IDWEEK 2025 | Poster #P1279

Five-year trend analysis of carbapenem-resistant Enterobacterales, including isolates carrying metallo-β-lactamase genes in United States, Europe and adjacent regions during 2020–2024

Mariana Castanheira[1], John H. Kimbrough[1], Zachary A. Kockler[1], Rodrigo E. Mendes[1]
[1] Element Iowa City (JMI Laboratories), IA, USA

Introduction

- Among carbapenem-resistant Enterobacterales (CRE) isolates, metallo-β-lactamase (MBL)-producing organisms are still an unmet need with limited therapeutic choices in clinical use in most countries.
- MBL-producing Enterobacterales were not very common until the emergence of NDM-1.
 - In the last few years NDM-producing Enterobacterales have disseminated globally.
- NDM-producing isolates are resistant to many antimicrobial agents since the genes encoding these enzymes are carried in mobile genetic element carrying resistance genes to various antimicrobial classes and/or belong to high-risk clones that are multidrug resistant.
- We evaluated the prevalence of CREs and MBL-carrying isolates in a 5-year surveillance study in European (includes Israel and Turkey) and US hospitals.

Materials and Methods

- A total of 38,614 Enterobacterales were consecutively collected during 2020–2024 in 42 European and 37 US hospitals.
 - Isolates were identified as the cause of infection
 - Isolates were limited to 1 per patient
- Susceptibility testing was performed by the CLSI reference broth microdilution method as described by the Clinical and Laboratory Standards Institute (CLSI) M07 (2024) and M100 (2025) documents.
- Quality control (QC) was performed according to the CLSI M100 (2025) criteria.
 - All QC MIC results were within acceptable ranges
- Categorical interpretations for all comparator agents were those criteria found in the CLSI M100 (2025), or the US FDA website.
- CRE isolates resistant to imipenem (encluding *Proteus mirabilis*, *P. penneri*, and indole-positive Proteeae) or meropenem were submitted to whole genome sequencing and data analysis for the detection of β-lactam resistance mechanisms.
- WGS was performed on a MiSeq or NextSeq (Illumina, San Diego, California, USA) instruments targeting a 30X coverage.
 - Sequences were de novo assembled
 - Analysis of β-lactam resistance mechanisms was performed in silico
- Statistical analysis was performed using the chi-square test, with significance defined as p<0.05.

Results

- Overall, 2.5% (979/38,614) isolates were CREs.
- CREs significantly increased from 2.1% (164/7,662) isolates in 2020 to 2.9% (216/7,405) in 2024 (*p*=0.001) (Figure 1).
- This increase was mostly noted in European hospitals where 3.3% (130/3,974) of the isolates from 2020 were CREs and these percentages were 4.3% (195/4,532) in 2024 and 5.0% (235/4,700) in 2023 (*p*=0.007).
 - The numbers of CRE isolates in the US were considerably lower (34 isolates in 2020 and 21 in 2024).
- Among all CREs, MBLs were observed among 344 isolates—35.1% of the CREs and 0.9% overall isolates.
 - MBL-producing isolates were 261 *K. pneumoniae*, 39 *Enterobacter cloacae* species complex, 17 *Escherichia coli* and other 7 species/species complexes.
- MBL carriers significantly increased overall from 0.4% (29/7,662) in 2020 to 1.5% (109/7,405) in 2024 (p<0.001).
- In Europe, MBL-producing isolates significantly increased from 0.7% (27/3,974) in 2020 to 2.3% (104/4,532) in 2024 (p<0.001).
- The numbers of MBL-carrying isolates in the USA were small (5 isolates in 2024 and 2 isolates in 2020) and yearly MBL rates were mostly <1% with no statistical significance (Table 1).
- The most common MBL genes detected were bla_{NDM-1}, detected alone (221 isolates) or with bla_{NDM-4} (1 isolate) in a total of 222 isolates (Table 1).
- Genes encoding NDM-5 and VIM-1 were observed in 66 and 42 isolates, respectively.

Conclusions

- In this evaluation of 5 years of a surveillance program, we observed a significant global increase in MBL-carriers among Enterobacterales that was mainly driven by an increase in MBL-carrying isolates from European countries.
- MBL-producing Enterobacterales are difficult-to-treat organisms and refractory to most of the antimicrobial armamentarium, thus monitoring MBL-carrying Enterobacterales isolates is an important endeavor to understand trends and potential spread of these isolates globally.

Figure 1. Percentage of CRE and MBL-producing Enterobacterales isolates by year overall and by continent

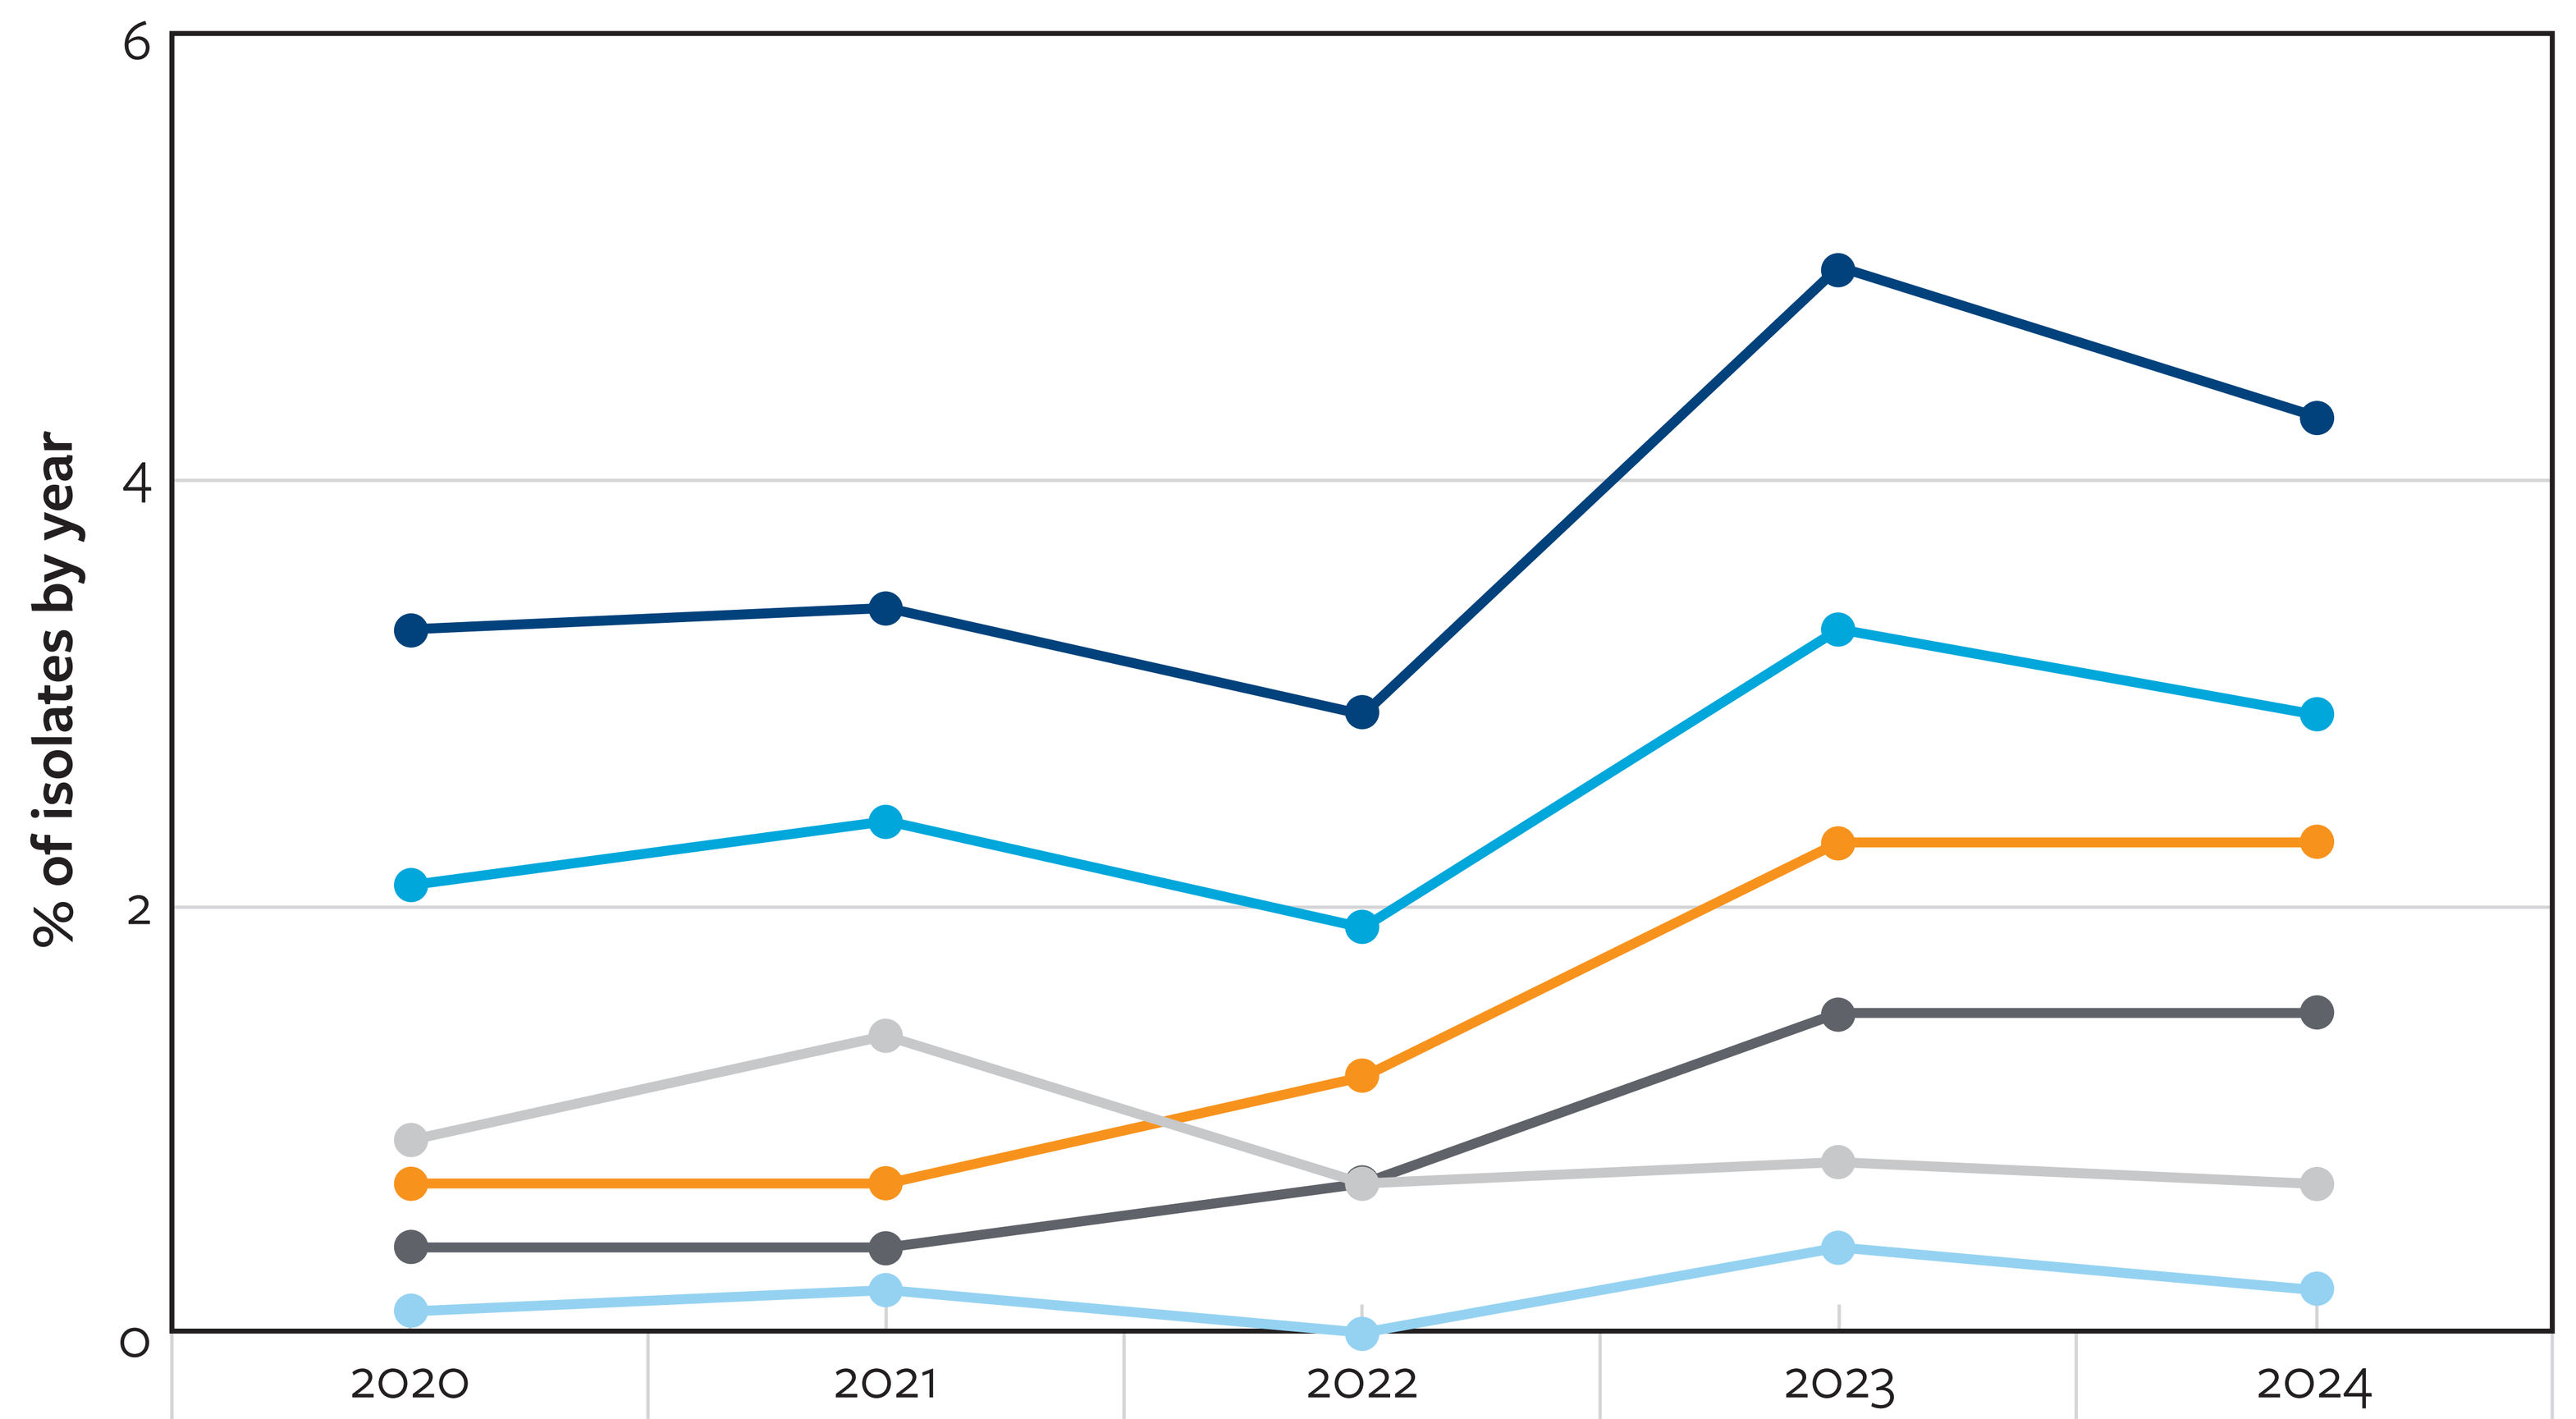
% of isolates by year
6
4
2
0

	2020	2021	2022	2023	2024
CRE-All*	2.1	2.4	1.9	3.3	2.9
CRE-USA	0.9	1.4	0.7	0.8	0.7
CRE-Europe*	3.3	3.4	2.9	5.0	4.3
MBL-All*	0.4	0.4	0.7	1.5	1.5
MBL-USA	0.1	0.2	0.0	0.4	0.2
MBL-Europe*	0.7	0.7	1.2	2.3	2.3

*Statistically significant trend (*p*<0.05)

Table 1. MBL-producing Enterobacterales isolates by enzyme and continent

MBL	No. of isolates overall	No. of isolates in European countries						No. of isolates in the USA					
		All years	2020	2021	2022	2023	2024	All years	2020	2021	2022	2023	2024
NDM-1	221	210	19	18	39	77	57	11	1	6		4	
NDM-1, NDM-4	1							1				1	
NDM-19	1	1			1								
NDM-5	66	57		2	2	17	36	9	1		1	4	3
NDM-6	1	1			1								
NDM-7	3	3			2		1						
NDM-7-like (M22I)[a]	2							2				2	
NDM-71	1	1					1						
NDM-9	2	2			2								
IMP-4	1							1		1			
VIM-1	42	40	8	7	6	12	7	2					2
VIM-4	3	3			1		2						
Total	344	318	27	27	54	106	104	26	2	7	1	11	5

Organisms include *Citrobacter freundii* species complex (1), *Enterobacter cloacae* species complex (39), *Escherichia coli* (17), *Klebsiella aerogenes* (3), *K. oxytoca* (6), *K. pneumoniae* (261), *Proteus mirabilis* (6), *Providencia rettgeri* (2), *P. stuartii* (4), and *Serratia marcescens* (5)
[a] This isolate has a bla_{NDM-7} with a mutation that generated a change in amino acid position 221 (M→I)

References

CLSI. 2024. M07Ed12. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. Institute. CaLS, Berwyn, PA.

CLSI. 2025. M100Ed35. Performance Standards for antimicrobial susceptability testing; 35th informational supplement. CLSI, Berwyn, PA.

Tamma PD, Aitken SL, Bonomo RA, Mathers AJ, van Duin D, Clancy CJ. 2022. Infectious Diseases Society of America 2022 Guidance on the Treatment of Extended-Spectrum beta-lactamase Producing Enterobacterales (ESBL-E), Carbapenem-Resistant Enterobacterales (CRE), and *Pseudomonas aeruginosa* with Difficult-to-Treat Resistance (DTR-*P. aeruginosa*). *Clin Infect Dis* 75:187–212.

Hidalgo-Tenorio C, Bou G, Oliver A, Rodriguez-Aguirregabiria M, Salavert M, Martinez-Martinez L. 2024. The Challenge of Treating Infections Caused by Metallo-beta-Lactamase-Producing Gram-Negative Bacteria: A Narrative Review. *Drugs* 84:1519–1539.

Funding

This research and poster presentation were sponsored by Shionogi & Co., Ltd.

Contact

element

SCAN ME

Mariana Castanheira, PhD
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
mariana.castanheira@element.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2025_25-SHI-03_P4_MLBs.pdf

Charges may apply. No personal information is stored.

IDWeek 2025, October 19–22, 2025, Atlanta, Georgia