IDWEEK 2025 | Poster #P1316

Activity of Aztreonam-Avibactam and Comparators against Difficult-to-Treat Resistant (DTR) Enterobacterales from United States Medical Centers (2020–2024)

Helio S. Sader[1], Rodrigo E. Mendes[1], Marisa L. Winkler[1], Dmitri Debabov[2], John H. Kimbrough[1], Mariana Castanheira[1]

[1]Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA; [2]AbbVie Inc., Chicago, Illinois, USA

CONCLUSIONS

ATM-AVI retained potent activity against DTR Enterobacterales from US medical centers and its activity was not adversely affected by clinically relevant CBases or resistance to agents used to treat multidrug-resistant Enterobacterales.

The activities of CAZ-AVI, MEM-VAB, and IMI-REL were compromised by the increased occurrence of MBL producers among DTR and CRE isolates.

INTRODUCTION

- Aztreonam-avibactam (ATM-AVI) was recently approved by the US FDA for treatment of complicated IAI and by the EMA in the European Union for treatment of adults with complicated IAI, complicated urinary tract infection (cUTI), hospital-acquired pneumonia, including ventilator-associated pneumonia, and infections due to aerobic Gram-negative bacteria in adults with limited treatment options.
- Aztreonam-avibactam (ATM-AVI) has demonstrated potent activity against multidrug-resistant (MDR) Enterobacterales worldwide, including metallo-β-lactamase (MBL) producers.
- We evaluated the activity of ATM-AVI and comparators against DTR Enterobacterales from US medical centers.

Table 1. Antimicrobial susceptibility of selected resistant subsets

Antimicrobial agent	% Susceptible per CLSI or US FDA criteria (no. of isolates)					
	DTR (307)	CAZ-AVI-NS DTR (89)	MEM-VAB-NS DTR (106)	Cefiderocol-NS DTR (20)	MBL-producer DTR (91)	CRE (450)
ATM-AVI	98.0	95.5	96.2	90.0	97.8	97.1
CAZ-AVI	71.0	0.0	21.7	20.0	5.5	74.4
MEM-VAB	65.5	6.7	0.0	20.0	8.8	70.9
IMI-REL	63.2	3.4	5.7	15.0	2.2	66.4
Cefiderocol	93.5	82.0	84.9	0.0	84.6	94.0
Gentamicin	54.4	59.6	53.8	65.0	59.3	61.6
Amikacin	57.0	56.2	51.9	65.0	54.9	66.2

Abbreviations: DTR, difficult-to-treat resistant; CAZ-AVI, ceftazidime-avibactam; NS, nonsusceptible; MEM-VAB, meropenem-vaborbactam; MBL, metallo-β-lactamase; CRE, carbapenem-resistant Enterobacterales; ATM-AVI, aztreonam-avibactam; IMI-REL, imipenem-relebactam.

Figure 1. Distribution of carbapenemase types among DTR and CRE isolates

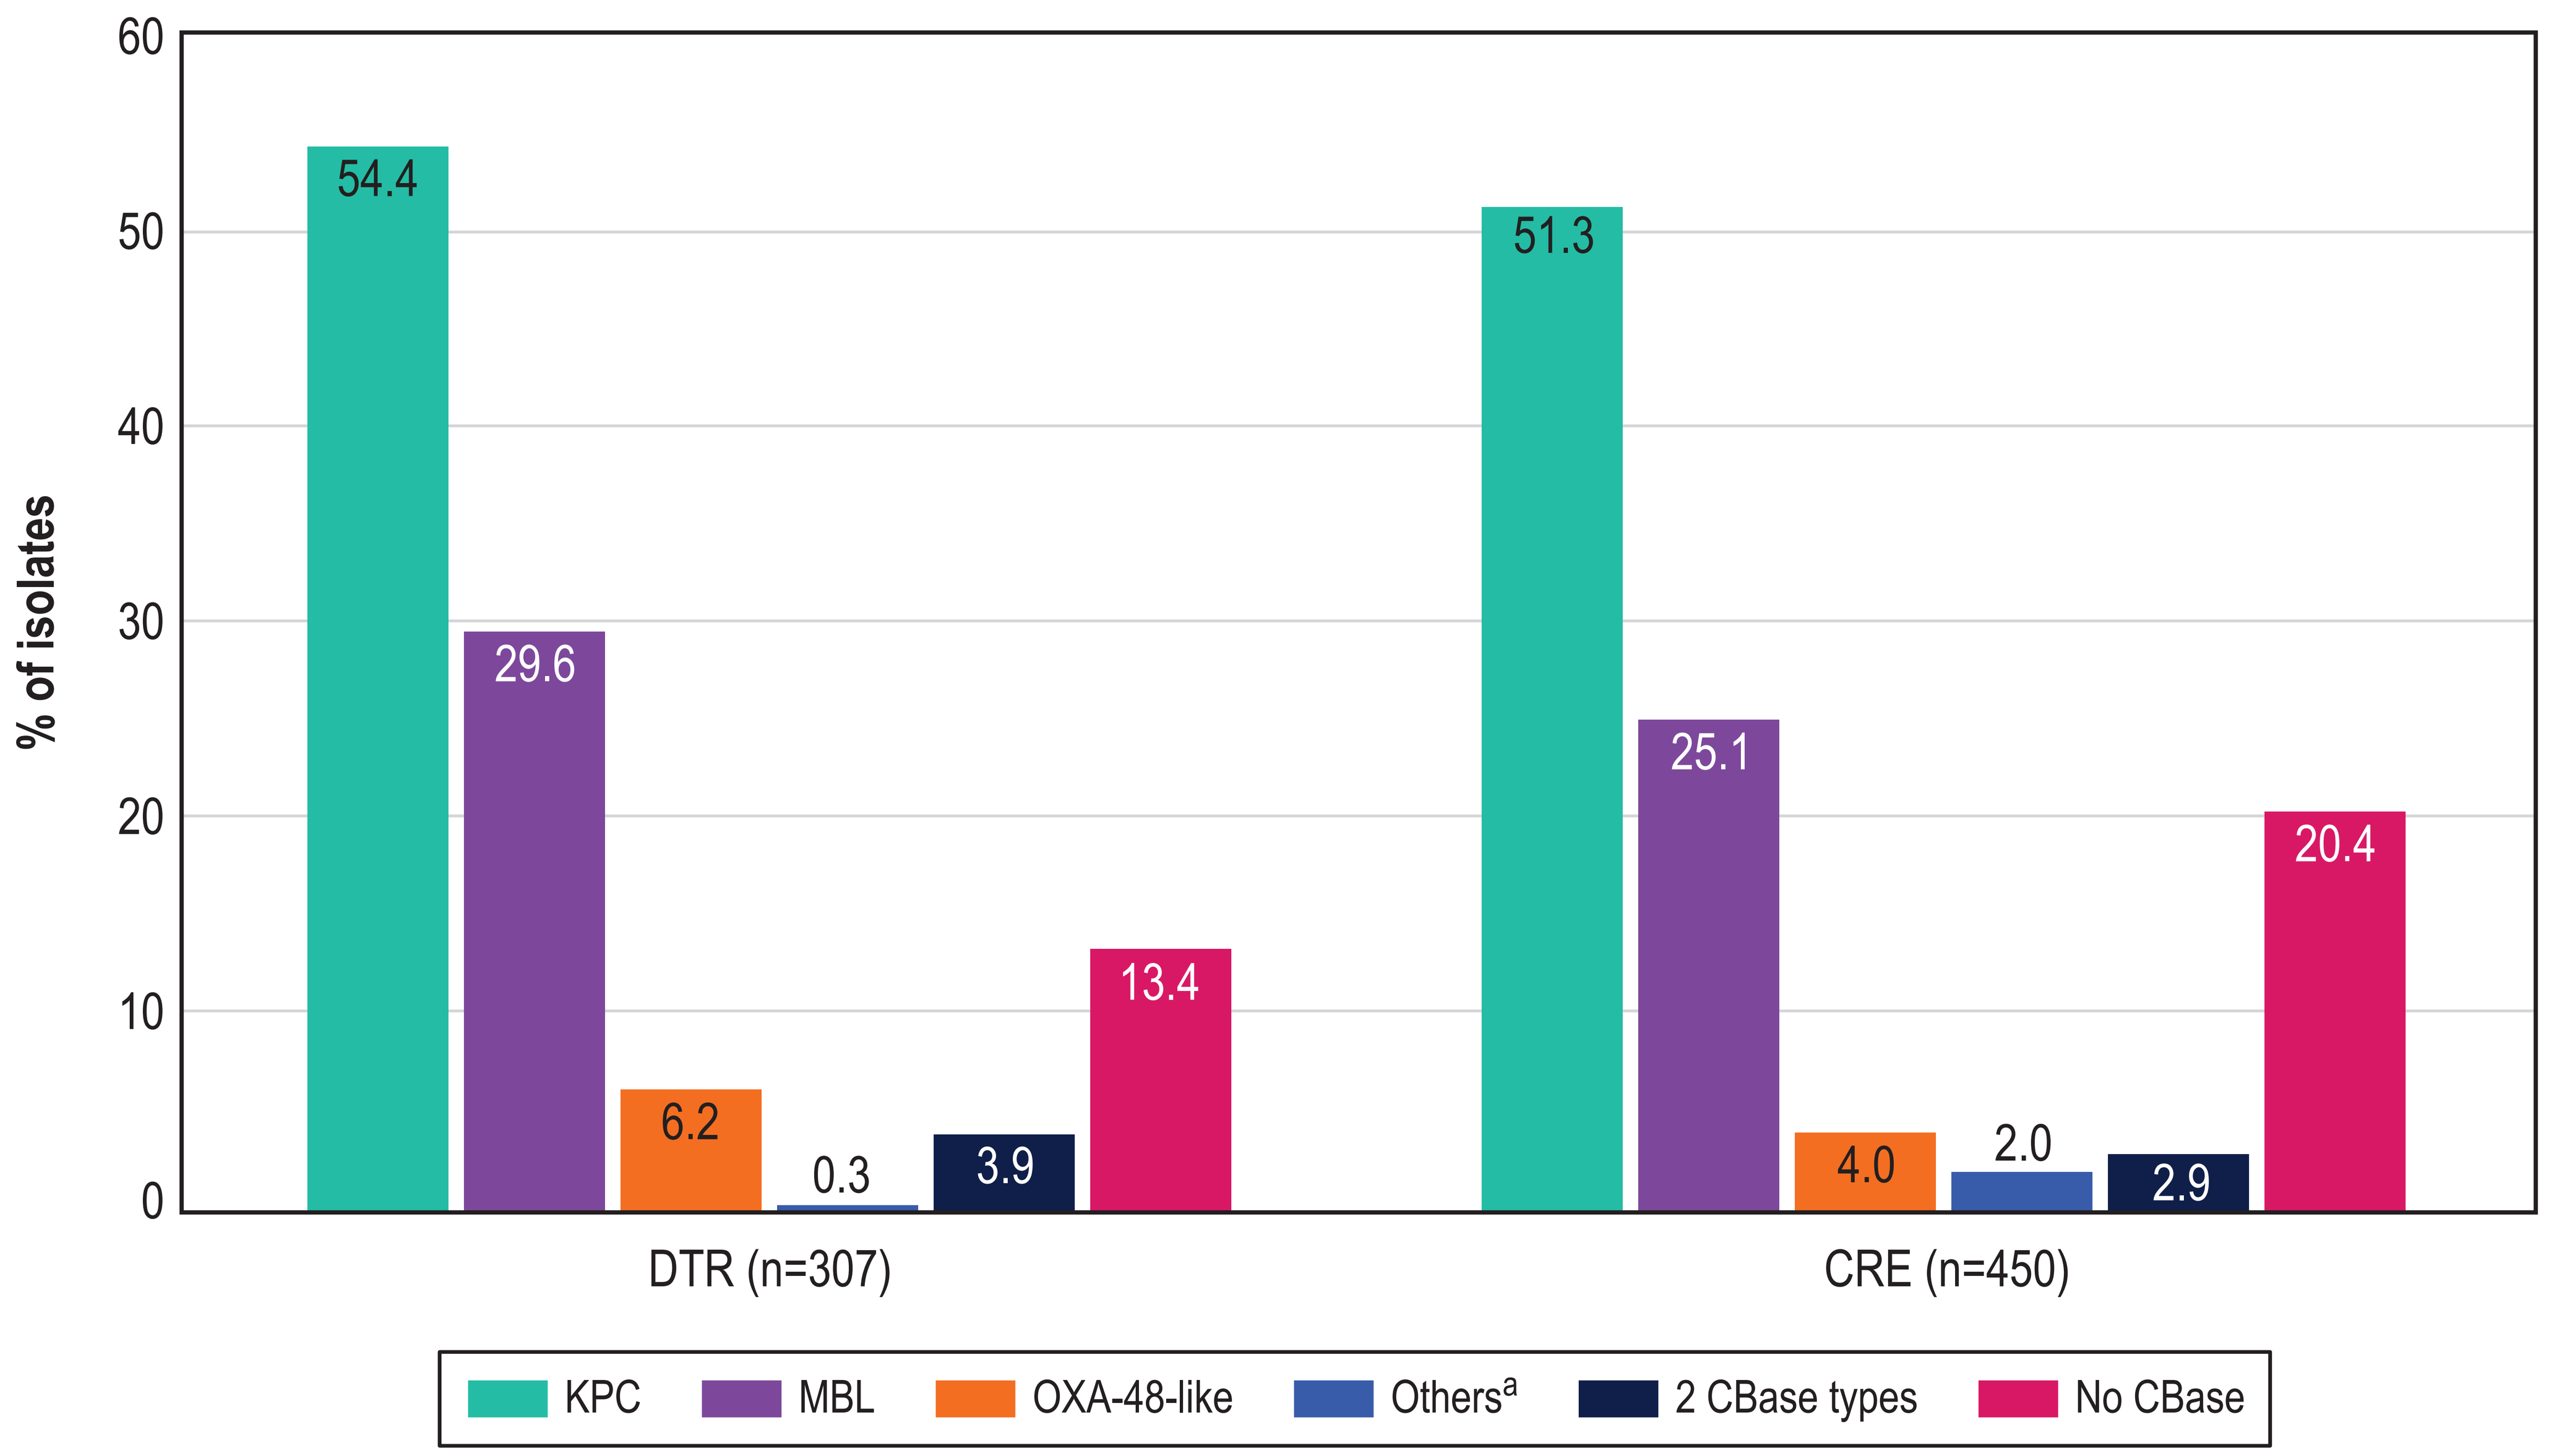
% of isolates
0
10
20
30
40
50
60
54.4
29.6
6.2
0.3
3.9
13.4
51.3
25.1
4.0
2.0
2.9
20.4
DTR (n=307)
CRE (n=450)
KPC
MBL
OXA-48-like
Others[a]
2 CBase types
No CBase

[a] Includes GES-6 (1 isolate) for DTR and SME-2 (5), SME-4 (2), GES-6 (1), and IMI-4 (1) for CRE.
Abbreviations: DTR, difficult-to-treat resistant; CRE, carbapenem-resistant Enterobacterales; KPC, *Klebsiella pneumoniae* carbapenemase; MBL, metallo-β-lactamases; OXA, oxacillinase; CBase, carbapenemase.

METHODS

- A total of 42,295 Enterobacterales isolates were consecutively collected (1/patient) from 85 US medical centers in 2020–2024.
- The collection included 450 carbapenem-resistant (CRE) and 307 difficult-to-treat resistant (DTR) Enterobacterales.
 - CRE was defined as resistant to meropenem and/or imipenem.
 - DTR was defined as a CRE isolate resistant to fluoroquinolones (levofloxacin and/or ciprofloxacin).
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of infection were included in the study.
- Isolates were susceptibility tested by CLSI M07 broth microdilution method.
- The ATM-AVI susceptible breakpoint of ≤4 mg/L, which was established by the US FDA and EMA for Enterobacterales, was applied.
- All CRE isolates were screened for β-lactamase genes by whole genome sequencing.

RESULTS

- ATM-AVI was active (MIC ≤4 mg/L) against 98.0% of DTR ($MIC_{50/90}$, 0.25/1 mg/L) and 97.1% of CRE ($MIC_{50/90}$, 0.25/1 mg/L) isolates (Table 1).
- ATM-AVI retained potent activity against DTR isolates nonsusceptible to ceftazidime-avibactam (CAZ-AVI; 95.5% susceptible [S]; $MIC_{50/90}$, 0.25/1 mg/L), meropenem-vaborbactam (MEM-VAB; 96.2% S; $MIC_{50/90}$, 0.25/1 mg/L), and/or cefiderocol (90.0% S; $MIC_{50/90}$, 0.5/4 mg/L; Table 1).
- Cefiderocol was active against 93.5% of DTR isolates, whereas CAZ-AVI, MEM-VAB, IMI-REL, and the aminoglycosides exhibited limited activity against these organisms (Table 1).
- The most common carbapenemase (CBase) genes identified among DTR (n=307) isolates were bla_{KPC} (54.4% of isolates) and bla_{NDM} (28.0%).
- DTR and CRE isolates exhibited similar frequencies of CBase types (Figure 1).
- An MBL gene was observed in 29.6% of DTR and 25.1% of CRE isolates (Figure 1).
- ATM-AVI ($MIC_{50/90}$, 0.12/0.5 mg/L) and cefiderocol ($MIC_{50/90}$, 2/8 mg/L) were active against 98.2% and 84.1% of MBL producers, respectively, whereas MEM-VAB and IMI-REL exhibited poor activity against these organisms (Figure 2).

Figure 2. Susceptibility stratified by β-lactamase type

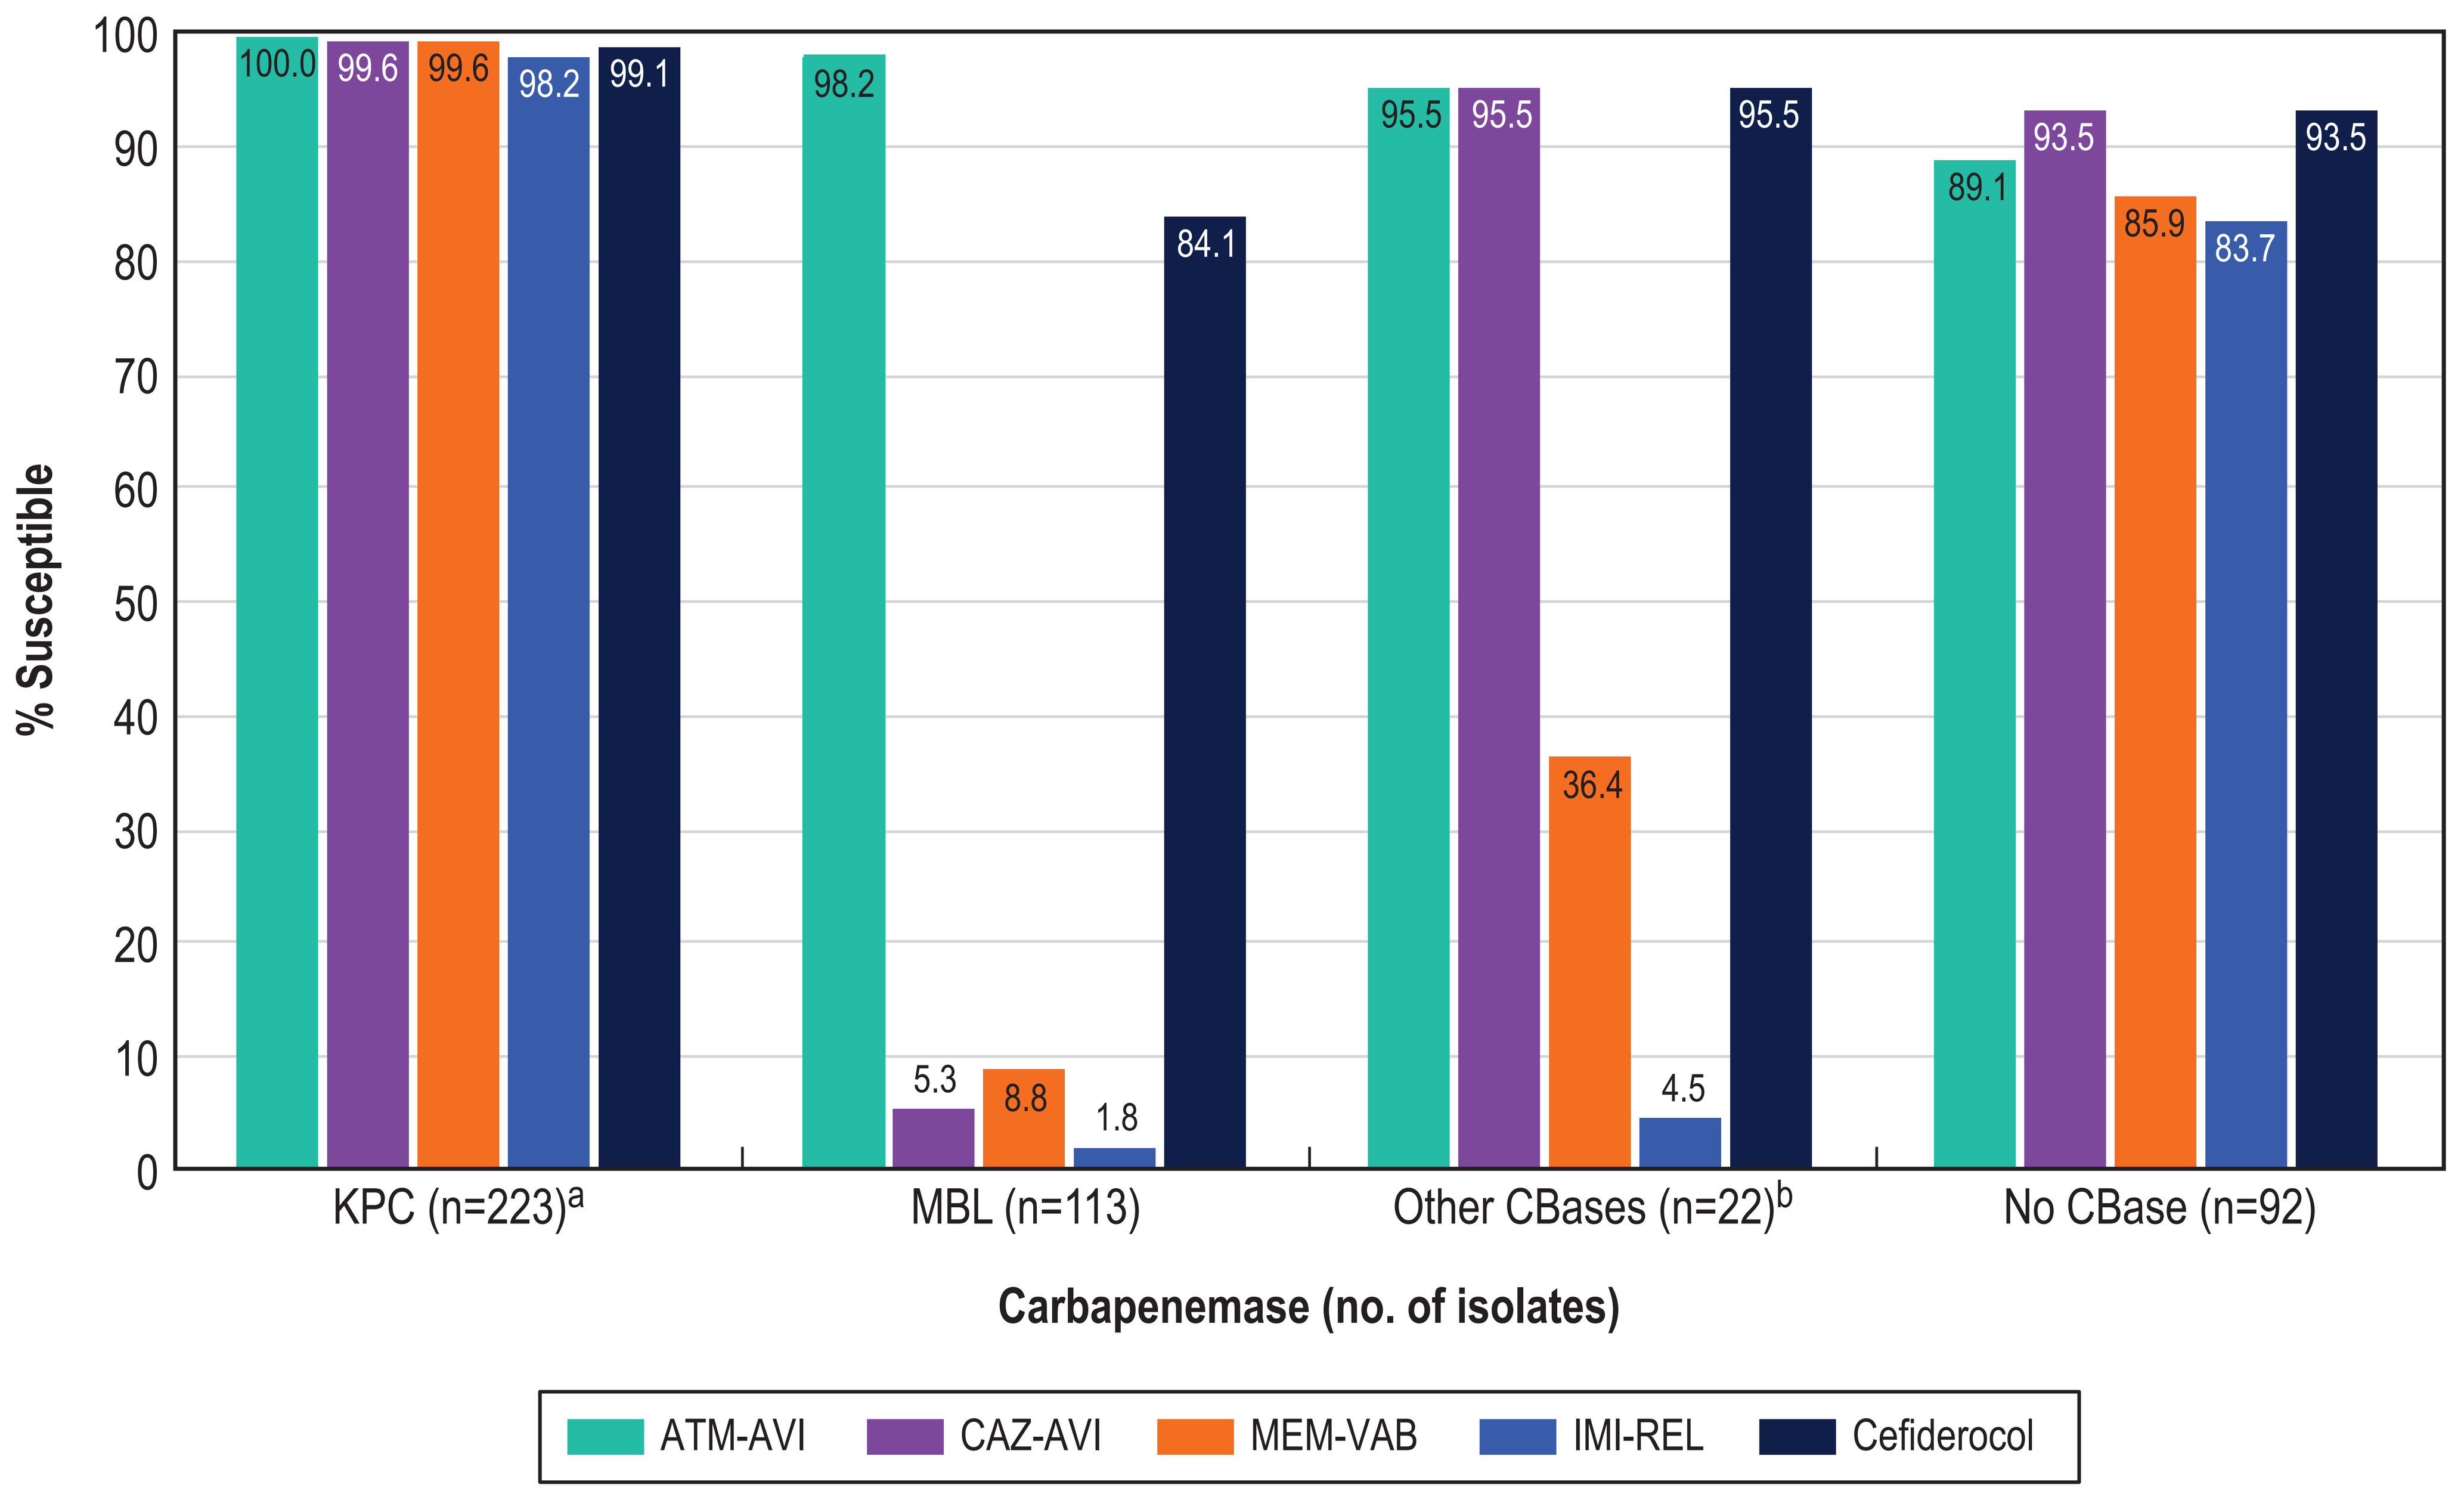
% Susceptible
0
10
20
30
40
50
60
70
80
90
100
100.0
99.6
99.6
98.2
99.1
98.2
5.3
8.8
1.8
84.1
95.5
95.5
36.4
4.5
95.5
89.1
93.5
85.9
83.7
93.5
KPC (n=223)[a]
MBL (n=113)
Other CBases (n=22)[b]
No CBase (n=92)
Carbapenemase (no. of isolates)
ATM-AVI
CAZ-AVI
MEM-VAB
IMI-REL
Cefiderocol

[a] Excludes MBL co-producers.
[b] Includes OXA-48-like (14), SME type (7), and IMI-4 (1); and excludes MBL co-producers.
Abbreviations: KPC, *Klebsiella pneumoniae* carbapenemase; MBL, metallo-β-lactamases; CBase, carbapenemase; ATM-AVI, aztreonam-avibactam; CAZ-AVI, ceftazidime-avibactam; MEM-VAB, meropenem-vaborbactam; IMI-REL, imipenem-relebactam.

Contact Information

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

Acknowledgements

This study at JMI Laboratories was supported by AbbVie Inc. (New York, NY). JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by AbbVie Inc.

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2025 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2025_25-ALG-02_P3_DTR.pdf

To submit a medical question, please visit www.abbviemedinfo.com

SCAN ME

References

1. CLSI. 2025. M100Ed35. Performance standards for antimicrobial susceptibility testing: 35th informational supplement. Clinical and Laboratory Standards Institute.
2. Kadri SS, Adjemian J, Lai YL, *et al.* Difficult-to-treat resistance in Gram-negative bacteremia at 173 US hospitals: Retrospective cohort analysis of prevalence, predictors, and outcome of resistance to all first-line agents. *Clin Infect Dis* 2018;67(12):1803–1814.
3. Sader HS, Mendes RE, Carvalhaes CG, Kimbrough JH, Castanheira M. Changing epidemiology of carbapenemases among carbapenem-resistant Enterobacterales from United States hospitals and the activity of aztreonam-avibactam against contemporary Enterobacterales (2019–2021). *Open Forum Infect Dis* 2023;10(2):ofad046.
4. Tamma PD, Heil EL, Justo JA *et al.* Infectious Diseases Society of America 2024 guidance on the treatment of antimicrobial-resistant Gram-negative infections. *Clin Infect Dis* 2024 ciae403.

abbvie