Activity of Omadacycline and Comparators Against Common Microbial Contaminants: *Cronobacter sakazakii*, *Pluralibacter gergoviae*, and *Burkholderia cepacia* species complex Isolates

Michael D. Huband[1], Paul R. Rhomberg[1], Allison Fuhrmeister[1], Jessica V. Pierce[2], and Mariana Castanheira[1]

[1]Element Iowa City (JMI Laboratories), North Liberty, Iowa, United States; [2]Paratek Pharmaceuticals, King of Prussia, Pennsylvania, USA

Introduction

- Microbial contaminants commonly found in consumer products (i.e., infant formula, cosmetics) include *Cronobacter sakazakii*, *Burkholderia cepacia* species complex (*B. cepacia*), and *Pluralibacter gergoviae* and are responsible for causing illnesses leading to major product recalls.
- *Cronobacter sakazakii* isolates that have been found to be in infant formula are resistant to desiccation and can cause severe illness in infants.
- *Burkholderia cepacia* and *Pluralibacter gergoviae* isolates that have been found to be in cosmetics are resistant to disinfectants and preservatives, respectively, as well as multiple antibacterial agents.
- Omadacycline is an FDA-approved tetracycline class antibacterial agent indicated for the treatment of acute bacterial skin and skin structure infection (ABSSSI) and community-acquired bacterial pneumonia (CABP) in adults against indicated bacterial pathogens.
- Omadacycline has previously demonstrated potent *in vitro* activity against bacterial isolates expressing common resistance mechanisms to tetracycline, penicillin, fluoroquinolone, macrolide, and β-lactam agents.
- In this study, the *in vitro* activity of omadacycline and comparator agents was investigated against a contemporary collection of *Burkholderia cepacia* isolates, and contemporary and historical isolates of *Cronobacter sakazakii*, and *Pluralibacter gergoviae*.

Materials and Methods

- Bacterial isolates tested included 104 *Burkholderia cepacia* species complex (2025), 31 *Cronobacter sakazakii* (2006–2024), and 85 *Pluralibacter gergoviae* (2001–2025) isolates.
- Bacterial isolates were recovered from patients with bloodstream infection (n=46; 20.9% overall), skin and skin structure infection (n=24; 10.9% overall), pneumonia in hospitalized patients (n=109; 49.5% overall), urinary tract infection (n=22; 10.0% overall), intra-abdominal infection (n=5; 2.3% overall), and other infection types (n=14; 6.4%).
- Bacterial isolate identifications were confirmed at Element Iowa City using standard microbiology methods and matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany).
- Broth microdilution susceptibility testing was performed according to Clinical and Laboratory Standards Institute M07 (CLSI; 2024) and M100 (2026) guidelines.
- MIC results were interpreted using CLSI (2026) and United States Food and Drug Administration (FDA) breakpoint criteria (2026).

Table 1. Antimicrobial activity of omadacycline and comparators against 31 *Cronobacter sakazakii* isolates

Antimicrobial agent	MIC (mg/L)			CLSI or US FDA[a]		
	MIC_{50}	MIC_{90}	MIC range	%S	%I	%R
Omadacycline	**1**	**2**	**0.5 to 8**	**96.8 [b]**	**3.2**	**0.0**
Tetracycline	1	2	0.5 to 4	100.0 [c]	0.0	0.0
Amikacin	≤4	≤4	≤4 to ≤4	100.0 [c]	0.0	0.0
Aztreonam	≤0.12	≤0.12	≤0.12 to ≤0.12	100.0 [c]	0.0	0.0
Cefepime	≤0.5	≤0.5	≤0.5 to ≤0.5	100.0 [c,d]	0.0	0.0
Ceftazidime	≤1	≤1	≤1 to ≤1	100.0 [c]	0.0	0.0
Ceftriaxone	≤0.25	≤0.25	≤0.25 to ≤0.25	100.0 [c]	0.0	0.0
Gentamicin	≤2	≤2	≤2 to ≤2	100.0 [c]	0.0	0.0
Meropenem	0.015	0.015	0.015 to 0.03	100.0 [c]	0.0	0.0
Levofloxacin	0.015	0.03	0.015 to 0.12	100.0 [c]	0.0	0.0
Piperacillin-tazobactam	1	2	≤0.5 to 8	100.0	0.0	0.0
Tobramycin	0.25	0.5	0.25 to 1	100.0 [c]	0.0	0.0
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	≤0.5 to >4	96.8 [c]		3.2

S, susceptible; I, intermediate; R, resistant.
[a] Criteria as published by CLSI (2026) and US FDA (2026).
[b] Omadacycline ABSSSI breakpoints for *E. cloacae* species complex and *K. pneumoniae* applied for comparison purposes.
[c] CLSI M100 standard is recognized.
[d] Intermediate is interpreted as susceptible-dose dependent.

Table 2. Antimicrobial activity of omadacycline and comparators against 85 *Pluralibacter gergoviae* isolates

Antimicrobial agent	MIC (mg/L)			CLSI or US FDA[a]		
	MIC_{50}	MIC_{90}	MIC range	%S	%I	%R
Omadacycline	**1**	**2**	**0.5 to 8**	**98.8 [b]**	**1.2**	**0.0**
Tetracycline	1	2	0.5 to >32	98.8 [c]	0.0	1.2
Amikacin	≤4	≤4	≤4 to >32	97.6 [c]	1.2	1.2
Aztreonam	≤0.12	0.5	≤0.12 to >16	92.9 [c]	2.4	4.7
Cefepime	≤0.5	≤0.5	≤0.5 to >16	97.6 [c,d]	0.0	2.4
Ceftazidime	≤2	≤2	≤2 to >16	94.1 [c]	1.2	4.7
Ceftriaxone	≤0.25	0.5	≤0.25 to >8	92.9 [c]	0.0	7.1
Gentamicin	≤2	≤2	≤2 to >8	95.3 [c]	1.2	3.5
Meropenem	0.03	0.06	0.015 to >8	97.6 [c]	0.0	2.4
Levofloxacin	0.03	0.06	0.015 to 4	97.6 [c]	0.0	2.4
Piperacillin-tazobactam	1	4	≤0.5 to >64	97.6	0.0	2.4
Tobramycin	0.25	0.5	≤0.12 to >8	92.9 [c]	2.4	4.7
Trimethoprim-sulfamethoxazole	≤0.5	≤0.5	≤0.5 to >2	94.1 [c]		5.9

S, susceptible; I, intermediate; R, resistant.
[a] Criteria as published by CLSI (2026) and US FDA (2026).
[b] Omadacycline ABSSSI breakpoints for *E. cloacae* species complex and *K. pneumoniae* applied for comparison purposes.
[c] CLSI M100 (2026) standard is recognized.
[d] Intermediate is interpreted as susceptible-dose dependent.

Discussion

Increasing numbers of recalls for consumer products due to microbial contamination highlight the need for effective antibacterial treatment options.

Novel antibacterial agents or combination therapies are needed for the treatment of infections caused by the *Burkholderia cepacia* species complex isolates.

Conclusions

Omadacycline demonstrated potent *in vitro* activity against *Cronobacter sakazakii* isolates, inhibiting 96.8% of all isolates at or below the current omadacycline susceptible breakpoint for Enterobacteriaceae.

Omadacycline demonstrated potent *in vitro* activity against *Pluralibacter gergoviae* isolates, inhibiting 98.8% of all isolates at or below the current omadacycline susceptible breakpoint for Enterobacteriaceae.

Limited activity was observed for omadacycline and all comparator agents against *Burkholderia cepacia* isolates.

Omadacycline may represent an additional treatment option (oral and intravenous dosing) for infections caused by *Cronobacter sakazakii* and *Pluralibacter gergoviae*; further investigation is warranted, particularly against isolates sourced from contaminated products.

Scan Me

Scan for a copy of this poster

https://www.jmilabs.com/data/posters/IDWeek2026_25-PAR-04_P1_MIC90.pdf

PARATEK

Results

- Applying FDA breakpoint interpretive criteria for omadacycline (ABSSSI and CABP) to other Enterobacteriaceae organism groups for comparison purposes resulted in susceptibilities of 96.8% for *Cronobacter sakazakii* ($MIC_{50/90}$, 1/2 mg/L) and 98.8% for *Pluralibacter gergoviae* ($MIC_{50/90}$, 1/2 mg/L) isolates (Tables 1–2 and Figure 1).
- Comparator agent susceptibilities for *Cronobacter sakazakii* ranged from 96.8%–100.0% (Table 1).
- Comparator agent susceptibilities for *Pluralibacter gergoviae* ranged from 92.9%–98.8% (Table 2).
- Omadacycline ($MIC_{50/90}$, 4/32 mg/L) inhibited 65.4% (68/104) of *Burkholderia cepacia* isolates at ≤4 mg/L; the current omadacycline susceptible breakpoint for Enterobacteriaceae (Table 3; Figure 1).
- All comparator agents showed limited activity against *Burkholderia cepacia* isolates (Table 3).

Table 3. Antimicrobial activity of omadacycline and comparators against 104 *Burkholderia cepacia* isolates

Antimicrobial agent	MIC (mg/L)		
	MIC_{50}	MIC_{90}	MIC range
Omadacycline	**4**	**32**	**0.5 to >32**
Amikacin	>32	>32	8 to >32
Aztreonam	>16	>16	1 to >16
Cefepime	16	>32	0.5 to >32
Ceftazidime	4	16	0.5 to >32
Colistin	>8	>8	8 to >8
Gentamicin	>16	>16	8 to >16
Meropenem	4	8	0.25 to >32
Levofloxacin	2	32	0.5 to >32
Piperacillin-tazobactam	4	128	≤0.25 to >128
Tigecycline	2	>8	0.25 to >8
Tobramycin	>16	>16	4 to >16
Trimethoprim-sulfamethoxazole	0.5	>4	≤0.12 to >4

No current breakpoints are available for *B. cepacia* species complex.

Figure 1. Cumulative percent inhibition results for omadacycline against *Burkholderia cepacia*, *Cronobacter sakazakii*, and *Pluralibacter gergoviae* isolates

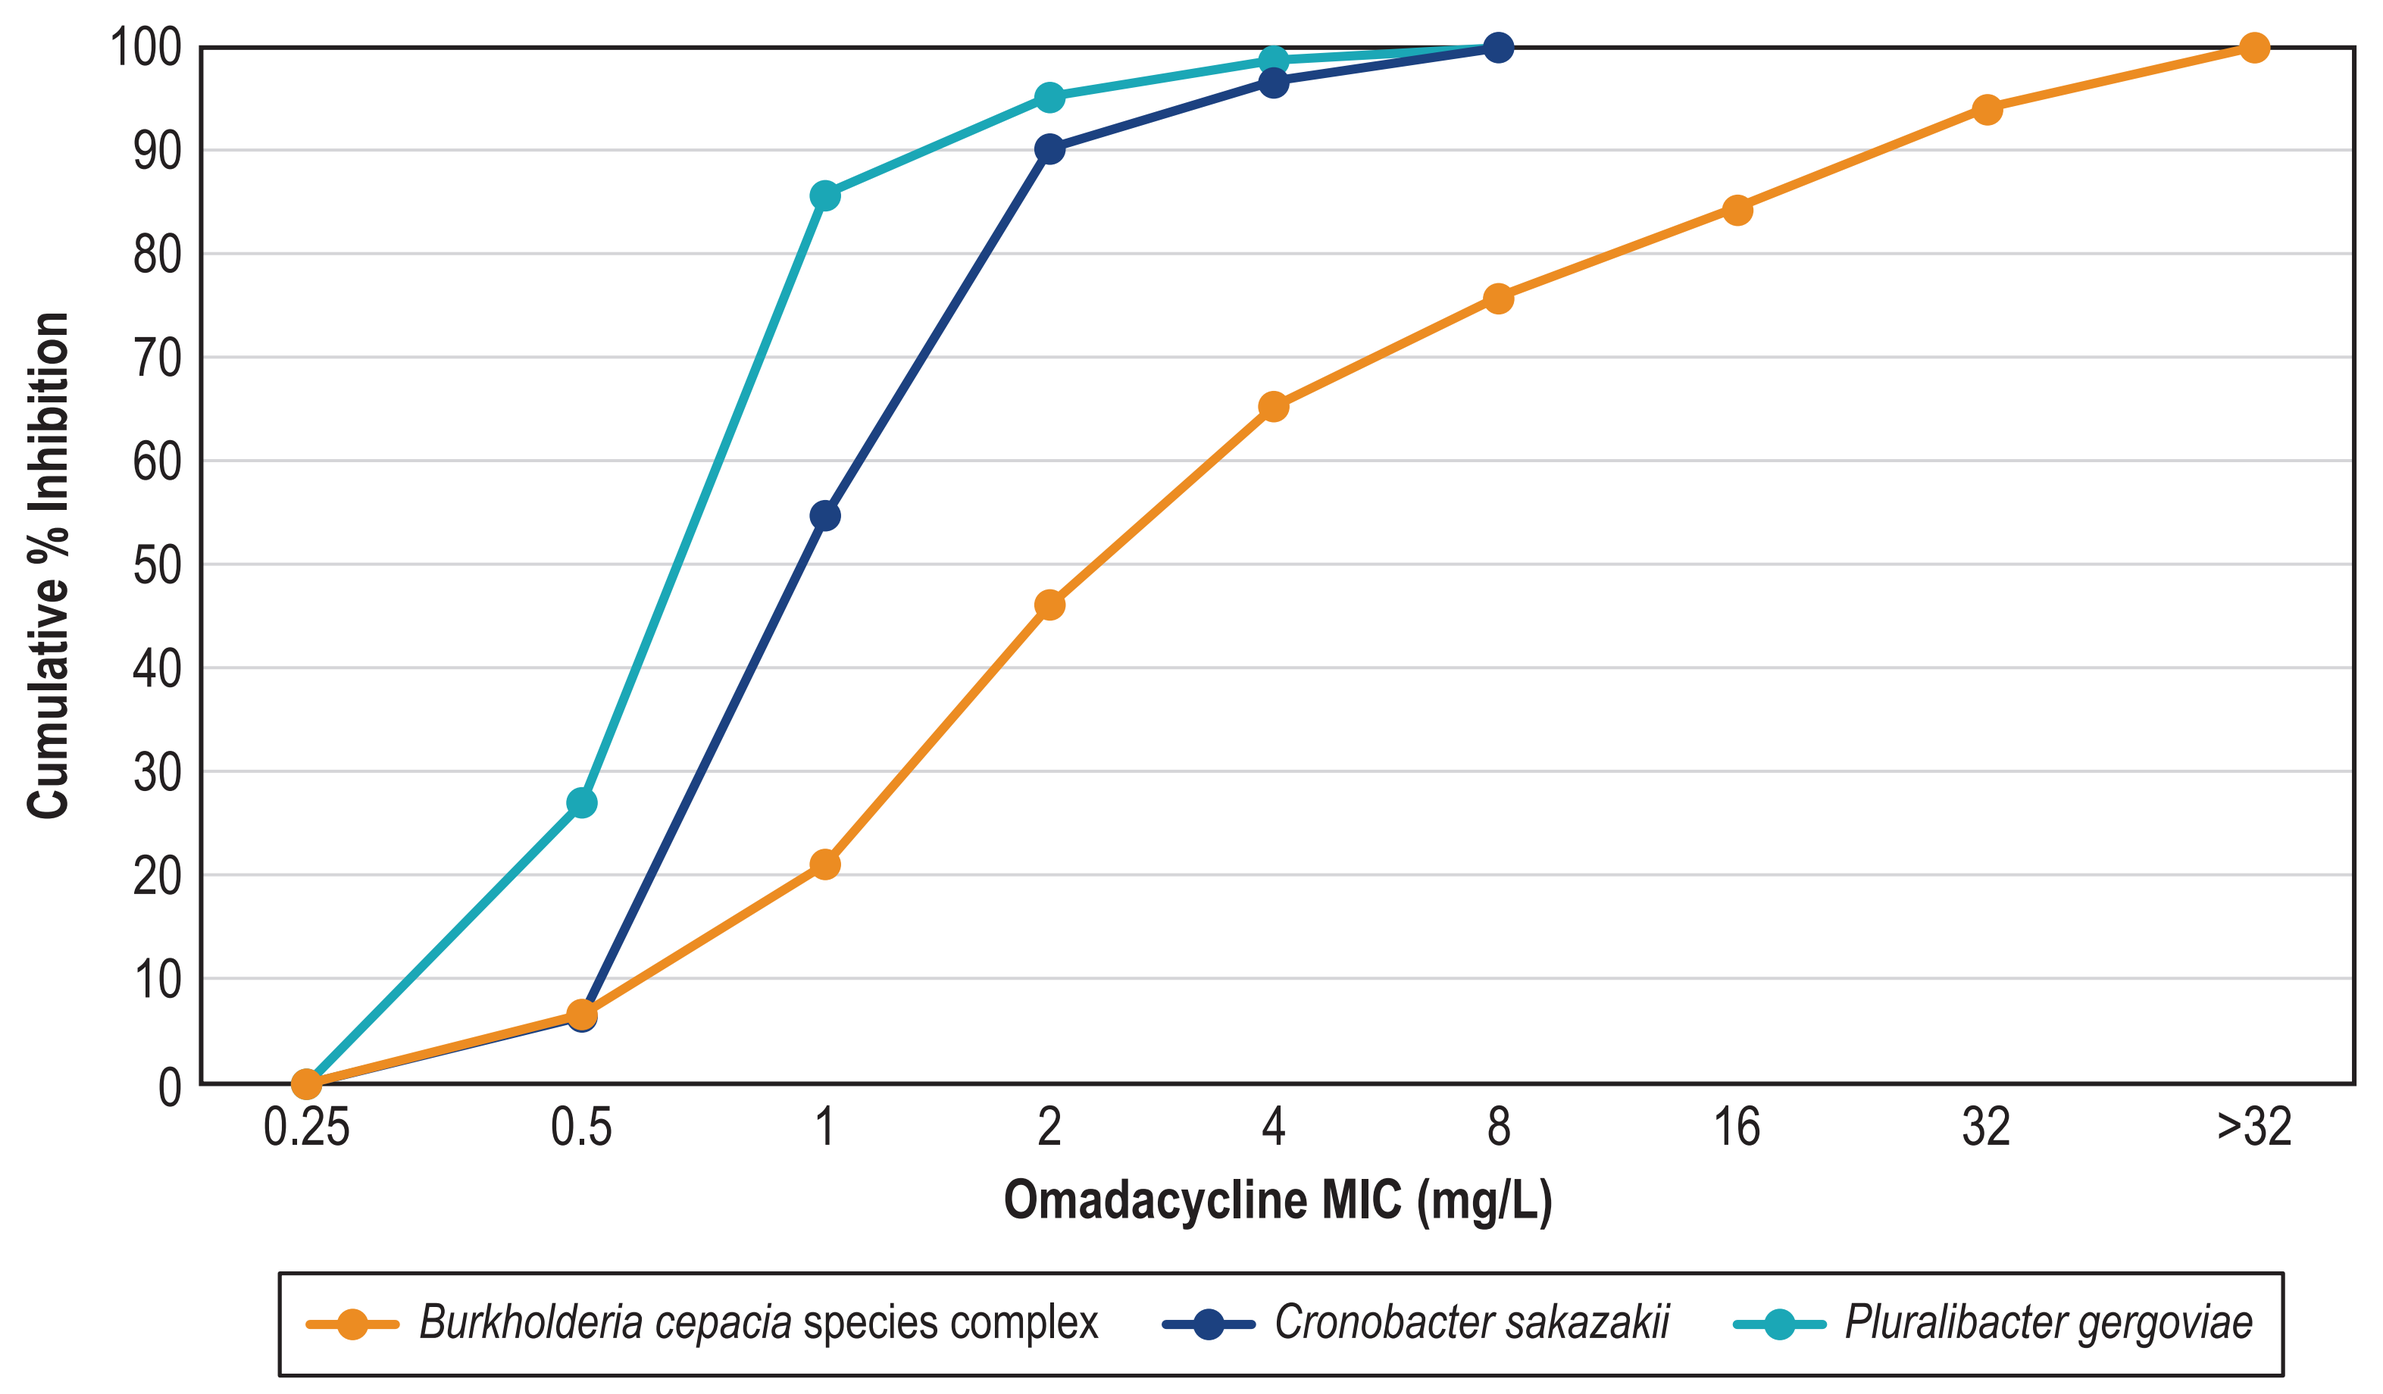
Cumulative % Inhibition
100
90
80
70
60
50
40
30
20
10
0
0.25
0.5
1
2
4
8
16
32
>32
Omadacycline MIC (mg/L)
Burkholderia cepacia species complex
Cronobacter sakazakii
Pluralibacter gergoviae

Acknowledgements

This study and poster presentation were supported by funding from Paratek Pharmaceuticals, Inc.

References

Clinical and Laboratory Standards Institute (2024). M07-A12. *Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard—eleventh edition*. Wayne, PA: CLSI.

Clinical and Laboratory Standards Institute (2026). M100-S36. *Performance standards for antimicrobial susceptibility testing: 36th informational supplement.* Wayne, PA: CLSI.

US Food and Drug Administration: https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria. Accessed July 2026.

Poster P-488 presented at IDWeek 2026, 21–24 October 2026, Washington, DC

Contact: Michael D. Huband, Director; Element Iowa City, 345 Beaver Kreek Center, North Liberty, IA 52317
319-665-3370 x 203; michael.huband@element.com