IDWeek 2026 | Poster #P-18

Activity of manogepix against mold isolates across geographic regions

Marisa L. Winkler, Samuel Edeker, Abby Klauer, Paul R. Rhomberg, Mariana Castanheira
Element Materials Technology (JMI Laboratories), North Liberty, Iowa, USA

Introduction

- Fosmanogepix is the prodrug of manogepix, a novel antifungal agent targeting the Gwt1 enzyme.
 - This is a different target and mechanism of action relative to azoles, echinocandins, and amphotericin B.
- Manogepix has activity against both yeasts and molds.
 - It is currently in phase 3 clinical trials for the treatment of invasive candidiasis and mold infections.
- The broad spectrum of activity of manogepix includes mold isolates from patients with no or few other treatment options such as *Fusarium* spp. and *Scedosporium*/*Lomentospora* spp.
- Here, we look at the *in vitro* activity of manogepix against molds from different geographic regions between 2017 and 2024.

Methods

- A total of 2,661 mold isolates were collected from 67 different hospitals in 25 countries.
 - 6 countries in the Asia-Pacific region (APAC), 15 countries in Europe, 2 countries in Latin America (LATAM), and 2 countries in North America (NA) (Figure 1).
- Only 1 isolate per patient episode was included.
- All isolates were identified by MALDI-TOF MS and/or DNA sequencing.
- Isolates were tested by CLSI reference broth microdilution method (M38) for manogepix, voriconazole, posaconazole, and amphotericin B.
 - Minimum effective concentration (MEC) was read for manogepix; minimum inhibitory concentration (MIC) was read for all comparators.
- CLSI breakpoints (M38M51S) or epidemiological cutoff values (ECVs, M57S) were applied for comparator agents as available; no breakpoints or epidemiological cutoff values are available for manogepix.

Figure 2. Infection source of collected mold isolates

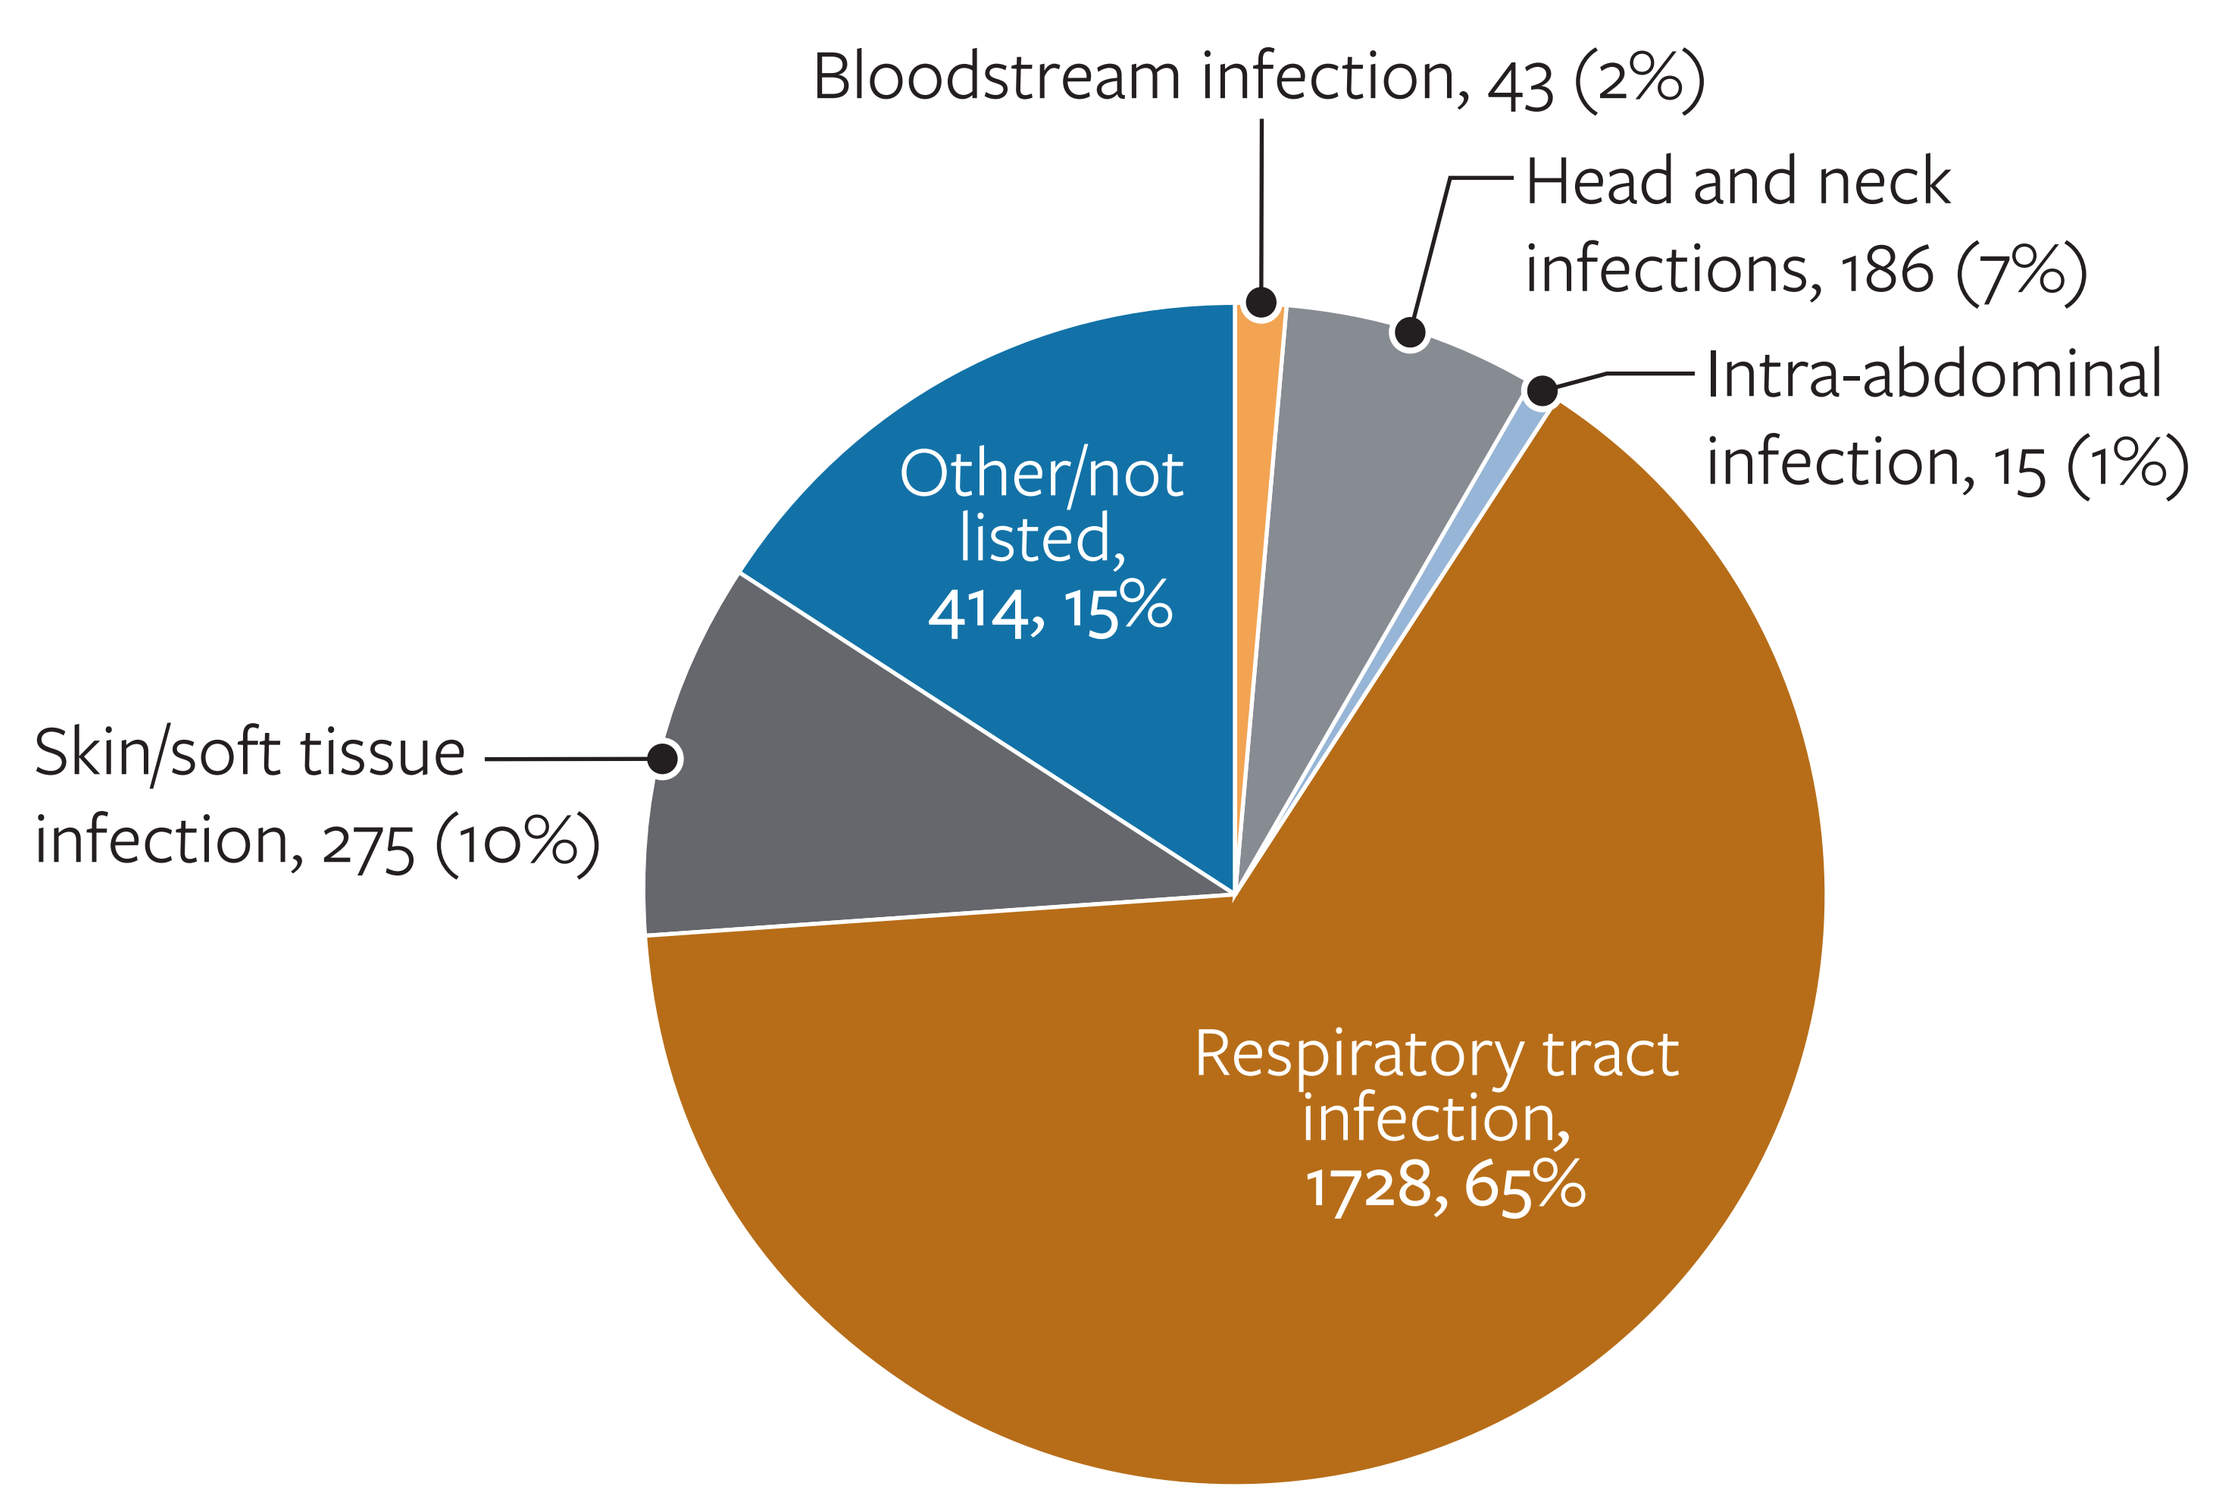
Bloodstream infection, 43 (2%)
Head and neck infections, 186 (7%)
Intra-abdominal infection, 15 (1%)
Other/not listed, 414, 15%
Skin/soft tissue infection, 275 (10%)
Respiratory tract infection, 1728, 65%

Results

- Isolate distribution was 40.9% Europe (1,087/2,661), 38.2% NA (1,017/2,661), 19.3% APAC (513/2,661), and 1.7% LATAM (44/2,661).
- There were 34 genera represented (Figure 1, Table 1).
 - *Aspergillus* spp. was the most common mold in all regions (78% APAC, 88% Europe, 88% LATAM, 78% NA).
 - Represented genera were less diverse in LATAM than in other regions, likely due to smaller number of isolates.
- Most isolates were from the respiratory tract (1,728/2,661, 64.9%), followed by other or no infection type listed (414/2,661, 15.6%), skin and soft tissue infection (275/2,661, 10.3%), head and neck infections (186/2,661, 7.0%), bloodstream infections (43/2,661, 1.6%), and intra-abdominal infections (15/2,661, 0.6%) (Figure 2).
 - Infection sources were similar among all geographic regions with the exception of less diversity in LATAM (data not shown).
- The MEC/MIC range and MEC/$MIC_{50/90}$ were the same or within one doubling dilution for manogepix for *Aspergillus* spp., *Fusarium* spp., Mucorales, and *Scedosporium*/*Lomentospora* spp. across all regions (Table 1).
 - There was more regional variability in MECs and MICs in the "other mold" category likely due to different genera and species.
- For non-Mucorales molds, manogepix $MEC_{50/90}$ was >32 times lower than the $MIC_{50/90}$ for comparator agents across all regions.
 - Against these organisms manogepix MEC_{50} was ≤0.008 to 0.03 mg/L and MEC_{90} was 0.03 to 0.5 mg/L; for comparator agents MIC_{50} ranged from 0.5 to >8 mg/L and MIC_{90} ranged from 2 to >8 mg/L.
- For Mucorales, MIC_{50} was lower for posaconazole than MEC_{50} for manogepix, but MIC/MEC_{90} were similar for the two agents across all regions.
- Amphotericin B $MIC_{50/90}$ was lower than $MEC_{50/90}$ for manogepix against Mucorales in all regions.

Figure 1. Geography of collected isolates

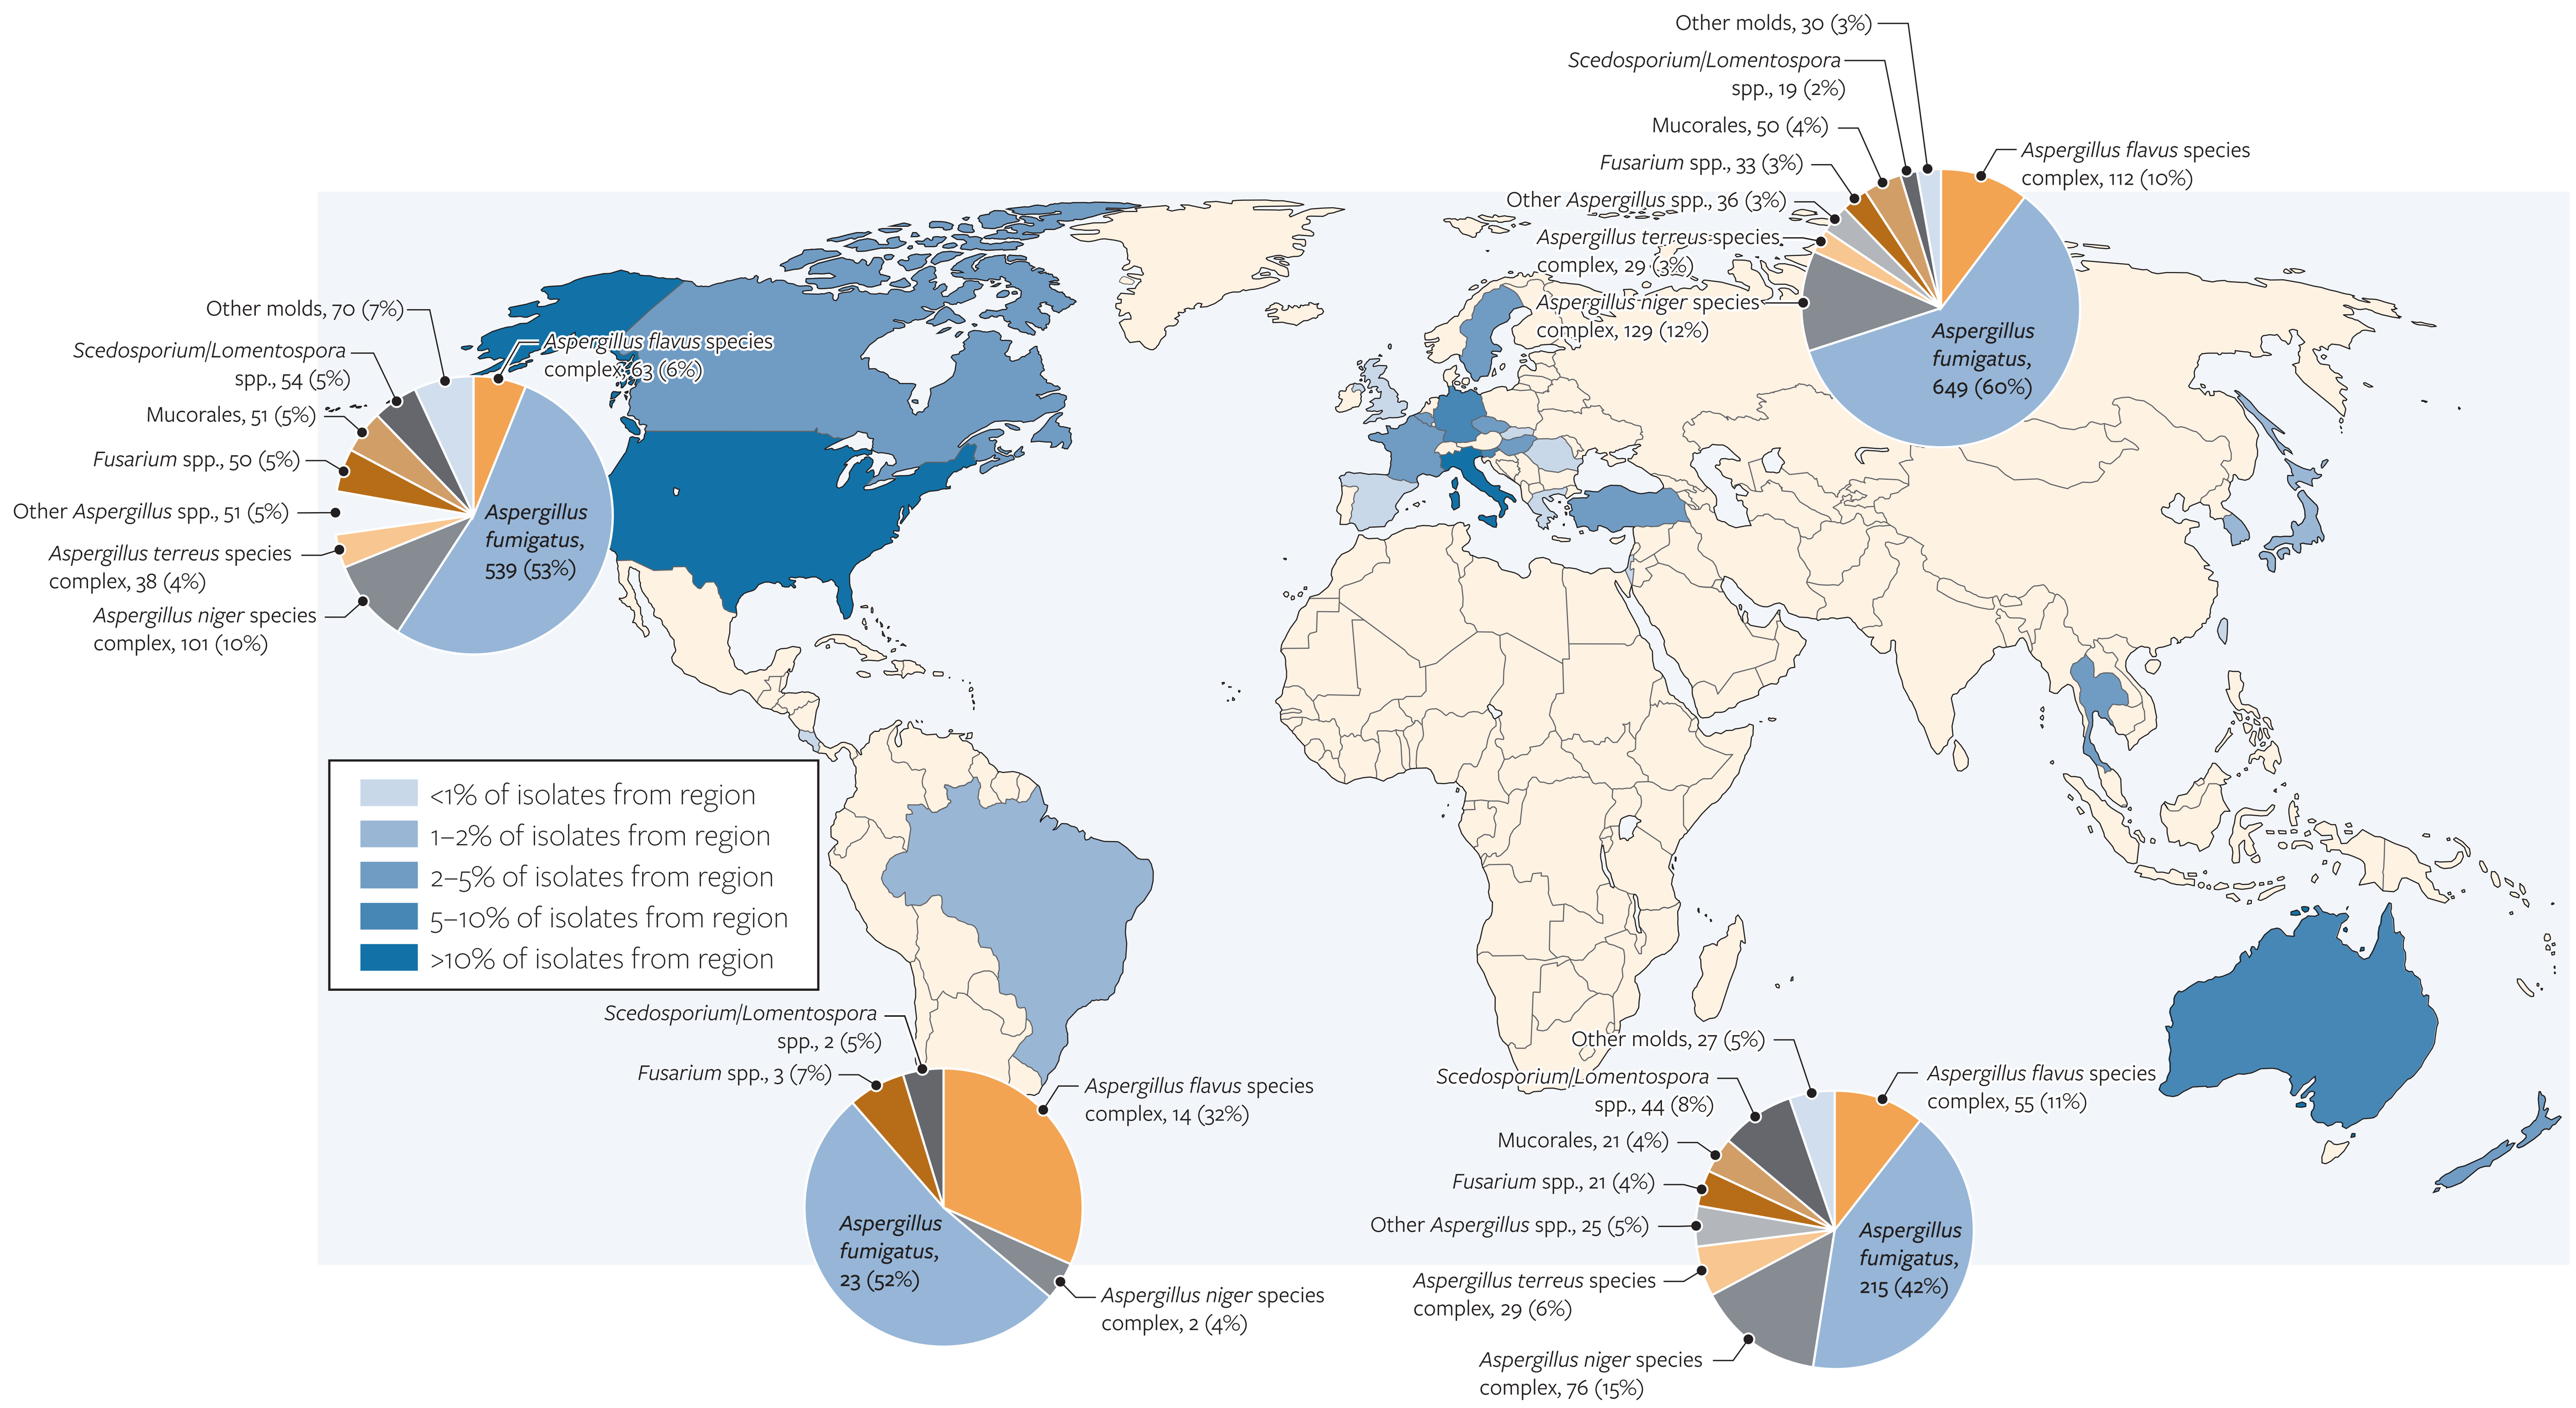
Other molds, 70 (7%)
Scedosporium/Lomentospora spp., 54 (5%)
Mucorales, 51 (5%)
Fusarium spp., 50 (5%)
Other Aspergillus spp., 51 (5%)
Aspergillus terreus species complex, 38 (4%)
Aspergillus niger species complex, 101 (10%)
Aspergillus flavus species complex, 63 (6%)
Aspergillus fumigatus, 539 (53%)
Other molds, 30 (3%)
Scedosporium/Lomentospora spp., 19 (2%)
Mucorales, 50 (4%)
Fusarium spp., 33 (3%)
Other Aspergillus spp., 36 (3%)
Aspergillus terreus species complex, 29 (3%)
Aspergillus niger species complex, 129 (12%)
Aspergillus flavus species complex, 112 (10%)
Aspergillus fumigatus, 649 (60%)
<1% of isolates from region
1–2% of isolates from region
2–5% of isolates from region
5–10% of isolates from region
>10% of isolates from region
Scedosporium/Lomentospora spp., 2 (5%)
Fusarium spp., 3 (7%)
Aspergillus flavus species complex, 14 (32%)
Aspergillus fumigatus, 23 (52%)
Aspergillus niger species complex, 2 (4%)
Other molds, 27 (5%)
Scedosporium/Lomentospora spp., 44 (8%)
Mucorales, 21 (4%)
Fusarium spp., 21 (4%)
Other Aspergillus spp., 25 (5%)
Aspergillus terreus species complex, 29 (6%)
Aspergillus niger species complex, 76 (15%)
Aspergillus flavus species complex, 55 (11%)
Aspergillus fumigatus, 215 (42%)

Table 1. MIC/MEC range and MIC/$MEC_{50/90}$ (mg/L) for manogepix, voriconazole, posaconazole, and amphotericin B in molds across geographic regions

	manogepix		voriconazole		posaconazole		amphotericin B	
Organism (n)	MEC range	$MEC_{50/90}$	MIC range	$MIC_{50/90}$	MIC range	$MIC_{50/90}$	MIC range	$MIC_{50/90}$
All organisms (2,661)	≤0.008 – >4	0.015/0.06	0.015 – >8	0.5/4	≤0.008 – >8	0.25/2	≤0.03 – >2	1/2
All organisms APAC (513)	≤0.008 – >4	0.015/0.06	0.03 – >8	0.5/8	0.015 – >8	0.25/>8	≤0.03 – >2	1/>2
All organisms EU (1,087)	≤0.008 – >4	0.015/0.03	0.03 – >8	0.5/4	0.03 – >8	0.25/1	≤0.03 – >2	1/2
All organisms LATAM (44)	≤0.008 – 0.06	0.015/0.03	0.25 – 8	0.5/1	0.06 – >8	0.25/1	0.25 – 2	1/2
All organisms NA (1,016)	≤0.008 – >4	0.015/0.06	0.015 – >8	0.5/8	≤0.008 – >8	0.25/4	≤0.03 – >2	1/2
All *Aspergillus* spp. (2,186)	≤0.008 – 0.25	0.015/0.03	0.03 – >8	0.5/1	0.015 – >8	0.25/0.5	≤0.03 – >2	1/2
Aspergillus APAC (400)	≤0.008 – 0.06	0.015/0.03	0.03 – >8	0.5/1	0.015 – >8	0.25/0.5	≤0.03 – >2	1/2
Aspergillus EU (955)	≤0.008 – 0.25	0.015/0.03	0.06 – >8	0.5/1	0.03 – >8	0.25/0.5	0.06 – >2	1/2
Aspergillus LATAM (39)	≤0.008 – 0.06	0.015/0.03	0.25 – 1	0.5/1	0.06 – 0.5	0.25/0.5	0.25 – 2	1/2
Aspergillus NA (792)	≤0.008 – 0.25	0.015/0.03	0.03 – >8	0.5/1	0.03 – >8	0.25/0.5	0.06 – >2	1/2
All *Fusarium* spp. (107)	≤0.008 – >4	0.015/0.06	2 – >8	8/>8	0.5 – >8	>8/>8	0.12 – >2	2/2
Fusarium APAC (21)	≤0.008 – 0.12	0.015/0.06	2 – >8	4/>8	1 – >8	>8/>8	0.12 – >2	1/2
Fusarium EU (33)	≤0.008 – 0.06	0.015/0.03	2 – >8	8/>8	0.5 – >8	>8/>8	0.25 – >2	2/>2
Fusarium LATAM (3)	0.008 – 0.015	0.015/*	2 – 8	4/*	1 – >8	>8/*	1 – 2	2/*
Fusarium NA (50)	≤0.008 – >4	0.015/0.06	2 – >8	8/>8	0.5 – >8	>8/>8	0.12 – >2	2/2
All Mucorales (122)	0.008 – >4	4/>4	0.5 – >8	>8/>8	0.06 – >8	0.5/>8	≤0.03 – >2	0.5/1
Mucorales APAC (21)	0.25 – >4	>4/>4	0.5 – >8	>8/>8	0.25 – >8	1/4	≤0.03 – 2	0.25/1
Mucorales EU (50)	0.008 – >4	4/>4	0.5 – >8	>8/>8	0.06 – >8	0.5/>8	0.06 – 2	0.5/1
Mucorales NA (51)	0.25 – >4	4/>4	2 – >8	>8/>8	0.12 – >8	1/>8	≤0.03 – 2	0.5/1
All SCED/LOMENT (119)	≤0.008 – 0.5	0.03/0.06	0.25 – >8	1/>8	0.5 – >8	>8/>8	0.25 – >2	>2/>2
SCED/LOMENT APAC (44)	≤0.008 – 0.12	0.015/0.06	0.25 – >8	1/>8	0.5 – >8	>8/>8	0.25 – >2	>2/>2
SCED/LOMENT EU (19)	0.008 – 0.06	0.03/0.06	0.5 – >8	1/2	1 – >8	4/>8	0.5 – >2	>2/>2
SCED/LOMENT LATAM (2)	0.015	0.015/*	1	1/*	4	4/*	0.5 – 1	0.5/*
SCED/LOMENT NA (54)	≤0.008 – 0.5	0.03/0.06	0.25 – >8	1/>8	0.5 – >8	4/>8	0.5 – >2	>2/>2
All other molds (127)	≤0.008 – >4	≤0.008/0.5	0.015 – >8	1/>8	≤0.008 – >8	0.5/>8	≤0.03 – >2	1/>2
Other molds APAC (27)[1]	≤0.008 – 1	0.015/0.5	0.12 – >8	0.5/>8	0.06 – >8	0.5/>8	0.06 – >2	0.5/>2
Other molds EU (30)[2]	≤0.008 – 0.06	≤0.008/0.015	0.03 – >8	2/>8	0.06 – >8	0.25/2	≤0.03 – >2	1/>2
Other molds NA (70)[3]	≤0.008 – >4	≤0.008/0.5	0.015 – >8	0.5/>8	≤0.008 – >8	0.5/>8	≤0.03 – >2	1/>2

*MIC_{50} only as < 10 organisms represented
[1] Genera include *Curvularia, Exophiala, Medicopsis, Monascus, Paecilomyces, Penicillium, Phaeoacremonium, Pleurostomophora, Purpureocillium, Rasamsonia, Sarocladium, Scopularipsis, Verruconis*
[2] Genera include *Aureobasidium, Exophiala, Paecilomyces, Penicillium, Rasamsonia, Trichoderma*
[3] Genera include *Acremonium, Alternaria, Aureobasidium, Chaetomium, Coprinellus, Coprinopsis, Curvularia, Exophiala, Medicopsis, Microascus, Paecilomyces, Penicillium, Pseudopithomyces, Purpureocillium, Rasamsonia, Sarocladium, Scopularipsis, Trichoderma, Verruconis*

Conclusions

- Manogepix is highly active *in vitro* against diverse mold species across geographic regions.
- There was significant diversity among genera of mold collected in all regions of the world except for LATAM.
 - This was likely due to the smaller number of collected isolates.
- The only regional differences observed among susceptibility patterns to manogepix or comparator agents was among less common molds.
 - This was likely due to different genera and species among this organism group in the different geographic regions.
- Continued development of fosmanogepix for the treatment of invasive mold infections worldwide is warranted based on these *in vitro* data.

Funding

This study was supported by Basilea Pharmaceutica International Ltd and this project has been funded in part with federal funds from the U.S. Department of Health and Human Services (HHS); Administration of Strategic Preparedness and Response (ASPR); Biomedical Advanced Research and Development Authority (BARDA), under OT number: 75A50124C00033. The contract and federal funding are not an endorsement of the study results, product, or company.

ML Winkler, S Edeker, A Klauer, PR Rhomberg, and M Castanheira were employees of Element Materials Technology (JMI Laboratories) at the time of this study, which was a paid consultant to Basilea in connection with the development of this poster.

Acknowledgments

The authors thank all of the SENTRY Program participants for providing the isolates used in this study.

References

CLSI, *M38M51S Ed4. Performance standards for antifungal susceptibility testing of molds*. 2026, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M57S Ed4. Epidemiological cutoff values for antifungal susceptibility testing*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M38 Ed3. Reference method for broth dilution antifungal susceptibility testing of filamentous fungi*. 2017, Clinical and Laboratory Standards Institute: Wayne, PA.

Contact

Marisa Winkler, MD, PhD
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: marisa.winkler@element.com

Scan Me

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2026_26-BAS-01_P2_Mano_Moulds_Across_Regions.pdf

Charges may apply. No personal information is stored.

IDWeek 2026, October 21–24, 2026, Washington, D.C.