IDWEEK 2025 | Poster #P1332

Activity of aztreonam-avibactam tested against genetically characterized non-carbapenemase-producing carbapenem-resistant Enterobacterales

Mariana Castanheira[1], John H. Kimbrough[1], Dmitri Debabov[2], Helio S. Sader[1]

[1] Element Iowa City (JMI Laboratories), IA, USA; [2] Abbvie Inc., Chicago, Illinois, USA

CONCLUSIONS

Carbapenem resistance among non-CPE CRE isolates seems to involve β-lactamase production with reduced access to the bacterial target due to changes in OMP and regulators that might increase drug efflux.

Despite the combinations of resistance mechanisms, aztreonam-avibactam and ceftazidime-avibactam were the most active agents tested, followed by meropenem-vaborbactam, imipenem-relebactam, and cefiderocol.

New β-lactam/β-lactamase inhibitors and cefiderocol are recommended for treatment of serious infections caused by CRE isolates and these agents display good activity against CRE isolates; however, the activity of these agents varies depending on the enzymes present.

Contact Information

Mariana Castanheira, PhD
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
mariana.castanheira@element.com

Acknowledgements

The authors thank the participant sites of the INFORM Program for providing the isolates.

Funding

This study was supported by Abbvie. Authors are employees of JMI Laboratories, which was paid consultant to Abbvie in connection with the development of this poster.

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2025 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2025_25-ALG-02_P1_non_CPE_CRE.pdf

To submit a medical question, please visit www.abbviemedinfo.com

SCAN ME

References

1. CLSI. M07Ed12. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically: eleventh edition. Wayne, PA, Clinical and Laboratory Standards Institute, 2012.
2. CLSI. M100Ed35. Performance standards for antimicrobial susceptibility testing: 35th informational supplement. Wayne, PA, Clinical and Laboratory Standards Institute, 2025.
3. Tamma PD, Aitken SL, Bonomo RA, Mathers AJ, van Duin D, Clancy CJ. Infectious Diseases Society of America 2022 Guidance on the Treatment of Extended-Spectrum β-lactamase Producing Enterobacterales (ESBL-E), Carbapenem-Resistant Enterobacterales (CRE), and Pseudomonas aeruginosa with Difficult-to-Treat Resistance (DTR-P. aeruginosa). Clin Infect Dis. 2022 Apr 19:ciac268. doi: 10.1093/cid/ciac268. Epub ahead of print. PMID: 35439291.

abbvie

INTRODUCTION

- Carbapenem-resistant Enterobacterales (CRE) isolates are considered a threat to human health.
- Most of the CRE isolates produce carbapenemases, but a portion of them do not carry these enzymes and instead have combinations of resistance mechanisms that elevate the carbapenem MIC values along with those of other β-lactams.
- Aztreonam-avibactam was recently approved by the United States Food and Drug Administration (US FDA) for the treatment of complicated intra-abdominal infections in adults with limited or no alternative treatment options.
 – This combination agent displays activity against CRE isolates producing class A enzymes, some class D and also class B metallo-β-lactamases but limited data is available for its activity against non-carbapenemase-producing (non-CPE) CRE isolates.
- We evaluated the activity of aztreonam-avibactam and comparator agents against 111 non-CPE CRE isolates collected over a 7-year period in US hospitals (2016–2023).

Figure 1. Summary of β-lactam resistance mechanisms among 111 non-CPE CRE isolates

				Amino acid alteration					
Organism	**US State**	**MLST**	**Beta-lactamase**	**PBP3**	**ACRR**	**OMPC**	**OMPF**	**RAMR**	**SOXR**
Citrobacter freundii	TX	402	TEM-1						
	FL	519							
Citrobacter youngae	KY	30							
Enterobacter asburiae	MO	484	OXA-17						
	KY	162-like							
Enterobacter cloacae	WI	50							
Enterobacter hormaechei	WI	108							
	WI	198							
	IL	254							
	WA	550							
	IL	742							
	CO	782							
	WI	108-like							
	NY	120-like							
	NY	636-like							
	MA	93-like							
Enterobacter kobei	MN	54							
	FL	855-like							
Enterobacter ludwigii	CO	853-like							
Enterobacter spp.	LA	483	CTX-M-15, OXA-1, TEM-1						
Escherichia coli	FL	131	CTX-M-15						
	KS	131	CTX-M-15, OXA-1						
	LA	131	CMY-2, TEM-1						
	LA	131	CTX-M-15						
	MI	131	CTX-M-15-like, TEM-1						
	TX	131	CTX-M-15, TEM-1						
	VA	131	CMY-2						
	VA	131	CTX-M-15, OXA-1						
	CA	405	CTX-M-15, OXA-1						
	IN	405	CTX-M-15, CTX-M-33	YRIK					
	TX	744	TEM-1						
	TX	12468	CTX-M-14						
	MN	617-like	CTX-M-15, OXA-1						
Hafnia paralvei	CO	—							
Klebsiella aerogenes	CO	4							
	NY	4							
	TX	4							
	CA	92							
	CA	93							
	CA	93							
	CA	93							
	CA	93							
	CA	93							
	CO	93							
	CO	93							
	NJ	93							
	TX	93							
	TX	93							
	MA	116							
	MN	128							
	NY	135							
	CO	151							
	KY	176							
	TX	240							
	TX	246							
	IN	385							
	NJ	477							
	CO	484							
	CA	517							
	CA	613							
	MI	441-like							
	NY	73-like							
	NJ	93-like							
	PA	93-like							
	PA	99-like							
Klebsiella michiganensis	TX	107	CTX-M-15, OXA-1, TEM-1						
	CA	194							

				Amino acid alteration					
Organism	**US State**	**MLST**	**Beta-lactamase**	**PBP3**	**ACRR**	**OMPC**	**OMPF**	**RAMR**	**SOXR**
Klebsiella oxytoca	NY	176							
	CA	287							
Klebsiella pneumoniae	FL	11	CTX-M-15, OXA-1, SHV-11						
	NY	15	SHV-1						
	WA	15	CTX-M-15, SHV-28, TEM-1						
	CA	16	CTX-M-15, SHV-1						
	CA	16	CTX-M-15, SHV-1						
	TX	25	CTX-M-71, SHV-11, TEM-1						
	TX	29	CTX-M-15, OXA-1, SHV-187, TEM-1						
	OH	37	CTX-M-15, OXA-1, SHV-11, TEM-1						
	CO	45	CTX-M-15, OXA-1, SHV-1, TEM-1						
	MI	45	SHV-1						
	NY	45	CTX-M-2, OXA-2, SHV-1, TEM-1						
	WA	45	SHV-1						
	KS	147	CTX-M-15, SHV-11						
	LA	147	CTX-M-15, OXA-1, SHV-11						
	NY	147	CTX-M-15, SHV-11, TEM-1						
	NY	152	CTX-M-15, OXA-1, SHV-1, TEM-1						
	PA	258	SHV-12-like						
	AZ	307	CTX-M-33, SHV-205						
	CA	307	CTX-M-15, OXA-1, SHV-28, TEM-1						
	FL	307	CTX-M-15, OXA-1, SHV-28, TEM-1						
	NC	307	CTX-M-15, OXA-1, SHV-28, TEM-1						
	NJ	307	CTX-M-15, OXA-1, SHV-28						
	NY	307	CTX-M-15, OXA-1, SHV-28, TEM-1						
	CA	348	CMY-2, CTX-M-14, SHV-11, TEM-1						
	MN	392	CTX-M-15, SHV-11, TEM-1						
	MA	1229	CTX-M-15, SHV-11						
	CO	1774	CTX-M-15, OXA-2, SHV-11						
	VA	1818	CTX-M-15, OXA-1, SHV-11, TEM-1						
	NY	1835	CTX-M-15, OXA-1, SHV-11, TEM-1						
	NY	2004	CTX-M-15, OXA-1, SHV-168, TEM-1						
	TX	3576	CTX-M-15, OXA-1-like, SHV-1, TEM-1						
	UT	3846	SHV-11						
	TX	4961	CTX-M-220, SHV-52-like, TEM-1						
	OR	5437	SHV-11						
Klebsiella quasipneumoniae	FL	1455	CTX-M-15, OXA-1						
Raoultella ornithinolytica	TX	105	CTX-M-14						
	NY	NT	CARB-2, FOX-5						
Serratia marcescens	OH	365							
	OH	491	CTX-M-15, OXA-1, TEM-1						
	NY	622							
	FL	381-like							
	NJ	386-like							
	FL	NT							

MATERIALS AND METHODS

- A total of 72,265 Enterobacterales isolates were collected during 2016–2023 in 62 US hospitals.
 – Isolates were identified as the cause of infection.
 – Isolates were limited to 1 per patient.
- Isolates were susceptibility tested against aztreonam-avibactam and comparator agents using the reference broth microdilution method as described by the Clinical and Laboratory Standards Institute (CLSI) M07 (2022) and M100 (2025) documents.
 – Avibactam and relebactam were tested at a fixed concentration of 4 mg/L.
 – Vaborbactam was tested at a fixed concentration of 8 mg/L.
- Cefiderocol powder was acquired from MedChem Express (Monmouth Junction, NJ) and tested using iron-depleted cation adjusted Mueller-Hinton broth.
- Quality control (QC) was performed according to the CLSI M100 (2025) criteria.
 – All QC MIC results were within acceptable ranges.
- Categorical interpretations for all comparator agents were those criteria found in the CLSI M100 (2025), or the US FDA website.
- Aztreonam-avibactam breakpoints were those approved by the US FDA.
- CRE isolates resistant to imipenem or meropenem were submitted to whole genome sequencing and data analysis for the detection of β-lactam resistance mechanisms.
- WGS was performed on MiSeq or NextSeq (Illumina, San Diego, California, USA) instruments targeting a 30X coverage.
 – Sequences were *de novo* assembled.
 – Analysis of β-lactam resistance mechanisms was performed *in silico.*

RESULTS

- Among 694 CREs collected during 2016–2023, 111 (16.0% of the CRE, 0.2% overall) were non-CPE.
- Non-CPE CRE isolates belonged to 6 genera, but *K. pneumoniae* (n=34) and *K. aerogenes* (n=30) were the dominant species (Figure 1).
- Non-CPE CRE isolates were observed in 49 hospitals distributed in all US Census divisions.
 – Notably, 10 isolates were collected in one hospital in Colorado.
- Multilocus sequence typing (MLST) analysis showed that some *E. coli* and *K. pneumoniae* non-CPE CRE isolates belonged to successful clones such as ST131 and ST307, but genetic diversity was noted for other species (Figure 1).
- A total of 47 isolates carried acquired β-lactamase genes, including all 13 *E. coli* and 28/34 *K. pneumoniae* (Figure 1).
 – 29/34 *K. pneumoniae* and 10/13 *E. coli* isolates carried CTX-M-encoding genes with most being CTX-M-15.
 – Acquired enzymes were not common among *K. aerogenes*, *Enterobacter*, *Citrobacter*, and *Serratia* species.
- Early terminations/lost start or stop codons in outer membrane protein (OMP) genes were noted in 64 isolates for a single gene (Figure 1).
 – 36 isolates displayed these disruptions for both *ompC/ompK36* and *ompF/ompK35*, including 10 *E. coli* and 10 *K. pneumoniae.*
- Disruptions in the genes encoding the efflux regulators RamR were noted among 6/30 *K. aerogenes*, 1/3 *C. freundii*, and 5/17 *E. cloacae* species complex.
- Aztreonam-avibactam and ceftazidime-avibactam were active against 95.5% and 97.3% of the isolates and had the highest susceptibility rates among the agents tested (Figure 2).
- Meropenem-vaborbactam, imipenem-relebactam, and cefiderocol inhibited 90.0%, 90.0%, and 90.1% of the non-CPE CRE isolates.
 – Other agents displaying activity against >80% of the isolates were gentamicin (80.2% susceptible), amikacin (84.7%), and tigecycline (91.0%).
 – 91.9% of the isolates were intermediate to colistin.
- The two isolates were resistant to aztreonam-avibactam: one *E. coli* producing CTX-M-15 and CTX-M-33 with disruptions in the genes encoding AcrA, OmpC, OmpF, and a PBP3 alteration YRIK and one *K. aerogenes* with OMP and RamR disruptions.
 – These isolates had cefiderocol MIC values >64 mg/L but were susceptible to meropenem-vaborbactam and imipenem-relebactam.

Figure 2. Susceptibility patterns of antimicrobial agents tested against 111 non-CPE CRE isolates collected during 2016–2023 in US hospitals

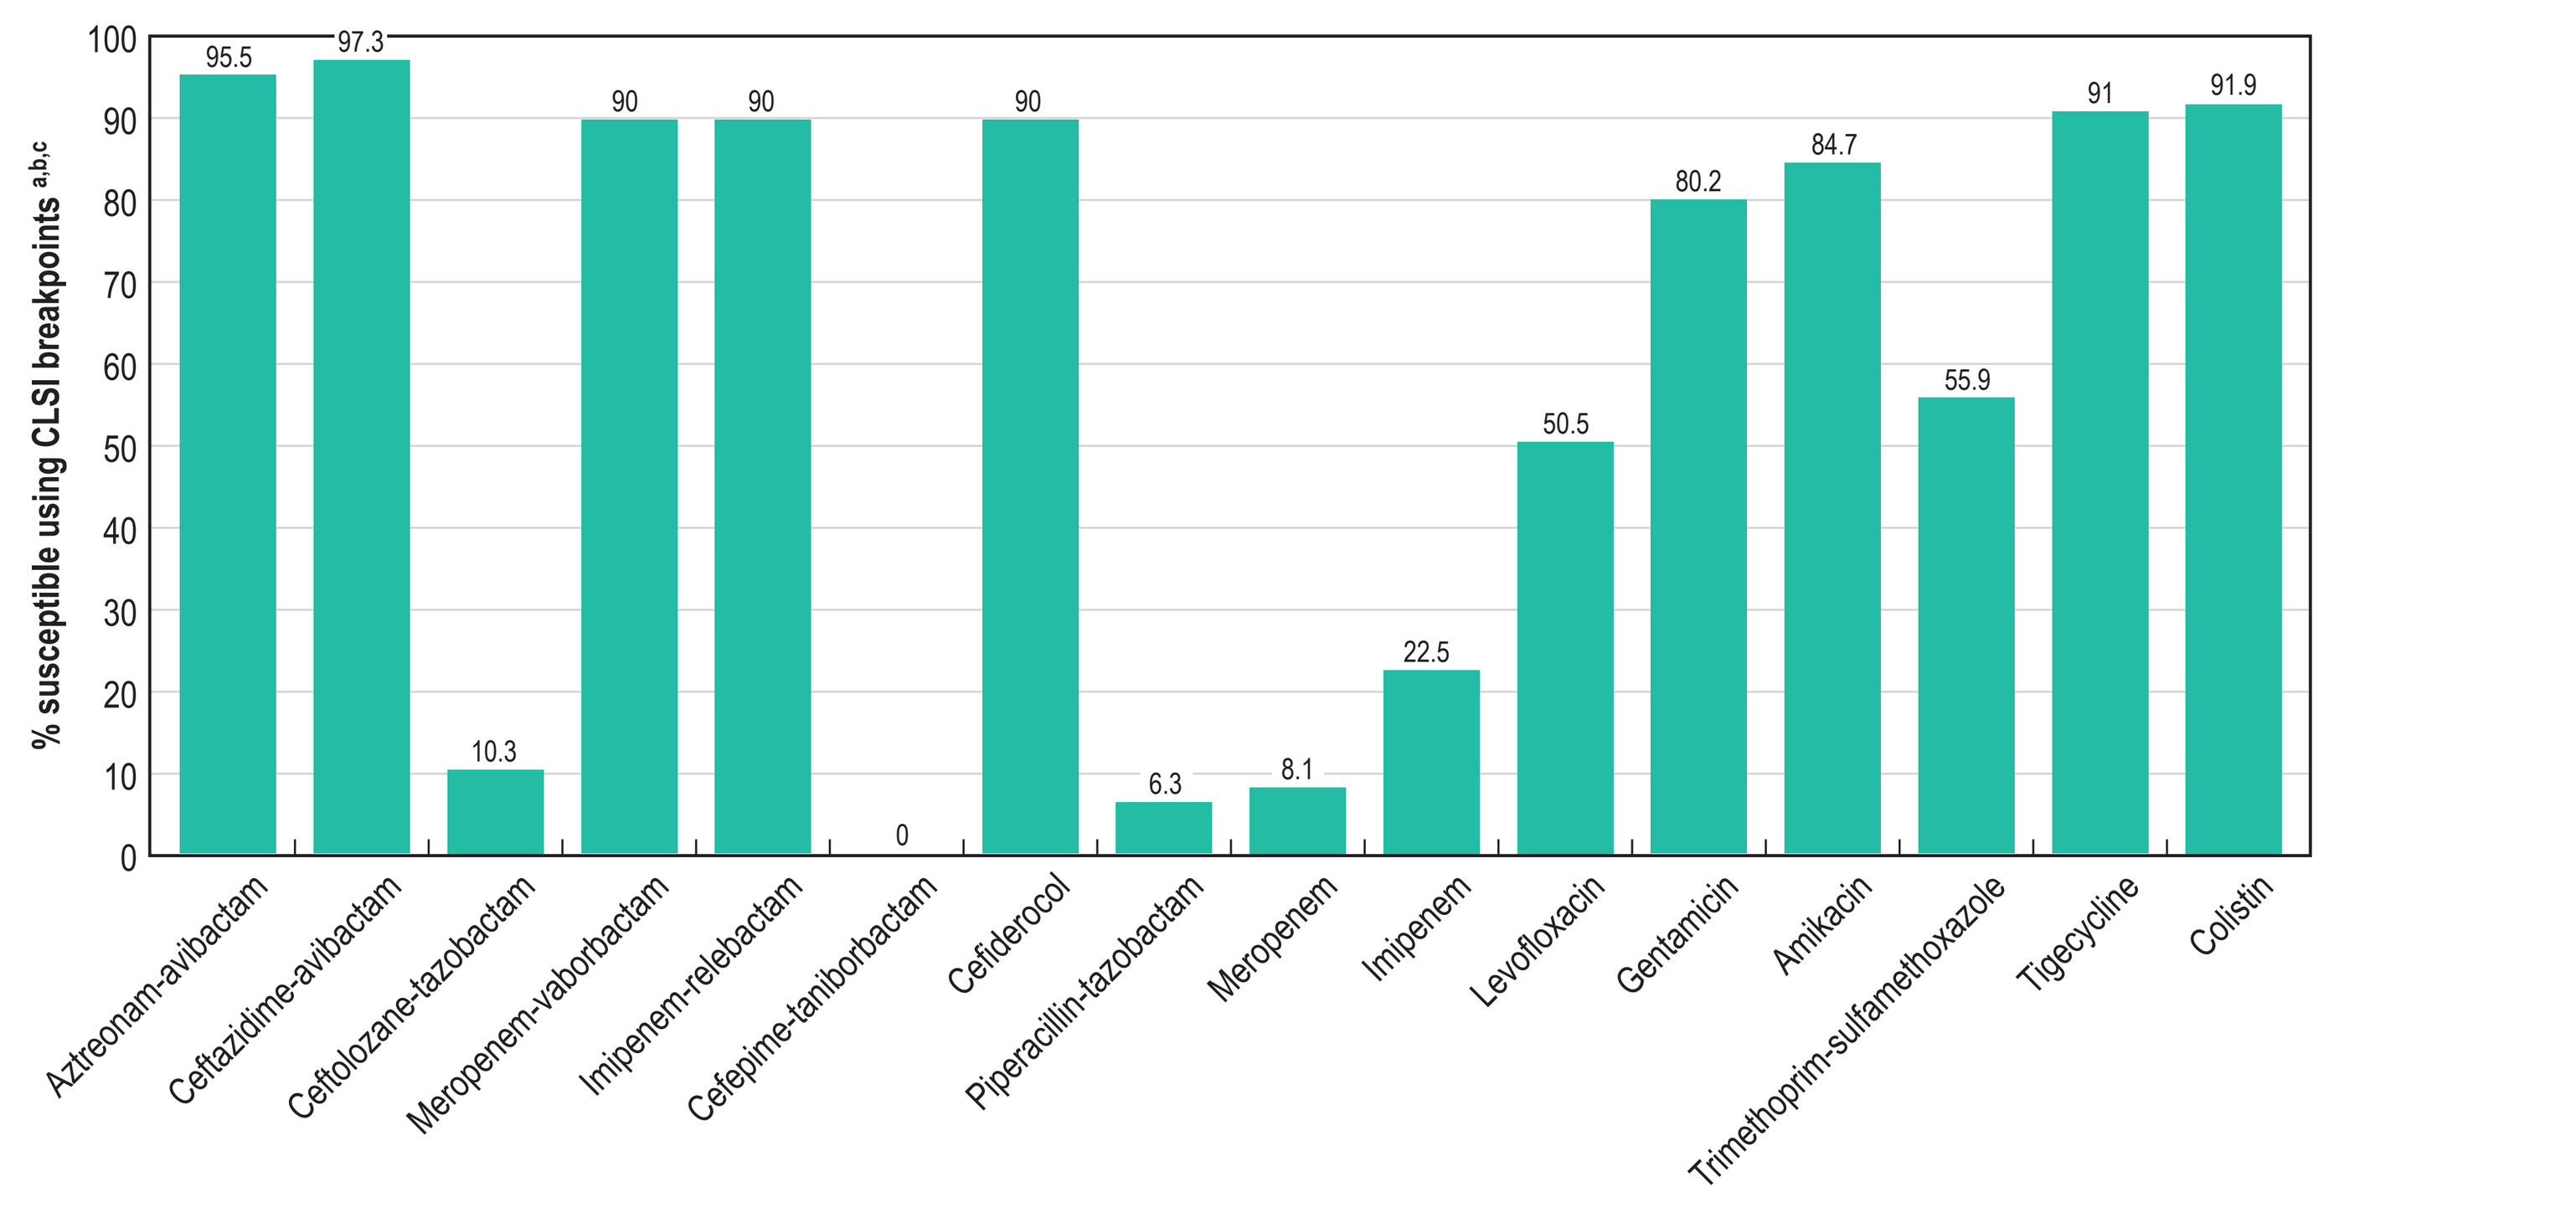
% susceptible using CLSI breakpoints a,b,c
0
10
20
30
40
50
60
70
80
90
100
95.5
97.3
10.3
90
90
0
90
6.3
8.1
22.5
50.5
80.2
84.7
55.9
91
91.9
Aztreonam-avibactam
Ceftazidime-avibactam
Ceftolozane-tazobactam
Meropenem-vaborbactam
Imipenem-relebactam
Cefepime-taniborbactam
Cefiderocol
Piperacillin-tazobactam
Meropenem
Imipenem
Levofloxacin
Gentamicin
Amikacin
Trimethoprim-sulfamethoxazole
Tigecycline
Colistin

[a] Aztreonam-avibactam and tigecycline using FDA breakpoints
[b] Cefepime alone breakpoints were applied for cefepime-taniborbactam for comparison purposes only
[c] Percentage intermediate for colistin