IDWeek 2026 | Poster #P-20

Activity of rezafungin against *Aspergillus* spp. that are non-wild-type to azole antifungal agents

Marisa L Winkler, Samuel Edeker, Abby L Klauer, Paul Rhomberg, Maura Karr, John H Kimbrough, Mariana Castanheira

Element Materials Technology (JMI Laboratories), North Liberty, Iowa, USA

Introduction

- *Aspergillus* spp. are molds that cause allergic reactions, invasive aspergillosis, and chronic pulmonary aspergillosis (CPA).
- Resistance rates of *Aspergillus* spp. to azoles are rising.
- Rezafungin (RZF) is a long-acting echinocandin with pharmacokinetic and pharmacodynamic properties allowing weekly dosing.
- Rezafungin is in Phase 2 clinical trials for the treatment of CPA.
- Rezafungin was non-inferior to standard antimicrobial regimen in the prophylaxis of invasive fungal diseases in allogeneic hematopoietic stem cell transplant patients in a Phase 3 clinical trial (ReSPECT).
- Our study investigated the *in vitro* activity of RZF against *Aspergillus* spp. isolates that are non-wild-type (NWT) to azole comparators.

Methods

- 1,115 *Aspergillus* spp. isolates were collected from 2014 to 2025 in 25 countries (Figure 1).
 - This included 65 medical centers.
- Only 1 isolate per patient episode was included.
- All isolates were identified by MALDI-TOF MS and/or DNA sequencing.
- Isolates were tested by CLSI reference broth microdilution method (M38) for rezafungin, anidulafungin, caspofungin, micafungin, itraconazole, voriconazole, and amphotericin B.
- Minimum inhibitory concentrations (MICs) for azoles and amphotericin B were interpreted by Clinical and Laboratory Standards Institute epidemiological cutoff values (ECVs); minimum effective concentrations (MECs) for echinocandins do not have CLSI interpretations defined.
- Non-wild-type (NWT) ECV criteria were defined as an MIC of >1 mg/L for voriconazole (VRC) and itraconazole (ITR) against *A. fumigatus*; >2 mg/L for voriconazole against *A. niger* species complex, and >4 mg/L for itraconazole against *A. niger* species complex (CLSI M57S 4th edition).
- All isolates that were NWT to ≥1 azole had whole genome sequencing performed with analysis of CYP51A and CYP51B genes.

Results

- Isolate infection sources were respiratory tract (810/1,115, 72.6%), skin and soft tissue infection (108/1,115, 9.7%), head and neck infections (75/1,115, 6.7%), bloodstream infections (12/1,115, 1.1%), and other or not defined (110/1,115, 9.9%).
- There were 4.9% of isolates that were NWT to ITR or VRC (55/1,115) (Table 1).
 - These NWT isolates were from 13 countries (Figure 2).
 - NWT isolates were either *A. fumigatus* (n=45) or *A. niger* species complex (n=10). All *A. flavus* species complex and *A. terreus* species complex isolates were WT for azoles.
- The $MEC_{50/90}$ and MEC range were comparable for rezafungin and anidulafungin against all organism groups in Table 1.
 - The $MEC_{50/90}$ is 6–7 doubling dilutions lower for rezafungin/anidulafungin than the $MIC_{50/90}$ for itraconazole and 5 doubling dilutions lower than for voriconazole among all collected *Aspergillus* spp. isolates.
 - For *A. fumigatus* isolates NWT to azoles, the rezafungin $MEC_{50/90}$ was 7 doubling dilutions and >8 doubling dilutions lower than the $MIC_{50/90}$ for itraconazole and 6 and >8 doubling dilutions lower than the $MIC_{50/90}$ for voriconazole.
 - For *A. niger* isolates NWT to azoles, the rezafungin $MEC_{50/90}$ was >10 doubling dilutions lower than the $MIC_{50/90}$ for itraconazole and 9 doubling dilutions lower than the $MIC_{50/90}$ for voriconazole.
- Among the isolates NWT for ≥1 azole, CYP51 alterations were identified in 23 *A. fumigatus* (23/45, 51.1%) and 4 *A. niger* species complex isolates (4/10, 40%) (Table 2).
 - Isolates with CYP51 alterations were from USA (8/27, 29.6%), Europe (11/27, 40.7%) and the Asia-Pacific region (8/27, 29.6%).
 - 8 of the isolates from Europe had promoter mutations, 2 of the 8 isolates from the Asia-Pacific region had promoter mutations, and none of the USA isolates had promoter mutations.
- Among the isolates with identified CYP51 alterations, rezafungin MECs ranged from 0.002 to 0.12 mg/L with $MEC_{50/90}$ 0.015/0.03 mg/L while voriconazole MICs ranged from 0.5 to >8 mg/L with $MIC_{50/90}$ 2/>8 mg/L.

Table 1. Antimicrobial activity of rezafungin and select comparator agents in mg/L tested against *Aspergillus* spp.

Organism (no. isolates)	Rezafungin		Anidulafungin		Itraconazole		Voriconazole		Amphotericin B	
	$MEC_{50/90}$	MEC Range	$MEC_{50/90}$	MEC Range	$MIC_{50/90}$ (%WT) [c]	MIC Range	$MIC_{50/90}$ (%WT) [c]	MIC Range	$MIC_{50/90}$ (%WT) [c]	MIC Range
Aspergillus spp. (1,115) [a]	0.015/0.03	≤0.002 to 0.12	0.015/0.03	≤0.002 to 0.12	1/1	0.06 to >8	0.5/1	0.06 to >8	1/2	≤0.03 to >2
Aspergillus spp. NWT azoles (55) [b]	0.015/0.03	≤0.002 to 0.12	0.015/0.06	0.004 to 0.12	4/>8	0.5 to >8	2/>8	0.5 to >8	1/2	0.12 to 2
All *A. fumigatus* (874)	0.015/0.03	≤0.002 to 0.12	0.015/0.03	≤0.002 to 0.12	1/1 (95.1)	0.12 to >8	0.5/0.5 (97.9)	0.12 to >8	1/2 (99.8)	0.06 to 4
Aspergillus fumigatus NWT azoles (45)	0.015/0.03	≤0.002 to 0.12	0.015/0.06	0.004 to 0.12	2/>8 (4.4)	0.5 to >8	1/>8 (60.0)	0.5 to >8	1/2 (100.0)	0.25 to 2
All *A. niger* species complex (49)	0.004/0.008	≤0.002 to 0.03	0.004/0.008	≤0.002 to 0.06	2/>8 (79.6)	0.12 to >8	1/2 (91.8)	0.12 to >8	0.12/0.25 (100.0)	≤0.03 to 0.5
A. niger species complex NWT azoles (10)	0.004/0.008	≤0.002 to 0.008	0.004/0.004	0.004	>8/>8 (0.0)	8 to >8	2/4 (60.0)	2 to >8	0.25/0.25 (100.0)	0.12 to 0.25

MEC, minimum effective concentration; MIC, minimum inhibitory concentration; WT, wild-type; NWT, non-wild-type
[a] Organisms include *A. flavus* species complex (164), *A. fumigatus* (874), *A. hiratsukae* (1), *A. lentulus* (1), *A. nidulans* species complex (5), *A. niger* species complex (49), *A. nomius* (2), *A. parasiticus* (2), *A. tamarii* (1), *A. terreus* species complex (13), *A. thermomutatus* (1), *A. unguis* (1), and unspeciated *Aspergillus* (1).
[b] Includes *A. fumigatus* (45) and *A. niger* species complex (10).
[c] ECV criteria published in CLSI M57S (2022), only applied to single species or species complex.

Conclusions

- Rezafungin has potent activity against *Aspergillus* spp. isolates over a 12-year period from diverse geographic areas.
- Rezafungin maintained low MECs against *Aspergillus* spp. isolates that were NWT to ≥1 azole antifungal agent.
 - Low MECs were also maintained against isolates with identified azole resistance mechanisms in CYP51 genes.
- Continued clinical development of rezafungin for the prevention and treatment of invasive aspergillosis infections is warranted based on this *in vitro* data.

Table 2. Summary of *Aspergillus* spp. isolates with CYP51A/B alterations, voriconazole MICs, and rezafungin MECs

Organism	Country	CYP51A	CYP51B	Voriconazole MIC (mg/L)	Rezafungin MEC (mg/L)
Aspergillus fumigatus	USA	I242V	wild-type	1	0.015
Aspergillus fumigatus	Belgium	TR34/L98H	wild-type	2	0.03
Aspergillus fumigatus	USA	I242V	wild-type	0.5	0.03
Aspergillus fumigatus	UK	TR34/L98H	wild-type	2	0.004
Aspergillus fumigatus	USA	G448S	wild-type	4	0.004
Aspergillus fumigatus	Belgium	Y121F, M172I, T289A, G448S, TR46	wild-type	≥8	0.004
Aspergillus fumigatus	USA	F46Y, M172V, N248T, D255E, E427K	wild-type	0.5	0.03
Aspergillus fumigatus	New Zealand	G138C	wild-type	8	0.004
Aspergillus fumigatus	UK	TR34/L98H	wild-type	2	0.015
Aspergillus fumigatus	USA	I242V	wild-type	0.5	0.03
Aspergillus fumigatus	France	H147Y	wild-type	≥8	0.015
Aspergillus fumigatus	USA	I242V	wild-type	0.5	0.015
Aspergillus fumigatus	USA	I242V	wild-type	1	0.015
Aspergillus fumigatus	Belgium	TR34/L98H	wild-type	4	0.03
Aspergillus fumigatus	Belgium	TR34/L98H	wild-type	8	0.12
Aspergillus fumigatus	France	TR34/L98H	wild-type	4	0.015
Aspergillus fumigatus	USA	G448S	wild-type	≥8	0.015
Aspergillus fumigatus	New Zealand	TR46, Y121F, T289A	wild-type	≥8	0.015
Aspergillus fumigatus	New Zealand	F46Y, M172V, E427K, G448S	wild-type	4	0.008
Aspergillus fumigatus	New Zealand	TR46, Y121F, T289A	wild-type	≥8	0.004
Aspergillus fumigatus	Czech Republic	G448S	wild-type	4	0.002
Aspergillus fumigatus	Czech Republic	TR46, Y121F, T289A, S363P, I364V, G448S	wild-type	≥8	0.008
Aspergillus fumigatus	Australia	wild-type	P383L	1	0.015
Aspergillus niger species complex	Thailand	A140V, T321A, P413S, L492M, I503F, Q504P	wild-type	2	0.002
Aspergillus niger species complex	Thailand	L383V	wild-type	≥8	0.002
Aspergillus niger species complex	Italy	A9V, L21F, T321A, I503V	wild-type	2	0.004
Aspergillus niger species complex	Australia	A9V, L21F, G52E, T321A, I503V	wild-type	2	0.002

MIC, minimum inhibitory concentration; MEC, minimum effective concentration

Figure 1. Map of geography of all collected *Aspergillus* spp. isolates

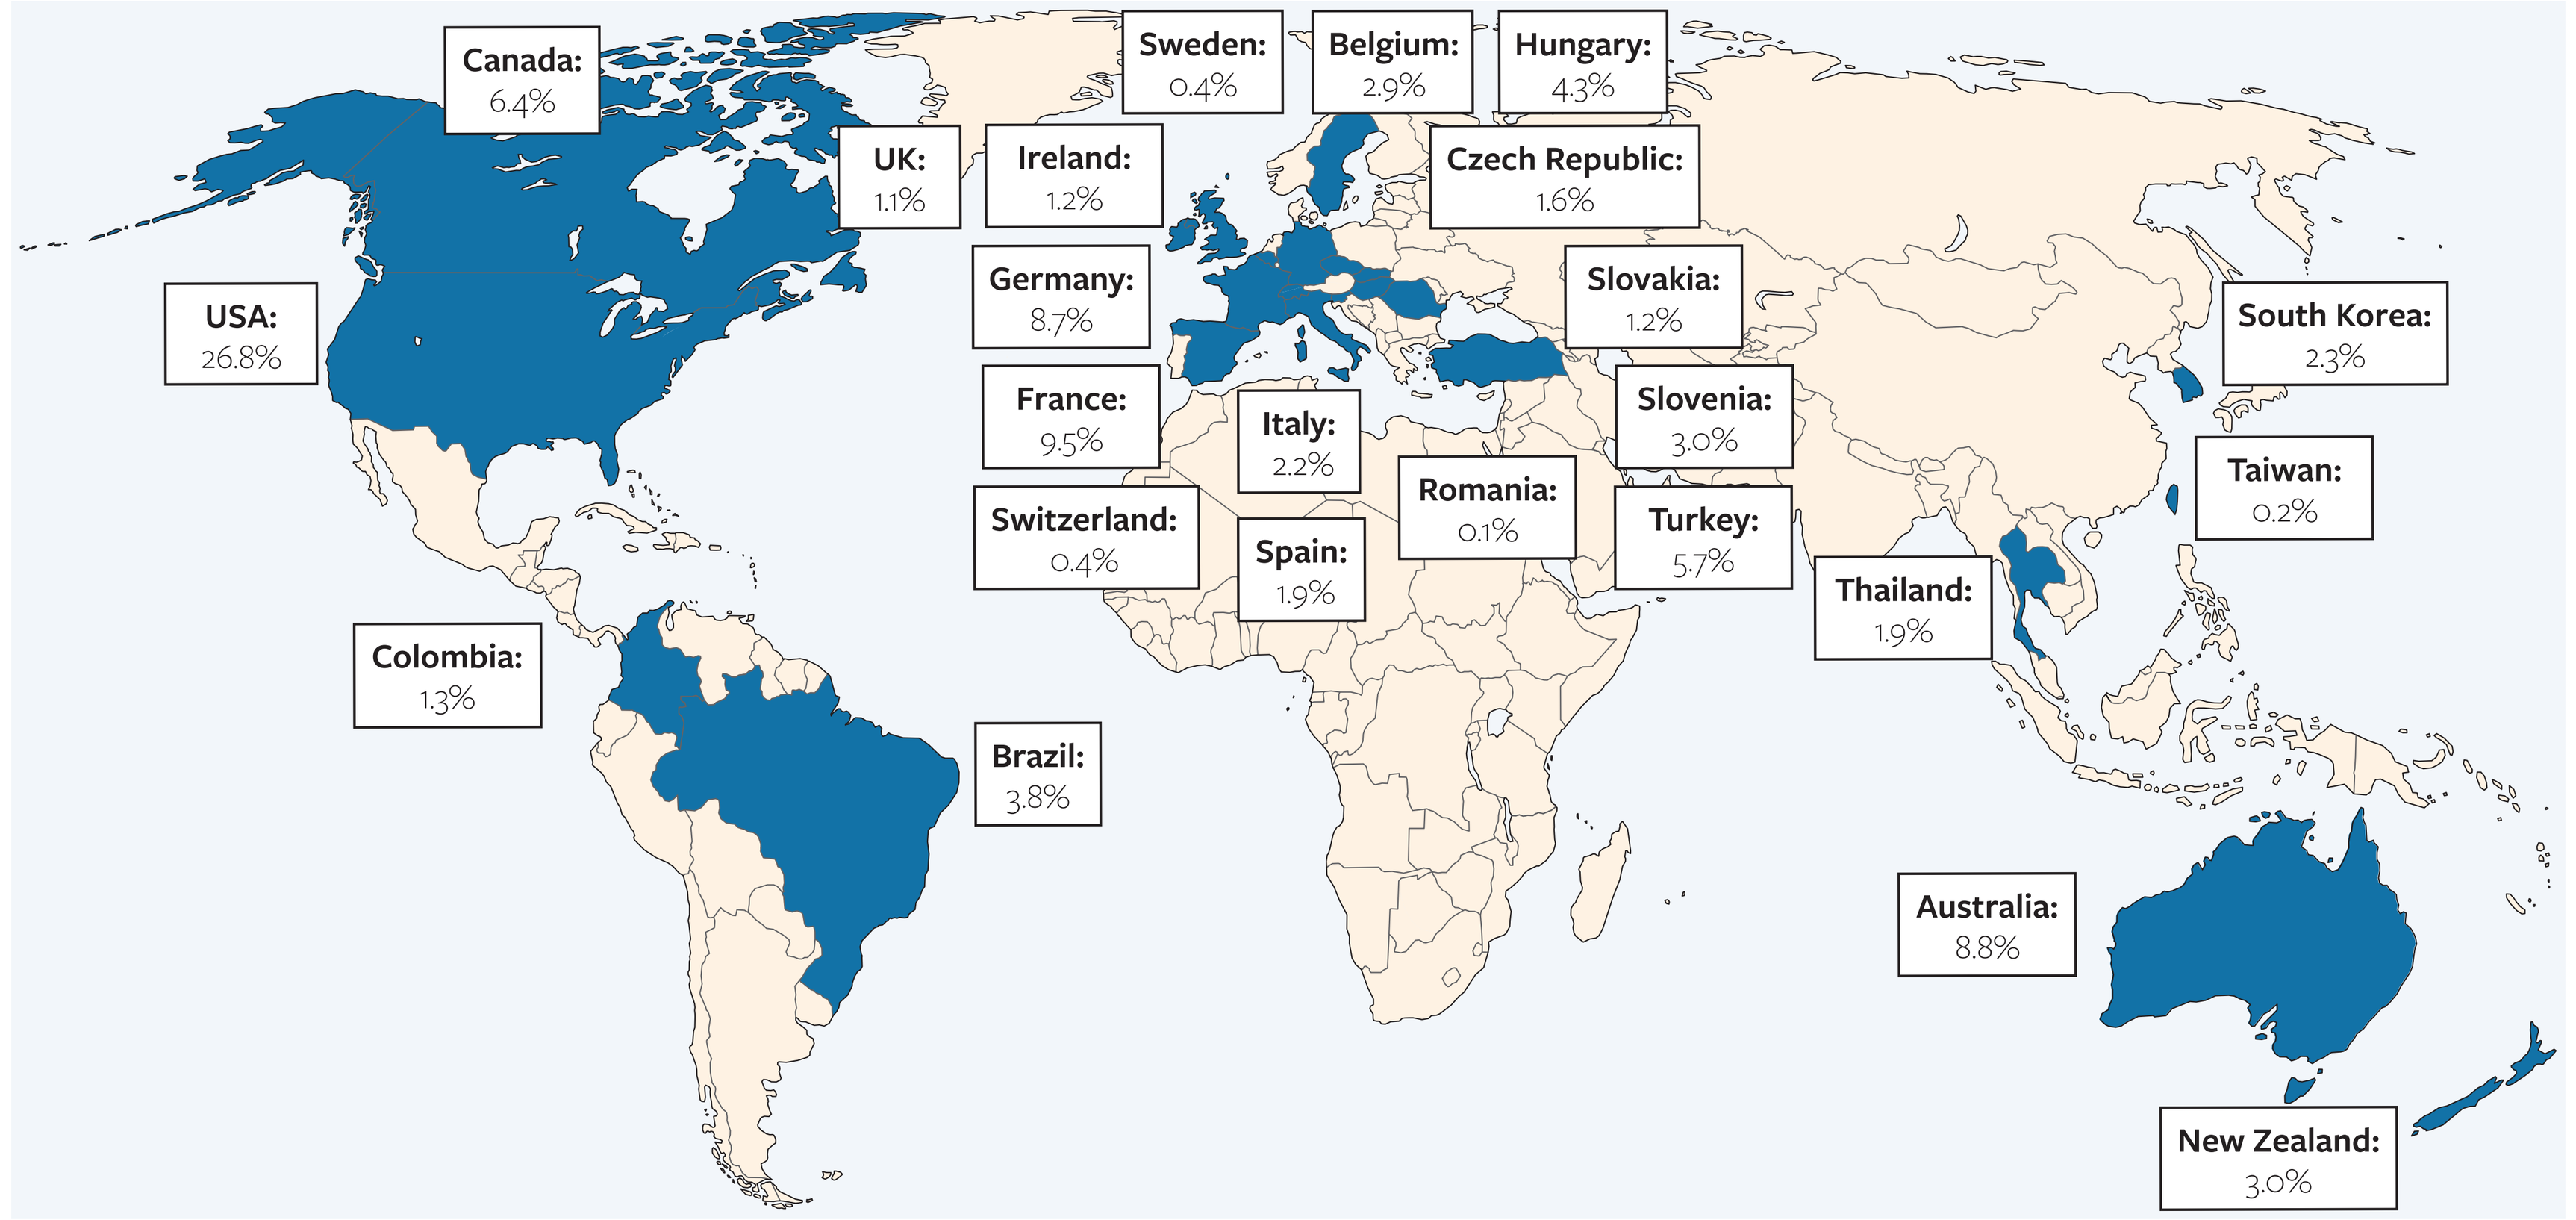
Canada: 6.4%
Sweden: 0.4%
Belgium: 2.9%
Hungary: 4.3%
UK: 1.1%
Ireland: 1.2%
Czech Republic: 1.6%
USA: 26.8%
Germany: 8.7%
Slovakia: 1.2%
South Korea: 2.3%
France: 9.5%
Italy: 2.2%
Slovenia: 3.0%
Taiwan: 0.2%
Romania: 0.1%
Switzerland: 0.4%
Spain: 1.9%
Turkey: 5.7%
Thailand: 1.9%
Colombia: 1.3%
Brazil: 3.8%
Australia: 8.8%
New Zealand: 3.0%

Figure 2. Map of geography of NWT *Aspergillus* spp. isolates

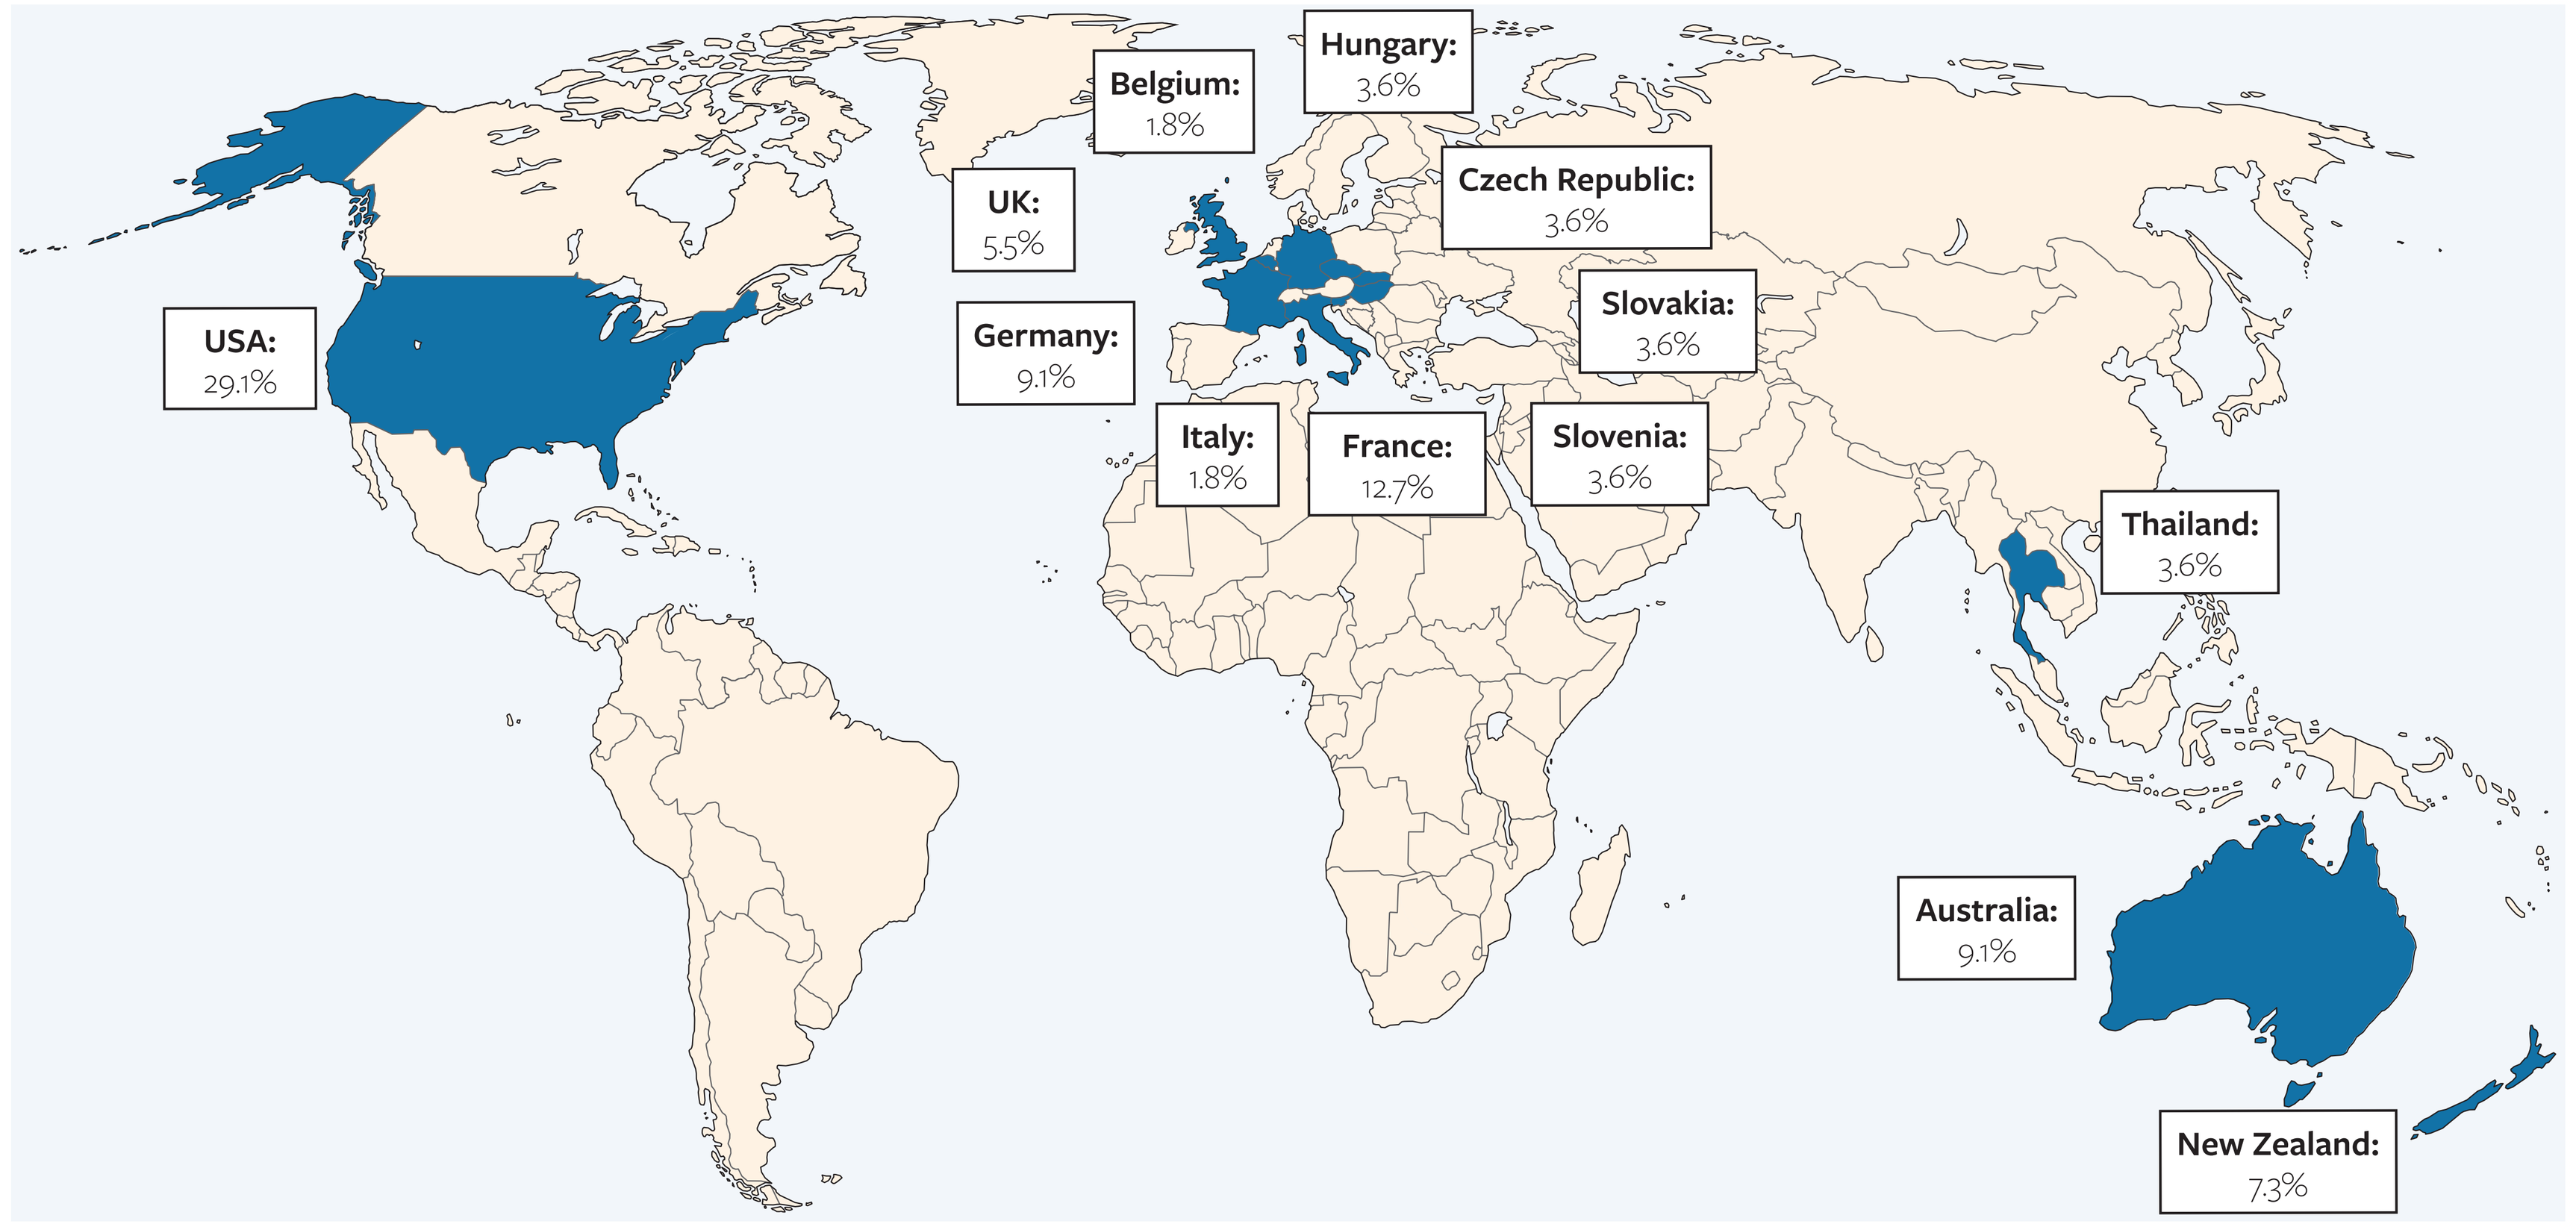
Hungary: 3.6%
Belgium: 1.8%
Czech Republic: 3.6%
UK: 5.5%
USA: 29.1%
Germany: 9.1%
Slovakia: 3.6%
Italy: 1.8%
France: 12.7%
Slovenia: 3.6%
Thailand: 3.6%
Australia: 9.1%
New Zealand: 7.3%

Funding

ML Winkler, S Edeker, AL Klauer, P Rhomberg, M Karr, JH Kimbrough, and M Castanheira were employees of Element Materials Technology (JMI Laboratories) at the time of this study. JMI Laboratories was a paid consultant to Melinta Therapeutics, LLC, a wholly owned subsidiary of CorMedix, in connection with the development of this poster.

Acknowledgments

The authors thank all of the SENTRY Program participants for providing the isolates used in this study.

References

CLSI, *M38 M51S Ed4. Performance standards for antifungal susceptibility testing of yeasts*. 2026, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M57S Ed4. Epidemiological cutoff values for antifungal susceptibility testing*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M38 Ed4. Reference method for broth dilution antifungal susceptibility testing of filamentous fungi*. 2017, Clinical and Laboratory Standards Institute: Wayne, PA.

Contact

element

Scan Me

Marisa Winkler, MD, PhD
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: marisa.winkler@element.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2026_25-MEL-08_P2_Reza_Aspergillus_Azoles.pdf

Charges may apply. No personal information is stored.

IDWeek 2026, October 21–24, 2026, Washington, D.C.