IDWeek 2026 | Poster #P-142

In Vitro Activity of Ceftobiprole and Comparator Agents Against Contemporary U.S. Clinical Isolates Collected in 2025 Following FDA Approval

Rodrigo E. Mendes[1], Marisa Winkler[1], Samir H. Moussa[2], Tatiana Wiktorowicz[3], Helio S. Sader[1], Mariana Castanheira[1]

[1] Element Iowa City (JMI Laboratories), North Liberty, IA, USA; [2] Innoviva Specialty Therapeutics, Inc., Waltham, MA, USA; [3] Basilea Pharmaceutica International Ltd, Allschwil, Switzerland

Background

- Ceftobiprole is an intravenous cephalosporin with activity against clinically important Gram-positive pathogens, including methicillin-resistant *Staphylococcus aureus* (MRSA), and selected Gram-negative and respiratory pathogens.
- The U.S. FDA approved ceftobiprole in April 2024 for adults with *S. aureus* bacteremia, including right-sided infective endocarditis; adults with acute bacterial skin and skin structure infections (ABSSSI); and adult and pediatric patients with community-acquired bacterial pneumonia (CABP) caused by susceptible isolates (Table 2).
- Post-approval surveillance provides ongoing assessment of antimicrobial activity against contemporary clinical isolates collected from patients in U.S. institutions and enables detection of potential changes in susceptibility patterns following clinical introduction.

Methods

Bacterial isolates

- A total of 4,126 clinical isolates were collected from 31 U.S. medical centers across all 9 U.S. census divisions during 2025 as part of the SENTRY Antimicrobial Surveillance Program (Figure 1).
- Isolates were primarily recovered from hospitalized patients with bloodstream infections (BSI; 45%), ABSSSI (20%), and respiratory tract infections (35%).

Susceptibility testing

- Isolates were tested for susceptibility by broth microdilution according to CLSI approved methods using cation-adjusted Mueller–Hinton broth (CAMHB).
- CLSI and U.S. FDA (for ceftobiprole) breakpoint criteria were applied for interpretation of MIC results. *E. coli* and *K. pneumoniae* displaying MIC results ≥2 µg/mL for aztreonam or ceftazidime or ceftriaxone were defined as presumptive extended-spectrum β-lactamase (ESBL) producers.

Contact

element

Rodrigo E. Mendes, PhD, FIDSA
Element Iowa City/JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317 USA
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo.mendes@element.com

INNOVIVA Specialty Therapeutics

Samir H. Moussa, PhD
Innoviva Specialty Therapeutics
Email: samir.moussa@istx.com

Scan Me

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2026_26-IST-01_P1_Ceftobiprole_US_2025.pdf

Charges may apply. No personal information is stored.

Results

- Against 1,606 *S. aureus* isolates, ceftobiprole demonstrated $MIC_{50/90}$ values of 0.5/1 µg/mL, and 99.7% susceptibility overall, including susceptibility against 100% of MSSA and 99.2% of MRSA isolates (Table 1 and Figure 2).
- Among the subset of *S. aureus* isolates recovered from BSI (n=553) and ABSSSI (n=548), ceftobiprole susceptibility remained high (99.1–100% susceptible) (Table 2).
- When tested against 199 β-hemolytic streptococci, ceftobiprole inhibited all isolates at an MIC of ≤0.03 µg/mL ($MIC_{50/90}$, 0.008/0.03 µg/mL), whereas the ceftobiprole $MIC_{50/90}$ values against Viridans group streptococci (VGS) were 0.06/0.25 µg/mL (Table 1).
- Among respiratory pathogens, ceftobiprole demonstrated activity against *S. pneumoniae* and *H. influenzae*, with $MIC_{50/90}$ values of 0.008/0.25 µg/mL and 0.03/0.12 µg/mL, respectively; susceptibility rates were 98.8% and 97.8%, respectively (Table 1).
- As expected, ceftobiprole demonstrated limited activity against presumed ESBL-producing isolates, whereas ceftobiprole retained activity against non-ESBL *E. coli* and *K. pneumoniae*, with susceptibility rates of 100% and 99.7%, respectively (Table 1).

Conclusions

- Ceftobiprole demonstrated potent *in vitro* activity against contemporary U.S. clinical isolates collected in 2025, including pathogens associated with all current FDA-approved indications.
- Against 1,606 *S. aureus* isolates, ceftobiprole remained highly active, with susceptibility rates of 99.7% overall, including 100% of MSSA and 99.2% of MRSA isolates.
- Ceftobiprole also demonstrated potent activity against β-hemolytic streptococci, *S. pneumoniae*, and *H. influenzae*, supporting continued activity against pathogens relevant to ABSSSI and CABP indications.
- Among Enterobacterales, ceftobiprole retained activity against presumed non-ESBL *E. coli* and *K. pneumoniae* isolates but showed limited activity against presumed ESBL-producing isolates, consistent with its expected antibacterial spectrum.
- These findings establish an important post-approval benchmark for ceftobiprole activity in the U.S. and provide a foundation for longitudinal surveillance of susceptibility trends during the ongoing monitoring program.

Acknowledgments

The authors express appreciation to all U.S. medical sites participating in the SENTRY Antimicrobial Surveillance Program for their most valuable support and contribution over the years. This study was performed by Element Iowa City (JMI Laboratories) and sponsored by Basilea Pharmaceutica International, Ltd., with additional support from ISTx, LLC, an affiliate of Innoviva Specialty Therapeutics, Inc.

This project has been funded in whole or in part with federal funds from the U.S. Department of Health and Human Services (HHS); Administration of Strategic Preparedness and Response (ASPR); Biomedical Advanced Research and Development Authority (BARDA), under contract number: HHSO100201600002C. The contract and federal funding are not an endorsement of the study results, product, or company.

Figure 1. Geographic representation of the U.S. ceftobiprole surveillance program. A total of 4,126 contemporary clinical isolates were collected from 31 participating medical centers representing all 9 U.S. Census divisions. The surveillance collection included pathogens recovered from bloodstream, respiratory tract, and skin and skin structure infections and was designed to provide a geographically representative assessment of ceftobiprole activity in the U.S.

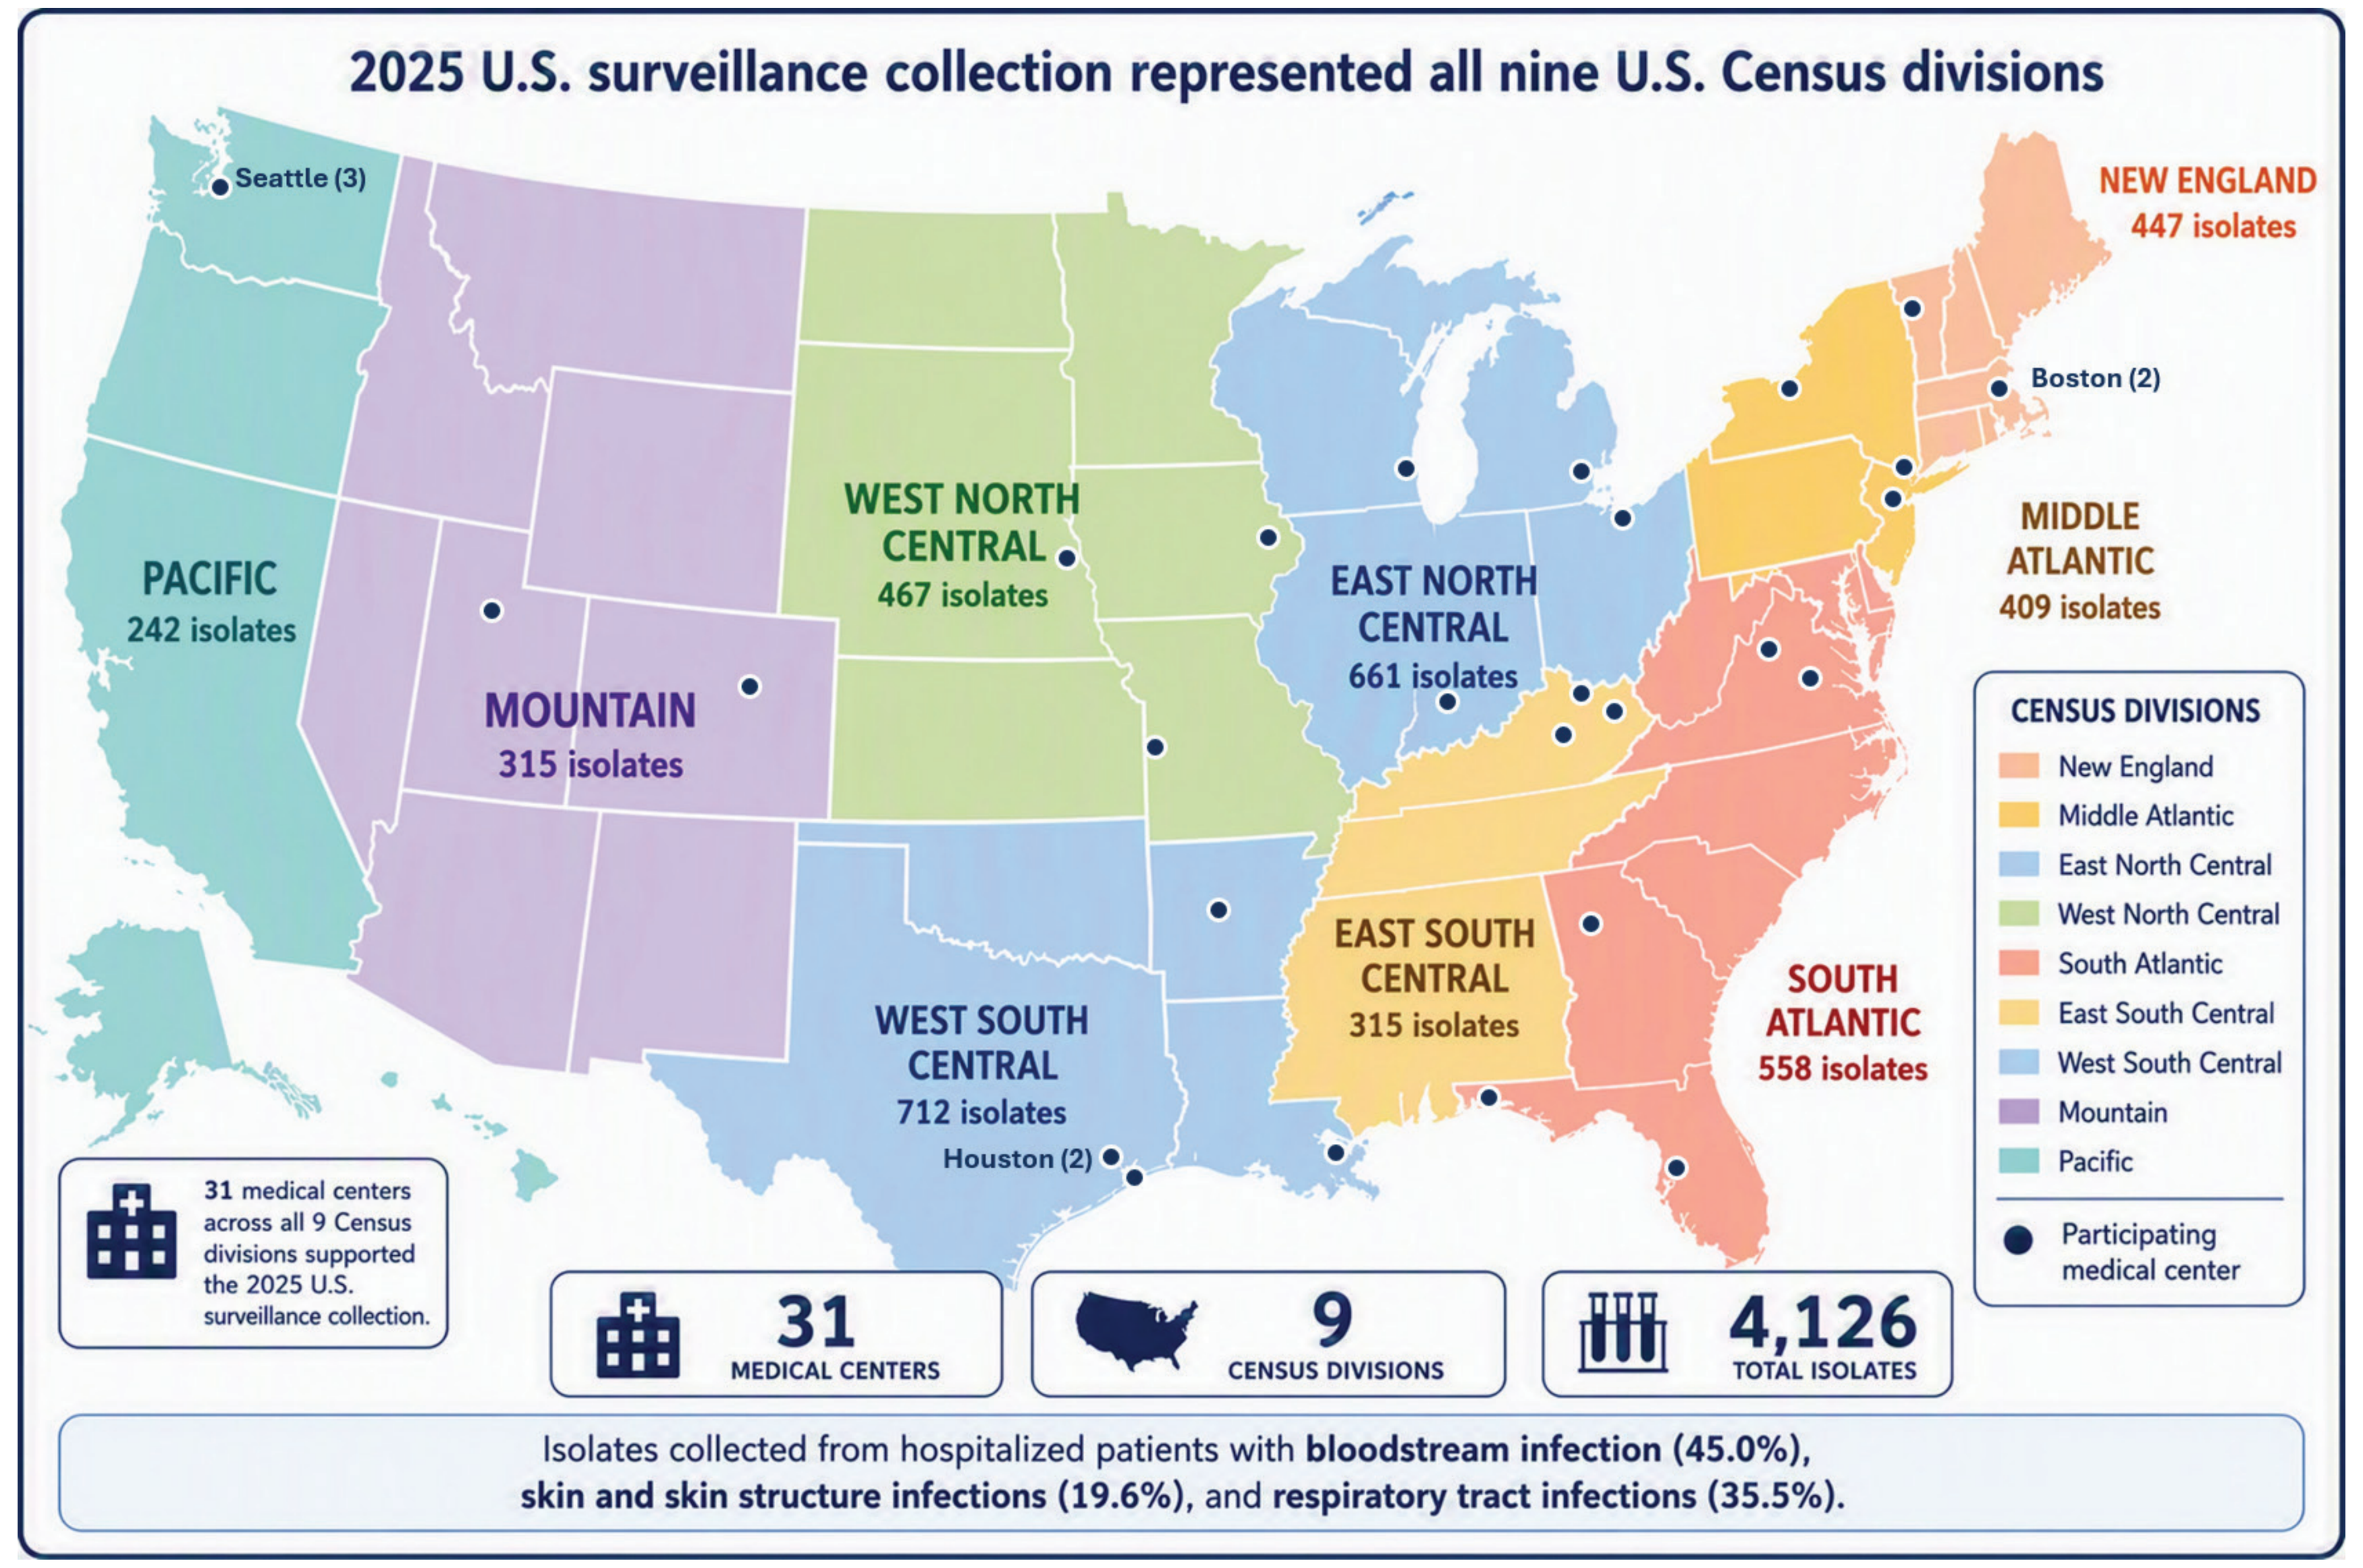
2025 U.S. surveillance collection represented all nine U.S. Census divisions
Seattle (3)
PACIFIC
242 isolates
MOUNTAIN
315 isolates
WEST NORTH CENTRAL
467 isolates
EAST NORTH CENTRAL
661 isolates
NEW ENGLAND
447 isolates
Boston (2)
MIDDLE ATLANTIC
409 isolates
WEST SOUTH CENTRAL
712 isolates
Houston (2)
EAST SOUTH CENTRAL
315 isolates
SOUTH ATLANTIC
558 isolates
CENSUS DIVISIONS
New England
Middle Atlantic
East North Central
West North Central
South Atlantic
East South Central
West South Central
Mountain
Pacific
Participating medical center
31 medical centers across all 9 Census divisions supported the 2025 U.S. surveillance collection.
31 MEDICAL CENTERS
9 CENSUS DIVISIONS
4,126 TOTAL ISOLATES
Isolates collected from hospitalized patients with bloodstream infection (45.0%), skin and skin structure infections (19.6%), and respiratory tract infections (35.5%).

Figure 2. Activity of ceftobiprole against contemporary MSSA and MRSA isolates collected in the U.S. MIC distributions demonstrate sustained *in vitro* activity against both MSSA and MRSA, with susceptibility rates of 100% and 99.2%, respectively. Although MRSA isolates exhibited a modest shift toward higher MIC values than MSSA isolates, the overwhelming majority remained at or below the FDA susceptible breakpoint (≤2 µg/mL).

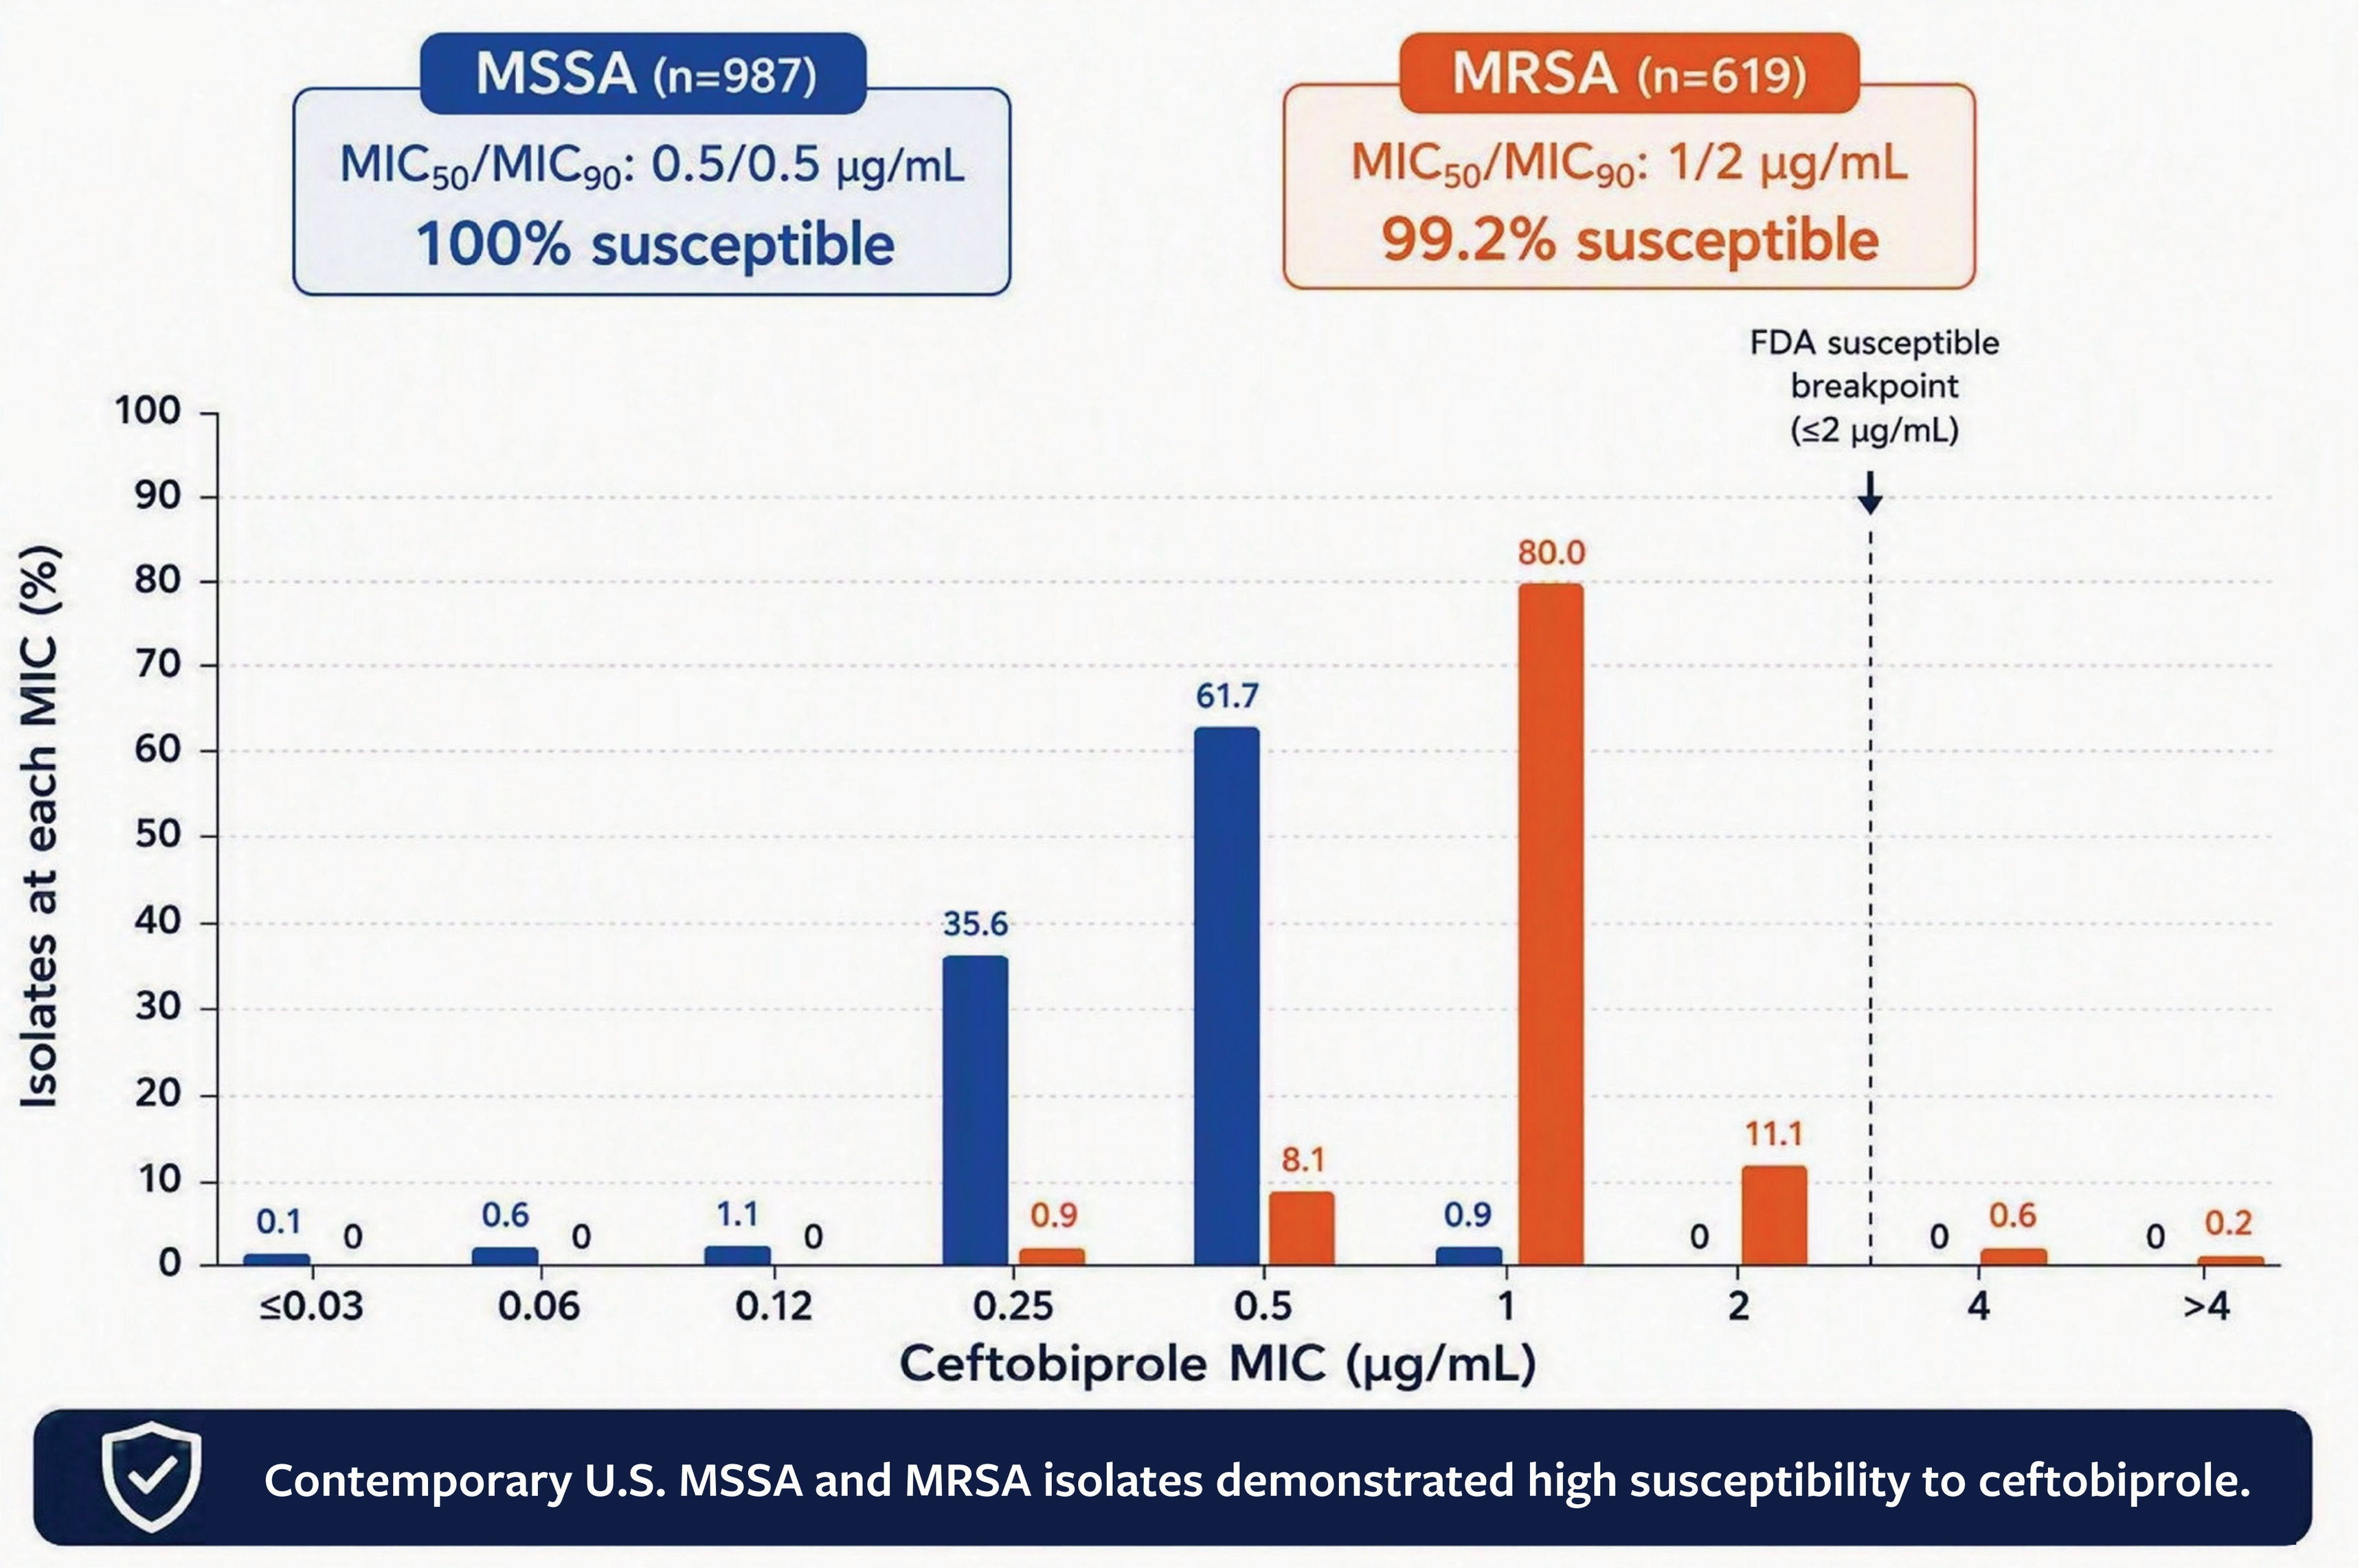
MSSA (n=987)
MIC50/MIC90: 0.5/0.5 µg/mL
100% susceptible
MRSA (n=619)
MIC50/MIC90: 1/2 µg/mL
99.2% susceptible
FDA susceptible breakpoint (≤2 µg/mL)
Isolates at each MIC (%)
0
10
20
30
40
50
60
70
80
90
100
0.1
0
0.6
0
1.1
0
35.6
0.9
61.7
8.1
0.9
80.0
0
11.1
0
0.6
0
0.2
≤0.03
0.06
0.12
0.25
0.5
1
2
4
>4
Ceftobiprole MIC (µg/mL)
Contemporary U.S. MSSA and MRSA isolates demonstrated high susceptibility to ceftobiprole.

Table 1. Activity of ceftobiprole and comparator agents against surveillance clinical isolates collected from U.S. sites in 2025

Species/group (No. tested)	MIC50/MIC90 in µg/mL (% susceptible by CLSI/FDA)[a]						
	BPR	**CPT**	**CLI**	**CRO**	**DAP**	**LZD**	**VAN**
S. aureus (1,606)	0.5/1 (99.7)	0.25/1 (98.6)	0.06/>2 (89.4)	4/>8 (61.5)[b]	0.25/0.25 (99.9)	1/2 (100)	1/1 (100)
Methicillin-susceptible (987)[b]	0.5/0.5 (100)	0.25/0.25 (100)	0.06/0.12 (96.9)	4/4 (100)[b]	0.25/0.25 (100)	1/2 (100)	1/1 (100)
Methicillin-resistant (619)[b]	1/2 (99.2)	0.5/1 (96.4)	0.06/>2 (77.4)	>8/>8 (0.0)[b]	0.25/0.25 (99.8)	1/2 (100)	1/1 (100)
	BPR	**CPT**	**A/C**	**AZI**	**CRO**	**LEV**	**PEN**
β-hemolytic streptococci (199)[c]	0.008/0.03 (100)	≤0.008/0.015 (100)	≤0.03/0.06 (100)	0.12/>4 (54.8)	0.03/0.06 (100)	0.5/1 (99.0)	0.015/0.06 (100)
Viridans group streptococci (96)[d]	0.06/0.25 (—)	0.015/0.06 (—)	0.12/1 (—)	2/>4 (42.1)	0.12/0.5 (96.9)	1/>4 (75.0)	0.12/1 (57.3)
S. pneumoniae (406)	0.008/0.25 (98.8)	≤0.008/0.12 (100)	≤0.03/2 (96.8)	0.06/>4 (59.5)	0.015/0.5 (98.0)	1/1 (100)	0.015/2 (96.3)
H. influenzae (182)	0.03/0.12 (97.8)	0.008/0.03 (100)	0.5/2 (93.4)	1/2 (97.3)	0.004/0.015 (100)	0.015/0.03 (98.9)	1/>4 (—)
	BPR	**A/C**	**CRO**	**IMI**	**P/T**	**LEV**	**GEN**
E. coli (807)	0.03/>16 (73.0)	8/32 (73.0)	≤0.06/>8 (72.4)	0.12/0.25 (99.5)	2/8 (92.7)	0.06/16 (67.4)	0.5/>16 (85.5)
Non-ESBL (560)	0.03/0.06 (100)	8/16 (83.6)	≤0.06/0.12 (100)	0.12/0.12 (100)	2/4 (97.5)	0.03/8 (82.5)	0.5/2 (91.2)
ESBL (247)	>16/>16 (11.7)	16/32 (49.0)	>8/>8 (9.7)	0.12/0.25 (98.4)	4/32 (81.8)	8/32 (33.2)	1/>16 (72.5)
K. pneumoniae (541)	0.03/>16 (73.0)	2/16 (78.6)	≤0.06/>8 (74.9)	0.12/0.25 (97.0)	4/32 (79.1)	0.06/4 (83.5)	0.25/>16 (87.8)
Non-ESBL (388)	0.03/0.06 (99.7)	2/8 (96.4)	≤0.06/0.12 (100)	0.12/0.25 (100)	4/8 (92.3)	0.06/0.25 (95.9)	0.25/0.5 (98.7)
ESBL (153)	>16/>16 (5.2)	16/32 (33.3)	>8/>8 (11.1)	0.12/2 (89.5)	16/>128 (45.8)	0.5/16 (52.3)	0.25/>16 (60.1)

BPR, ceftobiprole; CPT, ceftaroline; CLI, clindamycin; CRO, ceftriaxone; DAP, daptomycin; LZD, linezolid; VAN, vancomycin; A/C, amoxicillin-clavulanate (2:1 ratio); AZI, azithromycin; LEV, levofloxacin; PEN, parenteral penicillin; IMI, imipenem; P/T, piperacillin-tazobactam; GEN, gentamicin; ESBL, extended-spectrum β-lactamase; *E. coli* and *K. pneumoniae* displaying MIC results ≥2 µg/mL for aztreonam or ceftazidime or ceftriaxone were defined as presumptive ESBL producers, whereas those with MIC results <2 µg/mL for aztreonam and ceftazidime and ceftriaxone were defined as presumptive non-ESBL producers.
[a] Susceptibility for ceftobiprole based on breakpoints approved by the U.S. FDA, including the susceptible breakpoint for *S. pyogenes* applied to β-hemolytic streptococci. Susceptibility to comparator agents based on CLSI breakpoints. "—" breakpoint not available.
[b] Methicillin-susceptible and -resistant *S. aureus* isolates were defined based on oxacillin MIC results. Ceftriaxone susceptibility against *S. aureus* determined based on the oxacillin results used as a surrogate.
[c] *Streptococcus agalactiae* (79), *S. canis* (2), *S. dysgalactiae* (20), *S. equi* (1), and *S. pyogenes* (97).
[d] *Streptococcus anginosus* group (10), *S. constellatus* (4), *S. equinus* group (2), *S. gallolyticus* (4), *S. gordonii* (5), *S. intermedius* (4), *S. mitis* group (48), *S. mitis/oralis* (2), *S. mutans* (2), *S. parasanguinis* (5), *S. salivarius* group (4), *S. sanguinis* (5), and *S. vestibularis* (1).

Table 2. Ceftobiprole FDA-approved indications and *in vitro* activity against current indicated pathogens

Approved indication	Indicated pathogens	Ceftobiprole susceptibility in indication-specific isolate subsets (n)
S. aureus bacteremia, including right-sided infective endocarditis (adult)	MSSA and MRSA	*S. aureus*: **99.6% susceptible** (n = 553) MSSA: **100% susceptible** (n = 325) MRSA: **99.1% susceptible** (n = 228)
Acute bacterial skin and skin structure infections (adult)	MSSA, MRSA, and β-hemolytic streptococci	*S. aureus*: **100% susceptible** (n = 548) MSSA: **100% susceptible** (n = 346) MRSA: **100% susceptible** (n = 202) β-hemolytic streptococci: **100% susceptible** (n = 81)
Community-acquired bacterial pneumonia (adult and pediatric)	*S. pneumoniae* and *H. influenzae*	*S. pneumoniae*: **98.9% susceptibl**e (n = 355) *H. influenzae*: **97.8% susceptible** (n = 182)

References

Clinical and Laboratory Standards Institute. M07Ed12. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically. Wayne, PA, USA, 2024.

Clinical and Laboratory Standards Institute. M100Ed36. Performance standards for antimicrobial susceptibility testing. Wayne, PA, USA, 2026.

ZEVTERA Prescribing Information (ceftobiprole medocaril sodium; injection for intravenous use). Available at https://zevtera-assets.s3.amazonaws.com/prescribing-information.pdf.

IDWeek 2026, October 21–24, 2026, Washington, D.C.