In Vitro Activity of Omadacycline against 7,200 Bacterial Clinical Isolates Representing Multiple Infection Types from the United States (2024–2025)

Michael D. Huband[1], Marisa L. Winkler[1], Kelley A. Fedler[1], Krisztina M. Papp-Wallace[1], and Mariana Castanheira[1]

[1] Element Iowa City (JMI Laboratories), North Liberty, Iowa, United States

Introduction

1 Omadacycline *Aminomethylcycline (Tetracycline Class)* | FDA APPROVED 2018 | AB ABSSSI adult skin/skin structure infection | CP CABP adult community-acquired pneumonia

2 AMR Potent broad-spectrum in vitro activity against isolates expressing common resistance mechanisms: TET, PEN, FQ, MAC, β-LACTAM

3 Potent in vitro activity against key pathogens associated with ABSSSI or CABP
- GRAM-POSITIVE: Staphylococci, Streptococci, Enterococci
- GRAM-NEGATIVE: *Enterobacter cloacae*, *Klebsiella pneumoniae*, *Escherichia coli*, *Haemophilus influenzae*

4 Recently completed clinical trials
- DFI/OM: Diabetic foot infection & acute osteomyelitis, NCT04714411
- NTM: NTM infections, NCT04922554
- CABP: CABP, NCT04779242
- PK: IV/oral PK in children/adolescents, NCT05217537

5 THIS STUDY | 7,200 bacterial isolates | 2024–2025 surveillance programs SENTRY + Paratek KEYSTONE

Determined the in vitro antibacterial activity and susceptibility of omadacycline and tetracycline-class comparator agents doxycycline, minocycline, tetracycline, tigecycline against bacterial isolates collected during the surveillance programs.

Materials and Methods

- 7,200 bacterial isolates were collected in 2024–2025 from United States medical centers (9 Census Divisions), including 2,400 staphylococci, 2,200 *Enterobacterales*, 1,000 streptococci, 600 non-fermenters, 400 enterococci, 400 *Haemophilus* spp., and 200 *Moraxella catarrhalis* isolates.
- Bacterial isolate identifications were confirmed at Element Iowa City using standard microbiology methods and matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany).
- 1 isolate per patient infection episode was collected from patients with skin and skin structure infections ABSSSI (1,023 isolates; 14.2%), bloodstream infections (2,294 isolates; 31.9%), CABP (864 isolates; 12.0%), intra-abdominal infection (27 isolates; 0.4%), pneumonia in hospitalized patients (PIHP) (1,730 isolates; 24.0%), urinary tract infections (1,085 isolates; 15.1%), and other infections (177 isolates; 2.4%).
- Broth microdilution susceptibility testing was performed according to Clinical and Laboratory Standards Institute M07 (CLSI; 2024) and M100 (2026) guidelines.
- Omadacycline and tigecycline MIC results were interpreted using United States Food and Drug Administration (FDA) breakpoint criteria. Doxycycline, minocycline, and tetracycline interpretive criteria are defined by the CLSI M100 (2026) document.

Table 1. Antimicrobial activity of omadacycline and tetracycline class comparator agents against 3,800 Gram-positive bacterial isolates collected from medical centers in the United States (2024–2025)

Organism (no. tested)	Omadacycline			Doxycycline			Minocycline			Tetracycline			Tigecycline		
	$MIC_{50/90}$		% S (FDA)	$MIC_{50/90}$		% S (CLSI)	$MIC_{50/90}$		% S (CLSI)	$MIC_{50/90}$		% S (CLSI)	$MIC_{50/90}$		% S (FDA)
Staphylococcus aureus (2,000)	0.12	**0.12**	98.6[a] 96.6[b,c]	≤0.06	0.5	97.1	0.06	0.12	97.6	≤0.12	8	89.7	0.06	0.12	>99.9
MRSA (760)	0.12	**0.25**	96.4[a] 92.5[b,c]	≤0.06	4	93.7	0.06	1	95.4	≤0.12	>8	79.3	0.06	0.12	99.9
MSSA (1,240)	0.12	**0.12**	99.8[a] 99.2[b,c]	≤0.06	0.12	99.2	0.06	0.12	99.0	≤0.012	0.25	96.0	0.06	0.12	100.0
Tetracycline-R (178)	0.12	**0.5**	90.4[a]	4	8	67.4	1	>8	73.6	>8	>8	0.0	0.12	0.25	99.4
Staphylococcus capitis (11)	0.06	**0.12**	—[d]	≤0.06	1	100.0	0.06	0.06	100.0	0.25	>8	81.8	0.12	0.12	—
Staphylococcus epidermidis (187)	0.25	**0.5**	—[d]	0.25	4	93.6	0.12	0.25	97.9	0.5	>8	79.1	0.12	0.25	—
Staphylococcus haemolyticus (14)	0.12	**0.5**	—[d]	0.25	8	85.7	0.06	0.25	100.0	0.5	>8	50.0	0.12	0.25	—
Staphylococcus hominis (25)	0.06	**0.5**	—[d]	0.12	1	96.0	0.06	0.25	100.0	0.25	>8	88.0	0.06	0.12	—
Staphylococcus lugdunensis (81)	0.03	**0.06**	100.0[a]	≤0.06	≤0.06	100.0	≤0.03	0.06	100.0	≤0.12	≤0.12	97.5	0.03	0.06	—
Staphylococcus saprophyticus (68)	0.12	**0.25**		0.12	0.25	100.0	0.12	0.12	100.0	0.25	0.5	94.1	0.12	0.12	—
Streptococcus anginosus group (50)	0.03	**0.12**	96.0[a]	0.12	>1	—	0.12	>1	—	0.5	>4	64.0	0.03	0.06	100.0
Streptococcus pneumoniae (600)[d,e]	0.03	**0.06**	99.8[b]	0.06	>1	82.2	0.06	>1	—	0.25	>4	82.2	0.03	0.06	98.7
Penicillin-R (63)	0.06	**0.06**	100.0[b]	0.25	>1	55.6	0.12	>1	—	0.25	>4	55.6	0.03	0.06	98.4
Macrolide-R (230)	0.06	**0.06**	99.6[b]	0.12	>1	60.4	0.12	>1	—	0.25	>4	60.4	0.03	0.06	98.3
Tetracycline-R (107)	0.06	**0.06**	99.1[b]	>1	>1	0.0	>1	>1	—	>4	>4	0.0	0.03	0.06	98.1
S. pyogenes (199)	0.06	**0.12**	100.0[a]	0.12	>1	—	0.12	>1	—	0.25	>4	67.8	0.06	0.06	100.0
Macrolide-R (43)	0.12	**0.12**	100.0[a]	>1	>1	—	>1	>1	—	>4	>4	14.0	0.06	0.06	100.0
Tetracycline-R (63)	0.12	**0.12**	100.0[a]	>1	>1	—	>1	>1	—	>4	>4	0.0	0.06	0.06	100.0
Viridans group streptococci (201)	0.06	**0.12**	—	0.25	>1	—	0.12	>1	—	0.5	>4	62.2	0.03	0.06	100.0
Enterococcus faecalis (200)	0.06	**0.12**	100.0[a]	4	8	72.0	8	>8	37.5	>16	>16	25.5	0.06	0.12	99.0[f]
Tetracycline-R (146)	0.06	**0.25**	100.0[a]	4	8	62.3	8	>8	15.1	>16	>16	0.0	0.06	0.12	98.6[f]
Vancomycin-R (4)	0.12	—	100.0[a]	8	—	25.0	>8		0.0	>16		0.0	0.06		100.0[f]
E. faecium (200)	0.06	**0.12**	—[d]	4	16	56.0	8	>8	48.2	>16	>16	19.1	0.06	0.12	—
Tetracycline-R (155)	0.12	**0.12**	—[d]	8	16	43.2	8	>8	33.5	>16	>16	0.0	0.06	0.12	—
Vancomycin-R (120)	0.06	**0.12**	—[d]	4	16	56.7	8	>8	47.5	>16	>16	10.0	0.06	0.12	—

CLSI, Clinical and Laboratory Standards Institute; FDA, United States Food and Drug Administration; S, susceptible; MRSA, methicillin-resistant *S. aureus*; MSSA, methicillin-susceptible *S. aureus*; R, resistant.
[a] ABSSSI breakpoint applied.
[b] CABP breakpoint applied.
[c] US FDA breakpoints published for MSSA (methicillin-susceptible *S. aureus*) isolates were applied to all *S. aureus* isolates.
[d] Breakpoint interpretive criteria are unavailable.
[e] Oral dosing (penicillin MIC, ≥2 mg/L).
[f] FDA breakpoints for vancomycin-susceptible isolates were applied to all isolates.

Bold, omadacycline MIC_{90} values.
Green, susceptible according to CLSI or FDA breakpoint interpretive criteria.
Yellow, intermediate according to CLSI or FDA breakpoint interpretive criteria.
Orange, resistant according to CLSI or FDA breakpoint interpretive criteria.

Conclusions

Since first approved in 2018, omadacycline continues to demonstrate potent *in vitro* activity against medically important bacterial pathogens with no evidence of increasing MIC values over time including those expressing resistance to macrolides, oxacillin/penicillin, vancomycin, tetracycline, and β-lactams.

Omadacycline demonstrated potent *in vitro* activity against other non-indicated Gram-positive and Gram-negative bacterial pathogens including *A. baumannii*, *S. maltophilia*, staphylococci, viridans group streptococci, and *E. faecium*.

The results of this surveillance study support the continued use of omadacycline, especially in ABSSSI and CABP infections where resistant pathogens are likely to be encountered, and warrant further investigation in disease states where omadacycline is not an FDA-approved treatment.

Scan Me

Scan for a copy of this poster
https://www.jmilabs.com/data/posters/IDWeek2026_25-PAR-03_P1_Omadacycline_Surveillance.pdf

PARATEK

Results

Omadacycline demonstrated broad and potent in vitro activity against a wide range of medically important Gram-positive and Gram-negative bacterial pathogens collected from U.S. medical centers in 2024–2025 (Table 1 and Table 2).

GRAM-POSITIVE PATHOGENS (Table 1)

- *Staphylococcus aureus*, 2,000 isolates — MRSA (n=760), MSSA (n=1,240), Tetracycline-resistant (n=178) — 98.6% susceptible (S); MRSA: 96.4% S; MSSA: 99.8% S; Tet-R: 90.4% S
- *Staphylococcus lugdunensis*, 81 isolates (MIC50/90, 0.03/0.06 mg/L) — 100.0% susceptible (S)
- *Coagulase-negative staphylococci*, S. capitis, S. epidermidis, S. haemolyticus, S. hominis — Inhibited by ≤0.5 mg/L (using S. aureus ABSSSI breakpoint) — ≥90.0% susceptible (S)
- *Streptococcus anginosus group*, 50 isolates (MIC50/90, 0.03/0.12 mg/L) — Includes tetracycline-resistant strains — 96.0% susceptible (S)
- *Streptococcus pneumoniae*, 600 isolates — Penicillin-resistant (n=63), Macrolide-resistant (n=230), Tetracycline-resistant (n=107) — 99.8% susceptible (S); Pen-R: 100.0% S; Mac-R: 99.6% S; Tet-R: 99.1% S
- *Streptococcus pyogenes*, 199 isolates (MIC50/90, 0.03/0.06 mg/L) — Macrolide-resistant (n=43), Tetracycline-resistant (n=63) — 100.0% susceptible (S)
- *Enterococcus faecalis*, 200 isolates (MIC50/90, 0.06/0.12 mg/L) — Tetracycline-resistant (n=146), Vancomycin-resistant (n=4) — 100.0% susceptible (S)
- *Enterococcus faecium*, 200 isolates — Vancomycin-resistant (subset) — 99.0% inhibited by ≤0.25 mg/L

Omadacycline showed excellent activity across key Gram-positive pathogens, including resistant phenotypes.

GRAM-NEGATIVE PATHOGENS (Table 2)

- *Haemophilus influenzae*, 354 isolates — Includes 109 β-lactamase positive isolates — 99.4% susceptible (S)
- *Haemophilus parainfluenzae*, 46 isolates — 97.8% susceptible (S)
- *Enterobacter cloacae species complex*, 400 isolates — 94.5% susceptible (S)
- *Klebsiella pneumoniae*, 1,000 isolates — ESBL-phenotype (n=255), Carbapenem-resistant (n=18) — 92.2% susceptible (S); ESBL: 84.3% S; CR: 100.0% S
- *Acinetobacter baumannii species complex*, 400 isolates — 97.2% inhibited by ≤4 µg/mL
- *Stenotrophomonas maltophilia*, 200 isolates — 85.5% inhibited by ≤4 µg/mL

Omadacycline maintained high susceptibility rates across diverse Gram-negative pathogens, including MDR and β-lactamase producers.

S = Susceptible | MIC50/90 = MIC at which 50%/90% of isolates are inhibited
Breakpoints: ABSSSI = Acute Bacterial Skin and Skin Structure Infection; CABP = Community-Acquired Bacterial Pneumonia
Data from omadacycline surveillance program, U.S. medical centers, 2024–2025.

Table 2. Antimicrobial activity of omadacycline and tetracycline class comparator agents against 3,400 Gram-negative bacterial isolates collected from medical centers in the United States (2024–2025)

Organism (no. tested)	Omadacycline			Doxycycline			Minocycline			Tetracycline			Tigecycline		
	$MIC_{50/90}$		%S (FDA)	$MIC_{50/90}$		%S (CLSI)	$MIC_{50/90}$		%S (CLSI)	$MIC_{50/90}$		%S (CLSI)	$MIC_{50/90}$		%S (FDA)
Acinetobacter baumannii-calcoaceticus species complex (400)	0.25	**4**	—[a]	0.25	>8	—	≤0.25	8	77.0	2	>16	—	0.25	2	—
Enterobacter cloacae species complex (400)	1	**4**	94.5[b]	2	4	90.2	2	4	91.2	2	8	88.7	0.25	1	98.8
Escherichia coli (800)	0.5	**1**	—[a]	1	>8	78.9	0.5	4	91.6	2	>16	72.6	0.12	0.25	100.0
ESBL-phenotype (172)	1	**2**	—[a]	4	>8	58.7	1	8	84.9	>16	>16	47.1	0.25	0.25	100.0
Haemophilus influenzae (354)	0.5	**1**	99.4[c]	—	—	—	—	—	—	0.5	0.5	99.7	0.25	0.5	83.2
β-lactamase positive (109)	0.5	**1**	98.2[c]	—	—	—	—	—	—	0.5	0.5	99.1	0.25	0.5	79.8
Haemophilus parainfluenzae (46)	1	**2**	97.8[c]	—	—	—	—	—	—	0.5	1	95.7	0.25	1	—
Klebsiella pneumoniae (1,000)	1	**4**	92.2[b,c]	1	>8	73.5	1	16	83.3	1	>16	73.5	0.5	1	97.8
ESBL-phenotype (255)	2	**8**	84.3[b,c]	8	>8	42.0	2	16	74.1	>16	>16	40.8	0.5	1	96.5
Carbapenem-R (18)	1	**4**	100.0[b,c]	2	8	83.3	2	8	72.2	4	16	83.3	0.5	1	100.0
Moraxella catarrhalis (604)	≤0.12	**≤0.12**	—[a]	—	—	—	—	—	—	0.25	0.5	99.5	0.06	0.06	—
Stenotrophomonas maltophilia (200)	2	**8**	—[a]	—	—	—	0.5	2	89.9	—	—	—	1	4	—

CLSI, Clinical and Laboratory Standards Institute; ESBL, extended-spectrum β-lactamase; S, susceptible; FDA, United States Food and Drug Administration; R, resistant.
[a] Breakpoint interpretive criteria are unavailable.
[b] ABSSSI breakpoint applied.
[c] CABP breakpoint applied.

Bold, MIC_{90} values.
Green, susceptible according to CLSI or FDA breakpoint interpretive criteria.
Yellow, intermediate according to CLSI or FDA breakpoint interpretive criteria.
Orange, resistant according to CLSI or FDA breakpoint interpretive criteria.

Acknowledgements

This study has been funded in whole or in part by federal funds from the U.S. Department of Health and Human Services, Administration for Strategic Preparedness and Response, Biomedical Advanced Research and Development Authority (BARDA), under Contract No. 75A50120C00001 and Paratek Pharmaceuticals, Inc.

MD Huband, ML Winkler, KA Fedler, KM Papp-Wallace, and M Castanheira were employees of Element Materials Technology (JMI Laboratories) at the time of this study, which was a paid consultant to Paratek Pharmaceuticals in connection with the development of this poster.

References

Clinical and Laboratory Standards Institute (2024). M07-A12. *Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard—eleventh edition*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2026). M100-S36. *Performance standards for antimicrobial susceptibility testing: 36th informational supplement.* Wayne, PA: CLSI.

US Food and Drug Administration: https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria. Accessed July 2026.

Poster P-383 presented at IDWeek 2026, 21–24 October 2026, Washington, DC

Contact: Michael D. Huband, Director; Element Iowa City, 345 Beaver Kreek Center, North Liberty, IA 52317
319-665-3370 x 203; michael.huband@element.com