IDWeek 2026 | Poster #P-17

Activity of manogepix against *Aspergillus* spp. isolates with CYP51 alterations

Marisa L. Winkler, Samuel Edeker, Abby Klauer, Paul R. Rhomberg, Maura Karr, John H. Kimbrough, Mariana Castanheira

Element Materials Technology (JMI Laboratories), North Liberty, Iowa, USA

Introduction

- Fosmanogepix is the prodrug of manogepix, a novel antifungal agent targeting the Gwt1 enzyme.
 - This is a different target and mechanism of action relative to azoles, echinocandins, and amphotericin B.
- Fosmanogepix is currently in Phase 3 clinical trials for the treatment of invasive candidiasis and invasive mold infections.
- Given its novel mechanism of action, manogepix activity is not expected to be affected by typical resistance mechanisms that reduce susceptibility to other antifungal agents.
- Here, we look at the *in vitro* activity of manogepix against *Aspergillus* spp. isolates with identified CYP51 alterations leading to reduced azole susceptibility.

Methods

- Isolates were collected from the United States as a part of an antifungal surveillance program between 2017 and 2024.
 - There were 740 total isolates tested from all 9 Census regions and 18 states.
 - Isolates predominated from New England, Middle Atlantic, and East North Central with a smaller number from other regions (Figure 1).
- Only 1 isolate per patient episode was included.
- All isolates were identified by MALDI-TOF MS and/or DNA sequencing.
- Isolates were tested by CLSI reference broth microdilution method (M38) for manogepix, voriconazole, itraconazole, and amphotericin B.
 - Minimum effective concentration (MEC) was read for manogepix; minimum inhibitory concentration (MIC) was read for all comparators.
- CLSI breakpoints (M38M51S) or epidemiological cutoff values (ECVs, M57S) were applied for comparator agents as available; no breakpoints or epidemiological cutoff values are available for manogepix.
- Isolates that were non-wildtype (NWT) for ≥1 azole had CYP51A/B sequencing performed.

Results

- There were 16 *Aspergillus* spp. isolates with CYP51A or CYP51B alterations identified.
 - These were from 7 states and 6 Census regions (West South Central, East South Central, South Atlantic, Middle Atlantic, New England, and East North Central; Figure 2).
 - The most isolates were from Virginia (6) and Indiana (5) with 1 each from other states.
- The itraconazole MICs were 1 to >8 mg/L, voriconazole 0.5 to >8 mg/L, amphotericin B 0.25 to 4 mg/L (Table 1).
 - 15 of the isolates (93.8%) were NWT to itraconazole, 5 (31.3%) were NWT to voriconazole, and 1 (6.3%) was NWT to amphotericin B.
 - There were 3 isolates that were intermediate to voriconazole by breakpoint but WT by ECV.
- Isolates with CYP51 alterations were 12 *A. fumigatus*, one *A. brasiliensis*, one *A. niger*, one *A. welwitschiae*, and one *A. tubingensis*.
- All 16 isolates had CYP51A point alterations, but no promoter changes were observed. Only one *A. niger* isolate had a CYP51B point alteration.
- Manogepix MECs were 0.008 to 0.06 mg/L among the isolates with CYP51 changes.
 - The manogepix MECs were similar regardless of genetic background or MICs to antifungal comparator agents.
 - Overall manogepix MECs were similar at $MEC_{50/90}$ 0.015/0.03 mg/L (data not shown).

Conclusions

- Single amino acid changes in CYP51A predominated among *Aspergillus* spp. isolates that were NWT to azole antifungal agents collected in the United States.
- Isolates had elevated MICs to itraconazole and voriconazole, but maintained low MECs to manogepix.
- Fosmanogepix is a promising agent for future development in the treatment of invasive mold infections including infections caused by molds containing resistance mechanisms to current first-line therapies.

References

CLSI, *M38M51S Ed4. Performance standards for antifungal susceptibility testing of molds*. 2026, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M57S Ed4. Epidemiological cutoff values for antifungal susceptibility testing*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M38 Ed3. Reference method for broth dilution antifungal susceptibility testing of filamentous fungi*. 2017, Clinical and Laboratory Standards Institute: Wayne, PA.

Funding

This study was supported by Basilea Pharmaceutica International Ltd and this project has been funded in part with federal funds from the U.S. Department of Health and Human Services (HHS); Administration of Strategic Preparedness and Response (ASPR); Biomedical Advanced Research and Development Authority (BARDA), under OT number: 75A50124C00033. The contract and federal funding are not an endorsement of the study results, product, or company.

ML Winkler, S Edeker, A Klauer, PR Rhomberg, M Karr, JH Kimbrough, and M Castanheira were employees of Element Materials Technology (JMI Laboratories) at the time of this study, which was a paid consultant to Basilea in connection with the development of this poster.

Acknowledgments

The authors thank all of the SENTRY Program participants for providing the isolates used in this study.

Contact

element

Scan Me

Marisa Winkler, MD, PhD
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: marisa.winkler@element.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2026_26-BAS-01_P3_Mano_Aspergillus_CYP_US.pdf

Charges may apply. No personal information is stored.

Table 1. MEC and MIC (mg/L) for manogepix (MGX), voriconazole (VRC), itraconazole (ITR), posaconazole (PSC), and amphotericin B (AmB) in *Aspergillus* spp. isolates with CYP51 alterations

Organism	CYP51A	CYP51B	MGX	ITR	VRC	AmB
Aspergillus brasiliensis	V6I, M53I, K246Q	wildtype	0.015	>8	8	0.25
Aspergillus fumigatus	I242V	wildtype	0.015	2	1	1
Aspergillus fumigatus	A9T	wildtype	0.03	2	0.5	2
Aspergillus fumigatus	I242V	wildtype	0.015	2	0.5	2
Aspergillus fumigatus	G448S	wildtype	0.015	>8	4	1
Aspergillus fumigatus	I242V	wildtype	0.015	2	0.5	4
Aspergillus fumigatus	F46Y, M172V, N248T, D255E, E427K	wildtype	0.015	2	0.5	2
Aspergillus fumigatus	I242V	wildtype	0.015	2	0.5	1
Aspergillus fumigatus	F46Y, M172V, N248T, D255E, E427K	wildtype	0.008	>8	2	1
Aspergillus fumigatus	I242V	wildtype	0.015	2	0.5	1
Aspergillus fumigatus	I242V	wildtype	0.03	4	1	1
Aspergillus fumigatus	I242V	wildtype	0.03	1	1	1
Aspergillus fumigatus	G448S	wildtype	0.06	>8	>8	0.25
Aspergillus welwitschiae	W221R, H382R, L383V	I285V	0.015	8	2	1
Aspergillus tubingensis	V17A	wildtype	0.06	>8	4	1
Aspergillus niger	T57A, S346R, I507S, M511L	wildtype	0.008	8	2	0.25

NWT (non-wildtype) values in green; blue shading reflects isolates intermediate but WT to voriconazole

Figure 1. Geography and source of all 740 collected isolates

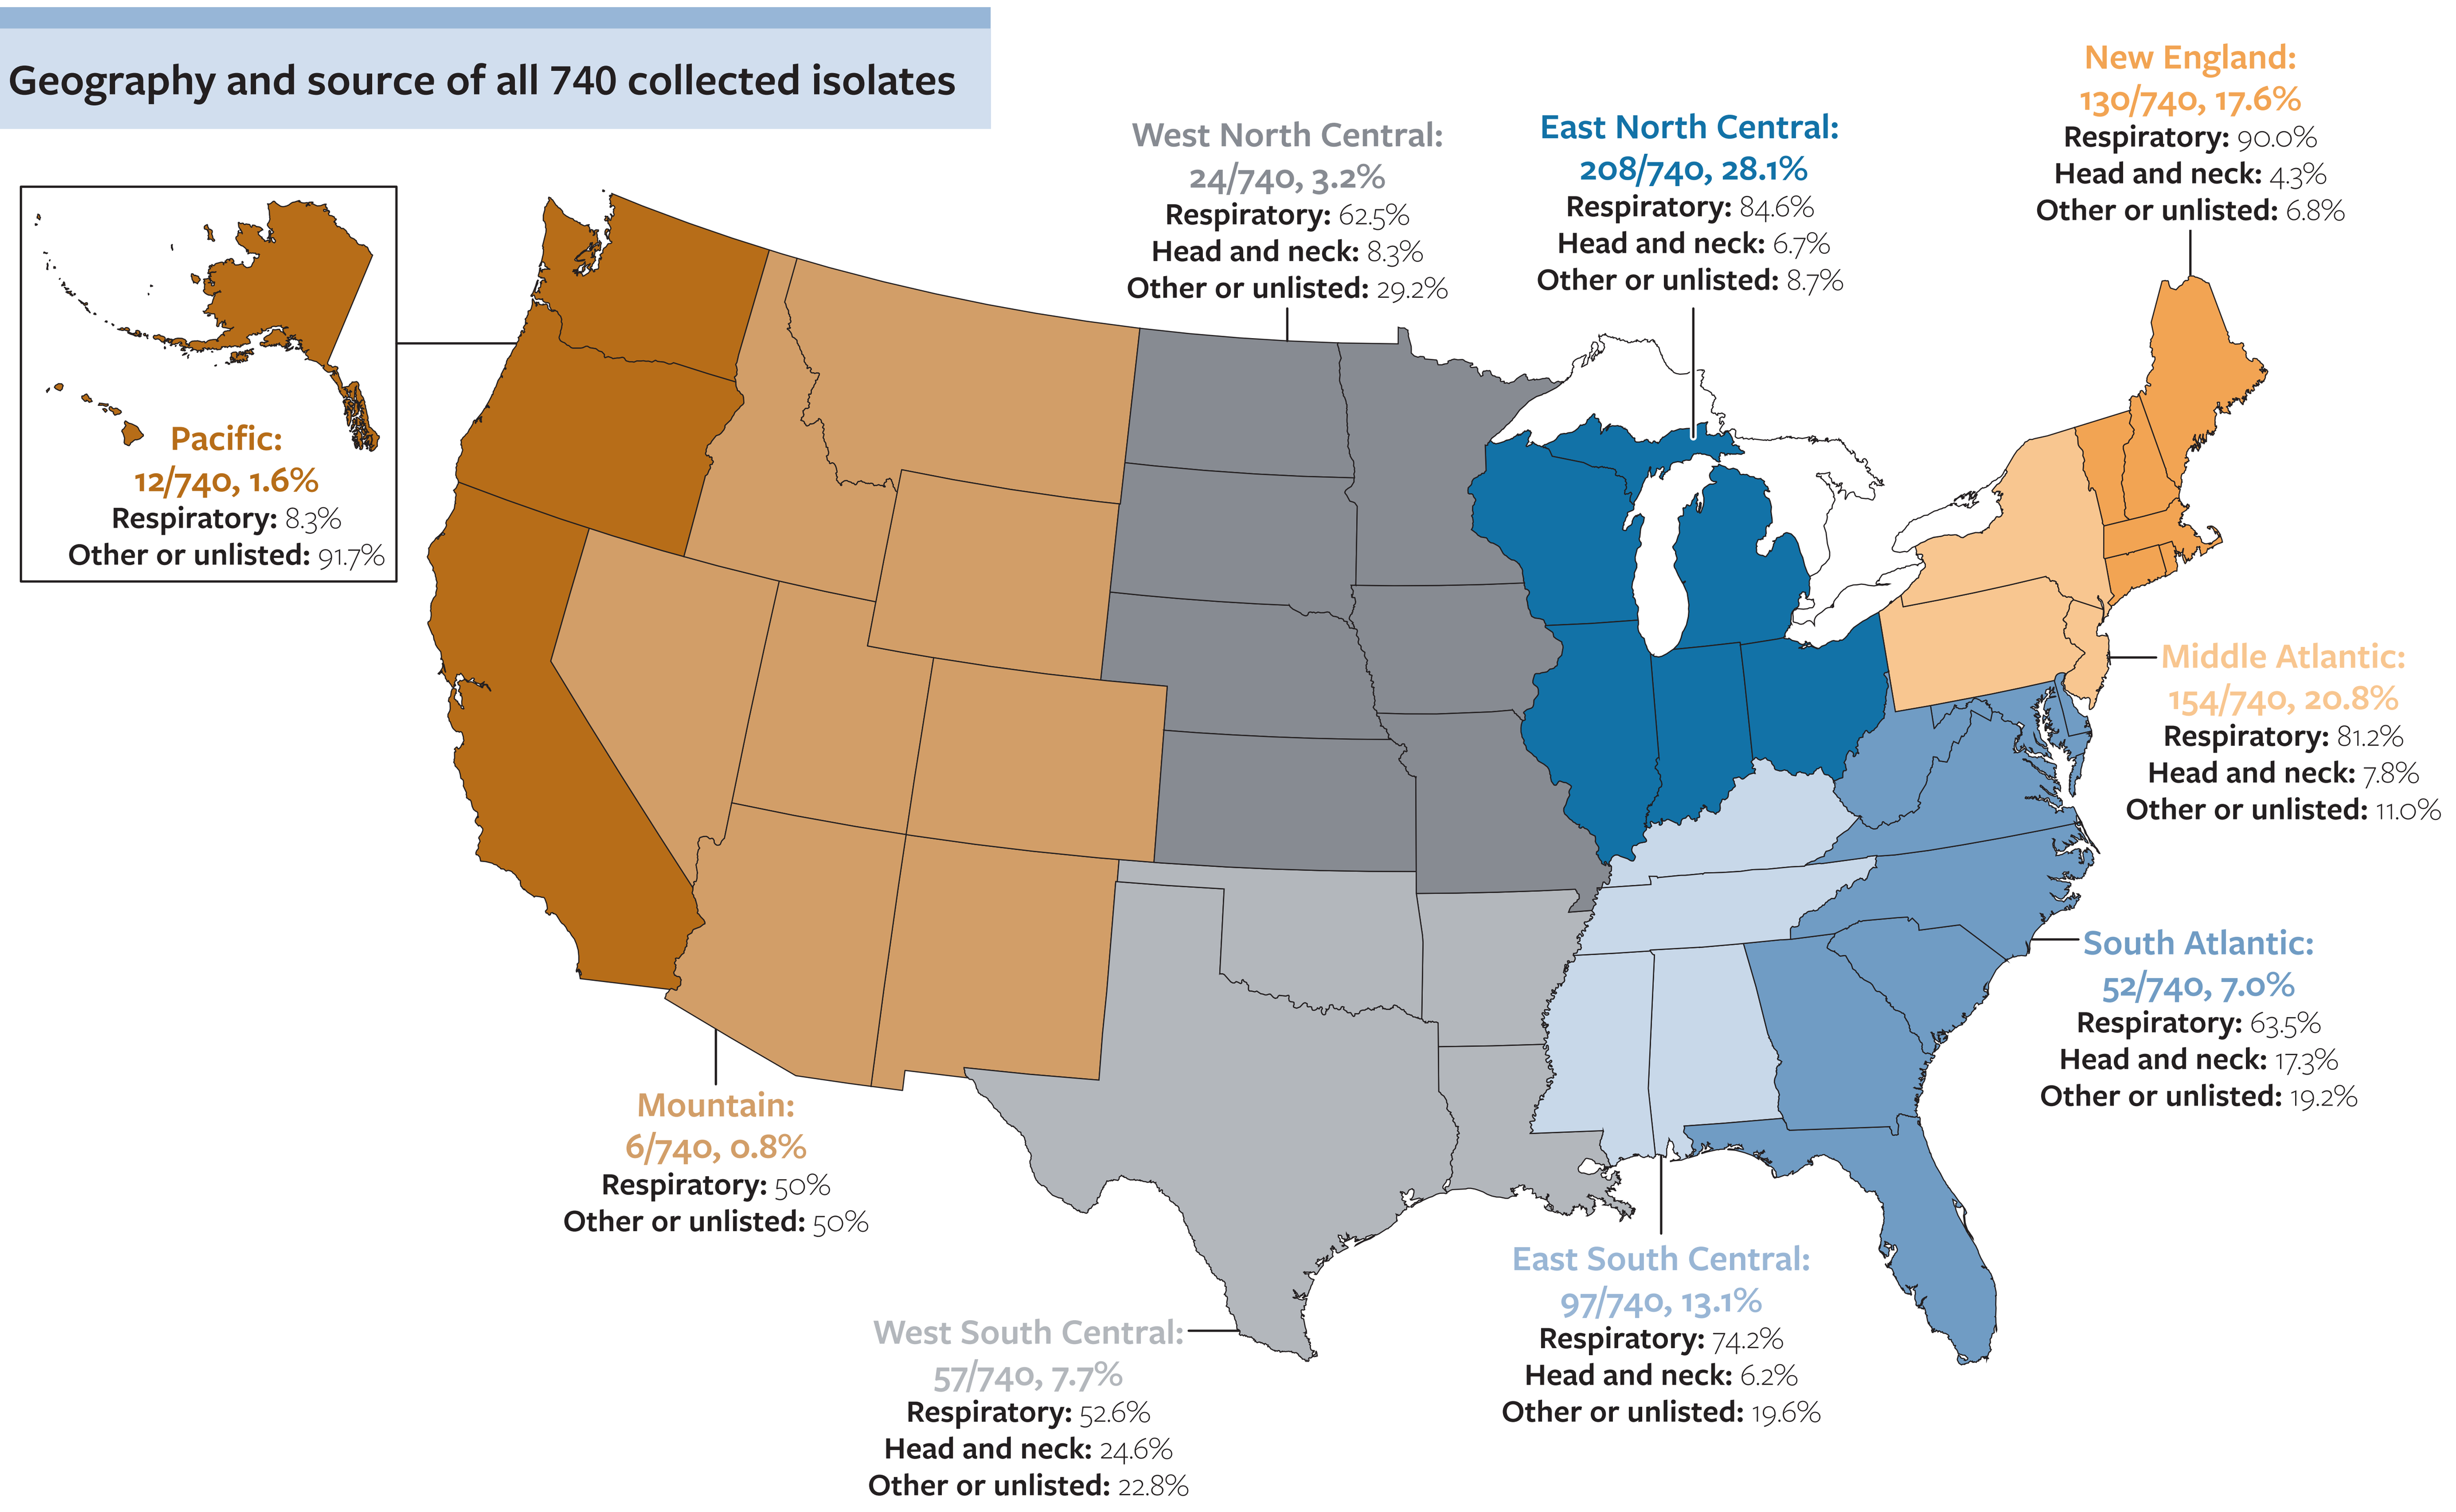
West North Central:
24/740, 3.2%
Respiratory: 62.5%
Head and neck: 8.3%
Other or unlisted: 29.2%
East North Central:
208/740, 28.1%
Respiratory: 84.6%
Head and neck: 6.7%
Other or unlisted: 8.7%
New England:
130/740, 17.6%
Respiratory: 90.0%
Head and neck: 4.3%
Other or unlisted: 6.8%
Pacific:
12/740, 1.6%
Respiratory: 8.3%
Other or unlisted: 91.7%
Middle Atlantic:
154/740, 20.8%
Respiratory: 81.2%
Head and neck: 7.8%
Other or unlisted: 11.0%
South Atlantic:
52/740, 7.0%
Respiratory: 63.5%
Head and neck: 17.3%
Other or unlisted: 19.2%
Mountain:
6/740, 0.8%
Respiratory: 50%
Other or unlisted: 50%
East South Central:
97/740, 13.1%
Respiratory: 74.2%
Head and neck: 6.2%
Other or unlisted: 19.6%
West South Central:
57/740, 7.7%
Respiratory: 52.6%
Head and neck: 24.6%
Other or unlisted: 22.8%

Figure 2. Geography of isolates with CYP alterations (states in blue) with shading reflective of number of isolates per state

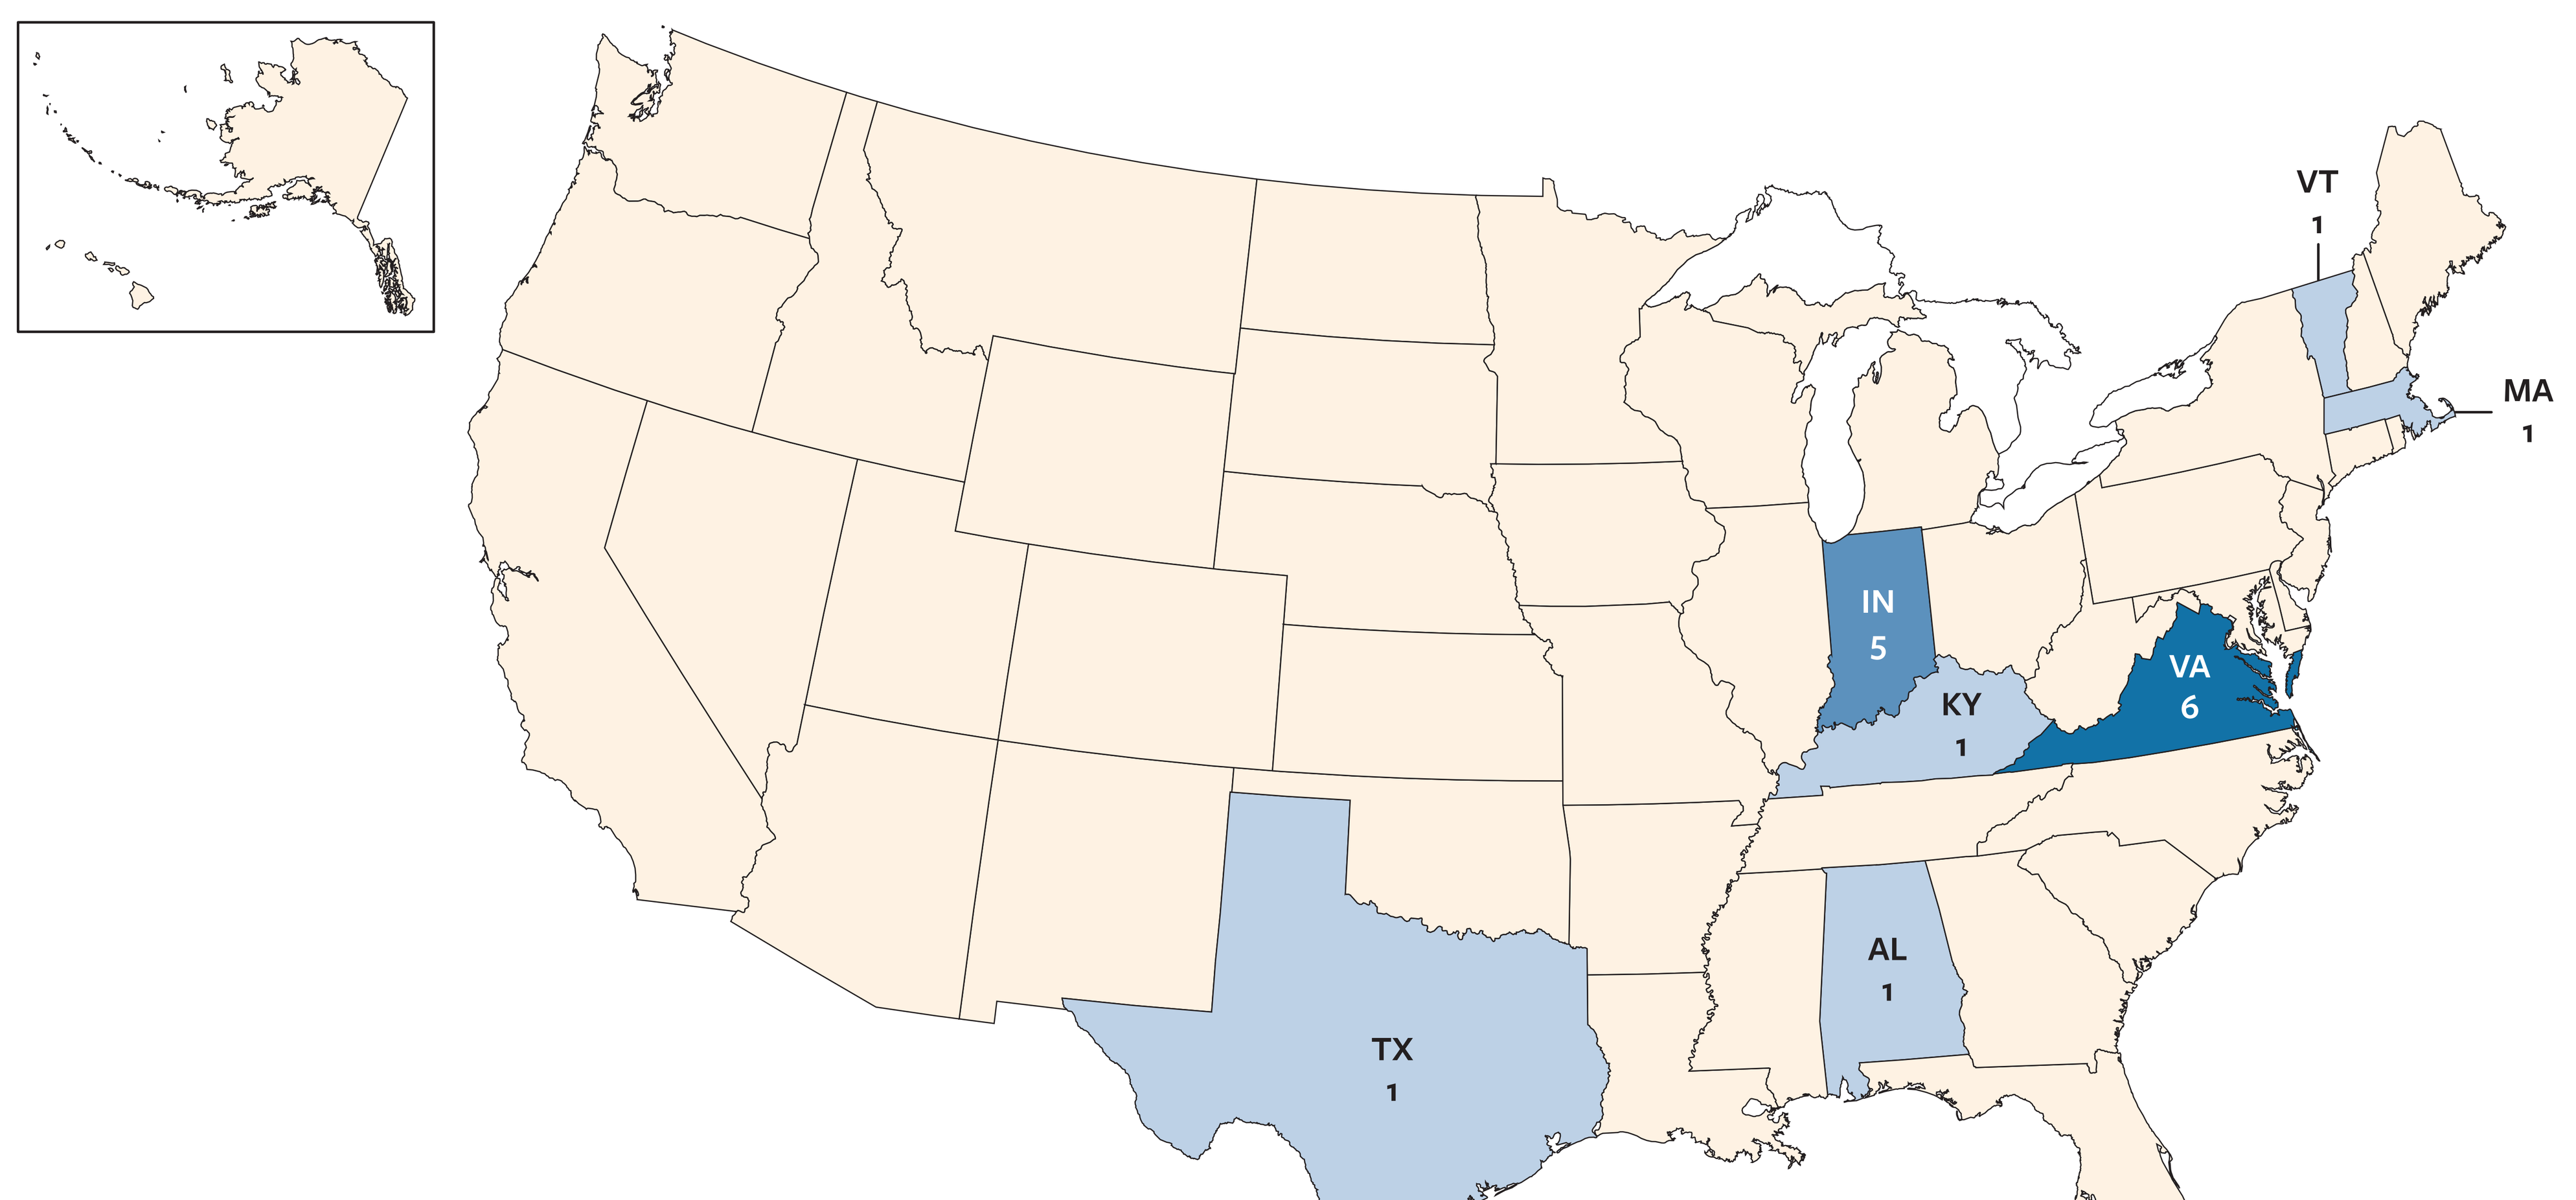
VT
1
MA
1
IN
5
KY
1
VA
6
AL
1
TX
1

IDWeek 2026, October 21–24, 2026, Washington, D.C.