IDWEEK 2026 | Poster #P-115

Activity of Ceftazidime-Avibactam against *Pseudomonas aeruginosa* from Patients with Ventilator-Associated Pneumonia (VAP) from United States Medical Centers (2021–2025)

Helio S. Sader[1], Krisztina M. Papp-Wallace[1], Todd Riccobene[2], Rodrigo E. Mendes[1], Mariana Castanheira[1]

[1] Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA; [2] AbbVie Inc., Chicago, Illinois, USA

CONCLUSIONS

CAZ-AVI, TOL-TAZ, and IMI-REL were highly active and exhibited similar coverage against a large contemporary collection of *P. aeruginosa* isolates from patients with VAP, non-VAP, and BSI.

Cross-resistance among these β-lactamase inhibitor combinations varied markedly, indicating that all three should be tested in the clinical laboratory and available for clinical use.

INTRODUCTION

- Treatment of ventilator-associated pneumonia (VAP) represents a major challenge.
- Mortality rates and the length of hospital stays are markedly higher among patients with VAP compared to other infections and an important factor that contributes to poor patient outcomes is a delay in the introduction of effective antimicrobial therapy.
- We evaluated the antimicrobial susceptibility of *Pseudomonas aeruginosa* causing VAP in United States (US) hospitals.
- Susceptibility results from VAP isolates were compared to isolates from patients with non-ventilator-associated pneumonia (non-VAP) and bloodstream infections (BSI).

METHODS

- A total of 7,576 *P. aeruginosa* isolates were consecutively collected from 85 US medical centers through the INFORM Program in 2021–2025.
- The organism collection included 793 isolates from VAP, 1,583 isolates from non-VAP, and 967 isolates from BSI.
- Isolates were susceptibility tested by CLSI broth microdilution in the monitoring laboratory (JMI Laboratories).
- Multidrug-resistant (MDR) was defined as nonsusceptibility to ≥1 agent in ≥3 antimicrobial classes and difficult-to-treat (DTR) was defined as nonsusceptibility to all the following: piperacillin-tazobactam, ceftazidime, cefepime, aztreonam, meropenem, imipenem, ciprofloxacin, and levofloxacin.

RESULTS

- The most active agents against *P. aeruginosa* from all three patient groups were ceftazidime-avibactam (CAZ-AVI; 95.8% to 98.2% susceptible), ceftolozane-tazobactam (TOL-TAZ; 95.1% to 98.3% susceptible), and imipenem-relebactam (IMI-REL; 97.0% to 98.9% susceptible) (Table 1).
- Susceptibility rates were generally lower among isolates from VAP compared to non-VAP and BSI, and lower among isolates from non-VAP compared to BSI (Table 1).
- Highest differences in susceptibility between isolates from VAP and BSI were observed for meropenem (73.5% vs. 89.6%), imipenem (74.0% vs. 89.6%), piperacillin-tazobactam (70.6% vs. 85.0%), cefepime (79.3% vs. 92.0%), and ceftazidime (76.7% vs. 88.5%; Table 1).
- The frequency of MDR and DTR isolates was markedly higher among VAP isolates (22.7% and 3.9% respectively) compared to non-VAP (14.8% and 1.4% respectively) and BSI (8.4% and 1.0% respectively; Figure 1).
- Ceftazidime-avibactam, ceftolozane-tazobactam, and imipenem-relebactam retained good activity against MDR and DTR *P. aeruginosa* from VAP and non-VAP (Table 2).
- CAZ-AVI retained activity against 51.3% of TOL-TAZ-nonsusceptible and 62.5% of IMI-REL-nonsusceptible isolates (Table 3).

Table 1. Antimicrobial susceptibility of *P. aeruginosa*

Antimicrobial agent	% Susceptible (no. of isolates tested)[a]		
	VAP (793)	Non-VAP [b] (1,583)	BSI (967)
Ceftazidime-avibactam	95.8	97.6	98.2
Ceftolozane-tazobactam	95.1	97.5	98.3
Imipenem-relebactam	97.0	98.6	98.9
Meropenem-vaborbactam [c]	89.5 [c]	94.8 [c]	96.7 [c]
Piperacillin-tazobactam	70.6	80.1	85.0
Ceftazidime	76.7	83.5	88.5
Cefepime	79.3	85.7	92.0
Meropenem	73.5	83.1	89.6
Imipenem	74.0	81.2	89.6
Levofloxacin	71.0	75.0	80.1
Tobramycin	89.0	89.8	96.7

Abbreviations: VAP, ventilator-associated pneumonia; BSI, bloodstream infections.
[a] Susceptibility per CLSI and/or US FDA criteria.
[b] Pneumonia excluding VAP.
[c] This compound is not approved for treatment of *P. aeruginosa* infections in the US. EUCAST breakpoint criteria were applied since neither CLSI nor US FDA publishes breakpoint criteria.

Figure 1. Frequency of MDR and DTR isolates

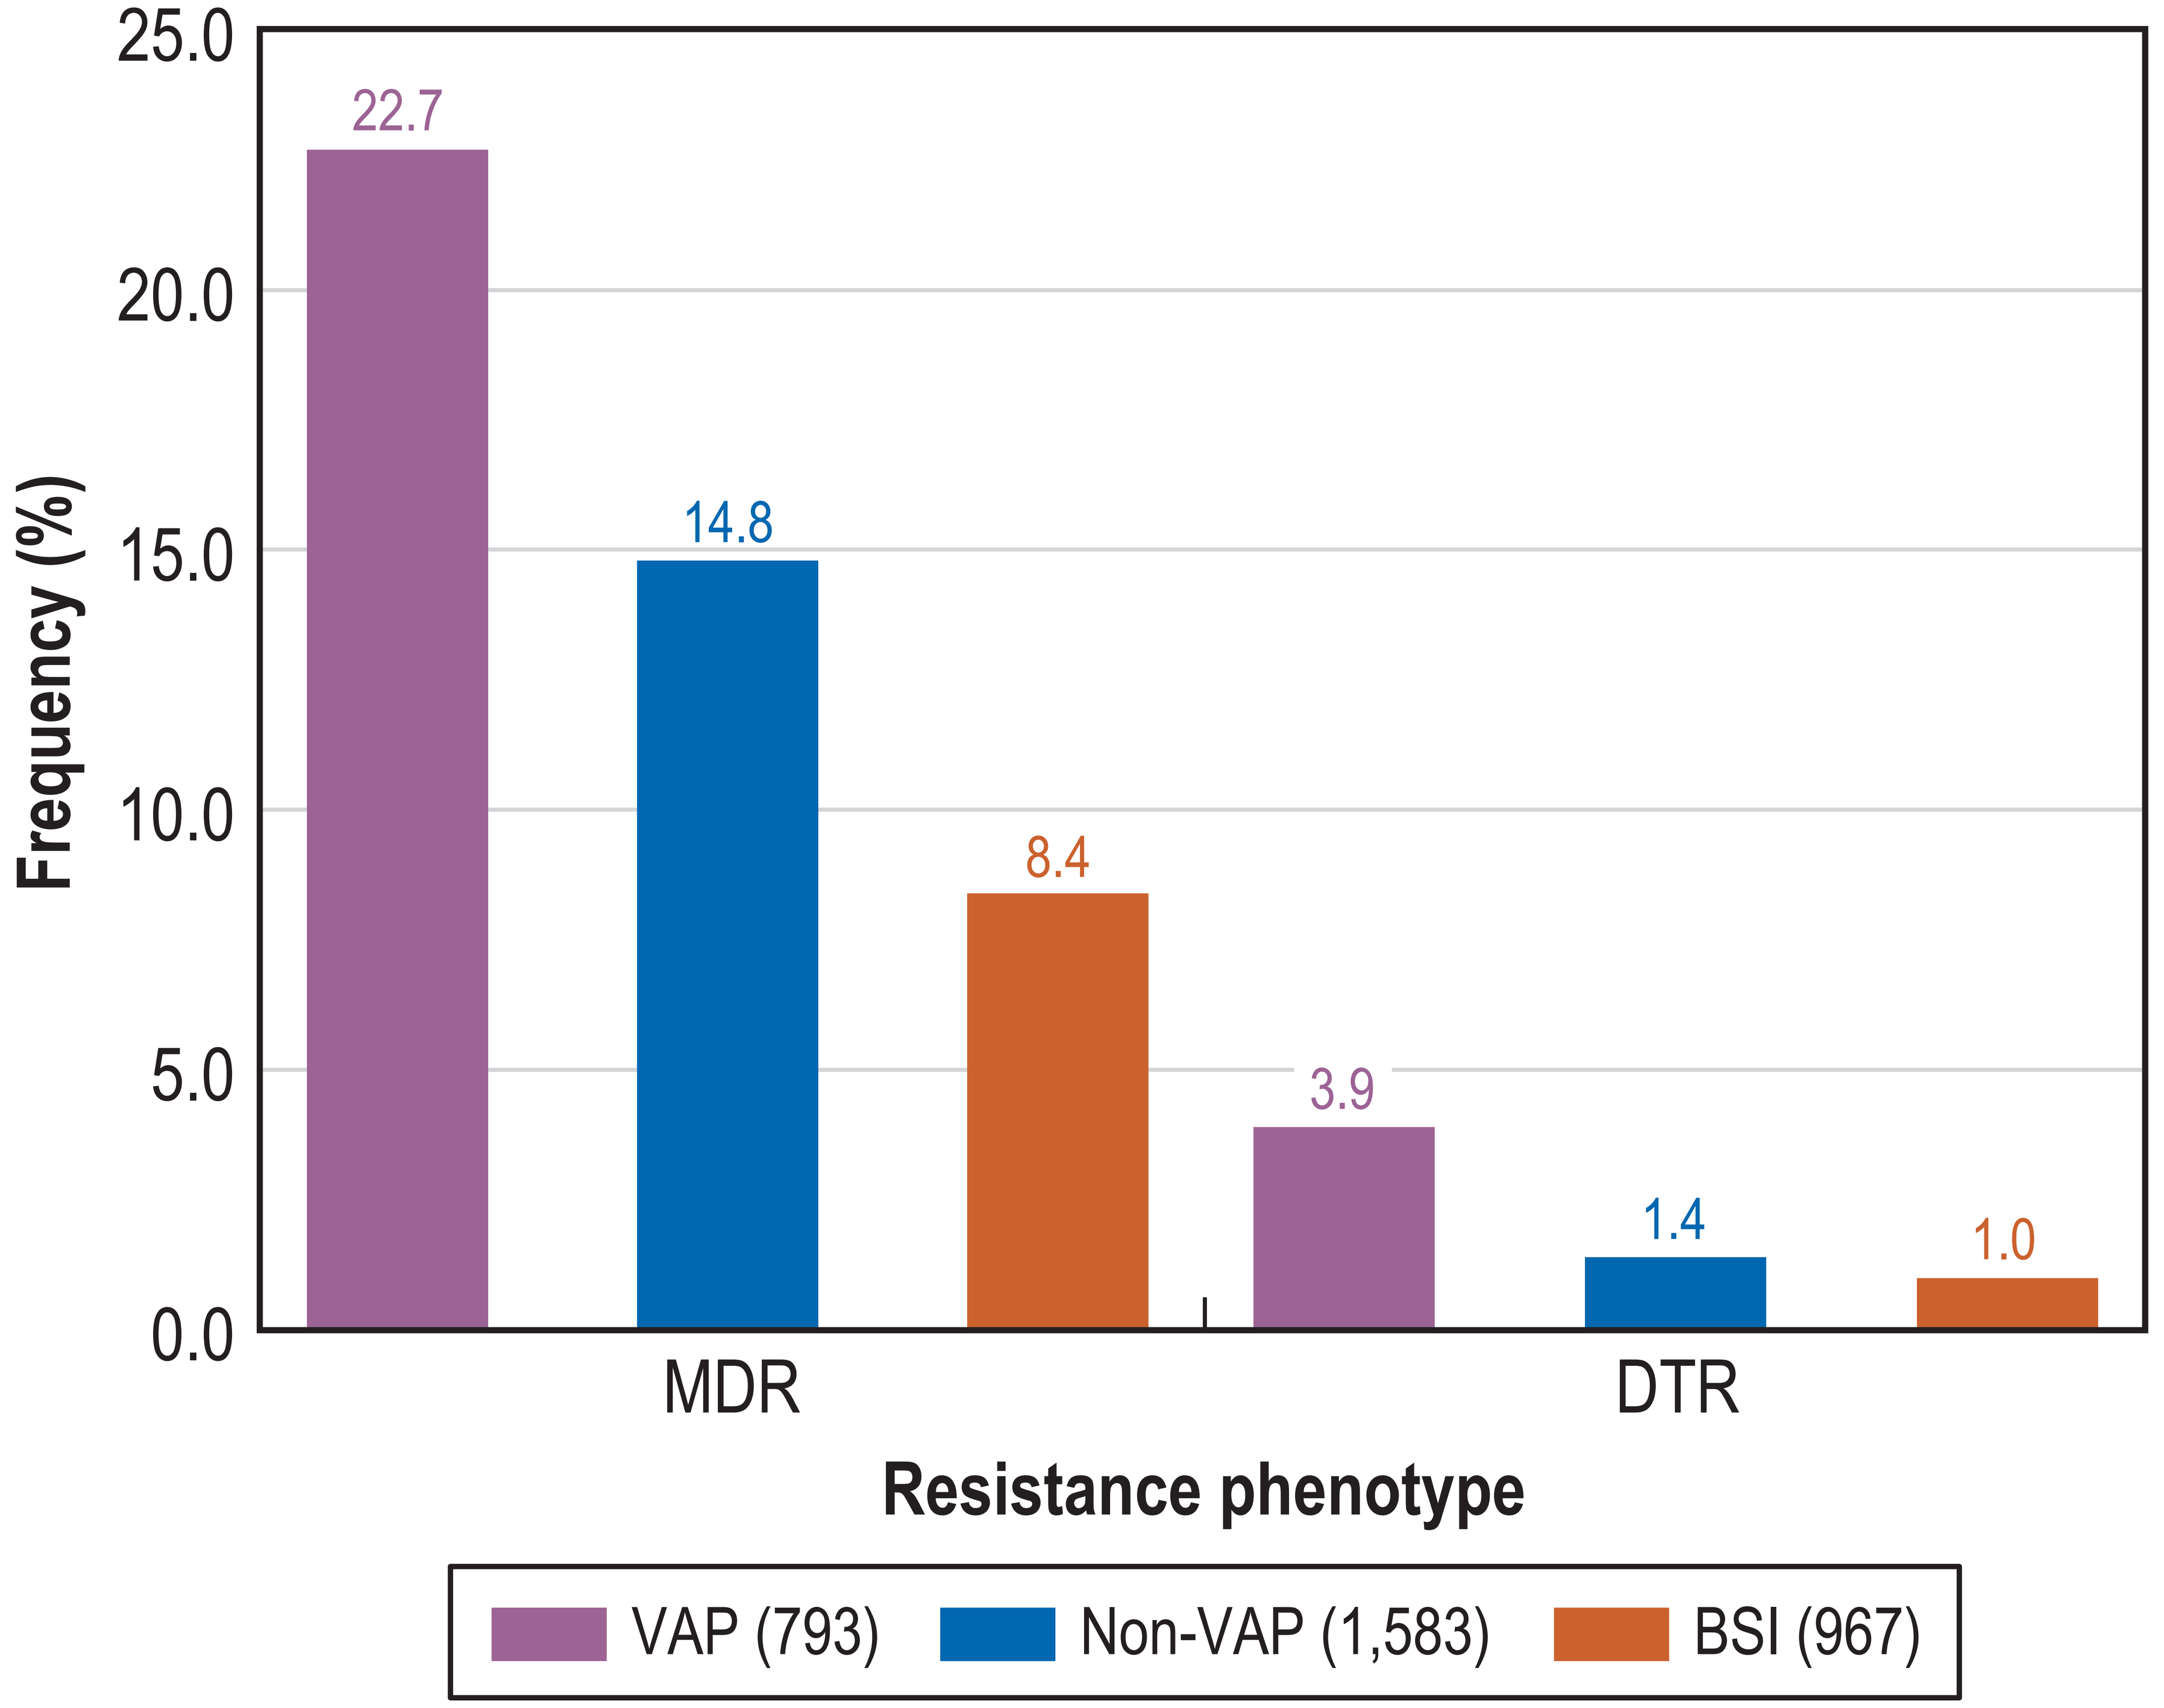
25.0
20.0
15.0
10.0
5.0
0.0
Frequency (%)
22.7
14.8
8.4
3.9
1.4
1.0
MDR
DTR
Resistance phenotype
VAP (793)
Non-VAP (1,583)
BSI (967)

Table 2. Antimicrobial susceptibility of MDR and DTR *P. aeruginosa*

Resistant subset / Antimicrobial agent	% Susceptible (no. of isolates tested)[a]		
	VAP	Non-VAP [b]	BSI
MDR	(180)	(235)	(81)
Ceftazidime-avibactam	81.7	84.7	79.0
Ceftolozane-tazobactam	78.3	84.3	80.0
Imipenem-relebactam	86.7	91.5	86.7
Tobramycin	70.6	58.7	70.4
DTR	(31)	(22)	(10)
Ceftazidime-avibactam	71.0	72.7	40.0
Ceftolozane-tazobactam	71.0	72.7	60.0
Imipenem-relebactam	77.4	72.7	60.0
Tobramycin	71.0	36.4	40.0

Abbreviations: VAP, ventilator-associated pneumonia; BSI, bloodstream infections.
[a] Susceptibility per CLSI and/or US FDA criteria.
[b] Pneumonia excluding VAP.

Table 3. Cross resistance rates among β-lactamase inhibitor combinations when testing *P. aeruginosa* isolates from VAP

Antimicrobial agent	% Susceptible by resistance phenotype (no. of isolates)		
	CAZ-AVI-NS (33)	TOL-TAZ-NS (39)	IMI-REL-NS (24)
CAZ-AVI	0.0	51.3	62.5
TOL-TAZ	42.4	0.0	50.0
IMI-REL	72.7	69.2	0.0

Abbreviations: CAZ-AVI, ceftazidime-avibactam; TOL-TAZ, ceftolozane-tazobactam; NS, nonsusceptible; IMI-REL, imipenem-relebactam.

Contact Information

Helio S. Sader, MD, PhD, FIDSA
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2026 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2026_26-ALG-02_P5_PSA_VAP.pdf

To submit a medical question, please visit www.abbviemedinfo.com

Scan Me

Acknowledgements

AbbVie funded this study and participated in the study design, research, analysis, data collection, interpretation of data, reviewing, and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship.

References

1. CLSI. 2026. M100ed36. Performance standards for antimicrobial susceptibility testing: 36th informational supplement. Clinical and Laboratory Standards Institute.
2. Sader HS, Mendes RE, Kimbrough JH, *et al.* Activity of aztreonam/avibactam and recently approved β-lactamase inhibitor combinations against Enterobacterales and *Pseudomonas aeruginosa* from intensive care unit and non-intensive care unit patients. Antibiotics (Basel). 2024;13(6):564.
3. Sader HS, Mendes RE, Lock J, *et al.* A 10-year trend of antimicrobial resistance in *Pseudomonas aeruginosa* from United States medical centers: results from the INFORM surveillance program (2015–2024). *Open Forum Infect Dis* 2026 [in press].
4. Tamma PD, Heil EL, Justo JA *et al.* Infectious Diseases Society of America 2024 guidance on the treatment of antimicrobial-resistant Gram-negative infections. *Clin Infect Dis* 2024 ciae403.

abbvie