IDWEEK 2025 | Poster #P1314

Activity of aztreonam-avibactam against Enterobacterales isolated from patients with intra-abdominal infection from Europe, Asia, and Latin America (2020–2024)

Helio S. Sader[1], Rodrigo E. Mendes[1], Katherine Perez[2], Gregory Stone[2], Marisa L. Winkler[1], Mariana Castanheira[1]
[1] Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA; [2] Pfizer Inc., New York, New York, USA

Introduction

- Aztreonam-avibactam (ATM-AVI) was recently approved by the US FDA for treatment of complicated intra-abdominal infection (IAI) and by the EMA in the European Union for treatment of adults with complicated IAI, complicated urinary tract infection (cUTI), hospital-acquired pneumonia, including ventilator-associated pneumonia, and infections due to aerobic Gram-negative bacteria in adults with limited treatment options.
- Aztreonam-avibactam (ATM-AVI) has demonstrated potent activity against multidrug-resistant (MDR) Enterobacterales worldwide, including metallo-β-lactamase (MBL) producers.
- We evaluated the activity of ATM-AVI against Enterobacterales isolated from patients with IAI.

Methods

- A total of 3,565 isolates (1/patient) were consecutively collected in 2020–2024 from patients with IAI in 52 medical centers from 28 countries as follows:
 - Western Europe (W-EU): 2,059 isolates from 25 centers in 10 countries
 - Eastern Europe (E-EU): 524 isolates from 7 centers in 5 countries
 - The Asia-Pacific region (APAC): 645 isolates from 12 centers in 7 countries
 - Latin America (LATAM): 337 isolates from 8 centers in 6 countries.
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of pneumonia were included in the study.
- Isolates were susceptibility tested by CLSI M07 broth microdilution method.
- The ATM-AVI susceptible breakpoint of ≤4 mg/L, which was established by the US FDA and EMA for Enterobacterales, was applied.
- Carbapenem-resistant Enterobacterales (CRE) were screened for carbapenemases (CBase) by whole genome sequencing.

Results

- The most common Enterobacterales species were *E. coli* (50.4%), *K. pneumoniae* (18.0%), and *E. cloacae* species complex (8.0%).
- Susceptibility of Enterobacterales stratified by species is shown in Table 1.
- Only 2 Enterobacterales exhibited ATM-AVI MIC >4 mg/L ($MIC_{50/90}$, ≤0.03/0.12 mg/L), 2 *E. coli* with ATM-AVI MIC of 8 mg/L.
- A multidrug-resistant (MDR) phenotype (not susceptible [S] to ≥3 classes per EUCAST) was observed in 14.6% of Enterobacterales (*n*=522).
- The most active β-lactamase inhibitor combination (BLIC) against MDR isolates were ATM-AVI (99.6% S), ceftazidime-avibactam (CAZ-AVI; 91.9% S), meropenem-vaborbactam (MEM-VAB; 91.6% S), and imipenem-relebactam (IMI-REL; 87.9% S; Table 1).
- Piperacillin-tazobactam (PIP-TAZ) was active against 82.2% of Enterobacterales and 38.1% of MDR, and meropenem was active against 97.4% of Enterobacterales and 82.2% of MDR.
- A CBase was identified in 92 isolates (89.3% of CREs), and included KPCs (44.7% of CREs), MBLs (40.8%), and OXA-48 type (15.5%); 11.7% of CRE had 2 CBase types and a CBase was not identified in 10.7% of CREs (Figure 1).
- Only ATM-AVI showed good activity against CRE ($MIC_{50/90}$, 0.25/1 mg/L, 100.0% S); CAZ-AVI, MEM-VAB, and IMI-REL were active against 59.2%, 57.3%, and 43.4% of CRE, respectively (Figure 2).
- All CBase producers were susceptible to ATM-AVI whereas susceptibility to CAZ-AVI, MEM-VAB, and IMI-REL were 55.4%, 54.3%, and 42.9%, respectively (Figure 2).
- The frequencies of CBases varied markedly by region (Figure 3).

Conclusions

- ATM-AVI exhibited almost complete activity against Enterobacterales causing IAI in Europe, the Asia-Pacific region, and Latin America.
- The activities of CAZ-AVI, MEM-VAB, and IMI-REL against CRE were compromised by the high occurrence of MBL and OXA-48-like producers.

Acknowledgments

This study at JMI Laboratories was supported by Pfizer Inc. (New York, NY). JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by Pfizer Inc.

References

Bonomo RA, Chow AW, Edwards MS, *et al*. Clinical practice guideline update by the Infectious Diseases Society of America on complicated intra-abdominal infections: Risk assessment, diagnostic imaging, and microbiological evaluation in adults, children, and pregnant people. *Clin Infect Dis* 2024; 79: S81–S87.

CLSI. 2025. M100Ed35. Performance standards for antimicrobial susceptibility testing: 35th informational supplement. Clinical and Laboratory Standards Institute.

Delp H, Gibson GA, Buckman SA. Aztreonam-avibactam for the treatment of intra-abdominal infections. *Expert Opin Pharmacother* 2024;25(14):1867–1872.

Sader HS, Kimbrough JH, Winkler ML, Mendes RE, Castanheira M. Activity of β-lactamase inhibitor combinations against Enterobacterales isolated from patients with intra-abdominal infection from United States medical centres (2019–2023). *Antibiotics* (*Basel*) 2025;14(6):544.

Figure 1. Carbapenemase distribution (no. of isolates)

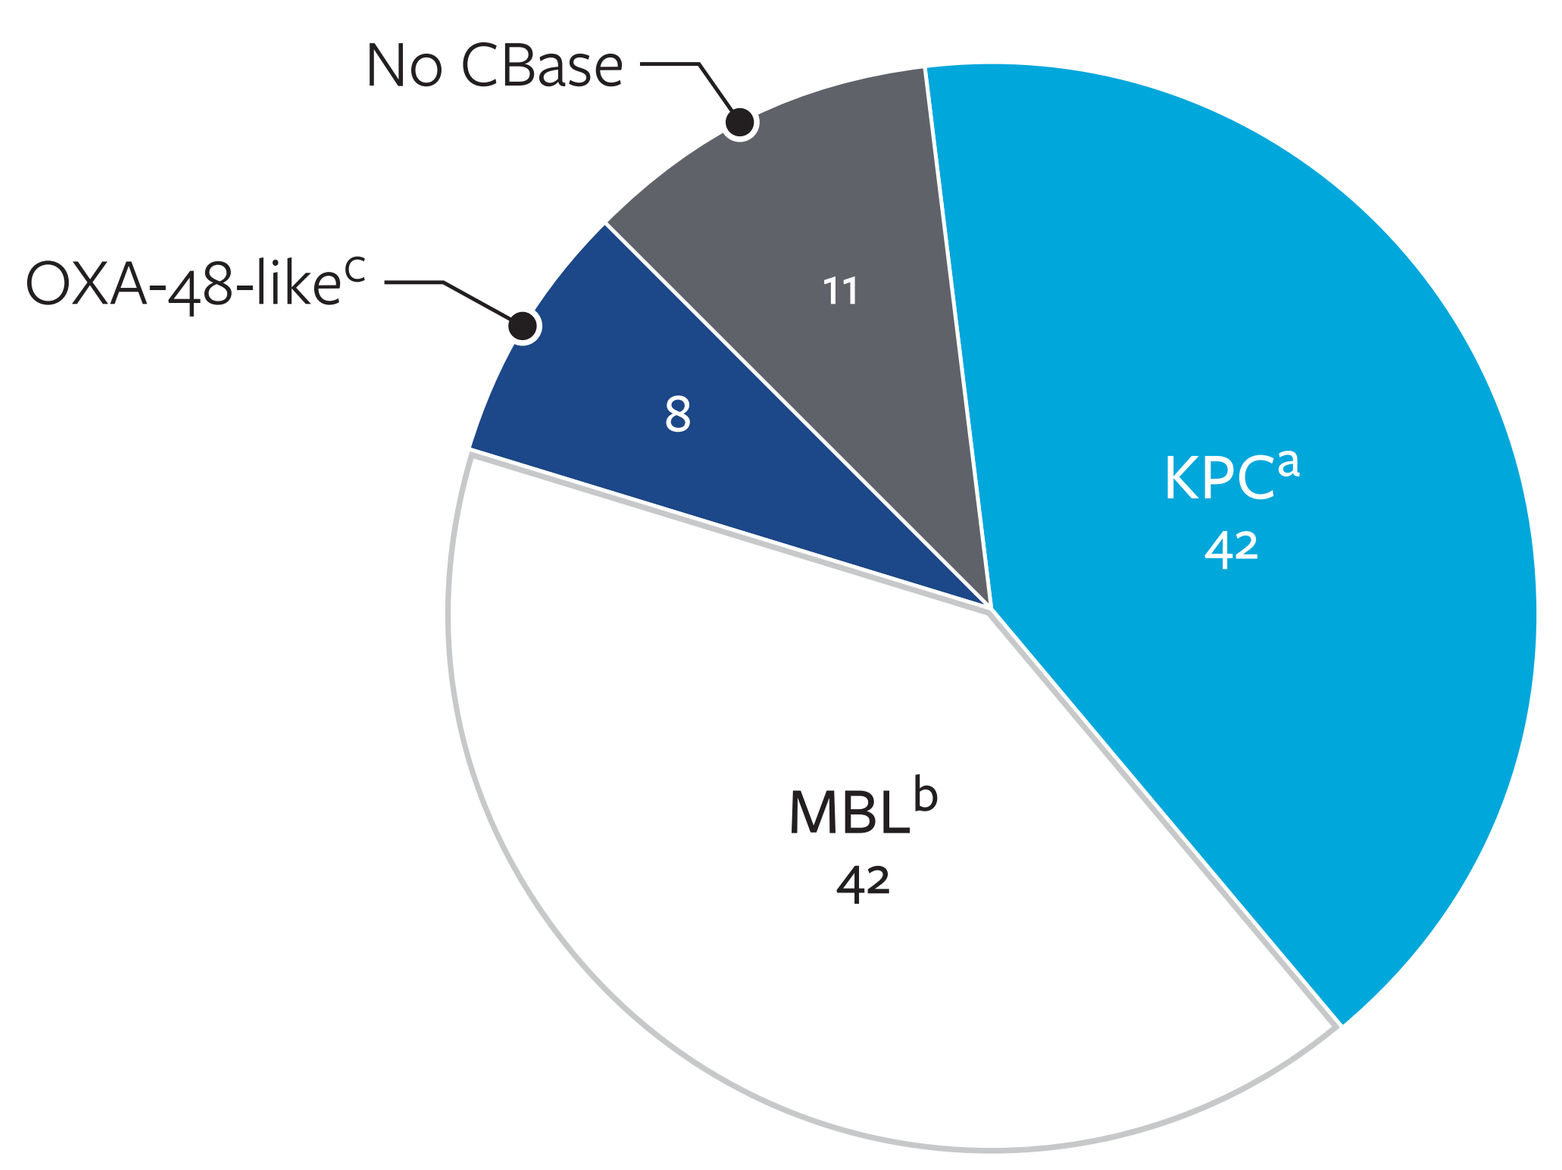
No CBase
OXA-48-like[c]
11
8
KPC[a]
42
MBL[b]
42

[a] Includes KPC-2 (22), KPC-3 (19), and KPC-4 (1); and excludes 4 MBL co-producers (KPC-2 plus NDM-1 [2], KPC-2 plus VIM-1 [1], and KPC-3 plus VIM-1 [1])
[b] Includes NDM-1 (23), NDM-5 (11), and VIM-1 (8). The MBL co-producers are included in this group.
[c] Includes OXA-48 (6), OXA-181 (1), and OXA-232 (1); and excludes 8 MBL co-producers (OXA-48 plus NDM-1 [2], OXA-48 plus NDM-5 [2], OXA-181 plus NDM-5 [1], OXA-232 plus NDM-1 [1], and OXA-232 plus NDM-5 (2).

Table 1. Activity of β-lactamase inhibitor combinations and ceftriaxone against Enterobacterales from intra-abdominal infections

Organism / group (no.)	% Susceptible per EUCAST (MIC_{90} in mg/L)					
	ATM-AVI	CAZ-AVI	MEM-VAB	IMI-REL	PIP-TAZ	CRO
Enterobacterales (3,565)	99.9 (0.12)	98.8 (0.5)	98.8 (0.06)	97.6 (0.5)	82.2 (64)	75.2 (>8)
MDR (522)	99.6 (0.5)	91.9 (2)	91.6 (4)	87.9 (8)	38.1 (>128)	11.1 (>8)
E. coli (1,798)	99.9 (0.06)	99.7 (0.25)	99.7 (0.03)	99.8 (0.12)	90.7 (8)	79.0 (>8)
K. pneumoniae (641)	100.0 (0.25)	95.6 (1)	94.5 (0.5)	93.3 (0.5)	69.6 (>128)	67.9 (>8)
E. cloacae complex (285)	100.0 (1)	98.2 (1)	98.9 (0.06)	98.0 (0.25)	60.7 (128)	57.9 (>8)
K. oxytoca (157)	100.0 (0.12)	98.7 (0.25)	100.0 (0.03)	98.9 (0.25)	81.4 (>128)	87.3 (4)
P. mirabilis (124)	100.0 (≤0.03)	99.2 (0.06)	100.0 (0.12)	77.0 (4)	99.2 (1)	91.9 (≤0.06)
C. freundii complex (117)	100.0 (0.5)	100.0 (0.5)	100.0 (0.03)	100.0 (0.12)	61.5 (128)	60.7 (>8)
Other species (443)	100.0 (0.25)	100.0 (0.5)	100.0 (0.06)	99.3 (1)	81.3 (64)	76.7 (>8)

Abbreviations: ATM-AVI, aztreonam-avibactam; CAZ-AVI, ceftazidime-avibactam; MEM-VAB, meropenem-vaborbactam; IMI-REL, imipenem-relebactam; PIP-TAZ, piperacillin-tazobactam; CRO, ceftriaxone; MDR, multidrug-resistant.

Figure 2. Activity of selected BLICs against CRE isolates stratified by carbapenemase type

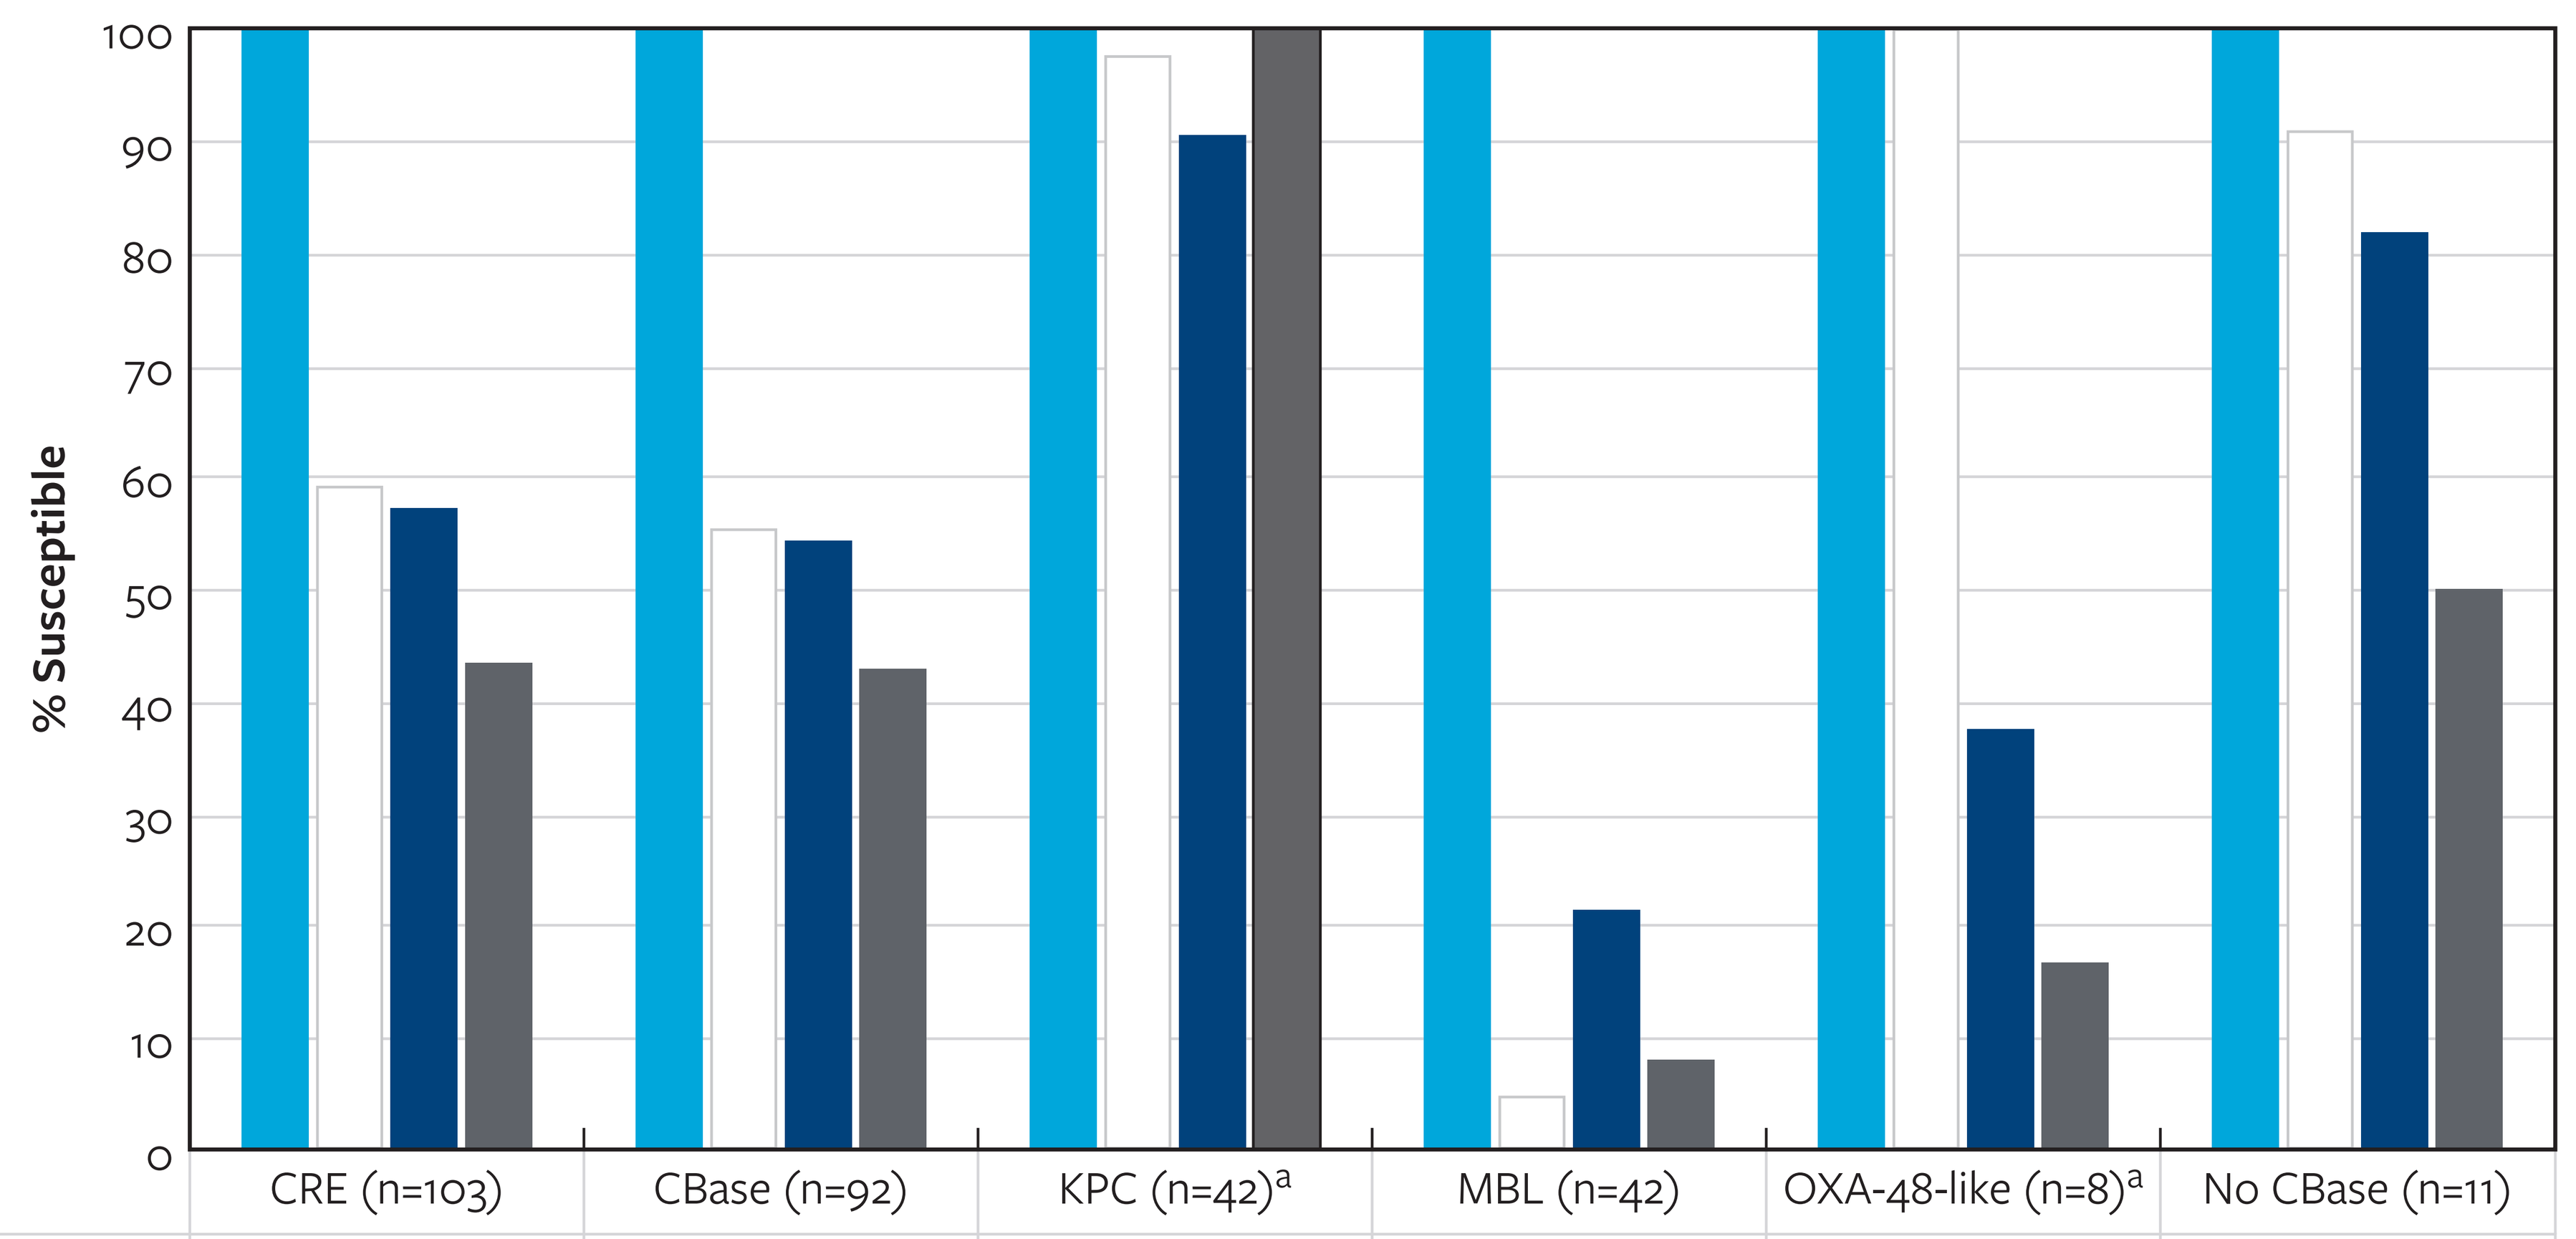
% Susceptible
0
10
20
30
40
50
60
70
80
90
100

	CRE (n=103)	CBase (n=92)	KPC (n=42)[a]	MBL (n=42)	OXA-48-like (n=8)[a]	No CBase (n=11)
ATM-AVI	100.0	100.0	100.0	100.0	100.0	100.0
CAZ-AVI	59.2	55.4	97.6	4.8	100.0	90.9
MEM-VAB	57.2	54.3	90.5	21.4	37.5	81.8
IMI-REL	43.4	42.9	100.0	8.0	16.7	50.0

[a] Excludes MBL co-producers.
Abbreviations: BLICs, β-lactamase inhibitor combinations; CRE, carbapenem-resistant Enterobacterales; CBase, carbapenemase; KPC, *Klebsiella pneumoniae* carbapenemase; MBL, metallo-β-lactamase; OXA, oxacillinase.

Figure 3. Distribution of carbapenemase types by region

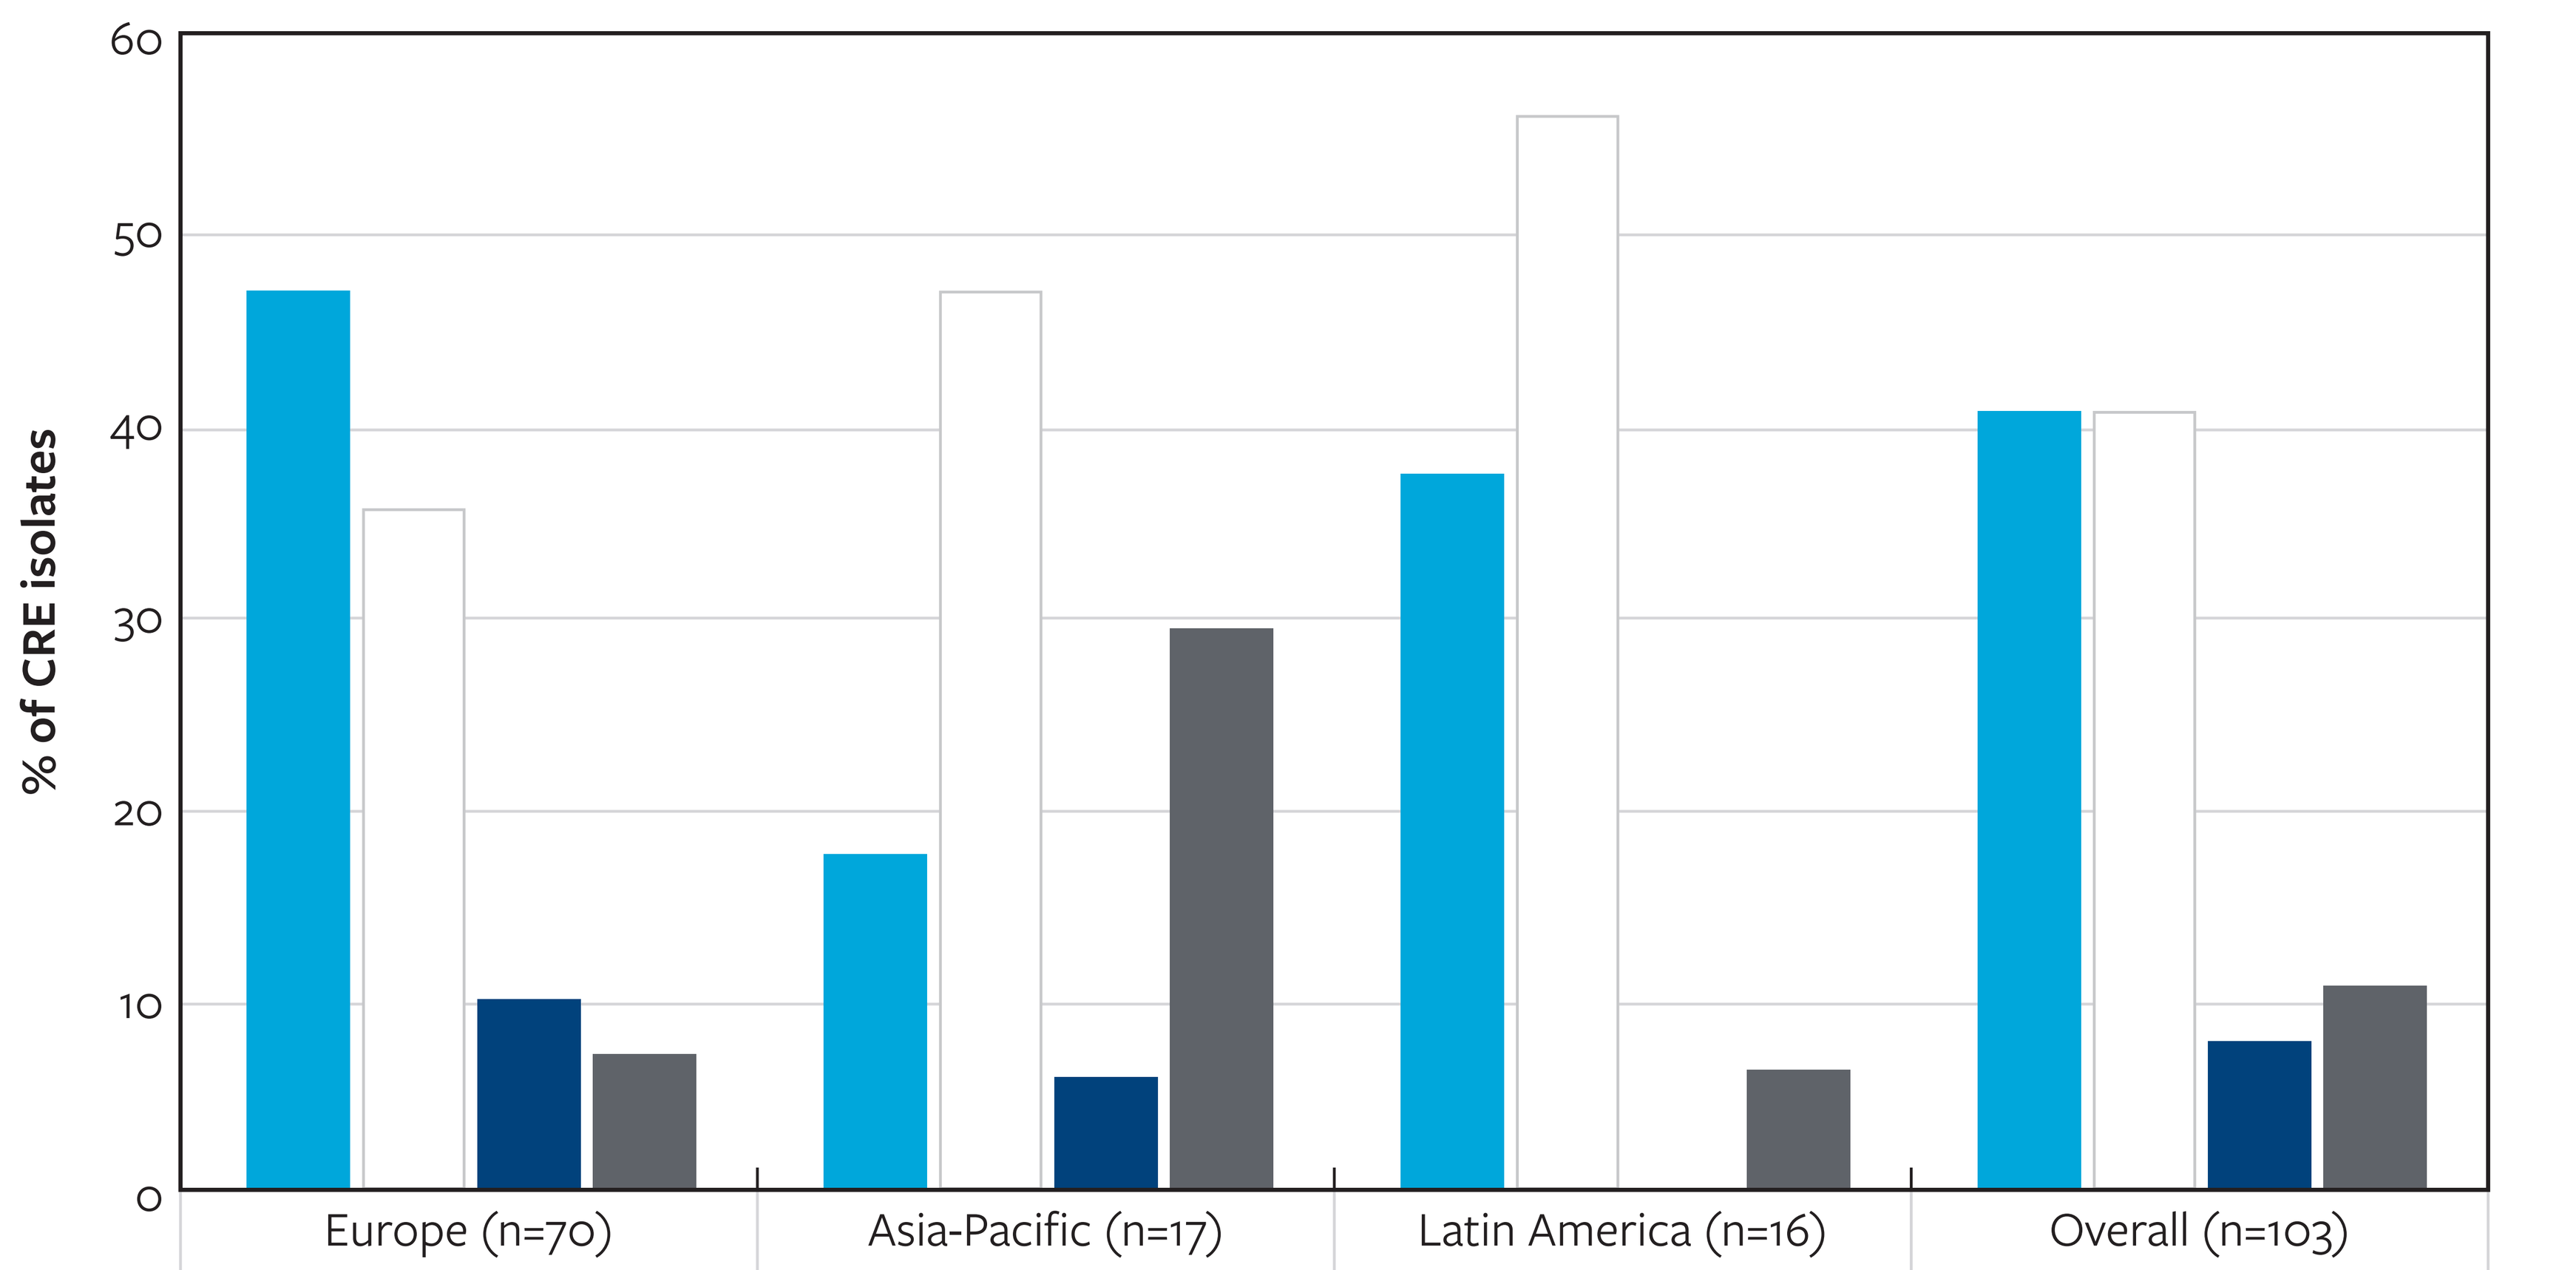
% of CRE isolates
0
10
20
30
40
50
60

	Europe (n=70)	Asia-Pacific (n=17)	Latin America (n=16)	Overall (n=103)
KPC[a]	47.1	17.6	37.5	40.8
MBL	35.7	47.1	56.3	40.8
OXA-48-like[a]	10.0	5.9	0.0	7.8
No CBase	7.1	29.4	6.3	10.7

[a] Excludes MBL co-producers.
Abbreviations: KPC, *Klebsiella pneumoniae* carbapenemase; MBL, metallo-β-lactamase; OXA, oxacillinase; CBase, carbapenemase.

Contact

element

SCAN ME

Pfizer.com

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2025_24-PZR-01_P3_ATM-AVI_IAI.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia