IDWeek 2026 | Poster #P-166

Activity of Cefiderocol-Xeruborbactam and Comparator Agents against Enterobacterales, Including Molecularly Characterized Isolates Recovered Globally in 2024

KM Papp-Wallace[1], JH Kimbrough[1], JM Maher[1], RE Mendes[1], HS Sader[1], and M Castanheira[1]

[1]Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA

Background

- Cefiderocol-xeruborbactam (FDX) is a siderophore-conjugated cephalosporin combined with a boronate β-lactamase inhibitor, and this combination shows wide-ranging activity against Gram-negatives.
- The activity of cefiderocol-xeruborbactam and comparators was investigated against Enterobacterales collected from the US, Europe, Turkey, and Israel in 2024.

Methods

- A total of 7,995 Enterobacterales were collected from 69 hospitals in 20 countries through the SENTRY Surveillance Program.
- Susceptibility testing was performed using broth microdilution with cation-adjusted Mueller-Hinton broth (CAMHB) for all agents except cefiderocol that was tested in iron-depleted CAMHB.
- Clinical Laboratory Standards Institute (CLSI) breakpoints were applied.
- Carbapenem-non-susceptible Enterobacterales (CNSE) with MICs ≥2 mg/L for imipenem/meropenem were screened for carbapenemase genes.

Results

- Cefiderocol-xeruborbactam demonstrated potent activity against Enterobacterales collected from the US (Figure 1) and Europe (Figure 2) with 100% of isolates with an MIC ≤4 mg/L and $MIC_{50/90}$ of ≤0.015/0.12 mg/L, while % susceptible varied from 83.1–99.9% for comparators (Table 1).
- All isolates, including multidrug-resistant (MDR), extensively drug-resistant (XDR), and CNSE with and without carbapenemases had cefiderocol-xeruborbactam MIC values ≤4 mg/L (Table 1 and Figure 3).
- Against MDR isolates, comparators were mostly effective with 88.7–99.6% of isolates testing susceptible (Table 1 and Figure 3).
- Conversely, XDR isolates were not as susceptible to most comparators (38.2–67.2%) except for cefiderocol at 96.3% and aztreonam-avibactam at 99.2% (Table 1 and Figure 3).
- Enterobacterales from Europe were less susceptible to imipenem-relebactam, meropenem-vaborbactam, and ceftazidime-avibactam compared to isolates from the US (Figure 3).
- European isolates were slightly more susceptible to cefiderocol and aztreonam-avibactam compared to US isolates (Figure 3).
- KPC- and MBL-producers were collected in the US and Europe, while OXA-48-like-producers and strain carrying more than one carbapenemase were only identified in European isolates (Figure 3).
- Comparators were largely active (80.6–98.4%) against KPC-producers (Table 1 and Figure 3).
- However, limited activity was reported with most comparators against MBL- (0–13.5%) and OXA-48-producers (22.0–52.0%), except cefiderocol for both categories (94.2–100%), aztreonam-avibactam for both categories (98.1–100%), ceftazidime-avibactam for OXA-48-producers (100%), and colistin for KPC- and MBL-producers (~81%) (Table 1 and Figure 3).
- Isolates carrying multiple carbapenemases were challenging to inhibit by most comparators (0–55.6% susceptible), except for cefiderocol at 92.1% and aztreonam-avibactam at 100% susceptible (Table 1 and Figure 3).
- Most comparators had activity against carbapenemase negative isolates (88.1–100%), except for colistin (Table 1 and Figure 3).

Conclusions

- Cefiderocol-xeruborbactam was highly active against Enterobacterales and demonstrated potent activity against XDR pathogens carrying multiple carbapenemases warranting further development for the treatment of infections caused by diverse Enterobacterales.

Acknowledgments

This study at Element Iowa City/JMI Laboratories was supported by Qpex Biopharma (San Diego, CA). Element Iowa City/JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by Qpex. This project has been funded in whole or in part with federal funds from the Department of Health and Human Services; Administration for Strategic Preparedness and Response; Center for the Biomedical Advanced Research and Development Authority (BARDA), under OTA number HHSO100201600026C.

References

CLSI. 2026. M100Ed36. Performance standards for antimicrobial susceptibility testing: 36th informational supplement. Clinical and Laboratory Standards Institute.

Contact

Krisztina M. Papp-Wallace, PhD
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: KrisztinaPappWallace@element.com

Scan Me

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2026_26-QPX-02_P1_CFDC-Xeru_Enterobacterales.pdf

Charges may apply. No personal information is stored.

Figure 1. U.S. Census division distribution of Enterobacterales and cefiderocol-xeruborbactam AST results (n=3,488)

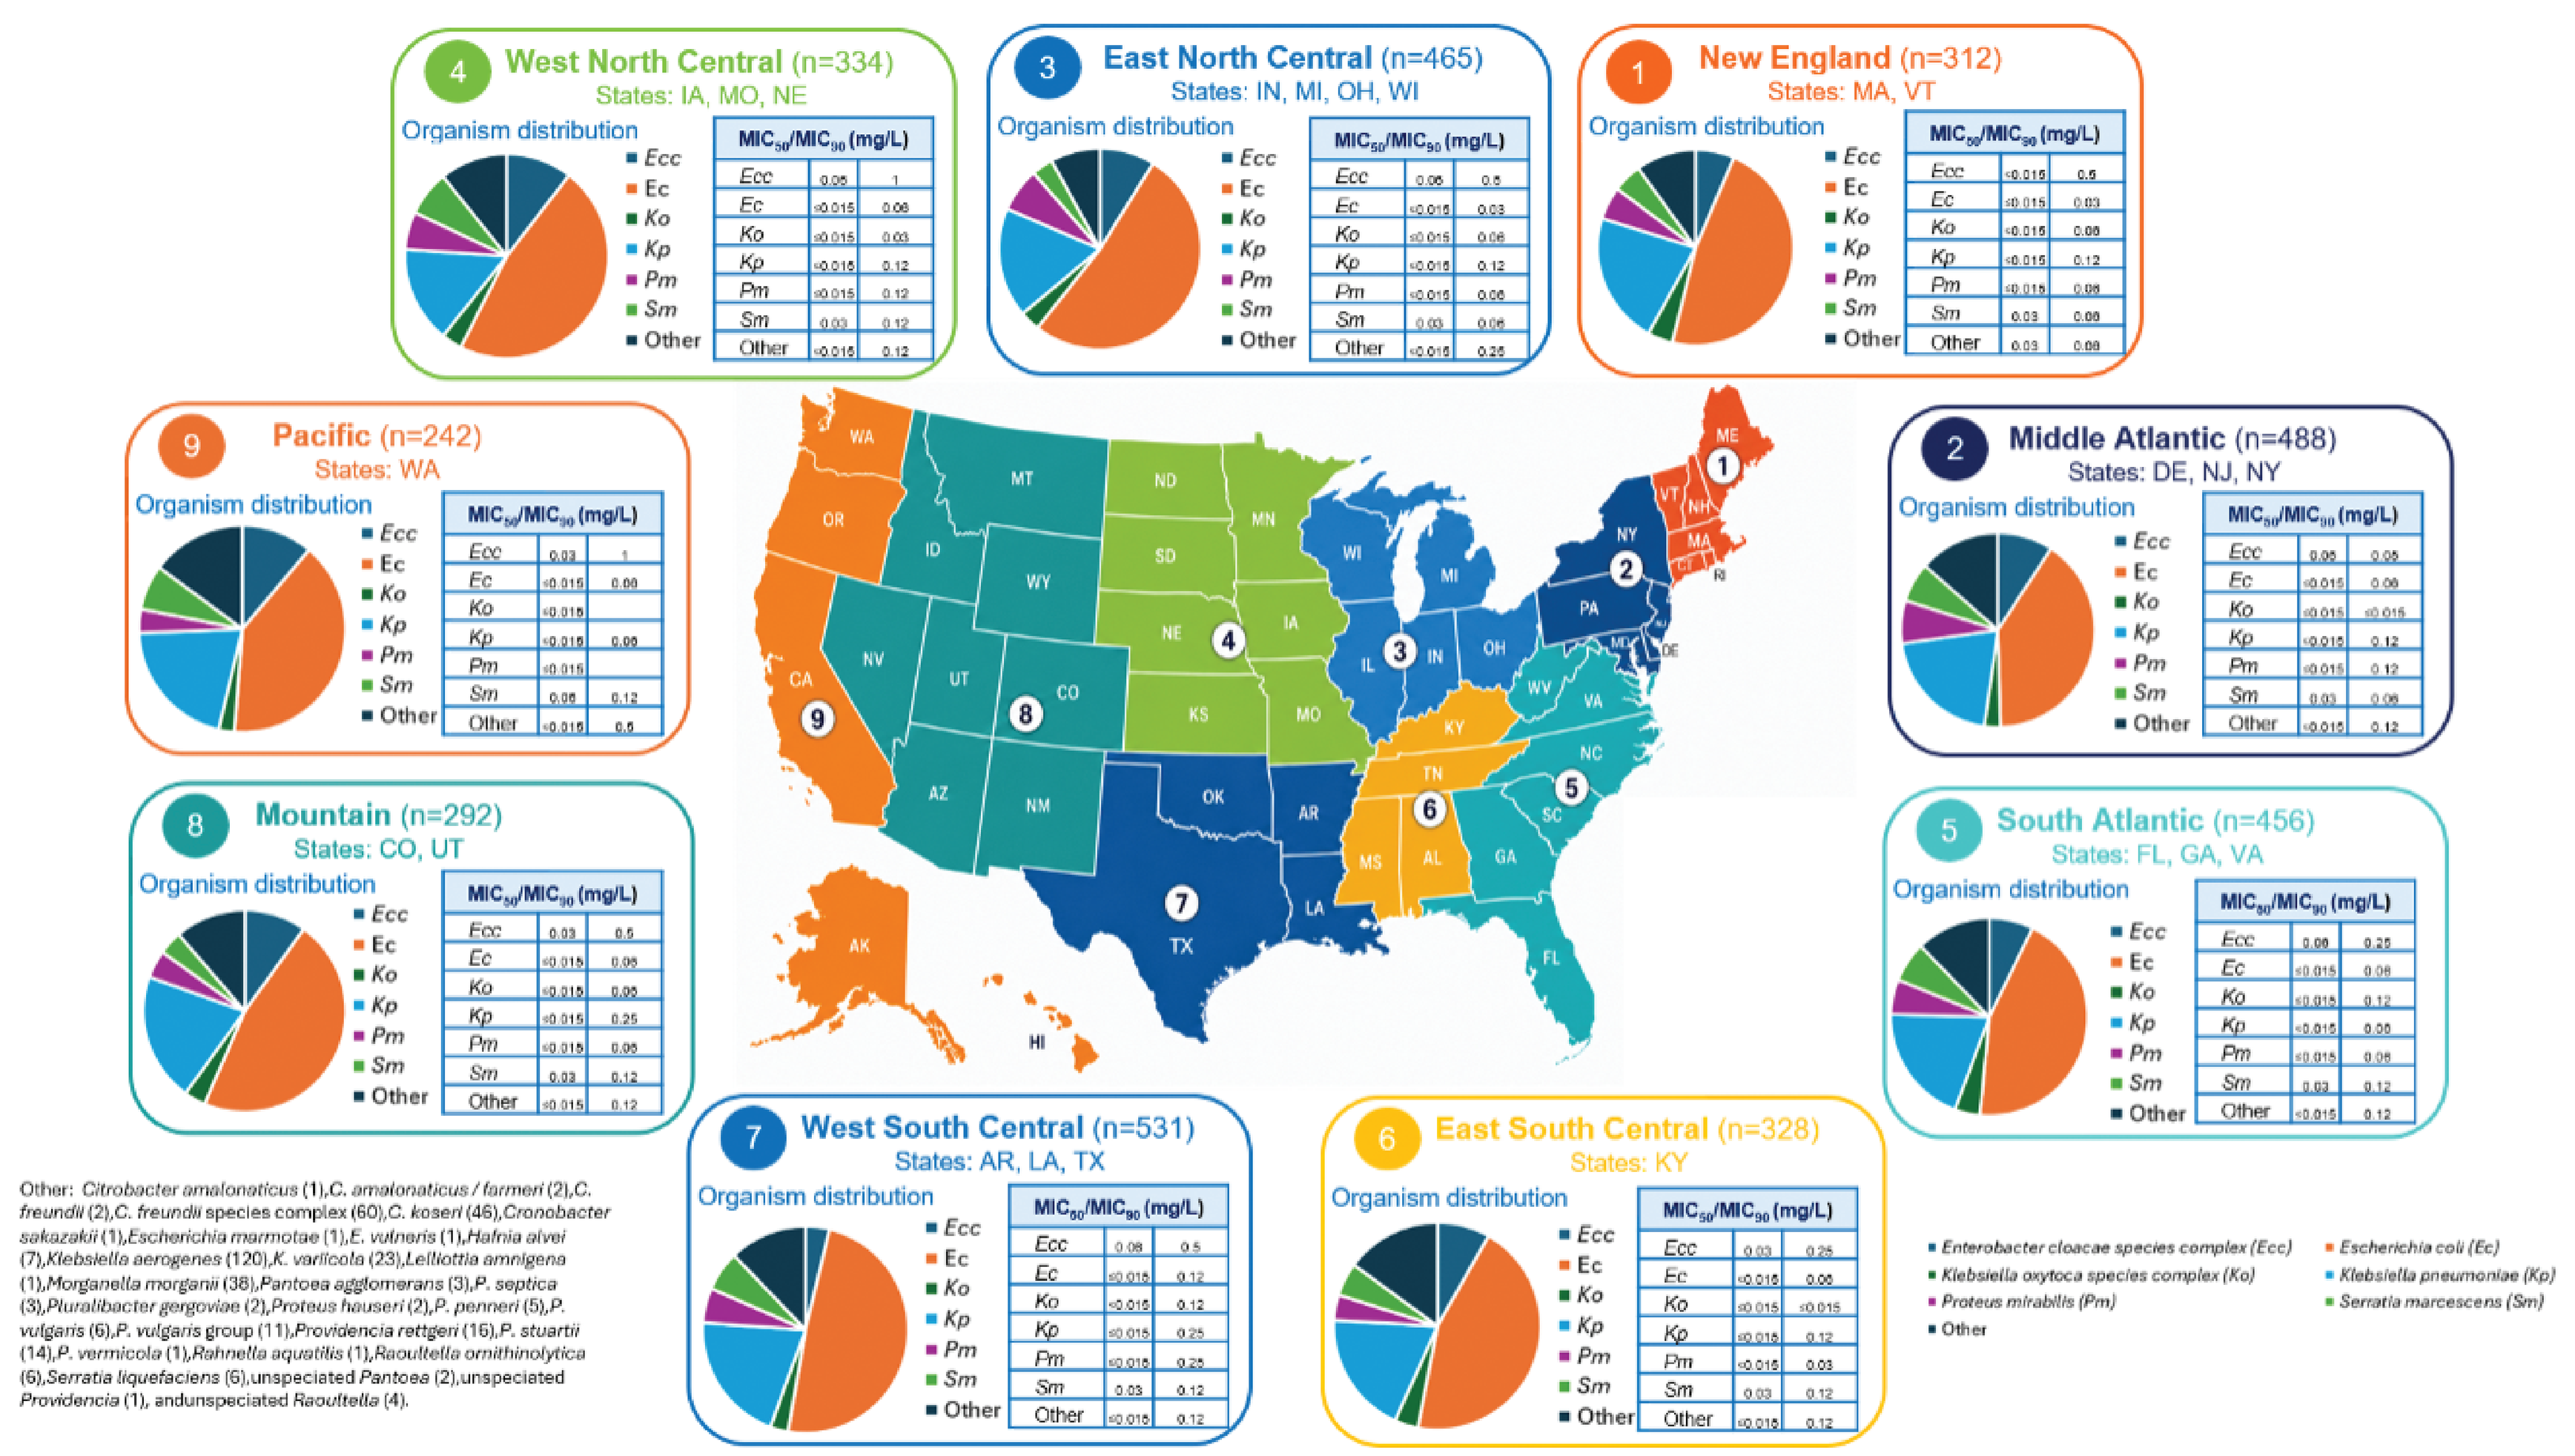
West North Central (n=334)
States: IA, MO, NE
East North Central (n=465)
States: IN, MI, OH, WI
New England (n=312)
States: MA, VT
Pacific (n=242)
States: WA
Middle Atlantic (n=488)
States: DE, NJ, NY
Mountain (n=292)
States: CO, UT
South Atlantic (n=456)
States: FL, GA, VA
West South Central (n=531)
States: AR, LA, TX
East South Central (n=328)
States: KY
Organism distribution
MIC50/MIC90 (mg/L)
Ecc
Ec
Ko
Kp
Pm
Sm
Other
Enterobacter cloacae species complex (Ecc)
Escherichia coli (Ec)
Klebsiella oxytoca species complex (Ko)
Klebsiella pneumoniae (Kp)
Proteus mirabilis (Pm)
Serratia marcescens (Sm)
Other

Figure 2. European distribution of Enterobacterales and cefiderocol-xeruborbactam AST results (n=4,547)

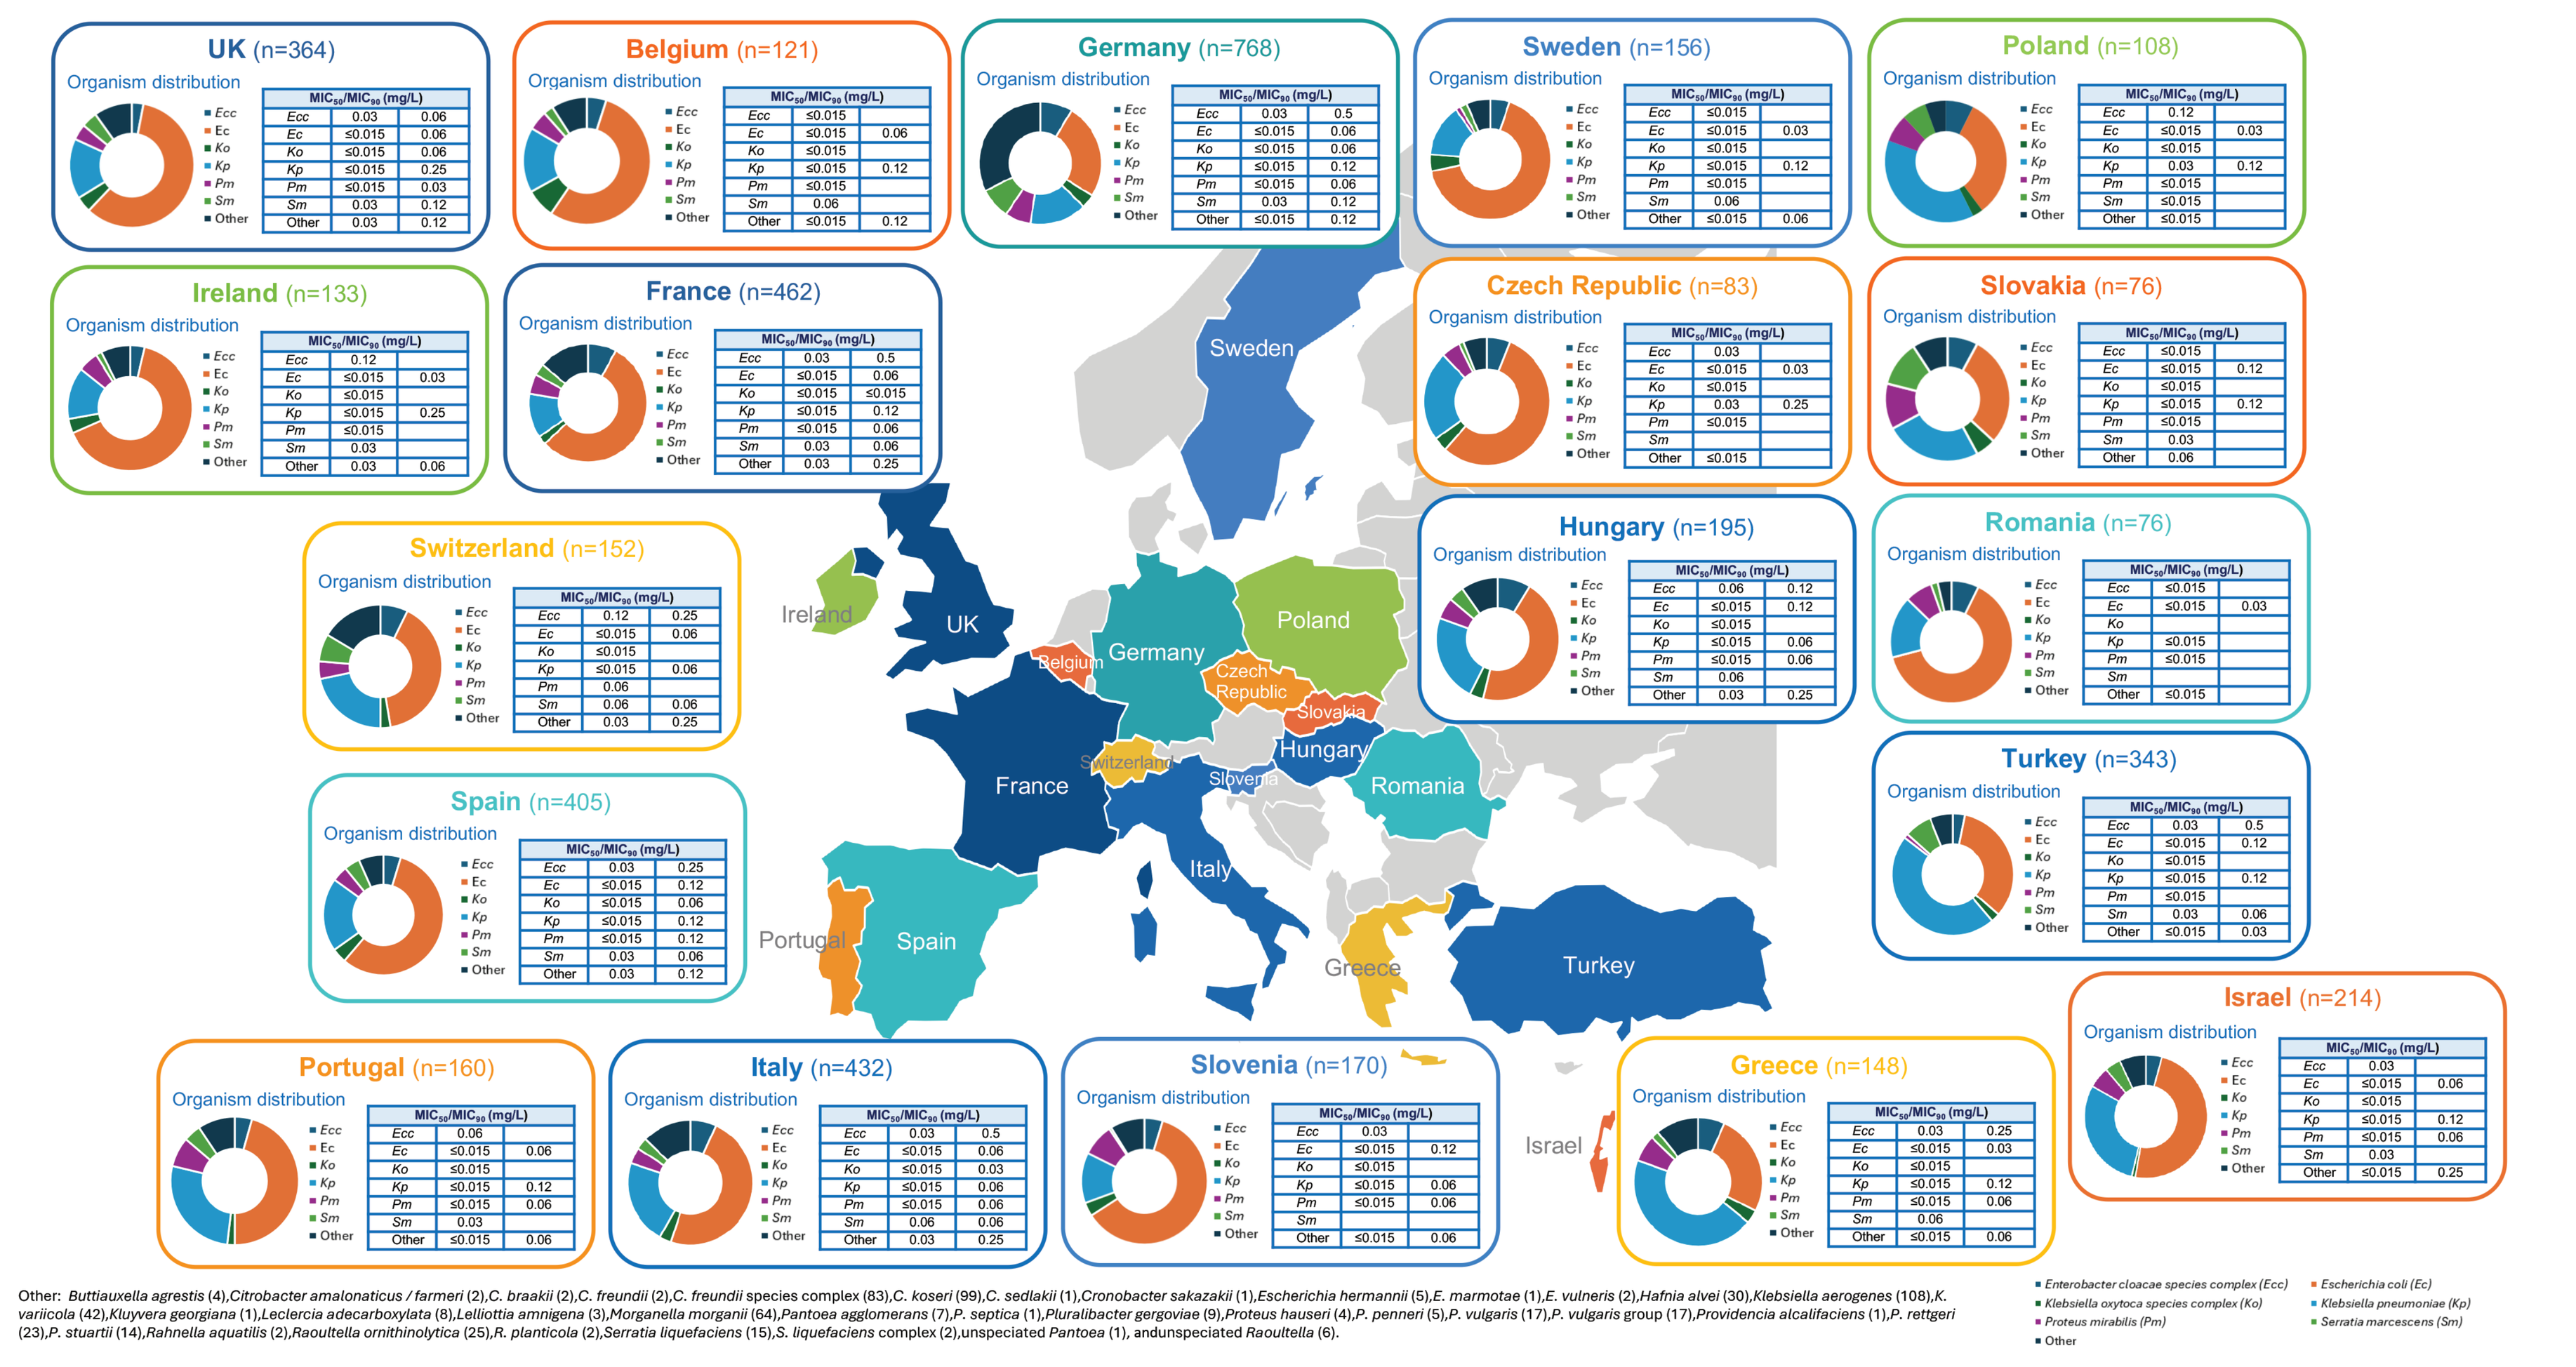
UK (n=364)
Belgium (n=121)
Germany (n=768)
Sweden (n=156)
Poland (n=108)
Ireland (n=133)
France (n=462)
Czech Republic (n=83)
Slovakia (n=76)
Switzerland (n=152)
Hungary (n=195)
Romania (n=76)
Turkey (n=343)
Spain (n=405)
Israel (n=214)
Portugal (n=160)
Italy (n=432)
Slovenia (n=170)
Greece (n=148)
Organism distribution
MIC50/MIC90 (mg/L)
Enterobacter cloacae species complex (Ecc)
Escherichia coli (Ec)
Klebsiella oxytoca species complex (Ko)
Klebsiella pneumoniae (Kp)
Proteus mirabilis (Pm)
Serratia marcescens (Sm)
Other

