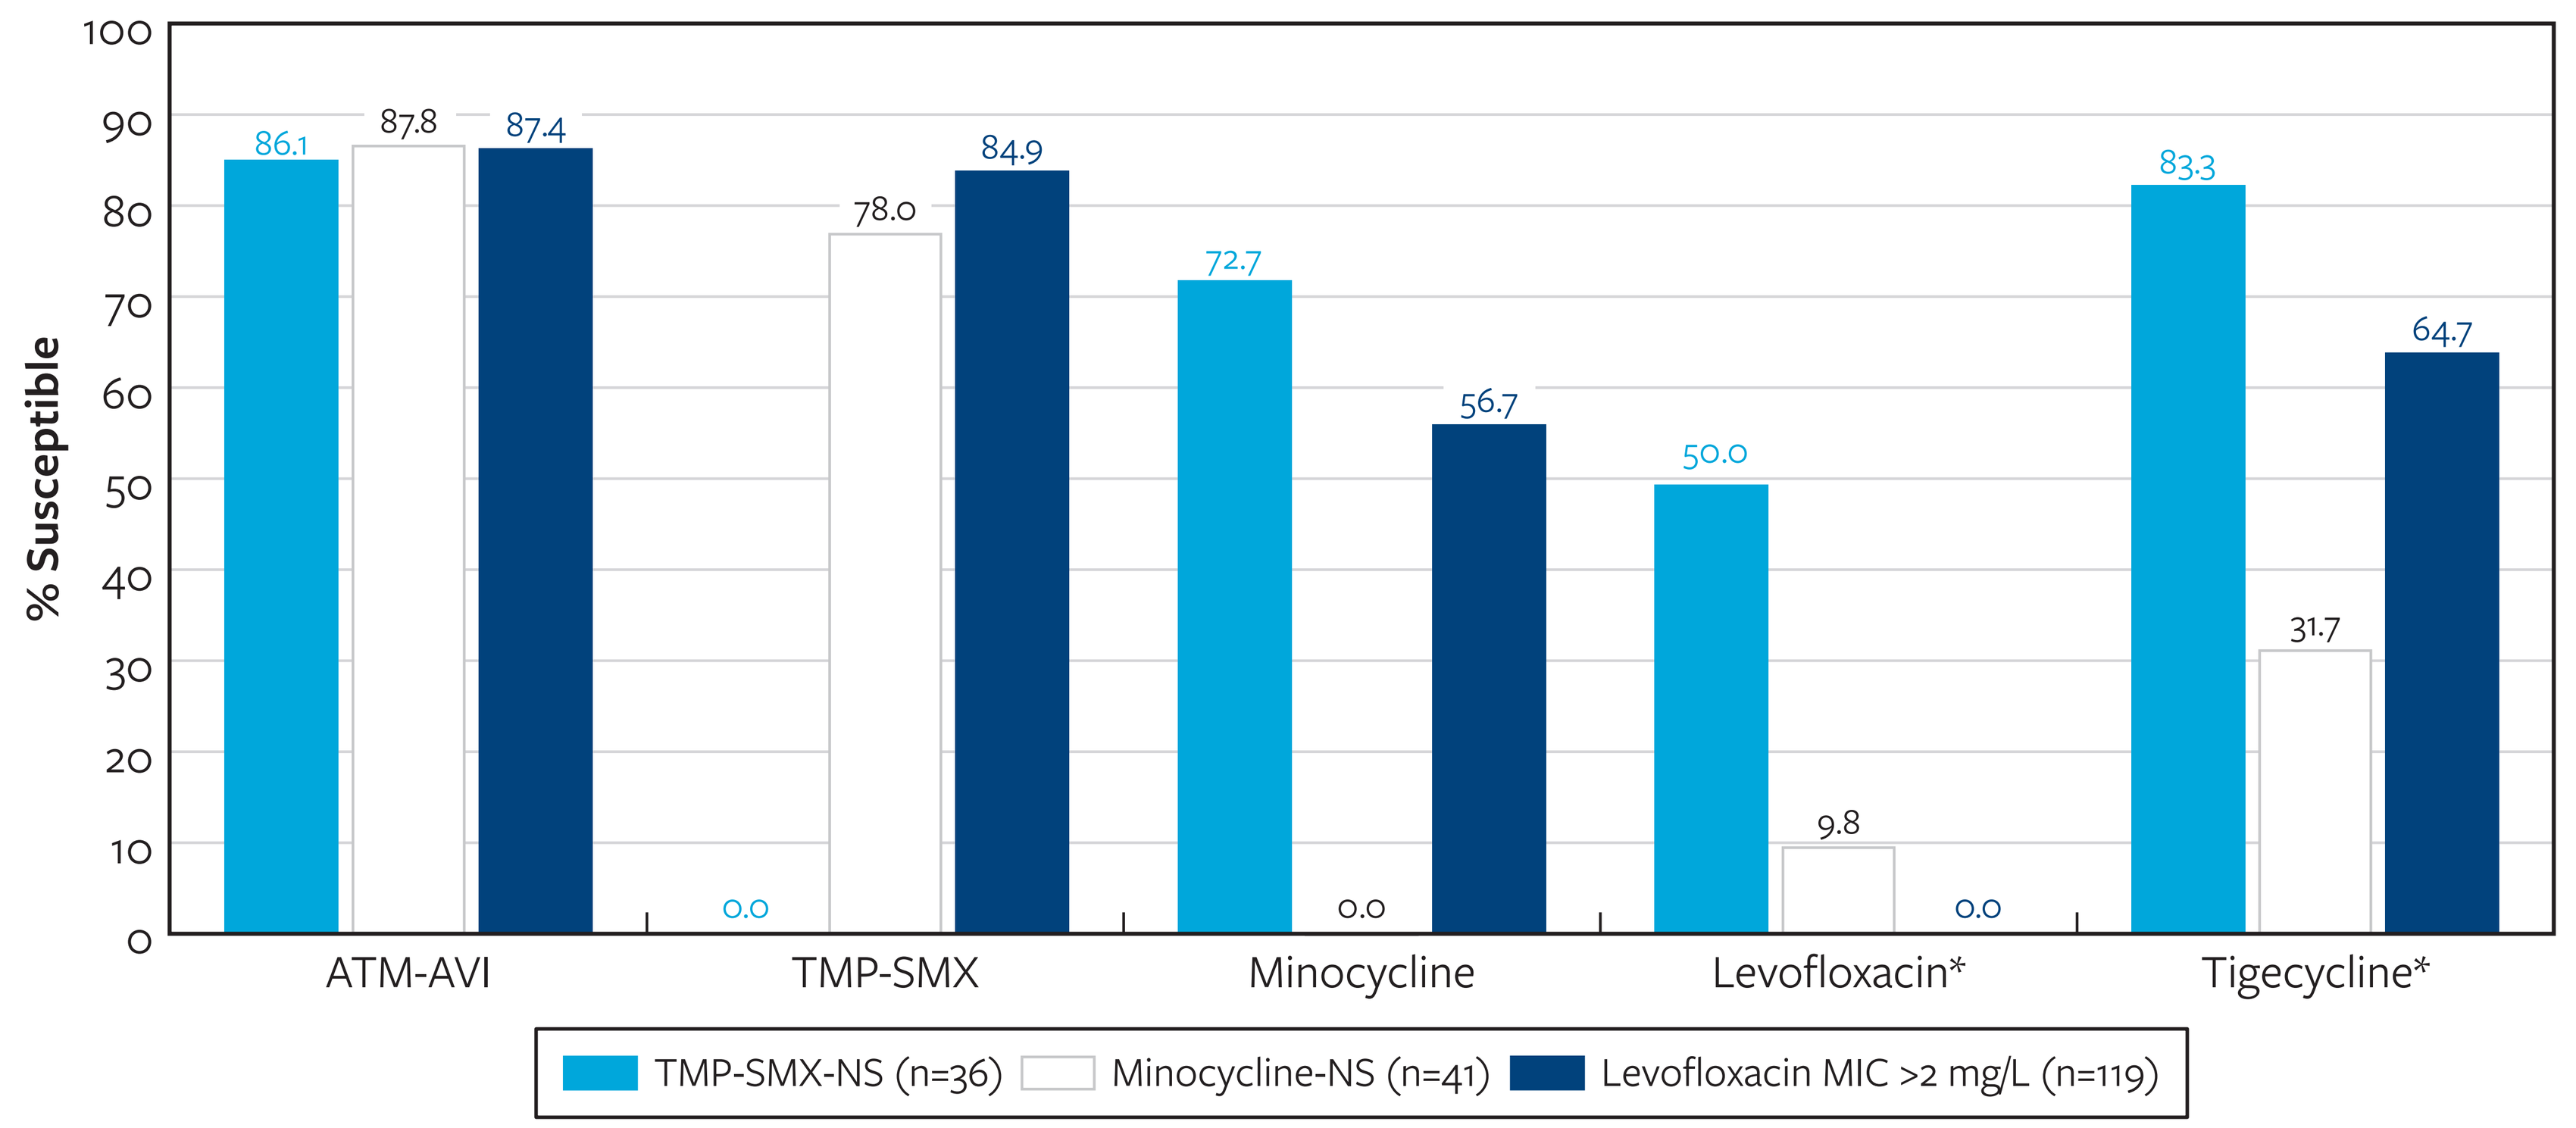
% Susceptible
100
90
80
70
60
50
40
30
20
10
0
86.1
87.8
87.4
0.0
78.0
84.9
72.7
0.0
56.7
50.0
9.8
0.0
83.3
31.7
64.7
ATM-AVI
TMP-SMX
Minocycline
Levofloxacin*
Tigecycline*
TMP-SMX-NS (n=36)
Minocycline-NS (n=41)
Levofloxacin MIC >2 mg/L (n=119)

* % inhibited at ≤2 mg/L.
Abbreviations: ATM-AVI, aztreonam-avibactam; TMP-SMX, trimethoprim-sulfamethoxazole; NS, not susceptible.

Acknowledgments

This study at JMI Laboratories was supported by Pfizer Inc. (New York, NY). JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by Pfizer Inc.

References

Barrasa H, Morán MA, Fernández-Ciriza L, *et al.* Optimizing antibiotic therapy for *Stenotrophomonas maltophilia* infections in critically ill patients: A pharmacokinetic/pharmacodynamic approach. *Antibiotics* (*Basel*) 2024;13(6):553.

CLSI. 2025. M100Ed35. Performance standards for antimicrobial susceptibility testing: 35th informational supplement. Clinical and Laboratory Standards Institute.

Sader HS, Mendes RE, Doyle TB, Winkler ML, Castanheira M. Antimicrobial susceptibility of clinical isolates of *Stenotrophomonas maltophilia* from Europe, Asia, and Latin America (2018–2023). *Int J Infect Dis* 2025;153:107803.

Sader HS, Castanheira M, Arends SJR, Doyle TB. Antimicrobial susceptibility of *Stenotrophomonas maltophilia* from United States medical centers (2019–2023). *Antimicrob Agents Chemother* 2025 2;69(4):e0012425.

Tamma PD, Heil EL, Justo JA, *et al.* Infectious Diseases Society of America 2024 guidance on the treatment of antimicrobial-resistant Gram-negative infections. *Clin Infect Dis* 2024 ciae403.

Contact

element

SCAN ME

Pfizer.com

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2025_24-PZR-01_P1_XM_Pneumo.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia