IDWEEK 2025 | Poster #P1333

Activity of Zosurabalpin and Comparator Antimicrobial Agents Tested Against *Acinetobacter baumannii-calcoaceticus* Complex (ABC) Isolates Collected Worldwide in 2024

Joshua M. Maher[1], Mariana Castanheira[1], Kelley Fedler[1], Severine Louvel[2]

[1] Element Iowa City (JMI Laboratories), North Liberty, IA, 52317, USA; [2] Roche Pharma Research and Early Development, Infectious Diseases, Roche Innovation Center Basel, F. Hoffmann La Roche Ltd, Grenzacherstrasse 124, 4070 Basel, Switzerland

Background

- Zosurabalpin (ZAB, RG6006) is a first-in-class tethered macrocyclic peptide antibiotic that targets intracellular transport of lipopolysaccharide inhibiting the $LptB_2FGC$ complex.
- Currently zosurabalpin is being developed for the treatment of carbapenem-resistant *Acinetobacter baumannii-calcoaceticus* complex (ABC) infections, a top priority pathogen on the Centers for Disease Control and Prevention (CDC) urgent threat list.
- This agent demonstrates *in vitro* activity against *Acinetobacter baumannii-calcoaceticus* complex clinical isolates, including carbapenem-resistant (CRABC) and multidrug-resistant (MDR) strains.
- The *in vitro* antimicrobial activity of zosurabalpin was monitored as part of the 2024 global SENTRY Antimicrobial Surveillance Program platform against 1,575 ABC bacterial isolates using Clinical and Laboratory Standards Institute (CLSI) reference susceptibility testing methods.

Results

- Zosurabalpin ($MIC_{50/90}$, 0.12/0.5 mg/L) inhibited all ABC isolates at ≤2 mg/L and 99.7% (1,571/1,575) at ≤1 mg/L.
- Resistance rates to standard-of-care agents varied by region, with 81.0%/75.0% MDR/CRABC seen in Latin American isolates (*n*=100), 69.9%/69.9% in Asia-Pacific isolates (*n*=166), 51.7%/50.1% in European isolates (*n*=747), and 31.5%/28.1% in United States isolates (*n*=562).
- Zosurabalpin activity was not impacted by differences in regional resistance rates and retained activity against resistant phenotypes (Figure 1) including CRABC (*n*=723 [45.9% overall]; $MIC_{50/90}$, 0.25/0.5 mg/L) and MDR (*n*=760 [48.3%]; $MIC_{50/90}$, 0.25/0.5 mg/L) isolates, inhibiting 99.7% at ≤1 mg/L and 100% at ≤2 mg/L.
- Among comparators, cefiderocol and sulbactam-durlobactam were the most active agents inhibiting 93.0% and 95.2% of the isolates, respectively.
 - Cefiderocol and sulbactam-durlobactam inhibited 86.3% and 89.6% of the CRABC isolates and 86.6% and 90.1% of the MDR isolates, respectively (Figure 2).
- Susceptibility rates for the remaining agents ranged from 51.4% for ampicillin-sulbactam to 96.4% for colistin (Figure 2).
 - The activity of amikacin, ampicillin-sulbactam, and minocycline comparators was markedly reduced against CRABC and MDR isolates, with susceptibility rates of 3.5–28.3%; colistin susceptibility rates were 93.6–93.7% against these resistant phenotypes (Figure 2).
- Cefiderocol-resistant (*n*=74), sulbactam-durlobactam-resistant (*n*=62), and 24 isolates resistant to both agents were all inhibited by zosurabalpin at ≤1 mg/L.

