IDWEEK 2025 | Poster #P1317

Antimicrobial Activities of Aztreonam-Avibactam and Comparator Agents Tested against Enterobacterales from European Hospitals Analysed by Infection Type (2022–2024)

Helio S. Sader[1], John H. Kimbrough[1], Gregory Stone[2], Katherine Perez[2], Rodrigo E. Mendes[1], Mariana Castanheira[1]
[1] Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA; [2] Pfizer Inc., New York, New York, USA

Introduction

- Aztreonam-avibactam (ATM-AVI) was recently approved by the US FDA (February 2025) for treatment of complicated intra-abdominal infection (IAI) and by the EMA in the European Union (April 2024) for treatment of adults with complicated IAI, complicated urinary tract infection (cUTI), hospital-acquired pneumonia, including ventilator-associated pneumonia, and infections due to aerobic Gram-negative bacteria in adults with limited treatment options.
- Aztreonam-avibactam (ATM-AVI) has demonstrated potent activity against multidrug-resistant (MDR) Enterobacterales worldwide, including metallo-β-lactamase (MBL) producers.
- We evaluated the activity of ATM-AVI and comparators against Enterobacterales causing infections in European medical centers.

Methods

- A total of 18,650 isolates were consecutively collected from 40 medical centers located in:
 - Western Europe (W-EU): 13,089 isolates from 25 centers in 10 countries including Belgium, France, Germany, Ireland, Italy, Portugal, Spain, Sweden, Switzerland, and the United Kingdom.
 - Eastern Europe and Mediterranean Region (E-EU): 5,561 isolates from 15 centers in 9 countries including the Czech Republic, Greece, Hungary, Israel, Poland, Romania, Slovakia, Slovenia, and Turkey.
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of infection were included in the study.
- Isolates were susceptibility tested by CLSI M07 broth microdilution method and susceptibility rates were based on EUCAST breakpoint criteria when available.
- The antimicrobial susceptibility and frequency of key resistance phenotypes were assessed and stratified by infection type as follows:
 - Bloodstream (BSI): 5,657 isolates; 30.3%
 - Pneumonia: 4,169 isolates; 22.4%
 - Skin and skin structure infection (SSSI): 1,379 isolates; 7.4%
 - Urinary tract infection (UTI): 5,833 isolates; 31.3%
 - Intra-abdominal infection (IAI): 1,612 isolates; 8.6%
- The ATM-AVI susceptible breakpoint of ≤4 mg/L, which was established by the US FDA and EMA for Enterobacterales, was applied.
- Carbapenem-resistant Enterobacterales (CRE) was defined as an MIC ≥4 mg/L for meropenem and/or imipenem and difficult-to-treat resistant (DTR) was defined as a CRE resistant to fluoroquinolone.
- All CRE isolates were screened for carbapenemases (CBase) by whole genome sequencing.

Results

- ATM-AVI was active against 99.9–100.0% of W-EU isolates ($MIC_{50/90}$, ≤0.03/0.12 mg/L) and 99.7–100.0% of E-EU isolates ($MIC_{50/90}$, ≤0.03/0.25 mg/L; Table 1) across all infection types.
- Susceptibility rates for comparator agents were similar among infection types in W-EU, whereas in E-EU susceptibility rates were markedly lower for isolates from pneumonia compared to other infection types (Table 1).
- Resistance to comparator agents as well as the frequencies of CRE, MDR, extensive-drug resistant (XDR), and DTR isolates were markedly higher among isolates from E-EU compared to W-EU for all infection types (Table 1 and Figures 1 and 2).
- ATM-AVI was active against 98.9% of CRE from W-EU and 99.8% from E-EU (Figure 3; $MIC_{50/90}$, 0.25/0.5 mg/L in both regions).
- All comparators exhibited limited activity against CRE, including ceftazidime-avibactam (CAZ-AVI; 66.1% susceptible [S] in W-EU and 49.3% S in E-EU) and meropenem-vaborbactam (MEM-VAB; 76.3% S in W-EU and 30.0% S in E-EU; Figure 3).
- A CBase was identified in 166 CREs from W-EU (93.8% of CREs) and 494 CREs from E-EU (96.3% of CREs; Figure 4).
- The frequencies of CBase types varied markedly by EU region; the most common CBase types were KPC (53.7% of CREs) and NDM (22.0%) in W-EU and NDM (48.1%) and OXA-48 (46.4%) in E-EU (Figure 4).
- Almost half (47.9%) of CRE isolates carrying an OXA-48-like also carried an NDM.
- An MBL gene was identified in 34.5% of CRE isolates from W-EU and 50.7% of CRE isolates from E-EU (Figure 4).

Conclusions

- ATM-AVI demonstrated potent activity against Enterobacterales, including CREs, from all infection types in W-EU and E-EU.
- The activities of CAZ-AVI and MEM-VAB were compromised by the elevated frequencies of MBLs and OXA-48 types, especially in E-EU.

Acknowledgments

This study at JMI Laboratories was supported by Pfizer Inc. (New York, NY). JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by Pfizer Inc.

Figure 1. Frequency of carbapenem-resistant Enterobacterales (CRE) stratified by European region and infection type

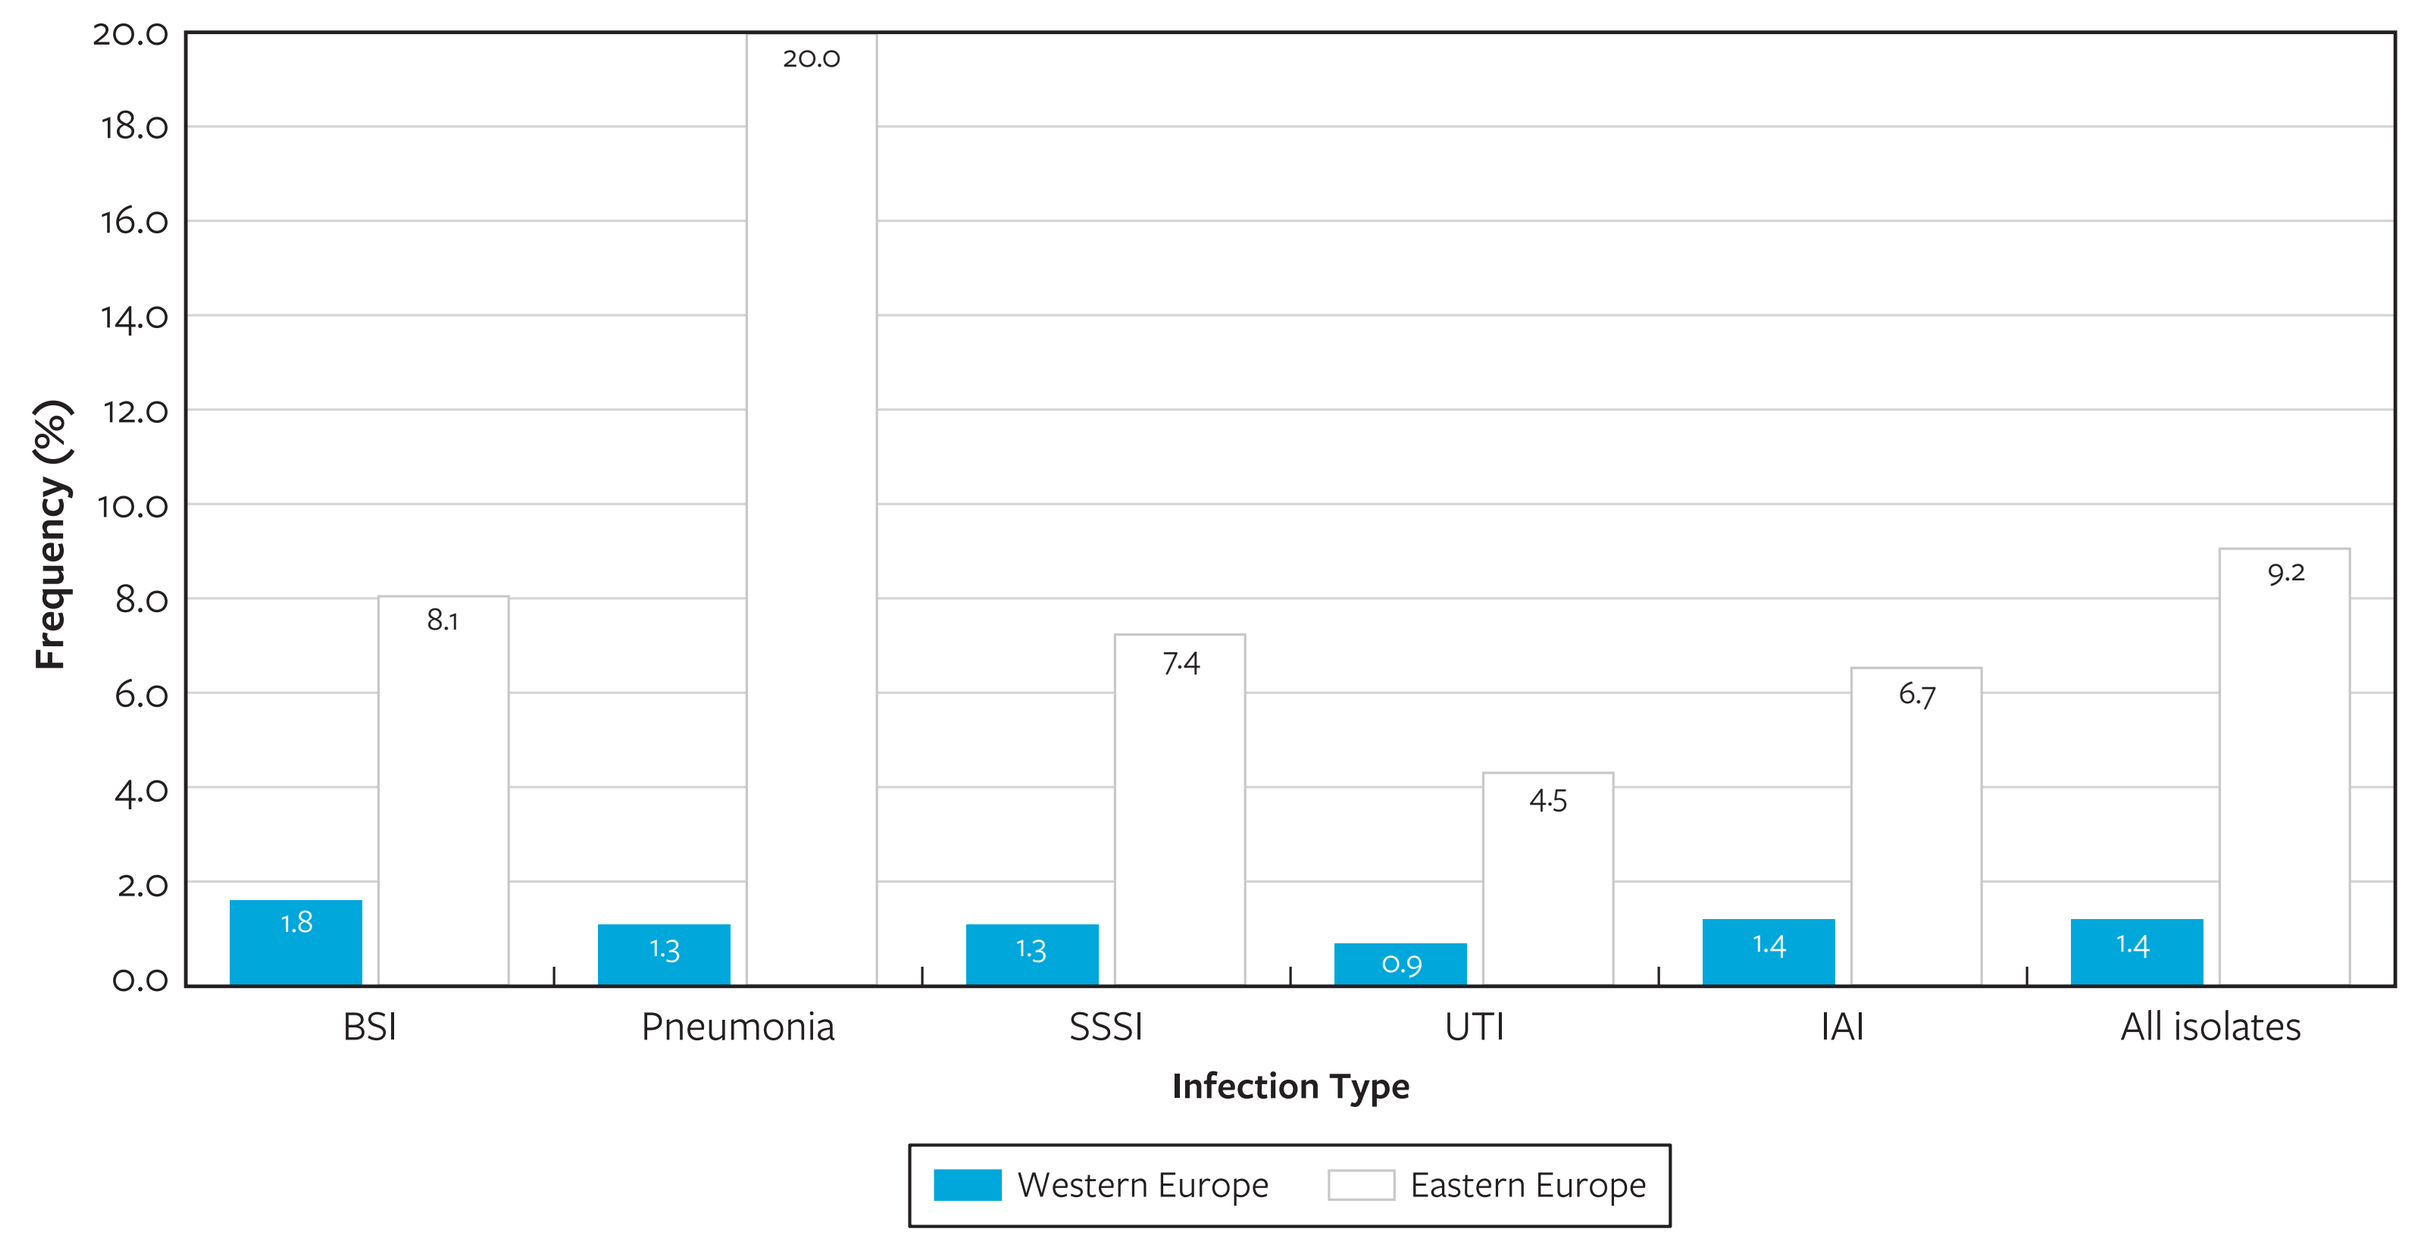
Frequency (%)
Infection Type
BSI
Pneumonia
SSSI
UTI
IAI
All isolates
1.8
8.1
1.3
20.0
1.3
7.4
0.9
4.5
1.4
6.7
1.4
9.2
Western Europe
Eastern Europe

Abbreviations: BSI, bloodstream infection; SSSI, skin and skin structure infection; UTI, urinary tract infection; IAI, intra-abdominal infection.

Figure 2. Frequencies of resistant subsets by European region

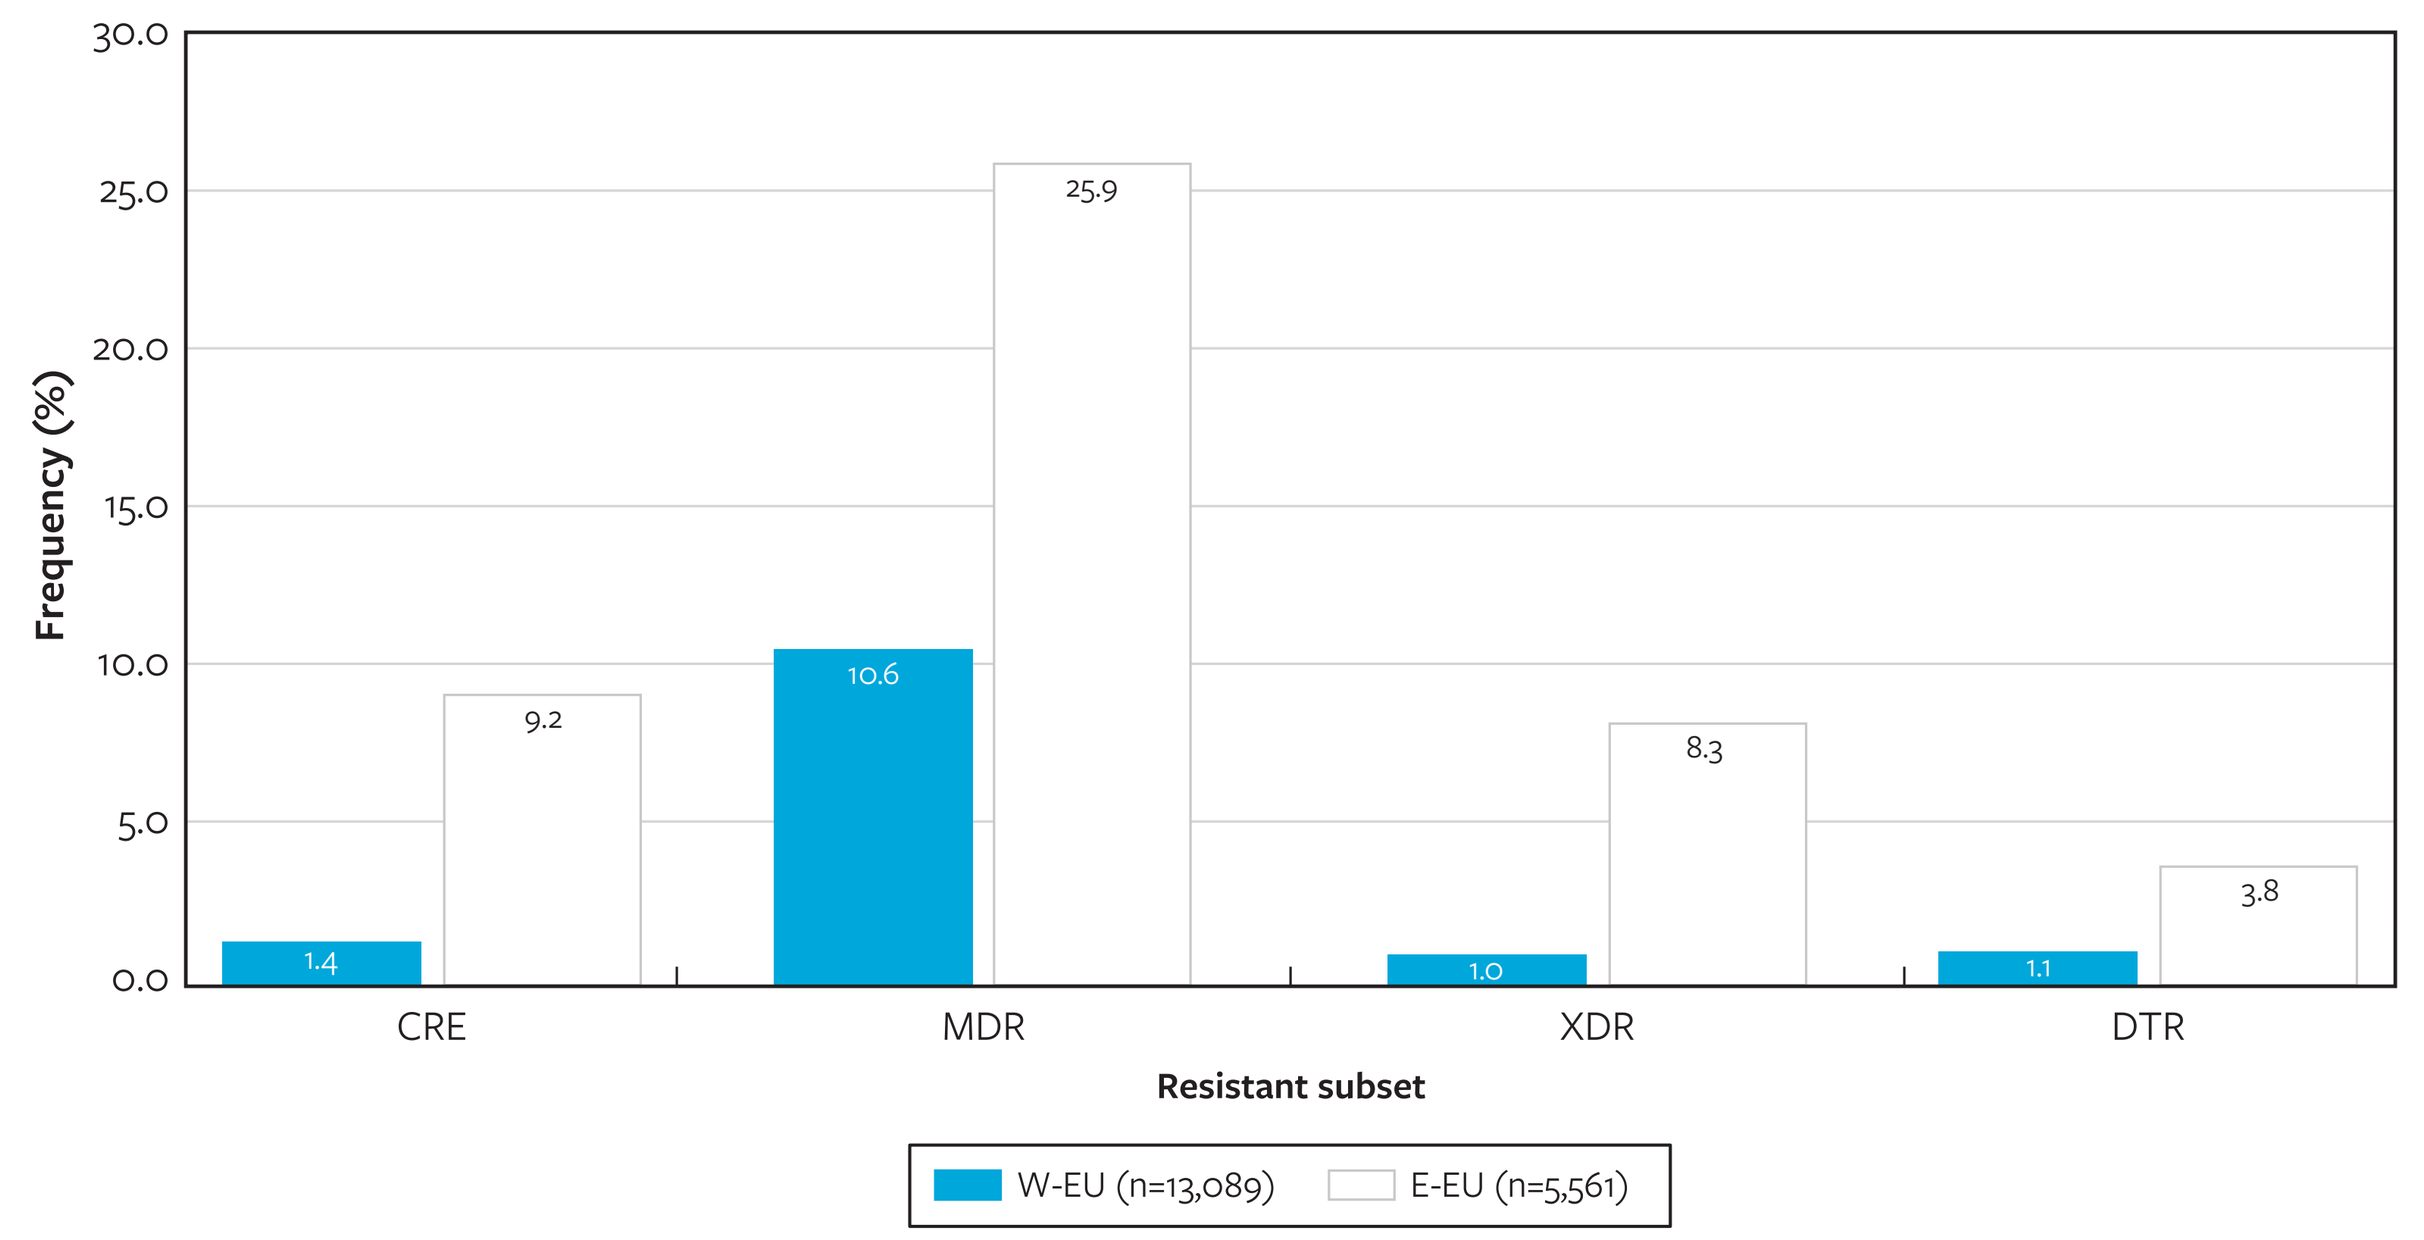
Frequency (%)
Resistant subset
CRE
MDR
XDR
DTR
1.4
9.2
10.6
25.9
1.0
8.3
1.1
3.8
W-EU (n=13,089)
E-EU (n=5,561)

Abbreviations: CRE, carbapenem-resistant Enterobacterales; MDR, multidrug-resistant; XDR, extensively drug-resistant; DTR, difficult-to-treat resistant; W-EU, Western Europe; E-EU, Eastern Europe.

Figure 3. Antimicrobial susceptibility of carbapenem-resistant Enterobacterales (CRE) stratified by European region

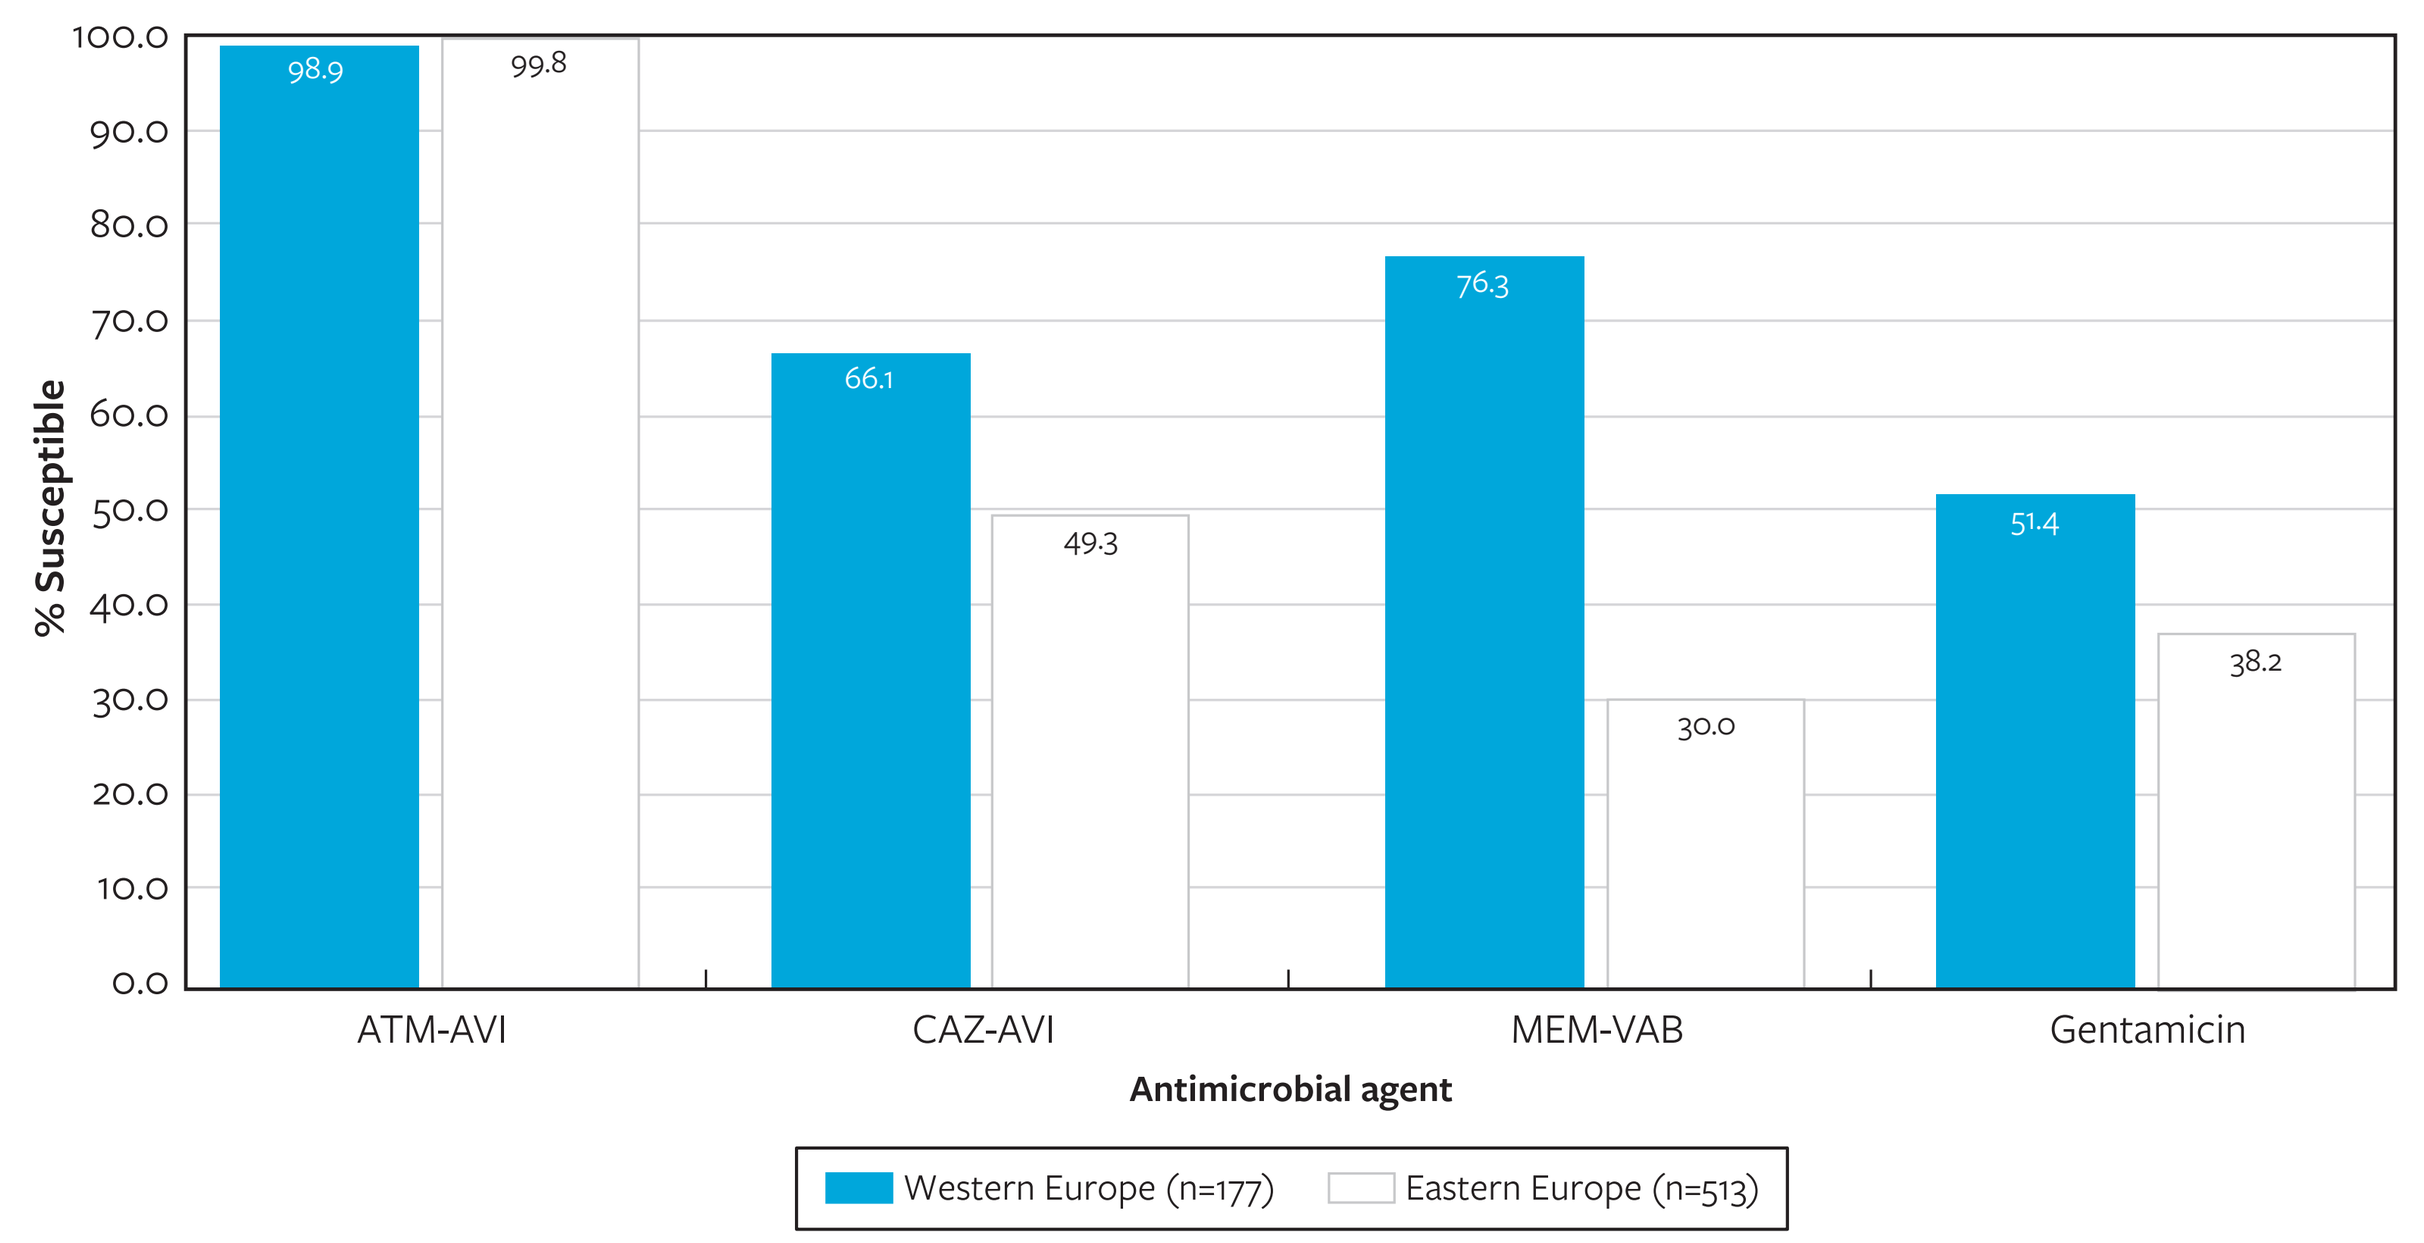
% Susceptible
Antimicrobial agent
ATM-AVI
CAZ-AVI
MEM-VAB
Gentamicin
98.9
99.8
66.1
49.3
76.3
30.0
51.4
38.2
Western Europe (n=177)
Eastern Europe (n=513)

Abbreviations: ATM-AVI, aztreonam-avibactam; CAZ-AVI, ceftazidime-avibactam; MEM-VAB, meropenem-vaborbactam.

Figure 4. Distribution of carbapenemase types by region

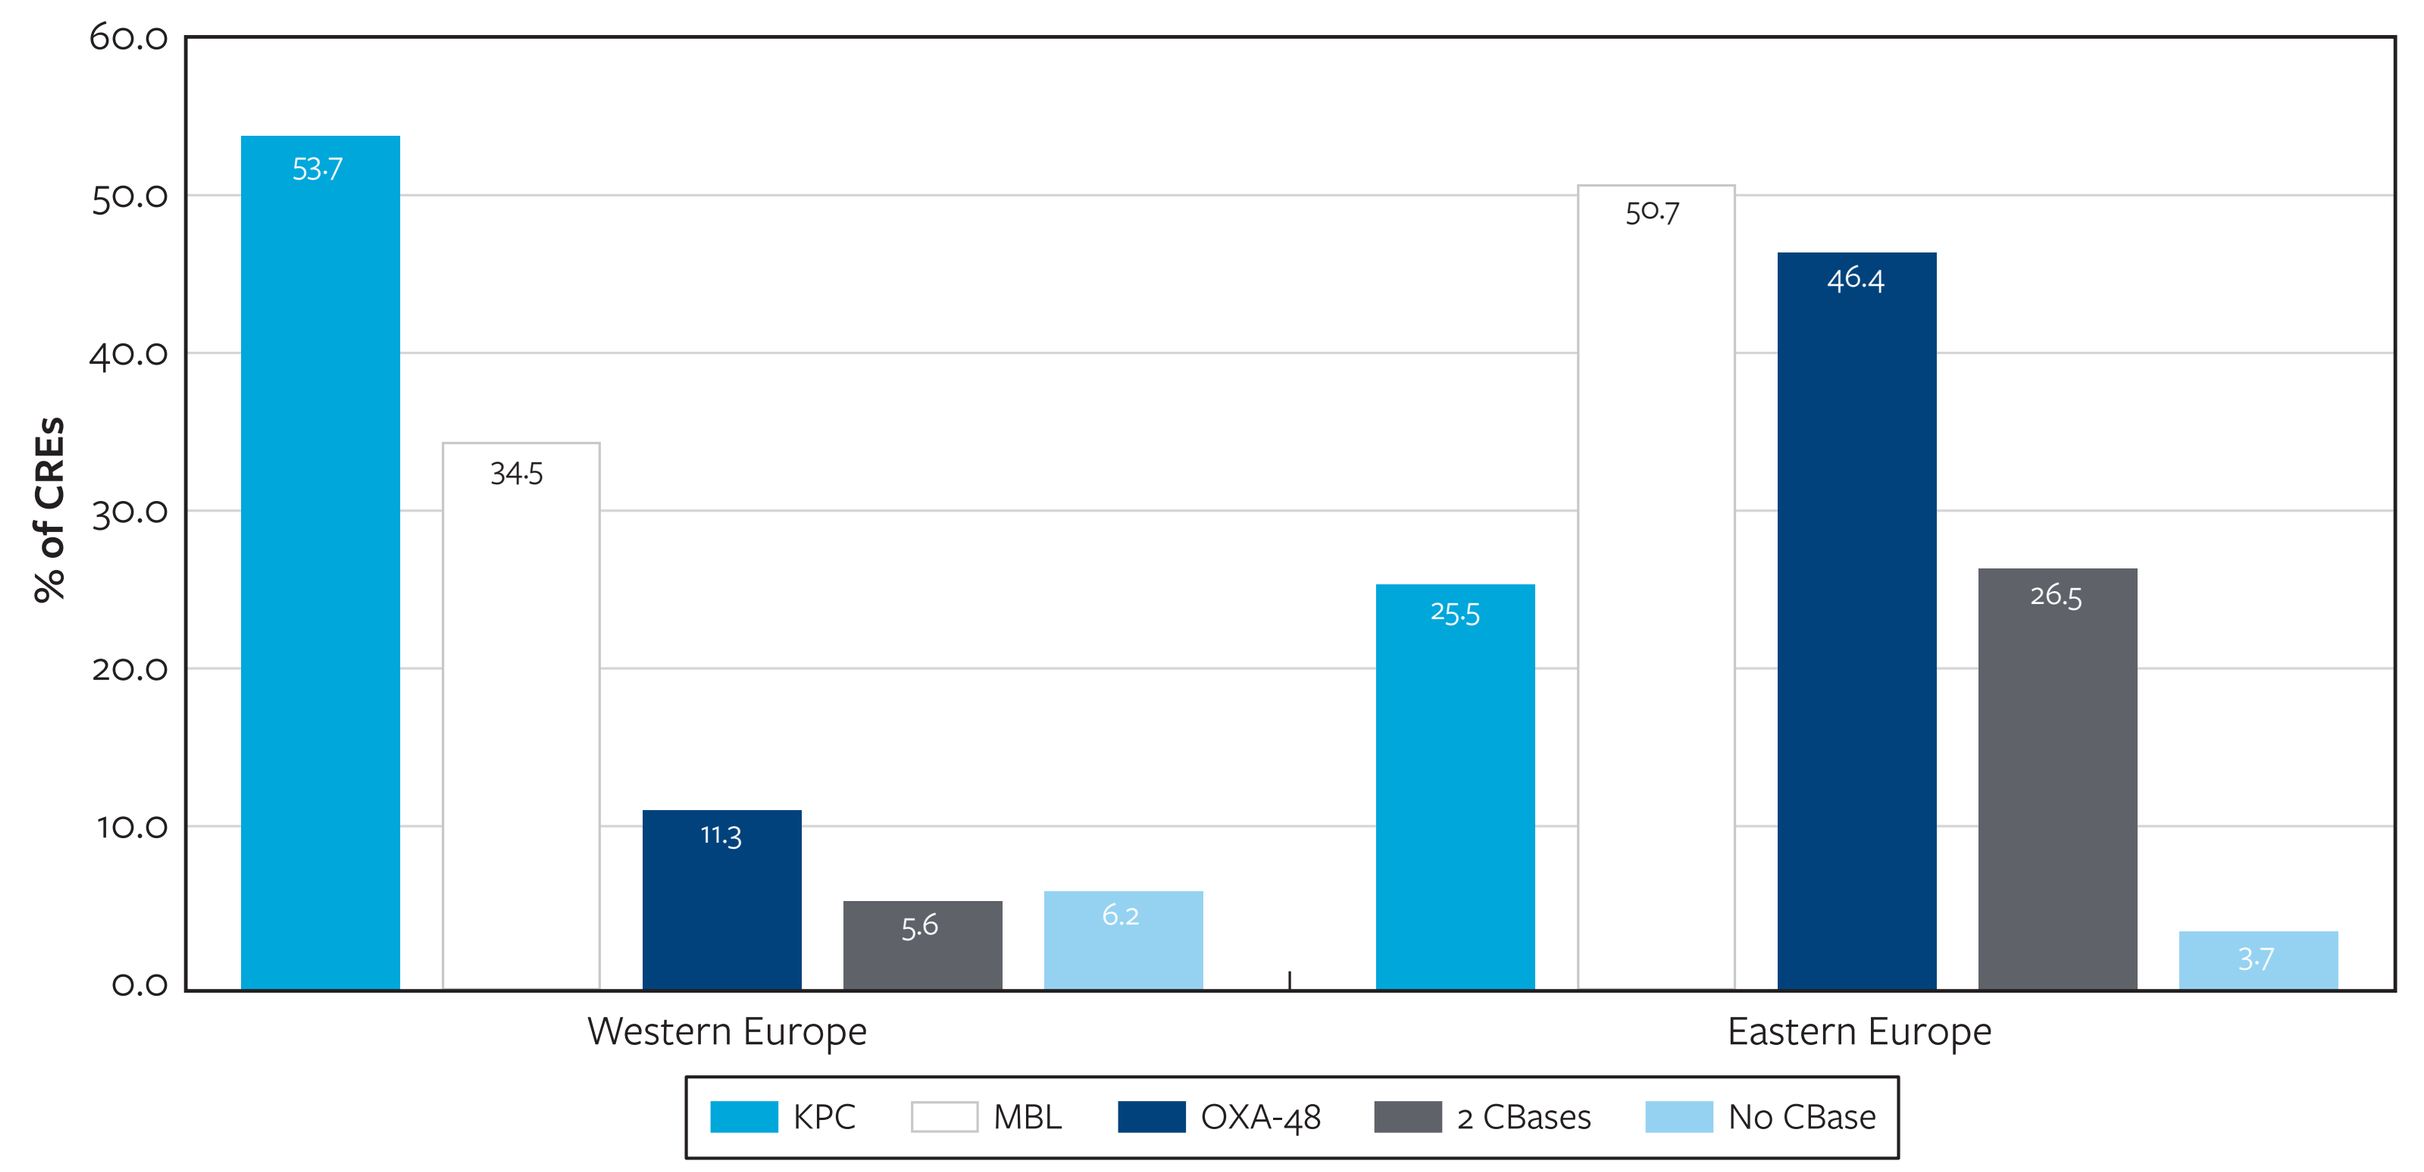
% of CREs
Western Europe
Eastern Europe
53.7
34.5
11.3
5.6
6.2
25.5
50.7
46.4
26.5
3.7
KPC
MBL
OXA-48
2 CBases
No CBase

Abbreviations: KPC, *Klebsiella pneumoniae* carbapenemase; MBL, metallo-β-lactamase; OXA, oxacillinase; CBase, carbapenemase.

Table 1. Antimicrobial susceptibility of Enterobacterales stratified by European region and infection type

Region/ Antimicrobial	% Susceptible per EUCAST criteria (no. of isolates)					
	BSI	Pneumonia	SSSI	UTI	IAI	All isolates
Western Europe	(4,181)	(2,965)	(863)	(3,810)	(1,270)	(13,089)
ATM-AVI	100.0	99.9	99.9	100.0	100.0	>99.9
CAZ-AVI	99.5	99.5	99.8	99.4	99.5	99.5
MEM-VAB	99.5	99.8	100.0	99.7	99.5	99.7
TOL-TAZ	94.5	93.1	92.6	96.0	92.0	94.2
PIP-TAZ	86.6	83.4	86.4	88.5	83.9	86.1
Meropenem	98.3	98.9	98.7	99.3	98.7	98.8
Ceftriaxone	80.7	81.5	80.3	81.0	81.0	81.0
Levofloxacin	81.5	90.6	84.9	81.7	86.7	84.3
Gentamicin	90.8	94.7	93.4	91.3	93.5	92.3
Eastern Europe	(1,476)	(1,204)	(516)	(2,023)	(342)	(5,561)
ATM-AVI	99.7	99.9	99.8	100.0	100.0	99.9
CAZ-AVI	95.2	90.9	96.9	97.1	96.2	95.2
MEM-VAB	93.8	86.3	94.8	97.0	95.3	93.5
TOL-TAZ	86.7	74.6	87.0	92.4	88.0	86.3
PIP-TAZ	77.2	65.0	78.9	84.2	78.4	77.4
Meropenem	92.2	80.1	92.8	95.6	94.2	91.0
Ceftriaxone	64.6	58.4	66.7	71.7	63.7	66.0
Levofloxacin	66.7	63.9	65.4	69.4	67.3	67.0
Gentamicin	82.2	78.5	81.4	85.7	88.6	83.0

Abbreviations: BSI, bloodstream infection; SSSI, skin and skin structure infection; UTI, urinary tract infection; IAI, intra-abdominal infection; ATM-AVI, aztreonam-avibactam; CAZ-AVI, ceftazidime-avibactam; MEM-VAB, meropenem-vaborbactam; TOL-TAZ, ceftolozane-tazobactam; PIP-TAZ, piperacillin-tazobactam.

References

CLSI. 2025. M100Ed35. Performance standards for antimicrobial susceptibility testing: 35th informational supplement. Clinical and Laboratory Standards Institute.

EUCAST 2025. Breakpoint tables for interpretation of MICs and zone diameters. Version 15.0. European Committee on Antimicrobial Susceptibility Testing.

Sader HS, Carvalhaes CG, Kimbrough JH, Mendes RE, Castanheira M. Activity of aztreonam-avibactam against Enterobacterales resistant to recently approved beta-lactamase inhibitor combinations collected in Europe, Latin America, and the Asia-Pacific Region (2020–2022). *Int J Antimicrob Agents* 2024;63(4):107113.

Sader HS, Kimbrough JH, Winkler ML, Castanheira M, Mendes RE. Aztreonam/avibactam activity against Enterobacterales from European medical centres: summary of 5 years of surveillance prior to approval for clinical use (2019–2023). *J Antimicrob Chemother* 2025;80(8):2070–2079.

Tamma PD, Heil EL, Justo JA *et al.* Infectious Diseases Society of America 2024 guidance on the treatment of antimicrobial-resistant Gram-negative infections. *Clin Infect Dis* 2024 ciae403.

Contact

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

SCAN ME
Pfizer.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2025_24-PZR-01_P2_ATM-AVI_Inf.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia