IDWeek 2026 | Poster #P-172

Reproducibility of Cefiderocol-Xeruborbactam in Cation Adjusted Mueller Hinton Broth (CAMHB) and Iron Depleted (ID) CAMHB Using Two Reading Endpoints

Mariana Castanheira[1], SJ Ryan Arends[1], John H. Kimbrough[1], Joshua M. Maher[1], Takafumi Hara[3], Yoshinori Yamano[3], Olga Lomovskaya[2]

[1]Element Iowa City/JMI Laboratories, North Liberty, Iowa; [2]Qpex Biopharma, San Diego, California; [3]Shionogi & Co., Ltd., Osaka, Japan

Introduction

- Production of β-lactamases is the main β-lactam resistance mechanism among Gram-negative species and to overcome this resistance mechanism, β-lactam agents can be combined with β-lactamase inhibitors.
- Xeruborbactam is a novel boronic acid β-lactamase inhibitor that restores the cefiderocol activity against isolates producing β-lactamases, including Ambler classes A, B, C, and D.
- Cefiderocol susceptibility testing using the reference broth microdilution method (BMD) is performed in iron-depleted (ID) Cation Adjusted Mueller Hinton broth (CAMHB) and read at significant reduction (SR) of growth.
- We investigated the reproducibility of the reference BMD when testing cefiderocol-xeruborbactam in CAMHB and IDCAMHB and reading SR and 100% inhibition endpoints.

Materials and Methods

- Cefiderocol-xeruborbactam was tested by BMD against 212 Gram-negative clinical isolates that included 90 *Acinetobacter baumannii-calcoaceticus* complex, 57 *Pseudomonas aeruginosa*, and 65 Enterobacterales.
- Susceptibility testing was performed by the CLSI reference broth microdilution method as described by the Clinical and Laboratory Standards Institute (CLSI) M07 (2024) and M100 (2026) documents.
- Quality control (QC) was performed according to the CLSI M100 (2026) criteria using recently approved cefiderocol-xeruborbactam QC ranges.
- Isolates were tested in 6 to 16 replicates using four media manufacturers/lots of CAMHB and IDCAMHB prepared with these same manufacturers/lots and results were read by the same technicians.
- Cefiderocol-xeruborbactam was tested in triplicate against a subset of 50 isolates using the same four CAMHB manufacturers/lots and read by 10 technicians with different experience levels.
- Endpoints were determined as significant reduction (SR) in growth as defined by CLSI for cefiderocol and 100% growth inhibition.
- Results were compared to the modal MIC for the same testing conditions/reading endpoint to determine percentage essential agreement at ±1 dilution.

Funding

This project has been funded in whole or in part with federal funds from the Department of Health and Human Services; Administration for Strategic Preparedness and Response; Center for the Biomedical Advanced Research and Development Authority (BARDA), under OTA number HHSO100201600026C.

IDWeek 2026, October 21–24, 2026, Washington, D.C.

Results

- The overall CAMHB essential agreement for on-scale values was 91.5% and 92.8% when reading at 100% growth inhibition and SR endpoints, respectively (Figure 1).
- The essential agreements for IDCAMHB when read at 100% growth inhibition and SR endpoints were 91.8% and 92.0%, respectively (Figure 1).
- The essential agreements varied among media manufacturers/lots, and they were lower for Oxoid CAMHB for the 100% endpoint (89.7% vs ≥91.5%; Figure 2).
- The Difco media displayed lower essential agreement values in IDCAMHB: 90.3% and 90.5% for 100% endpoint and SR, respectively, when compared to 91.5% to 93.2% for other media (Figure 2).
- The lowest essential agreements by species/groups were observed with *A. baumannii-calcoaceticus* complex isolates that ranged from 88.8% for IDCAMHB SR to 92.4% for CAMHB SR (Figure 3).
 - These values were ≥93.4% for *P. aeruginosa* and ≥91.2% for Enterobacterales (Figure 3).
- The essential agreement for the results for a subset of 50 isolates read by 10 readers was considerably higher for 100% growth inhibition (97.1%) compared to SR (91.6%) endpoint regardless of the reader's experience level (Figure 4).

Conclusions

- This robust evaluation of the reproducibility of cefiderocol-xeruborbactam BMD testing demonstrates that CAMHB and 100% growth inhibition endpoint reading had similar reproducibility compared to IDCAMHB SR, the recommended reading condition for cefiderocol BMD.
- Importantly, CAMHB and 100% growth inhibition endpoint reading displayed greater reproducibility compared to CAMHB and SR reading when results were determined by different readers.
- The BMD method of cefiderocol-xeruborbactam in CAMHB and 100% growth inhibition endpoint reading provides a more reliable and convenient method for testing this combination.

References

CLSI. 2024. M07Ed12. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. CLSI, Berwyn, PA.

CLSI. 2026. M100Ed36. Performance Standards for antimicrobial susceptibility testing; 36th informational supplement. CLSI, Berwyn, PA.

Drawz SM, Bonomo RA. Three decades of beta-lactamase inhibitors. Clin Microbiol Rev. 2010 Jan;23(1):160-201.

Lomovskaya O, Castanheira M, Lindley J, Rubio-Aparicio D, Nelson K, Tsivkovski R, Sun D, Totrov M, Loutit J, Dudley M. *In vitro* potency of xeruborbactam in combination with multiple β-lactam antibiotics in comparison with other β-lactam/β-lactamase inhibitor (BLI) combinations against carbapenem-resistant and extended-spectrum β-lactamase-producing *Enterobacterales*. Antimicrob Agents Chemother. 2023 Nov 15;67(11):e0044023.

Figure 1. Reproducibility of MIC results for cefiderocol-xeruborbactam compared to the modal MIC for 5,534 test results when testing 212 Gram-negative isolates

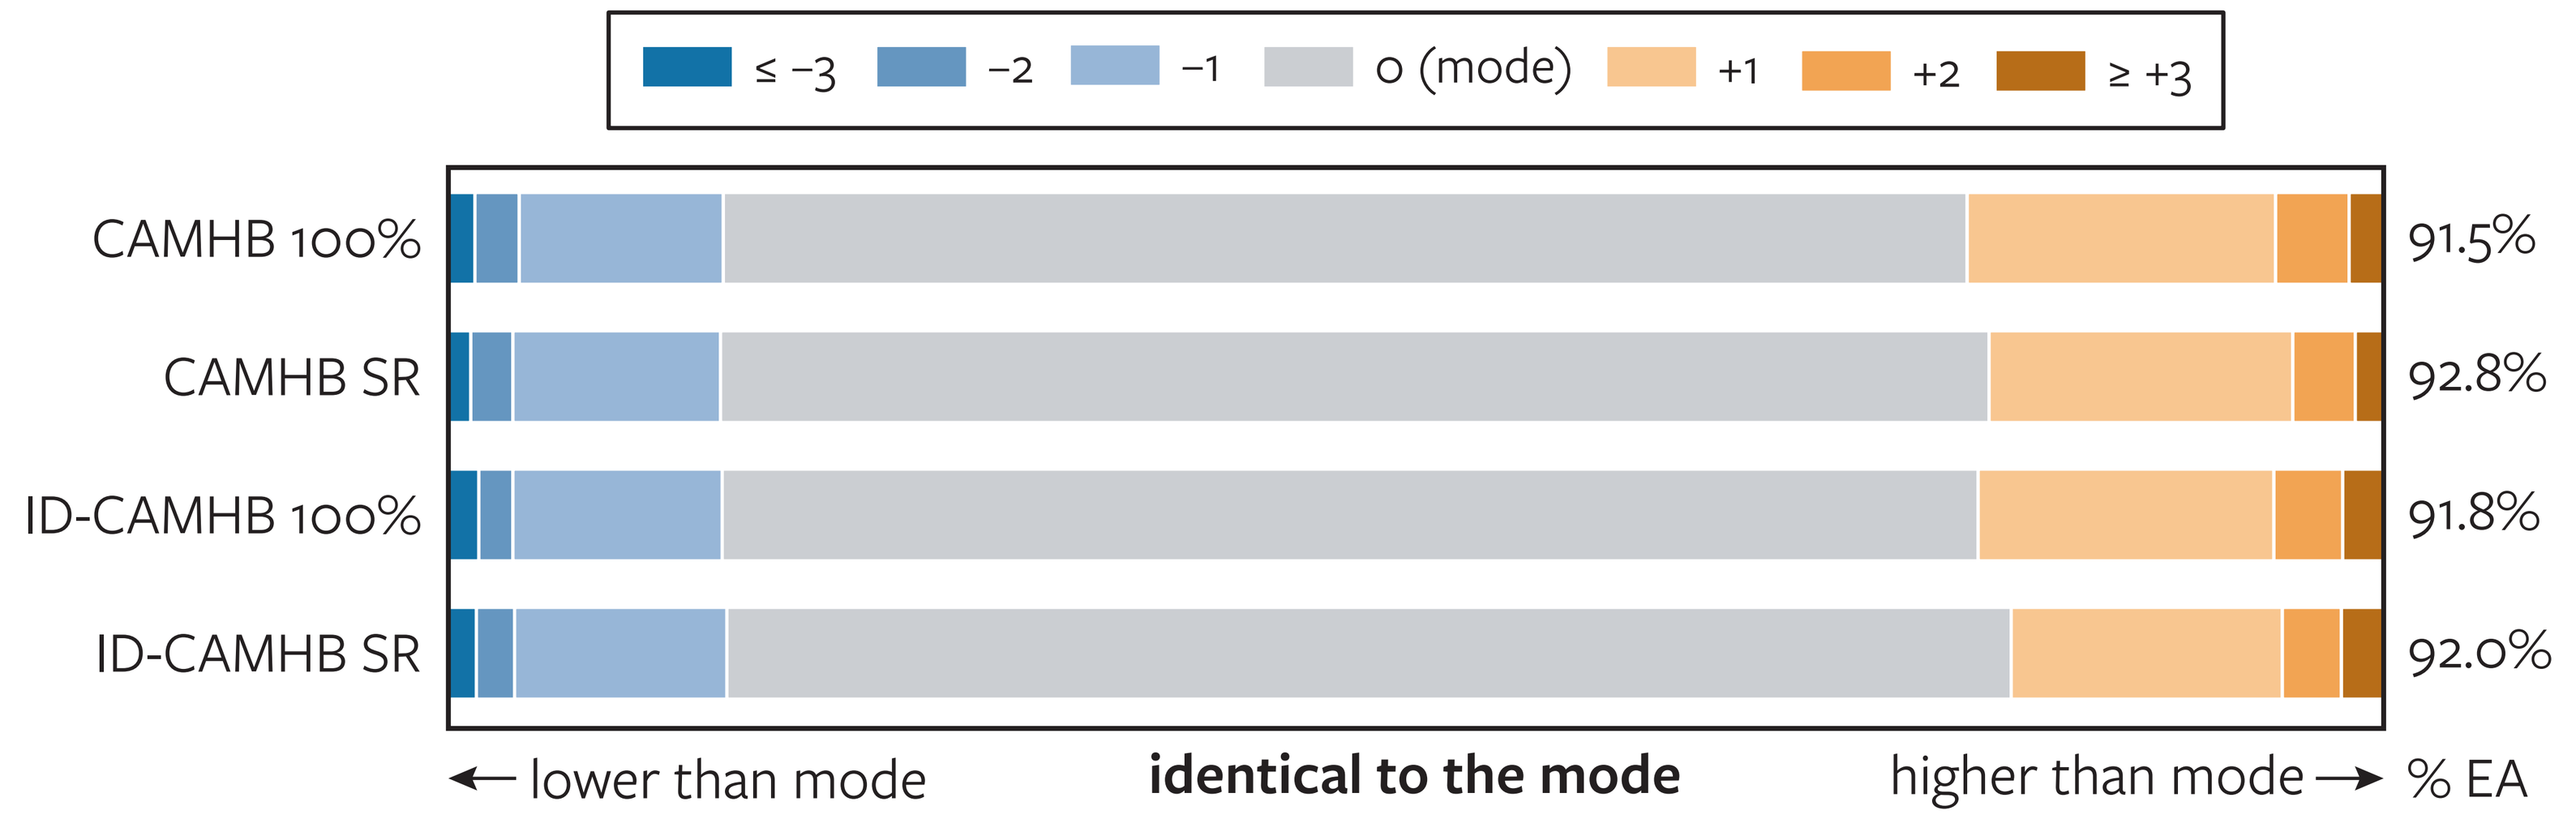
≤ −3
−2
−1
0 (mode)
+1
+2
≥ +3
CAMHB 100%
CAMHB SR
ID-CAMHB 100%
ID-CAMHB SR
91.5%
92.8%
91.8%
92.0%
lower than mode
identical to the mode
higher than mode
% EA

Figure 4. Essential agreement of MIC results for cefiderocol-xeruborbactam compared to the modal MIC for 50 Gram-negative isolates stratified by experience of the reader

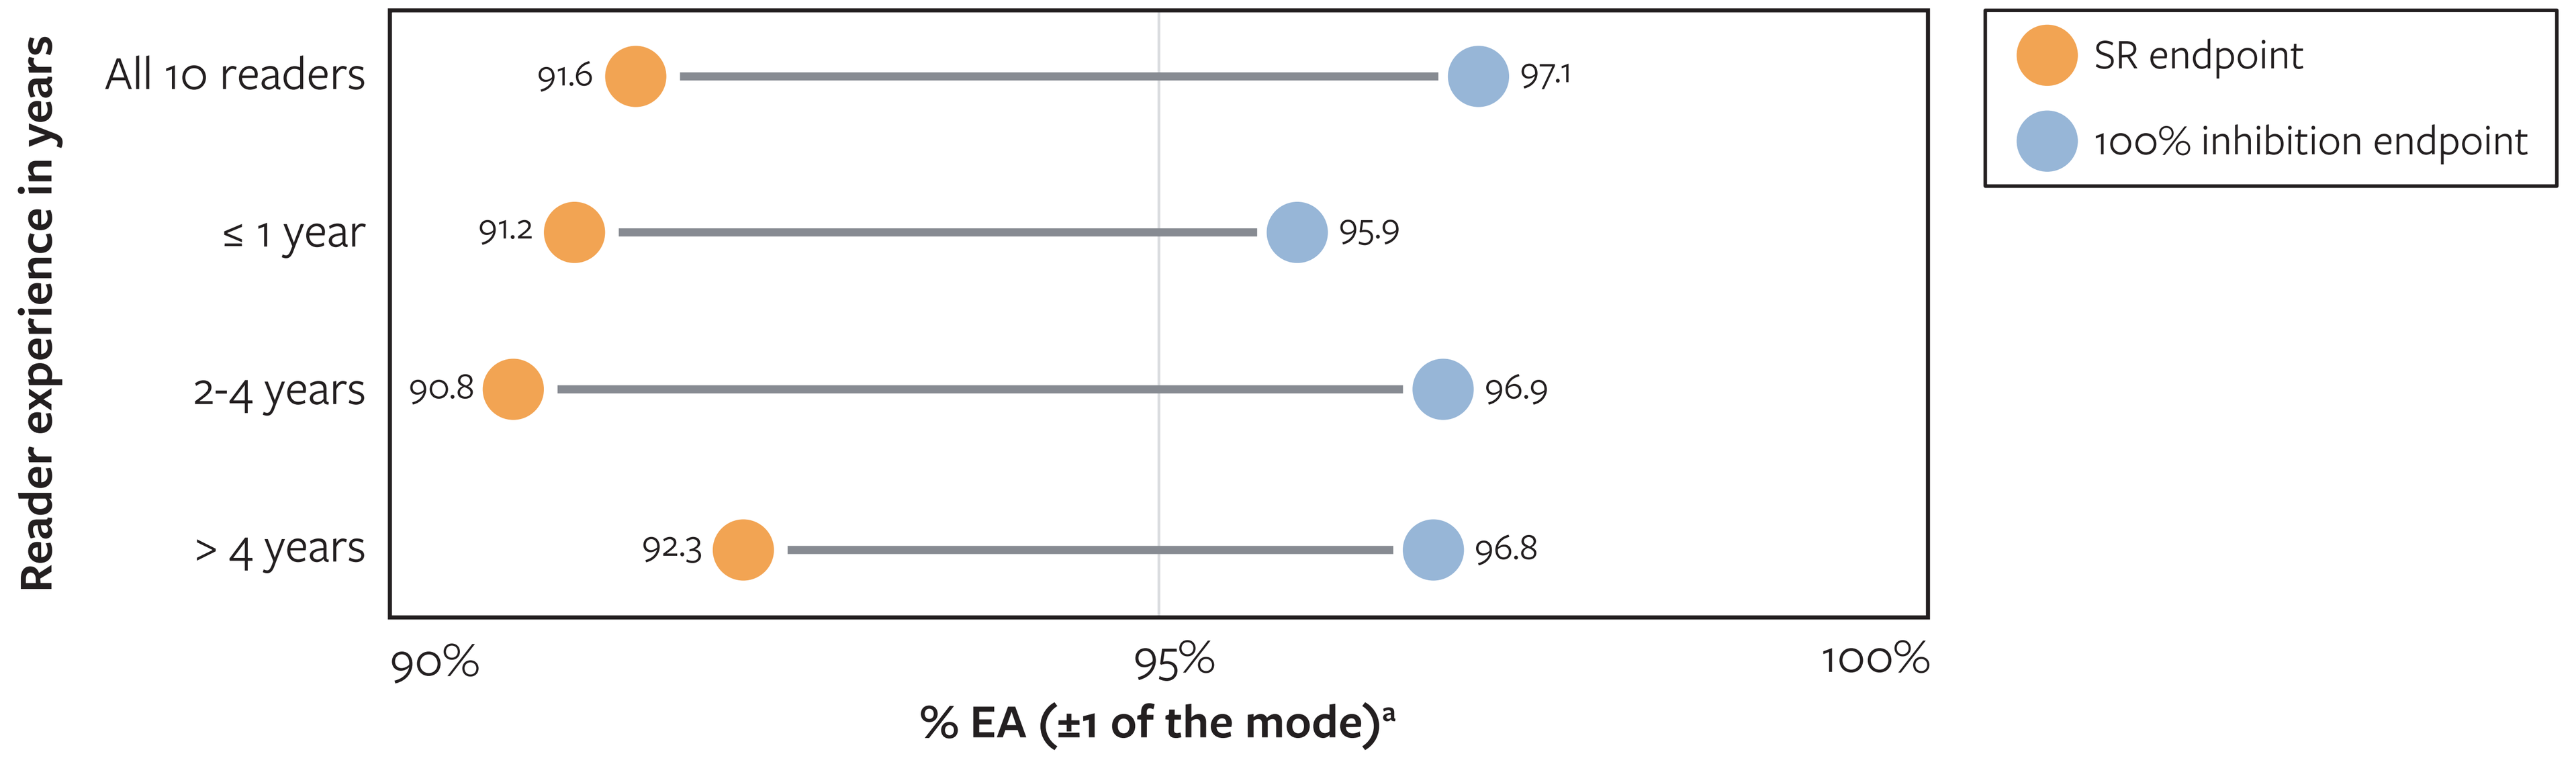
Reader experience in years
All 10 readers
≤ 1 year
2-4 years
> 4 years
91.6
97.1
91.2
95.9
90.8
96.9
92.3
96.8
90%
95%
100%
% EA (±1 of the mode)a
SR endpoint
100% inhibition endpoint

a % EA for single reader was calculated only for on-scale values.

