IDWEEK 2025 | Poster #P1340

Activity of Delafloxacin and Comparator Agents Against Bacterial Isolates from Patients with Diabetic Foot Infection in the United States and Europe (2017–2023)

Michael D Huband, Marisa L Winkler, Kelley A Fedler, Mariana Castanheira

Element Iowa City (JMI Laboratories), North Liberty, Iowa, United States

Introduction

- Delafloxacin is a fluoroquinolone antibacterial with oral and intravenous treatment options approved in 61 countries for treatment of acute bacterial skin and skin structure infection (ABSSSI) and community-acquired bacterial pneumonia (CABP) caused by specific pathogens in adults.
- Indicated organisms/organism groups for delafloxacin and ABSSSI in the United States include: staphylococci (*S. aureus* [SA] both methicillin-susceptible [MSSA] and methicillin-resistant [MRSA], *S. haemolyticus*, and *S. lugdunensis*), streptococci (*Streptococcus pyogenes*, *S. agalactiae*, and *S. anginosus* group), *Enterococcus faecalis*, Enterobacterales (*Enterobacter cloacae, Escherichia coli*, and *Klebsiella pneumoniae*), and *Pseudomonas aeruginosa*.
- In this study, the *in vitro* activity of delafloxacin, ciprofloxacin, levofloxacin, moxifloxacin, minocycline, meropenem-vaborbactam, and oritavancin were determined against bacterial isolates from patients with diabetic foot infection (DFI) in the United States (US) and Europe (2017–2023).

Materials and Methods

- ABSSSI isolates from patients with DFI were collected in the SENTRY Antimicrobial Surveillance Program (2017–2023) from medical centers located in the US and Europe.
- Broth microdilution susceptibility testing was conducted according to Clinical and Laboratory Standards Institute (CLSI) methods and CLSI, EUCAST, and FDA breakpoint interpretive criteria (2025) were applied.
- Bacterial isolate identifications were determined at participating SENTRY sites and confirmed at Element Iowa City (JMI Laboratories) using matrix-assisted laser-desorption ionization time-of-flight mass spectrometry (as needed).

Results

- The most common DFI pathogens from patients with ABSSSI in the SENTRY Antimicrobial Surveillance Program based on prevalence during 2017–2023 were *S. aureus* (38%), Enterobacterales (32%), *P. aeruginosa* (11%), enterococci (8%), streptococci (7%), other staphylococci (2%), and other non-fermenters (2%) (Figure 1).
- MRSA rates in DFI patients were 39.0% (60/154) in the US and 25.9% (42/162) in Europe.
- Overall, 87.7% (FDA) of *S. aureus* isolates from patients with DFI in the United States were susceptible to delafloxacin (Table 1), including 96.7% of MSSA and 73.3% of MRSA.
- 85.2% of *S. aureus* isolates from DFI patients in Europe were susceptible to delafloxacin (Table 1), including 96.9% of MSSA and 45.2% of MRSA.
- Comparator fluoroquinolone susceptibilities against *S. aureus* isolates from the US and Europe were 66.9% (CLSI/EUCAST/FDA) and 75.4% (CLSI/EUCAST/FDA), respectively, for levofloxacin, and 66.9% (CLSI/EUCAST/FDA) and 75.3% (CLSI/EUCAST/FDA), respectively, for moxifloxacin (Table 1).
- All *S. aureus* isolates were susceptible (100.0%; CLSI/EUCAST/FDA) to oritavancin (Table 1).
- All (100.0%) *S. haemolyticus* (n=2), *S. lugdunensis* (n=4), *S. pyogenes* (n=6), and *S. anginosus* group (n=4) isolates from patients with DFI were susceptible (ABSSSI breakpoints) to delafloxacin (data not shown).
- Susceptibility of *Streptococcus agalactiae* isolates (*n*=34) to delafloxacin was 97.1%/100.0% (EUCAST/FDA) using ABSSSI breakpoints (Table 1).
- 83.3% of *E. faecalis* isolates from DFI were susceptible to delafloxacin (FDA), levofloxacin (CLSI/EUCAST/FDA), and moxifloxacin (FDA) (Table 1).
- Overall, 60.8%/69.4% (EUCAST/FDA), 79.9% (CLSI/EUCAST/FDA), and 82.8% (CLSI/EUCAST/FDA) of all Enterobacterales isolates from patients with DFI in the US and Europe were susceptible to delafloxacin, ciprofloxacin, and levofloxacin, respectively (Table 2).
- 36.9% (116/314) of Enterobacterales isolates from patients with DFI in the US and Europe were *E. cloacae* species complex (12.4%), *E. coli* (17.8%), or *K. pneumoniae* (6.7%).
- Against indicated organism groups, 87.2% (FDA) of *E. cloacae* species complex isolates, 62.5%/71.4% (EUCAST/FDA) of *E. coli* isolates, and 57.1% (FDA) of *K. pneumoniae* isolates from patients with DFI in the US and Europe were susceptible to delafloxacin (Table 2).
- 75.0% (FDA) of *Citrobacter* spp. isolates and 76.6% (FDA) of *Proteus mirabilis* isolates from patients with DFI in the US and Europe were susceptible (ABSSSI breakpoints) to delafloxacin (Table 2).
- Overall, 66.3% (FDA), 79.3% (CLSI/EUCAST/FDA), and 68.5%/79.3%/68.5% (CLSI/EUCAST/FDA) of *P. aeruginosa* isolates from patients with DFI in the US and Europe were susceptible to delafloxacin, ciprofloxacin, and levofloxacin, respectively (Table 3).
- Susceptibility of *P. aeruginosa* isolates from patients with DFI to delafloxacin was higher in the US (74.4% [FDA]) compared to Europe (59.2% [FDA]) (Table 3).

Conclusions

- The most prevalent pathogen in patients with DFI was *S. aureus* (38%) of which delafloxacin was the most active fluoroquinolone (86.3% susceptible [EUCAST/FDA]) tested.
 - Against MRSA, delafloxacin was active against 73.3% of isolates from patients with DFI in the United States and 45.2% of isolates from Europe.
- Delafloxacin was active against other Gram-positive DFI pathogens including *S. haemolyticus*, *S. lugdunensis*, *S. pyogenes*, and *S. anginosus* group isolates (100.0% susceptible [FDA]), *S. agalactiae* (97.1%/100.0% susceptible [EUCAST/FDA]), and *E. faecalis* (83.3% susceptible [FDA]).
- Overall, 87.2% (FDA) of *E. cloacae* species complex, 62.5%/71.4% (EUCAST/FDA) of *E. coli*, 57.1% (FDA) of *K. pneumoniae*, and 66.3% (FDA) of *P. aeruginosa* isolates from patients with DFI were susceptible to delafloxacin.
- Delafloxacin may serve as a useful treatment option (either alone or in combination) in patients with DFI including those where a broad-spectrum agent is desired. Additional clinical studies in patients with DFI are needed.

Funding

This study was supported by Melinta Therapeutics. MD Huband, ML Winkler, KA Fedler, and M Castanheira are/were employees of Element Iowa City (JMI Laboratories) at the time of this study and were paid consultants to Melinta Therapeutics in connection with the development of this poster.

Table 1. Activity of delafloxacin and comparators against Gram-positive isolates from patients with diabetic foot infection in the United States and Europe (2017–2023)

