IDWeek 2026 | Poster #P-141

In vitro Activity of Cefiderocol Against a Collection of Infrequently Isolated Gram-Negative Bacterial Isolates from the SENTRY Antimicrobial Surveillance Program

Joshua M. Maher [1], Rodrigo E. Mendes [1], Mariana Castanheira [1], Shannin C Zevian [1], Boudewijn L.M. DeJonge [2], Hidenori Yamashiro [3], Christopher Longshaw [4], Yoshinori Yamano [3]

[1] Element Iowa City (JMI Laboratories), North Liberty, IA 52317, USA; [2] Shionogi & Co., Ltd, 3 Chome-1-8 Doshomachi, Chuo Ward, Osaka, 541-0045, Japan; [3] Shionogi Inc., 400 Campus Dr, Florham Park, NJ 07932; [4] Shionogi BV, 50 Eastbourne Terrace, London W2 6LG, United Kingdom

Background

- Due to increased utilization of MALDI-TOF and sequencing based identification methods, rare and uncommon Gram-negative species, including non-fermentative Gram-negative bacteria (NFGNB), are becoming better understood and identified.
- Rare Enterobacterales and NFGNB species can lead to healthcare-associated or opportunistic infections, posing a significant public health risk, particularly for immunocompromised patients.
- These species possess diverse acquired and intrinsic resistance mechanisms, limiting antibiotic treatment options and contributing to the pervasive burden of antimicrobial resistance.
- Cefiderocol is a siderophore-conjugated cephalosporin that has demonstrated activity against Enterobacterales and NFGNB and maintains activity against multi-drug resistant and carbapenemase-producing strains; however, little data exists on cefiderocol's activity against rare or infrequently isolated species.
- The activity of cefiderocol and comparator agents was evaluated against a collection of 1,483 infrequently isolated Enterobacterales and NFGNB isolates collected in the global SENTRY Antimicrobial Surveillance Program.

Methods

- 1,483 isolates of infrequently recovered species—including 281 Enterobacterales from 24 genera and 1,202 NFGNB from 30 genera (Figure 1)—were collected from 141 medical centers across the United States, European, Latin America, and Asia-Pacific regions between 2013–2023 during the SENTRY Antimicrobial Surveillance Program.
- Isolates were selected to maximize phylogenetic and phenotypic diversity and originated primarily from patients with pneumonia (*n*=441) or urinary tract (*n*=143), bloodstream (*n*=410), gastrointestinal tract (*n*=12), or skin/soft tissue infections (*n*=301).
- Susceptibility testing followed Clinical and Laboratory Standards Institute (CLSI) methods, using cation-adjusted Mueller–Hinton broth (CAMHB) for comparator agents and iron-depleted CAMHB for cefiderocol.
- Where possible, susceptibility rates were determined using 2025 CLSI breakpoints.

Acknowledgments

The authors express appreciation to all US medical sites participating in the SENTRY Antimicrobial Surveillance Program for their most valuable support and contribution over the years. We also thank Lalitagauri Deshpande, Michael Janechek, and Maria Morabe for technical support and/or assistance with manuscript preparation. This study was performed by Element Iowa City (JMI Laboratories) and sponsored by Shionogi, Inc.

Results

- Cefiderocol displayed potent activity against Enterobacterales isolates (inhibiting all isolates at ≤1 mg/L), including indole-positive *Proteus* spp. ($MIC_{50/90}$, 0.015/0.03 mg/L, 100% susceptible per CLSI), non-*E. coli Escherichia* spp. (0.03/0.06 mg/L, 100%), *Pantoea* spp. (0.12/0.25 mg/L, 100%), and *Serratia* spp. (0.12/0.5 mg/L, 100%; Tables 1 and 2).
- Cefiderocol and meropenem displayed the highest susceptibility rates against rare Enterobacterales isolates (100% susceptible per CLSI for both); the remaining agents displayed rates of 99.3–69.6% (Table 2).
- Cefiderocol displayed activity against *Acinetobacter* spp. ($MIC_{50/90}$, 0.06/0.5 mg/L, 99.1% inhibited at ≤2 mg/L), *Achromobacter* spp. (0.015/0.25 mg/L), *Aeromonas* spp. (0.25/4 mg/L), *Elizabethkingia* spp. (0.5/1 mg/L), and *Pseudomonas* spp. (non-*P. aeruginosa*; 0.12/0.5 mg/L), although less active against *Burkholderia gladioli* (4/64 mg/L).
- Cefiderocol ($MIC_{50/90}$, 0.25/1 mg/L) and levofloxacin (1/2 mg/L) were the sole agents with activity against Flavobacteriales non-fermenters, including *Elizabethkingia* spp. isolates.
- Against all 1,202 NFGNB, cefiderocol ($MIC_{50/90}$, 0.06/1 mg/L) displayed potency greater than meropenem (0.25/4 mg/L), piperacillin-tazobactam (1/64 mg/L), cefepime (2/>16 mg/L), amikacin (2/>32 mg/L), and levofloxacin (0.25/2 mg/L).

Conclusions

- This study provides a contemporary evaluation of antimicrobial susceptibility against a collection of rare Enterobacterales and NFGNB bacterial isolates from 54 distinct genera and highlights the potential utility of cefiderocol in combating infections caused by these rare species.

References

CLSI. M100Ed36E. *Performance Standards for Antimicrobial Susceptibility Testing*. Wayne, PA, Clinical and Laboratory Standards Institute, 2026.

CLSI. *Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically*. 12th ed. CLSI standard M07. Clinical and Laboratory Standards Institute, 2024.

EUCAST. EUCAST Guidance Document on when there are no breakpoints. February 2024 ed2024.

Figure 1. Taxonomic overview and species-level diversity among 1,483 rare Gram-negative isolates

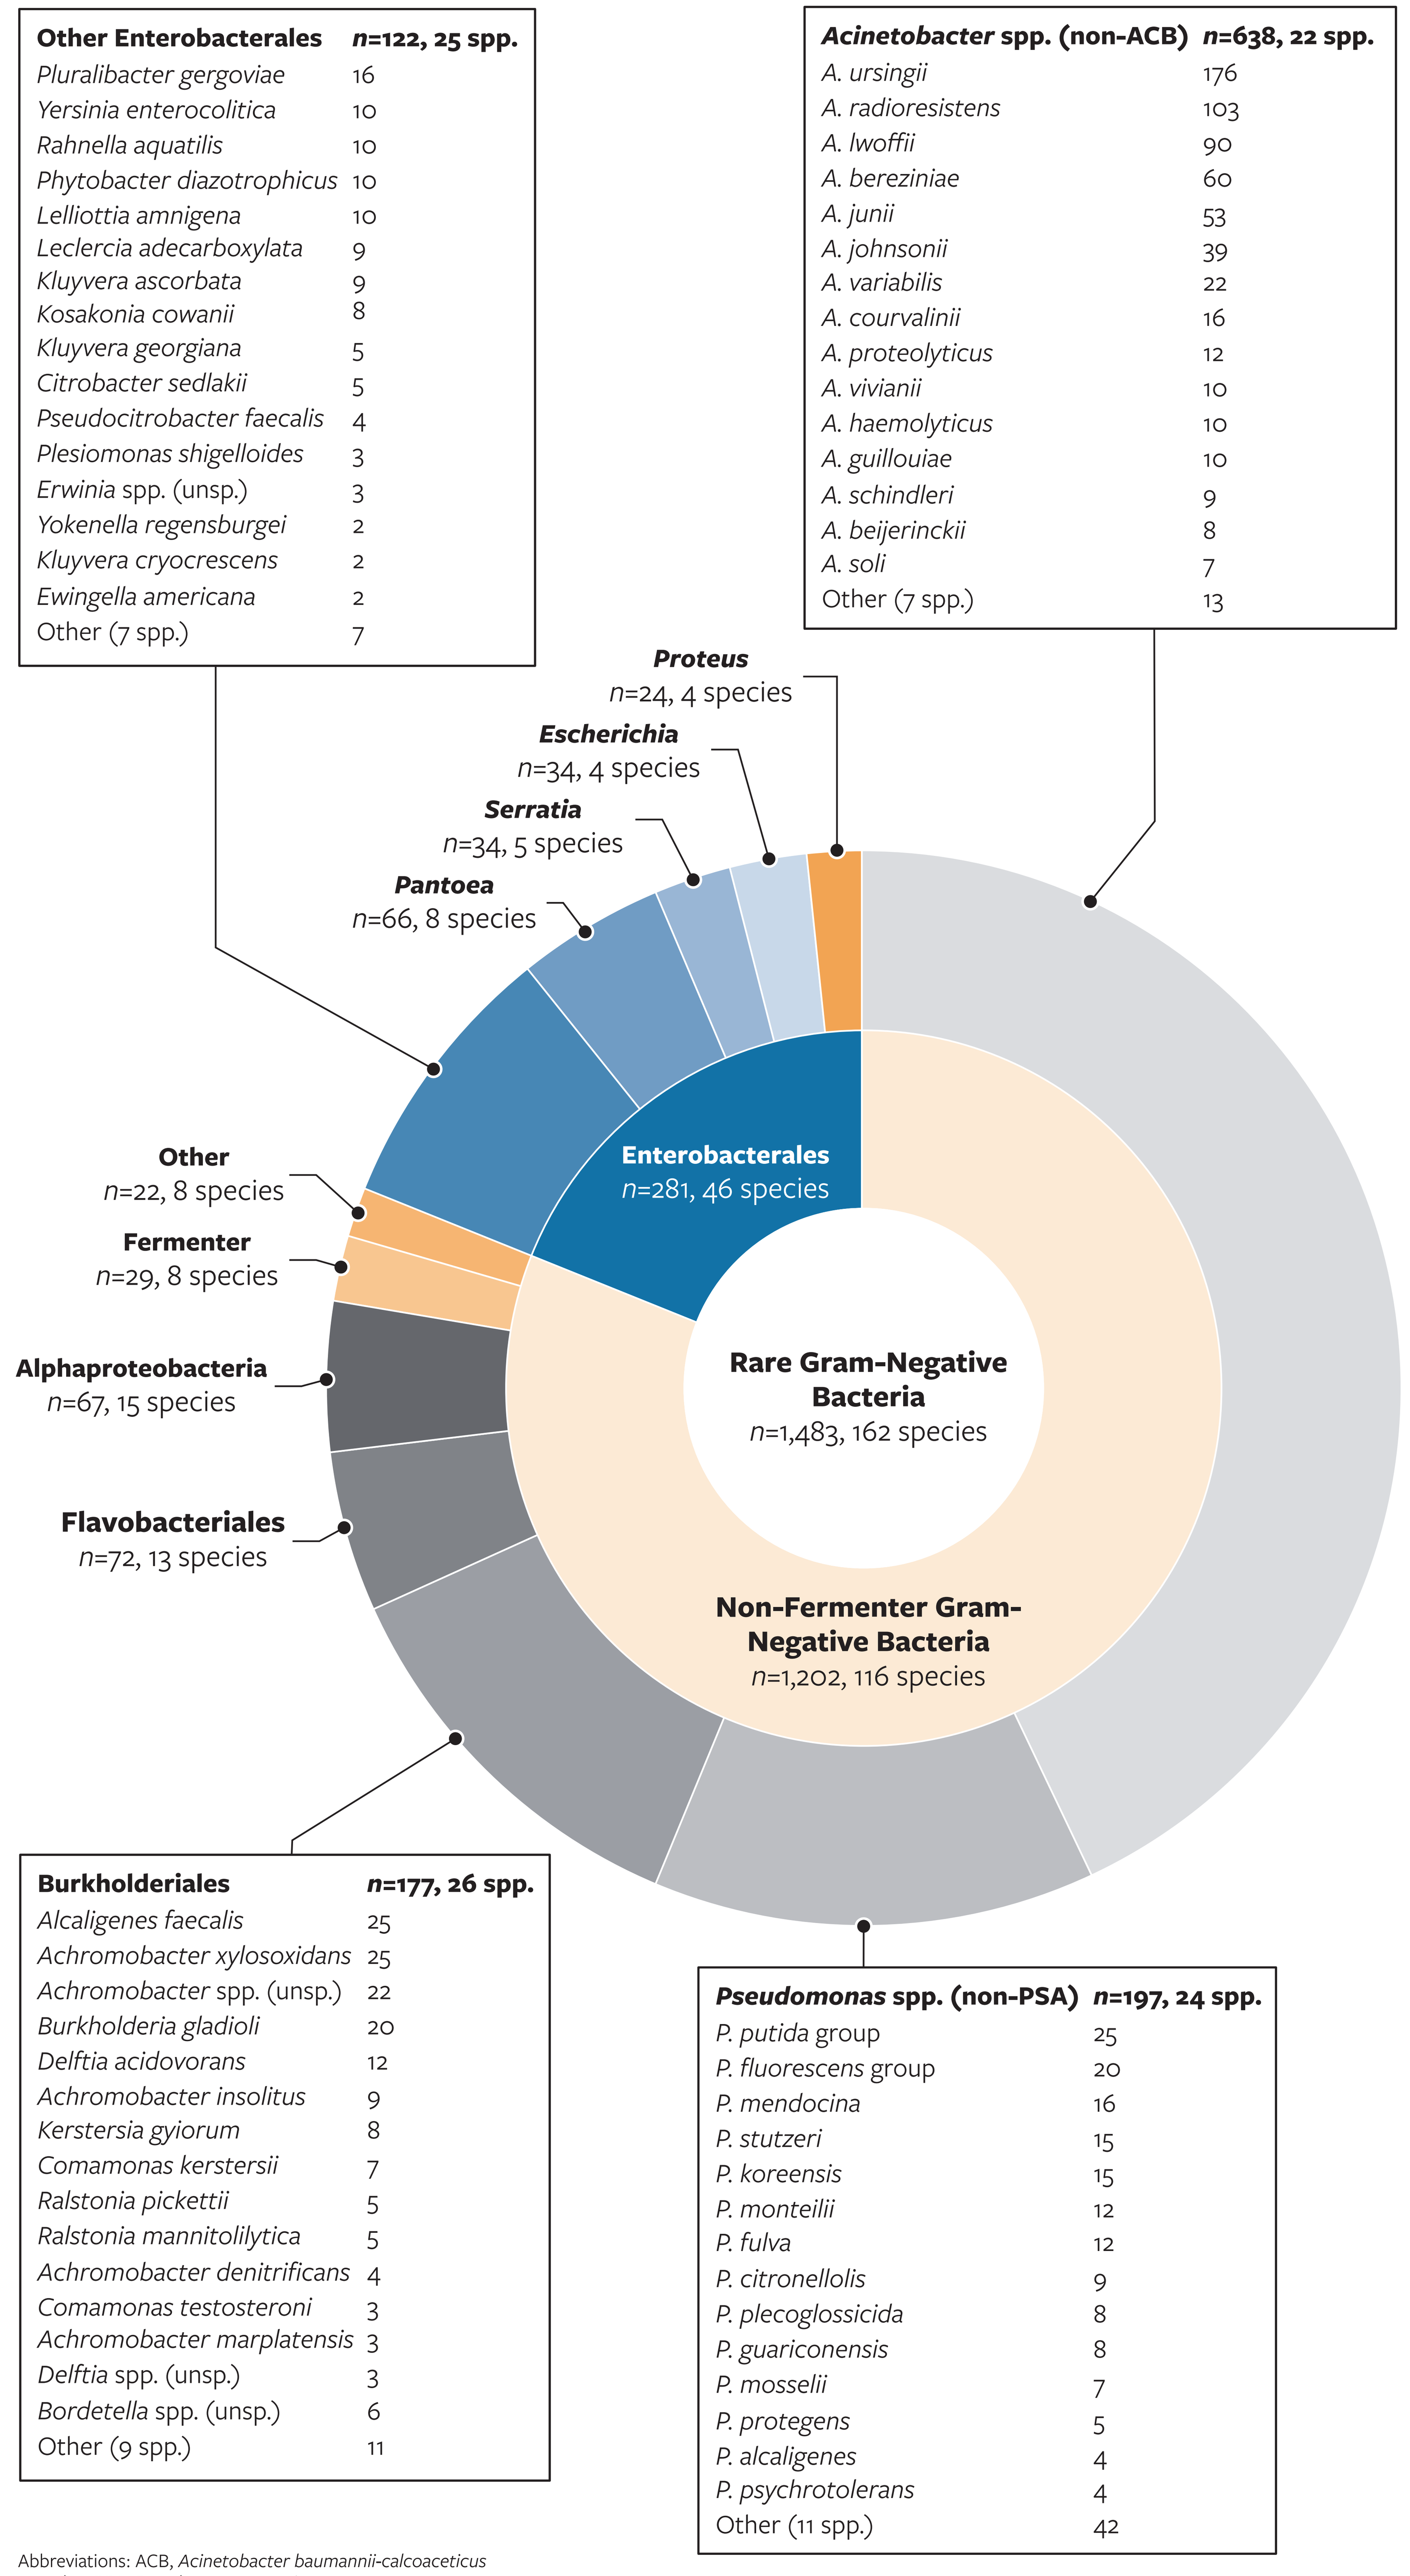

Other Enterobacterales n=122, 25 spp.
Pluralibacter gergoviae 16
Yersinia enterocolitica 10
Rahnella aquatilis 10
Phytobacter diazotrophicus 10
Lelliottia amnigena 10
Leclercia adecarboxylata 9
Kluyvera ascorbata 9
Kosakonia cowanii 8
Kluyvera georgiana 5
Citrobacter sedlakii 5
Pseudocitrobacter faecalis 4
Plesiomonas shigelloides 3
Erwinia spp. (unsp.) 3
Yokenella regensburgei 2
Kluyvera cryocrescens 2
Ewingella americana 2
Other (7 spp.) 7
Acinetobacter spp. (non-ACB) n=638, 22 spp.
A. ursingii 176
A. radioresistens 103
A. lwoffii 90
A. bereziniae 60
A. junii 53
A. johnsonii 39
A. variabilis 22
A. courvalinii 16
A. proteolyticus 12
A. vivianii 10
A. haemolyticus 10
A. guillouiae 10
A. schindleri 9
A. beijerinckii 8
A. soli 7
Other (7 spp.) 13
Proteus n=24, 4 species
Escherichia n=34, 4 species
Serratia n=34, 5 species
Pantoea n=66, 8 species
Other n=22, 8 species
Fermenter n=29, 8 species
Alphaproteobacteria n=67, 15 species
Flavobacteriales n=72, 13 species
Enterobacterales n=281, 46 species
Rare Gram-Negative Bacteria n=1,483, 162 species
Non-Fermenter Gram-Negative Bacteria n=1,202, 116 species
Burkholderiales n=177, 26 spp.
Alcaligenes faecalis 25
Achromobacter xylosoxidans 25
Achromobacter spp. (unsp.) 22
Burkholderia gladioli 20
Delftia acidovorans 12
Achromobacter insolitus 9
Kerstersia gyiorum 8
Comamonas kerstersii 7
Ralstonia pickettii 5
Ralstonia mannitolilytica 5
Achromobacter denitrificans 4
Comamonas testosteroni 3
Achromobacter marplatensis 3
Delftia spp. (unsp.) 3
Bordetella spp. (unsp.) 6
Other (9 spp.) 11
Pseudomonas spp. (non-PSA) n=197, 24 spp.
P. putida group 25
P. fluorescens group 20
P. mendocina 16
P. stutzeri 15
P. koreensis 15
P. monteilii 12
P. fulva 12
P. citronellolis 9
P. plecoglossicida 8
P. guariconensis 8
P. mosselii 7
P. protegens 5
P. alcaligenes 4
P. psychrotolerans 4
Other (11 spp.) 42

Abbreviations: ACB, *Acinetobacter baumannii-calcoaceticus* complex; PSA, *Pseudomonas aeruginosa*.

Table 1. Antimicrobial activity of cefiderocol and comparator agents against the main organisms and organism groups tested

		$MIC_{50/90}$ in mg/L										
Organism/Group	Total	Cefiderocol	Imipenem-Relebactam	Aztreonam	Ceftriaxone	Cefepime	Piperacillin-Tazobactam	Meropenem	Levofloxacin	Amikacin	Ampicillin-Sulbactam	Colistin
Enterobacterales	**281**	**0.06/0.25**	**0.12/0.5**	**≤0.12/0.5**	**≤0.06/2**	**≤0.5/≤0.5**	**1/4**	**≤0.06/≤0.06**	**≤0.12/0.25**	**1/2**	**4/16**	**≤0.5/>8**
Indole-positive *Proteus* spp.	25	0.015/0.03	1/2	≤0.03/0.12	≤0.06/>8	≤0.12/0.5	1/1	0.06/0.12	0.12/0.5	2/8	8/32	>8/>8
***Escherichia* spp. (non-*E. coli*)**	34	0.03/0.06	0.12/0.25	0.06/0.25	≤0.06/0.12	≤0.12/≤0.12	1/4	≤0.015/0.03	≤0.03/0.06	1/4	2/8	0.12/0.25
***Pantoea* spp.**	66	0.12/0.25	0.12/0.5	≤0.12/0.25	≤0.06/0.25	≤0.5/≤0.5	1/2	≤0.06/≤0.06	≤0.12/≤0.12	1/2	4/16	≤0.5/≤0.5
***Serratia* spp.**	34	0.12/0.5	0.25/0.5	≤0.12/2	0.5/4	≤0.5/≤0.5	2/4	≤0.06/≤0.06	≤0.12/0.5	2/2	8/32	>8/>8
Other enterics [a]	122	0.06/0.25	0.12/0.25	≤0.12/1	0.12/1	≤0.5/≤0.5	1/4	≤0.06/≤0.06	≤0.12/0.25	1/2	2/32	0.25/>8
Non-Fermenter	**1202**	**0.06/1**	**0.25/1**	**>16/>16**	**8/>8**	**2/>16**	**1/64**	**0.25/4**	**0.25/2**	**2/>32**	**4/>32**	**≤0.5/>8**
***Acinetobacter* spp. (non-ACB)**	638	0.06/0.5	0.12/0.5	>16/>16	8/>8	1/16	≤0.5/32	0.25/1	≤0.12/0.5	1/4	2/8	≤0.5/2
Flavobacteriales [b]	72	0.25/1	>8/>8	>16/>16	>8/>8	>16/>16	8/64	16/>32	1/2	>32/>32	>32/>32	>8/>8
Elizabethkingia spp.	13	0.5/1	>8/>8	>16/>16	>8/>8	>32/>32	8/16	32/32	1/2	>32/>32	64/>64	>8/>8
Burkholderiales [c]	177	0.03/16	0.5/2	>16/>16	>8/>8	16/>16	≤0.5/32	0.12/16	1/>4	16/>32	8/>32	1/>8
Achromobacter spp.	63	0.015/0.25	1/2	>16/>16	>8/>8	>16/>16	≤0.5/8	0.12/4	4/>4	>32/>32	8/>32	1/2
Burkholderia gladioli	20	4/64	0.25/0.5	>16/>16	8/>8	16/>16	1/2	0.5/1	0.5/1	2/4	4/16	>8/>8
Fermenter non-enteric	29	0.25/8	not tested	≤0.12/>16	2/>8	≤0.5/8	4/>64	0.12/>32	≤0.12/0.5	2/8	>32/>32	1/>8
Aeromonas spp.	25	0.25/4	not tested	≤0.12/>16	2/>8	≤0.5/8	8/>64	0.12/>32	≤0.12/0.5	2/8	>32/>32	≤0.5/>8
***Pseudomonas* spp. (non-PSA)**	197	0.12/0.5	0.5/2	16/>16	8/>8	2/8	8/16	1/8	0.5/2	1/2	>32/>32	≤0.5/4
Alphaproteobacteria [d]	67	1/>64	0.5/1	>16/>16	>8/>8	16/>16	>64/>64	0.25/4	0.25/4	8/16	16/>32	>8/>8
Other non-fermenters [e]	22	0.008/1	not tested	0.5/>16	0.12/>8	≤0.5/8	≤0.5/64	0.25/2	0.25/4	2/>32	4/32	8/>8

Abbreviations: ACB, *Acinetobacter baumannii-calcoaceticus* complex; PSA, *Pseudomonas aeruginosa*.
[a] Other enterics include the following genera: *Cedecea, Citrobacter, Edwardsiella, Erwinia, Ewingella, Kluyvera, Kosakonia, Leclercia, Lelliottia, Metakosakonia, Phytobacter, Plesiomonas, Pluralibacter, Pseudocitrobacter, Rahnella, Rouxiella, Scandinavium,* and *Yersinia.*
[b] Members of the order Flavobacteriales include the following genera: *Chryseobacterium, Elizabethkingia, Empedobacter,* and *Myroides.*
[c] Members of the order Burkholderiales include the following genera: *Achromobacter, Alcaligenes, Bordetella, Burkholderia, Comamonas, Delftia, Herbaspirillum, Kerstersia, Pandoraea,* and *Ralstonia.*
[d] Members of the class Alphaproteobacteria include the following genera: *Agrobacterium, Brevundimonas, Ochrobactrum, Paracoccus, Rhizobium, Roseomonas, Sphingobium,* and *Sphingomonas.*
[e] Other non-fermenters include: *Oligella, Shewanella, Stenotrophomonas,* and *Sphingobacterium.*

Table 2. Susceptibility rates of cefiderocol and comparator agents against main organisms and organism groups

		(% susceptible per CLSI breakpoint criteria)										
Organism/Group	Total	Cefiderocol	Imipenem-Relebactam	Aztreonam	Ceftriaxone	Cefepime	Piperacillin-Tazobactam	Meropenem	Levofloxacin	Amikacin	Ampicillin-Sulbactam	Colistin [a]
Enterobacterales	**281**	**100.0**	**96.0**	**98.6**	**88.9**	**99.3**	**99.3**	**100.0**	**98.6**	**97.2**	**79.7**	**69.6**
Indole-positive *Proteus* spp.	25	100.0	57.1	100.0	68.0	96.0	100.0	100.0	96.0	88.0	56.0	4.0
Escherichia spp. (non-*E. coli*)	34	100.0	100.0	97.1	97.1	97.1	97.1	100.0	97.0	100.0	91.2	94.1
Pantoea spp.	66	100.0	100.0	100.0	100.0	100.0	98.5	100.0	98.5	100.0	83.3	93.8
Serratia spp.	34	100.0	96.0	97.1	70.6	100.0	100.0	100.0	97.1	97.1	70.6	8.8
Other enterics [b]	122	100.0	100.0	98.4	90.0	100.0	100.0	100.0	100.0	96.7	82.0	80.3
Non-Fermenter	**923**	98.5 [c]	—	—	—	—	—	—	—	—	—	—
Acinetobacter spp. (non-ACB)	638	99.1 [c]	—	—	73.0	87.5	89.5	98.1	98.9	95.9	93.1	92.5
Achromobacter spp.	63	98.4 [c]	—	0.0	1.8	23.8	92.1	92.1	47.6	12.7	—	—
Aeromonas spp.	25	88.0 [c]	—	84.0	38.9	84.0	64.0	84.0	92.0	96.0	—	—
Pseudomonas spp. (non-PSA) [d]	197	98.0 [c]	—	54.1	—	92.9	90.9	70.4	92.9	99.0	—	93.9

% Susceptible

≥95.0%	94.9–90.0%	89.9–60%	59.9%–40%	39.9–0%

Abbreviations: ACB, *Acinetobacter baumannii-calcoaceticus* complex; PSA, *Pseudomonas aeruginosa*.
[a] Colistin MIC results were interpreted according to CLSI criteria, and % value represents % intermediate.
[b] Other Enterobacterales include the following genera: *Cedecea, Citrobacter, Edwardsiella, Erwinia, Ewingella, Kluyvera, Kosakonia, Leclercia, Lelliottia, Metakosakonia, Phytobacter, Plesiomonas, Pluralibacter, Pseudocitrobacter, Rahnella, Rouxiella, Scandinavium,* and *Yersinia.*
[c] % susceptible per EUCAST Gram-negative PK-PD cut-off (≤2 mg/L).
[d] % intermediate (interpreted as susceptible, increased exposure) per EUCAST *Pseudomonas* spp. breakpoint criteria for all agents excluding meropenem (% susceptible).

Contact

element

Joshua Maher, PhD
Element Materials Technology/JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317 USA
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: joshua.maher@element.com

Scan Me

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2026_25-SHI-08_P1_Rare_GN.pdf

Charges may apply. No personal information is stored.

IDWeek 2026, October 21–24, 2026, Washington, D.C.