Figure 2. Essential agreement of MIC results for cefiderocol-xeruborbactam compared to the modal MIC for 212 Gram-negative isolates stratified by media lot/manufacturer

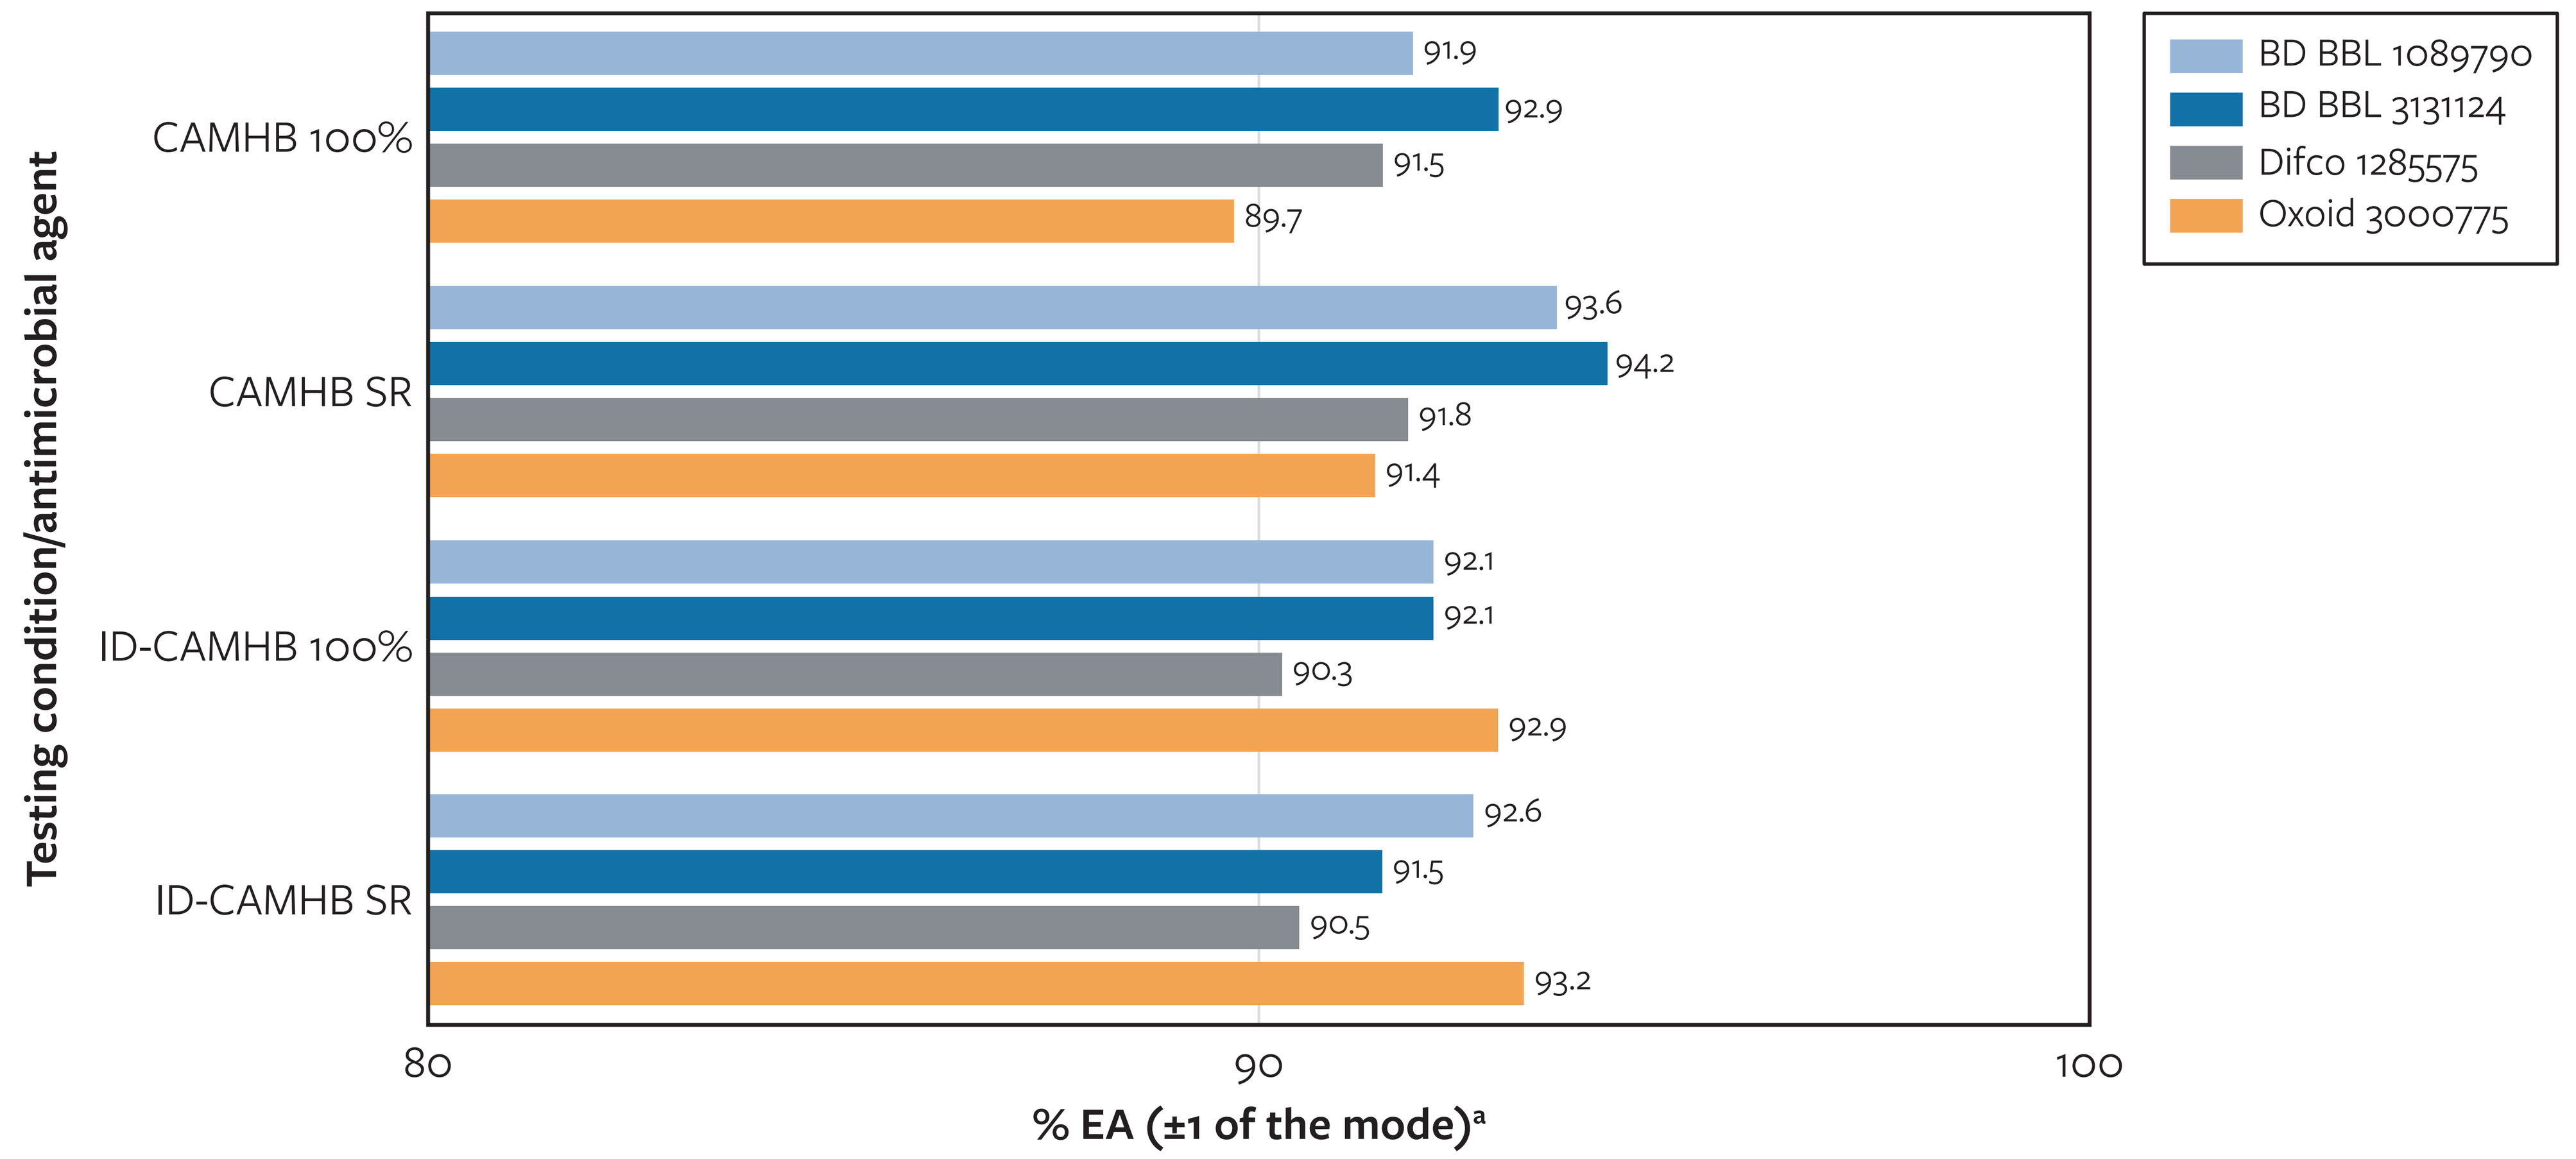
Testing condition/antimicrobial agent
CAMHB 100%
91.9
92.9
91.5
89.7
CAMHB SR
93.6
94.2
91.8
91.4
ID-CAMHB 100%
92.1
92.1
90.3
92.9
ID-CAMHB SR
92.6
91.5
90.5
93.2
80
90
100
% EA (±1 of the mode)a
BD BBL 1089790
BD BBL 3131124
Difco 1285575
Oxoid 3000775

a % EA for single reader was calculated only for on-scale values.

Figure 3. Essential agreement of MIC results for cefiderocol-xeruborbactam compared to the modal MIC for 212 Gram-negative isolates stratified by bacterial species/group

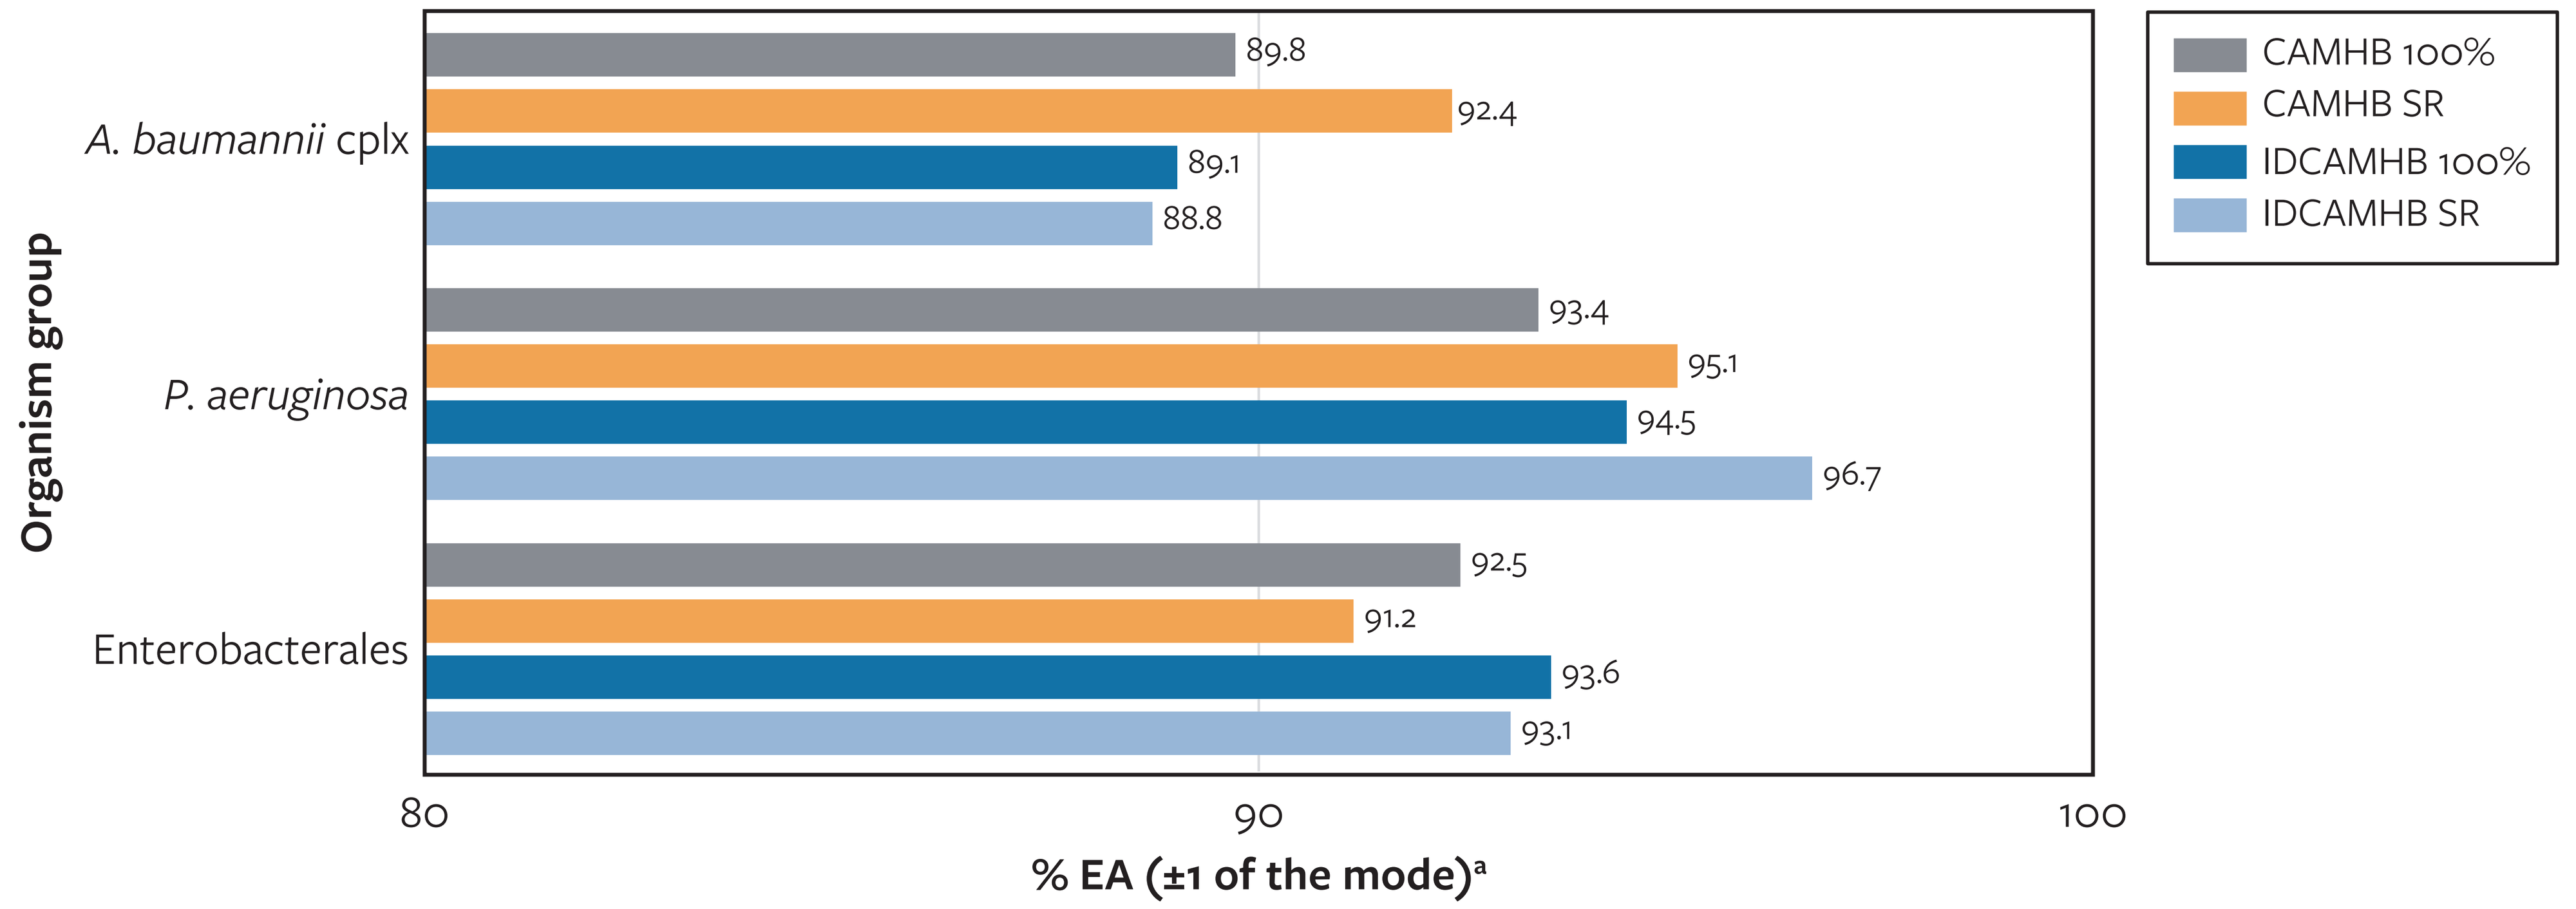
Organism group
A. baumannii cplx
89.8
92.4
89.1
88.8
P. aeruginosa
93.4
95.1
94.5
96.7
Enterobacterales
92.5
91.2
93.6
93.1
80
90
100
% EA (±1 of the mode)a
CAMHB 100%
CAMHB SR
IDCAMHB 100%
IDCAMHB SR

a % EA for single reader was calculated only for on-scale values.

Contact

element

Scan Me

Mariana Castanheira, PhD
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
mariana.castanheira@element.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2026_25-QPX-09_P1_Repro_Studies.pdf

Charges may apply. No personal information is stored.

Acknowledgments

Takafumi Hara and Yoshinori Yamano are employees of Shionogi. Olga Lomovskaya is an employee of QPEX.