IDWEEK 2025 | Poster #P1320

Antimicrobial susceptibility of *S. maltophilia* and *B. cepacia* species complex isolated from patients with pneumonia in United States hospitals (2022–2024)

Helio S. Sader[1], Marisa L. Winkler[1], Rodrigo E. Mendes[1], Dmitri Debabov[2], Mariana Castanheira[1]

[1] *Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA;* [2] *AbbVie Inc., Chicago, Illinois, USA*

CONCLUSIONS

ATM-AVI exhibited potent activity and broad coverage against *S. maltophilia* from US hospitals, and its activity was not adversely affected by resistance to other agents.

The β-lactamase inhibitor combinations CAZ-AVI and MEM-VAB were the most active agents against *B. cepacia* based on CLSI breakpoints published for Enterobacterales.

Appropriate assessment of breakpoints for these organisms is urgently needed to provide better guidance of antimicrobial therapy for infections caused by *S. maltophilia* and *B. cepacia*.

INTRODUCTION

- The occurrences of *S. maltophilia* and *B. cepacia* infections, mainly pneumonia, have increased continuously in the last few years.
- Aztreonam-avibactam (ATM-AVI) was recently approved by the US FDA for treatment of complicated IAI and by the EMA in the European Union for treatment of adults with complicated IAI, complicated urinary tract infection (cUTI), hospital-acquired pneumonia, including ventilator-associated pneumonia, and infections due to aerobic Gram-negative bacteria in adults with limited treatment options.
- We evaluated the *in vitro* activities of aztreonam-avibactam (ATM-AVI) and comparators against *S. maltophilia* and *B. cepacia* causing pneumonia in United States medical centers.

METHODS

- A total of 958 clinical isolates, including 830 *S. maltophilia* and 128 *B. cepacia*, were consecutively collected from patients with pneumonia in 65 United States (US) medical centers in 2022–2024.
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of infection were included in the study.
- Isolates were susceptibility tested by Clinical and Laboratory Standards Institute (CLSI) M07 broth microdilution methods at a monitoring laboratory.
- CLSI breakpoints were applied for *S. maltophilia* when available.
- Enterobacterales breakpoints were applied to *B. cepacia* and susceptible [S]/resistant breakpoints of ≤4/≥16 mg/L were applied for ATM-AVI against both organisms for comparison.
- Susceptibility results were stratified by year and infection type.

RESULTS

- The most active agents against *S. maltophilia* were trimethoprim-sulfamethoxazole (TMP-SMX; 97.2% S), minocycline (92.6% S), and ATM-AVI (92.2% inhibited at ≤4 mg/L; Table 1 and Figure 1).
- ATM-AVI retained potent activity against isolates non-S (NS) to other agents commonly used to treat *S. maltophilia* infections, including TMP-SMX (91.3% inhibited at ≤4 mg/L of ATM-AVI) and minocycline (91.8% inhibited at ≤4 mg/L; Figure 2).
- Levofloxacin showed moderate activity (81.2% inhibited at ≤2 mg/L), tigecycline inhibited 90.0% of isolates at ≤2 mg/L (42.5% at ≤0.5 mg/L), and both ceftazidime ($MIC_{50/90}$, >32/>32 mg/L; 13.5% inhibited at ≤4 mg/L) and colistin ($MIC_{50/90}$, 8/>8 mg/L; 37.0% inhibited at ≤2 mg/L) exhibited limited activity against *S. maltophilia* (Table 1).
- The most active agents against *B. cepacia* were ceftazidime-avibactam (CAZ-AVI; 97.7% inhibited at ≤8 mg/L), meropenem-vaborbactam (MEM-VAB; 96.1% inhibited at ≤4 mg/L), and TMP-SMX (82.8% inhibited at ≤2 mg/L); ATM-AVI inhibited 64.1% at ≤4 mg/L (Table 1).
- CAZ-AVI and MEM-VAB were active against 91.4% of *B. cepacia* isolates with ceftazidime MIC >4 mg/L (data not shown).

Table 1. Antimicrobial susceptibility of *S. maltophilia* and *B. cepacia* from patients with pneumonia

Antimicrobial agent	MIC_{50}	MIC_{90}	%S	%I	%R
S. maltophilia (830)					
Aztreonam-avibactam[a]	4	4	92.2	7.3	0.5
TMP-SMX[b]	≤0.12	0.5	97.2		2.8
Minocycline[b]	0.5	1	92.6	4.8	2.6
Levofloxacin[b]	1	8	81.2	8.2	10.6
Tigecycline[a]	1	2	90.0	7.2	2.8
Ceftazidime[a]	>32	>32	13.5	6.0	80.5
Colistin[a]	8	>8		37.0	63.0
B. cepacia (128)					
Aztreonam-avibactam[a]	4	16	64.1	25.8	10.2
Ceftazidime-avibactam[a]	2	4	97.7		2.3
Meropenem-vaborbactam[a]	0.5	2	96.1	2.3	1.6
Ceftolozane-tazobactam[a]	2	16	67.2	16.4	16.4
Ceftazidime[a]	4	16	72.7	11.7	15.6
Meropenem[a]	4	8	10.9	38.3	50.8
TMP-SMX[a]	0.5	>4	82.8	3.9	13.3
Minocycline[a]	2	8	81.7	9.8	8.5
Levofloxacin[a]	2	>32	5.5	18.7	75.8
Tigecycline[a]	2	>8	63.3	14.0	22.7

[a] CLSI (2025) and/or US FDA breakpoints for Enterobacterales were applied for comparison.
[b] Based on CLSI (2025) breakpoints for *S. maltophilia*.
Abbreviations: TMP-SMX, trimethoprim-sulfamethoxazole.

Figure 1. Aztreonam-avibactam MIC distributions for *S. maltophilia* and *B. cepacia* from patients with pneumonia

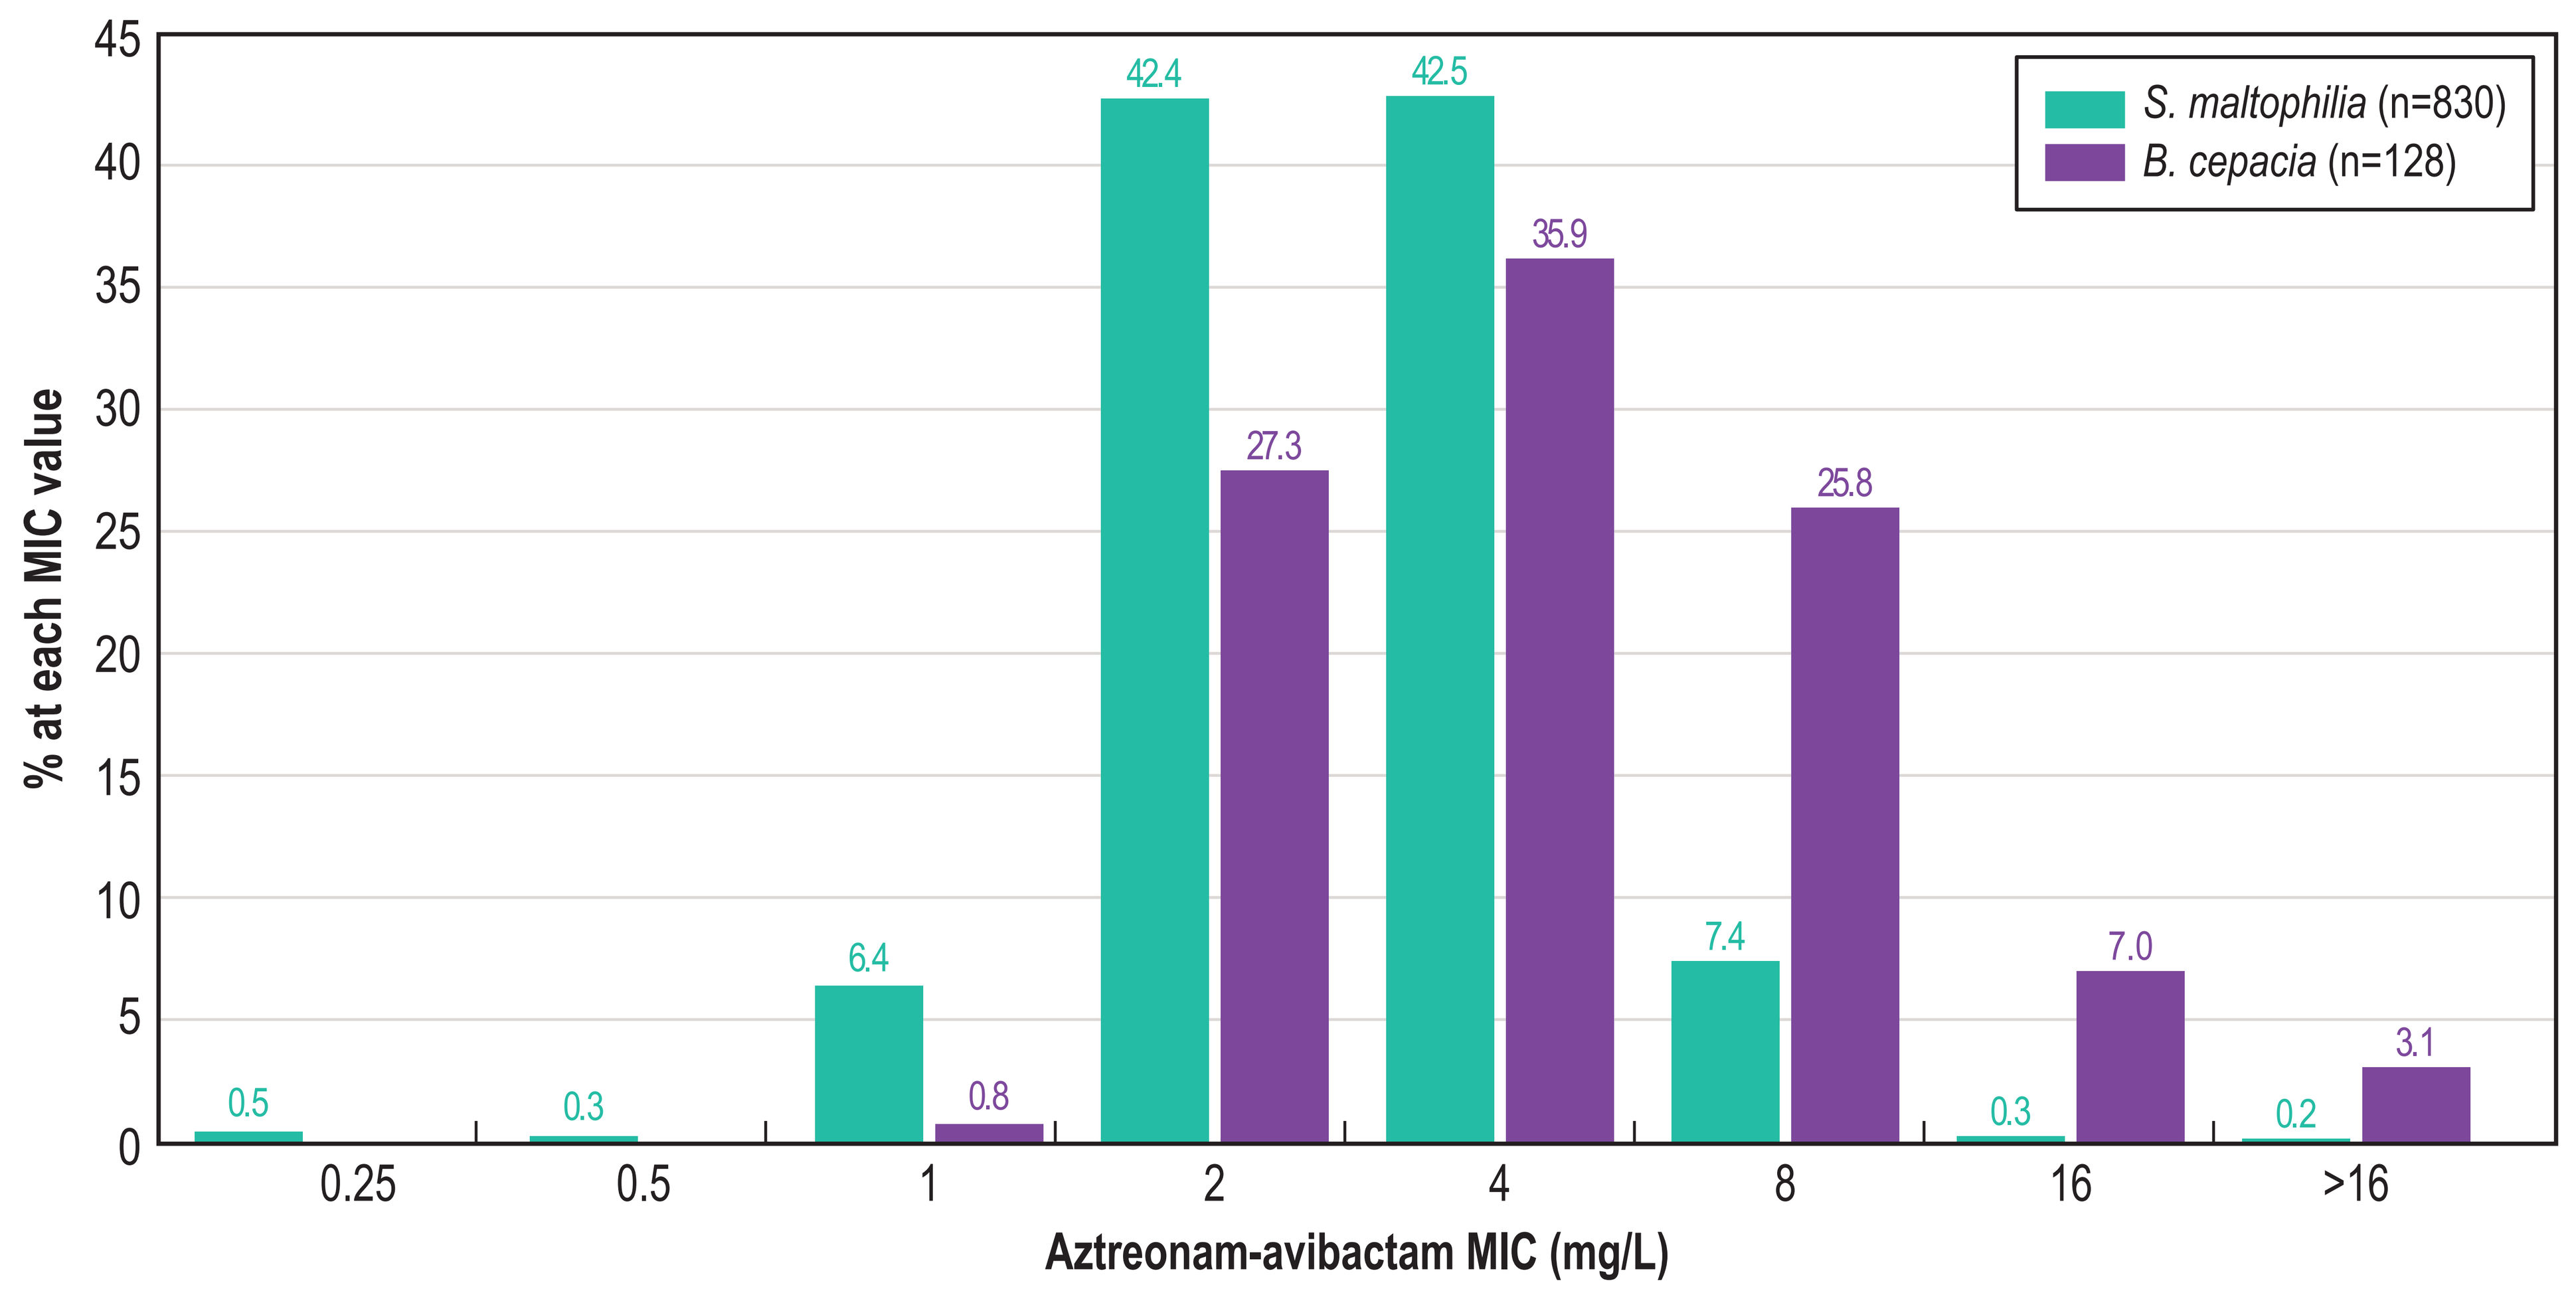
% at each MIC value
45
40
35
30
25
20
15
10
5
0
0.25
0.5
1
2
4
8
16
>16
Aztreonam-avibactam MIC (mg/L)
S. maltophilia (n=830)
B. cepacia (n=128)
0.5
0.3
6.4
0.8
42.4
27.3
42.5
35.9
7.4
25.8
0.3
7.0
0.2
3.1

Figure 2. Activity of aztreonam-avibactam and comparators against *S. maltophilia* resistant subsets

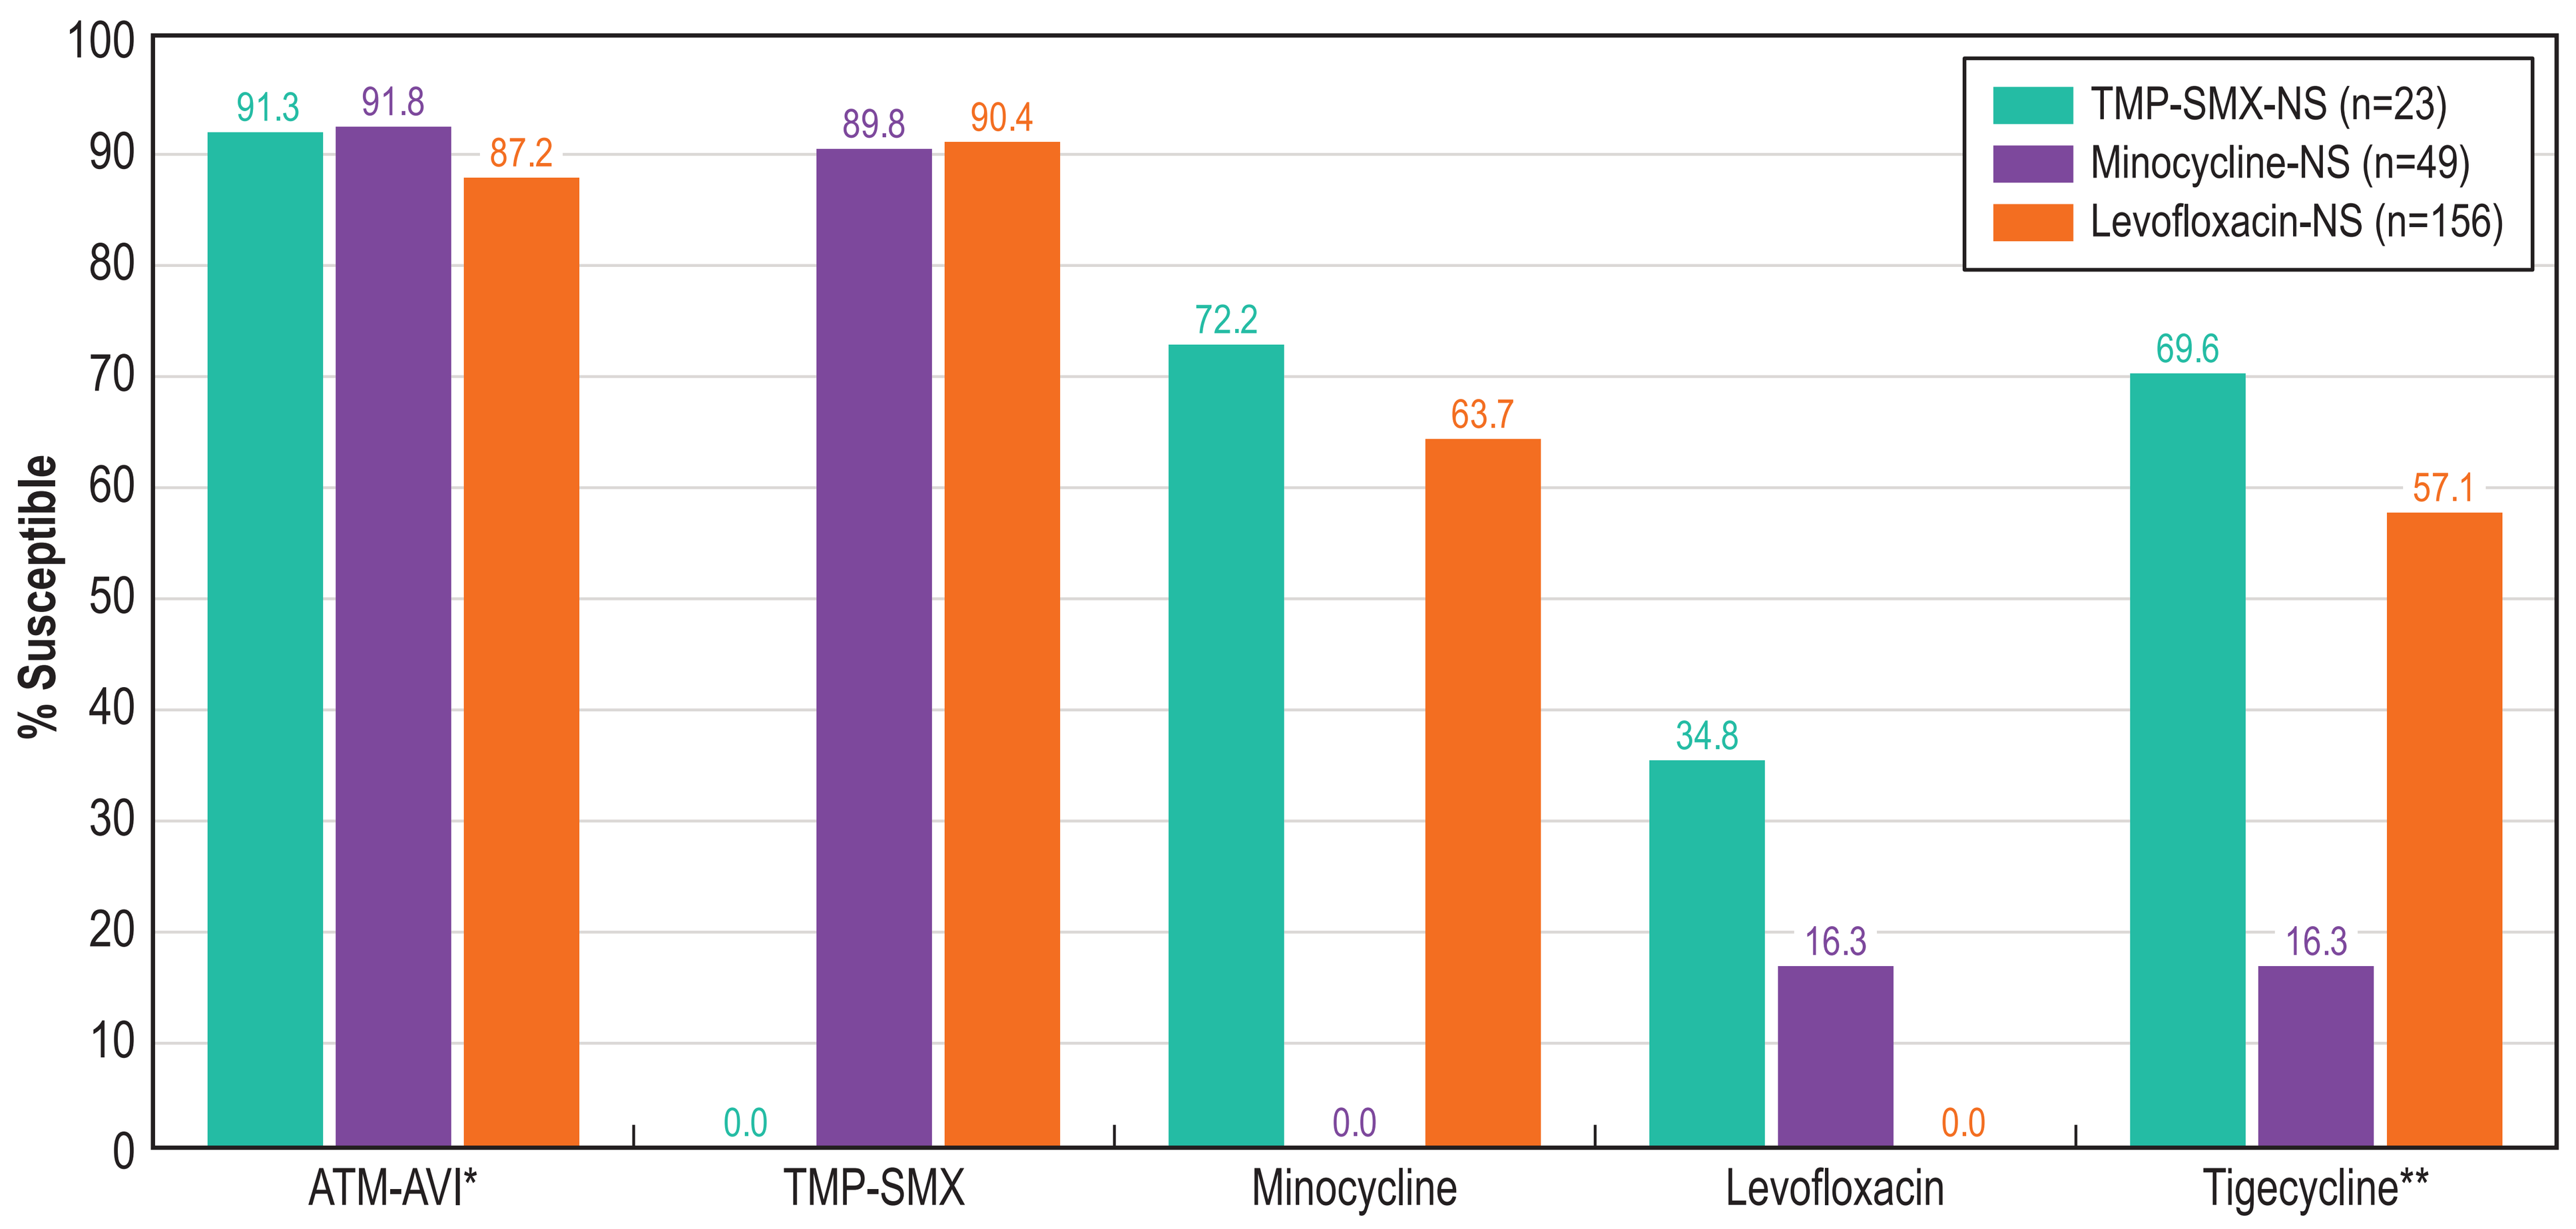
% Susceptible
100
90
80
70
60
50
40
30
20
10
0
ATM-AVI*
TMP-SMX
Minocycline
Levofloxacin
Tigecycline**
TMP-SMX-NS (n=23)
Minocycline-NS (n=49)
Levofloxacin-NS (n=156)
91.3
91.8
87.2
0.0
89.8
90.4
72.2
0.0
63.7
34.8
16.3
0.0
69.6
16.3
57.1

* % inhibited at ≤4 mg/L. ** % inhibited at ≤2 mg/L.
Abbreviations: ATM-AVI, aztreonam-avibactam; TMP-SMX, trimethoprim-sulfamethoxazole; NS, nonsusceptible.

Contact Information

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2025 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2025_25-ALG-02_P6_A6_XM_BC.pdf

To submit a medical question, please visit www.abbviemedinfo.com

SCAN ME

Acknowledgements

This study at JMI Laboratories was supported by AbbVie Inc. (New York, NY). JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by AbbVie Inc.

References

1. Barrasa H, Morán MA, Fernández-Ciriza L, *et al.* Optimizing antibiotic therapy for *Stenotrophomonas maltophilia* infections in critically ill patients: A pharmacokinetic/pharmacodynamic approach. *Antibiotics (Basel)* 2024;13(6):553.
2. CLSI. 2025. M100Ed35. Performance standards for antimicrobial susceptibility testing: 35th informational supplement. Clinical and Laboratory Standards Institute.
3. Jorth P, Manuel C, McLemore T, *et al.* Evaluation of antimicrobial susceptibility testing methods for *Burkholderia cepacia* complex isolates from people with and without cystic fibrosis. *J Clin Microbiol* 2025; 63(2): e0148024.
4. Sader HS, Castanheira M, Arends SJR, Doyle TB. Antimicrobial susceptibility of *Stenotrophomonas maltophilia* from United States medical centers (2019–2023). *Antimicrob Agents Chemother* 2025 2;69(4):e0012425.
5. Tamma PD, Heil EL, Justo JA, *et al.* Infectious Diseases Society of America 2024 guidance on the treatment of antimicrobial-resistant Gram-negative infections. *Clin Infect Dis* 2024 ciae403.

abbvie