IDWEEK 2026 | Poster #P-132

Antimicrobial Activity of Aztreonam-Avibactam against Enterobacterales Isolated from Patients with Ventilator-Associated Pneumonia (VAP)

Mariana Castanheira[1], Marisa L. Winkler[1], Dmitri Debabov[2], Rodrigo E. Mendes[1], Helio S. Sader[1]

[1] Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA; [2] AbbVie Inc., Chicago, Illinois, USA

CONCLUSIONS

ATM-AVI exhibited potent activity against Enterobacterales isolated from patients with VAP, non-VAP, and BSI, and retained potent activity against CRE, MDR, and XDR isolates.

The activities of CAZ-AVI, MEM-VAB, and IMI-REL against CRE and XDR isolates were compromised by the increased frequency of MBL producers.

Resistance to ceftolozane-tazobactam and old β-lactam agents, such as ceftazidime and piperacillin-tazobactam, was higher among isolates from VAP compared to non-VAP and BSI.

INTRODUCTION

- Aztreonam-avibactam (ATM-AVI) was recently approved for clinical use by the United States (US) Food and Drug Administration (FDA) and has demonstrated potent activity against carbapenem-resistant Enterobacterales (CRE), including MBL producers.
- We evaluated the activities of ATM-AVI and other β-lactamase inhibitor combinations against Enterobacterales causing ventilator-associated pneumonia (VAP) in US medical centers.

METHODS

- Enterobacterales isolates were consecutively collected from 92 US medical centers in 2018–2025.
- The organism collection included 2,106 isolates from VAP, 3,204 isolates from patients with non-ventilator-associated pneumonia (non-VAP), and 16,404 isolates from bloodstream infection (BSI).
- Isolates were susceptibility tested by Clinical and Laboratory Standards Institute (CLSI) M07 broth microdilution methods at a monitoring laboratory.
- Cefiderocol was tested in iron-depleted media against CRE only.
- Susceptibility results from VAP isolates were compared to isolates from non-VAP and BSI.
- CRE was defined as resistant to meropenem and/or imipenem, multidrug-resistant (MDR) as nonsusceptible to ≥1 agent in ≥3 antimicrobial classes, and extensively drug-resistant (XDR) as susceptible to ≤2 classes.
- CRE isolates from VAP (*n*=43) were screened for carbapenemases (CBases) by whole genome sequencing.

Table 1. Antimicrobial susceptibility of Enterobacterales

Antimicrobial agent	% Susceptible (no. of isolates tested)[a]		
	VAP (2,106)	Non-VAP [b] (3,204)	BSI (16,404)
Aztreonam-avibactam	>99.9	99.9	99.9
Ceftazidime-avibactam	99.4	99.6	99.6
Meropenem-vaborbactam	99.5	99.7	99.6
Imipenem-relebactam	95.4	95.2	94.9
Ceftolozane-tazobactam	89.4	91.8	95.4
Piperacillin-tazobactam	79.7	83.0	89.5
Ceftazidime	80.7	82.6	84.1
Cefepime	85.8	87.0	86.0
Meropenem	98.1	98.2	99.0
Imipenem	93.2	92.8	93.4
Levofloxacin	86.6	84.0	78.8
Amikacin	96.2	95.6	94.8
Gentamicin	92.1	91.7	89.9

Abbreviations: VAP, ventilator-associated pneumonia; BSI, bloodstream infections.
[a] Susceptibility per CLSI and/or US FDA criteria.
[b] Pneumonia excluding VAP.

Table 2. Antimicrobial susceptibility of isolates from VAP

Antimicrobial agent	% Susceptible (no. of isolates tested)[a]		
	CRE (43)	MDR (539)	XDR (14)
Aztreonam-avibactam	100.0	100.0	100.0
Ceftazidime-avibactam	74.4	97.6	50.0
Meropenem-vaborbactam	76.7	98.0	38.3
Imipenem-relebactam	76.7	95.4	33.3
Cefiderocol	97.7	NT[b]	90.0
Levofloxacin	51.2	60.5	14.3
Amikacin	65.1	90.4	21.4
Gentamicin	60.5	73.5	14.3

Abbreviations: VAP, ventilator-associated pneumonia; CRE, carbapenem-resistant Enterobacterales; MDR, multidrug-resistant; XDR, extensively drug-resistant.
[a] Susceptibility per CLSI and/or US FDA criteria.
[b] NT, not tested.

RESULTS

- The most common Enterobacterales species from VAP were *K. pneumoniae* (23.0%), *S. marcescens* (17.0%), *E. coli* (16.4%), and *E. cloacae* (14.2%; Figure 1).
- ATM-AVI was highly active against VAP isolates ($MIC_{50/90}$, 0.06/0.25 mg/L; >99.9% susceptible [S]) and retained activity against 100.0% of CRE, MDR, and XDR isolates from VAP (Tables 1 and 2).
- Ceftazidime-avibactam (CAZ-AVI) and meropenem-vaborbactam (MEM-VAB) were very active against Enterobacterales (99.4–99.5% S) and MDR isolates (97.6–98.0% S) from VAP but exhibited limited activity against CRE (74.4–76.7% S) and XDR isolates (38.3–50.0%; Table 2).
- Susceptibility to ceftolozane-tazobactam, piperacillin-tazobactam, and ceftazidime was lower among isolates from VAP compared to non-VAP and BSI (Table 1).
- CRE rates were higher among isolates from VAP (2.0%) compared to non-VAP (1.7%) and BSI (0.9%; Figure 2).
- MDR rates were higher among isolates from VAP (25.6%) compared to non-VAP (22.9%) and BSI (22.6%; Figure 2).
- A CBase was identified in 69.8% of CREs from VAP and the most common CBases were KPC (48.8% of CREs), NDM (14.0%), and VIM (7.0%).

Figure 1. Frequency of Enterobacterales species isolated from patients with VAP

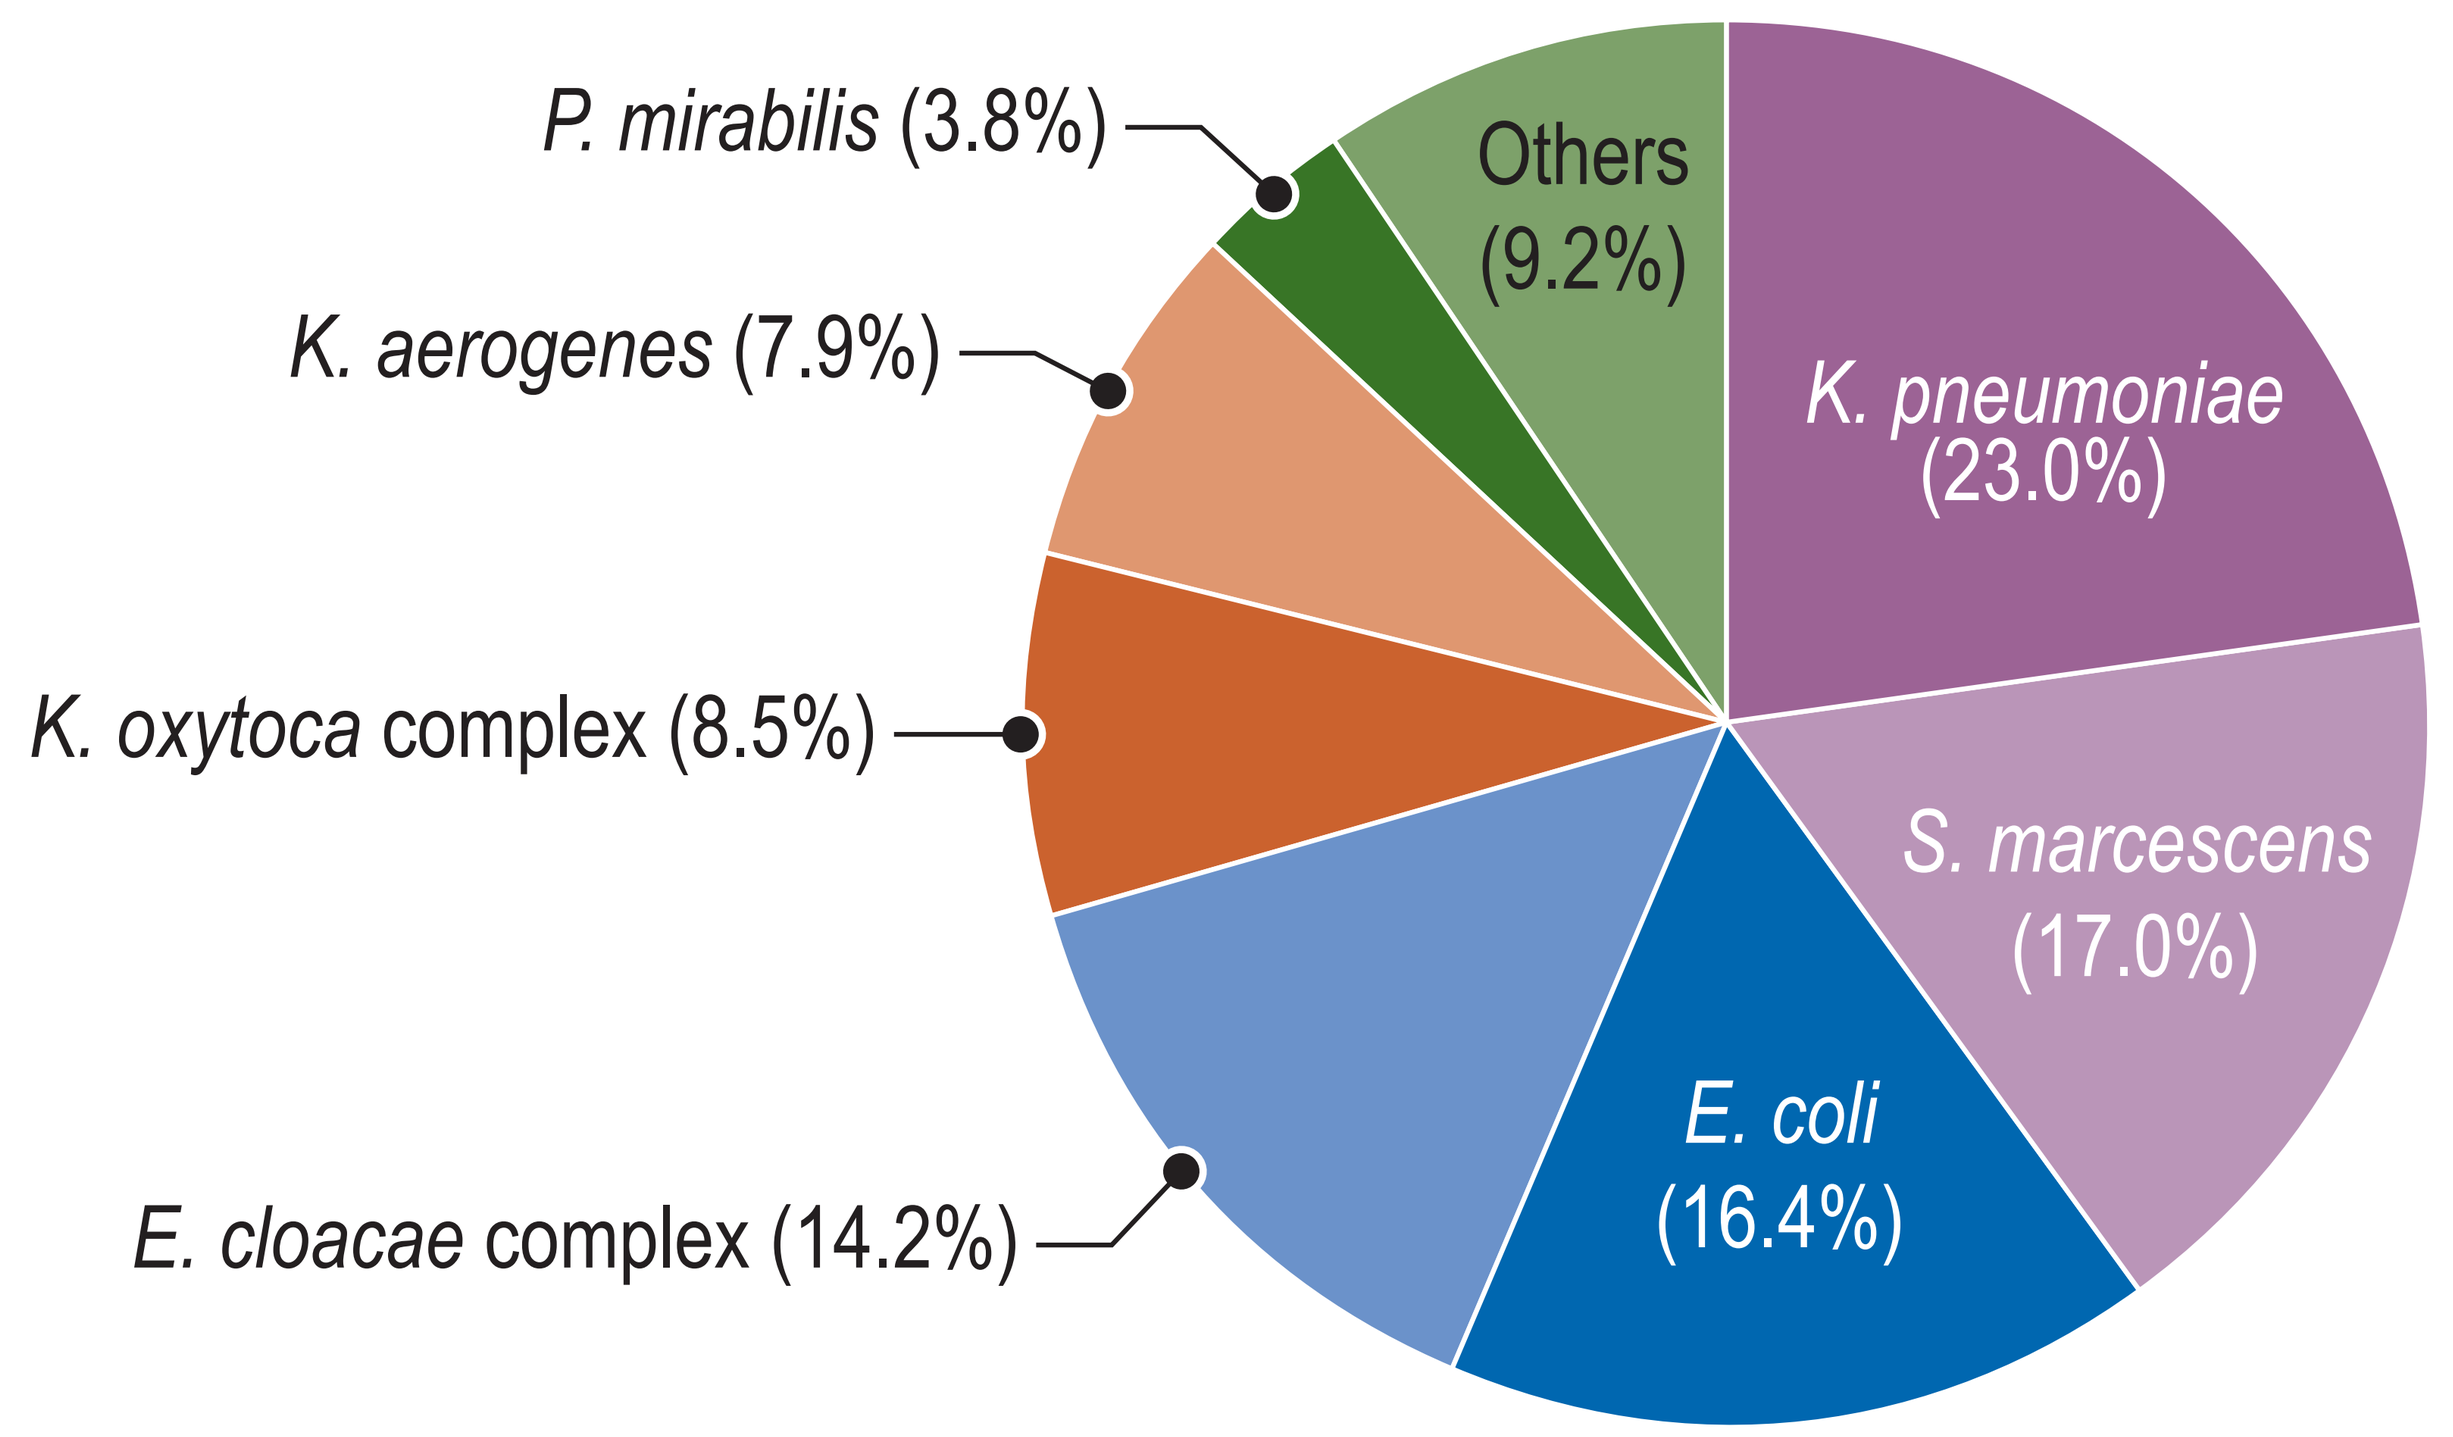
P. mirabilis (3.8%)
Others (9.2%)
K. aerogenes (7.9%)
K. pneumoniae (23.0%)
K. oxytoca complex (8.5%)
S. marcescens (17.0%)
E. coli (16.4%)
E. cloacae complex (14.2%)

Figure 2. Frequency of resistant subsets

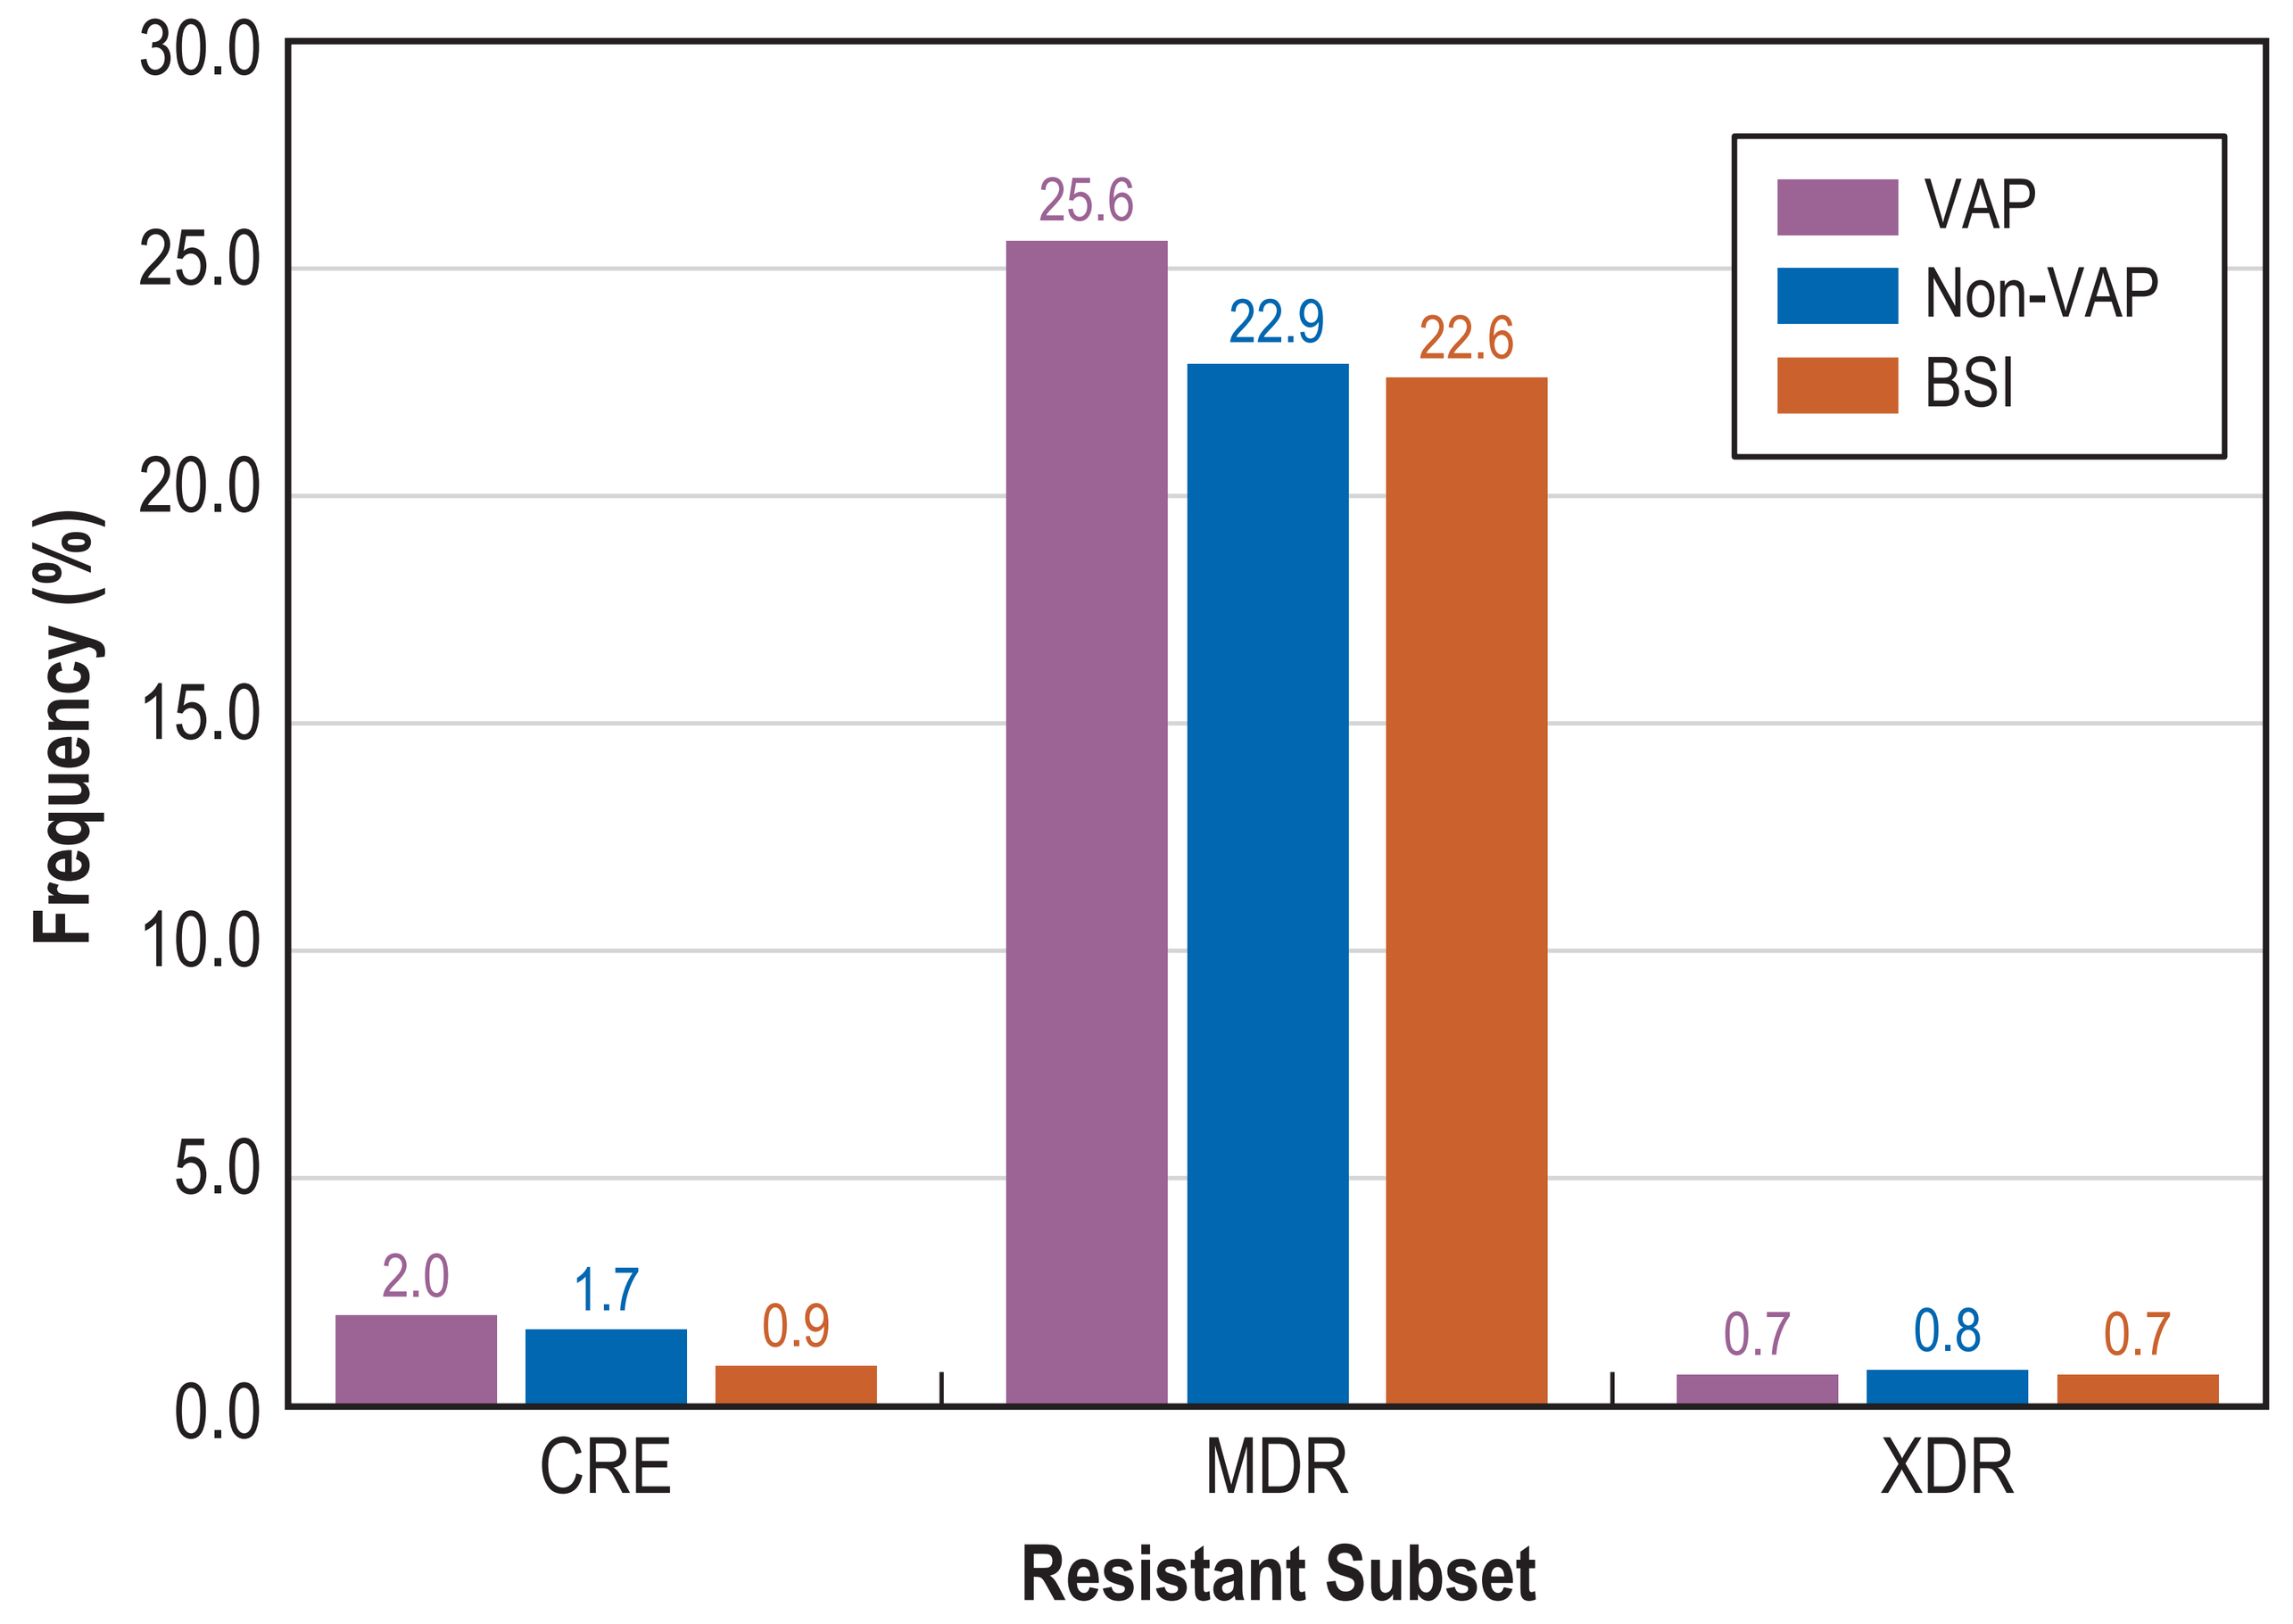
30.0
25.0
20.0
15.0
10.0
5.0
0.0
Frequency (%)
VAP
Non-VAP
BSI
2.0
1.7
0.9
25.6
22.9
22.6
0.7
0.8
0.7
CRE
MDR
XDR
Resistant Subset

Abbreviations: VAP, ventilator-associated pneumonia; BSI, bloodstream infection; CRE, carbapenem-resistant Enterobacterales; MDR, multidrug-resistant; XDR, extensively drug-resistant.

Contact Information

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2026 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2026_26-ALG-02_P3_ATM-AVI_VAP.pdf

To submit a medical question, please visit www.abbviemedinfo.com

Scan Me

Acknowledgements

AbbVie funded this study and participated in the study design, research, analysis, data collection, interpretation of data, reviewing, and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship.

References

1. CLSI. 2026. M100Ed36. Performance standards for antimicrobial susceptibility testing: 36th informational supplement. Clinical and Laboratory Standards Institute.
2. Sader HS, Kimbrough JH, Doyle TB et al. Frequency, antimicrobial susceptibility, and molecular characterization of carbapenem-resistant enterobacterales stratified by United States census divisions: Results from the INFORM program (2018–2022). *Open Forum Infect Dis* 2025; 12: ofaf005.
3. Sader HS, Mendes RE, Kimbrough JH, et al. Multidrug-resistant Enterobacterales in United States hospitals: Frequency and susceptibility to newer β-lactamase inhibitor combinations (2020–2024). Int J Antimicrob Agents. 2026;67(8):107835.
4. Tamma PD, Heil EL, Justo JA *et al.* Infectious Diseases Society of America 2024 guidance on the treatment of antimicrobial-resistant Gram-negative infections. *Clin Infect Dis* 2024 ciae403.

abbvie