IDWEEK 2026 | Poster #P-116

Antimicrobial Susceptibility of KPC-Producing Enterobacterales and Molecular Characterization of Ceftazidime-Avibactam-Resistant Strains from United States Medical Centers (2018–2025)

Helio S. Sader[1], John H. Kimbrough[1], Krisztina M. Papp-Wallace[1], John Lock[2], Mariana Castanheira[1], Rodrigo E. Mendes[1]

[1] Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA; [2] AbbVie Inc., Chicago, Illinois, USA

CONCLUSIONS

The prevalence of KPC variants that confer CAZ-AVI resistance appears to be extremely low nationwide.

CAZ-AVI remains highly active against KPC-producing Enterobacterales causing infections in US medical centers.

CAZ-AVI-resistant, KPC-producing Enterobacterales isolates were mainly *K. pneumoniae* (50.0%) and we did not identify a predominant MLST or KPC variant among these isolates.

INTRODUCTION

- There have been several reports of KPC variants that confer resistance to ceftazidime-avibactam (CAZ-AVI), but the prevalence of these variants among KPC-producing Enterobacterales has not been assessed in large nationwide surveillance.
- We aim to evaluate the frequency of KPC variants that confer resistance to CAZ-AVI in US medical centers.
- Thus, we assessed the activity of CAZ-AVI against a large collection of KPC-producing Enterobacterales from US medical centers and characterized the CAZ-AVI-resistant strains.

METHODS

- Among 70,530 Enterobacterales isolates consecutively collected in 2018–2025 from 93 US medical centers, 763 (1.1%) were carbapenem-resistant (CRE) and 448 (0.6%) were KPC producers (metallo-β-lactamase [MBL] co-producers were excluded).
- Isolates were susceptibility tested by CLSI broth microdilution.
- Cefiderocol was tested in iron-depleted media.
- CRE was defined as resistant (MIC ≥4 mg/L) to meropenem and/or imipenem.
- All isolates with decreased susceptibility to broad-spectrum cephalosporins and/or carbapenems were screened for carbapenemase genes by whole genome sequencing and molecular characterization.
- Isolates identified as carrying KPC encoding genes were evaluated in this analysis.

RESULTS

- A total of 448 KPC producers were identified among 70,530 Enterobacterales (0.6%).
- 87.2% of KPC producers (391/448) were categorized as CRE (meropenem and/or imipenem MIC ≥4 mg/L).
- The most common KPC-producing species were *Klebsiella pneumoniae* (*n*=242; 54.0%) and *Enterobacter cloacae* species complex (*n*=73; 16.3%).
- 97.8% of KPC producers were CAZ-AVI-susceptible and 15.8% were meropenem-susceptible (Table 1 and Figure 1).
- CAZ-AVI susceptibility rates among KPC-producing Enterobacterales species ranged from 95.7% (*Serratia marcescens*) to 100.0% (*Klebsiella oxytoca* species complex) and did not show any clear trends over the years (Table 2).
- Susceptibility to amikacin and gentamicin improved markedly during the investigation (Table 2 and Figure 1).
- Ten KPC-producing isolates (2.2%) were CAZ-AVI-resistant and 7 of them were meropenem-susceptible (Table 3).
- These 10 isolates were from 5 species and produced 9 distinct KPC variants (Table 3).
- Among KPC-producing *K. pneumoniae*, 5 isolates (2.1%) were CAZ-AVI-resistant; these 5 isolates produced KPC-3, KPC-31 (2 isolates), KPC-53, and KPC-58 and belonged to 4 multilocus sequence types (MLST; Table 3).

Figure 1. Yearly activity of selected agents against KPC-producing Enterobacterales

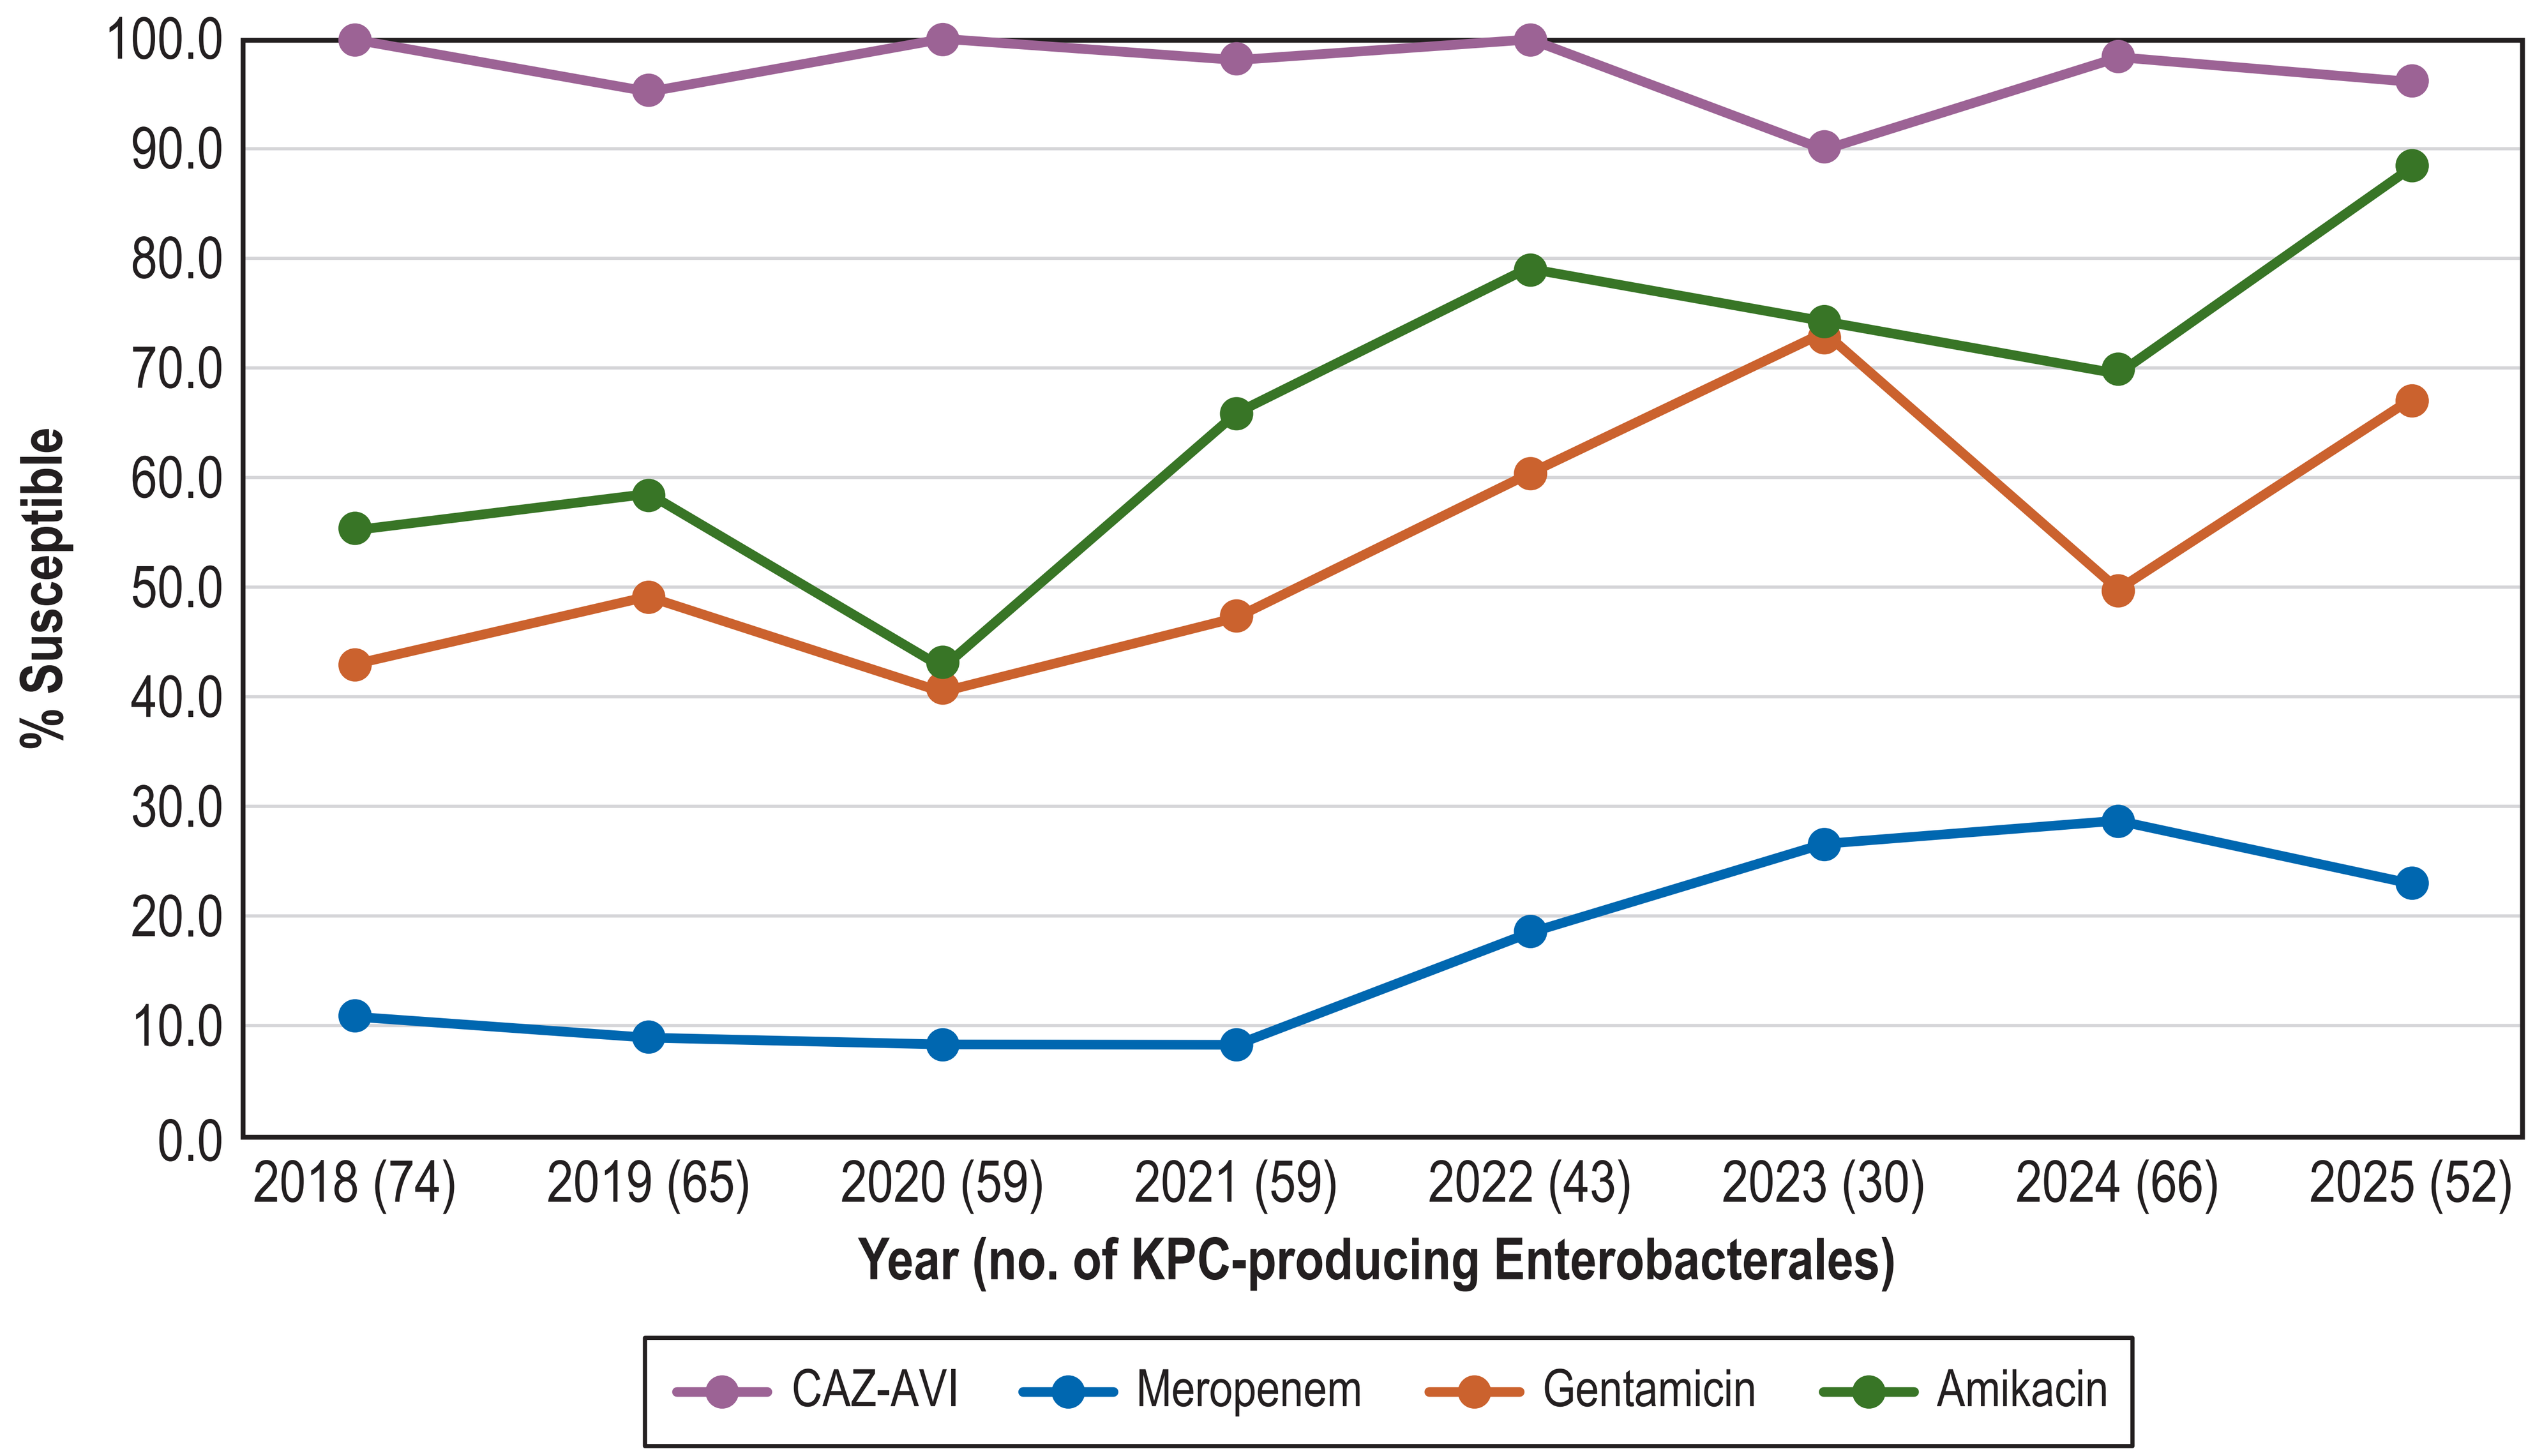
% Susceptible
100.0
90.0
80.0
70.0
60.0
50.0
40.0
30.0
20.0
10.0
0.0
2018 (74)
2019 (65)
2020 (59)
2021 (59)
2022 (43)
2023 (30)
2024 (66)
2025 (52)
Year (no. of KPC-producing Enterobacterales)
CAZ-AVI
Meropenem
Gentamicin
Amikacin

Table 1. Antimicrobial susceptibility of KPC-producing Enterobacterales stratified by species

Organism (no. of isolates)	% Susceptible					
	CAZ-AVI	MEM-VAB	IMI-REL	Cefiderocol	MEM	Amikacin
Enterobacterales (448)	97.8	99.1	97.0	99.0	15.8	65.0
K. pneumoniae (242)	97.9	99.6	97.5	99.6	9.5	53.7
E. cloacae complex (73)	97.3	95.8	97.0	96.2	28.8	84.9
E. coli (28)	96.4	100.0	100.0	94.7	42.9	75.0
K. oxytoca complex (27)	100.0	100.0	100.0	100.0	7.4	88.9
S. marcescens (23)	95.7	100.0	91.3	100.0	13.0	73.9
Other species (55)	98.2	100.0	93.6	100.0	18.2	67.3

Abbreviations: CAZ-AVI, ceftazidime-avibactam; MEM, meropenem; VAB, vaborbactam; IMI-REL, imipenem-relebactam.

Table 2. Antimicrobial susceptibility of KPC-producing Enterobacterales stratified by year

Antimicrobial	% Susceptible by year (no. of isolates)							
	2018 (74)	2019 (65)	2020 (59)	2021 (59)	2022 (43)	2023 (30)	2024 (66)	2025 (52)
CAZ-AVI	100.0	95.4	100.0	98.3	100.0	90.0	98.5	96.2
MEM-VAB	98.4	96.8	100.0	100.0	97.7	100.0	100.0	100.0
IMI-REL	93.8	94.9	100.0	98.3	95.3	100.0	98.5	96.2
Cefiderocol	100.0	98.3	100.0	96.4	100.0	95.8	100.0	100.0
Meropenem	10.8	9.2	8.5	8.5	18.6	26.7	28.8	23.1
Gentamicin	43.2	49.2	40.7	47.5	60.5	73.3	50.0	67.3
Amikacin	55.4	58.5	42.4	66.1	79.1	73.3	69.7	88.5

Abbreviations: CAZ-AVI, ceftazidime-avibactam; MEM-VAB, meropenem-vaborbactam; IMI-REL, imipenem-relebactam.

Table 3. Molecular characterization of ceftazidime-avibactam-resistant, KPC-producing Enterobacterales

Collection #	Year	Species	CAZ-AVI MIC [a]	MEM MIC [b]	KPC	MLST [c]
1106489	2019	*K. pneumoniae*	32	0.5	KPC-58	258
1111548	2019	*K. pneumoniae*	16	>32	KPC-3	258
1117040	2019	*K. pneumoniae*	>32	0.12	KPC-31	6450
1197406	2021	*E. cloacae* complex	16	4	KPC-4	597
1272771	2023	*C. freundii* complex	>32	0.06	KPC-44	22
1275465	2023	*E. coli*	>32	0.12	KPC-256	361
1294369	2023	*K. pneumoniae*	16	0.06	KPC-53	307
1416974	2024	*S. marcescens*	>32	0.5	KPC-140	882
1443067	2025	*K. pneumoniae*	>32	8	KPC-31	268
1466268	2025	*E. cloacae* complex	>32	0.12	KPC-74 (S181_P182insS)	66

[a] Ceftazidime-avibactam MIC in mg/L.
[b] Meropenem MIC in mg/L.
[c] Multilocus sequence typing.

Contact Information

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2026 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2026_26-ALG-02_P4_CAZ-AVI_KPC.pdf

To submit a medical question, please visit www.abbviemedinfo.com

Scan Me

Acknowledgements

AbbVie funded this study and participated in the study design, research, analysis, data collection, interpretation of data, reviewing, and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship.

References

1. CLSI. 2026. M100Ed36. Performance standards for antimicrobial susceptibility testing: 36th informational supplement. Clinical and Laboratory Standards Institute.
2. Hobson CA, Pierrat G, Tenaillon O, et al. *Klebsiella pneumoniae* carbapenemase variants resistant to ceftazidime-avibactam: An evolutionary overview. *Antimicrob Agents Chemother.* 2022;66(9):e0044722.
3. Sader HS, Kimbrough JH, Doyle TB et al. Frequency, antimicrobial susceptibility, and molecular characterization of carbapenem-resistant Enterobacterales stratified by United States census divisions: Results from the INFORM program (2018–2022). *Open Forum Infect Dis* 2025; 12: ofaf005.
4. Shields RK, Chen L, Cheng S, Chavda KD, et al. Emergence of ceftazidime-avibactam resistance due to plasmid-borne *bla*$_{kpc\text{-}3}$ mutations during treatment of carbapenem-resistant *Klebsiella pneumoniae* infections. *Antimicrob Agents Chemother.* 2017;61(3):e02097-16.

abbvie