IDWeek 2026 | Poster #P-19

Activity of manogepix against yeast and mold isolates from 2025 in the United States

Marisa L. Winkler, Samuel Edeker, Abby Klauer, Paul R. Rhomberg, Mariana Castanheira
Element Materials Technology (JMI Laboratories), North Liberty, Iowa, USA

Introduction

- Fosmanogepix is the prodrug of manogepix, a novel antifungal agent targeting the fungal Gwt1 enzyme.
 - Gwt1 is an acyltransferase in the glycosylphosphatidylinositol (GPI) biosynthesis pathway, which is required for anchoring of cell wall proteins in fungi.
- Manogepix has activity against both yeasts and molds.
 - It is currently in phase 3 clinical trials for the treatment of invasive candidiasis and mold infections.
- Here, we look at the *in vitro* activity of manogepix against yeast and molds from the United States collected as a part of a surveillance program in 2025.

Methods

- A total of 427 isolates from the United States were collected and tested.
 - 358 yeast and 69 molds.
- Only 1 isolate per patient episode was included.
- All isolates were identified by MALDI-TOF MS and/or DNA sequencing.
- Isolates were tested by CLSI reference broth microdilution method (M27 or M38) for manogepix, micafungin (yeast only), fluconazole (yeast only), itraconazole (molds only), voriconazole, and amphotericin B.
 - Minimum effective concentration (MEC) was read for manogepix against molds; minimum inhibitory concentration (MIC) was read for all comparators against yeast and molds and for manogepix against yeast.
- CLSI breakpoints (M27M44S, M38M51S) or epidemiological cutoff values (ECVs, M57S) were applied for comparator agents as available; no breakpoints or epidemiological cutoff values are available for manogepix.

Contact

Marisa Winkler, MD, PhD
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: marisa.winkler@element.com

Scan Me

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2026_26-BAS-01_P4_Mano_Yeast_Mould_US_2025.pdf

Charges may apply. No personal information is stored.

Results

- Isolates were from all 9 U.S census divisions (Figure 1).
 - There were no molds from census divisions 4 (West North Central) or 6 (East South Central).
- There were 18 yeast species (Table 1) and 15 mold species represented (Table 2).
- Yeast isolates were from bloodstream infections (244/358, 68.2%), skin and soft tissue infections (39/358, 10.9%), intra-abdominal infections (23/358, 6.4%), respiratory tract infections (22/358, 6.1%), head and neck infections (7/358, 2.0%), urinary tract infections (6/358, 1.7%), or other infection type including aspirate or genital tract (17/358, 4.7%) (Figure 2).
- Mold isolates were from respiratory tract infections (50/69, 72.5%), head and neck infections (11/69, 15.9%), skin and soft tissue infections (5/69, 7.2%), bloodstream infections (1/69, 1.4%), and no listed infection type (2/69, 2.9%) (Figure 2).
- For yeast, manogepix $MIC_{50/90}$ was 1 to 2 doubling dilutions lower than micafungin for all species with the exception of *C. krusei* against which manogepix does not have activity (Table 1). Manogepix $MIC_{50/90}$ was 3–4 doubling dilutions lower than fluconazole for all *Candida* spp. besides *C. krusei*. Manogepix $MIC_{50/90}$ was similar to voriconazole for most *Candida* spp. except for *C. auris* where manogepix was 6 doubling dilutions more active and *C. lusitaniae* where voriconazole was more active by 1 to 3 doubling dilutions. Manogepix $MIC_{50/90}$ was also lower than that of amphotericin B against all species besides *C. krusei*. Manogepix $MIC_{50/90}$ was lower than all comparators besides voriconazole against less common yeast.
 - There were 1 *C. albicans* and 5 *C. glabrata* non-susceptible to micafungin and 2 *C. albicans*, 6 *C. glabrata*, 1 *C. parapsilosis*, and 3 *C. tropicalis* resistant to fluconazole; manogepix MICs among these isolates were 0.004–0.12 mg/L (data not shown).
- For molds, manogepix $MEC_{50/90}$ was lower than $MIC_{50/90}$ for all comparators by 5 to 8 doubling dilutions.
 - There was 1 *Rhizopus oryzae* isolate with a manogepix MEC of 4 mg/L, voriconazole MIC >8 mg/L, itraconazole 4 mg/L, amphotericin B 0.06 mg/L.
 - Among the 3 *A. fumigatus* non-susceptible to voriconazole, manogepix MECs were 0.015 to 0.03 mg/L (data not shown).

Conclusions

- Manogepix is highly active *in vitro* against yeast and mold collected from diverse patient infection types across the United States.
 - These isolates were geographically diverse including yeast from all 9 census divisions and molds from 7/9 census divisions.
- Against most organisms manogepix MICs or MECs were lower than the MICs of comparator antifungal agents.
 - Higher MICs were observed against *C. krusei* among yeast and higher MECs in 1 *Rhizopus oryzae* among molds.
- These data support the continued development of fosmanogepix for the treatment of invasive fungal infections in the United States.

References

CLSI, *M38M51S Ed4. Performance standards for antifungal susceptibility testing of molds*. 2026, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M57S Ed4. Epidemiological cutoff values for antifungal susceptibility testing*. 2022, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M38 Ed3. Reference method for broth dilution antifungal susceptibility testing of filamentous fungi*. 2017, Clinical and Laboratory Standards Institute: Wayne, PA.

CLSI, *M27 Ed4. Reference method for broth dilution antifungal susceptibility testing of yeasts*. 2017, Clinical and Laboratory Standards Institute: Wayne, PA.

Figure 1. Geography of collected isolates

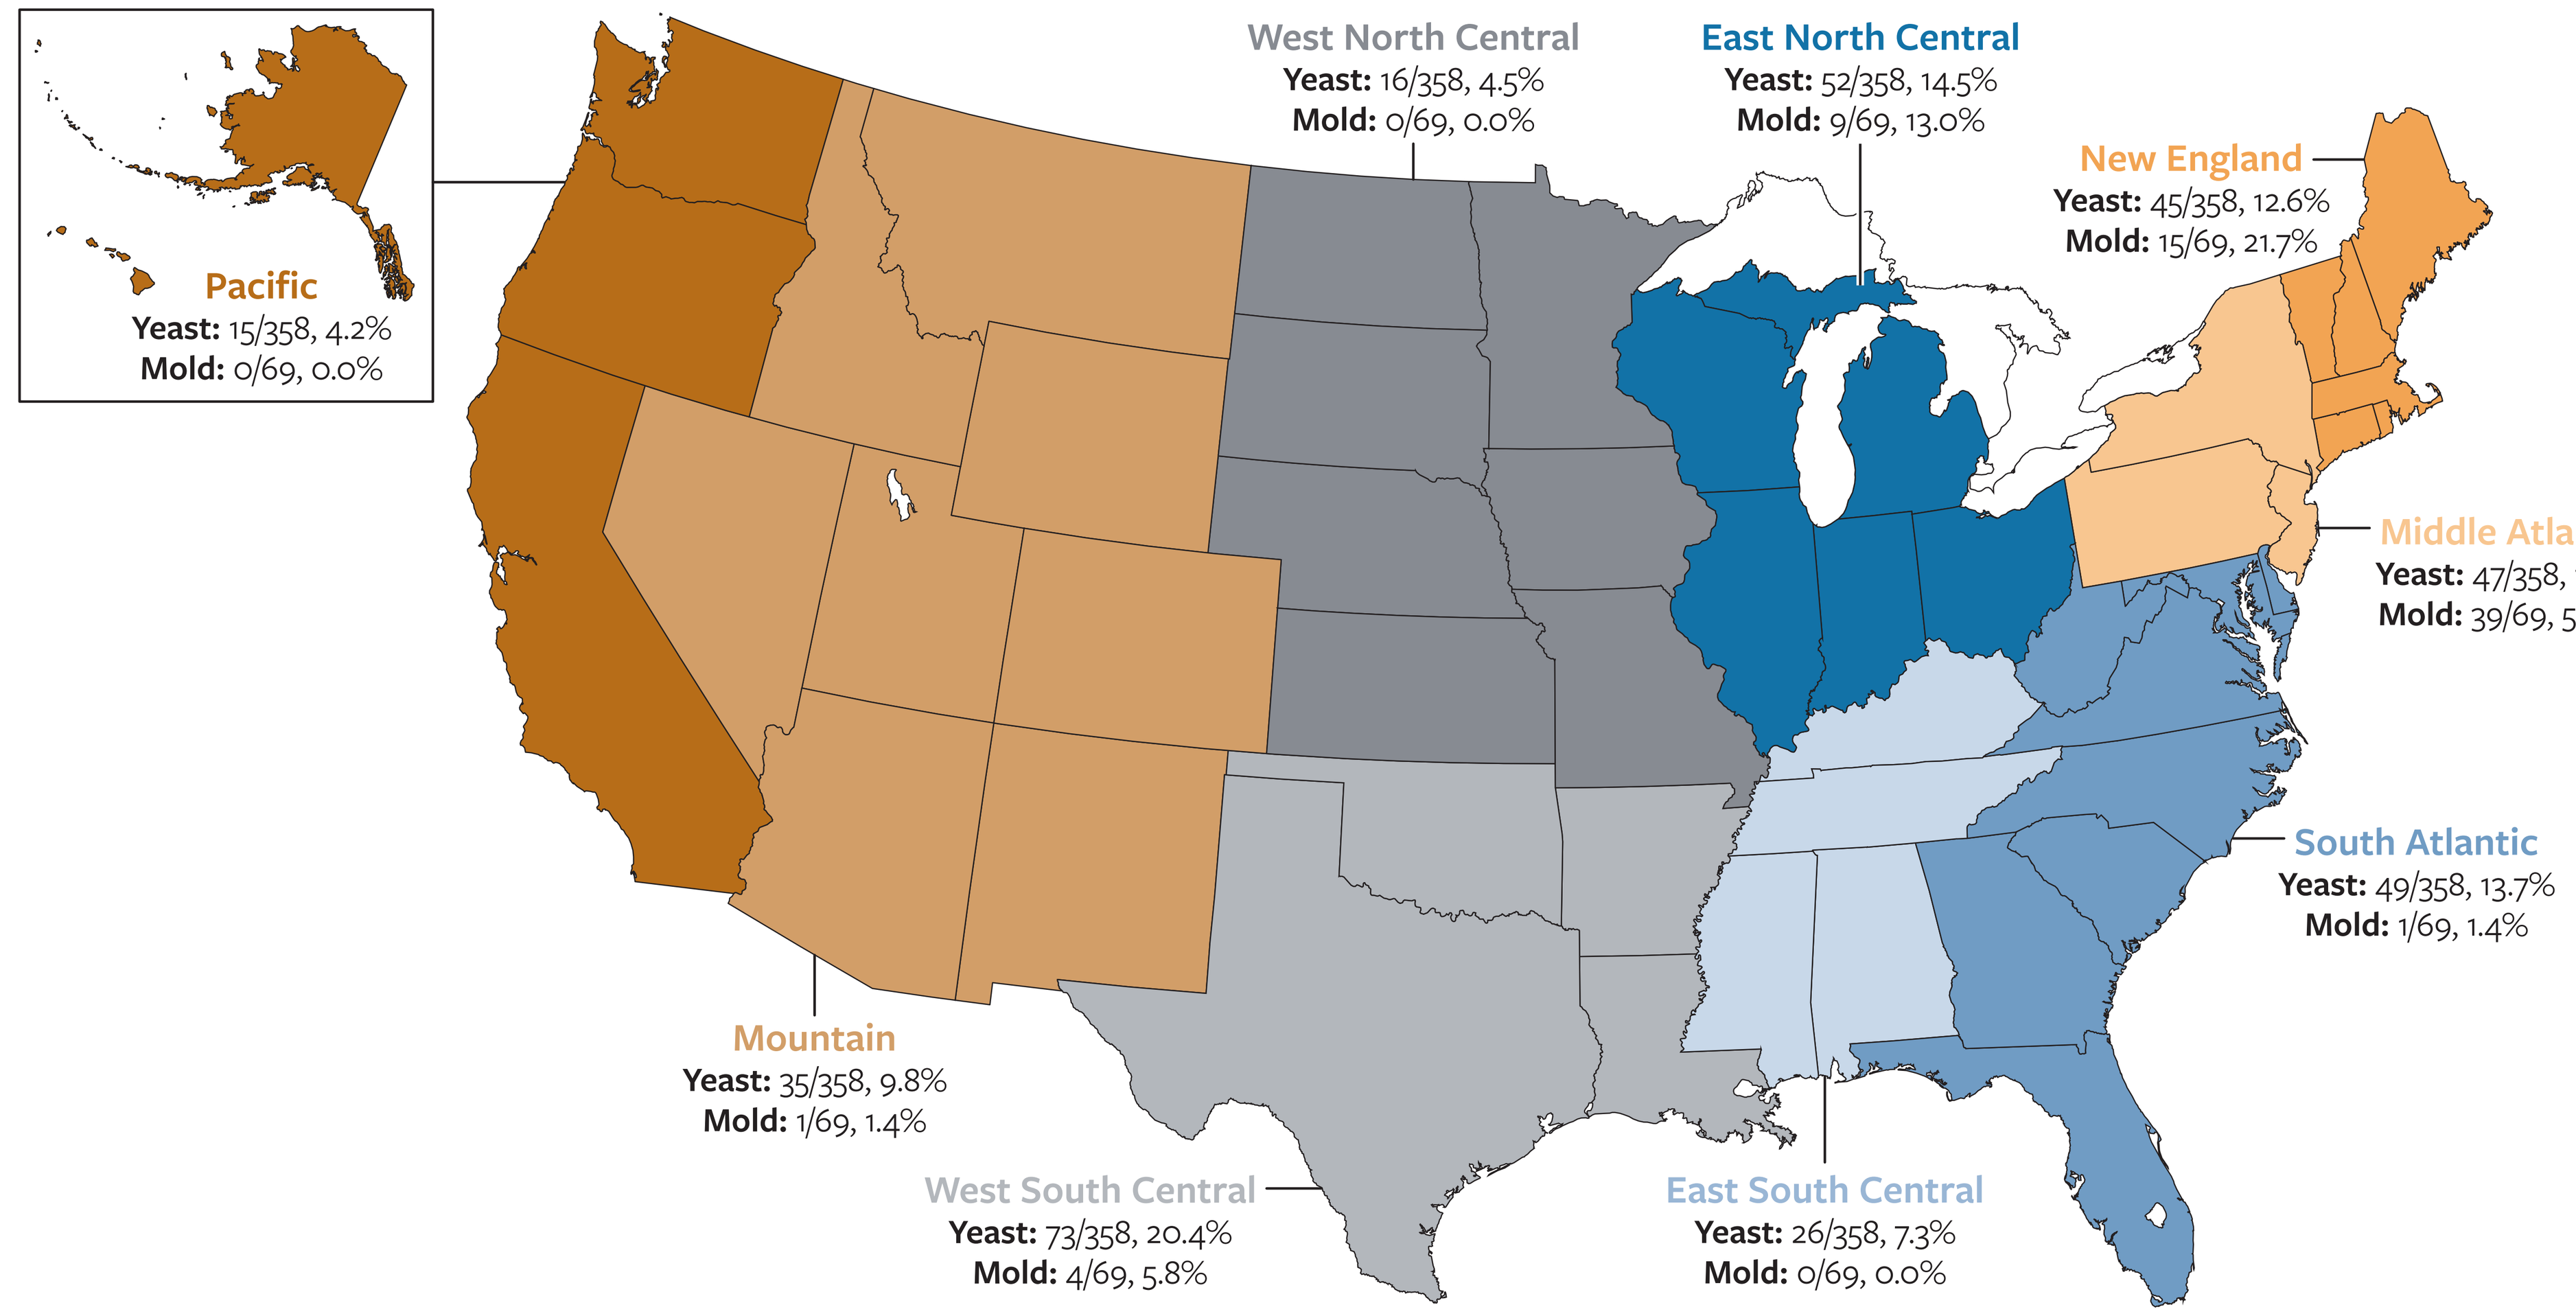
Pacific
Yeast: 15/358, 4.2%
Mold: 0/69, 0.0%
West North Central
Yeast: 16/358, 4.5%
Mold: 0/69, 0.0%
East North Central
Yeast: 52/358, 14.5%
Mold: 9/69, 13.0%
New England
Yeast: 45/358, 12.6%
Mold: 15/69, 21.7%
Middle Atlantic
Yeast: 47/358, 13.1%
Mold: 39/69, 56.5%
South Atlantic
Yeast: 49/358, 13.7%
Mold: 1/69, 1.4%
Mountain
Yeast: 35/358, 9.8%
Mold: 1/69, 1.4%
West South Central
Yeast: 73/358, 20.4%
Mold: 4/69, 5.8%
East South Central
Yeast: 26/358, 7.3%
Mold: 0/69, 0.0%

Figure 2. Infection source of collected yeast (A) and mold (B) isolates

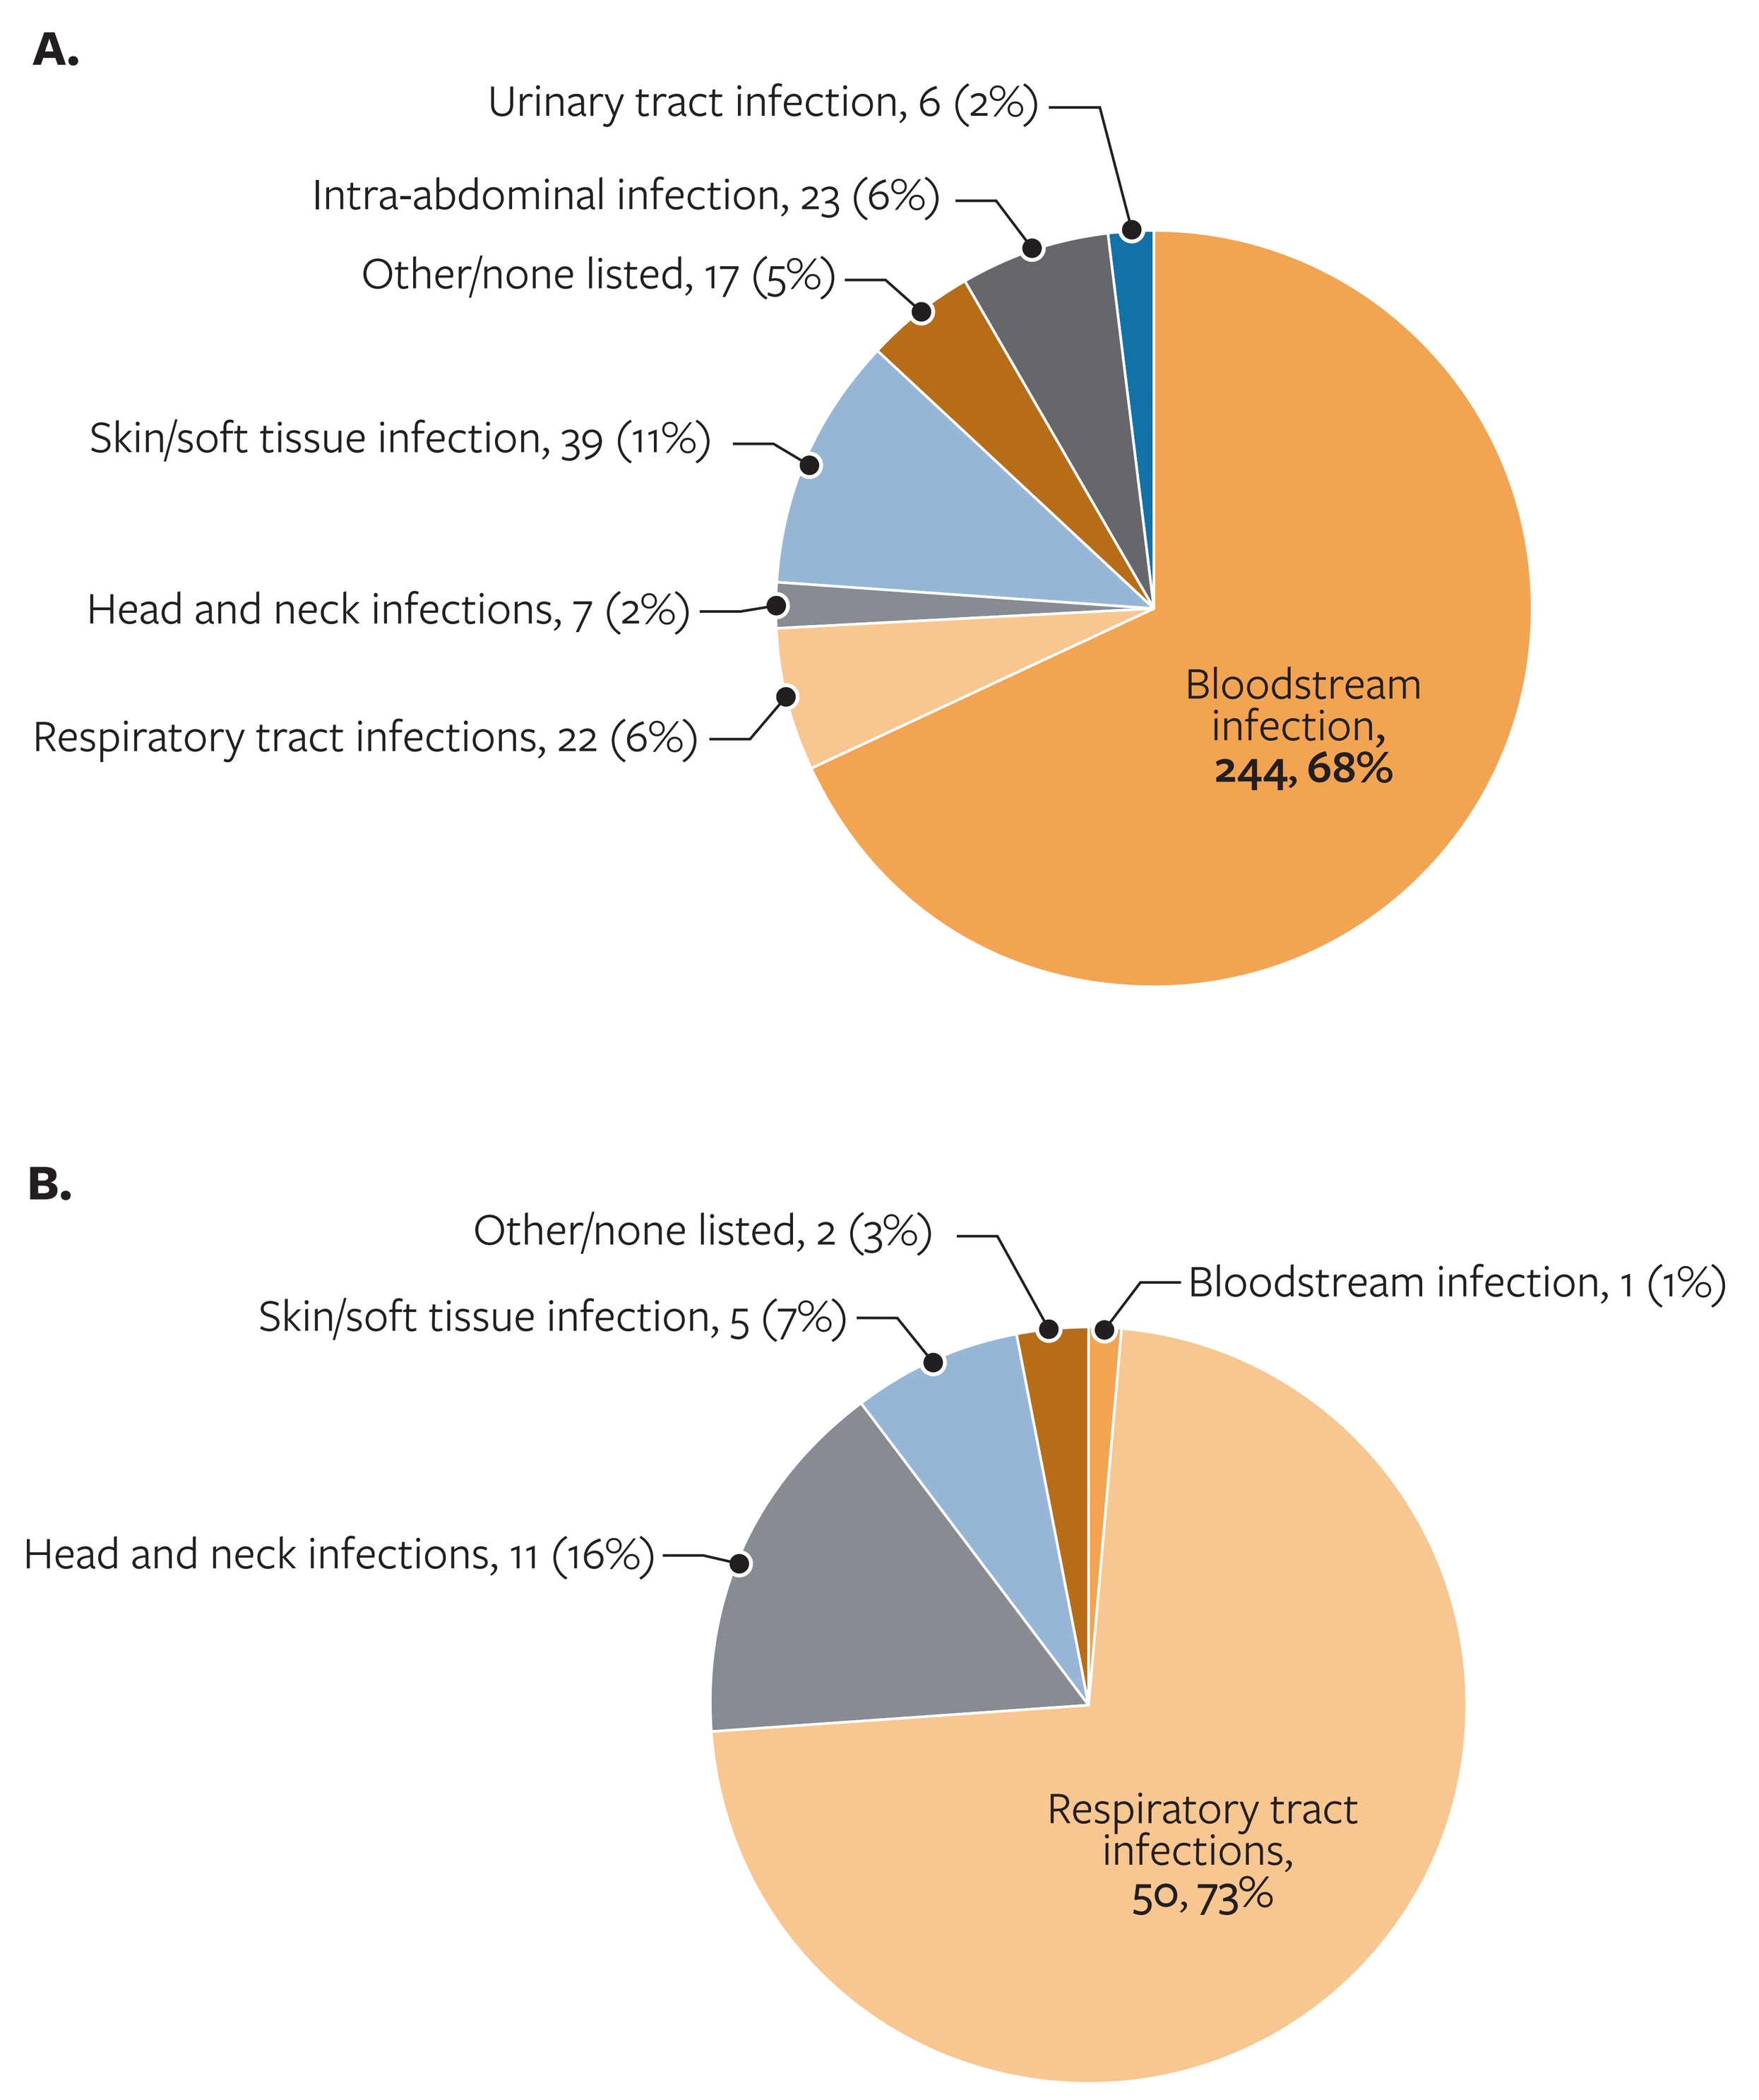
A.
Urinary tract infection, 6 (2%)
Intra-abdominal infection, 23 (6%)
Other/none listed, 17 (5%)
Skin/soft tissue infection, 39 (11%)
Head and neck infections, 7 (2%)
Respiratory tract infections, 22 (6%)
Bloodstream infection, 244, 68%
B.
Other/none listed, 2 (3%)
Bloodstream infection, 1 (1%)
Skin/soft tissue infection, 5 (7%)
Head and neck infections, 11 (16%)
Respiratory tract infections, 50, 73%

Table 1. MIC/MEC range (mg/L) and MIC/$MEC_{50/90}$ (mg/L) for manogepix (MGX), micafungin (MCG), fluconazole (FLC), voriconazole (VRC), and amphotericin B (AmB) in yeast

	MGX		MCG (%S/WT)[1]		FLC (%S/WT)[1]		VRC (%S/WT)[1]		AmB (%S/WT)[1]	
Organism (n)	MIC range	$MIC_{50/90}$	MIC range	$MIC_{50/90}$	MIC range	$MIC_{50/90}$	MIC range	$MIC_{50/90}$	MIC range	$MIC_{50/90}$
All yeast (358)	0.001 – >4	0.015/0.12	0.008 – >4	0.03/1	0.06 – >128	0.5/64	≤0.002 – >8	0.015/0.25	0.12 – 2	1/1
All *Candida* spp. (339)	0.001 – >4	0.015/0.06	0.008 – 4	0.03/1	0.06 – >128	0.5/64	≤0.002 – >8	0.015/0.25	0.12 – 2	1/1
C. albicans (91)	0.002 – 0.03	0.008/0.008	0.008 – 0.5 (98.9/98.9)	0.015/0.03	0.06 – 8 (97.8/96.7)	0.12/0.25	≤0.002 – 0.12 (100.0/96.7)	0.004/0.008	0.12 – 1 (NA/100.0)	0.5/1
C. glabrata (84)	0.008 – 0.12	0.03/0.12	0.008 – 1 (94.0/94.0)	0.015/0.03	0.25 – 128 (92.9/89.3)	4/16	0.008 – 4 (NA/89.3)	0.06/0.5	0.25 – 1 (NA/100.0)	1/1
C. parapsilosis (40)	0.004 – 0.03	0.015/0.015	0.5 – 2 (100.0/100.0)	1/2	0.12 – 16 (97.5/97.5)	0.25/0.5	≤0.002 – 0.25 (97.5/NA)	0.008/0.015	0.25 – 1 (NA/100.0)	0.5/1
C. dubliniensis (29)	0.002 – 0.008	0.004/0.008	0.008 – 0.03 (NA/100.0)	0.03/0.03	0.06 – 0.25 (NA/100.0)	0.12/0.25	≤0.002 – 0.015 (NA/NA)	0.004/0.008	0.12 – 0.5 (NA/100.0)	0.25/0.5
C. auris (25)	0.008 – 0.12	0.015/0.06	0.06 – 1 (100.0/96.0)	0.25/0.5	64 – >128 (0.0/NA)	128/>128	0.25 – >8 (NA/NA)	1/2	0.5 – 2 (44.0/NA)	2/2
C. tropicalis (25)	0.008 – 0.03	0.015/0.03	0.008 – 0.06 (100.0/100.0)	0.03/0.06	0.12 – >128 (88.0/88.0)	0.25/16	0.004 – 2 (88.0/88.0)	0.015/0.25	0.25 – 1 (NA/100.0)	1/1
C. lusitaniae (12)	0.015 – 0.5	0.06/0.25	0.06 – 4 (NA/91.7)	0.25/0.25	0.12 – 128 (NA/83.3)	0.5/128	0.004 – 1 (NA/NA)	0.008/0.5	0.5 – 1 (NA/100.0)	0.5/0.5
Cryptococcus neoformans var. grubii (12)	0.12 – 0.5	0.25/0.25	4 – >4 (NA/NA)	>4/>4	2 – 4 (NA/100.0)	4/4	0.03 – 0.06 (NA/100.0)	0.06/0.06	0.25 – 1 (NA/33.3)	1/1
C. krusei (10)	4 – >4	>4/>4	0.03 – 0.12 (100.0/100.0)	0.12/0.12	16 – 32 (NA/NA)	16/32	0.12/0.25 (100.0/100.0)	0.25/0.25	2 (NA/100.0)	2/2
Other *Candida* spp. (23) [2]	0.001 – 1	0.015/1	0.03 – 1	0.25/0.5	0.25 – 128	1/4	0.004 – 4	0.03/0.12	0.25 – 2	1/2
Other non-*Candida* yeast (7) [3]	0.015 – 0.25	0.03/NA	0.03 – >4	0.25/NA	0.25 – 4	1/NA	0.008 – 0.12	0.03/NA	0.25 – 1	1/NA

MGX, manogepix; MCG, micafungin; S, susceptible; WT, wildtype; VRC, voriconazole; ITR, itraconazole; AmB, amphotericin B; NA, not applicable
[1] CLSI M27M44S, CLSI M57S, or for *C. auris* https://www.cdc.gov/candida-auris/hcp/laboratories/antifungal-susceptibility-testing.html
[2] Includes *C. fermentati* (2), *C. guilliermondii* (5), *C. kefyr* (6), *C. metapsilosis* (5), *C. orthopsilosis* (3), *C. pelliculosa* (2)
[3] Includes *Lodderomyces elongisporus* (1), *Saccharomyces cerevisiae* (5), and *Trichosporon asahii* (1)

Table 2. MIC/MEC range (mg/L) and MIC/$MEC_{50/90}$ (mg/L) for manogepix (MGX), voriconazole (VRC), itraconazole (ITR), and amphotericin B (AmB) in molds

	MGX		VRC		ITR		AmB	
Organism (n)	MEC range	$MEC_{50/90}$	MIC range (%S/WT)[1]	$MIC_{50/90}$	MIC range (%S/WT)[1]	$MIC_{50/90}$	MIC range (%S/WT)[1]	$MIC_{50/90}$
All molds (69)	≤0.0005 – 4	0.008/0.03	0.06 – >8	0.5/2	0.008 – >8	1/8	≤0.03 – >4	1/2
All *Aspergillus* spp. (59)	0.002 – 0.06	0.008/0.03	0.12 – 2	0.5/1	0.008 – >8	1/2	0.12 – 2	1/2
Aspergillus fumigatus (42)	0.008 – 0.03	0.015/0.03	0.25 – 2 (92.9/97.6)	0.5/0.5	0.5 – 4 (NA/97.6)	1/1	0.25 – 2 (NA/100.0)	1/2
Aspergillus niger species complex (11)	0.002 – 0.008	0.004/0.008	0.5 – 2 (NA/100.0)	1/2	1 – >8 (NA/81.8)	2/8	0.12 – 0.25 (NA/100.0)	0.12/0.25
Other *Aspergillus* spp. (6) [2]	0.008 – 0.06	0.008/*	0.12 – 1	0.5/*	0.008 – 0.05	0.5/*	0.5 – 2	1/*
Other molds (10) [3]	≤0.0005 – 4	0.004/0.015	0.06 – >8	1/>8	0.12 – >8	4/>8	≤0.03 – >4	1/>4

*MIC_{50} only as < 10 organisms represented
MGX, manogepix; VRC, voriconazole; ITR, itraconazole; AmB, amphotericin B
[1] CLSI M38M51S, CLSI M57S
[2] Includes *Aspergillus flavus* species complex (3), *A. sclerotiorum* (1), *A. thermomutatus* (1), and *A. versicolor* species complex (1)
[3] Genera include *Exophiala dermatitidis* (1), *Exophiala phaeomuriformis* (1), *Paecilomyces variotii* (1), *Penicillium chrysogenum* (1), *Phialemonum obovatum* (1), *Rhizopus oryzae* (1), *Scedosporium boydii* (1), *Scopulariopsis brevicaulis* (1), *Scedosporium* spp. (2)

Funding

This study was supported by Basilea Pharmaceutica International Ltd and this project has been funded in part with federal funds from the U.S. Department of Health and Human Services (HHS); Administration of Strategic Preparedness and Response (ASPR); Biomedical Advanced Research and Development Authority (BARDA), under OT number: 75A50124C00033. The contract and federal funding are not an endorsement of the study results, product, or company.

ML Winkler, S Edeker, A Klauer, PR Rhomberg, and M Castanheira were employees of Element Materials Technology (JMI Laboratories) at the time of this study, which was a paid consultant to Basilea in connection with the development of this poster.

Acknowledgments

The authors thank all of the SENTRY Program participants for providing the isolates used in this study.

IDWeek 2026, October 21–24, 2026, Washington, D.C.