IDWEEK 2025 | Poster #P1319

Antimicrobial Activity of Aztreonam-Avibactam and Molecular Characterization of Enterobacterales Resistant to Ceftazidime-Avibactam and/or Meropenem-Vaborbactam from United States Medical Centers (2016–2024)

Helio S. Sader[1], John H. Kimbrough[1], Zachary Kockler[1], Dmitri Debabov[2], Rodrigo E. Mendes[1], Mariana Castanheira[1]

[1]Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA; [2]AbbVie Inc., Chicago, Illinois, USA

CONCLUSIONS

ATM-AVI demonstrated potent activity against isolates NS to CAZ-AVI and/or MEM-VAB as well as against CRE isolates, including MBL producers.

The frequency of MBL producers among CRE isolates increased markedly during the study period (2016–2024).

The activities of CAZ-AVI, MEM-VAB, and IMI-REL have been compromised by the increasing occurrence of MBL producers in US medical centers.

Contact Information

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

Acknowledgements

This study at JMI Laboratories was supported by AbbVie Inc. (New York, NY). JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by AbbVie Inc.

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2025 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2025_25-ALG-02_P4_Ar_CAZ-AVI_MEM-VAB-R.pdf

To submit a medical question, please visit www.abbviemedinfo.com

SCAN ME

References

1. CLSI. 2025. M100Ed35. Performance standards for antimicrobial susceptibility testing: 35th informational supplement. Clinical and Laboratory Standards Institute.
2. Sader HS, Mendes RE, Carvalhaes CG, Kimbrough JH, Castanheira M. Changing epidemiology of carbapenemases among carbapenem-resistant Enterobacterales from United States hospitals and the activity of aztreonam-avibactam against contemporary Enterobacterales (2019–2021). *Open Forum Infect Dis* 2023;10(2):ofad046.
3. Tamma PD, Heil EL, Justo JA *et al.* Infectious Diseases Society of America 2024 guidance on the treatment of antimicrobial-resistant Gram-negative infections. *Clin Infect Dis* 2024 ciae403.

INTRODUCTION

- Recently available β-lactamase inhibitor combination (BLIC) agents such as ceftazidime-avibactam (CAZ-AVI), meropenem-vaborbactam (MEM-VAB), and imipenem-relebactam (IMI-REL) represented a significant improvement in the treatment of CRE infections, especially with KPC producers.
- However, MEM-VAB and IMI-REL have limited activity against OXA-48–like producers and none of these newer BLIC agents are active against the MBLs, including the NDM.
- Aztreonam-avibactam (ATM-AVI) was recently approved by the US FDA and by the EMA in the European Union, and has demonstrated potent activity against CRE, including MBL producers.
- We evaluated activity of ATM-AVI and comparators against isolates nonsusceptible (NS) to ceftazidime-avibactam (CAZ-AVI) and/or meropenem-vaborbactam (MEM-VAB) and against MBL producers.

Figure 1. Finlandogram showing the activity of aztreonam-avibactam against selected resistant subsets

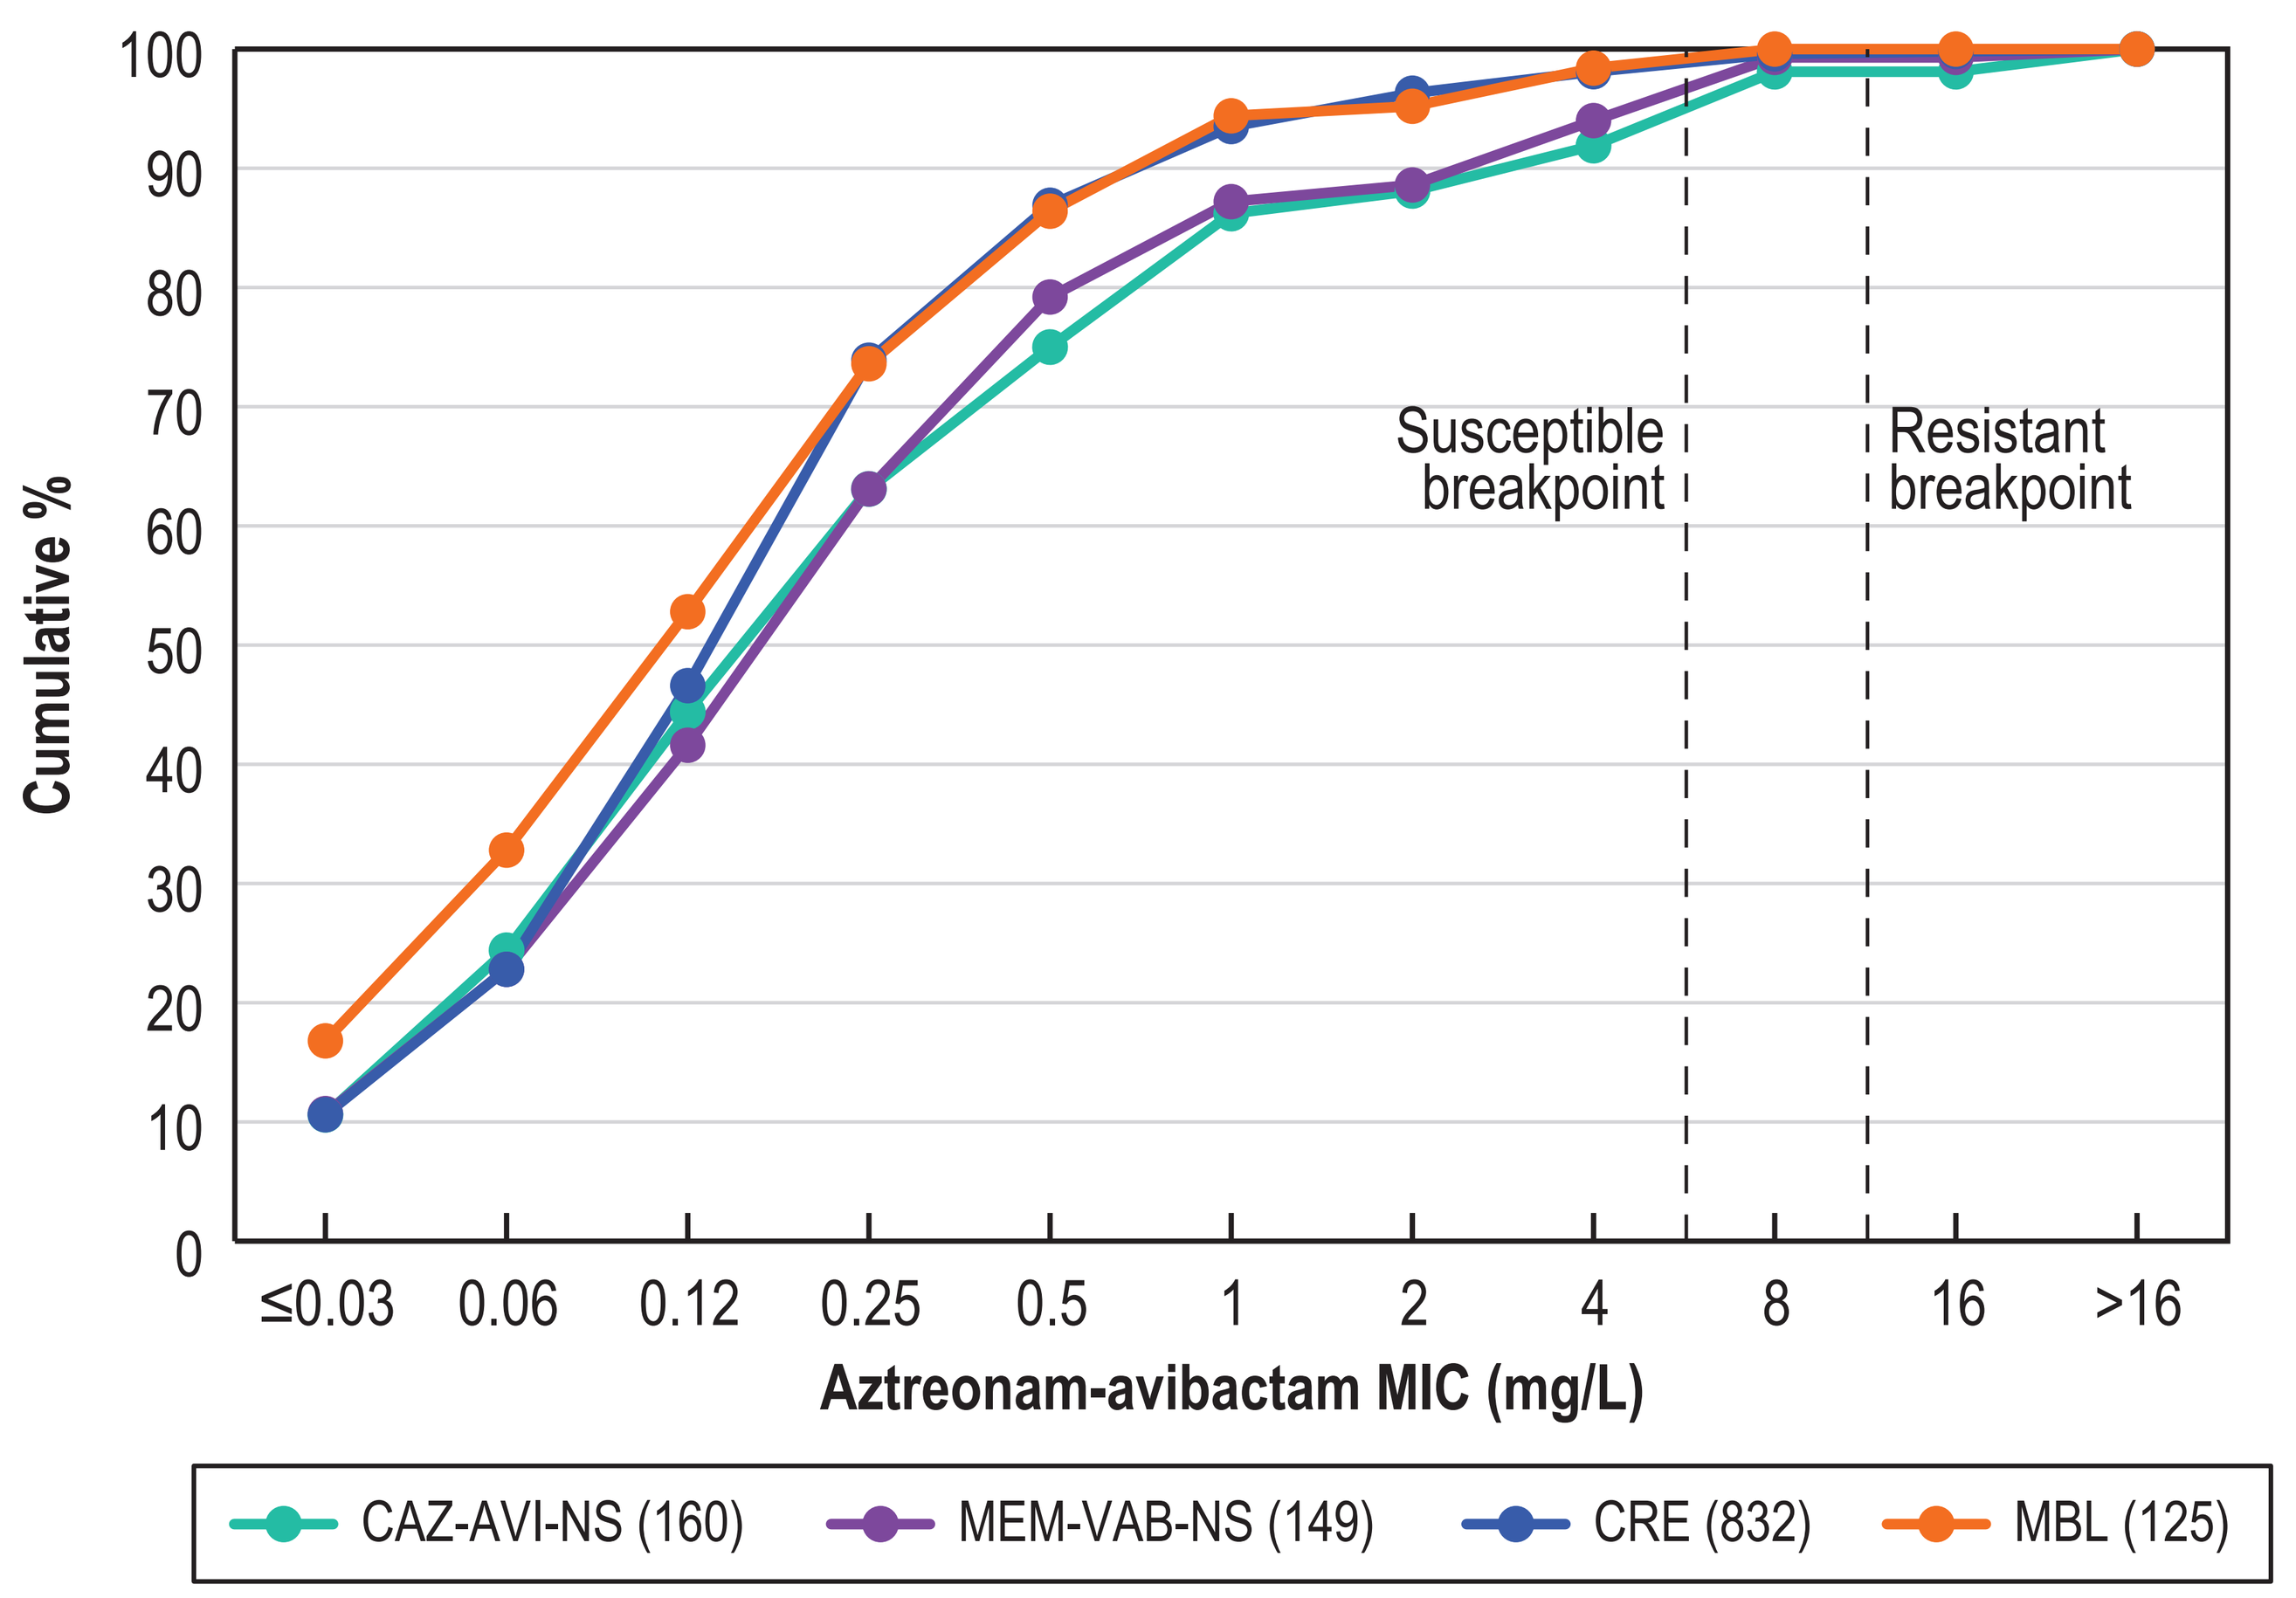
Cumulative %
100
90
80
70
60
50
40
30
20
10
0
≤0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
>16
Aztreonam-avibactam MIC (mg/L)
Susceptible breakpoint
Resistant breakpoint
CAZ-AVI-NS (160)
MEM-VAB-NS (149)
CRE (832)
MBL (125)

Abbreviations: CAZ-AVI, ceftazidime-avibactam; NS, nonsusceptible; MEM-VAB, meropenem-vaborbactam; CRE, carbapenem-resistant Enterobacterales; MBL, metallo-β-lactamase.

Figure 2. Distribution of carbapenemase types

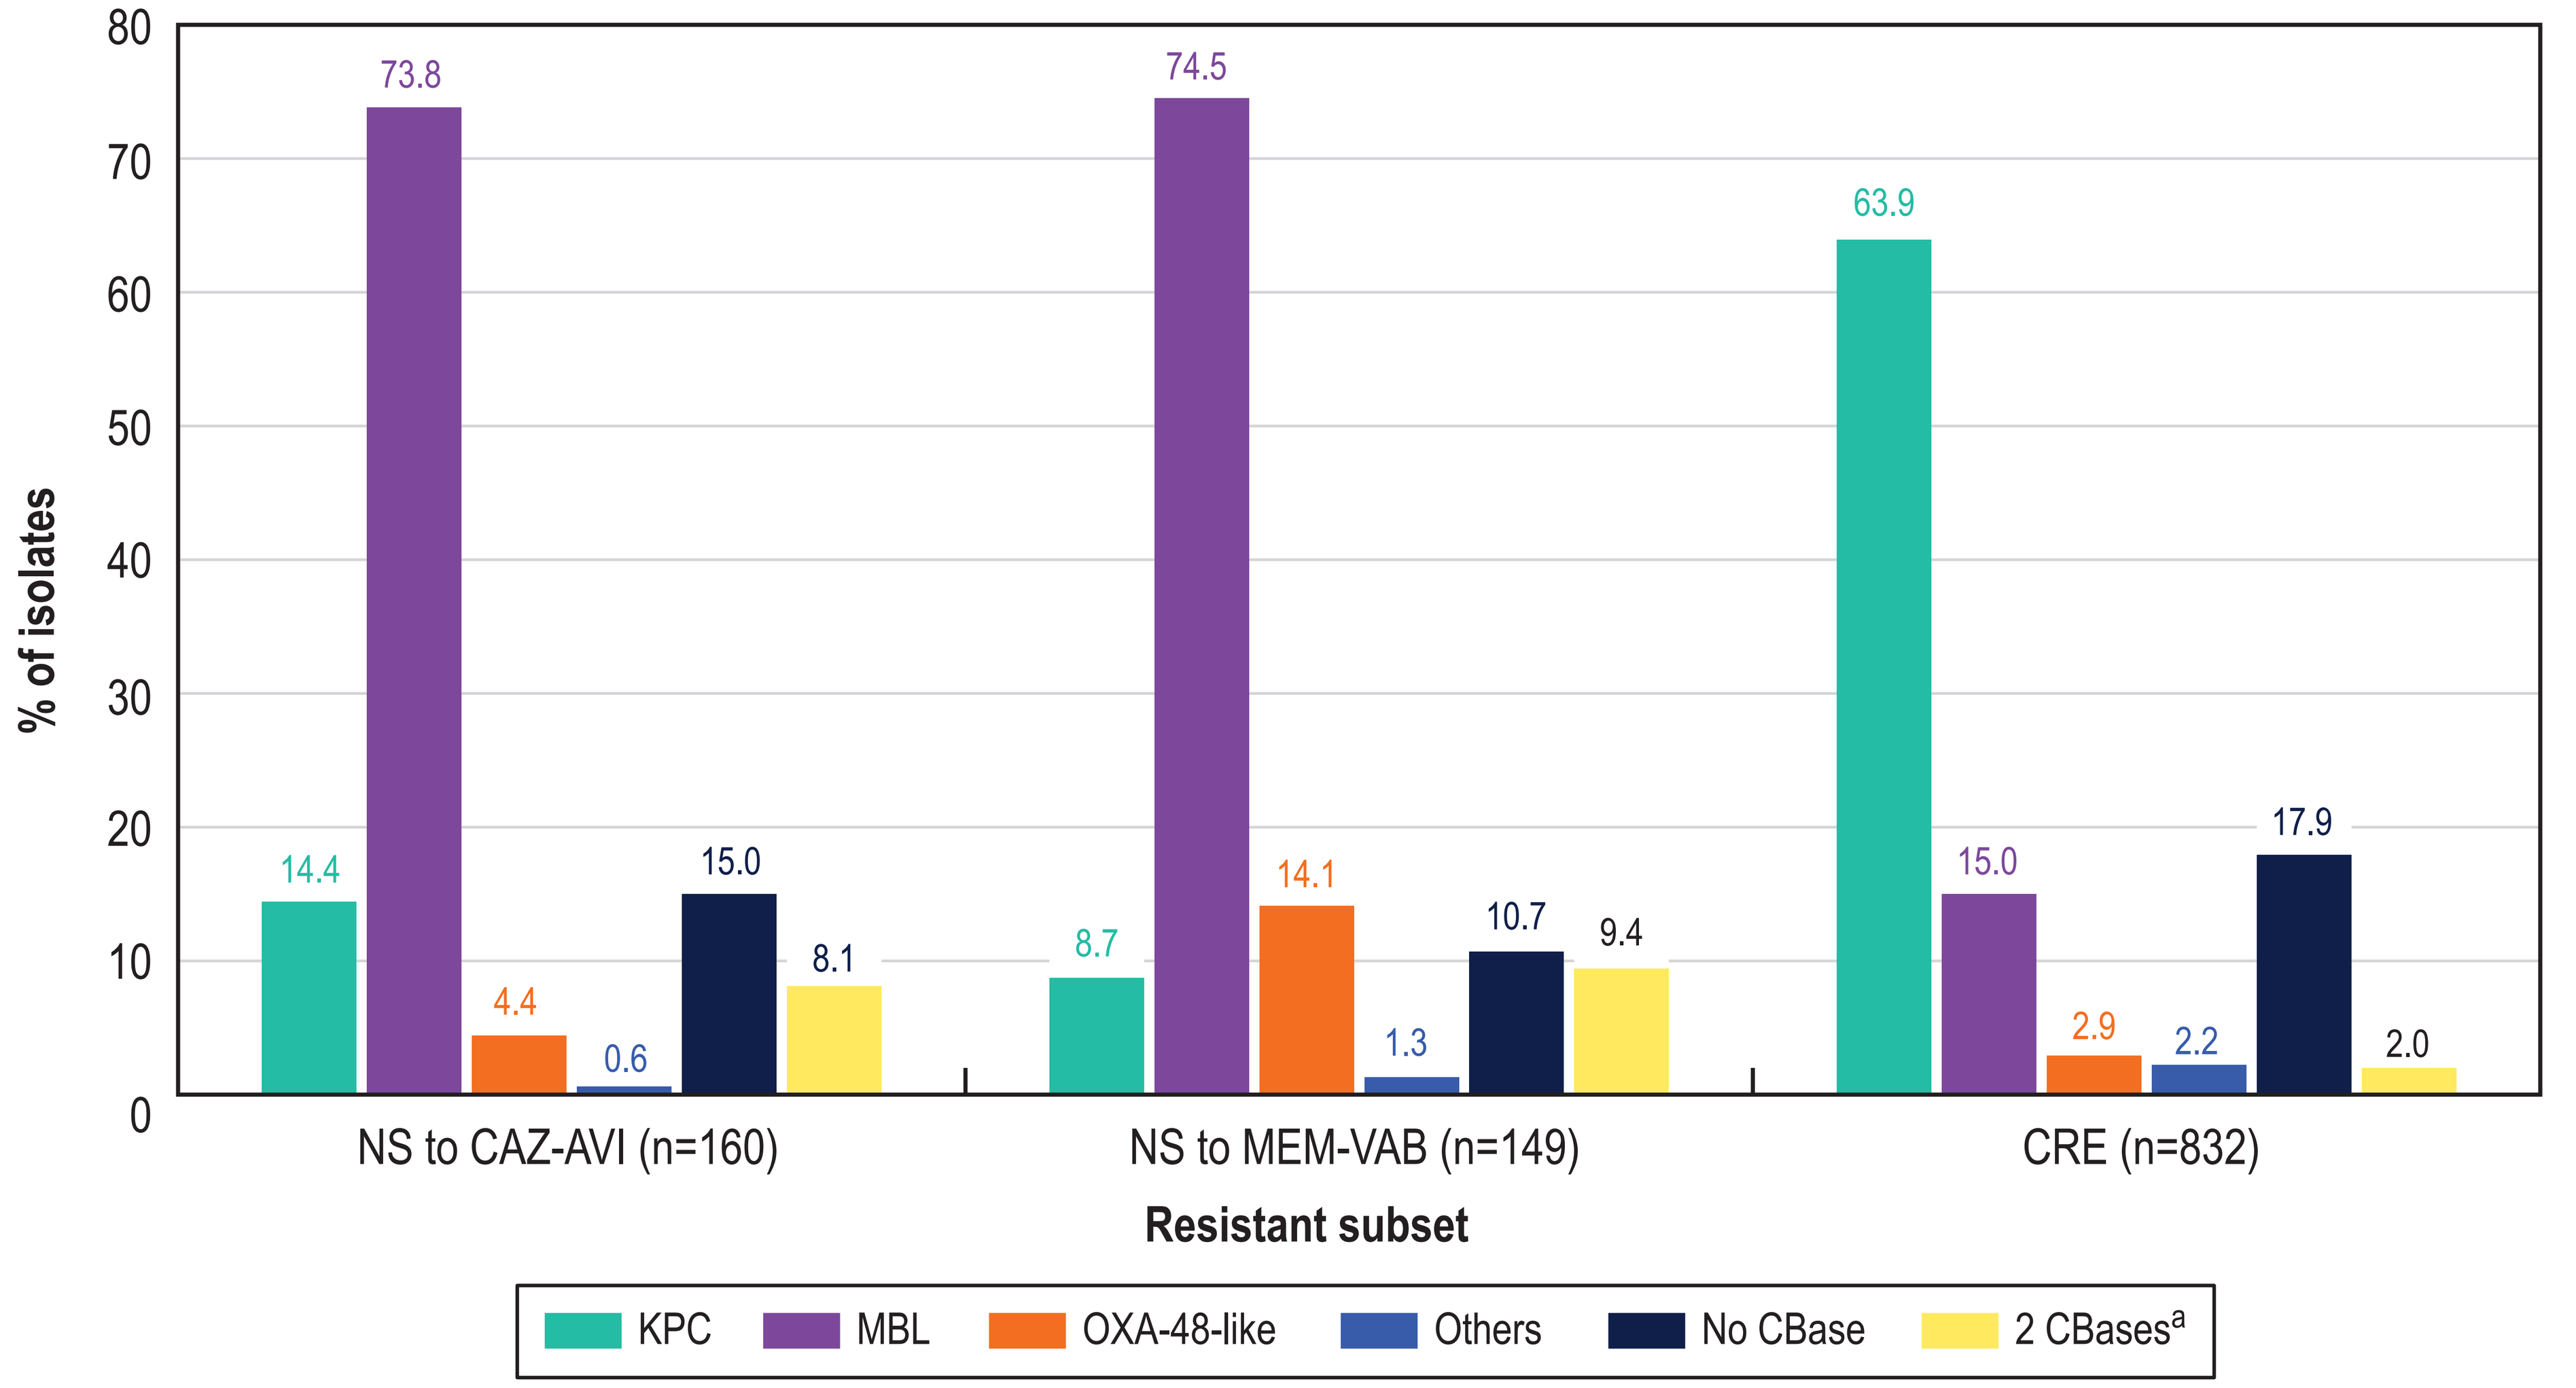
% of isolates
80
70
60
50
40
30
20
10
0
14.4
73.8
4.4
0.6
15.0
8.1
8.7
74.5
14.1
1.3
10.7
9.4
63.9
15.0
2.9
2.2
17.9
2.0
NS to CAZ-AVI (n=160)
NS to MEM-VAB (n=149)
CRE (n=832)
Resistant subset
KPC
MBL
OXA-48-like
Others
No CBase
2 CBases[a]

[a] Two carbapenemase genes were identified in 17 isolates, including NDM plus KPC (7), NDM plus OXA-48-like (7), KPC plus GES (1), KPC plus IMP (1), and 2 KPCs (1).

METHODS

- A total of 80,927 Enterobacterales isolates were consecutively collected (1/patient) in 2016–2024 from 103 US medical centers.
- Among these isolates, 160 (0.24%) were nonsusceptible (NS) to CAZ-AVI, 149 were NS to MEM-VAB, 115 (0.14%) were NS to both CAZ-AVI and MEM-VAB, and 832 isolates were carbapenem-resistant (CRE).
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of infection were included in the study.
- Isolates were susceptibility tested by CLSI M07 broth microdilution method.
- Cefiderocol was tested in iron-depleted media.
- The ATM-AVI susceptible breakpoint of ≤4 mg/L, which was established by the US FDA and EMA for Enterobacterales, was applied.
- All CRE isolates were screened for β-lactamases by whole genome sequencing.

RESULTS

- ATM-AVI was active (MIC ≤4 mg/L) against 91.9% of CAZ-AVI-NS, 94.0% of MEM-VAB-NS, 98.1% of CRE, and 98.4% of MBL-producing isolates (Table 1 and Figure 1).
- Cefiderocol retained activity against 78.8% of CAZ-AVI-NS, 83.2% of MEM-VAB-NS, 94.7% of CRE, and 82.1% of MBL-producing isolates (Table 1).
- Amikacin was the most active compound among non-β-lactams against CAZ-AVI-NS (63.1% S), MEM-VAB-NS (56.4% S), and CRE (61.1% S); whereas gentamicin (58.4% S) was slightly more active than amikacin (57.6% S) against MBL producers based on current CLSI breakpoints (Table 1).
- A carbapenemase (CBase) was identified in 136 (85.0%) isolates NS to CAZ-AVI, 133 (90.6%) isolates NS to MEM-VAB, and 683 (80.1%) CREs (Figure 1).
- NDM was the most common CBase type among isolates NS to CAZ-AVI (n=106; 66.3%) and isolates NS to MEM-VAB (n=106; 71.1%), whereas KPC (n=532; 63.9% of CREs) and NDM (n=113; 13.6%) were the most common CBase types among CREs (Figure 2).
- The frequency of MBL producers among CRE increased from 1.7% in 2016 to 43.8% in 2023 and decreased to 32.6% in 2024 (Figure 3).

Figure 3. Frequency of MBL producers among CRE over the years

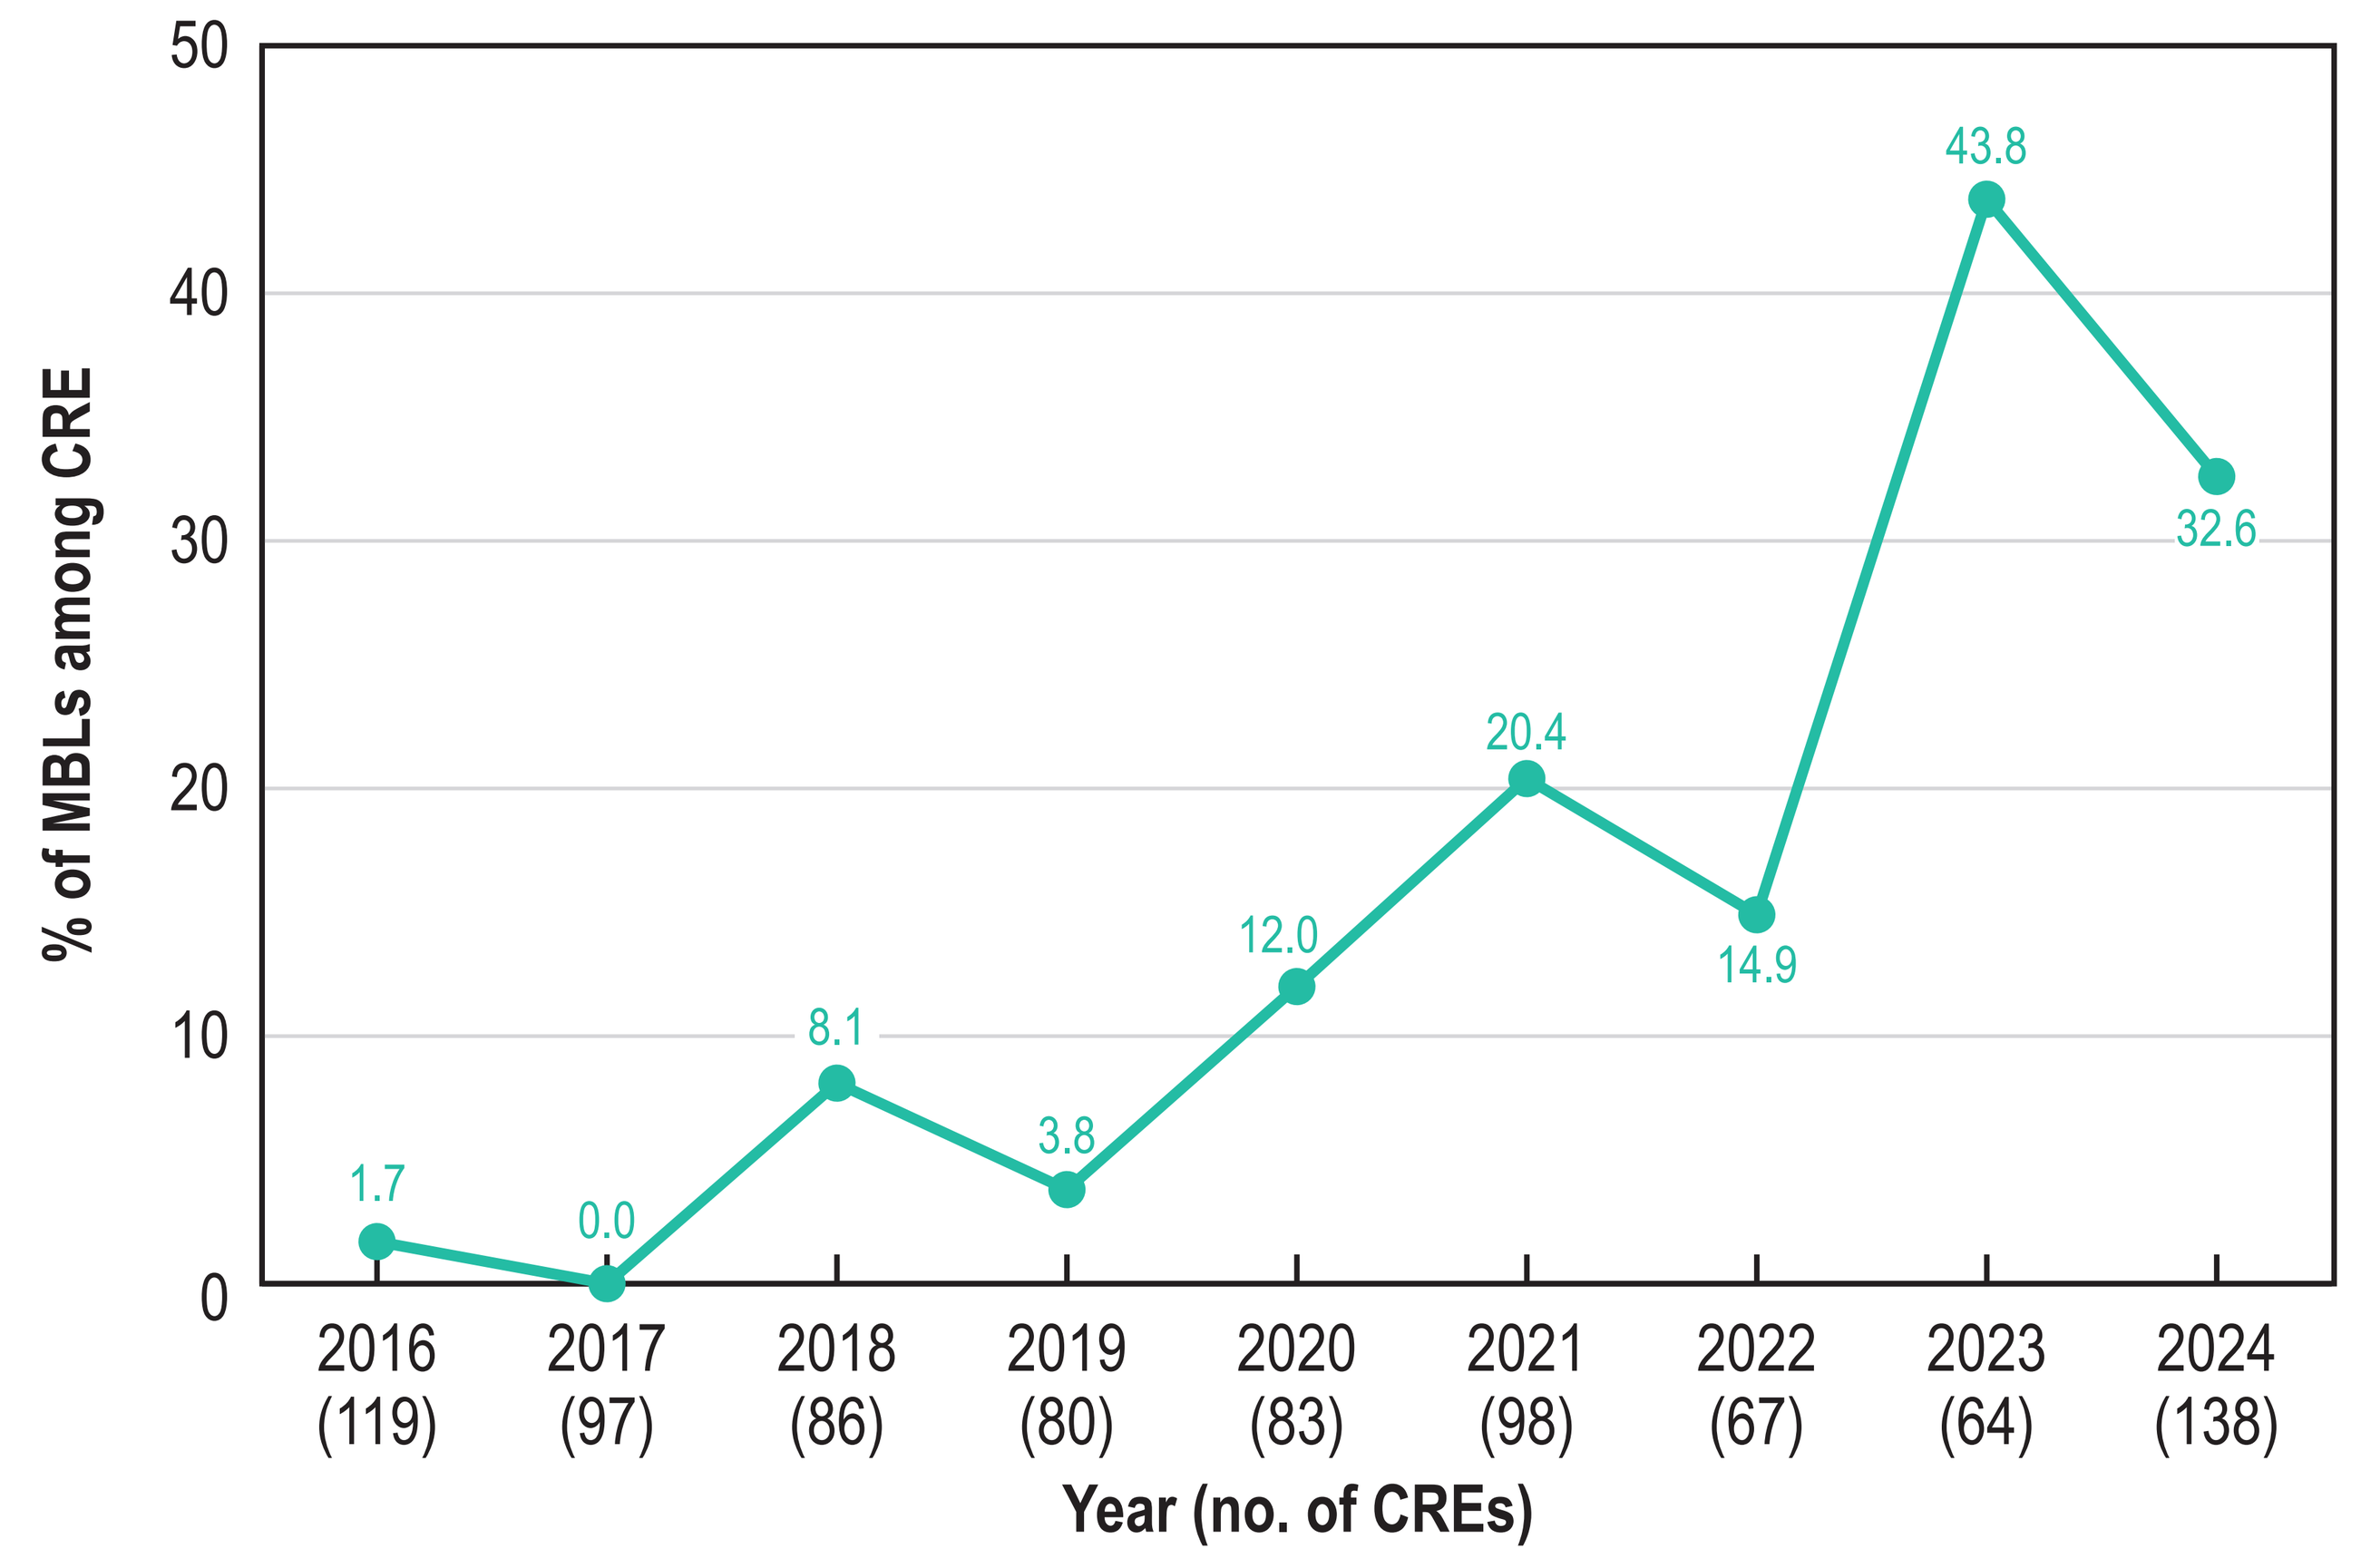
% of MBLs among CRE
50
40
30
20
10
0
1.7
0.0
8.1
3.8
12.0
20.4
14.9
43.8
32.6
2016 (119)
2017 (97)
2018 (86)
2019 (80)
2020 (83)
2021 (98)
2022 (67)
2023 (64)
2024 (138)
Year (no. of CREs)

Table 1. Antimicrobial susceptibility of resistant subsets

Antimicrobial agent	% Susceptible per CLSI and/or US FDA (no. of isolates)				
	CAZ-AVI-NS (160)	MEM-VAB-NS (149)	CAZ-AVI-NS and MEM-VAB-NS (115)	CRE (832)	MBL producers (125)
ATM-AVI	91.9	94.0	93.9	98.1	98.4
CAZ-AVI	0.0	22.8	0.0	84.3	5.6
MEM-VAB	25.8	0.0	0.0	79.1	9.8
IMI-REL	13.5	6.7	2.6	74.8	2.4
Cefiderocol	78.8	83.2	80.9	94.7	82.1
Levofloxacin	24.4	18.8	20.9	25.8	19.2
Gentamicin	60.0	54.4	58.3	55.6	58.4
Amikacin	63.1	56.4	58.3	61.1	57.6

Abbreviations: CAZ-AVI, ceftazidime-avibactam; NS, nonsusceptible; CRE, carbapenem-resistant Enterobacterales; MBL, metallo-β-lactamase; MEM-VAB, meropenem-vaborbactam; ATM-AVI, aztreonam-avibactam; IMI-REL, imipenem-relebactam.