IDWEEK 2025 | Poster #P1278

Activity of cefiderocol and comparator agents against carbapenem-resistant Enterobacterales, including molecularly characterized isolates recovered from United States hospitals in 2020–2024

Mariana Castanheira, John H. Kimbrough, Joshua M. Maher, Rodrigo E. Mendes
Element Iowa City (JMI Laboratories), IA, USA

Introduction

- Carbapenem-resistant Enterobacterales (CRE) are often multidrug resistant.
- The Infectious Diseases Society of America (IDSA) recommends meropenem-vaborbactam, ceftazidime-avibactam, imipenem-relebactam, and cefiderocol for the treatment of serious infections caused by KPC-producing organisms.
- The treatment of isolates producing metallo-β-lactamases (MBLs) is more challenging and cefiderocol is among the agents recommended by the IDSA for the treatment of these isolates.
- Cefiderocol is a siderophore cephalosporin that has increased stability to hydrolysis by β-lactamases, including serine-carbapenemases and MBLs.
- We evaluated the activity of cefiderocol and comparator agents against CRE isolates collected in US hospitals from 2020 to 2024.

Materials and Methods

- A total of 18,597 Enterobacterales were collected from 35 US hospitals during 2020–2024.
 - Isolates were identified as the cause of infection.
 - Isolates were limited to 1 per patient.
- Susceptibility testing was performed by the CLSI reference broth microdilution method as described by the Clinical and Laboratory Standards Institute (CLSI) M07 (2022) and M100 (2025) documents.
 - Cefiderocol was tested in iron-depleted cation-adjusted Mueller-Hinton broth.
- Quality control (QC) was performed according to the CLSI M100 (2025) criteria.
 - All QC MIC results were within acceptable ranges.
- Categorical interpretations for all comparator agents were those criteria found in the CLSI M100 (2025) or the US FDA website.
- CRE isolates resistant to imipenem (excluding *Proteus mirabilis*, *P. penneri*, and indole-positive Proteeae) or meropenem were submitted to whole genome sequencing and data analysis for the detection of β-lactam resistance mechanisms.
- WGS was performed on MiSeq or NextSeq (Illumina, San Diego, California, USA) instruments targeting a 30X coverage.
 - Sequences were *de novo* assembled.
 - Analysis of β-lactam resistance mechanisms was performed *in silico*.

Results

- Among surveillance Enterobacterales isolates, 176 (0.9% overall) were CREs.
 - A total of 76 isolates were *K. pneumoniae* and 100 isolates belonged to 9 other species/complexes.
- CRE isolates were observed in all US Census Divisions, except for the West North Central, with percentages ranging from 0.3% in New England to 2.0% in the Mid-Atlantic (Figure 1).
- Serine-carbapenemase-encoding genes were detected in 106 isolates, with bla_{KPC-2} (n=41) and bla_{KPC-3} (n=52) being the most common.
- MBL-encoding genes were observed in 29 (16.5% of the CRE) isolates and bla_{NDM-1} was noted in 13 isolates alone or with other carbapenemases.
 - MBL-producing isolates were detected in all US Census Divisions but the Pacific and West North Central divisions.
- A total of 41 CRE isolates carried no carbapenemase genes.
- Cefiderocol inhibited 96.0% of the CREs at the CLSI/FDA breakpoint (Figure 2).
 - Ceftazidime-avibactam, impenem-relebactam, and meropenem-vaborbactam were active against 81.2%, 72.2%, and 75.0% of the CRE isolates, respectively.
 - The activity of non-β-lactam agents ranged from 31.2% to 96.6% (tigecycline).
- Cefiderocol was active against 99.1%, 89.7%, and 92.7% of the isolates producing serine carbapenemases, MBLs, and carbapenemase-negative isolates, respectively.
 - β-lactam/β-lactamase inhibitor combinations had limited activity against MBL isolates.
 - Other comparators, with the exception of tigecycline (93.1% susceptible), inhibited <60% of the MBL-producing isolates.
 - 79.3% of the MBL-producing isolates were intermediate to colistin.
- Carbapenemase-negative isolates were more susceptible to comparator agents when compared to isolates producing carbapenemases.
 - The agents with lower susceptibility rates against carbapenemase-negative isolates were levofloxacin and imipenem-relebactam (53.7% and 75.6% susceptible).

Figure 1. Distribution of CRE isolates in the US Census Divisions

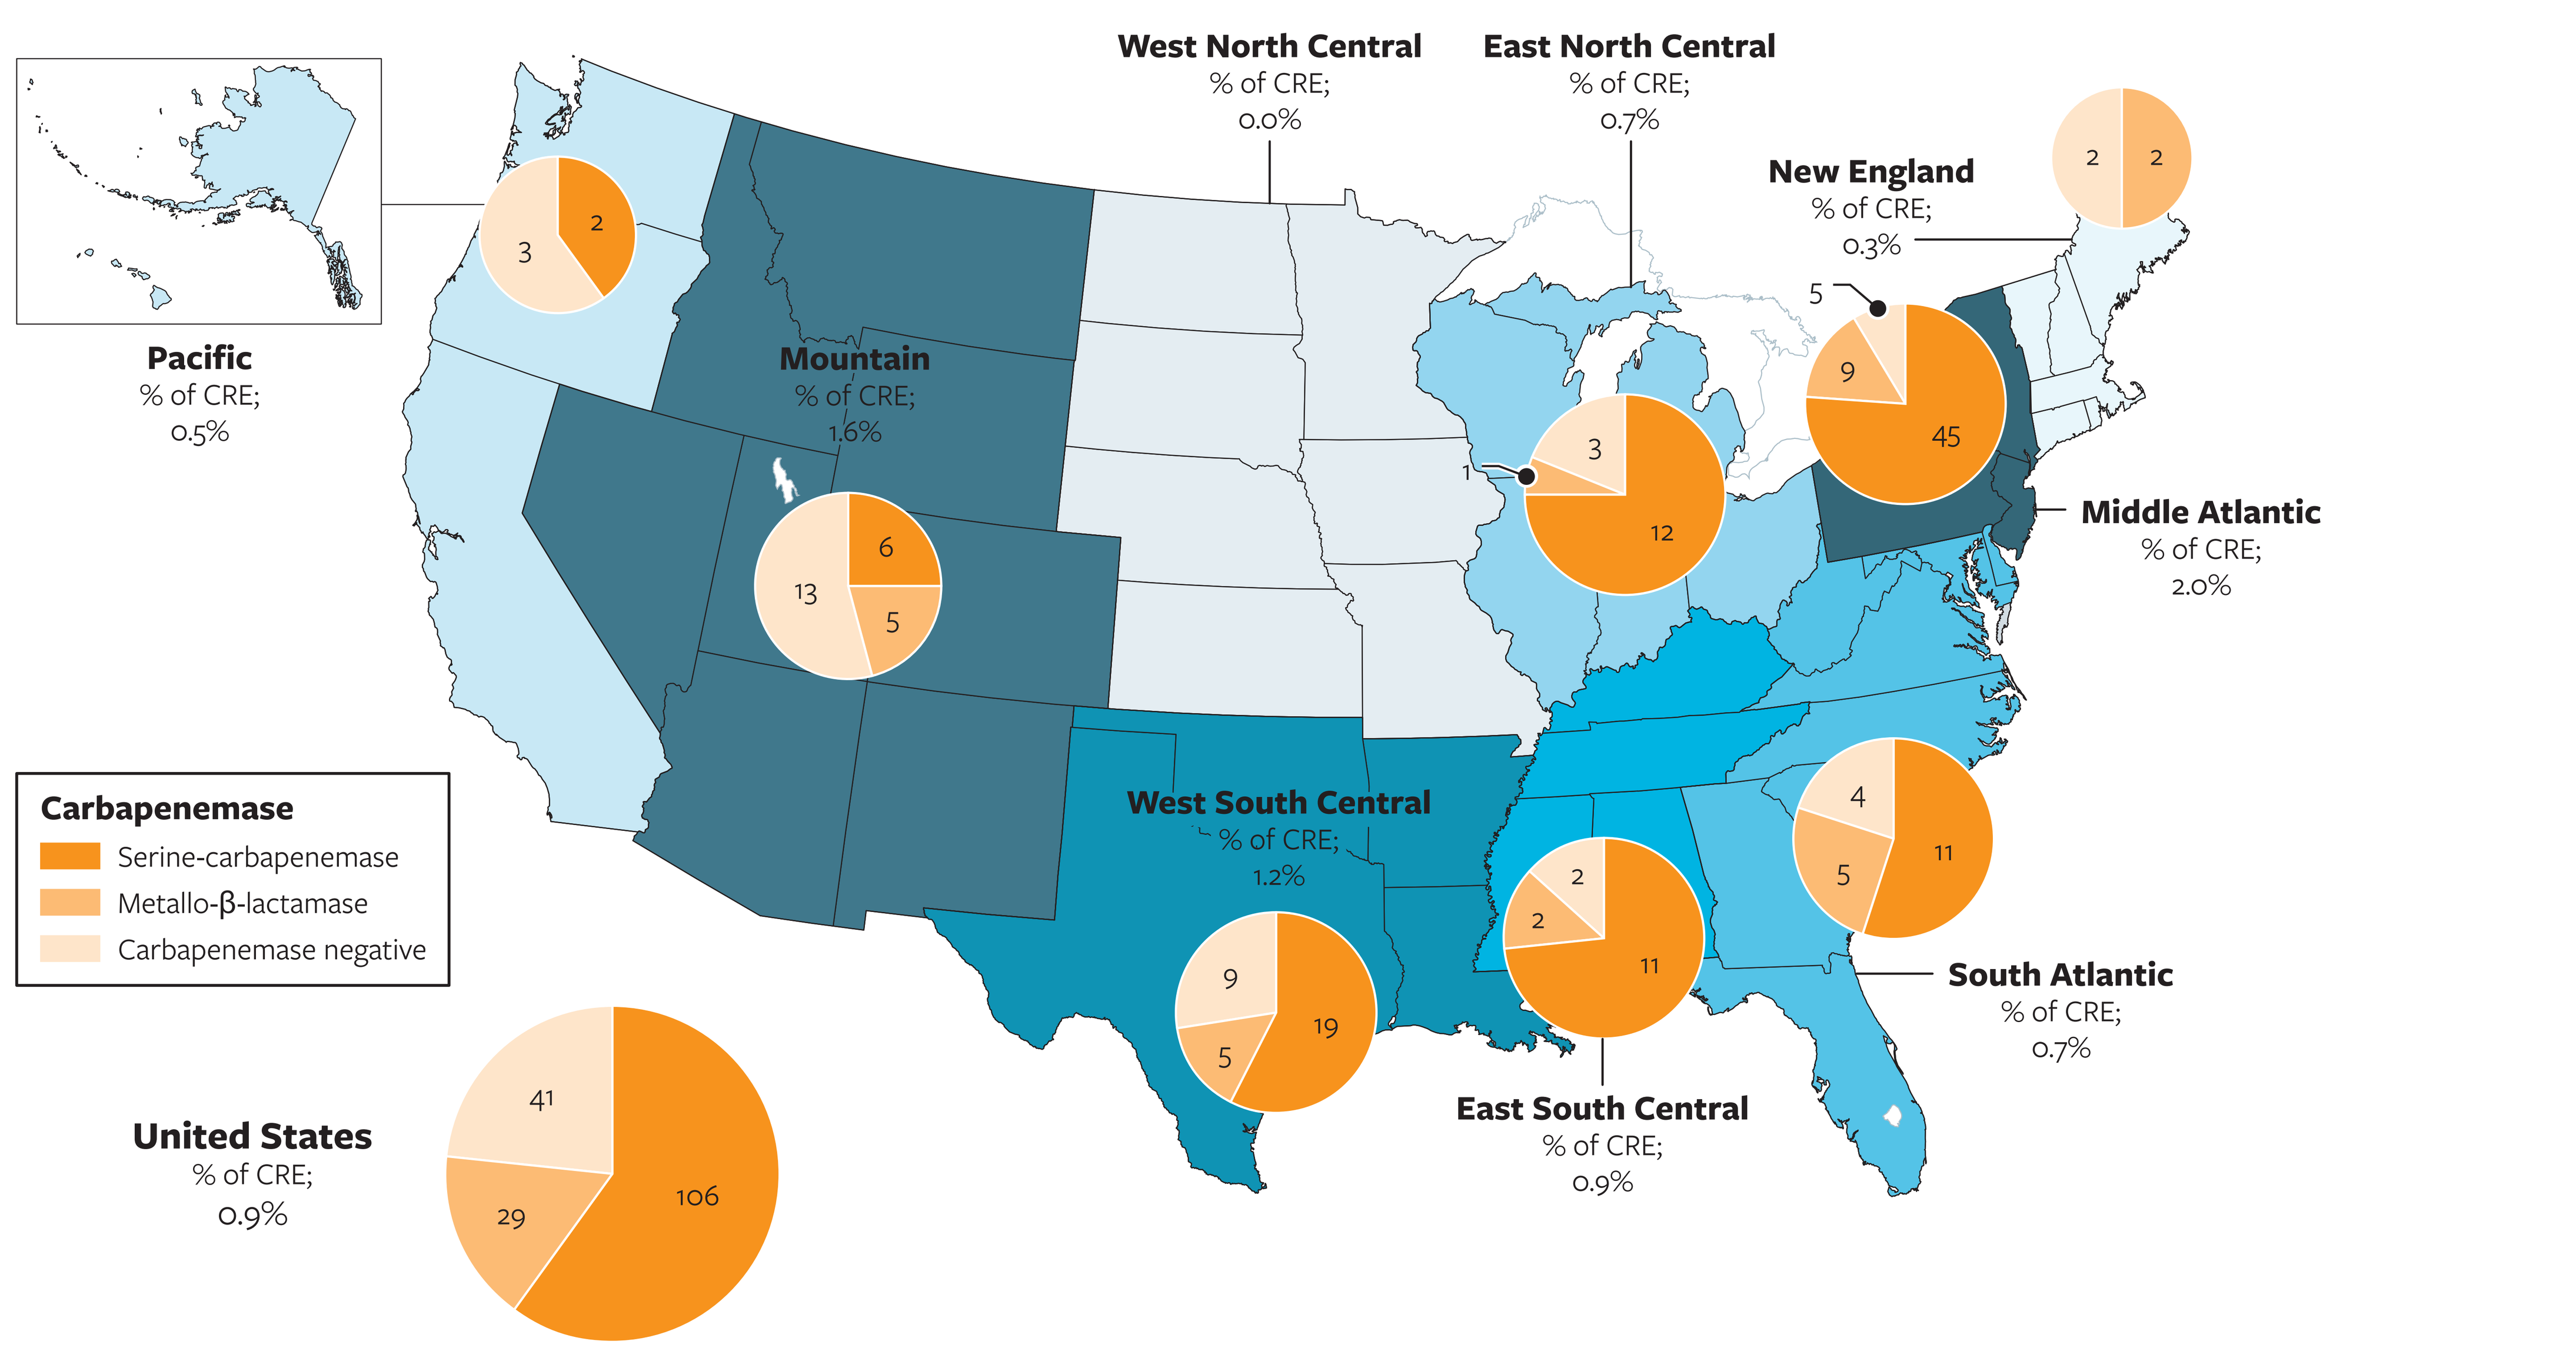
West North Central
% of CRE;
0.0%
East North Central
% of CRE;
0.7%
New England
% of CRE;
0.3%
Pacific
% of CRE;
0.5%
Mountain
% of CRE;
1.6%
Middle Atlantic
% of CRE;
2.0%
West South Central
% of CRE;
1.2%
East South Central
% of CRE;
0.9%
South Atlantic
% of CRE;
0.7%
United States
% of CRE;
0.9%
Carbapenemase
Serine-carbapenemase
Metallo-β-lactamase
Carbapenemase negative
3
2
6
13
5
1
3
12
5
9
45
2
2
9
19
5
2
2
11
4
11
5
41
29
106

Figure 2. Antimicrobial activity of cefiderocol and comparator agents against CRE isolates from US hospitals

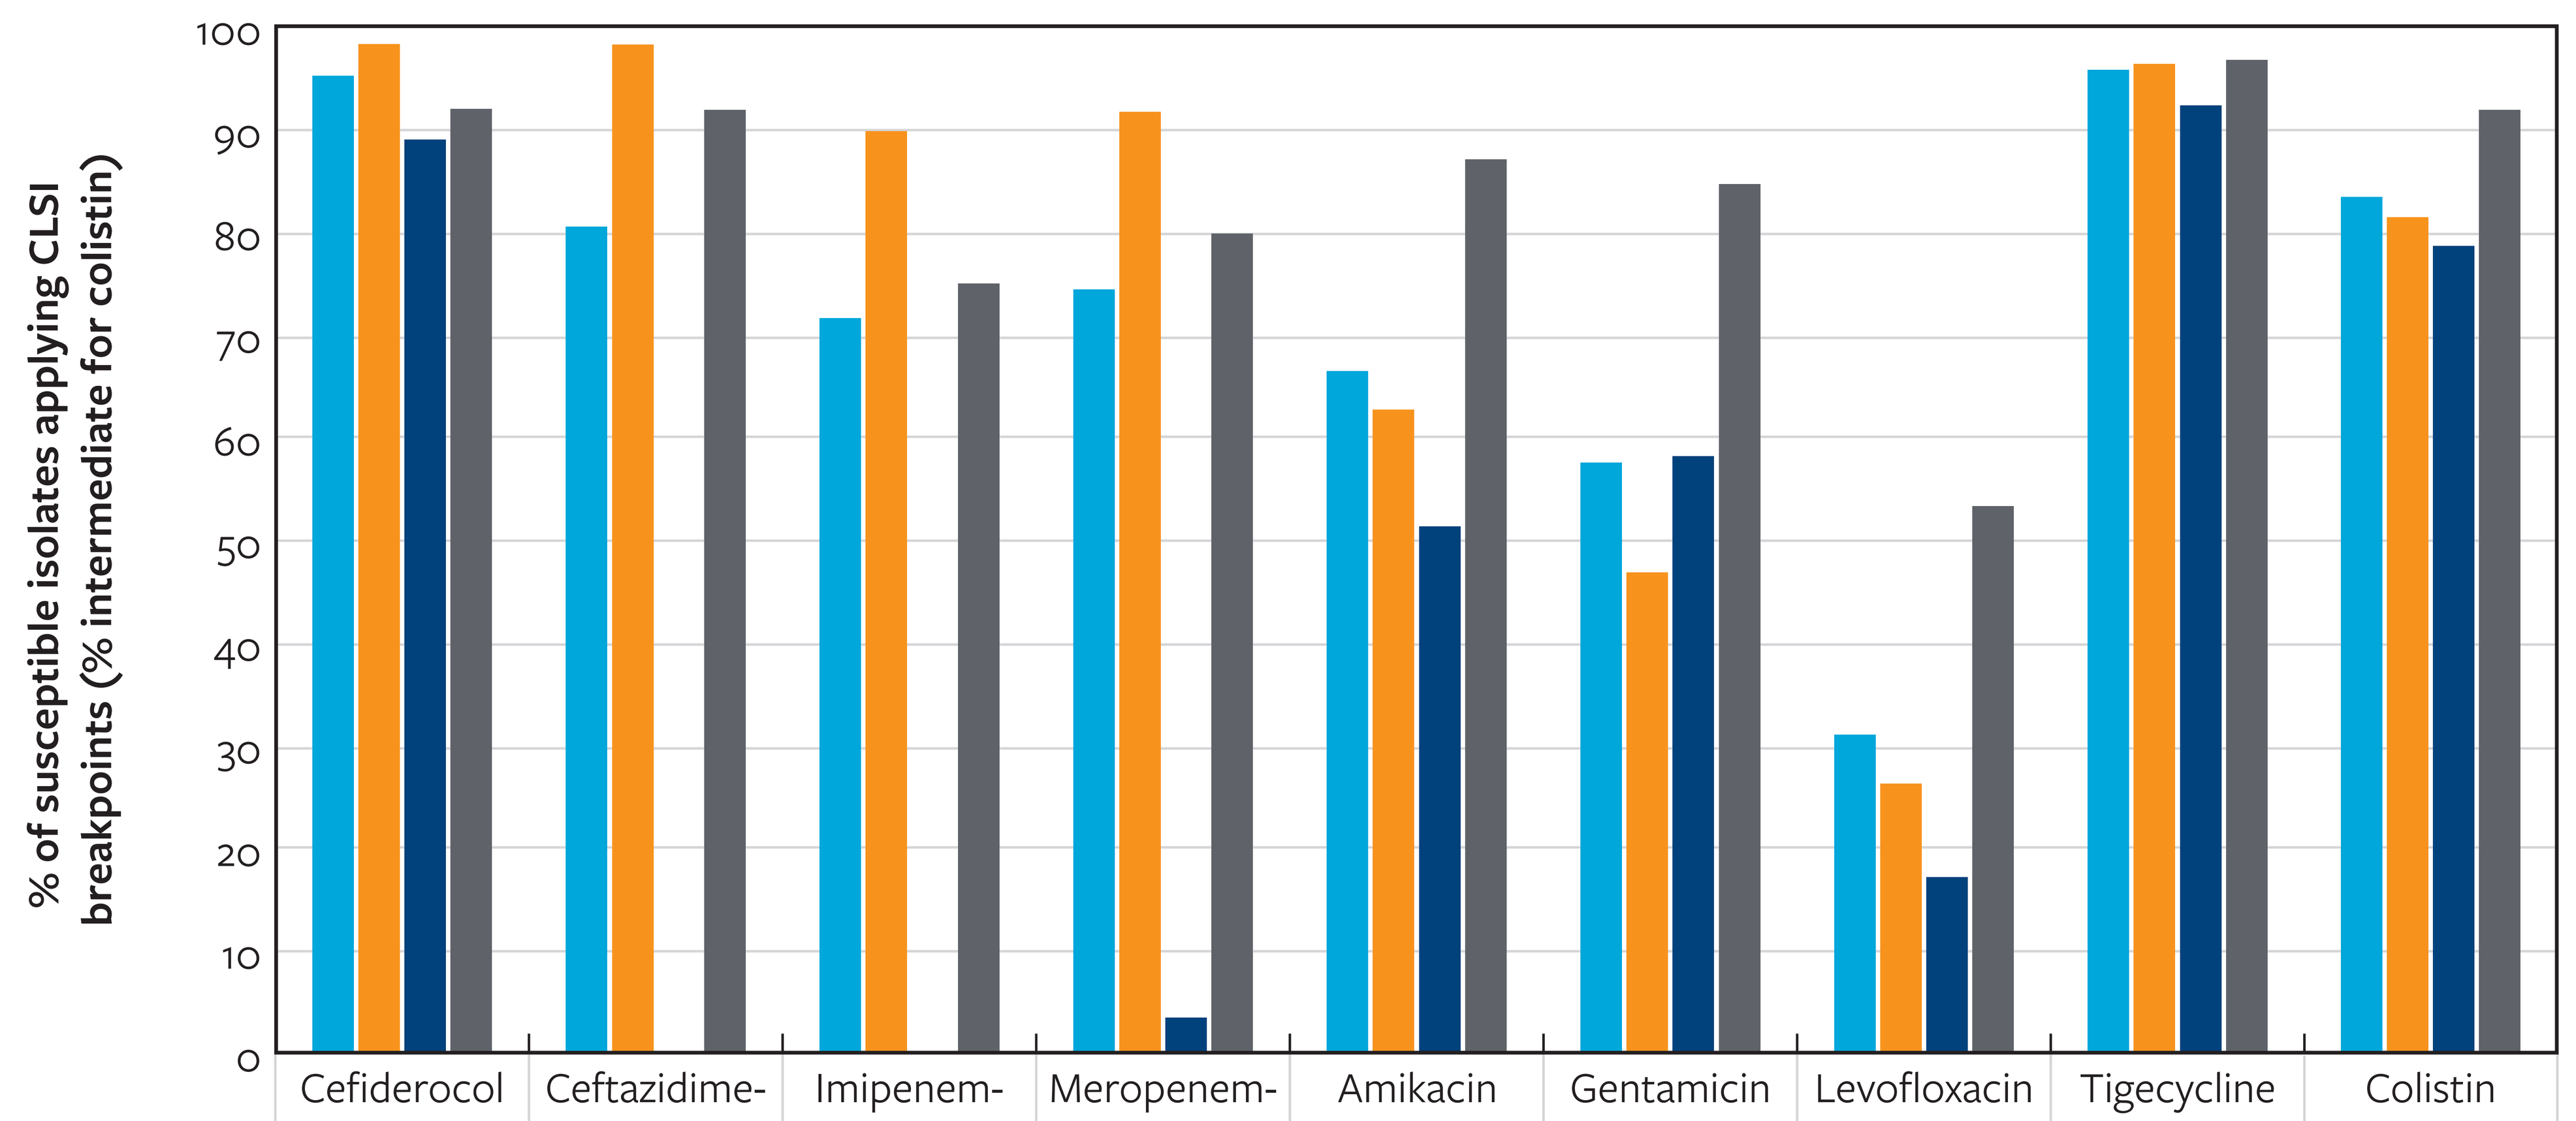
% of susceptible isolates applying CLSI breakpoints (% intermediate for colistin)
100
90
80
70
60
50
40
30
20
10
0

	Cefiderocol	Ceftazidime-avibactam	Imipenem-relebactam	Meropenem-vaborbactam	Amikacin	Gentamicin	Levofloxacin	Tigecycline	Colistin
All CRE (n=176)	96.0	81.2	72.2	75.0	67.0	58.0	31.2	96.6	84.1
Serine-carbapenemase (n=106)	99.1	99.1	90.6	92.5	63.2	47.2	26.4	97.2	82.1
Metallo-β-lactamase (n=29)	89.7	0.0	0.0	3.4	51.7	58.6	17.2	93.1	79.3
Carbapenemase-negative (n=41)	92.7	92.7	75.6	80.5	87.8	85.4	53.7	97.6	92.7

Conclusions

- CRE isolates are observed in all but one US Census Division and MBL-producing Enterobacterales isolates were noted in all but two, highlighting the broad dissemination of these isolates.
- Cefiderocol displayed activity against most CRE isolates, including isolates carrying MBL genes that are resistant to multiple comparator agents and CRE isolates with no carbapenemases.
- Cefiderocol should be considered as an important option for the treatment of infections caused by these resistant organisms.

References

CLSI. 2024. M07Ed12. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. Institute. CaLS, Berwyn, PA.

CLSI. 2025. M100Ed35. Performance Standards for antimicrobial susceptibility testing; 35th informational supplement. CLSI, Berwyn, PA.

Tamma PD, Heil EL, Justo JA, Mathers AJ, Satlin MJ, Bonomo RA. 2024. Infectious Diseases Society of America 2024 Guidance on the Treatment of Antimicrobial-Resistant Gram-Negative Infections. *Clin Infect Dis* doi:10.1093/cid/ciae403.

Kaye KS, Naas T, Pogue JM, Rossolini GM. 2023. Cefiderocol, a Siderophore Cephalosporin, as a Treatment Option for Infections Caused by Carbapenem-Resistant Enterobacterales. *Infect Dis Ther* 12:777–806.

Funding

This research and poster presentation were sponsored by Shionogi & Co., Ltd.

Contact

SCAN ME

Mariana Castanheira, PhD
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
mariana.castanheira@element.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2025_25-SHI-03_P1_CREs.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia