IDWEEK 2025 | Poster #P1313

A 10-year Trend on Antimicrobial Resistance of *Pseudomonas aeruginosa* from United States Medical Centers: Results from the INFORM Surveillance Program (2015–2024)

Helio S. Sader[1], Rodrigo E. Mendes[1], John Lock[2], Marisa L. Winkler[1], Mariana Castanheira[1]

[1] Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA; [2] AbbVie Inc., Chicago, Illinois, USA

CONCLUSIONS

CAZ-AVI, TOL-TAZ, and IMI-REL remain very active against *P. aeruginosa* from US hospitals.

Susceptibility to CAZ-AVI and TOL-TAZ decreased slightly during the study period.

P. aeruginosa susceptibility to meropenem, levofloxacin, and tobramycin improved since 2017, with an accompanying reduction of the frequency of MDR, XDR, and DTR isolates until 2023–2024.

INTRODUCTION

- *Pseudomonas aeruginosa* is a common cause of healthcare-associated infections, including pneumonia, skin and skin structure infection (SSSI), bloodstream infections (BSI), and complicated urinary tract infection (UTI).
- Hospital-associated *P. aeruginosa* infections are associated with high rates of antimicrobial resistance and elevated morbidity and mortality.
- IDSA guidelines currently recommend the use of newer β-lactam/β-lactamase inhibitor combinations (BLICs), such as ceftazidime-avibactam (CAZ-AVI), ceftolozane-tazobactam (TOL-TAZ), and imipenem-relebactam (IMI-REL), for treatment of infections due to difficult-to-treat resistant (DTR) *P. aeruginosa*.
- Several studies have shown that these three BLICs are highly active against multidrug-resistant (MDR) and DTR *P. aeruginosa*, but studies comparing the antipseudomonal activity of these compounds are scarce.
- We evaluated the antimicrobial susceptibility of *P. aeruginosa* isolates from United States (US) medical centers in the last 10 years.

METHODS

- A total of 14,303 *P. aeruginosa* isolates (1/patient) were consecutively collected from 56 US medical centers in 2015–2024.
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of infection were included in the study.
- Only centers that contributed to isolates for ≥8 years were included.
- Isolates were susceptibility tested by Clinical and Laboratory Standards Institute (CLSI) M07 broth microdilution methods at a monitoring laboratory.
- Susceptibility results were stratified by year and infection type.
- MDR was defined as resistance to ≥3 classes, extensively drug-resistant (XDR) as susceptibility to ≤2 classes, and DTR as nonsusceptible to piperacillin-tazobactam (PIP-TAZ), cephalosporins, carbapenems, fluoroquinolones, and aminoglycosides.

RESULTS

- The most active compounds were CAZ-AVI (97.0% susceptible [S]), TOL-TAZ (97.6%S), and IMI-REL (97.9%S; Table 1).
- Nonsusceptibility (NS) rates for CAZ-AVI and TOL-TAZ increased from 1.8% and 1.4% in 2015 to 3.2% and 2.5% in 2024, respectively (Figure 1).
- PIP-TAZ NS rates increased from 17.0% in 2015 to 22.5% in 2024 with marked variation during this period (Figure 1).
- NS rates for meropenem, levofloxacin, and tobramycin increased from 2015 to 2017 and then decreased progressively until 2023 or 2024 (Figure 1).
- The frequency of MDR, XDR, and DTR isolates increased until 2017, decreased from 2017 to 2022 or 2023, and then increased again in 2024 (Figure 2).
- CAZ-AVI was active against 82.9% of MDR, 66.1% of XDR, and 65.3% of DTR isolates, TOL-TAZ was active against 84.4% of MDR, 64.1% of XDR, and 71.9% of DTR isolates, and IMI-REL was active against 87.4% of MDR, 60.7% of XDR, and 64.8% of DTR isolates (Table 2).
- Susceptibility rates were lower among isolates from pneumonia compared to other infections (Figure 3).

Table 1. Overall antimicrobial susceptibility of *P. aeruginosa* (2015–2024)

Antimicrobial agent	No. of isolates tested	MIC_{50}	MIC_{90}	Susceptibility per CLSI		
				%S	%I	%R
Ceftazidime-avibactam	14,303	2	4	97.0		3.0
Ceftolozane-tazobactam	13,374	0.5	2	97.6	0.9	1.5
Imipenem-relebactam[a]	5,914[b]	0.25	1	97.9	1.3	0.8
Meropenem-vaborbactam[b]	13,093	0.5	8	91.6[b]		8.4[b]
Piperacillin-tazobactam	14,302	4	>64	80.2	5.8	14.0
Meropenem	14,303	0.5	8	80.6	5.8	13.6
Imipenem	14,298	1	8	79.3	4.2	16.4
Ceftazidime	14,303	2	32	84.2	4.0	11.8
Cefepime	14,302	2	16	85.3	9.3	5.4
Levofloxacin	14,288	0.5	>4	68.7	9.9	21.4
Tobramycin	14,302	0.5	2	89.0	4.4	6.6

[a] Imipenem-relebactam tested only from 2020 to 2024.
[b] Meropenem-vaborbactam is not approved by the US FDA for treatment of *P. aeruginosa* infections; susceptibility based on EUCAST criteria.
Abbreviations: CLSI, Clinical and Laboratory Standards Institute; S, susceptible; I, intermediate, and R, resistant.

Table 2. Antimicrobial susceptibility of *P. aeruginosa*

Antimicrobial agent	% Susceptible per CLSI (no. of isolates)				
	All isolates		Resistant subsets (2015–2024)		
	2015 (1,354)	2024 (1,122)	MDR (2,459)	XDR (691)	DTR (568)
Ceftazidime-avibactam	98.2	96.8	82.9	66.1	65.3
Ceftolozane-tazobactam	98.6	97.5	84.4	64.1	71.9
Imipenem-relebactam[a]	[a]	98.7	87.4	60.7	64.8
Piperacillin-tazobactam	83.0	77.5	18.6	3.6	0.0
Meropenem	81.4	81.5	22.5	3.8	0.0
Imipenem	79.7	79.4	26.2	6.7	0.0
Ceftazidime	86.6	82.2	33.3	5.5	0.0
Cefepime	88.3	84.2	30.5	5.8	0.0
Levofloxacin	65.1	76.4	17.9	4.9	0.0
Tobramycin	89.4	92.0	56.7	41.2	46.5

[a] Imipenem-relebactam tested only from 2020 to 2024. A total of 5,914 isolates were tested, including 741 MDR, 122 XDR, and 165 DTR.

Figure 1. Frequency of nonsusceptibility to selected agents

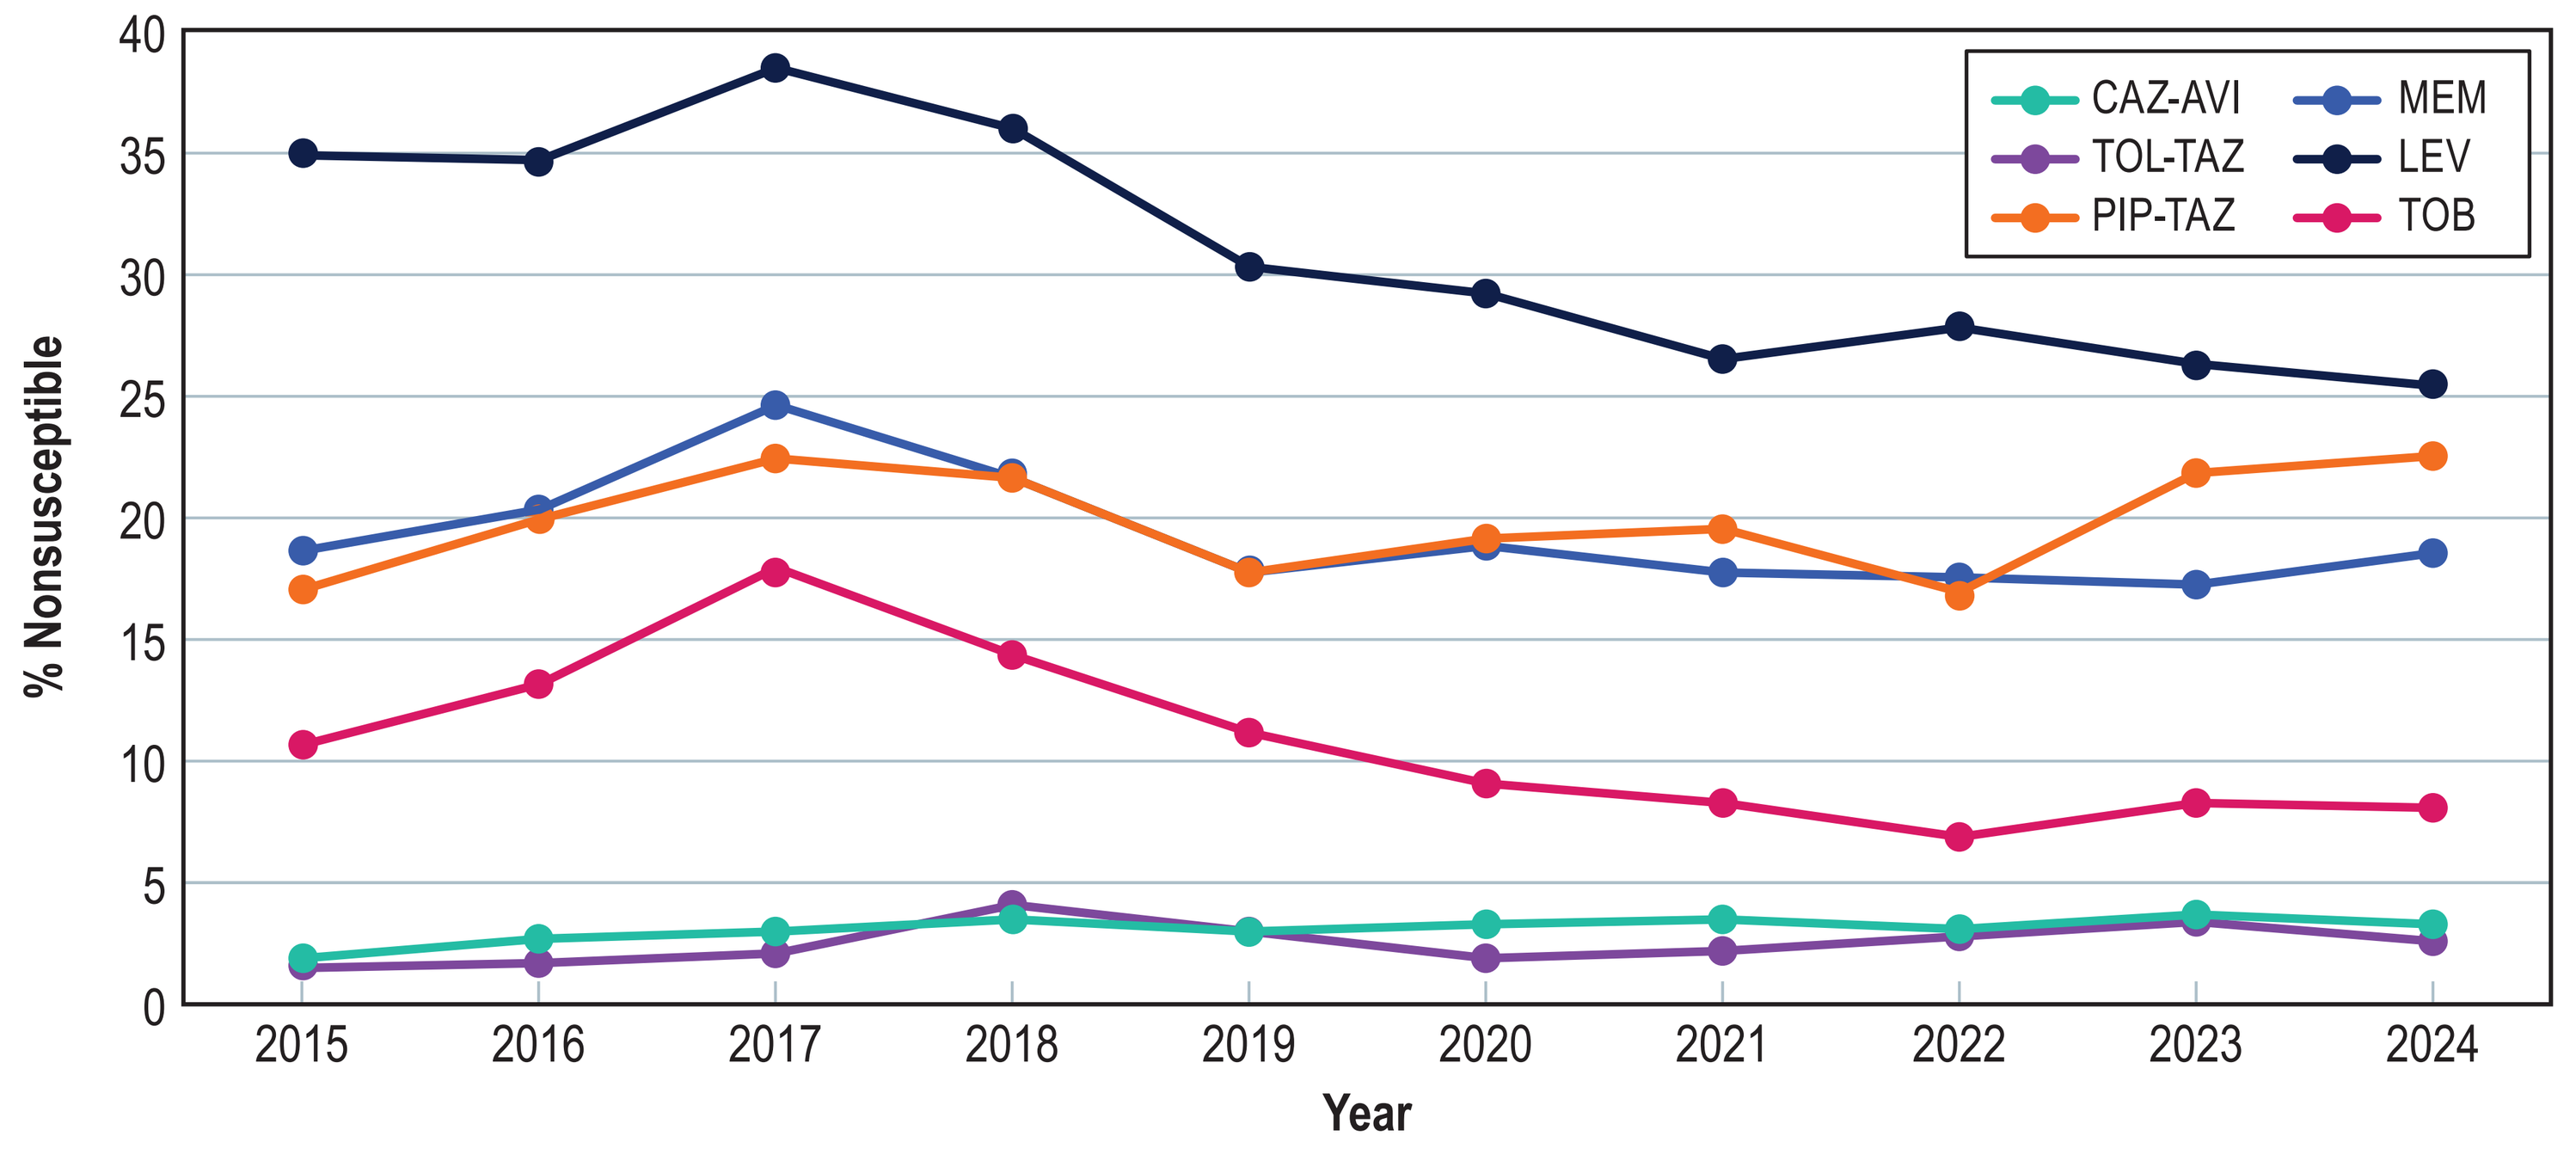
% Nonsusceptible
Year
CAZ-AVI
TOL-TAZ
PIP-TAZ
MEM
LEV
TOB

Abbreviations: CAZ-AVI, ceftazidime-avibactam; TOL-TAZ, ceftolozane-tazobactam; PIP-TAZ, piperacillin-tazobactam; MEM, meropenem; LEV, levofloxacin; TOB, tobramycin.

Figure 2. Frequency of resistant subsets

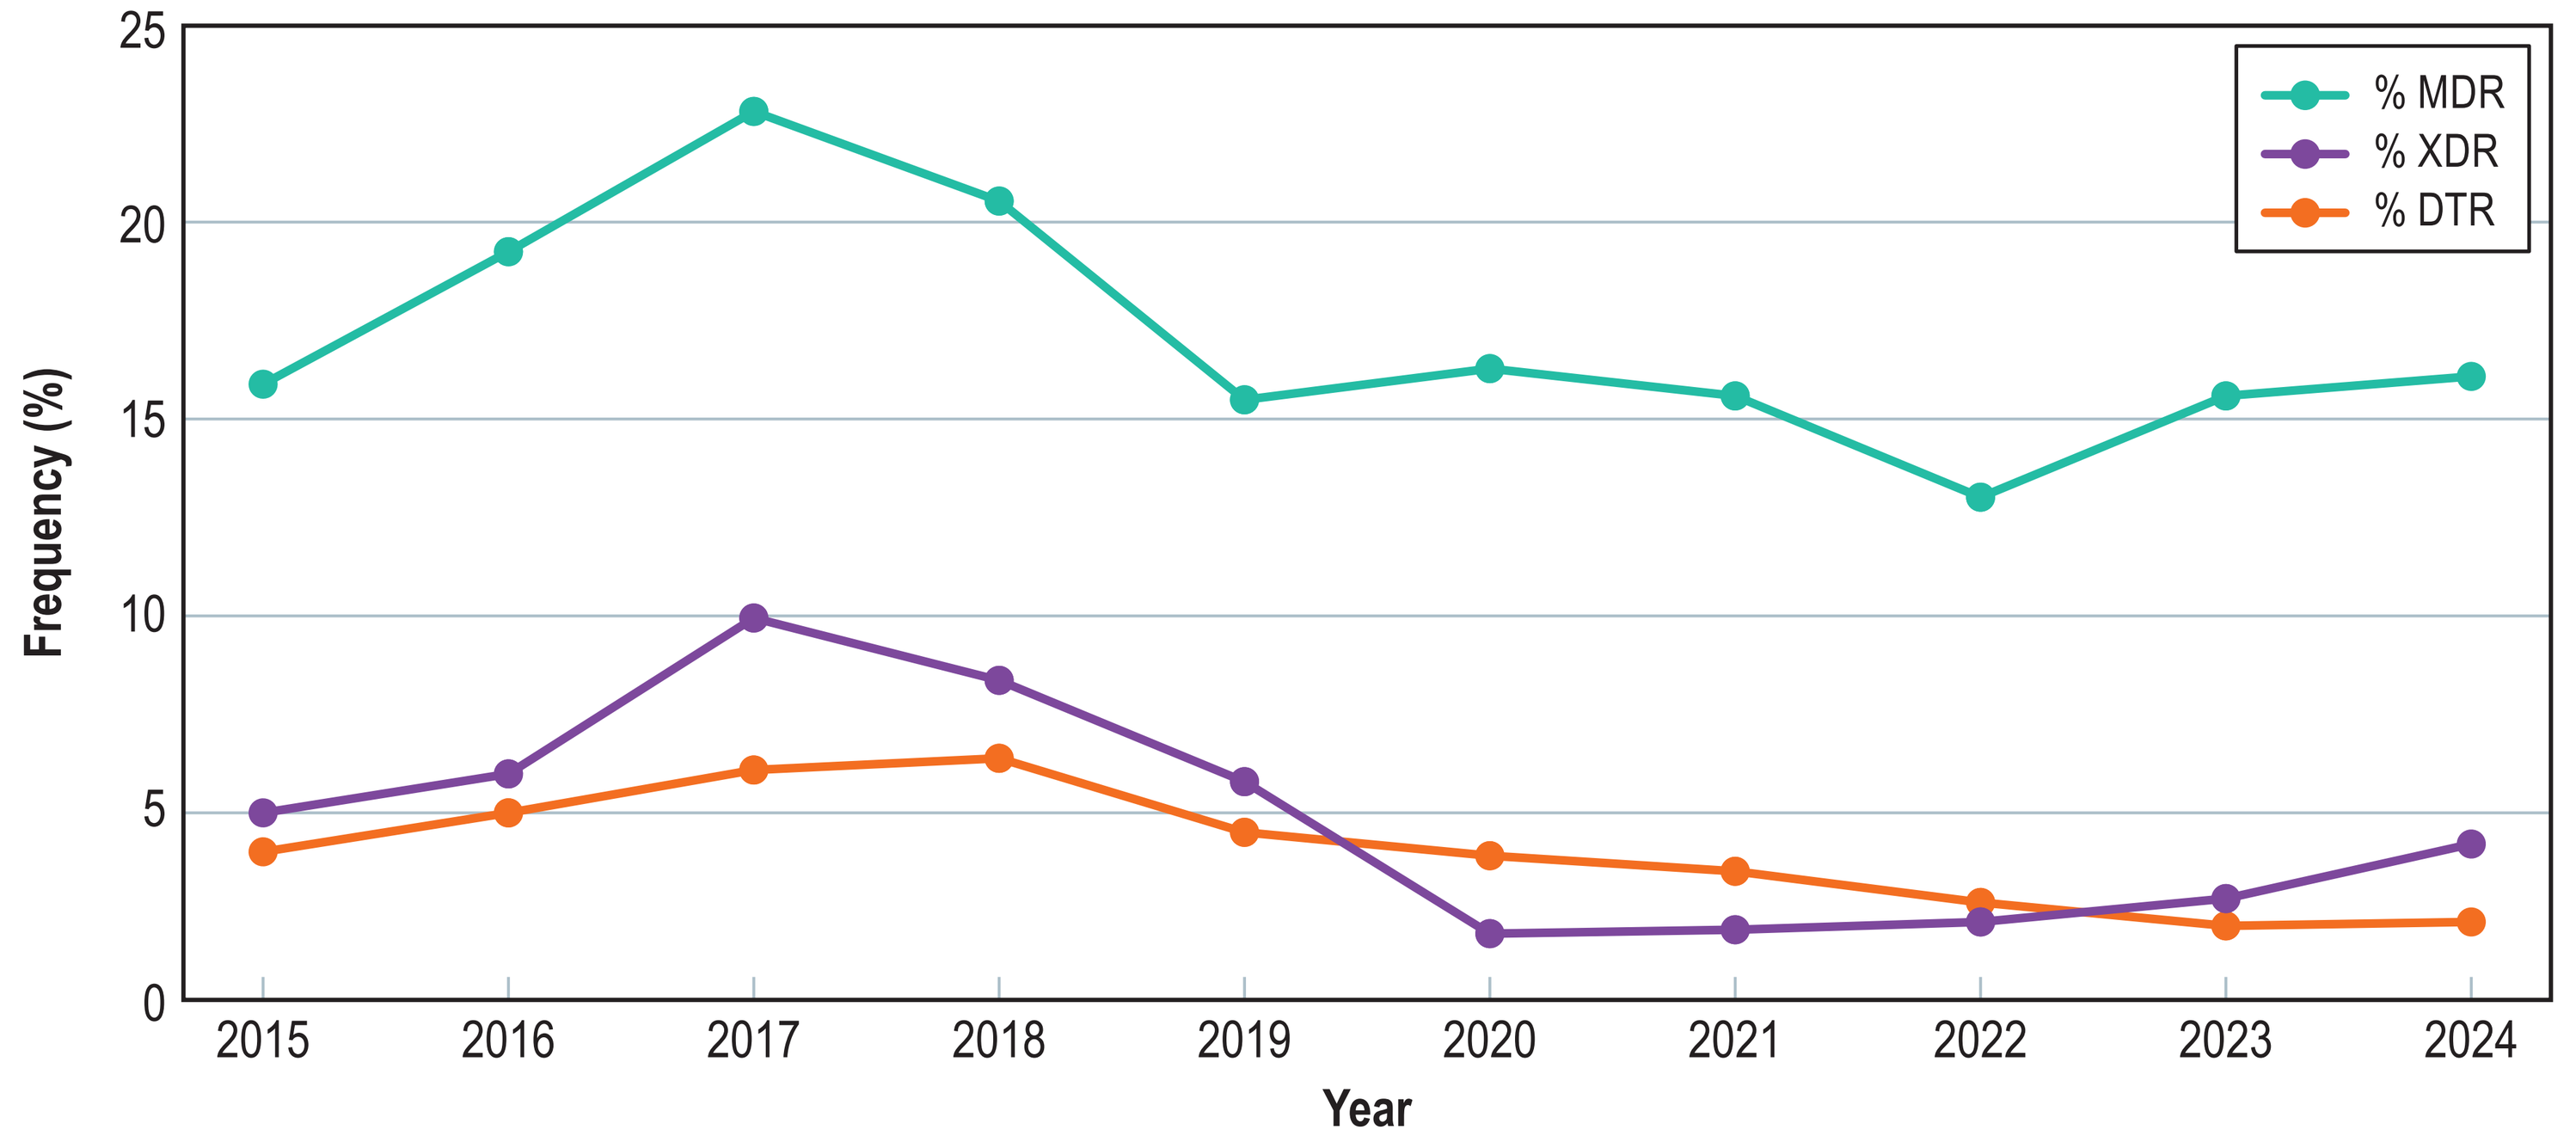
Frequency (%)
Year
% MDR
% XDR
% DTR

Abbreviations: MDR, multidrug-resistant; XDR, extensively drug-resistant; DTR, difficult-to-treat resistant.

Figure 3. Susceptibility to selected antimicrobial agents stratified by infection type

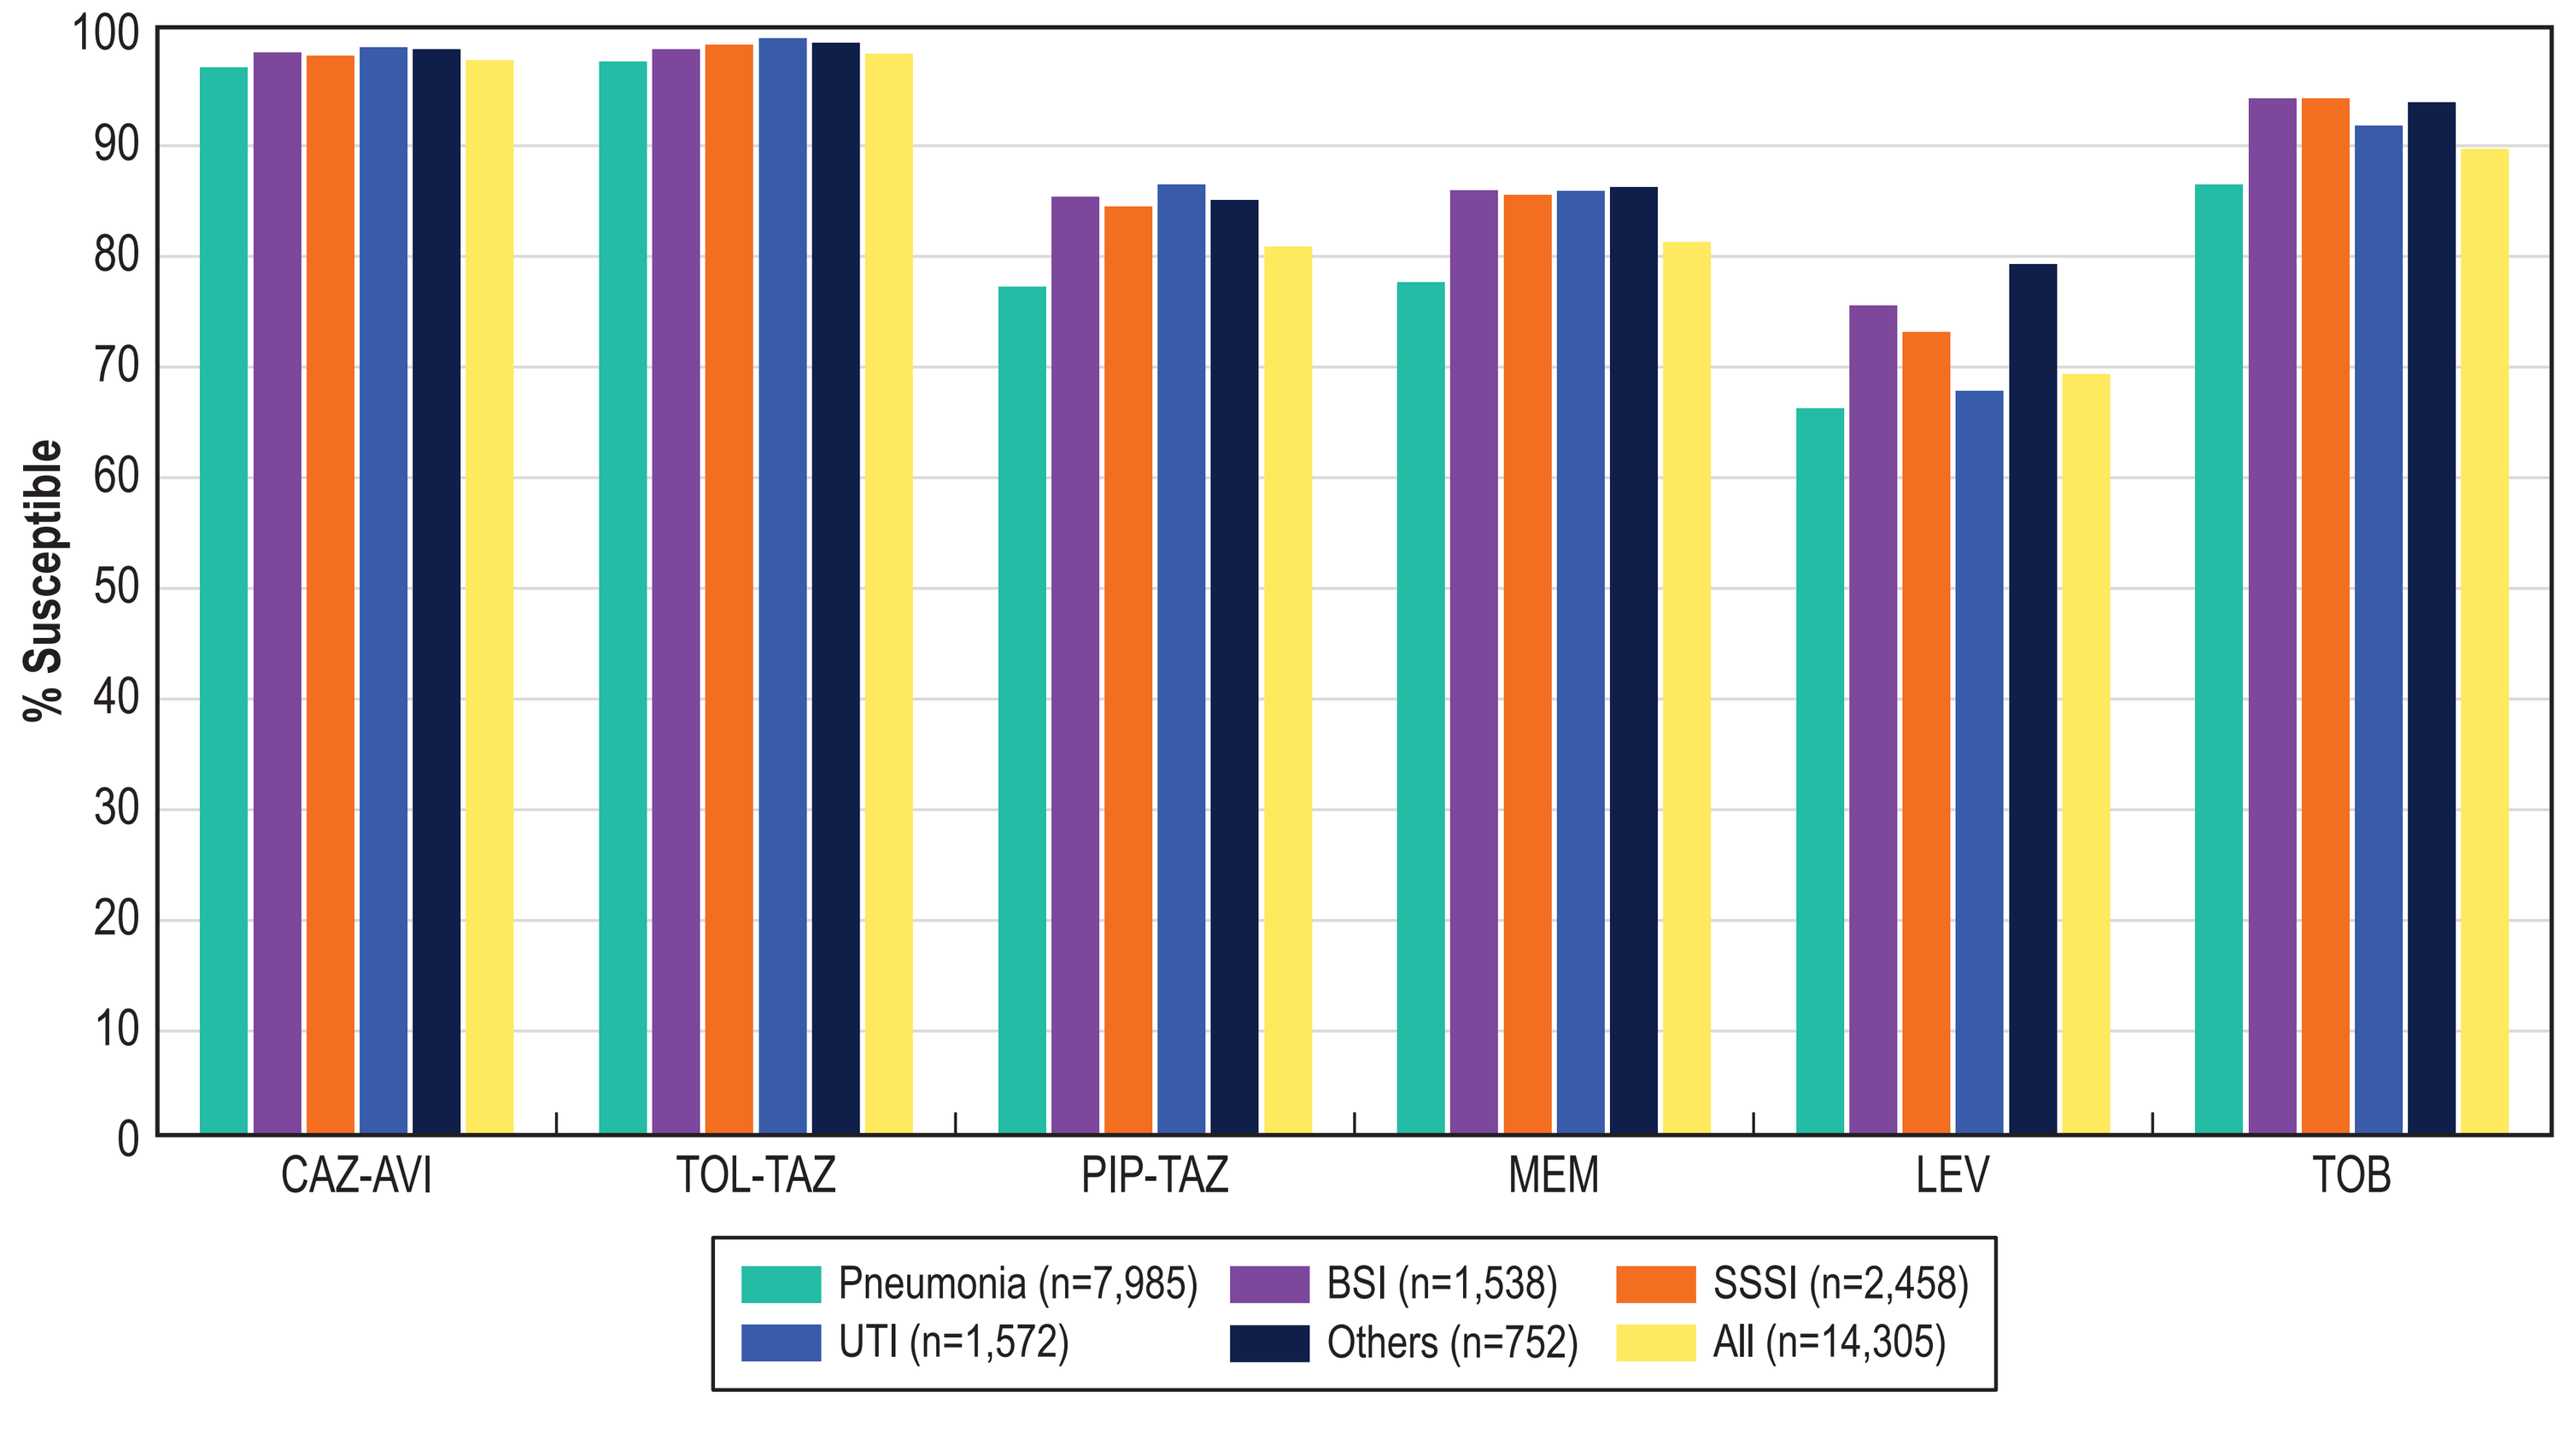
% Susceptible
CAZ-AVI
TOL-TAZ
PIP-TAZ
MEM
LEV
TOB
Pneumonia (n=7,985)
BSI (n=1,538)
SSSI (n=2,458)
UTI (n=1,572)
Others (n=752)
All (n=14,305)

Abbreviations: CAZ-AVI, ceftazidime-avibactam; TOL-TAZ, ceftolozane-tazobactam; PIP-TAZ, piperacillin-tazobactam; MEM, meropenem; LEV, levofloxacin; TOB, tobramycin; BSI, bloodstream infection; SSSI, skin and skin structure infection; UTI, urinary tract infection.

Contact Information

Helio S. Sader, MD, PhD
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

Acknowledgements

This study at JMI Laboratories was supported by AbbVie Inc. (New York, NY). JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by AbbVie Inc.

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2025 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2025_25-ALG-02_P2_PSA_10-year.pdf

SCAN ME

To submit a medical question, please visit www.abbviemedinfo.com

References

1. CLSI. 2025. M100Ed35. Performance standards for antimicrobial susceptibility testing: 35th informational supplement. Clinical and Laboratory Standards Institute.
2. Tamma PD, Aitken SL, Bonomo RA, *et al.* Infectious Diseases Society of America 2022 guidance on the treatment of extended-spectrum β-lactamase producing Enterobacterales (ESBL-E), carbapenem-resistant enterobacterales (CRE), and *Pseudomonas aeruginosa* with difficult-to-treat resistance (DTR). *Clin Infect Dis* 2022;75(2):187–212.
3. Tamma PD, Heil EL, Justo JA *et al.* Infectious Diseases Society of America 2024 guidance on the treatment of antimicrobial-resistant Gram-negative infections. *Clin Infect Dis* 2024 ciae403.