IDWEEK 2026 | Poster #P-75

Antimicrobial Susceptibility and Molecular Characterization of Carbapenem-Resistant *Klebsiella pneumoniae* from United States Medical Centers (2020–2025)

Helio S. Sader[1], Mariana Castanheira[1], Todd Riccobene[2], John H. Kimbrough[1], Krisztina M. Papp-Wallace[1]

[1] *Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA;* [2] *AbbVie Inc., Chicago, Illinois, USA*

CONCLUSIONS

- ATM-AVI demonstrated potent and consistent activity against CR-KPN over time.
- The proportion of CR-KPN isolates that produce MBL increased drastically during the study period.
- The activities of CAZ-AVI, MEM-VAB, and IMI-REL decreased markedly during the study period due to the increasing frequency of MBL producers.
- These results emphasize the importance of comprehensive surveillance programs to monitor the epidemiology of resistance mechanisms to antimicrobial agents.

INTRODUCTION

- The frequency of metallo-β-lactamase (MBL)-producing Enterobacterales has been increasing in the United States (US) and effective antibiotics to treat infections caused by these organisms are limited.
- Aztreonam-avibactam (ATM-AVI) was recently (February 2025) approved by the US Food and Drug Administration (FDA) and has shown potent activity against MBL-producing Enterobacterales.
- We evaluated the antimicrobial susceptibility and carbapenemase (CBase) production of carbapenem-resistant *K. pneumoniae* (CR-KPN) isolates from US hospitals.

METHODS

- A total of 10,254 *K. pneumoniae* isolates were consecutively collected from 86 US medical centers in 2020–2025.
- Among those, 298 (2.9%) were CR-KPN, defined as resistant (MIC ≥4 mg/L) to meropenem and/or imipenem.
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of infection were included in the study.
- Isolates were susceptibility tested by Clinical and Laboratory Standards Institute (CLSI) M07 broth microdilution methods at a monitoring laboratory.
- β-lactamase screening and characterization of resistance mechanisms were performed on all CR-KPN isolates by whole genome sequencing.
- Susceptibility results were stratified by year and CBase type.

Table 1. Antimicrobial susceptibility of CR-KPN from US hospitals (2020–2025)

Antimicrobial agent	mg/L		Susceptibility[a]		
	MIC_{50}	MIC_{90}	%S	%I	%R
Aztreonam-avibactam	0.12	0.5	98.7	0.7	0.7
Ceftazidime-avibactam	1	>32	70.5		29.5
Meropenem-vaborbactam	0.5	>32	63.8	3.7	32.6
Imipenem-relebactam	0.25	>8	61.6 [b]	3.0	35.4
Cefiderocol	1	4	96.6	1.0	2.3
Aztreonam	>16	>16	7.4	1.7	90.9
Ciprofloxacin	>4	>4	17.1	3.7	79.1
Levofloxacin	16	>32	16.8	11.1	72.1
Gentamicin	1	>16	61.1	4.4	34.6
Amikacin	4	>32	58.1	9.1	32.9
Tigecycline	0.5	2	96.3	3.7	0.0
Colistin	0.25	1		92.2	7.8

[a] Based on breakpoint criteria as published by CLSI and/or US FDA
[b] All Enterobacterales species were included in the analysis, but CLSI and EUCAST exclude organisms from the family Morganellaceae.
Abbreviations: CR-KPN, carbapenem-resistant *K. pneumoniae*; S, susceptible; I, intermediate; R, resistant.

Figure 1. Antimicrobial susceptibility of CR-KPN from US hospitals stratified by year

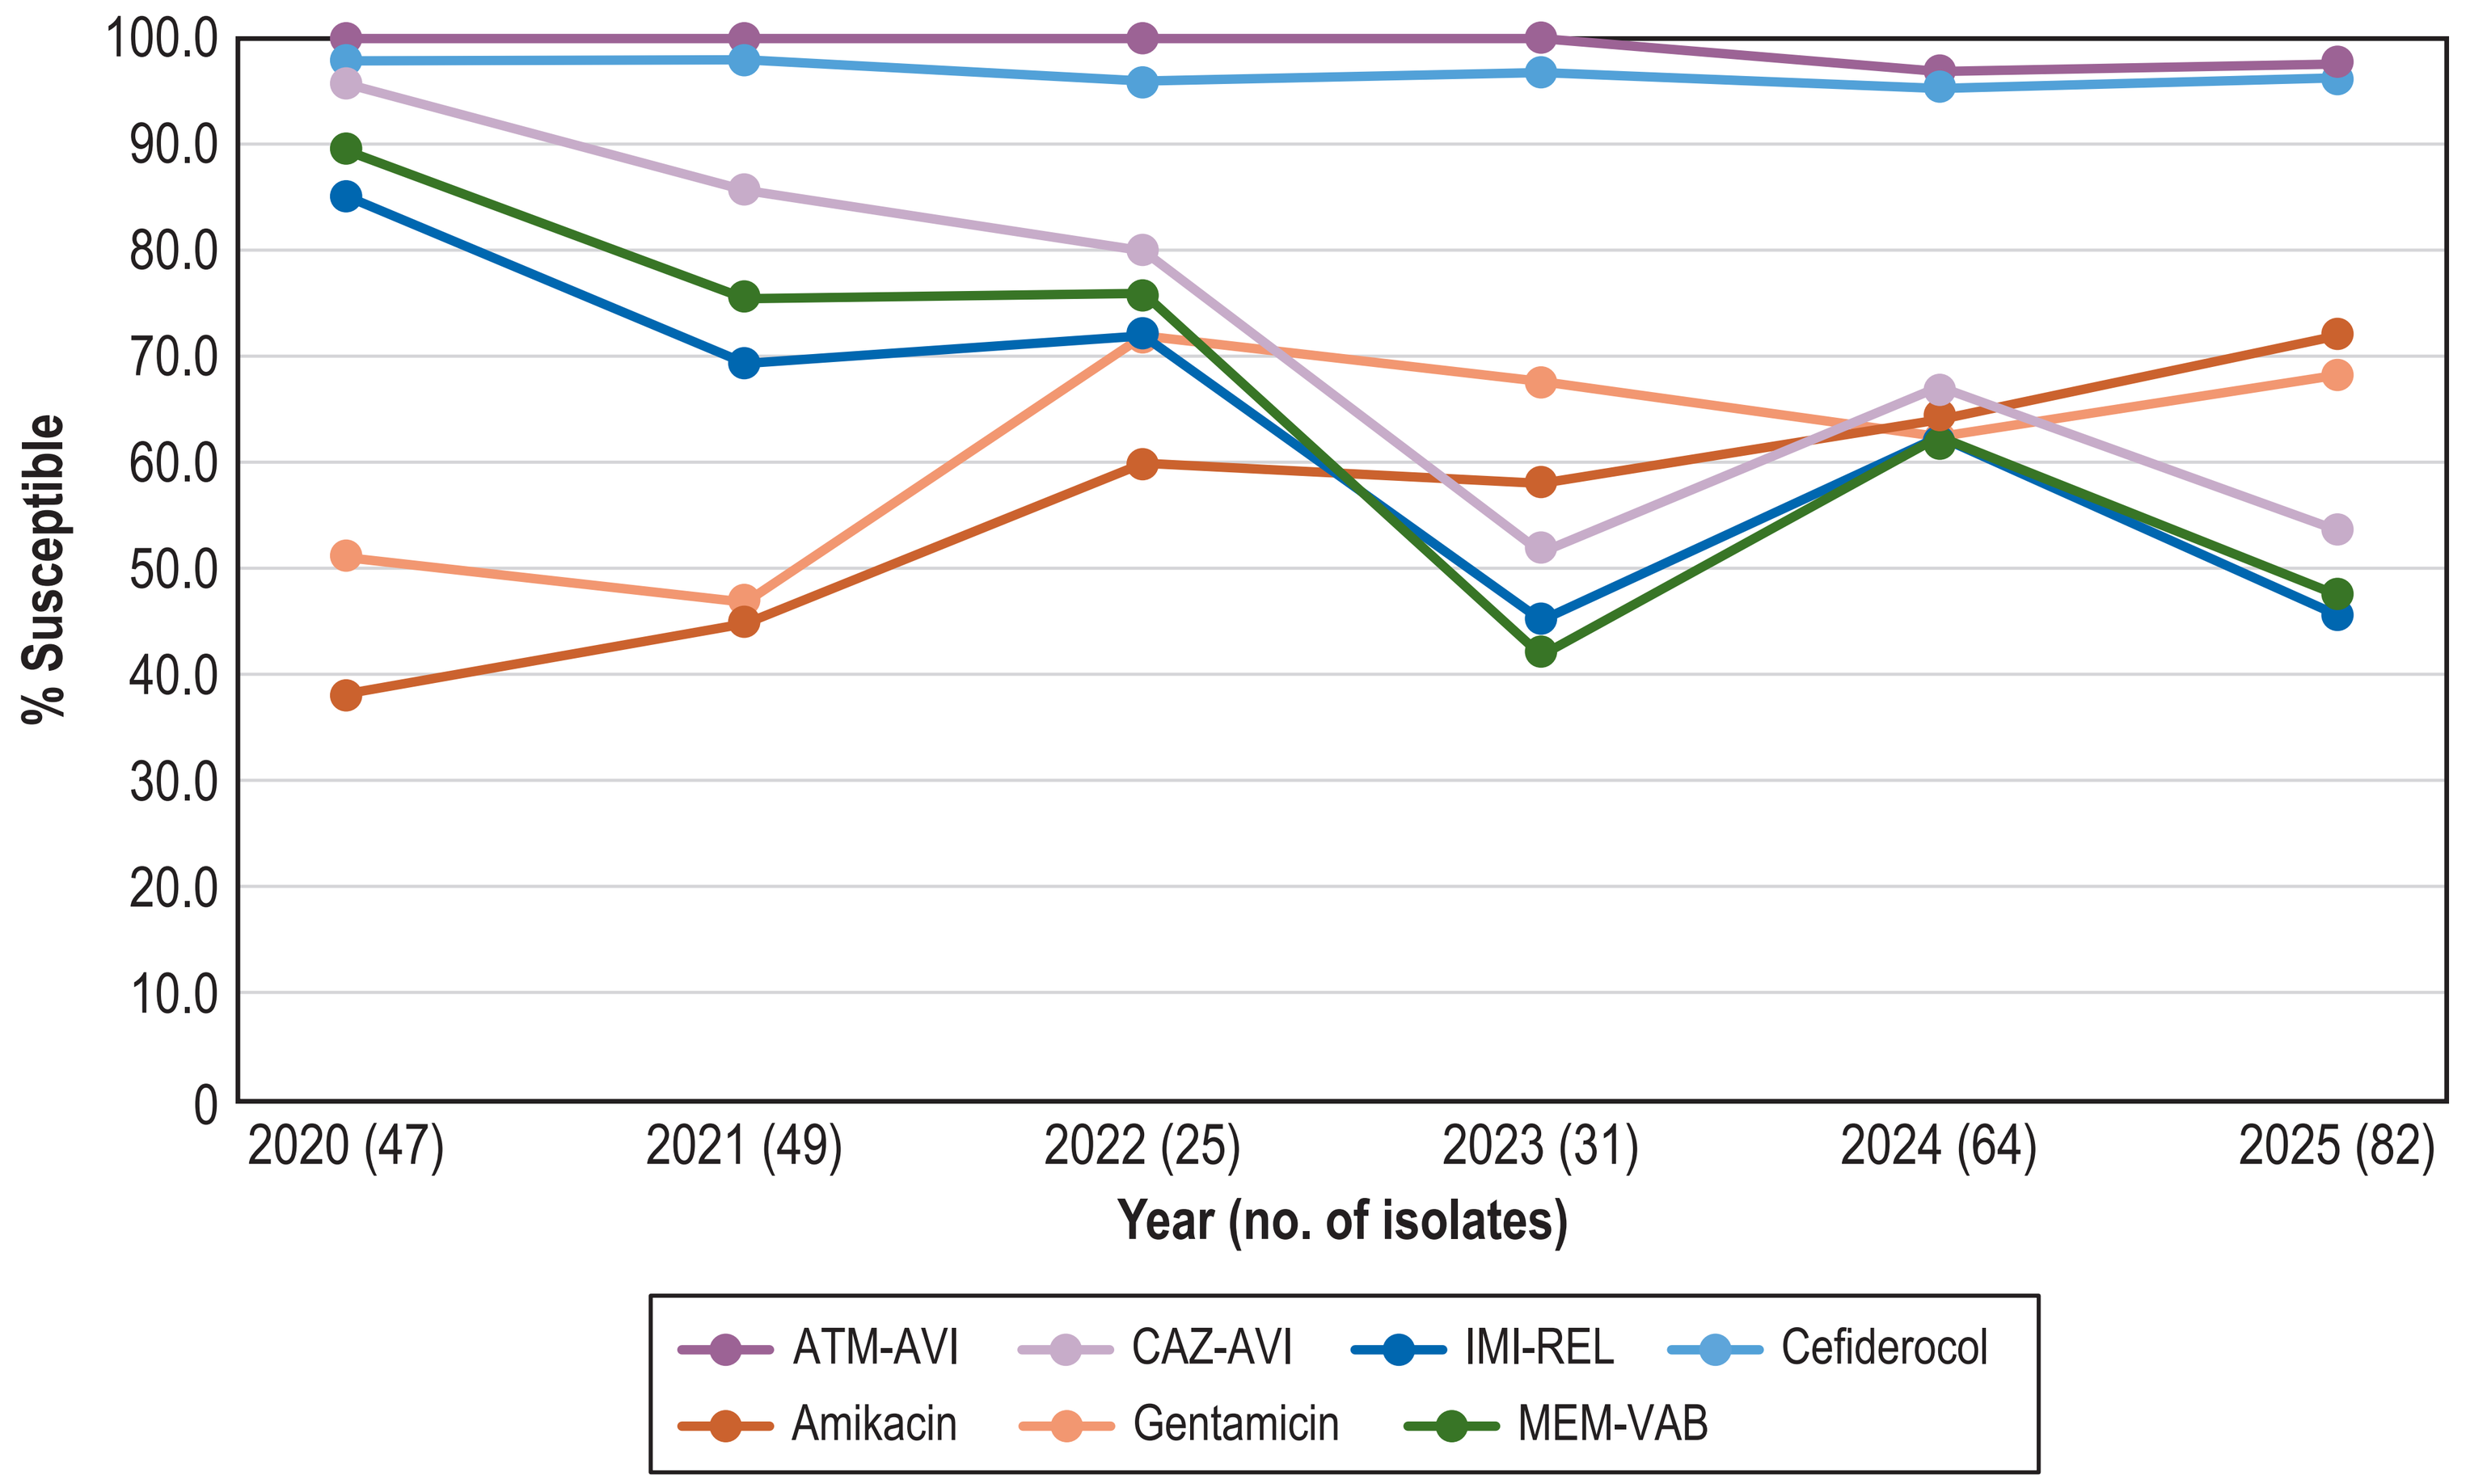

% Susceptible
Year (no. of isolates)
2020 (47)
2021 (49)
2022 (25)
2023 (31)
2024 (64)
2025 (82)
ATM-AVI
CAZ-AVI
IMI-REL
Cefiderocol
Amikacin
Gentamicin
MEM-VAB

RESULTS

- The most active agents against CR-KPN were ATM-AVI ($MIC_{50/90}$, 0.12/0.25; 98.7% susceptible [S]), cefiderocol ($MIC_{50/90}$,1/4 mg/L; 96.6%S), and ceftazidime-avibactam (CAZ-AVI; $MIC_{50/90}$, 1/>32 mg/L; 70.5%S; Table 1).
- ATM-AVI and cefiderocol displayed consistent activity over the years, whereas the activities of CAZ-AVI, meropenem-vaborbactam (MEM-VAB), and imipenem-relebactam (IMI-REL) decreased markedly during the investigation (Figure 1).
- Notably, the activities of amikacin (Figure 1) and gentamicin improved substantially during the same period.
- The proportion of KPC producers among CR-KPN decreased from 78.7% in 2020 to 34.1% in 2025, while the proportion of MBL producers increased from 6.4% to 43.9% in the same period (Figure 2).
- All β-lactamase inhibitor combinations (BLICs) and cefiderocol exhibited potent activity against KPC producers, but only ATM-AVI (100.0%S) and cefiderocol (93.1%S) retained good activity against MBL producers (Figure 3).
- The most common sequence types among CR-KPN were 258 (14.7%), 147 (13.1%), and 45 (7.2%).

Figure 2. Annual frequency of carbapenemase (CBase) types among CR-KPN isolates

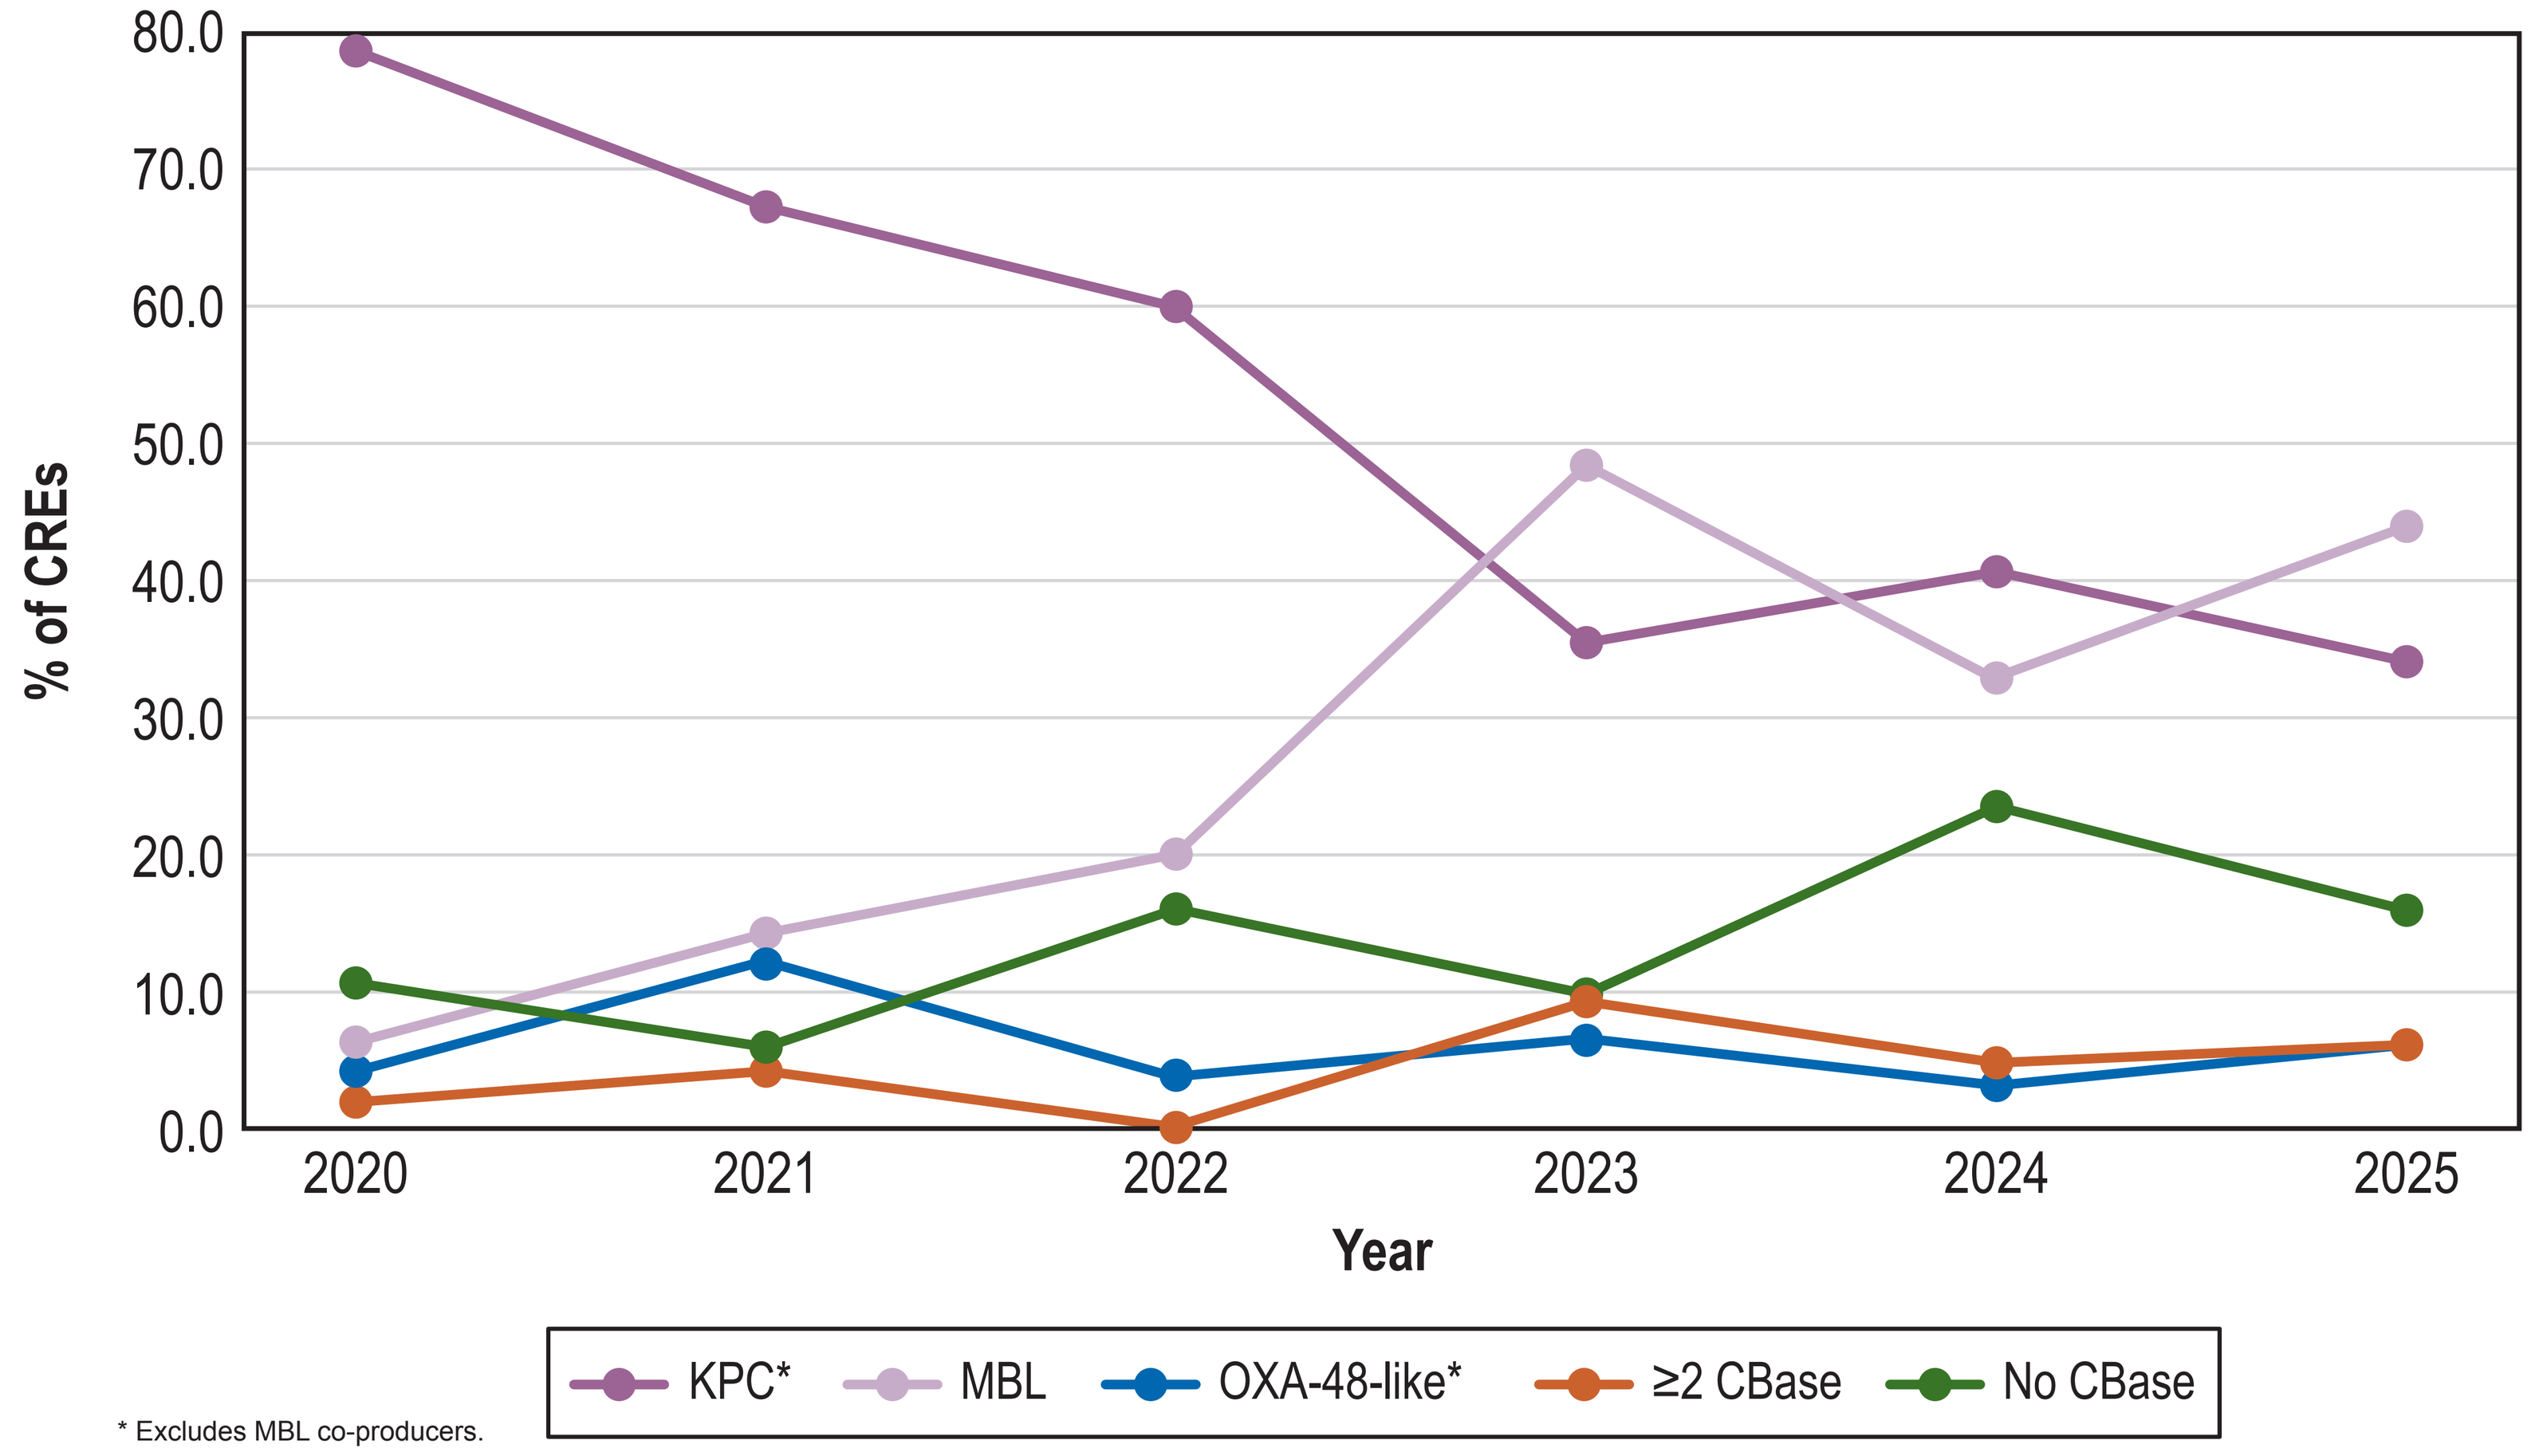

% of CREs
Year
KPC*
MBL
OXA-48-like*
≥2 CBase
No CBase

* Excludes MBL co-producers.

Figure 3. Activity of ATM-AVI and comparators against CR-KPN isolates stratified by carbapenemase type

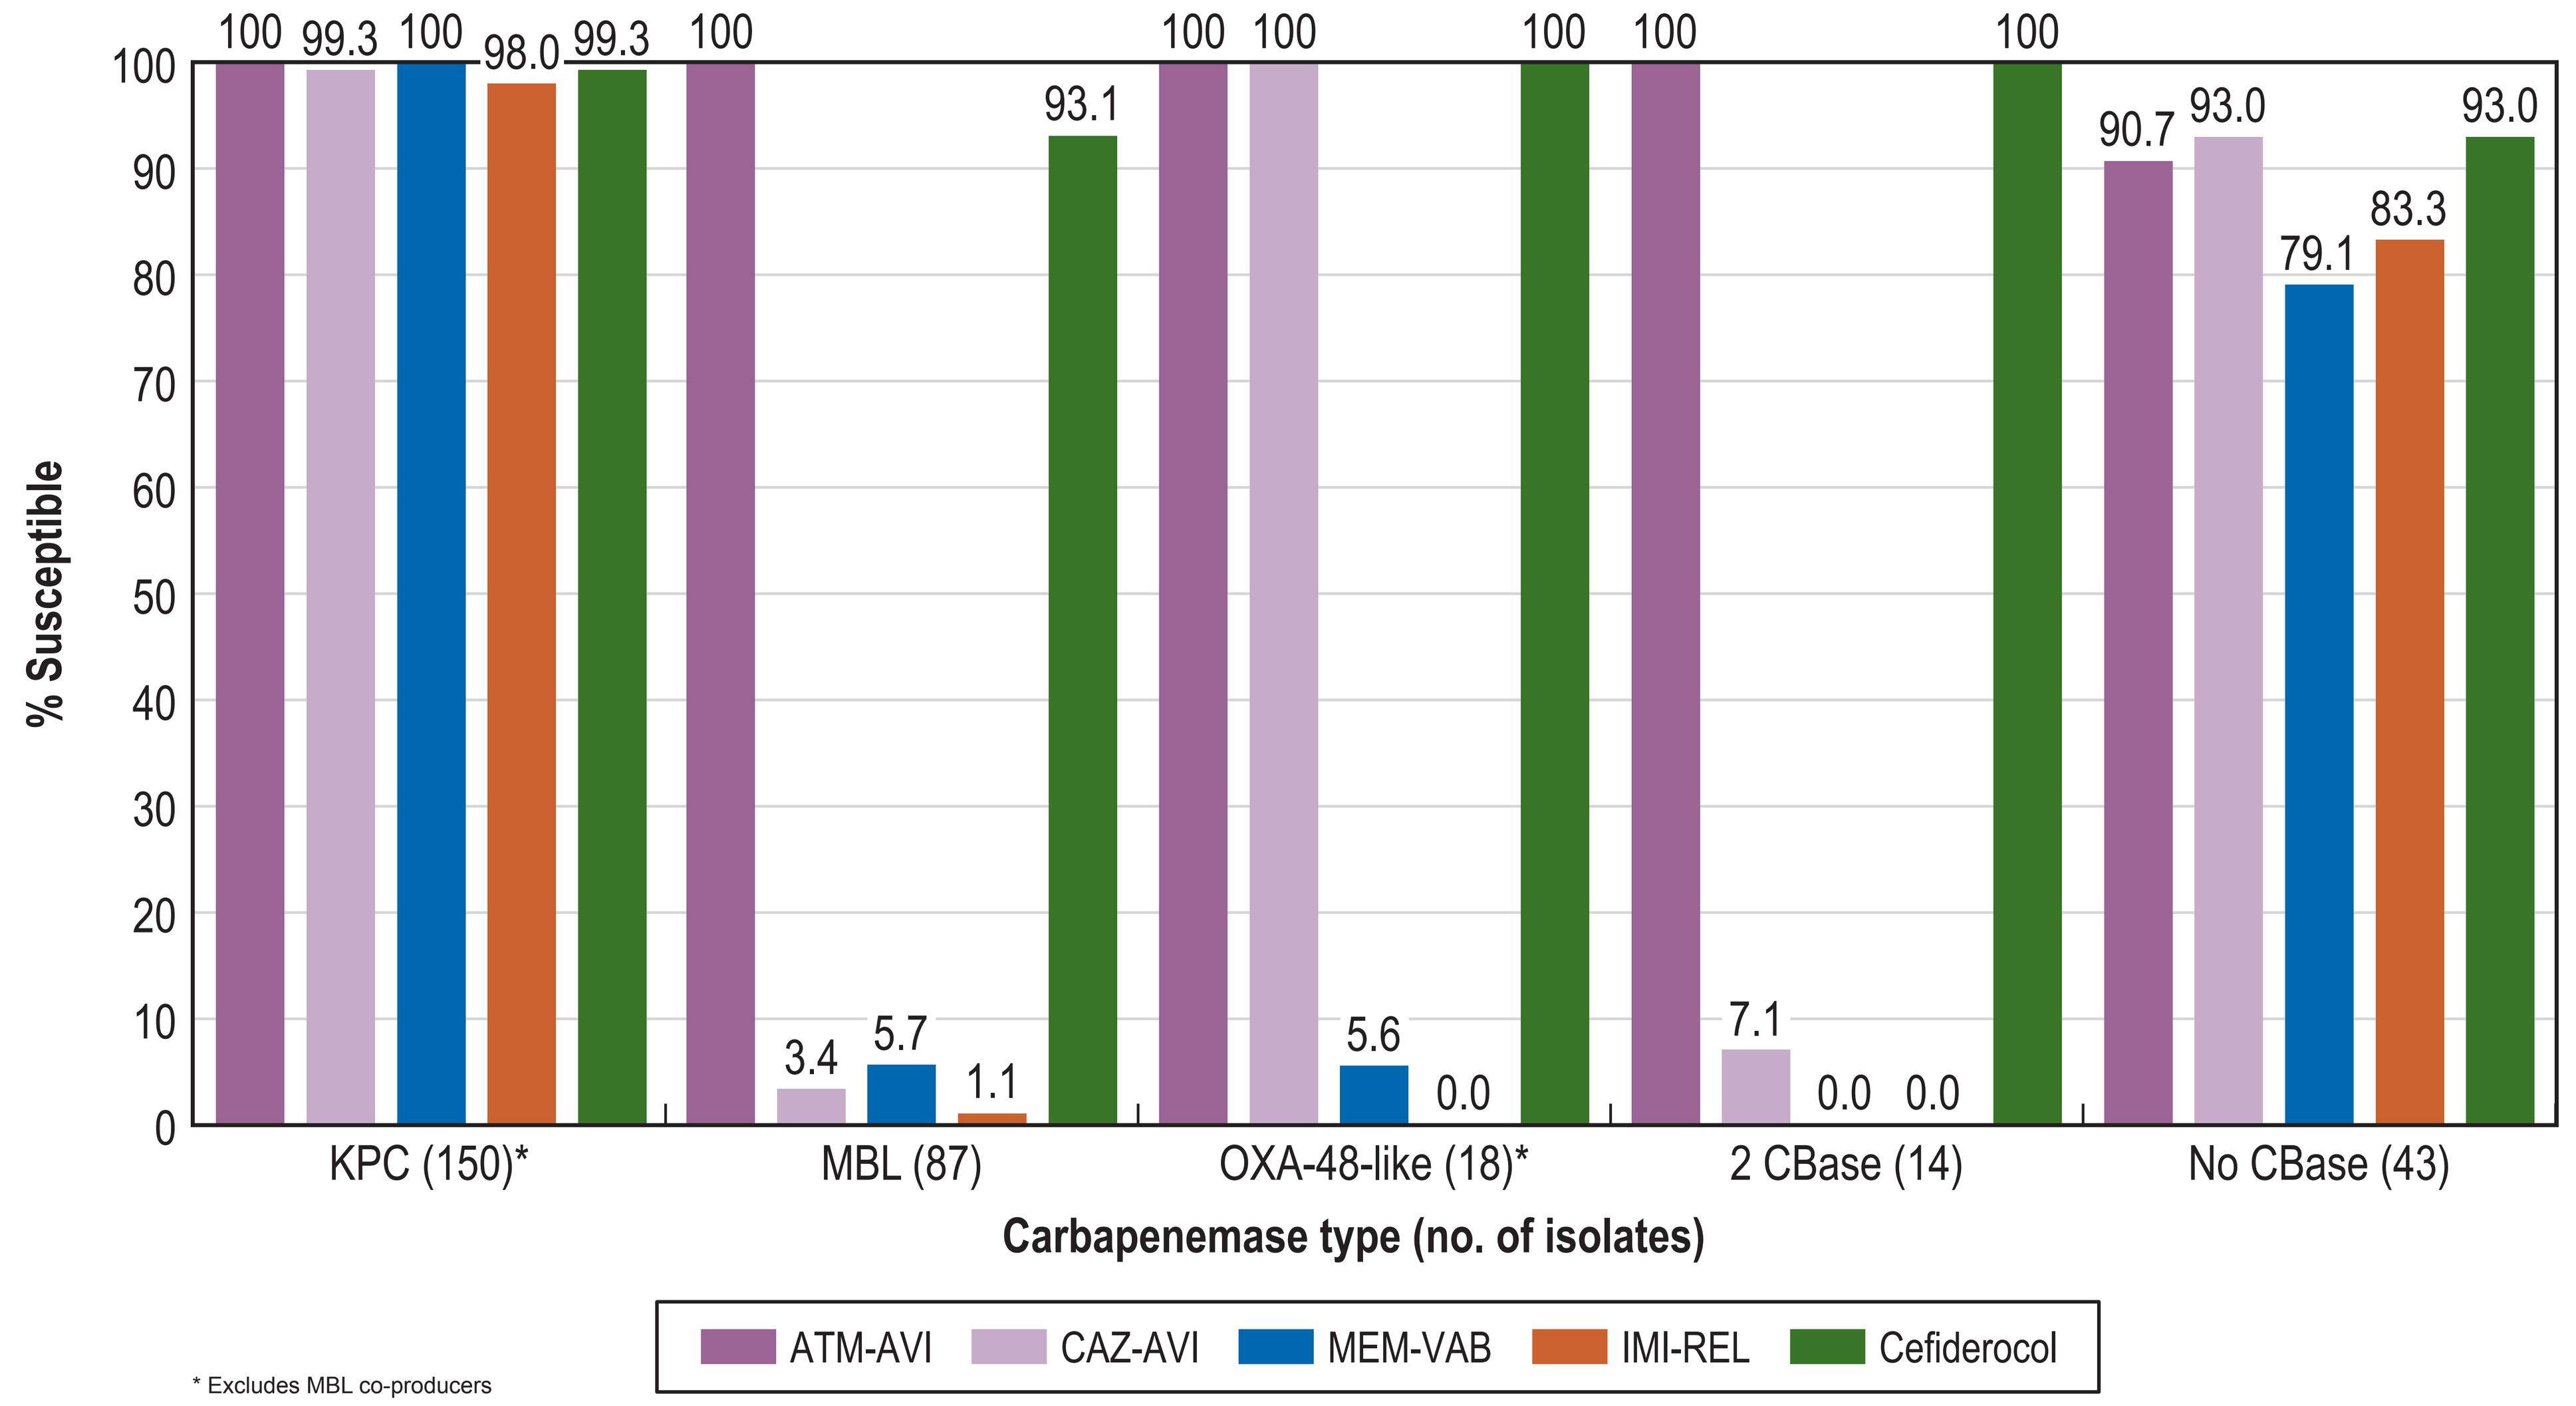

% Susceptible
100 99.3 100 98.0 99.3
100 3.4 5.7 1.1 93.1
100 100 5.6 0.0 100
100 7.1 0.0 0.0 100
90.7 93.0 79.1 83.3 93.0
KPC (150)*
MBL (87)
OXA-48-like (18)*
2 CBase (14)
No CBase (43)
Carbapenemase type (no. of isolates)
ATM-AVI
CAZ-AVI
MEM-VAB
IMI-REL
Cefiderocol

* Excludes MBL co-producers

Contact Information

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2026 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2026_26-ALG-02_P2_CR_KPN.pdf

To submit a medical question, please visit www.abbviemedinfo.com

Scan Me

Acknowledgements

AbbVie funded this study and participated in the study design, research, analysis, data collection, interpretation of data, reviewing, and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship.

References

1. CLSI. 2026. M100Ed35. Performance standards for antimicrobial susceptibility testing: 36th informational supplement. Clinical and Laboratory Standards Institute.
2. Sader HS, Kimbrough JH, Doyle TB et al. Frequency, antimicrobial susceptibility, and molecular characterization of carbapenem-resistant enterobacterales stratified by United States census divisions: Results from the INFORM program (2018–2022). *Open Forum Infect Dis* 2025; 12: ofaf005.
3. Tamma PD, Heil EL, Justo JA *et al.* Infectious Diseases Society of America 2024 guidance on the treatment of antimicrobial-resistant Gram-negative infections. *Clin Infect Dis* 2024 ciae403.