Methods

- A total of 1575 bacterial isolates were collected in 109 medical centers located in 30 countries in Europe (747 isolates; 47.4% overall; 19 countries), Latin America (100 isolates; 6.3% overall; 4 countries), the Asia-Pacific region (166 isolates; 10.5% overall; 7 countries) and in the United States (562 isolates; 35.7% overall; 9 US census divisions).
- Isolates originated from patients hospitalized with pneumonia (*n*=592), skin and skin structure infections (*n*=317), bloodstream infections (*n*=208), urinary tract infections (*n*=183), and other infections (*n*=275).
- Zosurabalpin was tested in cation-adjusted Mueller-Hinton broth (CAMHB) supplemented with 20% heat-inactivated horse serum (HoS), as approved by the CLSI.
- Comparator agents were tested using CLSI reference methods and CLSI and FDA interpretive breakpoint criteria were applied.
- CRABC were resistant to imipenem and/or meropenem and MDR were resistant to ≥3 antimicrobial classes applying CLSI breakpoints.

Conclusions

- ABC isolates are often CRABC and/or MDR and therapeutic options to treat infections caused by these isolates remain limited.
- Zosurabalpin sustained potent activity against antimicrobial-resistant and MDR phenotypes, including isolates resistant to new agents including cefiderocol and sulbactam-durlobactam.
- The potent *in vitro* activity of zosurabalpin observed against a large collection of clinical ABC isolates establishes this agent as a potential option to combat the growing threat of antimicrobial resistance and further development is warranted.

Acknowledgments

The authors would like to thank Arielle Watson, Gina Bartleson, and Jill Arends for editorial assistance during preparation of this poster. The project described herein was funded in part with federal funds from the U.S. Department of Health and Human Services (HHS); Administration for Strategic Preparedness and Response (ASPR); and Biomedical Advanced Research and Development Authority (BARDA), under contract number HHSO100201600038C. The contract and federal funding are not an endorsement of the study results, product, or company.

Figure 1. Distribution of 1,575 *A. baumannii-calcoaceticus* complex isolate resistant phenotypes

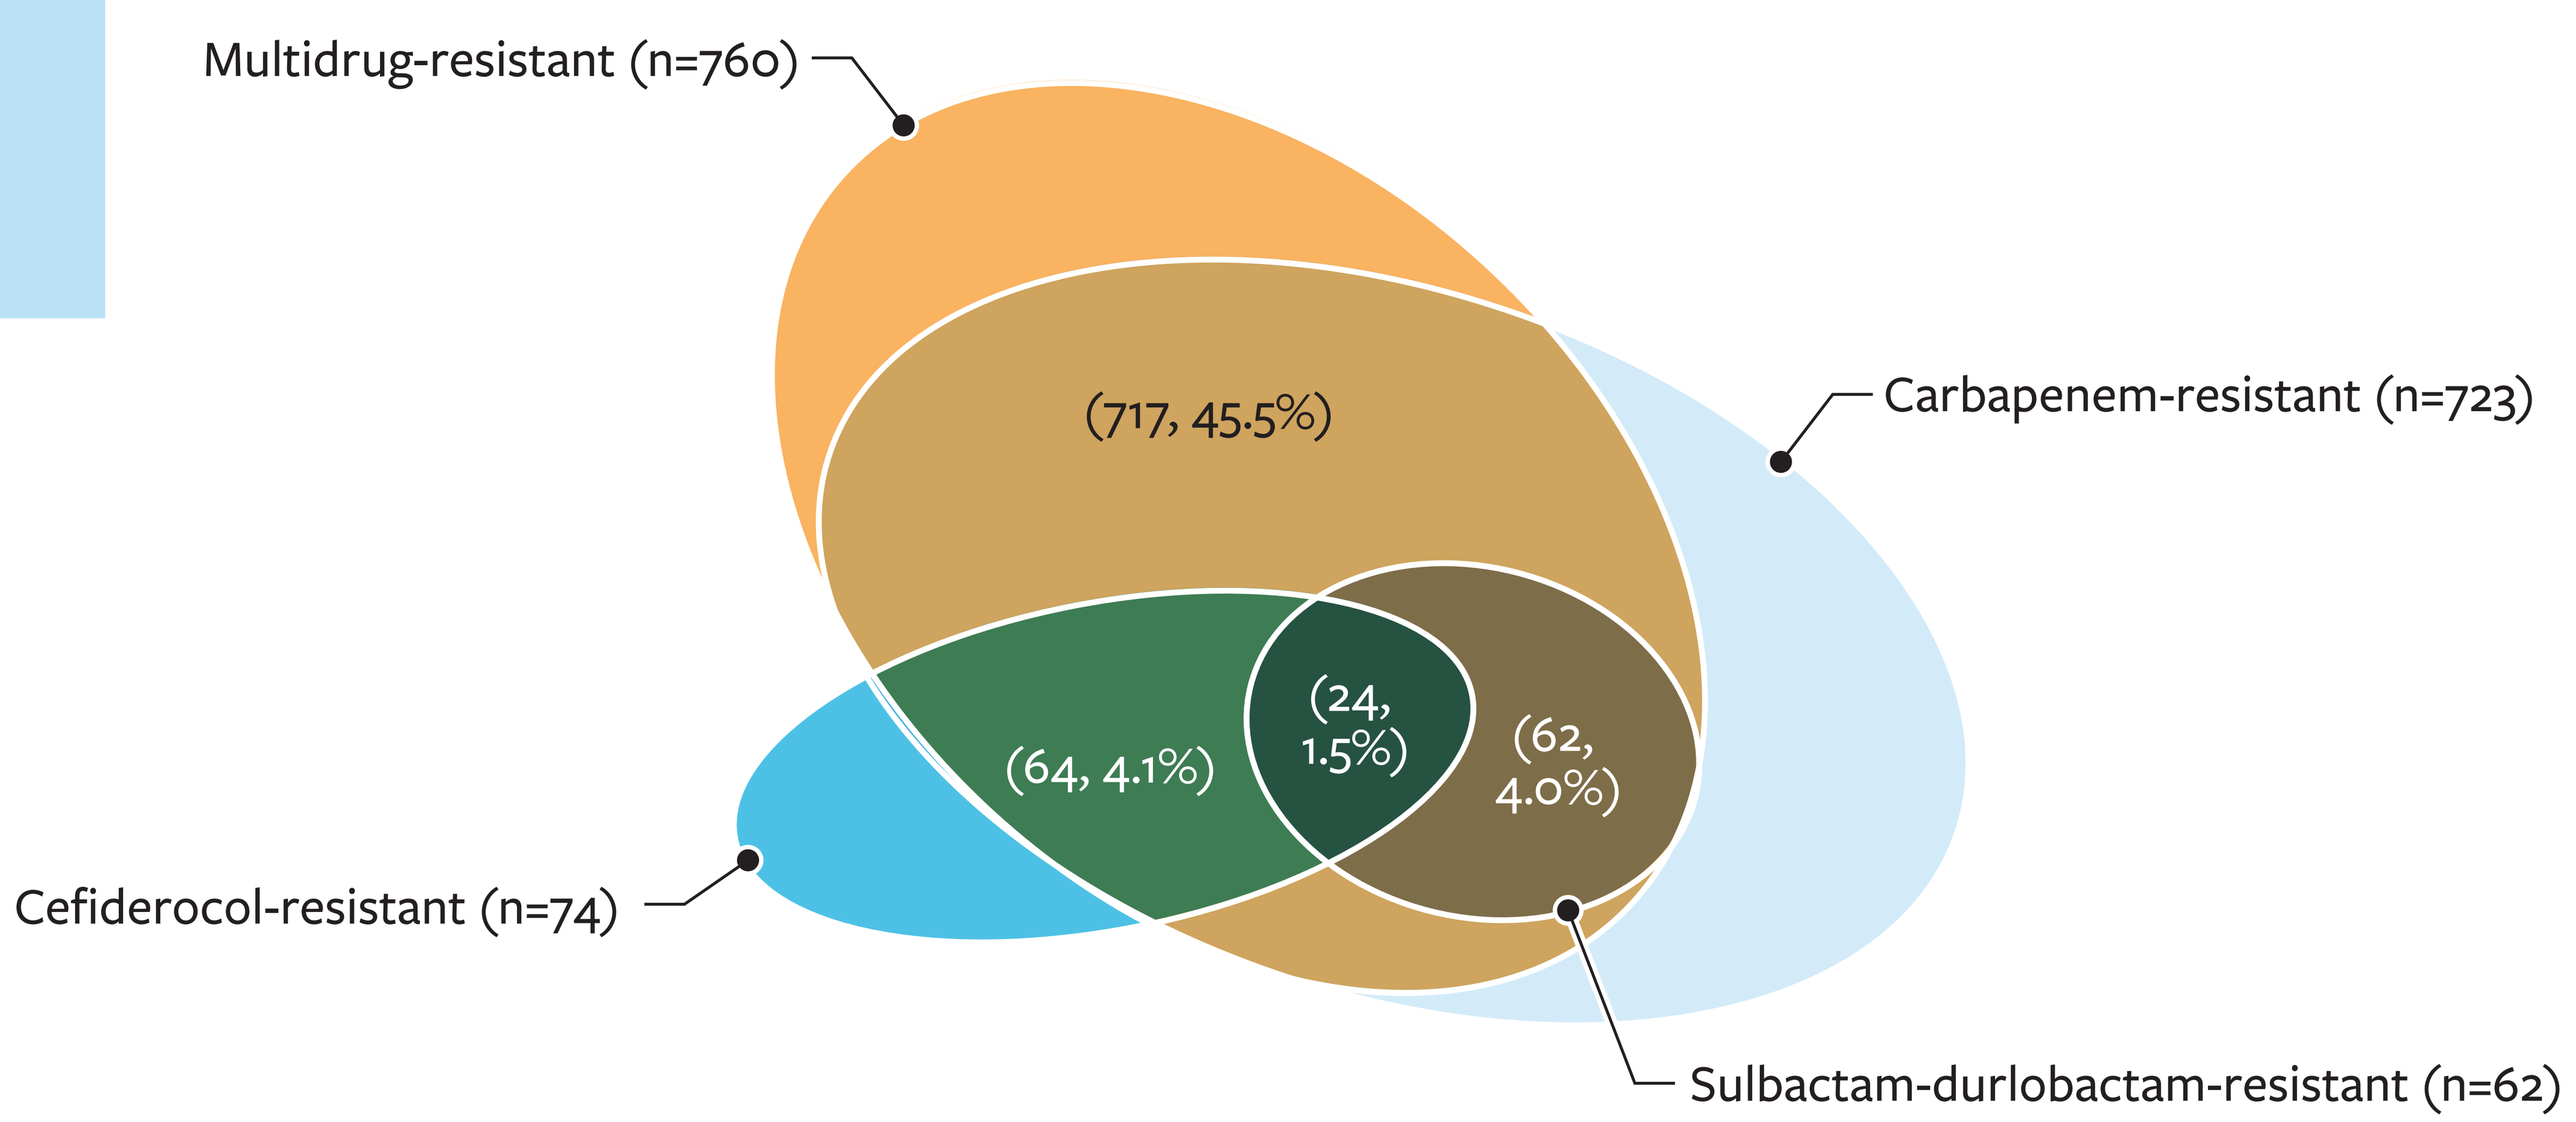

Multidrug-resistant (n=760)
Carbapenem-resistant (n=723)
Cefiderocol-resistant (n=74)
Sulbactam-durlobactam-resistant (n=62)
(717, 45.5%)
(64, 4.1%)
(24, 1.5%)
(62, 4.0%)

	MDR	CRABC	FDC-R	SUD-R
MDR	760	717	67	62
CRABC	717	723	64	62
FDC-R	67	64	74	24
SUD-R	62	62	24	62

Abbreviations: CRABC, carbapenem-resistant *A. baumannii-calcoaceticus* complex; FDC, cefiderocol; MDR, multidrug-resistant; R, resistant; SUD, sulbactam-durlobactam.