Antimicrobial Agent (Region; no. tested)	MIC (mg/L)			CLSI[a]		EUCAST[a]		US FDA[a]	
	MIC_{50}	MIC_{90}	MIC range	%S	%R	%S	%R	%S	%R
***Staphylococcus aureus* (United States; *n*=154)**									
Delafloxacin	0.008	0.5	0.001 to 4			87.7 [b]	12.3	87.7 [b]	7.8
Levofloxacin	0.25	>4	0.06 to >4	66.9	33.1	66.9 [c]	33.1	66.9 [d]	22.7
Moxifloxacin	≤0.06	4	≤0.06 to >4	66.9	29.9	66.9	33.1	66.9 [e]	29.9
Minocycline	0.06	0.12	≤0.03 to >8	98.7	1.3	98.1	1.9	98.7 [e]	1.3
Oritavancin	0.03	0.03	≤0.008 to 0.12	100.0		100.0	0.0	100.0 [e]	
Methicillin-resistant *Staphylococcus aureus* (United States; *n*=60)									
Delafloxacin	0.25	1	0.002 to 4			73.3 [b]	26.7	73.3 [b]	18.3
Levofloxacin	4	>4	0.12 to >4	35.0	65.0	35.0 [c]	65.0	35.0 [d]	46.7
Moxifloxacin	2	>4	≤0.06 to >4	35.0	60.0	35.0	65.0	35.0 [e]	60.0
Minocycline	0.06	0.5	≤0.03 to >8	98.3	1.7	96.7	3.3	98.3 [e]	1.7
Oritavancin	0.03	0.03	≤0.008 to 0.12	100.0		100.0	0.0	100.0 [e]	
***Staphylococcus aureus* (Europe; *n*=182)**									
Delafloxacin	0.008	0.5	≤0.0005 to 8			85.2 [b]	14.8	85.2 [b]	6.6
Levofloxacin	0.25	>4	0.06 to >4	75.4	24.6	75.4 [c]	24.6	75.4 [d]	22.0
Moxifloxacin	≤0.06	4	≤0.06 to >4	75.3	22.5	75.3	24.7	75.3 [e]	23.5
Minocycline	0.06	0.12	≤0.03 to 8	98.9	0.0	97.9	2.2	98.9 [e]	0.0
Oritavancin	0.03	0.06	≤0.008 to 0.12	100.0		100.0	0.0	100.0 [e]	
Methicillin-resistant *Staphylococcus aureus* (Europe; *n*=42)									
Delafloxacin	0.5	2	0.004 to 8			45.2 [b]	54.8	45.2 [b]	23.8
Levofloxacin	>4	>4	0.12 to >4	19.0	81.0	19.0 [c]	81.0	19.0 [d]	73.8
Moxifloxacin	4	>4	≤0.06 to >4	19.0	76.2	19.0	81.0	19.0 [e]	76.2
Minocycline	0.06	0.25	0.06 to 8	95.2	0.0	92.9	7.1	95.2 [e]	0.0
Oritavancin	0.015	0.06	≤0.008 to 0.12	100.0		100.0	0.0	100.0 [e]	
***Staphylococcus aureus* (United States and Europe; *n*=336)**									
Delafloxacin	0.008	0.5	≤0.0005 to 8			86.3 [b]	13.7	86.3 [b]	7.1
Levofloxacin	0.25	>4	0.06 to >4	75.3	28.6	75.3 [c]	28.6	75.3 [d]	22.3
Moxifloxacin	≤0.06	4	≤0.06 to >4	71.4	25.9	71.4	28.6	71.4 [e]	25.9
Minocycline	0.06	0.12	≤0.03 to >8	98.8	0.6	97.9	2.1	98.8 [e]	0.6
Oritavancin	0.03	0.06	≤0.008 to 0.12	100.0		100.0	0.0	100.0 [e]	
***Streptococcus agalactiae* (United States and Europe; *n*=34)**									
Delafloxacin	0.015	0.03	0.008 to 0.06			97.1 [b]	2.9	100.0 [b]	
Levofloxacin	1	1	0.5 to 2	100.0	0.0	100.0 [c]	0.0	100.0	0.0
Moxifloxacin	0.12	0.25	0.06 to 0.25			100.0	0.0		
Erythromycin	4	>16	0.03 to >16	33.3	66.7	33.3	66.7	33.3	66.7 [e]
Oritavancin	0.06	0.06	0.03 to 0.25	100.0		100.0	0.0	100.0 [e]	
***Enterococcus faecalis* (United States and Europe; *n*=24)**									
Delafloxacin	0.12	0.5	0.03 to 1			IE [f]	IE	83.3	12.5
Levofloxacin	1	>4	0.5 to >4	83.3	16.7	83.3 [g]	16.7	83.3	16.7 [e]
Moxifloxacin	0.25	>4	0.06 to >4					83.3	16.7
Oritavancin	0.015	0.03	0.004 to 0.03	100.0 [h]				100.0 [e,h]	
Vancomycin	1	2	0.5 to 2	100.0	0.0	100.0	0.0	100.0 [e]	0.0

[a] Criteria as published by CLSI (2025), EUCAST (2025), and US FDA (2025).
[b] Using ABSSSI breakpoints.
[c] An arbitrary susceptible breakpoint of ≤0.001 mg/L and/or >50 mm has been published by EUCAST indicating that susceptible should not be reported for this organism-agent combination and intermediate should be interpreted as susceptible, increased exposure.
[d] US FDA breakpoints were applied to all *Staphylococcus aureus*, but they are only approved for MSSA (methicillin-susceptible *S. aureus*) isolates.
[e] CLSI M100 standard is recognized.
[f] IE = Insufficient evidence that the organism or group is a good target for therapy with the agent.
[g] Uncomplicated urinary tract infection only.
[h] Vancomycin-susceptible isolates only.

Table 2. Activity of delafloxacin and comparators against Enterobacterales isolates from patients with diabetic foot infections in the United States and Europe (2017–2023)

Antimicrobial Agent (Region; no. tested)	MIC (mg/L)			CLSI[a]		EUCAST[a]		US FDA[a]	
	MIC_{50}	MIC_{90}	MIC range	%S	%R	%S	%R	%S	%R
Enterobacterales (United States; *n*=180)									
Delafloxacin	0.12	2	0.008 to >16			62.7 [b]	37.3	72.2 [c,d]	21.7
Ciprofloxacin	≤0.03	4	≤0.03 to >4	83.3	14.4	83.3 [e]	16.7	83.3 [f]	14.4
Levofloxacin	0.06	4	≤0.03 to >16	85.0	13.3	85.0	13.3	85.0 [f]	13.3
Minocycline (*n*=115)	2	32	0.25 to >32	60.0	25.2			60.0 [f]	25.2
Meropenem-vaborbactam	0.03	0.06	≤0.015 to 0.12	100.0		100.0	0.0	100.0 [f]	
Enterobacterales (Europe; *n*=134)									
Delafloxacin	0.12	4	0.008 to >16			58.2 [b]	41.8	65.7 [c,d]	27.6
Ciprofloxacin	≤0.03	4	≤0.03 to >4	75.4	21.6	75.4 [e]	21.6	75.4 [f]	21.6
Levofloxacin	0.06	4	≤0.03 to >16	79.9	14.9	79.9	14.9	79.9 [f]	14.9
Minocycline (*n*=133)	2	16	0.25 to >32	70.7	17.3			70.7 [f]	17.3
Meropenem-vaborbactam	0.03	0.06	≤0.015 to 8	99.3	0.0	100.0	0.0	99.3 [f]	0.0
Enterobacterales (United States and Europe; *n*=314 [g])									
Delafloxacin	0.12	4	0.008 to >16			60.8 [b]	39.2	69.4 [c,d]	24.2
Ciprofloxacin	≤0.03	4	≤0.03 to >4	79.9	17.5	79.9 [e]	20.1	79.9 [f]	17.5 [f]
Levofloxacin	0.06	4	≤0.03 to >16	82.8	14.0	82.8	14.0	82.8 [f]	14.0
Minocycline (*n*=248)	2	16	0.25 to >32	65.7	21.0			65.7 [f]	21.0
Meropenem-vaborbactam	0.03	0.06	≤0.015 to 8	99.7	0.0	100.0	0.0	99.7 [f]	0.0
***Citrobacter* spp. (United States and Europe; *n*=28 [g])**									
Delafloxacin	0.06	2	0.015 to 8					75.0 [c,d]	14.3
Ciprofloxacin	≤0.03	0.12	≤0.03 to 1	96.4	3.6	96.4 [e]	3.6	96.4 [f]	3.6 [f]
Levofloxacin	0.06	1	≤0.03 to 2	89.3	3.6	89.3	3.6	89.3 [f]	3.6
Minocycline (*n*=17)	2	4	0.5 to >32	94.1	5.9			94.1 [f]	5.9
Meropenem-vaborbactam	0.015	0.03	≤0.015 to 0.03	100.0	0.0	100.0	0.0	100.0 [f]	0.0
***Enterobacter cloacae* and *E. cloacae* species complex (United States and Europe; *n*=39)**									
Delafloxacin	0.12	2	0.008 to >16					87.2 [c,d]	10.3
Ciprofloxacin	≤0.03	0.5	≤0.03 to 4	89.7	7.7	89.7 [e]	7.7	89.7 [f]	7.7 [f]
Levofloxacin	0.06	0.25	≤0.03 to 2	94.8	2.6	94.8	2.6	94.8 [f]	2.6
Minocycline (*n*=31)	2	4	0.5 to >32	93.5	3.2			93.5 [f]	3.2
Meropenem-vaborbactam	0.015	0.03	≤0.015 to 0.25	100.0	0.0	100.0	0.0	100.0 [f]	0.0
***Escherichia coli* (United States and Europe; *n*=56)**									
Delafloxacin	0.06	4	0.008 to >16			62.5	37.5	71.4 [c,d]	25.0
Ciprofloxacin	≤0.03	>4	≤0.03 to >4	71.4	23.2	71.4 [e]	23.2	71.4 [f]	23.2 [e]
Levofloxacin	0.03	16	≤0.03 to >16	76.8	23.2	76.8	23.2	76.8 [f]	23.2
Minocycline (*n*=49)	1	8	0.25 to >32	84.6	7.7			84.6 [f]	7.7
Meropenem-vaborbactam	≤0.015	0.03	≤0.015 to 0.03	100.0	0.0	100.0	0.0	100.0 [f]	0.0
***Klebsiella pneumoniae* (United States and Europe; *n*=21)**									
Delafloxacin	0.12	>16	0.03 to >16					57.1 [c,d]	42.9
Ciprofloxacin	≤0.03	>16	≤0.03 to >16	52.4	42.9	52.4 [e]	47.6	52.4 [f]	42.9 [f]
Levofloxacin	0.06	32	≤0.03 to 32	71.4	28.6	71.4	28.6	71.4 [f]	28.6
Minocycline (*n*=16)	2	16	1 to >32	87.5	12.5			87.5 [f]	12.5
Meropenem-vaborbactam	0.03	0.06	≤0.015 to 8	95.2	4.8	100.0	0.0	95.2 [f]	4.8
***Proteus mirabilis* (United States and Europe; *n*=47 [g])**									
Delafloxacin	0.06	2	0.015 to >16					76.6 [c,d]	19.1
Ciprofloxacin	0.03	4	≤0.03 to >16	74.5	25.5	74.5 [e]	25.5	74.5 [f]	25.5 [f]
Levofloxacin	0.06	4	0.03 to >32	74.5	17.0	74.5	17.0	74.5 [f]	17.0
Minocycline (*n*=41)	16	>32	2 to >32	9.8	73.2			9.8 [f]	73.2
Meropenem-vaborbactam	0.06	0.12	0.03 to 0.25	100.0	0.0	100.0	0.0	100.0 [f]	0.0

[a] Criteria as published by CLSI (2025), EUCAST (2025), and US FDA (2025).
[b] *E. coli* breakpoints applied to all Enterobacterales.
[c] Using ABSSSI breakpoints.
[d] US FDA breakpoints published for *E. coli*, *K. pneumoniae*, and *E. cloacae* were applied to all Enterobacterales isolates.
[e] Using non-meningitis breakpoints.
[f] CLSI M100 standard is recognized.
[g] Organisms include *Citrobacter freundii* species complex (15), *C. koseri* (13), *Enterobacter cloacae* (14), *E. cloacae* species complex (25), *Escherichia coli* (56), *E. hermannii* (2), *Klebsiella aerogenes* (5), *K. oxytoca* (23), *K. pneumoniae* (21), *K. variicola* (1), *Morganella morganii* (32), *Proteus hauseri* (2), *P. mirabilis* (47), *P. penneri* (2), *P. vulgaris* (9), *P. vulgaris* group (16), *Providencia rettgeri* (4), *P. stuartii* (5), *Serratia liquefaciens* (2), *S. marcescens* (17), and unspeciated *Raoultella* (2).

Table 3. Activity of delafloxacin and comparators against *Pseudomonas aeruginosa* isolates from patients with diabetic foot infections in the United States and Europe (2017–2023)

Antimicrobial Agent (Region; no. tested)	MIC (mg/L)			CLSI[a]		EUCAST[a]		US FDA[a]	
	MIC_{50}	MIC_{90}	MIC range	%S	%R	%S	%R	%S	%R
***Pseudomonas aeruginosa* (United States; *n*=43)**									
Delafloxacin	0.5	2	0.12 to 16			IE [b]	IE [b]	74.4 [c]	18.6
Ciprofloxacin	0.12	1	0.06 to 8	86.0	9.3	86.0 [d]	14.0	86.0 [e]	9.3
Levofloxacin	0.5	4	0.25 to 16	79.1	16.3	83.7 [d]	16.3	79.1 [e]	16.3
Meropenem-vaborbactam	0.25	2	0.06 to 8			100.0	0.0		
***Pseudomonas aeruginosa* (Europe; *n*=49)**									
Delafloxacin	0.5	16	0.015 to >16			IE [b]	IE [b]	59.2 [c]	28.6
Ciprofloxacin	0.12	>4	≤0.03 to >16	73.5	24.5	73.5 [d]	26.5	73.5 [e]	24.5
Levofloxacin	1	32	0.06 to >32	59.2	24.5	75.5 [d]	24.5	59.2 [e]	24.5
Meropenem-vaborbactam	0.5	8	0.06 to >32			91.8	8.2		
***Pseudomonas aeruginosa* (United States and Europe; *n*=92)**									
Delafloxacin	0.5	8	0.015 to >16			IE [b]	IE [b]	66.3 [c]	23.9
Ciprofloxacin	0.12	8	≤0.03 to >16	79.3	17.4	79.3 [d]	21.7	79.3 [e]	17.4
Levofloxacin	0.5	16	0.06 to >32	68.5	20.7	79.3 [d]	20.7	68.5 [e]	20.7
Meropenem-vaborbactam	0.5	2	0.06 to >32			95.7	4.3		

[a] Criteria as published by CLSI (2025), EUCAST (2025), and US FDA (2025).
[b] IE = Insufficient evidence that the organism or group is a good target for therapy with the agent.
[c] Using ABSSSI breakpoints.
[d] An arbitrary susceptible breakpoint of ≤0.001 mg/L and/or >50 mm has been published by EUCAST indicating that susceptible should not be reported for this organism-agent combination and intermediate should be interpreted as susceptible, increased exposure.
[e] CLSI M100 standard is recognized.

Figure 1. Prevalence of DFI pathogens from patients in the United States and Europe (SENTRY 2017 to 2023)

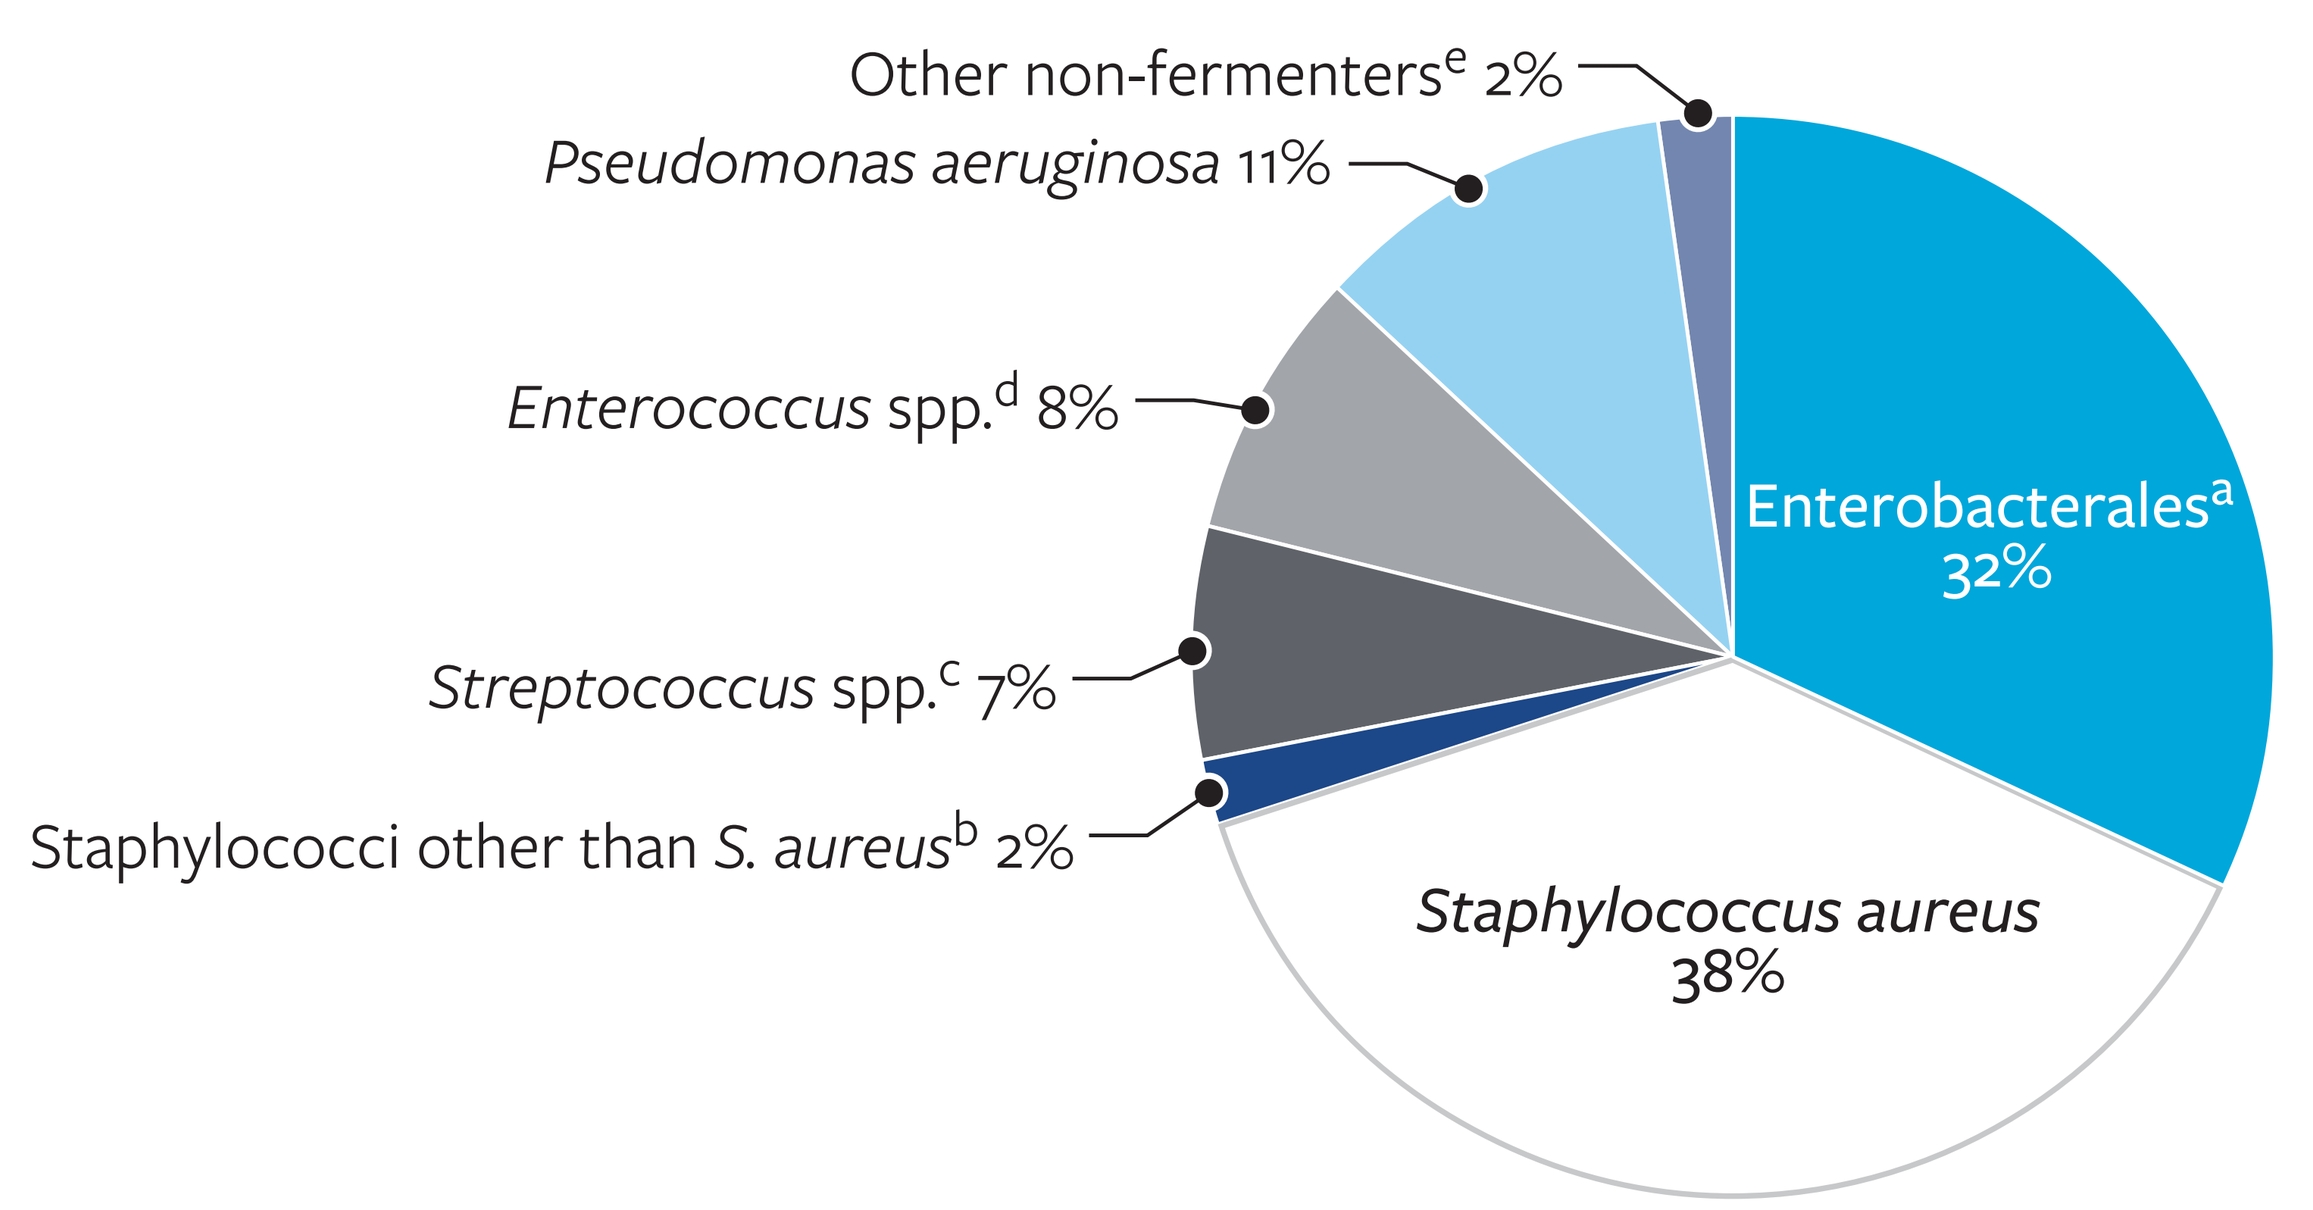
Other non-fermenters[e] 2%
Pseudomonas aeruginosa 11%
Enterococcus spp.[d] 8%
Enterobacterales[a] 32%
Streptococcus spp.[c] 7%
Staphylococci other than S. aureus[b] 2%
Staphylococcus aureus 38%

[a] Contains *Citrobacter freundii* species complex (12), *C. koseri* (6), *Enterobacter cloacae* (12), *E. cloacae* species complex (21), *Escherichia coli* (56), *E. hermannii* (2), *Klebsiella aerogenes* (4), *K. oxytoca* (19), *K. pneumoniae* (14), *K. variicola* (1), *Morganella morganii* (24), *Proteus hauseri* (2), *P. mirabilis* (40), *P. penneri* (2), *P. vulgaris* (4), *P. vulgaris* group (8), *Providencia rettgeri* (1), *P. stuartii* (3), *Serratia liquefaciens* (2), *S. marcescens* (17), and unspeciated *Raoultella* (2).
[b] Contains *Staphylococcus capitis* (2), *S. epidermidis* (4), *S. haemolyticus* (2), *S. lugdunensis* (8), *S. pseudintermedius* (1), *S. simulans* (1).
[c] Contains *Streptococcus agalactiae* (38), *S. anginosus* group (6), *S. canis* (1), *S. dysgalactiae* (10), *S. pyogenes* (4).
[d] Contains *Enterococcus avium* (2), *E. casseliflavus* (1), *E. faecalis* (44), *E. faecium* (16), *E. raffinosus* (2).
[e] Contains *Acinetobacter baumannii-calcoaceticus* species complex (8), *A. courvalinii* (1), *Stenotrophomonas maltophilia* (4).

Acknowledgments

The authors thank the participating medical centers for providing the bacterial isolates.

References

CLSI 2024. M07Ed12. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. Clinical and Laboratory Standards Institute, Wayne, PA.

CLSI. 2023. M100Ed35. Performance standards for antimicrobial susceptibility testing: 35th informational supplement. Clinical and Laboratory Standards Institute, Wayne, PA.

United States Food and Drug Administration (2025). Antibacterial susceptibility test interpretive criteria. Accessed July 2025. https://www.fda.gov/Drugs/Development ApprovalProcess/DevelopmentResources/ucm575163.htm

Contact

element

SCAN ME

To obtain a PDF of this poster:

Scan the QR code or visit https://www .jmilabs.com/data/posters/IDWeek2025 _25-MEL-02_P1_Delafloxacin.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia