IDWEEK 2026 | Poster #P-118

Aztreonam-Avibactam Activity against Gram-negative Bacteria Isolated from Patients Hospitalized with Pneumonia in United States Medical Centers (2023–2025)

Helio S. Sader[1], John H. Kimbrough[1], John Lock[2], Krisztina M. Papp-Wallace[1], Mariana Castanheira[1]

[1] *Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA;* [2] *AbbVie Inc., Chicago, Illinois, USA*

CONCLUSIONS

ATM-AVI exhibited potent activity against Enterobacterales and *S. maltophilia* isolated from patients with pneumonia in US hospitals.

ATM-AVI activity was not adversely affected by clinically relevant carbapenemases, including MBLs.

The activities of CAZ-AVI and MEM-VAB against Enterobacterales were compromised by the increased occurrence of MBL producers among CRE isolates.

CAZ-AVI and TOL-TAZ demonstrated potent activity against *P. aeruginosa* from patients with pneumonia.

INTRODUCTION

- Aztreonam-avibactam (ATM-AVI) was approved by the United States (US) Food and Drug Administration (FDA) in 2025 for the treatment of intra-abdominal infections and has demonstrated potent activity against carbapenem-resistant Enterobacterales (CRE), including metallo-β-lactamase (MBL) producers.
- We evaluated the frequency and antimicrobial susceptibility of selected Gram-negative bacilli (GNB) causing pneumonia in US hospitals.

Table 1. Activity of selected antimicrobials

Organism (no.)	% Susceptible per CLSI and/or US FDA					
	ATM-AVI	CAZ-AVI	MEM-VAB	TOL-TAZ	PIP-TAZ	MEM
Enterobacterales (4,474)	99.9	99.2	99.3	90.3	79.5	97.8
K. pneumoniae (1,154)	99.8	97.8	97.8	90.7	75.4	95.1
E. coli (777)	99.9	99.6	99.7	94.9	86.0	99.6
S. marcescens (745)	100.0	99.7	99.9	96.9	87.2	98.7
E. cloacae complex (604)	99.8	99.8	99.7	74.5	68.0	98.2
MDR (1,203)	99.7	96.8	97.3	64.3	36.8	91.7
CRE (96)	96.9	67.7	66.7	7.3	2.1	9.4
P. aeruginosa (2,807)	(80.2) [a]	96.1	92.3 [b]	96.2	74.8	79.1

[a] % inhibited at ≤8 mg/L.
[b] Based on EUCAST breakpoint for *P. aeruginosa* (≤8 mg/L).
Abbreviations: ATM, aztreonam; AVI, avibactam; CAZ, ceftazidime; MEM, meropenem; VAB, vaborbactam; TOL, ceftolozane; TAZ, tazobactam; PIP, piperacillin.

Table 2. Antimicrobial susceptibility of *S. maltophilia* and *B. cepacia*

Organism (no.) / antimicrobial agent	MIC in mg/L		Susceptibility per CLSI[a]	
	MIC_{50}	MIC_{90}	%Susceptible	%Resistant
S. maltophilia (779)				
Aztreonam-avibactam	4	4	(92.9/99.6)[b]	
Cefiderocol	0.12	0.5	98.7	
Levofloxacin	1	8	79.8	11.9
TMP-SMX [c]	0.25	1	96.7	3.3
Tigecycline	1	4	(89.6)[d]	
Minocycline	0.5	1	90.9	2.1
B. cepacia (25)				
Aztreonam-avibactam	4	8	(56.0/96.0)[b]	
Ceftazidime-avibactam	2	4	(100.0)[e]	
Ceftolozane-tazobactam	2	8	(88.0)[e]	
Meropenem-vaborbactam	0.5	1	(96.0/96.0)[b]	
Cefiderocol	0.06	0.5	(94.7)[e]	
Ceftazidime	4	8	(92.0)[e]	
Meropenem	4	4	(48.0)[e]	
Levofloxacin	2	>32	(24.0)[e]	
Tobramycin	>16	>16	(8.0)[e]	
Tigecycline	2	8	(68.0)[d]	

[a] Criteria as published by CLSI (2026) unless noted.
[b] % inhibited at ≤4/≤8 mg/L for comparison.
[c] Trimethoprim-sulfamethoxazole.
[d] % inhibited at ≤2 mg/L for comparison.
[e] *P. aeruginosa* breakpoints were applied for comparison.

Figure 1. Carbapenemase (CBase) distribution among CREs

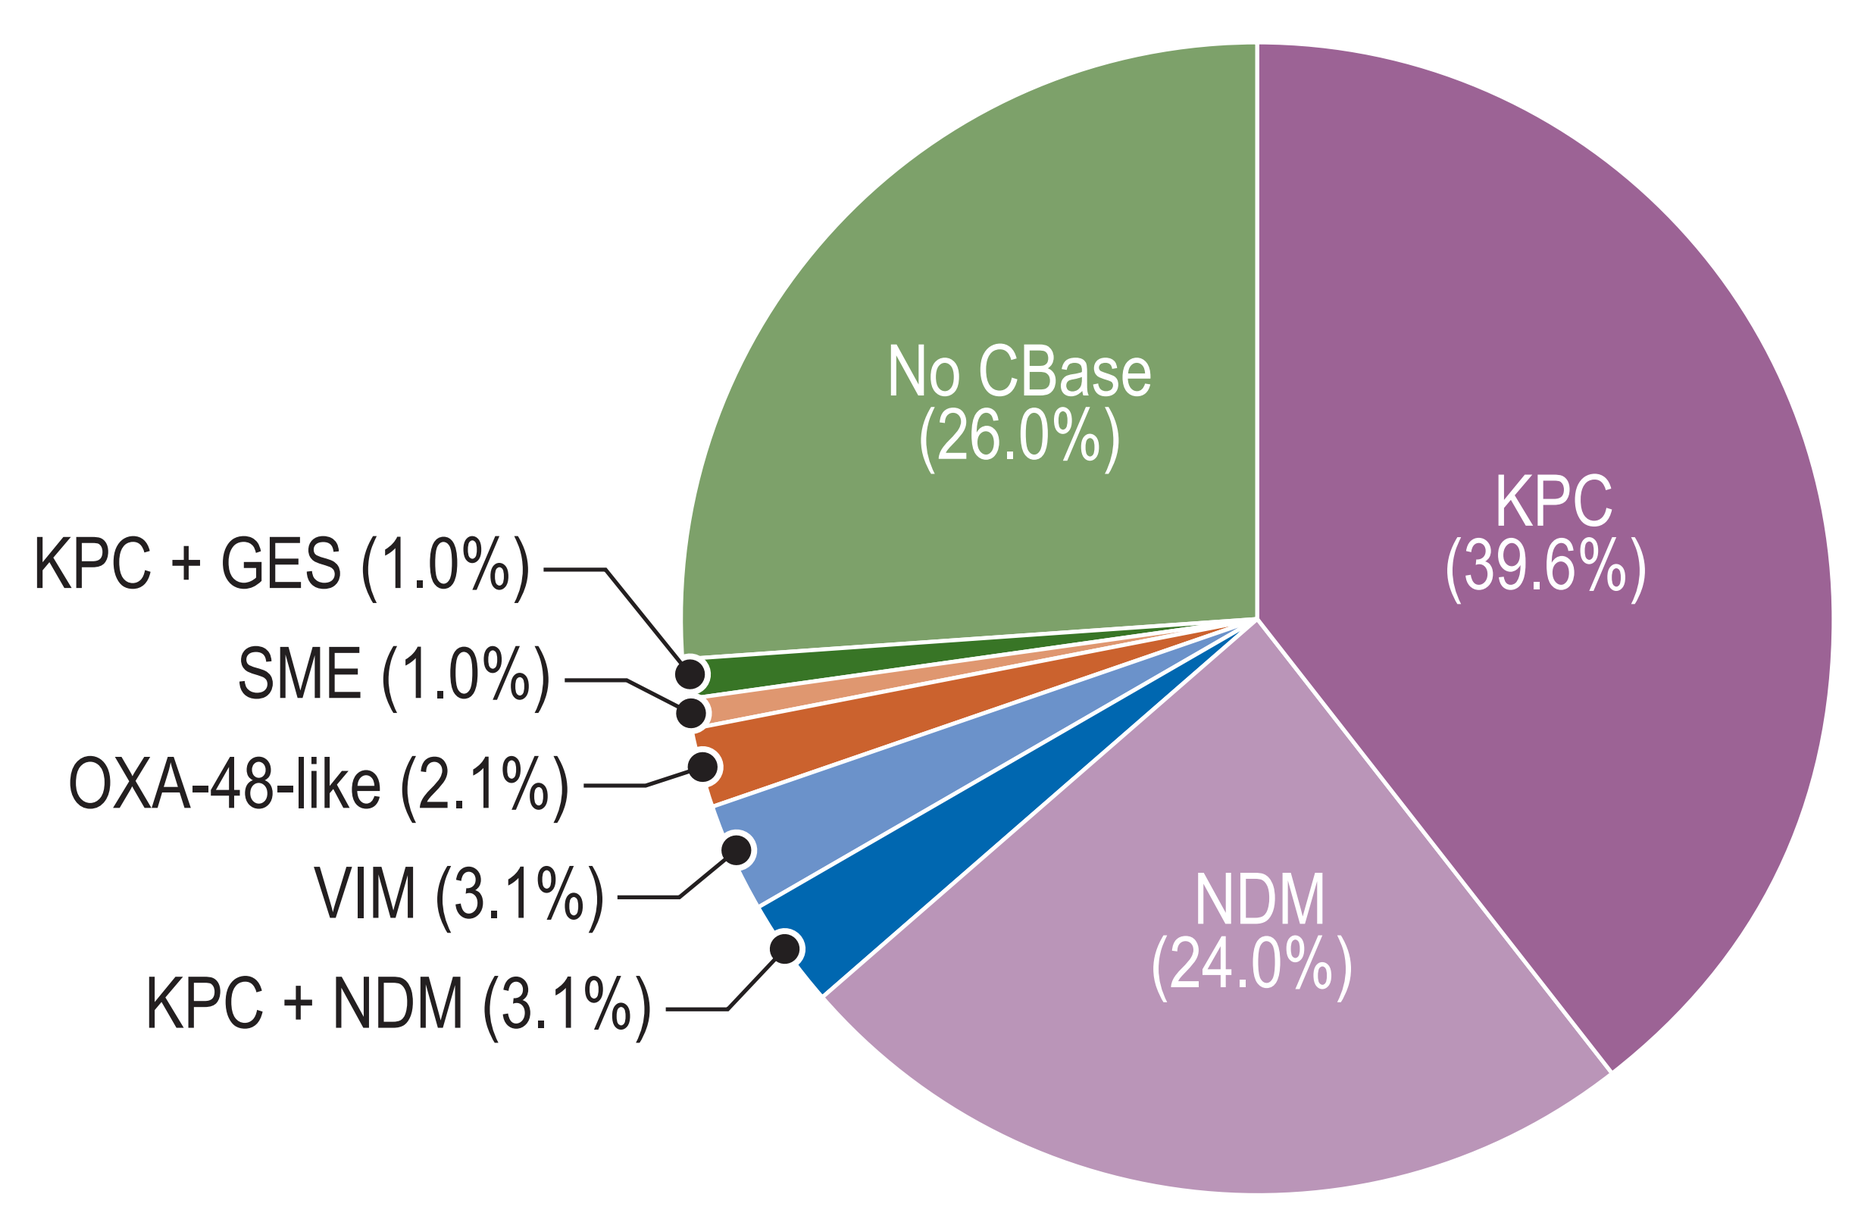
No CBase (26.0%)
KPC (39.6%)
NDM (24.0%)
KPC + GES (1.0%)
SME (1.0%)
OXA-48-like (2.1%)
VIM (3.1%)
KPC + NDM (3.1%)

METHODS

- Gram-negative bacilli (GNB) were consecutively collected from patients with pneumonia (1/patient) in 69 US medical centers in 2023–2025 via the INFORM Program.
- The organism collection included 4,474 Enterobacterales, 2,807 *Pseudomonas aeruginosa*, 779 *Stenotrophomonas maltophilia*, and 25 *Burkholderia cepacia* complex; other non-fermentative GNB were not evaluated.
- Only bacterial isolates determined to be significant by local criteria as the reported probable cause of infection were included in the study.
- Isolates were susceptibility tested by Clinical and Laboratory Standards Institute (CLSI) M07 broth microdilution methods at a monitoring laboratory (JMI Laboratories).
- Cefiderocol was tested in iron-depleted media against CRE only.
- CRE was defined as resistant to meropenem and/or imipenem and multidrug-resistant (MDR) as nonsusceptible to ≥1 agent in ≥3 antimicrobial classes.
- CRE isolates were screened for carbapenemases (CBases) by whole genome sequencing.

RESULTS

- ATM-AVI was active against 99.9% of Enterobacterales and retained activity against 96.9% of CRE (*n*=96) and 99.7% of MDR (*n*=1,203) Enterobacterales (Table 1).
- Ceftazidime-avibactam (CAZ-AVI) and meropenem-vaborbactam (MEM-VAB) were active against 99.2% and 99.3% of Enterobacterales, respectively, but exhibited limited activity against CREs (67.7% and 66.7% susceptible, respectively; Table 1).
- Cefiderocol was active against 94.8% of CREs, whereas amikacin and gentamicin inhibited only 74.0% and 67.7% of CREs, respectively (Table 1).
- The most common CBases among CREs were KPC (43.8% of CREs), NDM (27.1%), and VIM (3.1%; Figure 1).
- An MBL was identified in 30.2% of CREs (Figure 1).
- A CBase gene was not identified in 26.0% of CREs while 4 isolates (4.2%) had 2 CBase genes (Figure 1).
- ATM-AVI was active against 100.0% of KPC and MBL producers (Figure 2).
- CAZ-AVI and MEM-VAB were highly active against KPC producers but showed limited activity against MBL producers (Figure 2).
- The most active agents against *P. aeruginosa* were CAZ-AVI (96.1% susceptible) and ceftolozane-tazobactam (96.2% susceptible; Table 1).
- The most active agents against *S. maltophilia* were ATM-AVI (92.9%/99.6% inhibited at ≤4/≤8 mg/L), cefiderocol (98.7% susceptible), and trimethoprim-sulfamethoxazole (96.7% susceptible; Table 2).

Figure 2. Antimicrobial susceptibility of CRE isolates stratified by carbapenemase type

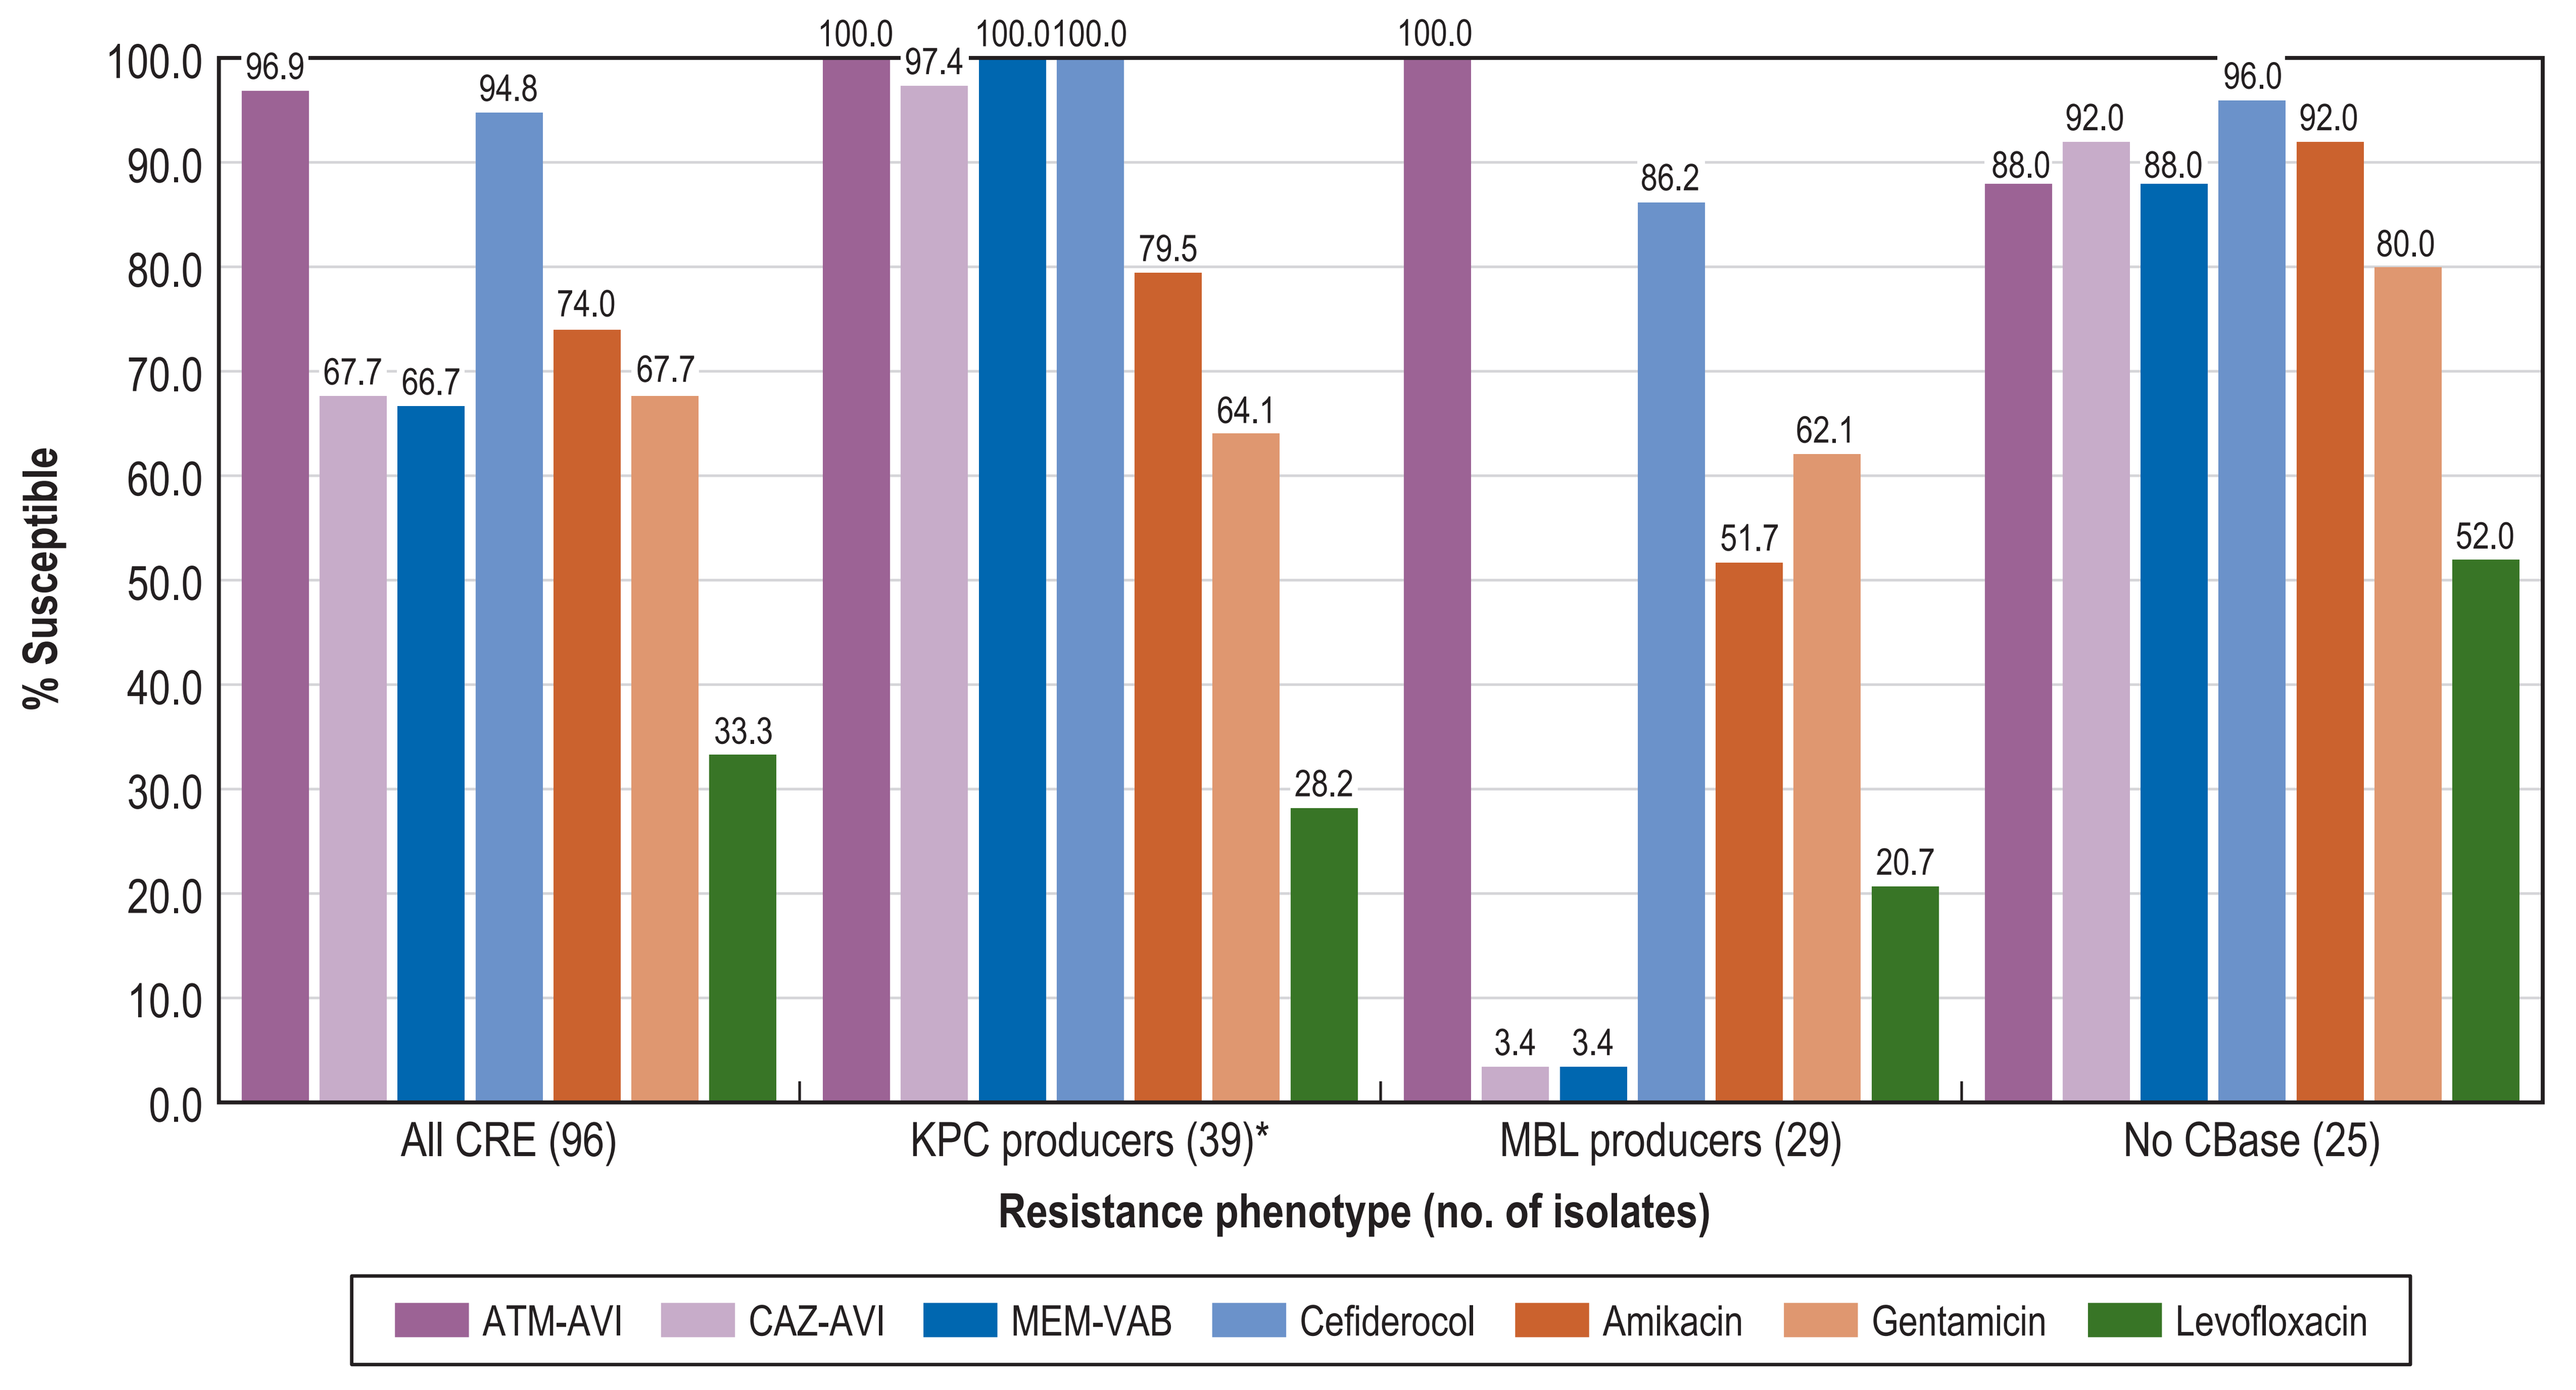
% Susceptible
100.0
90.0
80.0
70.0
60.0
50.0
40.0
30.0
20.0
10.0
0.0
96.9
67.7
66.7
94.8
74.0
67.7
33.3
100.0
97.4
100.0
100.0
79.5
64.1
28.2
100.0
3.4
3.4
86.2
51.7
62.1
20.7
88.0
92.0
88.0
96.0
92.0
80.0
52.0
All CRE (96)
KPC producers (39)*
MBL producers (29)
No CBase (25)
Resistance phenotype (no. of isolates)
ATM-AVI
CAZ-AVI
MEM-VAB
Cefiderocol
Amikacin
Gentamicin
Levofloxacin

* Exclude MBL co-producers.

Contact Information

Helio S. Sader, MD, PhD
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: helio.sader@element.com

Scan QR code or utilize the following link to download an electronic version of this presentation and other AbbVie IDWeek 2026 scientific presentations:

https://www.jmilabs.com/data/posters/IDWeek2026_26-ALG-02_P6_ATM-AVI_Pneumo.pdf

To submit a medical question, please visit www.abbviemedinfo.com

Scan Me

Acknowledgements

AbbVie funded this study and participated in the study design, research, analysis, data collection, interpretation of data, reviewing, and approval of the publication. All authors had access to relevant data and participated in the drafting, review, and approval of this publication. No honoraria or payments were made for authorship.

References

1. CLSI. 2026. M100Ed36. Performance standards for antimicrobial susceptibility testing: 36th informational supplement. Clinical and Laboratory Standards Institute.
2. Sader HS, Kimbrough JH, Doyle TB *et al.* Frequency, antimicrobial susceptibility, and molecular characterization of carbapenem-resistant Enterobacterales stratified by United States census divisions: Results from the INFORM program (2018–2022). *Open Forum Infect Dis* 2025; 12: ofaf005.
3. Sader HS, Mendes RE, Ryan Arends SJ, *et al.* Activity of Aztreonam-avibactam and other beta-lactamase inhibitor combinations against Gram-negative bacteria isolated from patients hospitalized with pneumonia in United States medical centers (2020–2022). *BMC Pulm Med.* 2025;25(1):38.
4. Tamma PD, Heil EL, Justo JA *et al.* Infectious Diseases Society of America 2024 guidance on the treatment of antimicrobial-resistant Gram-negative infections. *Clin Infect Dis* 2024 ciae403.

abbvie