IDWEEK 2025 | Poster #P1315

Activity of Aztreonam-Avibactam and Ceftazidime-Avibactam against Enterobacterales and *Pseudomonas aeruginosa* Causing Infections in Immunosuppressed Patients from United States Medical Centers (2019–2024)

Helio S. Sader[1], Mariana Castanheira[1], Todd Riccobene[2], John H. Kimbrough[1], Rodrigo E. Mendes[1]

[1]*Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA;* [2]*AbbVie Inc., Chicago, Illinois, USA*

CONCLUSIONS

ATM-AVI demonstrated almost complete activity against Enterobacterales (99.9% S) and retained potent activity against CRE (95.8% S) and CBase producers (100.0% S).

The activities of CAZ-AVI, MEM-VAB, and IMI-REL were compromised by the increased occurrence of MBL producers among CRE isolates.

CAZ-AVI, TOL-TAZ, and IMI-REL were highly active against *P. aeruginosa* from immunosuppressed patients.

INTRODUCTION

- Aztreonam-avibactam (ATM-AVI) was recently approved by the US FDA (February 2025) for treatment of complicated intra-abdominal infections (IAI) and by the EMA in the European Union (April 2024) for treatment of adults with complicated IAI, complicated urinary tract infection (cUTI), hospital-acquired pneumonia, including ventilator-associated pneumonia, and infections due to aerobic Gram-negative bacteria in adults with limited treatment options.
- Immunosuppression is associated with a higher incidence of infection with multidrug-resistant (MDR) pathogens, including carbapenem-resistant Enterobacterales (CRE), and it is also associated with higher mortality among patients with CRE infection.
- ATM-AVI has demonstrated potent activity against MDR Enterobacterales worldwide, including metallo-β-lactamase (MBL) producers.
- We evaluated the antimicrobial susceptibility of Enterobacterales and *P. aeruginosa* isolated from immunosuppressed patients in US medical centers.

METHODS

- A total of 51,992 Enterobacterales and 9,524 *P. aeruginosa* isolates were consecutively collected (1/patient) from 75 US medical centers in 2019–2024 as part of the INFORM Program.
- Isolates from patients hospitalized in hematology, oncology, and transplant units, including 2,407 Enterobacterales and 485 *P. aeruginosa*, were evaluated.
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of infection were included in the study.
- The ATM-AVI susceptible breakpoint of ≤4 mg/L was applied for Enterobacterales as established by the US FDA and EMA. CLSI breakpoints were applied to comparators.
- Multidrug resistance was defined as nonsusceptibility for ≥3 antibiotic classes.
- CRE isolates, defined as MIC ≥4 mg/L for meropenem and/or imipenem, were screened for β-lactamase genes by whole genome sequencing.

RESULTS

- Enterobacterales were mainly from bloodstream infection (BSI; 53.6%) and urinary tract infection (UTI; 19.9%) and *P. aeruginosa* were mainly from BSI (37.9%) and pneumonia (35.0%; Figure 1).
- ATM-AVI, ceftazidime-avibactam (CAZ-AVI), and meropenem-vaborbactam (MEM-VAB) were highly active against Enterobacterales (99.9–99.4% susceptible [S]), including MDR isolates (99.6–98.1% S; Table 1).
- Only ATM-AVI exhibited good activity against CRE isolates (95.8% S); CAZ-AVI was active against 79.2% and both MEM-VAB and imipenem-relebactam (IMI-REL) were active against 70.8% of CREs (Table 1).
- All (100.0%) carbapenemase (CBase)-producing CRE isolates were susceptible to ATM-AVI while 73.5% were susceptible to CAZ-AVI, 61.8% to MEM-VAB, and 58.8% to IMI-REL (Table 1).
- Ceftolozane-tazobactam (TOL-TAZ) showed good activity against *E. coli* (95.7% S) and *K. pneumoniae* (92.8% S), but limited activity against *E. cloacae* species complex (75.9% S; Table 2) and MDR organisms (72.4% S; Table 1).
- The most common CBases were KPC (41.7% of CREs), NDM (12.5%), and OXA-48-like types (10.4%; Figure 2).
- MBL represented 23.5% of CBases and was identified in 16.7% of CREs (Figure 2).
- The most active agents against *P. aeruginosa* were CAZ-AVI (95.7% S), TOL-TAZ (94.8% S), IMI-REL (97.4% S), and tobramycin (91.5% S; Table 2).
- Piperacillin-tazobactam (PIP-TAZ) and meropenem were active against 81.4% and 82.5% of *P. aeruginosa*, respectively, and ATM-AVI inhibited 78.6% of *P. aeruginosa* at ≤8 mg/L (Table 2).

Figure 1. Distribution of Enterobacterales (A) and *P. aeruginosa* (B) by infection type

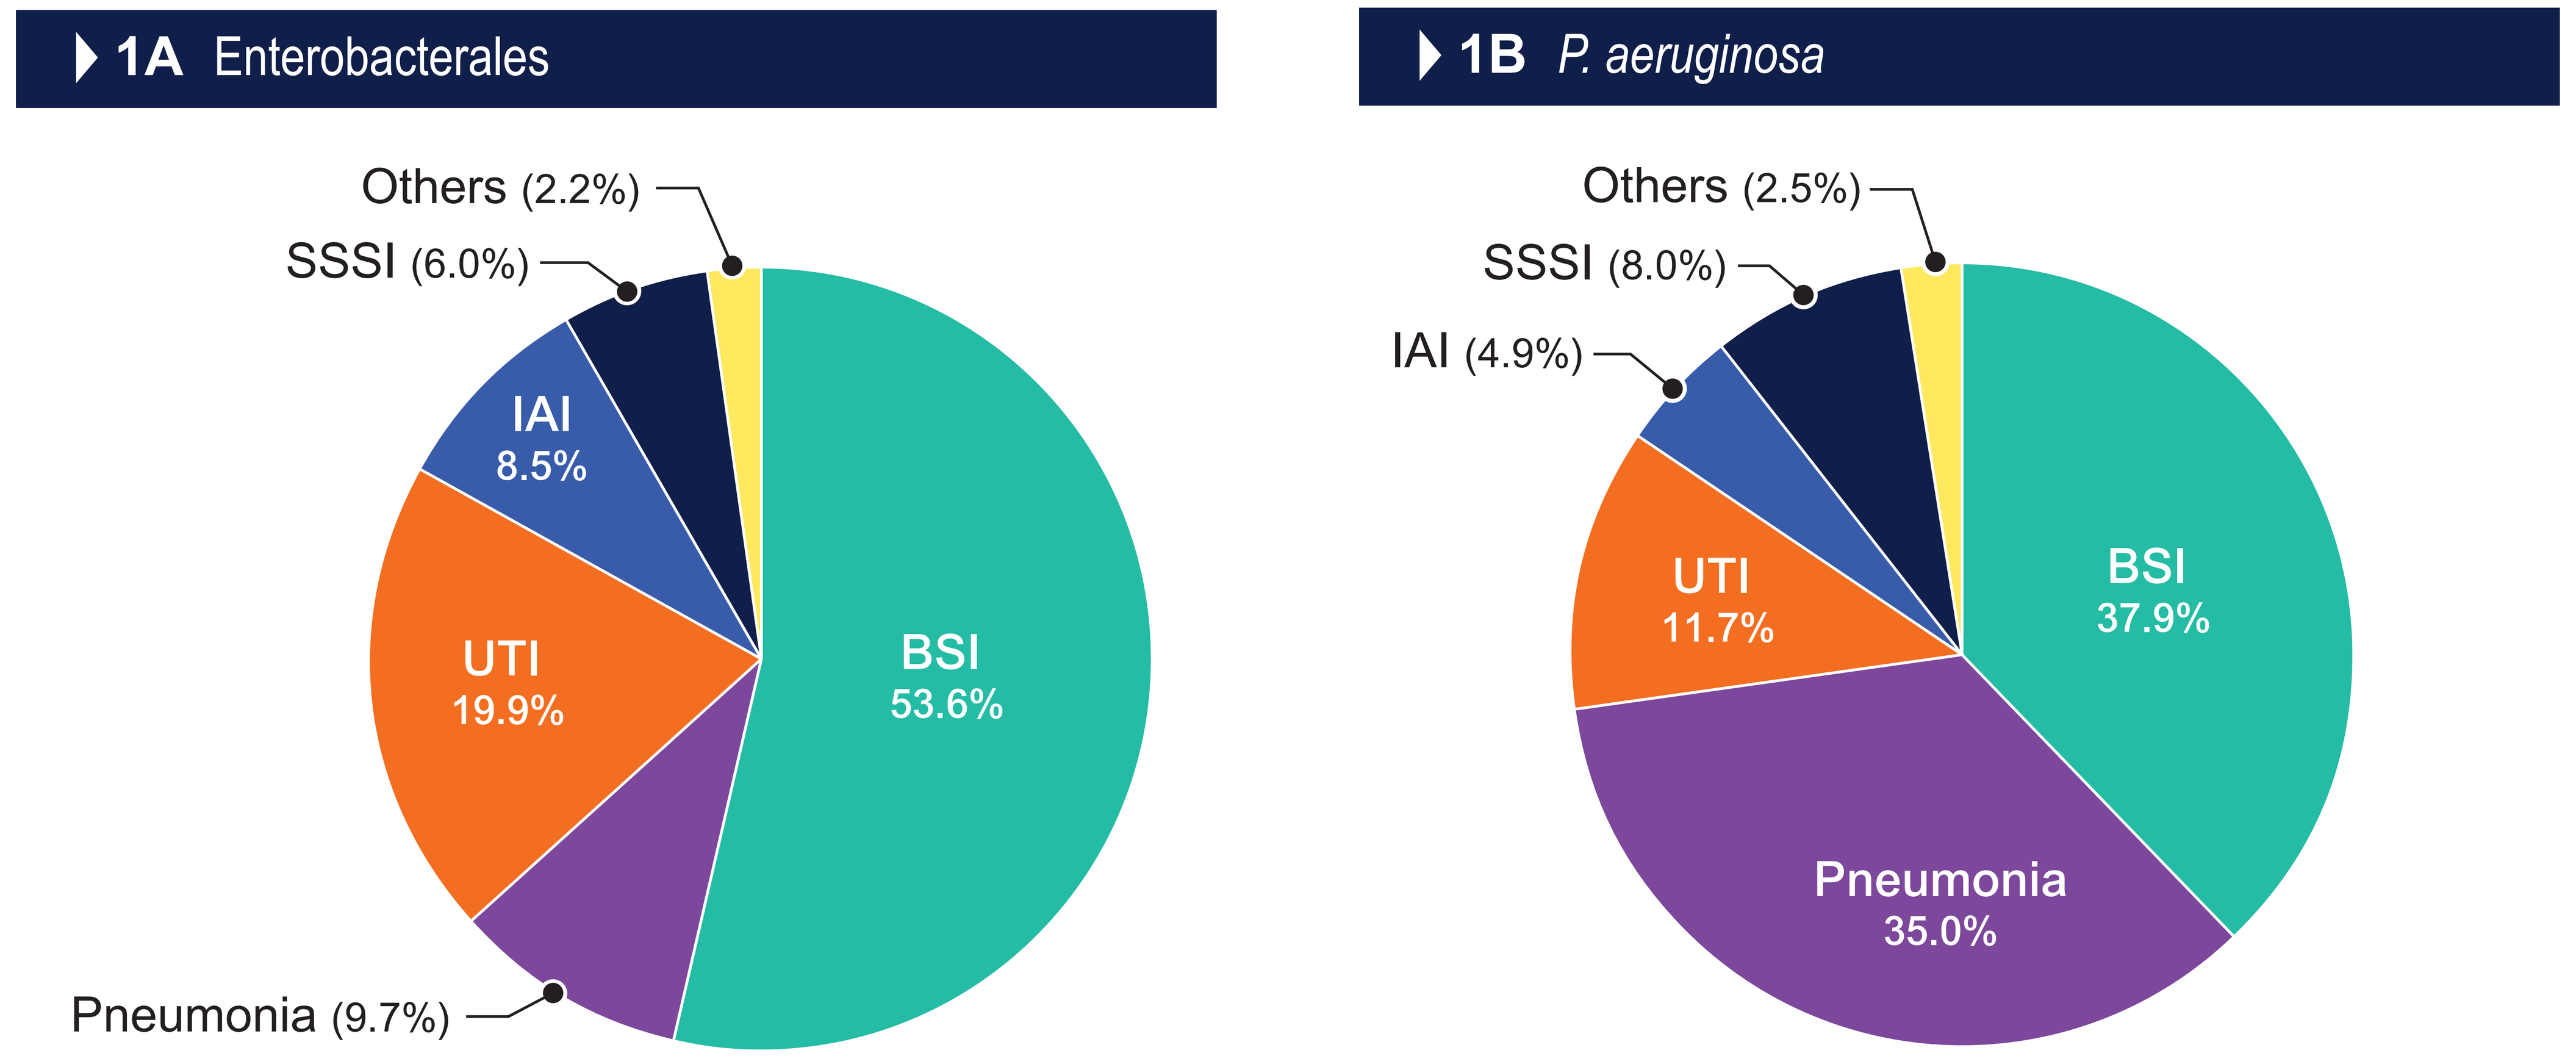
1A Enterobacterales
Others (2.2%)
SSSI (6.0%)
IAI 8.5%
UTI 19.9%
BSI 53.6%
Pneumonia (9.7%)
1B P. aeruginosa
Others (2.5%)
SSSI (8.0%)
IAI (4.9%)
UTI 11.7%
BSI 37.9%
Pneumonia 35.0%

Abbreviations: BSI, bloodstream infection; UTI, urinary tract infection; IAI, intra-abdominal infection; SSSI, skin and skin structure infection.

Figure 2. Distribution of carbapenemase types among CRE isolates

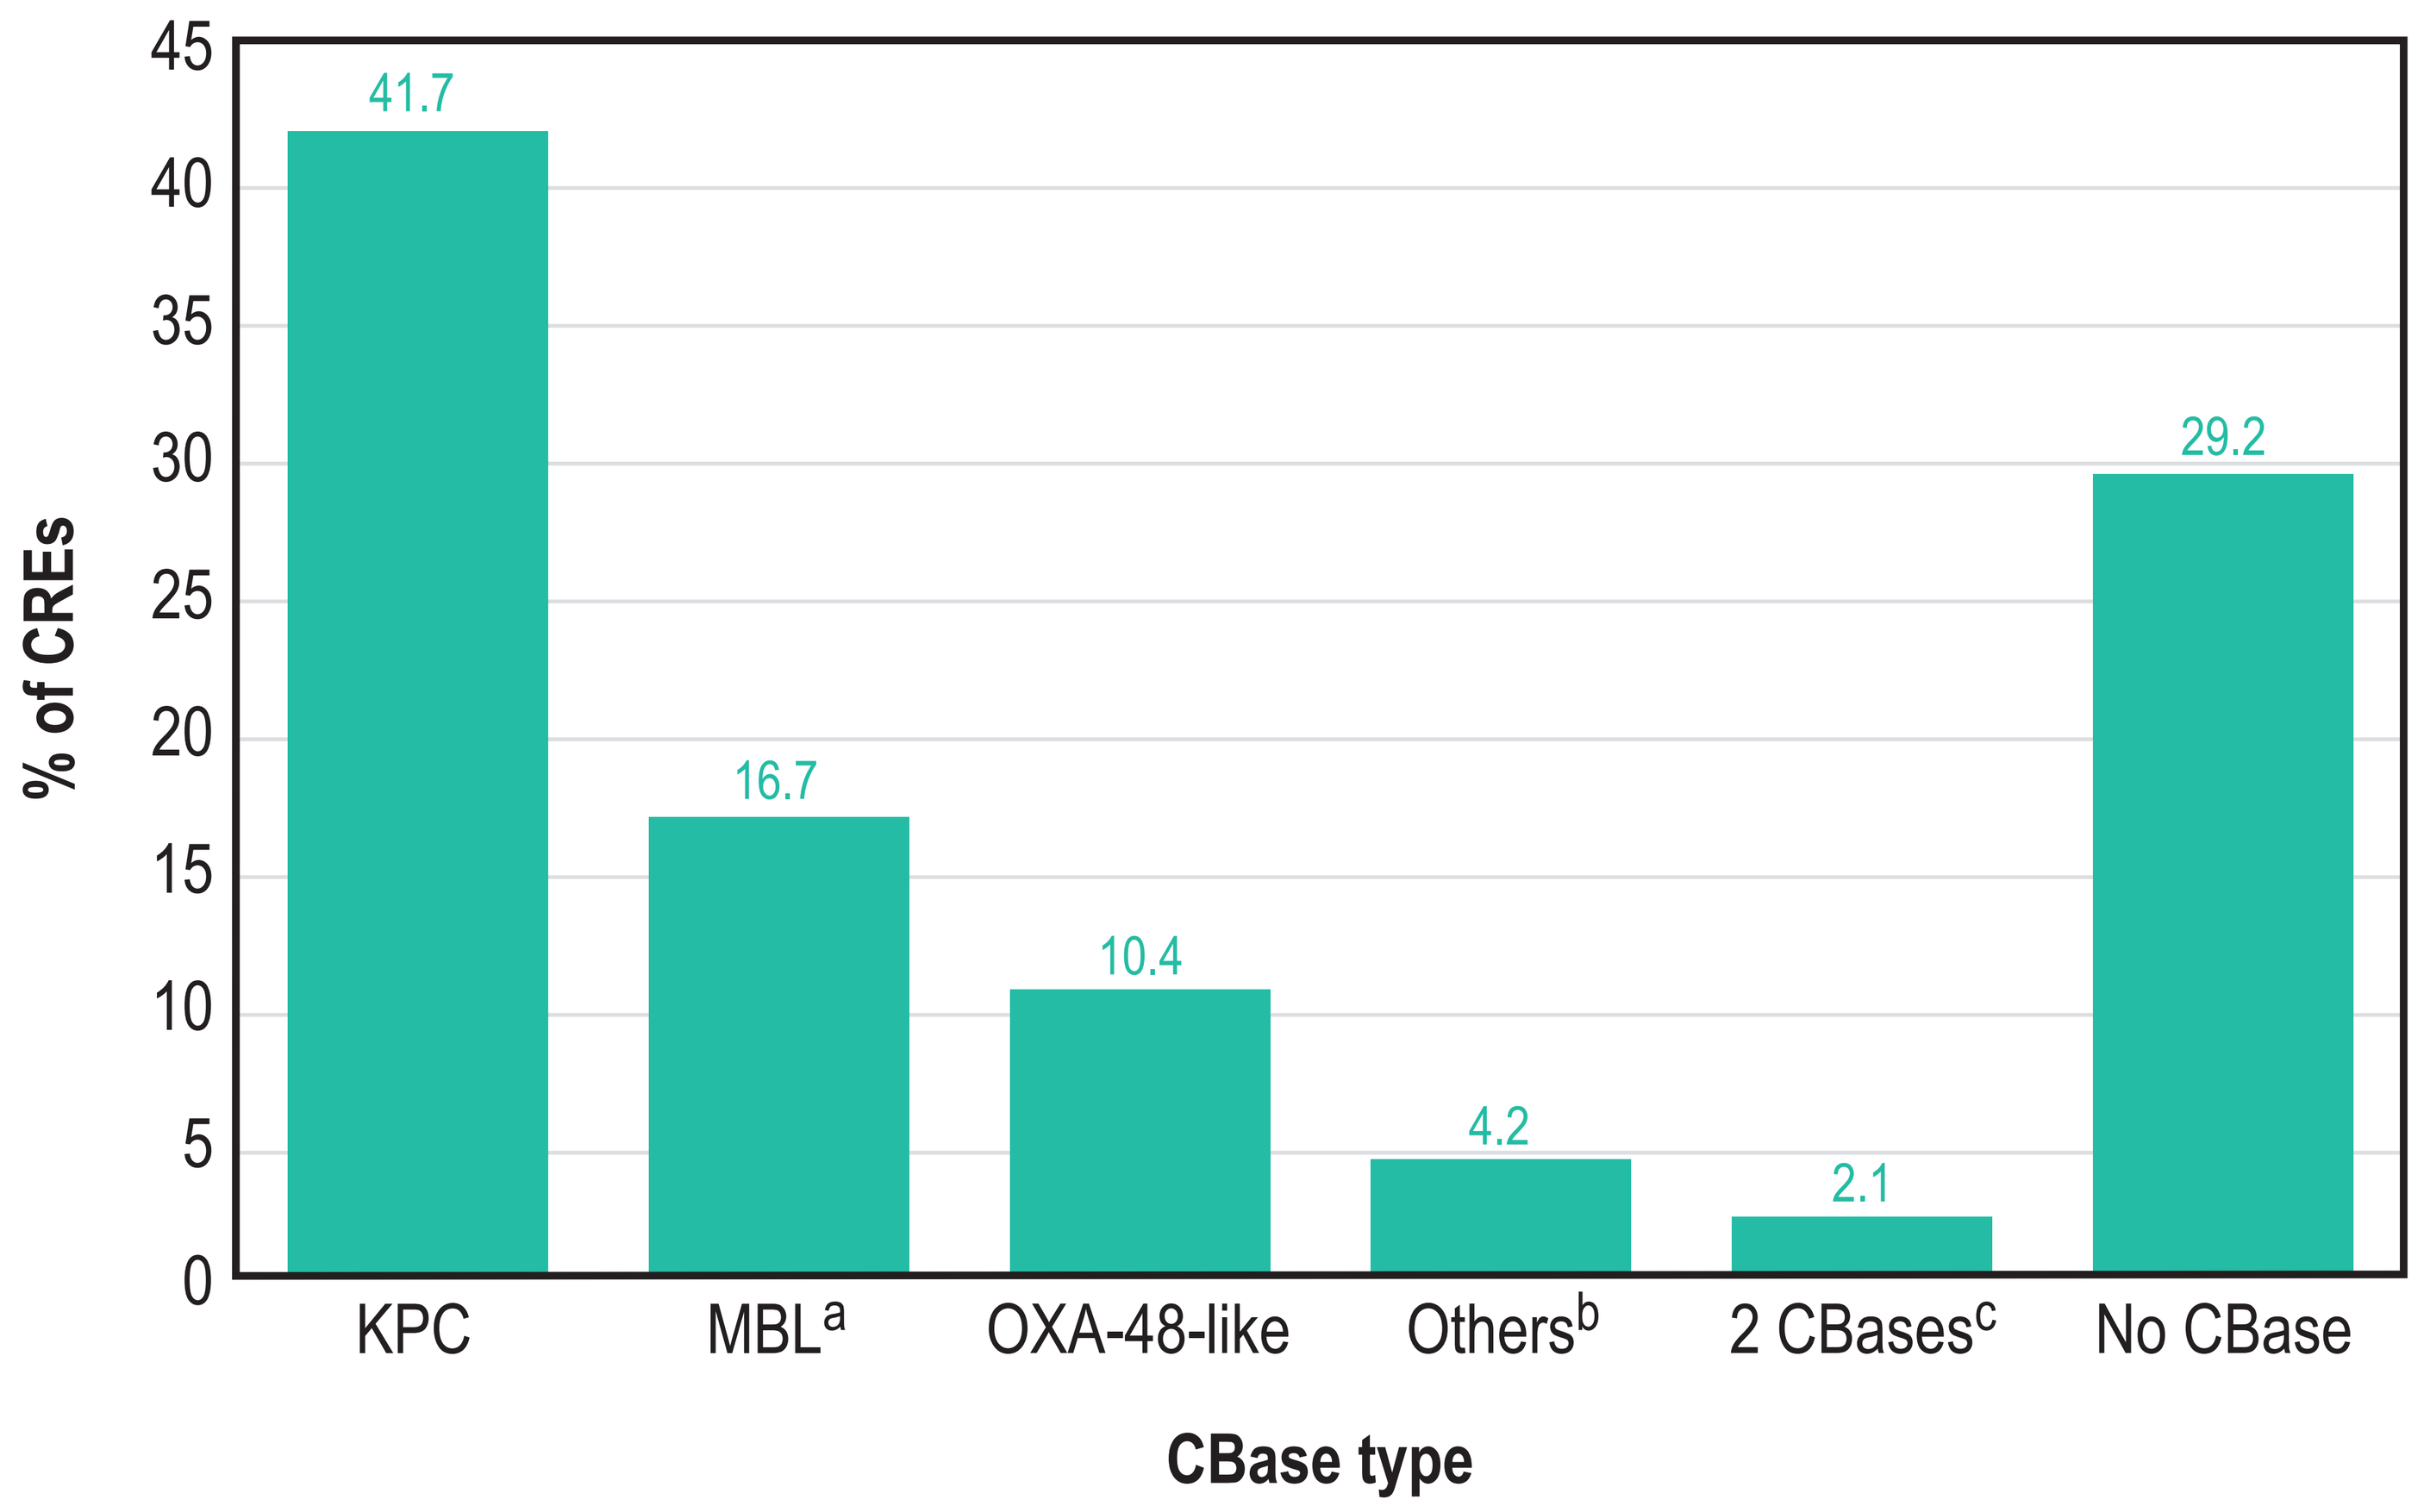
% of CREs
45
40
35
30
25
20
15
10
5
0
41.7
16.7
10.4
4.2
2.1
29.2
KPC
MBL[a]
OXA-48-like
Others[b]
2 CBases[c]
No CBase
CBase type

Abbreviations: CRE, carbapenem-resistant Enterobacterales; KPC, *Klebsiella pneumoniae* carbapenemase; MBL, metallo-β-lactamases; OXA, oxacillinase; CBase, carbapenemase.
[a] Includes IMP-4 (2 isolates), NDM-1 (2), and NDM-5 (4).
[b] Includes a GES-6 and an IMI-4.
[c] Includes a KPC-2 + GES-6.

Table 1. Antimicrobial susceptibility of Enterobacterales and resistant subsets of isolates from immunosuppressed patients

Antimicrobial	% Susceptible per CLSI and/or US FDA criteria (no. of isolates)			
	Enterobacterales (2,407)	MDR (773)	CRE (48)	CBase (34)[a]
Aztreonam-avibactam	99.9	99.6	95.8	100.0
Ceftazidime-avibactam	99.5	98.4	79.2	73.5
Ceftolozane-tazobactam	91.1	72.4	0.0	0.0
Meropenem-vaborbactam	99.4	98.1	70.8	61.8
Imipenem-relebactam	96.7	97.2	70.8	58.8
Piperacillin-tazobactam	81.2	48.1	0.0	0.0
Ampicillin-sulbactam	43.7	3.6	0.0	0.0
Ceftriaxone	73.0	24.7	0.0	0.0
Cefepime	80.9	43.7	0.0	0.0
Meropenem	97.9	93.4	0.0	0.0
Levofloxacin	71.7	36.5	29.2	26.5
Gentamicin	87.1	62.5	50.0	44.1
Amikacin	94.4	86.5	70.8	64.7

[a] Carbapenemase-producing CRE isolates.
Abbreviations: CLSI, Clinical and Laboratory Standards Institute; US FDA, United States Food and Drug Administration; MDR, multidrug-resistant; CRE, carbapenem-resistant Enterobacterales; CBase, carbapenemase.

Table 2. Antimicrobial susceptibility of selected species collected from immunosuppressed patients

Antimicrobial	% Susceptible per CLSI and/or US FDA criteria (no. of isolates)			
	E. coli (906)	*K. pneumoniae* (583)	*E. cloacae* (286)	*P. aeruginosa* (485)
Aztreonam-avibactam	99.8	100.0	99.7	(78.6)[a]
Ceftazidime-avibactam	99.4	99.7	98.6	95.7
Ceftolozane-tazobactam	95.7	92.8	75.9	94.8
Meropenem-vaborbactam	99.4	98.9	98.9	—[b]
Imipenem-relebactam	99.4	98.4	98.4	97.4
Piperacillin-tazobactam	87.7	77.8	67.4	81.4
Cefepime	75.1	76.3	87.4	85.8
Meropenem	98.9	96.4	97.6	82.5
Levofloxacin	52.8	75.0	93.0	71.3
Gentamicin	83.3	85.4	93.7	—[b]
Amikacin	89.8	97.3	98.3	—[b]
Tobramycin	—[b]	—[b]	—[b]	91.5

[a] % inhibited at ≤8 mg/L, the CLSI breakpoint for aztreonam. [b] Not tested or no breakpoint published by CLSI and/or US FDA.
Abbreviations: CLSI, Clinical and Laboratory Standards Institute; US FDA, United States Food and Drug Administration

Contact Information

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2025 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2025_25-ALG-02_P5_Immunossup.pdf

To submit a medical question, please visit www.abbviemedinfo.com

SCAN ME

Acknowledgements

This study at JMI Laboratories was supported by AbbVie Inc. (New York, NY). JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by AbbVie Inc.

References

1. Bar-Yoseph H, Korytny A, Andrawus EA, *et al.* Risk factors for mortality among carbapenem-resistant Enterobacteriaceae carriers with focus on immunosuppression. *J Infect* 2019;78(2):101–105.
2. CLSI. 2025. M100Ed35. Performance standards for antimicrobial susceptibility testing: 35th informational supplement. Clinical and Laboratory Standards Institute.
3. Sader HS, Mendes RE, Carvalhaes CG, Kimbrough JH, Castanheira M. Changing epidemiology of carbapenemases among carbapenem-resistant Enterobacterales from United States hospitals and the activity of aztreonam-avibactam against contemporary Enterobacterales (2019–2021). *Open Forum Infect Dis* 2023;10(2):ofad046.
4. Tamma PD, Heil EL, Justo JA *et al.* Infectious Diseases Society of America 2024 guidance on the treatment of antimicrobial-resistant Gram-negative infections. *Clin Infect Dis* 2024 ciae403.

abbvie