IDWEEK 2026 | Poster #P-117

Aztreonam-avibactam activity against Enterobacterales from United States medical centers: Summary of 5 years of surveillance prior to approval for clinical use (2020–2024)

Helio S. Sader[1], Rodrigo E. Mendes[1], Dmitri Debabov[2], John H. Kimbrough[1], Krisztina M. Papp-Wallace[1], Mariana Castanheira[1]

[1] *Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA;* [2] *AbbVie Inc., Chicago, Illinois, USA*

CONCLUSIONS

ATM-AVI demonstrated almost complete activity against Enterobacterales and retained potent activity against CRE, including MBL producers.

Importantly, ATM-AVI activity was not adversely affected by clinically relevant carbapenemases or resistance to agents used to treat multidrug-resistant Enterobacterales.

The activities of CAZ-AVI, MEM-VAB, and IMI-REL were compromised by the increased occurrence of MBL producers among CRE isolates from US medical centers.

These results provide a valuable benchmark for monitoring the *in vitro* activity of ATM-AVI after regulatory approval and emphasize the importance of comprehensive surveillance programs to monitor the emergence of resistance mechanisms to newly approved antimicrobial agents.

INTRODUCTION

- Aztreonam-avibactam (ATM-AVI) was approved by the United States (US) Food and Drug Administration (FDA) to treat intra-abdominal infections in February 2025.
- We evaluated the *in vitro* activity of ATM-AVI against Enterobacterales from US hospitals during the 5-year period prior to its approval for clinical use.

METHODS

- A total of 42,225 isolates (1/patient) were consecutively collected in 2020–2024 from 85 US medical centers.
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of infection were included in the study.
- Isolates were susceptibility tested by Clinical and Laboratory Standards Institute (CLSI) M07 broth microdilution methods at a monitoring laboratory.
- Susceptibility results were stratified by year.
- Carbapenem-resistant Enterobacterales (CRE; *n*=406 [1.0%]) and isolates with elevated ATM-AVI MICs (>4 mg/L) were molecularly characterized.

RESULTS

- ATM-AVI was active against 99.9% of Enterobacterales and exhibited potent activity against CRE ($MIC_{50/90}$, 0.25/1 mg/L; 97.0% susceptible [S]; Table 1).
- Ceftazidime-avibactam (CAZ-AVI), meropenem-vaborbactam (MEM-VAB), and imipenem-relebactam (IMI-REL) were very active against Enterobacterales overall (93.8–99.7% S) but showed moderate activity against CRE (67.5–76.1% S; Table 1).
- CRE susceptibility to CAZ-AVI decreased from 89.2% in 2020 to 69.1% in 2024 (Table 1 and Figure 1).
- CRE susceptibility to MEM-VAB decreased from 83.1% in 2020 to 66.0% in 2024 (Table 1 and Figure 1).
- Cefiderocol ($MIC_{50/90}$, 1/4 mg/L) was active against 94.1% of CREs (Table 1).
- The most common carbapenemases (CBase) among CREs were KPC (52.7% of CREs), NDM (21.2%), and OXA-48-like (4.4%).
- A CBase was not identified in 20.7% of CREs.
- The frequency of KPC decreased from 67.5% in 2020 to 39.4% in 2024 (Figure 2).
- The frequency of MBL ranged from 12.0% in 2020 to 43.8% in 2023 (23.4% overall; Figure 2).
- ATM-AVI, CAZ-AVI, MEM-VAB, IMI-REL, and cefiderocol were very active (≥98.1% S) against KPC producers (Table 2).
- Only ATM-AVI (97.9% S) and cefiderocol (84.2% S) showed good activity against MBL producers (Table 2).
- Only 27 isolates (<0.1%) had ATM-AVI MIC >4 mg/L; 55.6% and 66.7% of them were meropenem-S and CAZ-AVI-S, respectively.
- Decreased susceptibility to ATM-AVI was mainly due to PBP3 alterations and production of CMY and/or CTX-M among *E. coli*, and porin alterations plus β-lactamase production in *E. cloacae* and *Klebsiella* spp.

Table 1. Yearly susceptibility of CRE isolates

Antimicrobial agent	% Susceptible per CLSI and/or US FDA (no. of isolates)					
	2020 (83)	2021 (98)	2022 (67)	2023 (64)	2024 (94)	All years (406)
Aztreonam-avibactam	100.0	96.9	97.0	100.0	92.6	97.0
Ceftazidime-avibactam	89.2	78.6	83.6	57.8	69.1	76.1
Meropenem-vaborbactam	83.1	76.5	80.6	53.1	66.0	72.4
Imipenem-relebactam	77.1	70.4	74.6	48.4	63.8	67.5
Cefiderocol	92.8	92.9	95.5	93.8	95.7	94.1
Amikacin	55.4	59.2	79.1	64.1	70.2	65.0
Gentamicin	51.8	55.1	76.1	70.3	63.8	62.3
Levofloxacin	25.3	28.6	41.8	31.2	34.0	31.8
Tigecycline	94.0	91.8	98.5	96.9	97.9	95.6
Colistin	84.3	84.7	83.6	78.1	87.2	84.0

Table 2. Antimicrobial susceptibility of CRE isolates stratified by carbapenemase type

Antimicrobial agent	% Susceptible per CLSI and/or US FDA (no. of isolates)				
	All CRE (406)[a]	KPC producers (206)[a]	MBL producers (95)	Other carbapenemases (21)[a,b]	No carbapenemase identified (84)
Aztreonam-avibactam	97.0	100.0	97.9	95.2	89.3
Ceftazidime-avibactam	76.1	99.5	6.3	95.2	92.9
Meropenem-vaborbactam	72.4	99.5	10.5	38.1	84.5
Imipenem-relebactam	67.5	98.1	2.1	4.8	82.1
Cefiderocol	94.1	99.0	84.2	95.2	92.9
Amikacin	65.0	60.7	55.8	57.1	88.1
Gentamicin	62.3	51.9	66.3	61.9	83.3
Levofloxacin	31.8	27.2	18.9	42.9	54.8
Tigecycline	95.6	97.6	94.7	85.7	94.0
Colistin	84.0	83.5	86.3	61.9	88.1

[a] Excludes MBL co-producers
[b] Includes isolates with OXA-48-like (13), SME (7), or IMI (1).

Figure 1. Yearly susceptibility of CRE isolates

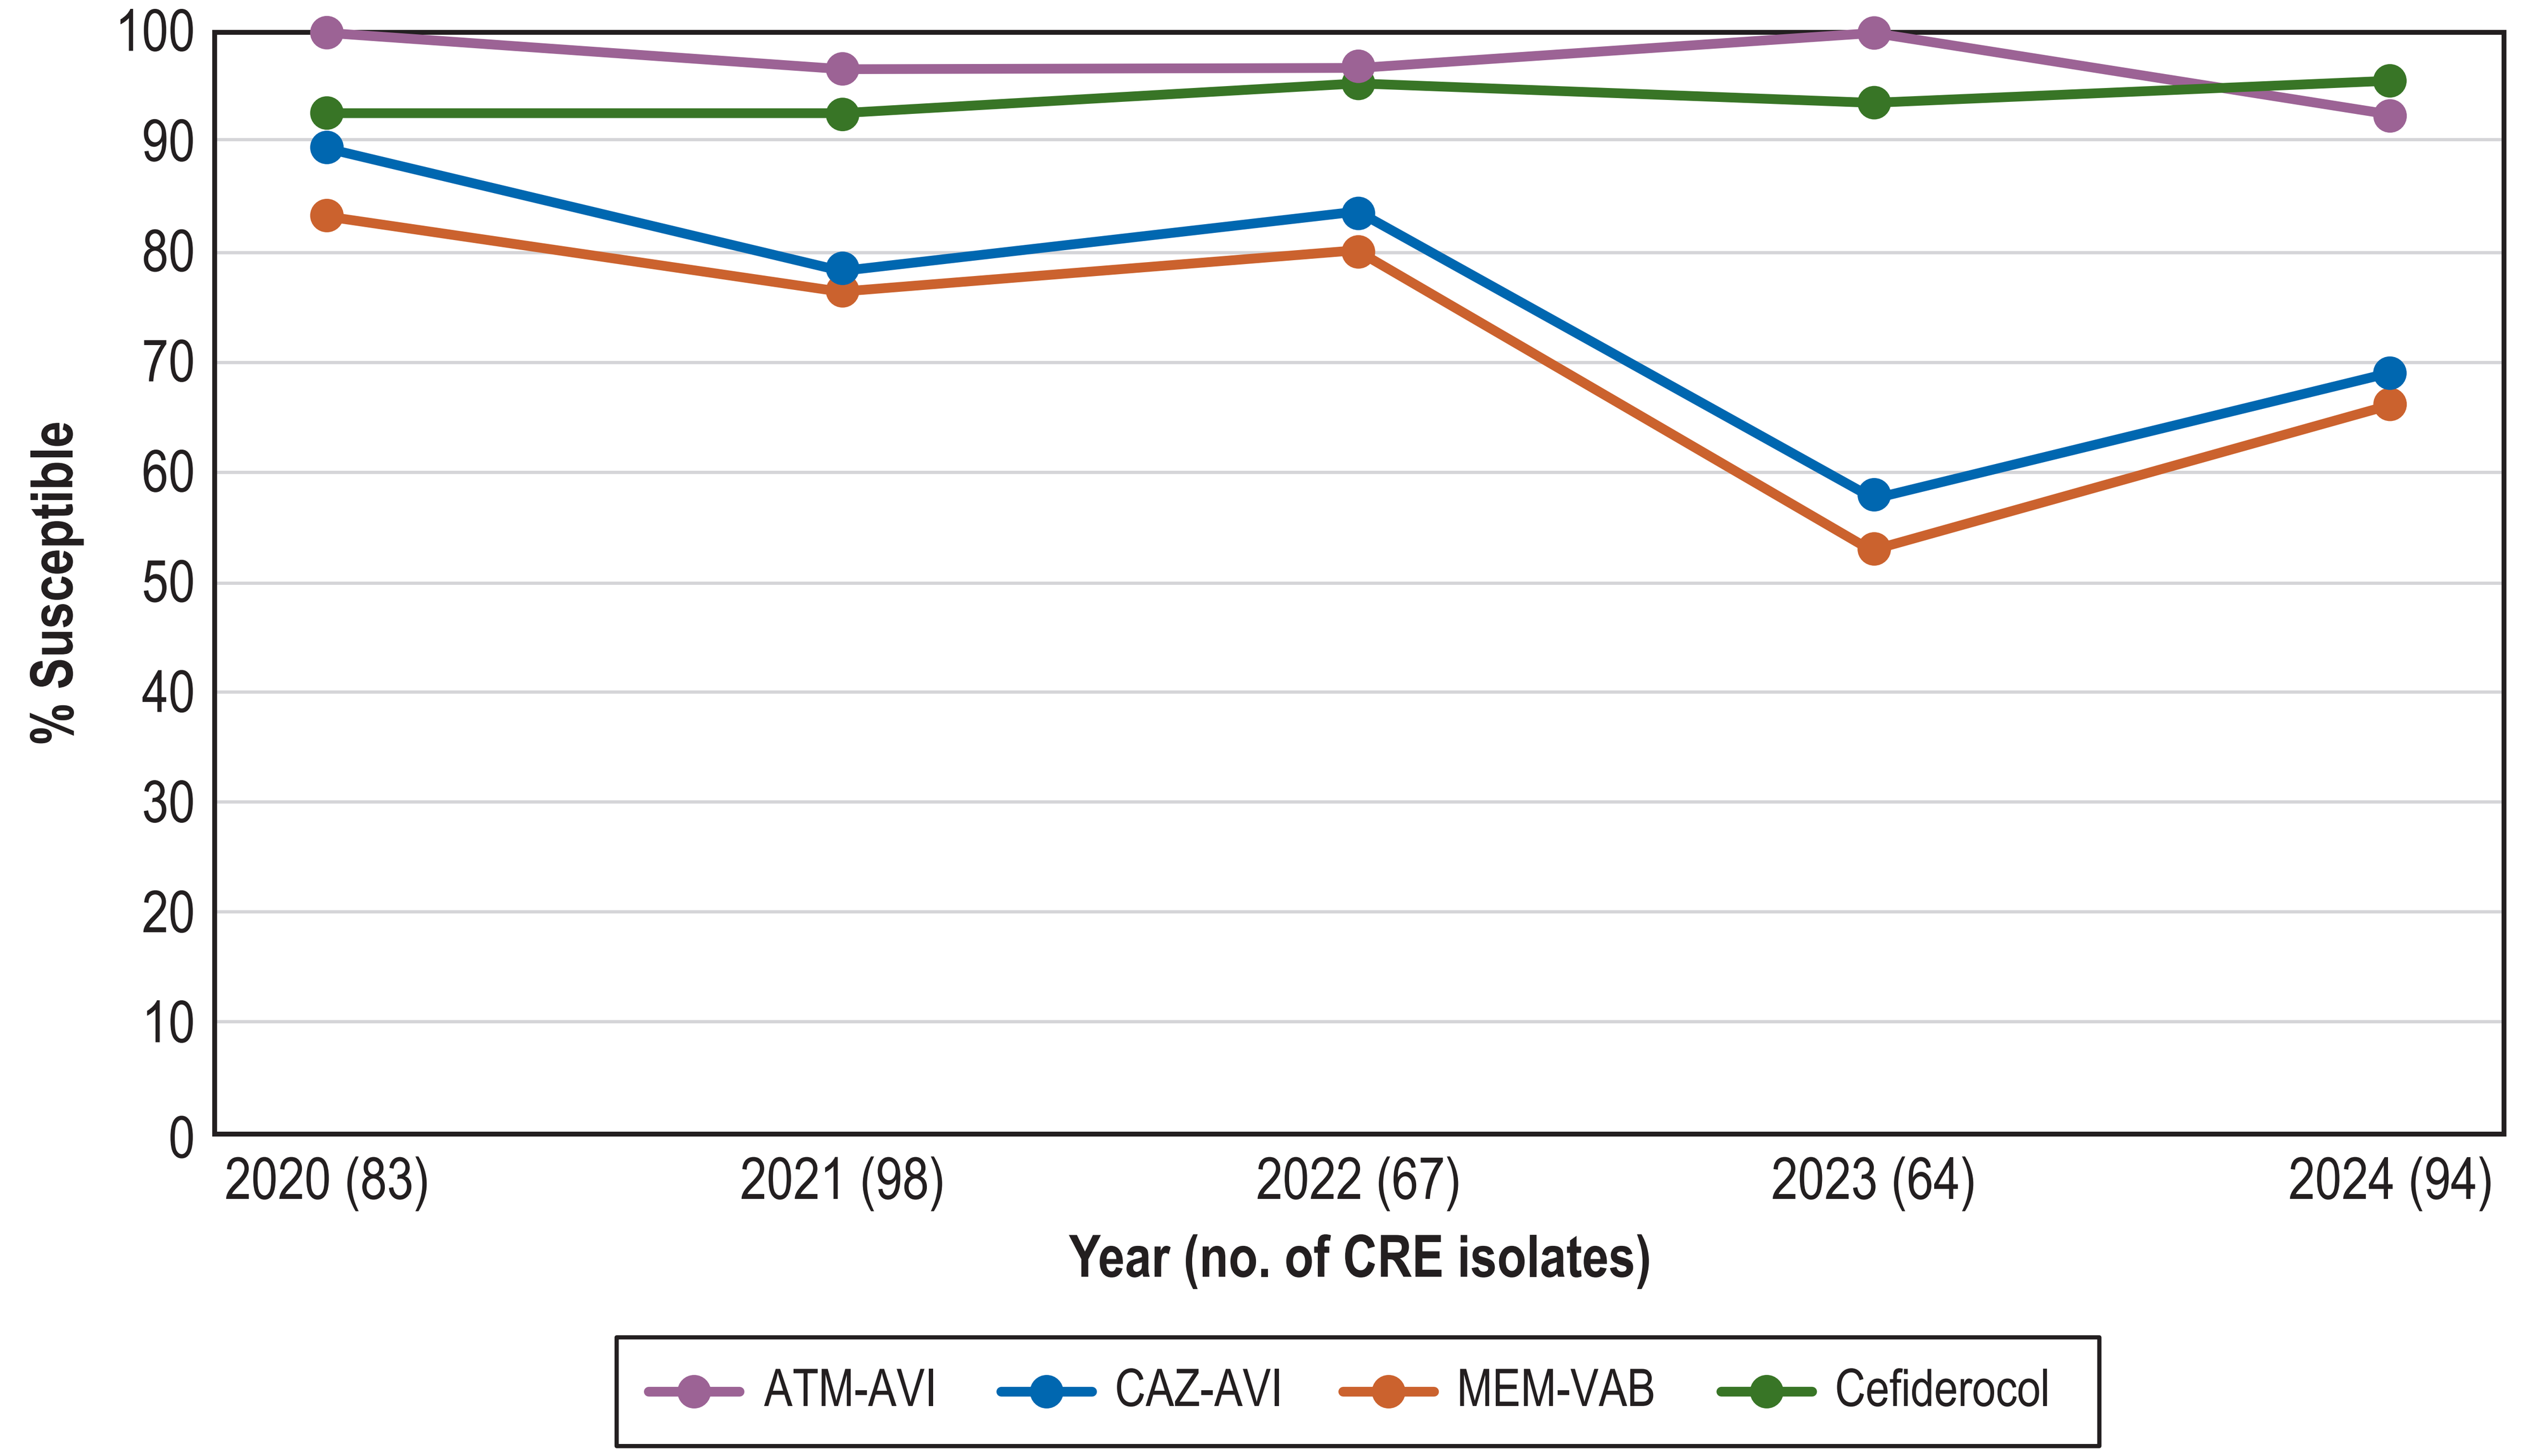
100
90
80
70
60
50
40
30
20
10
0
% Susceptible
2020 (83)
2021 (98)
2022 (67)
2023 (64)
2024 (94)
Year (no. of CRE isolates)
ATM-AVI
CAZ-AVI
MEM-VAB
Cefiderocol

Figure 2. Yearly frequency of carbapenemases (CBases) among CREs

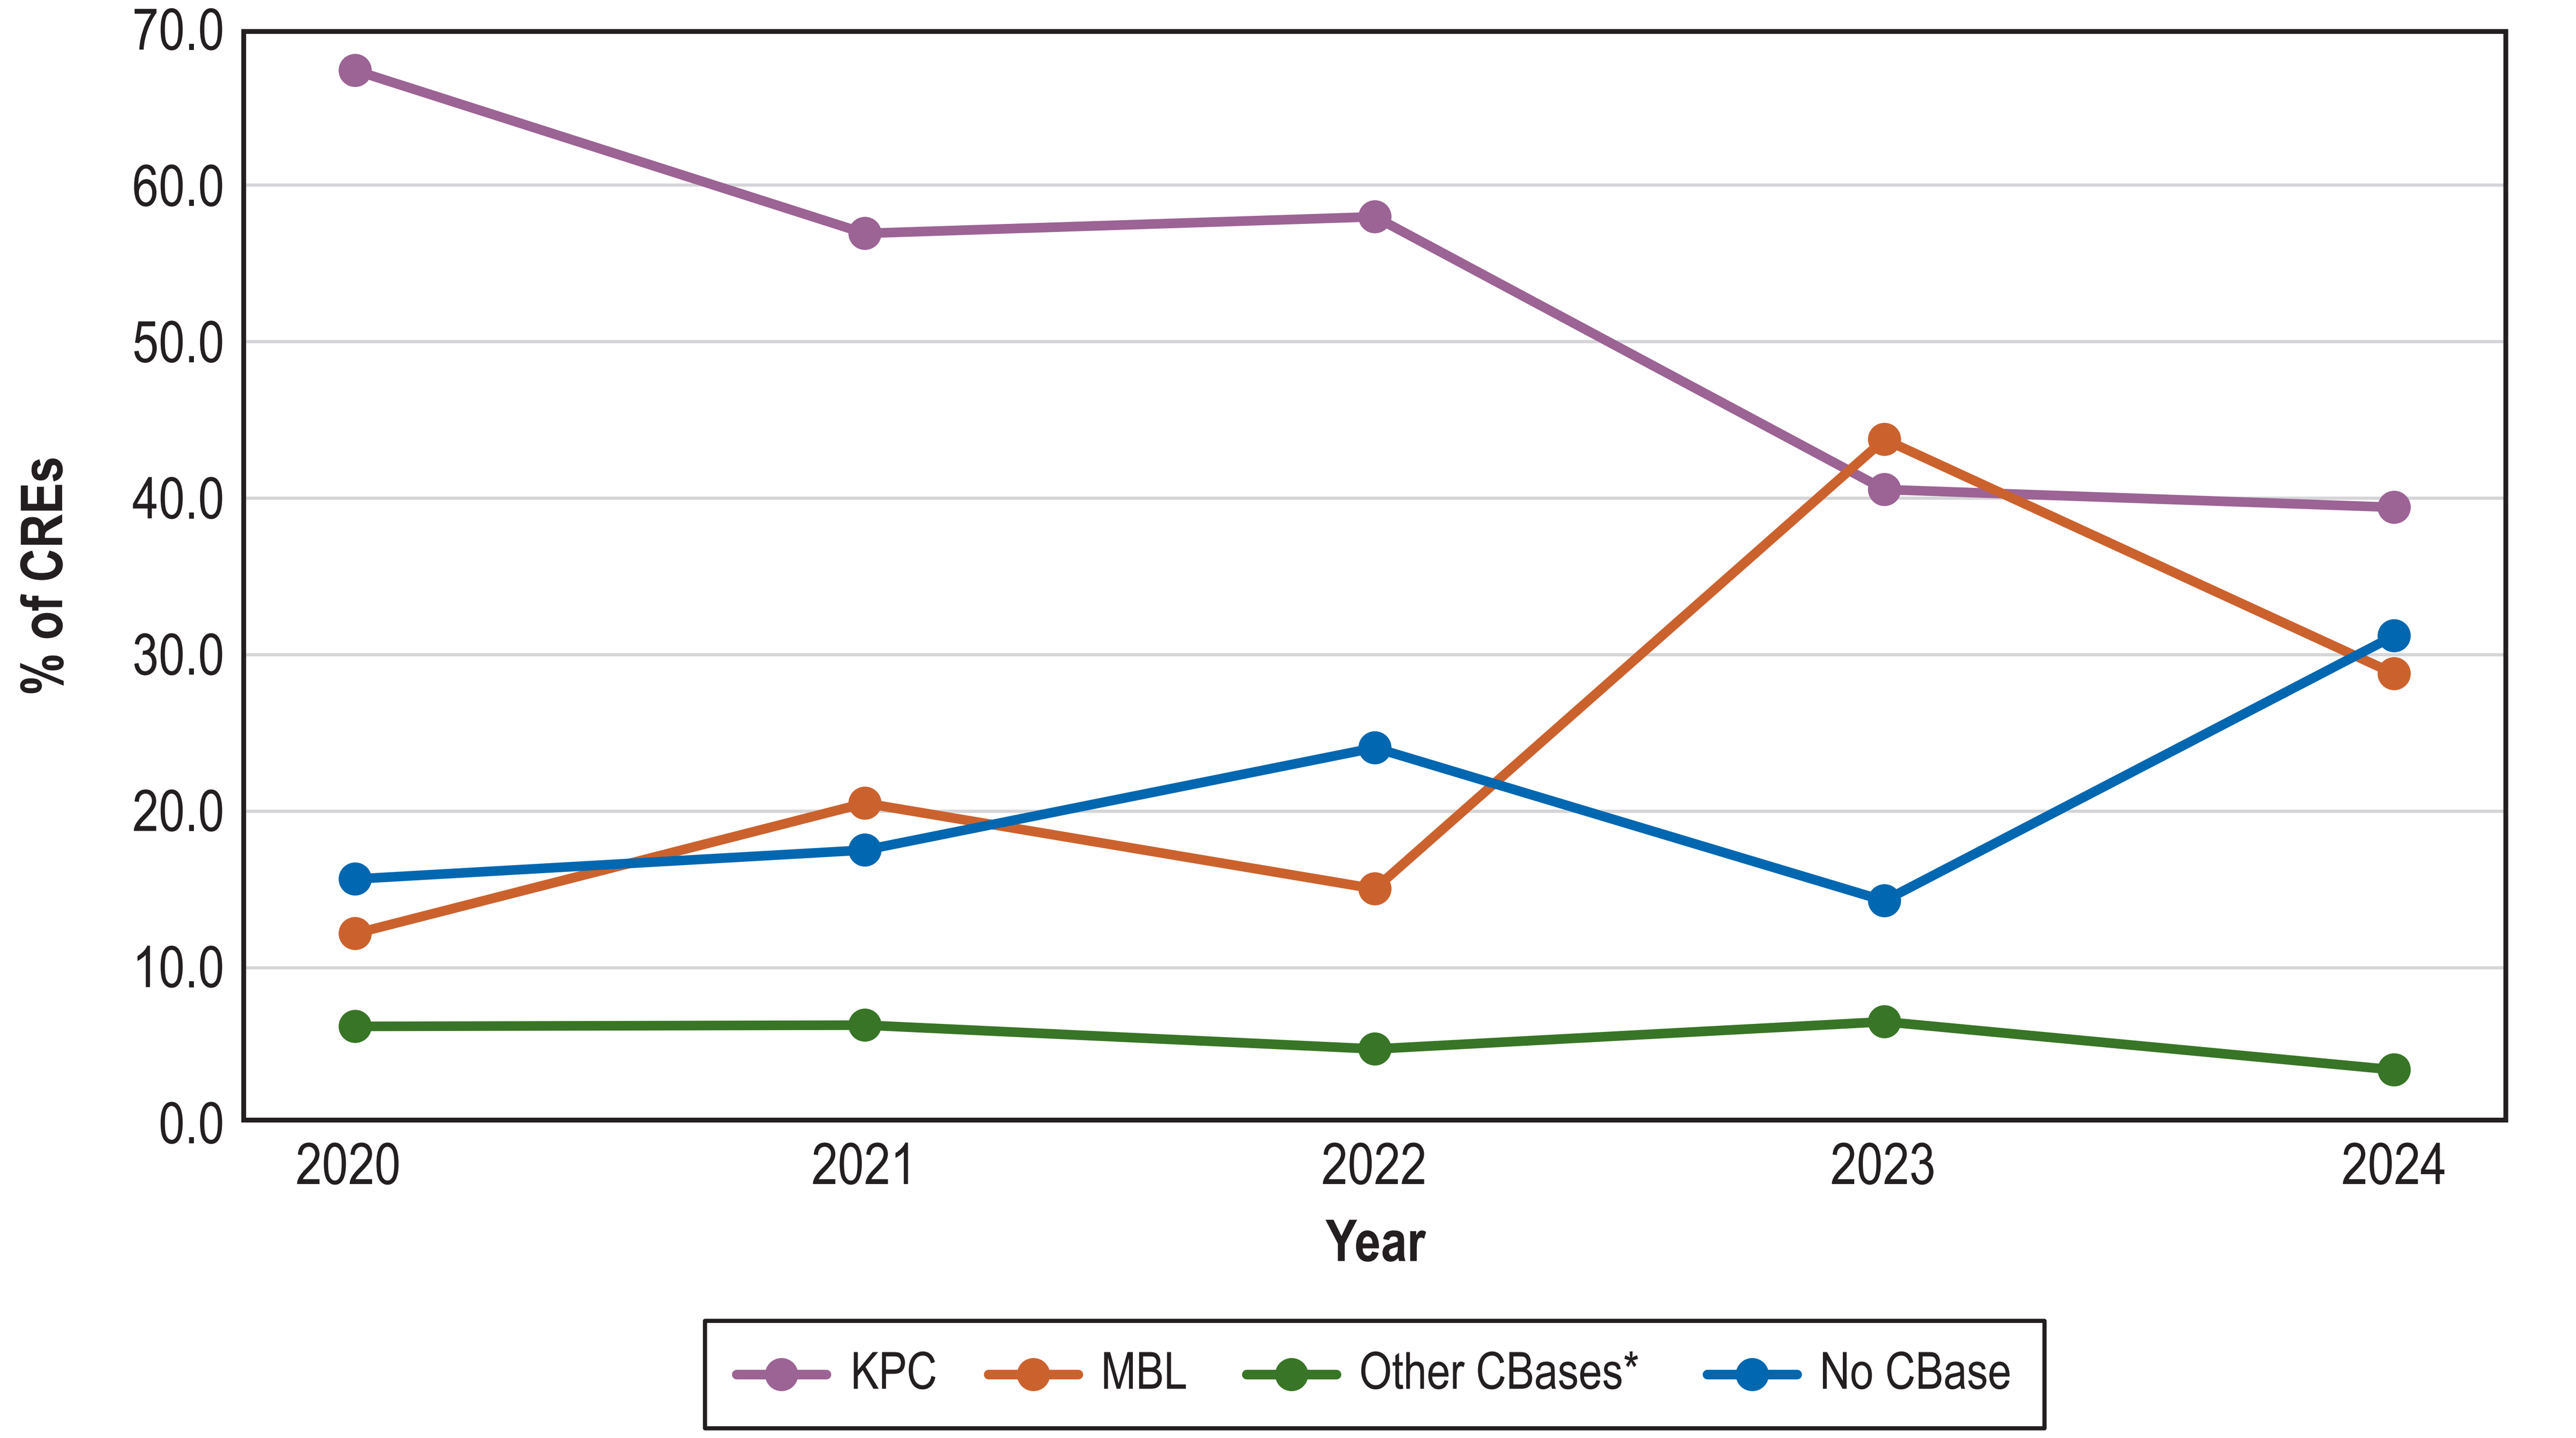
70.0
60.0
50.0
40.0
30.0
20.0
10.0
0.0
% of CREs
2020
2021
2022
2023
2024
Year
KPC
MBL
Other CBases*
No CBase

* Includes isolates with OXA-48-like (13), SME (7), or IMI (1); excludes MBL co-producers.

Contact Information

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2026 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2026_26-ALG-02_P1_USA_5Y.pdf

To submit a medical question, please visit www.abbviemedinfo.com

Scan Me

Acknowledgements

AbbVie funded this study and participated in the study design, research, analysis, data collection, interpretation of data, reviewing, and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship.

References

1. CLSI. 2026. M100Ed35. Performance standards for antimicrobial susceptibility testing: 36th informational supplement. Clinical and Laboratory Standards Institute.
2. Sader HS, Kimbrough JH, Doyle TB et al. Frequency, antimicrobial susceptibility, and molecular characterization of carbapenem-resistant enterobacterales stratified by United States census divisions: Results from the INFORM program (2018-2022). *Open Forum Infect Dis* 2025; 12: ofaf005.
3. Tamma PD, Heil EL, Justo JA *et al.* Infectious Diseases Society of America 2024 guidance on the treatment of antimicrobial-resistant Gram-negative infections. *Clin Infect Dis* 2024 ciae403.

abbvie