Figure 2. Activity of zosurabalpin and comparator agents against 1,575 *A. baumannii-calcoaceticus* complex isolates and resistant phenotypes

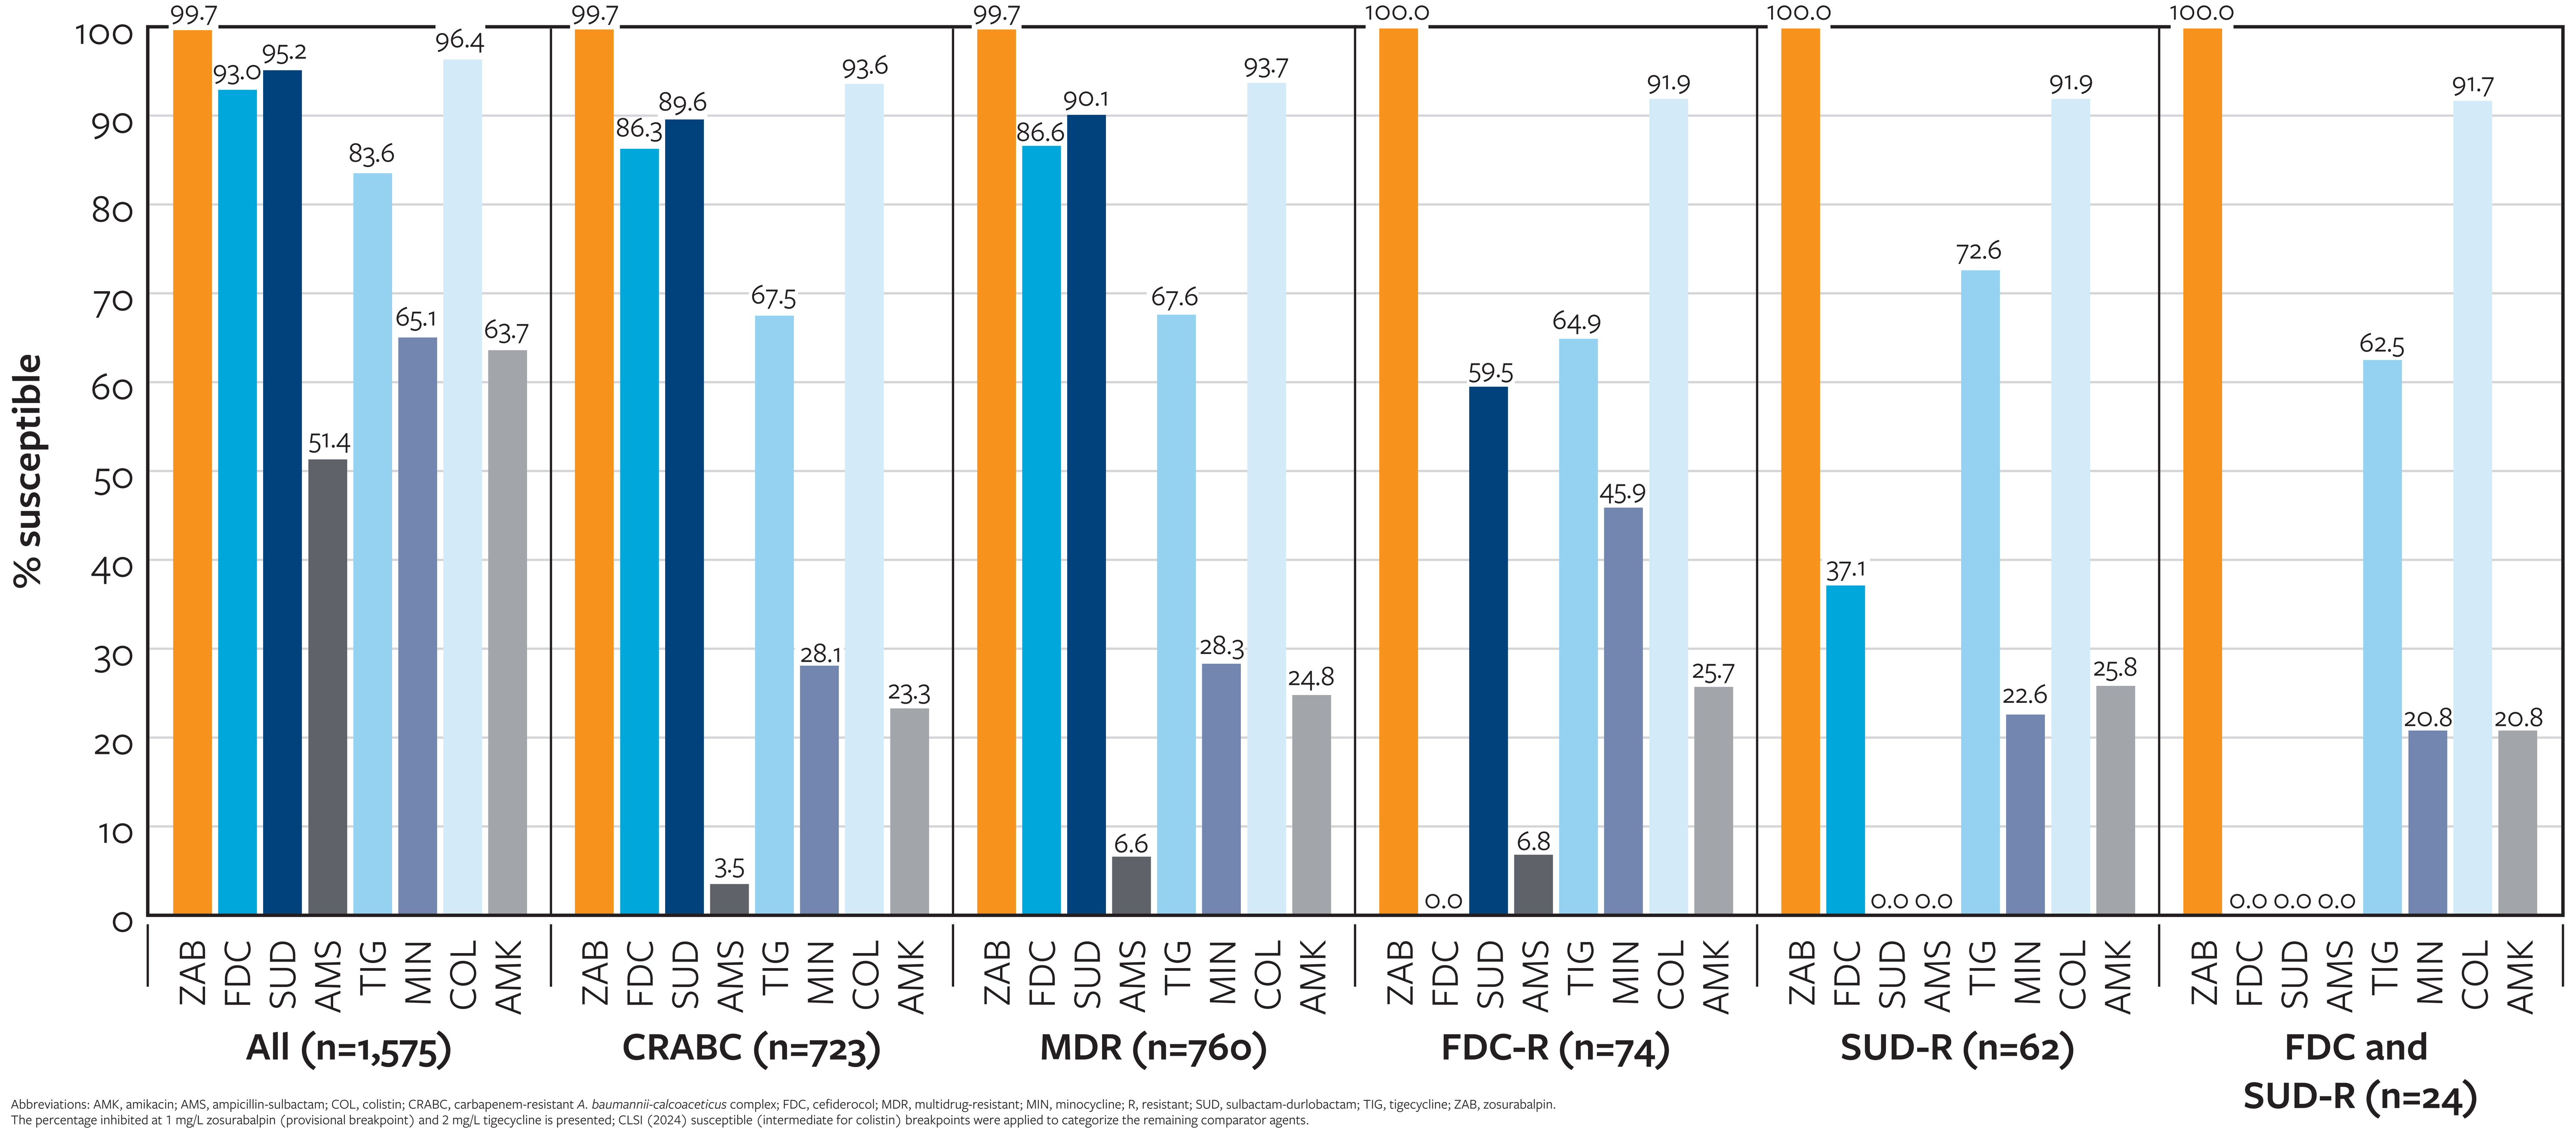

% susceptible
0 10 20 30 40 50 60 70 80 90 100
ZAB FDC SUD AMS TIG MIN COL AMK
All (n=1,575)
99.7 93.0 95.2 51.4 83.6 65.1 96.4 63.7
CRABC (n=723)
99.7 86.3 89.6 3.5 67.5 28.1 93.6 23.3
MDR (n=760)
99.7 86.6 90.1 6.6 67.6 28.3 93.7 24.8
FDC-R (n=74)
100.0 0.0 59.5 6.8 64.9 45.9 91.9 25.7
SUD-R (n=62)
100.0 37.1 0.0 0.0 72.6 22.6 91.9 25.8
FDC and SUD-R (n=24)
100.0 0.0 0.0 0.0 62.5 20.8 91.7 20.8

Abbreviations: AMK, amikacin; AMS, ampicillin-sulbactam; COL, colistin; CRABC, carbapenem-resistant *A. baumannii-calcoaceticus* complex; FDC, cefiderocol; MDR, multidrug-resistant; MIN, minocycline; R, resistant; SUD, sulbactam-durlobactam; TIG, tigecycline; ZAB, zosurabalpin.
The percentage inhibited at 1 mg/L zosurabalpin (provisional breakpoint) and 2 mg/L tigecycline is presented; CLSI (2024) susceptible (intermediate for colistin) breakpoints were applied to categorize the remaining comparator agents.

References

CLSI. M100Ed35E. *Performance Standards for Antimicrobial Susceptibility Testing*. Wayne, PA, Clinical and Laboratory Standards Institute, 2025.

CLSI. *Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically*. 12th ed. CLSI standard M07. Clinical and Laboratory Standards Institute, 2024.

FDA. 2025. FDA-recognized antimicrobial susceptibility test interpretive criteria. Available at https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria.

Magiorakos AP, et al. 2012. *Multidrug-resistant, extensively drug-resistant and pandrug-resistant bacteria: an international expert proposal for interim standard definitions for acquired resistance.* Clin Microbiol Infect 18:268–281.

Contact

element

SCAN ME

Mariana Castanheira, PhD, FIDSA, FAAM
Element Materials Technology/
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317 USA
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: mariana.castanheira@element.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2025_25-ROC-04_P1_ZAB_Surveillance.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia