IDWeek 2026 | Poster #P-169

Activity of IV fosfomycin against Enterobacterales collected from US medical centers during 2020–2023

KM Papp-Wallace[1], JM Maher[1], M Rodriguez[2], L Bjork[2], ML Winkler[1], M Castanheira[1], HS Sader[1]

[1]Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA; [2]Meitheal Pharmaceuticals, Chicago, Illinois, USA

Background

- Intravenous (IV) fosfomycin was recently approved by the US FDA for the treatment of complicated urinary tract infections, including acute pyelonephritis caused by *Escherichia coli* and *Klebsiella pneumoniae*.
- Fosfomycin inhibits the MurA enzyme, UDP-N-acetylglucosamine-enolpyruvyltransferase, which is responsible for catalyzing the initial step in peptidoglycan synthesis; inhibition of MurA leads to cell lysis and death.
- The activity of IV fosfomycin and comparators was investigated against Enterobacterales isolates collected from the United States from 2020 to 2023.

Methods

- A total of 1,022 Enterobacterales isolates were consecutively collected from 74 hospitals across the US.
- Susceptibility testing was performed using broth microdilution with cation-adjusted Mueller-Hinton broth (CAMHB) for all agents except IV fosfomycin for which agar dilution was used.
- Breakpoints from CLSI were applied for comparators while FDA breakpoints were used for IV fosfomycin.
- The organisms were collected through the SENTRY Surveillance Program.

Results

- Among 1,022 U.S. Enterobacterales isolates, organism distribution varied by census division, while fosfomycin MIC_{50}/MIC_{90} values remained consistent for some organisms and varied for others across regions, including for *E. coli* and *K. pneumoniae* (Figure 1).
- IV fosfomycin was highly active against Enterobacterales with 81.0% and 94.1% of isolates having MICs ≤8 mg/L (*E. coli* susceptibility (S) breakpoint) or ≤32 mg/L (*K. pneumoniae* S breakpoint) and $MIC_{50/90}$ values of 4/16 mg/L (Table 1).
- Comparators displayed similar activity with 78.2–97.5% of isolates testing susceptible; ceftazidime-avibactam (CZA) was the most active agent overall against Enterobacterales (Table 1).
- Against isolates with complex phenotypes, including multidrug-resistant (MDR), extended-spectrum-β-lactamase (ESBL)-phenotype, and carbapenem-non-susceptible (NS), IV fosfomycin was active with 58.7–86% of isolates having MICs ≤8 mg/L or ≤32 mg/L, respectively, with colistin (COL) being the most active agent with 88.1–93.8% of isolates being susceptible (Table 1 and Figure 2).
- Against specific species, 97.3% of *E. coli* and 87.0% of *K. pneumoniae* isolates were susceptible to IV fosfomycin (Figure 2).
- Versus all drug-resistant subsets of *E. coli*, IV fosfomycin was more active than other comparators, except colistin, with >90.6% of isolates being susceptible (Table 1 and Figure 2).
- IV fosfomycin demonstrated equivalent activity (60.7% inhibition) to CZA and gentamicin against carbapenem-NS *K. pneumoniae* (Table 1 and Figure 2).
- IV fosfomycin also performed well against other Enterobacterales with 80.6% and 95.6% having MICs ≤8 mg/L or ≤32 mg/L, respectively (Table 1).

Figure 1. U.S. Census Division distribution of Enterobacterales and IV fosfomycin AST results (n=1,022)

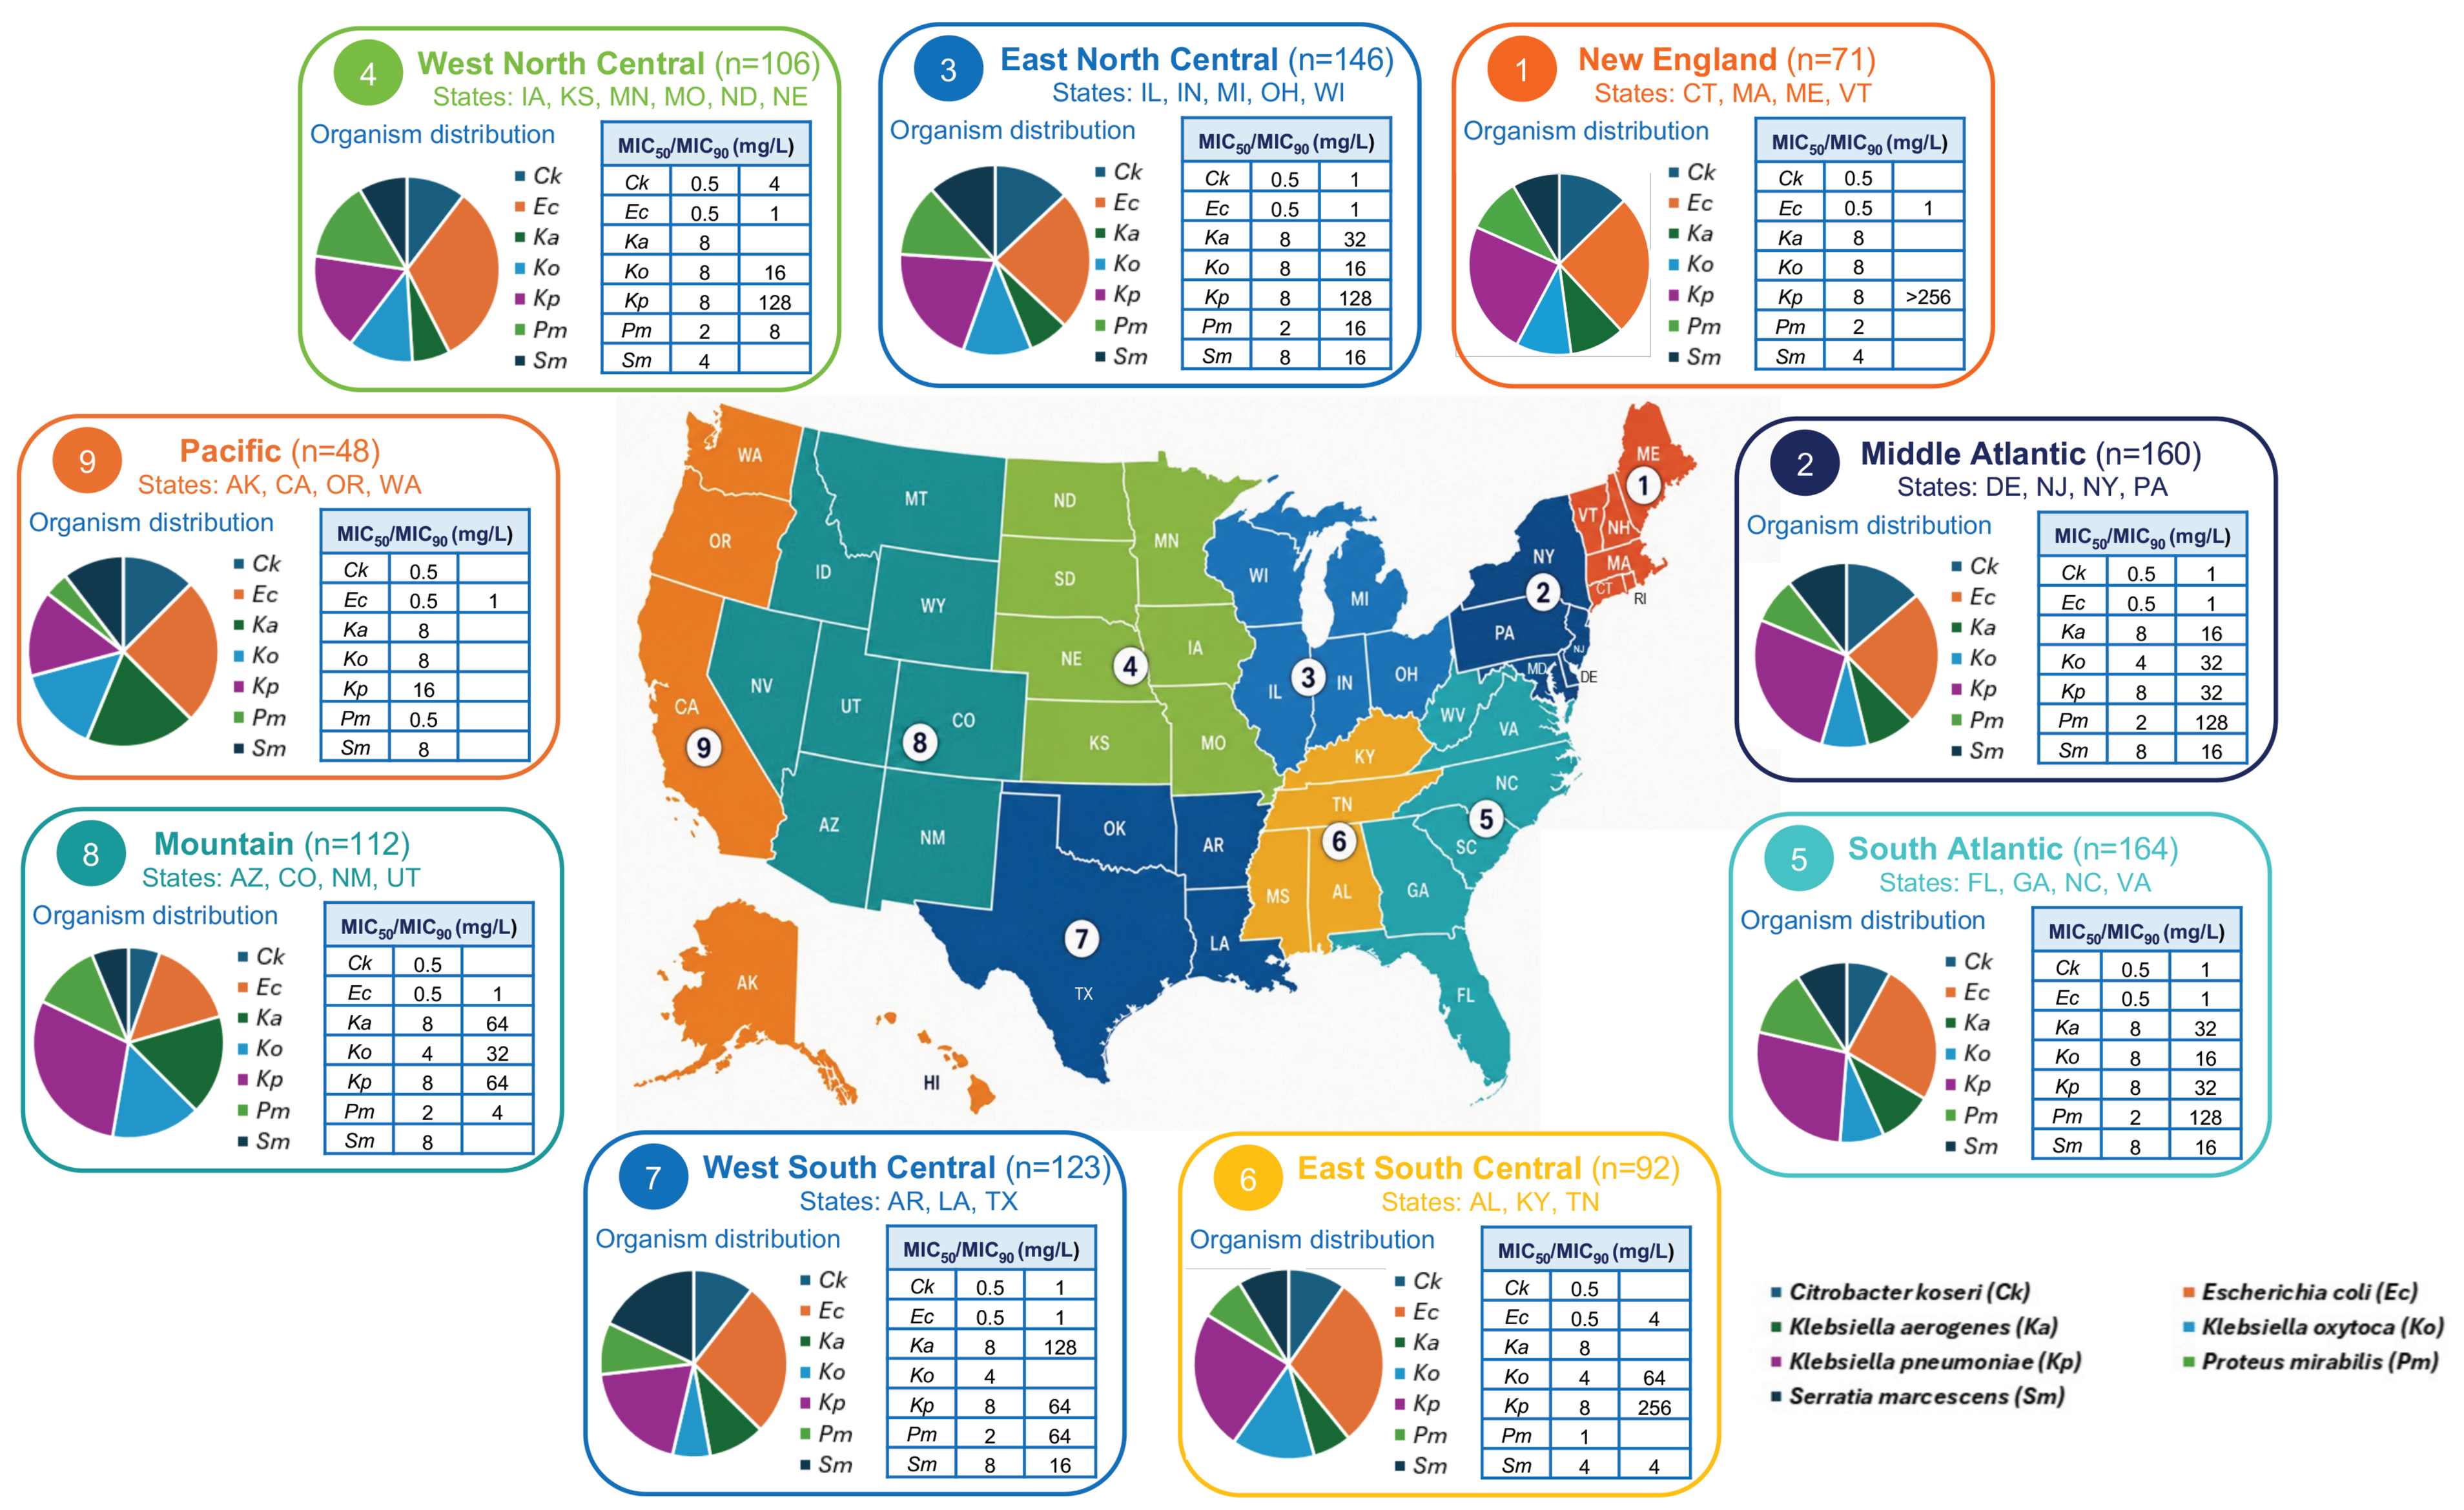
1 New England (n=71)
States: CT, MA, ME, VT
Organism distribution
MIC50/MIC90 (mg/L)
Ck 0.5; Ec 0.5 1; Ka 8; Ko 8; Kp 8 >256; Pm 2; Sm 4
2 Middle Atlantic (n=160)
States: DE, NJ, NY, PA
Ck 0.5 1; Ec 0.5 1; Ka 8 16; Ko 4 32; Kp 8 32; Pm 2 128; Sm 8 16
3 East North Central (n=146)
States: IL, IN, MI, OH, WI
Ck 0.5 1; Ec 0.5 1; Ka 8 32; Ko 8 16; Kp 8 128; Pm 2 16; Sm 8 16
4 West North Central (n=106)
States: IA, KS, MN, MO, ND, NE
Ck 0.5 4; Ec 0.5 1; Ka 8; Ko 8 16; Kp 8 128; Pm 2 8; Sm 4
5 South Atlantic (n=164)
States: FL, GA, NC, VA
Ck 0.5 1; Ec 0.5 1; Ka 8 32; Ko 8 16; Kp 8 32; Pm 2 128; Sm 8 16
6 East South Central (n=92)
States: AL, KY, TN
Ck 0.5; Ec 0.5 4; Ka 8; Ko 4 64; Kp 8 256; Pm 1; Sm 4 4
7 West South Central (n=123)
States: AR, LA, TX
Ck 0.5 1; Ec 0.5 1; Ka 8 128; Ko 4; Kp 8 64; Pm 2 64; Sm 8 16
8 Mountain (n=112)
States: AZ, CO, NM, UT
Ck 0.5; Ec 0.5 1; Ka 8 64; Ko 4 32; Kp 8 64; Pm 2 4; Sm 8
9 Pacific (n=48)
States: AK, CA, OR, WA
Ck 0.5; Ec 0.5 1; Ka 8; Ko 8; Kp 16; Pm 0.5; Sm 8
Citrobacter koseri (Ck)
Klebsiella aerogenes (Ka)
Klebsiella pneumoniae (Kp)
Serratia marcescens (Sm)
Escherichia coli (Ec)
Klebsiella oxytoca (Ko)
Proteus mirabilis (Pm)

Figure 2. Antimicrobial activity of IV fosfomycin and comparators against *E. coli* and *K. pneumoniae* with phenotypic subsets

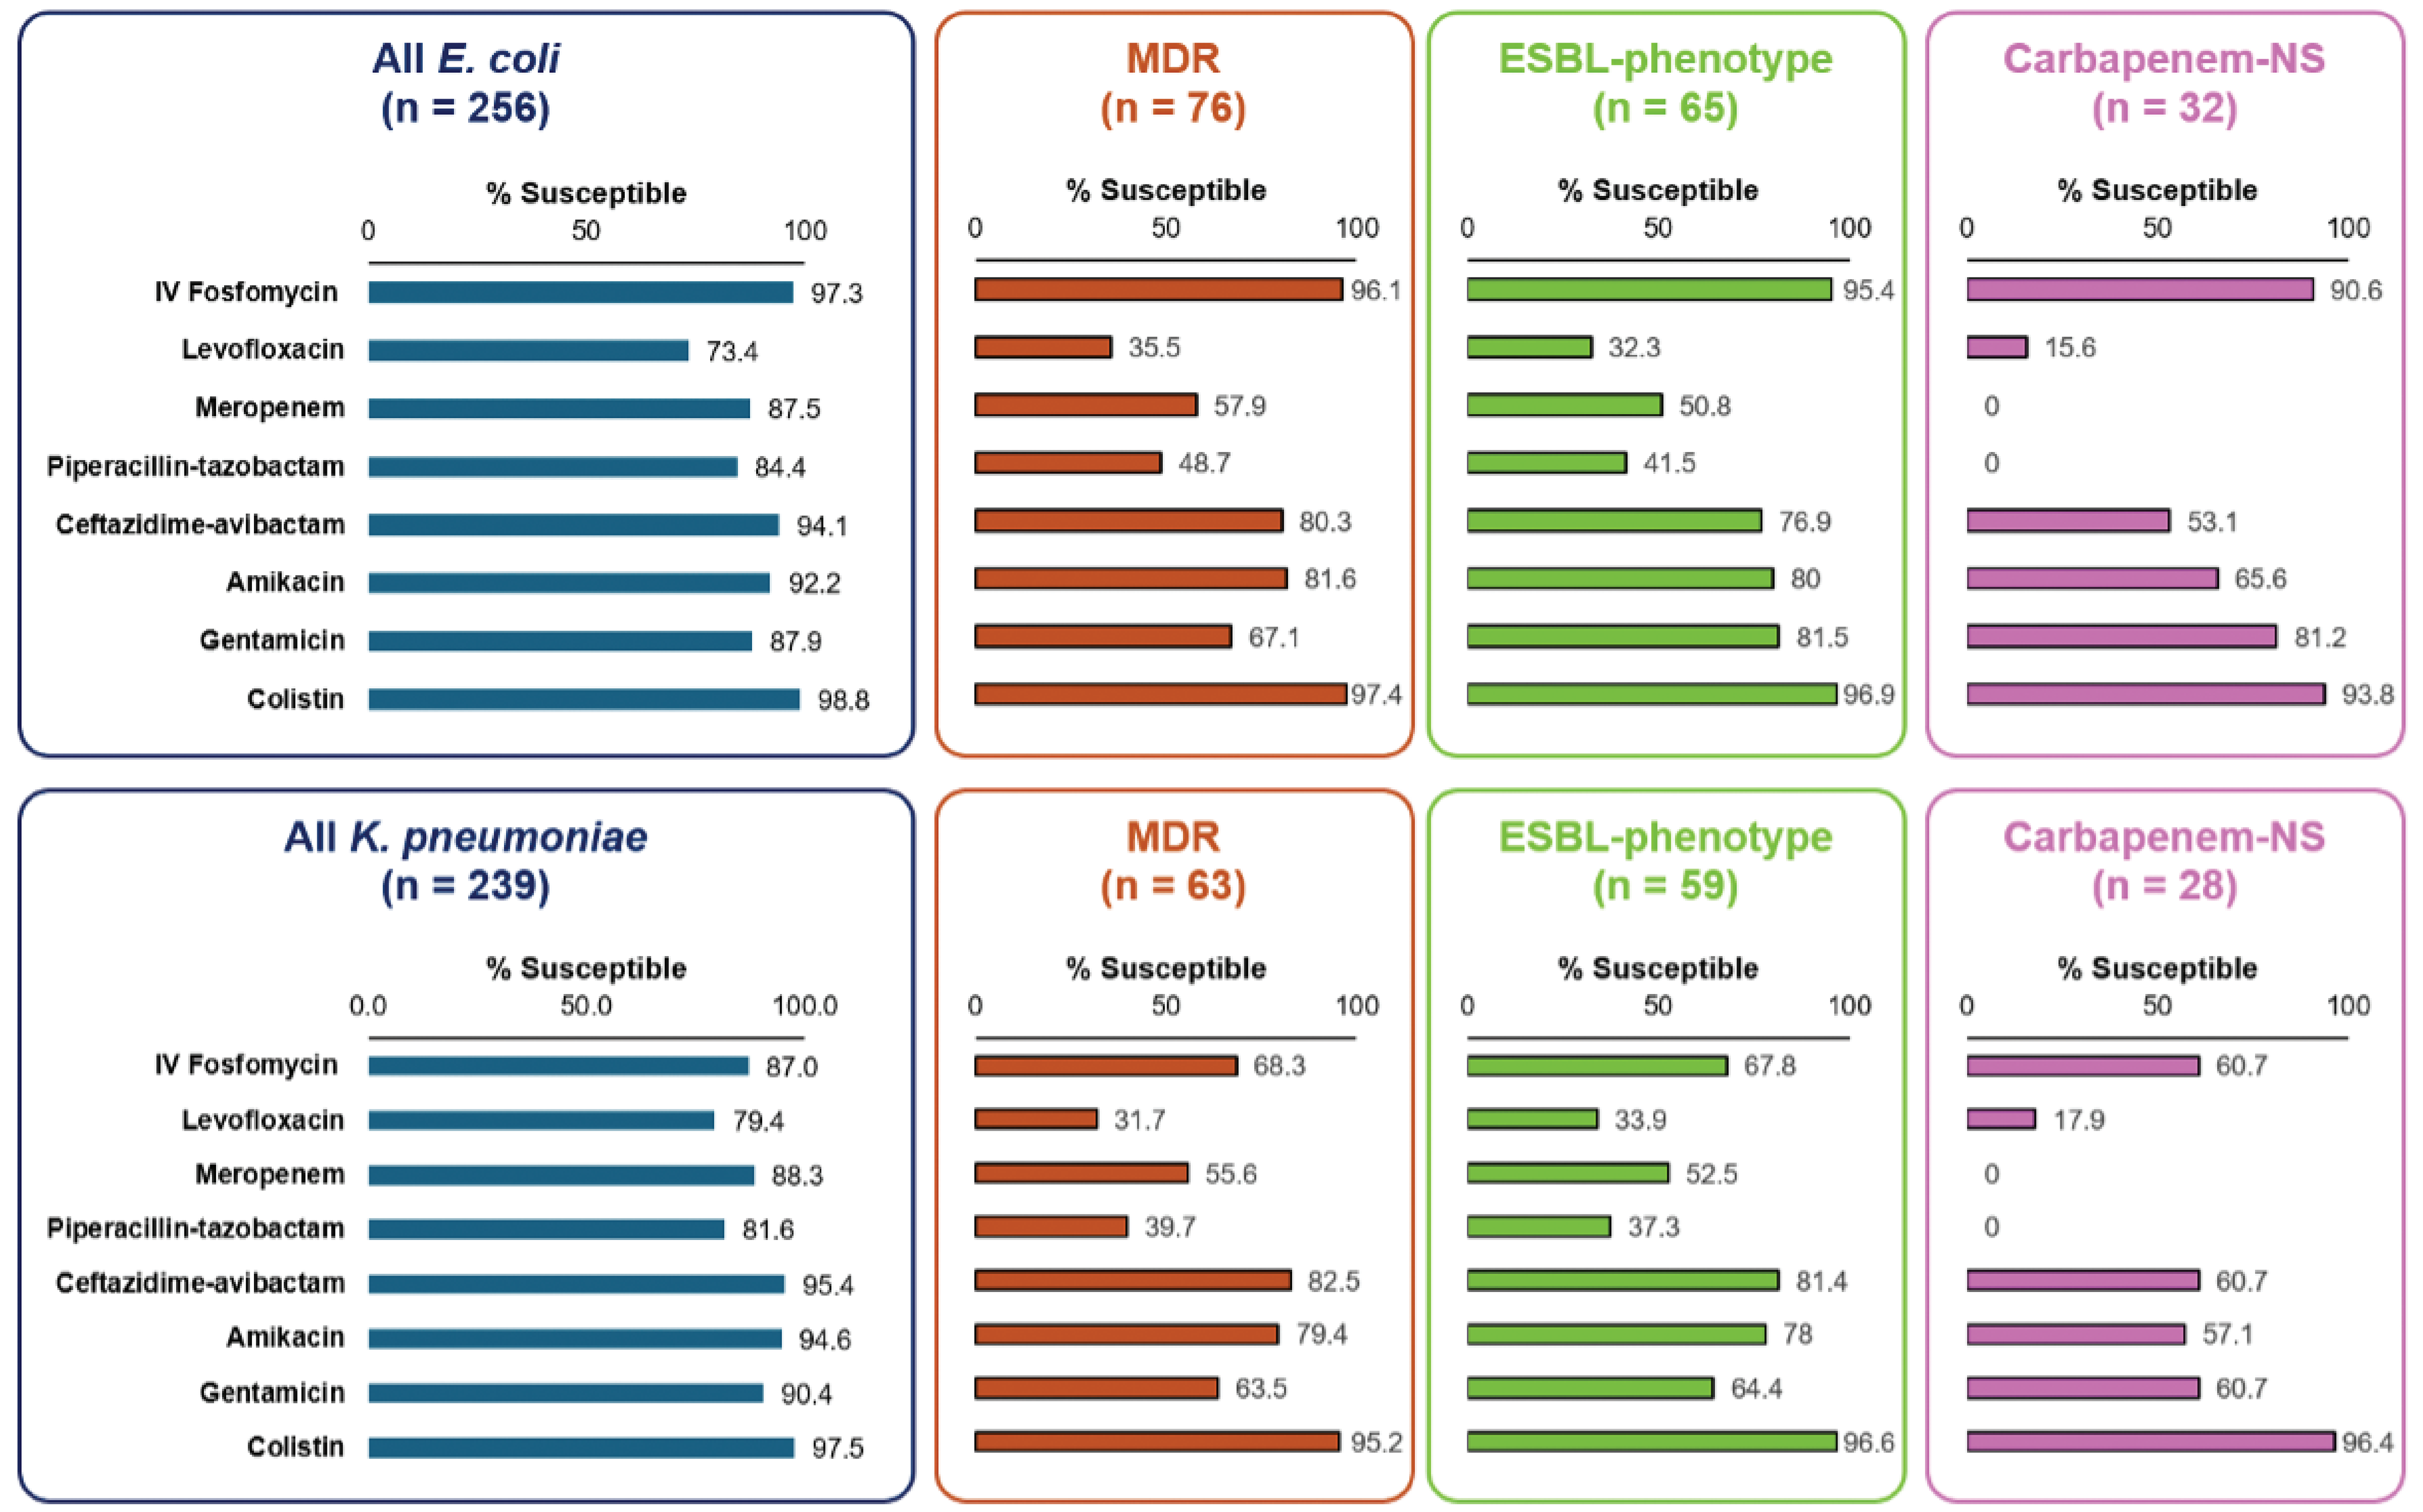
% Susceptible
IV Fosfomycin
Levofloxacin
Meropenem
Piperacillin-tazobactam
Ceftazidime-avibactam
Amikacin
Gentamicin
Colistin
All E. coli (n = 256): 97.3, 73.4, 87.5, 84.4, 94.1, 92.2, 87.9, 98.8
MDR (n = 76): 96.1, 35.5, 57.9, 48.7, 80.3, 81.6, 67.1, 97.4
ESBL-phenotype (n = 65): 95.4, 32.3, 50.8, 41.5, 76.9, 80, 81.5, 96.9
Carbapenem-NS (n = 32): 90.6, 15.6, 0, 0, 53.1, 65.6, 81.2, 93.8
All K. pneumoniae (n = 239): 87.0, 79.4, 88.3, 81.6, 95.4, 94.6, 90.4, 97.5
MDR (n = 63): 68.3, 31.7, 55.6, 39.7, 82.5, 79.4, 63.5, 95.2
ESBL-phenotype (n = 59): 67.8, 33.9, 52.5, 37.3, 81.4, 78, 64.4, 96.6
Carbapenem-NS (n = 28): 60.7, 17.9, 0, 0, 60.7, 57.1, 60.7, 96.4

Table 1. Antimicrobial activity of IV fosfomycin and comparators against Enterobacterales

Phenotype (No.)	IV fosfomycin	Levofloxacin	Meropenem	Piperacillin-tazobactam	Ceftazidime-avibactam	Amikacin	Gentamicin	Colistin
	MIC_{50}/MIC_{90} in mg/L (% susceptible by FDA/CLSI criteria) [a]							
All (1,022)	4/16 81.0-94.1%	0.06/4 84.7%	0.03/0.06 93.8%	2/32 86.3% [b]	0.12/0.5 97.5%	2/4 95.1%	0.5/1 92.5%	0.25/>8 78.2%
MDR (193)	4/64 68.9-86.0%	1/32 47.2%	0.06/32 67.4%	32/>128 39.4% [b]	0.25->32 86.5%	2/8 83.4%	0.5/>16 68.9%	0.25/>8 88.1%
ESBL-phenotype (145)	4/128 67.6-82.8%	4/32 40.7%	0.06/>32 57.9%	64/>128 40.7% [b]	0.25/>32 82.1%	2/16 80.0%	0.5/>16 71.0%	0.25/0.25 93.8%
Carbapenem-NS (63)	4/64 58.7-76.2%	16/>32 20.6%	16/>32 0.0%	>128/>128 1.6% [b]	2/>32 58.7%	4/16 61.9%	1/>16 71.4%	0.25/0.25 93.7%
E. coli (256)	0.5/1 97.3%	0.03/16 73.4%	≤0.015/4 87.5%	2/>128 84.4% [b]	0.12/1 94.1%	2/4 92.2%	0.5/16 87.9%	0.25/0.25 98.8%
MDR (76)	0.5/4 96.1%	8/32 35.5%	0.03/32 57.9%	16/>128 48.7% [b]	0.12/>32 80.3%	2/8 81.6%	1/>16 67.1%	0.25/0.25 97.4%
ESBL-phenotype (65)	0.5/4 95.4%	16/32 32.3%	1/32 50.8%	128/>128 41.5% [b]	0.25/>32 76.9%	2/16 80.0%	1/>16 81.5%	0.25/0.25 96.9%
Carbapenem-NS (32)	1/4 90.6%	16/32 15.6%	8/>32 0.0%	>128/>128 0.0% [b]	4/>32 53.1%	4/16 65.6%	1/16 81.2%	0.25/0.25 93.8%
K. pneumoniae (239)	8/64 87.0%	0.06/8 79.4%	0.03/8 88.3%	4/>128 81.6% [b]	0.12/1 95.4%	1/2 94.6%	0.25/2 90.4%	0.25/0.25 97.5%
MDR (63)	16/>256 68.3%	1/>32 31.7%	0.06/>32 55.6%	32/>128 39.7% [b]	0.25/>32 82.5%	1/16 79.4%	0.5/>16 63.5%	0.25/0.25 95.2%
ESBL-phenotype (59)	16/>256 67.8%	2/>32 33.9%	0.5/>32 52.5%	>128/>128 37.3% [b]	0.25/>32 81.4%	1/16 78.0%	0.5/>16 64.4%	0.25/0.25 96.6%
Carbapenem-NS (28)	32/>256 60.7%	4/>32 17.9%	>32/>32 0.0%	>128/>128 0.0% [b]	2/>32 60.7%	2/>32 57.1%	0.5/>16 60.7%	0.12/0.25 96.4%
Other enterics (527) [c]	4/16 80.6-95.6%	0.06/0.5 92.6%	0.03/0.06 99.4%	2/16 89.4% [b]	0.12/0.25 100%	2/4 96.8%	0.5/1 95.6%	0.25/>8 59.4%

MDR was defined as resistance to at least one drug in three or more antimicrobial categories.
Abbreviations: MDR, multidrug-resistant; ESBL, extended-spectrum β-lactamase; NS, non-susceptible.
[a] MIC results were interpreted according to CLSI criteria (for colistin % susceptible value is % intermediate), except for fosfomycin for which FDA Antibacterial Susceptibility Test Interpretive Criteria were used. For all organisms and other enterics, IV fosfomycin results were interpreted according to *E. coli* (susceptible, ≤8 mg/L) and *K. pneumoniae* FDA breakpoints (susceptible, ≤32 mg/L) and the range is shown.
[b] Intermediate is interpreted as susceptible-dose dependent.
[c] Species included are *C. kosei* (n=108), *K. aerogenes* (n=100), *K. oxytoca* species complex (n=107), *Proteus mirabilis* (n=106), and *S. marcescens* (n=106).

%Susceptible: ≥95% | 94.9-90% | 89.9-60% | 59.9-40% | 39.9-0%

Conclusions

- IV fosfomycin demonstrated potent activity against Enterobacterales, including isolates with drug-resistant phenotypes.

Acknowledgments

This study at Element Iowa City/JMI Laboratories was supported by Meitheal Pharmaceuticals (Chicago, IL). Element Iowa City/JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by Meitheal.

References

CLSI. 2026. M100Ed36. Performance standards for antimicrobial susceptibility testing: 36th informational supplement. Clinical and Laboratory Standards Institute.

https://www.fda.gov/drugs/development-resources/antibacterial-susceptibility-test-interpretive-criteria

Silver, LL. (2017) Fosfomycin: Mechanism and Resistance. *Cold Spring Harb Perspect Med*. 7(2):a025262.

Contact

element

Scan Me

Krisztina M. Papp-Wallace, PhD
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: KrisztinaPappWallace@element.com

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2026_26-MEH-02_P1_Fosfomycin_vs_Enterics.pdf

Charges may apply. No personal information is stored.

IDWeek 2026, October 21–24, 2026, Washington, D.C.