Figure 3. Antimicrobial activity of cefiderocol-xeruborbactam and comparators against Enterobacterales stratified by phenotype, genotype, and region

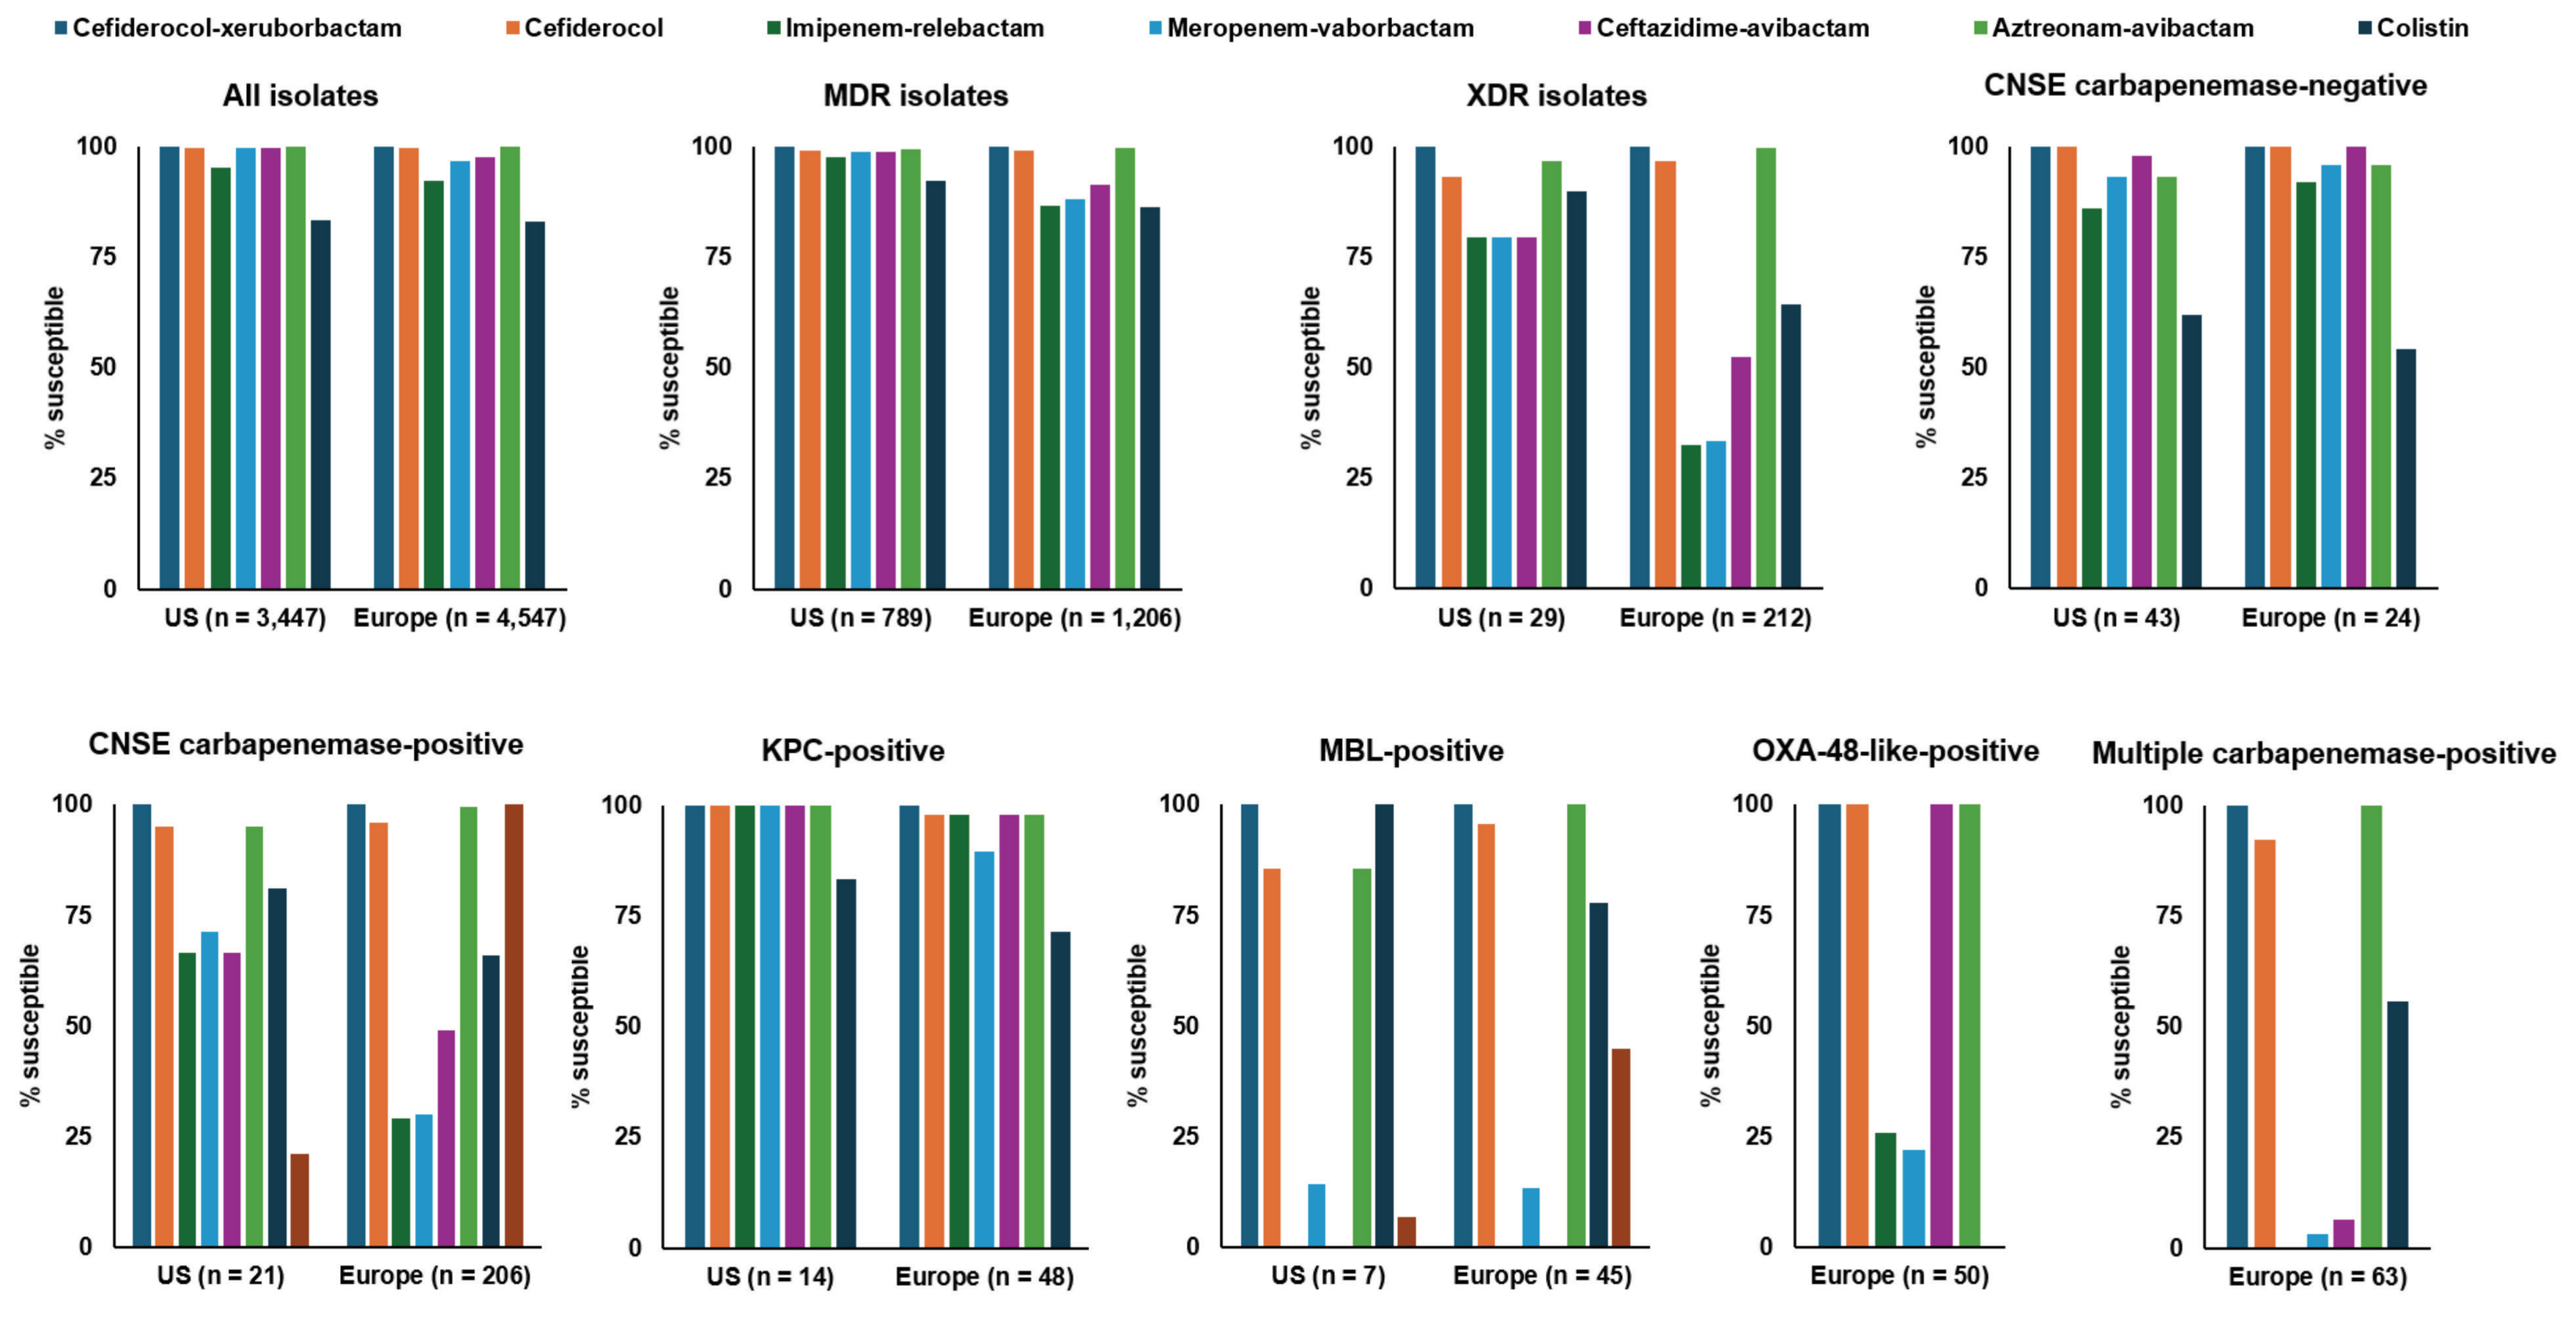
Cefiderocol-xeruborbactam
Cefiderocol
Imipenem-relebactam
Meropenem-vaborbactam
Ceftazidime-avibactam
Aztreonam-avibactam
Colistin
All isolates
US (n = 3,447)
Europe (n = 4,547)
MDR isolates
US (n = 789)
Europe (n = 1,206)
XDR isolates
US (n = 29)
Europe (n = 212)
CNSE carbapenemase-negative
US (n = 43)
Europe (n = 24)
CNSE carbapenemase-positive
US (n = 21)
Europe (n = 206)
KPC-positive
US (n = 14)
Europe (n = 48)
MBL-positive
US (n = 7)
Europe (n = 45)
OXA-48-like-positive
Europe (n = 50)
Multiple carbapenemase-positive
Europe (n = 63)
% susceptible
0
25
50
75
100

Table 1. Antimicrobial activity of cefiderocol-xeruborbactam and comparators against Enterobacterales

%Susceptible: ≥99% | 98.9-90% | 89.9-60% | 59.9-40% | 39.9-0%

Phenotype/genotype (No.)	Cefiderocol-xeruborbactam [b]	Cefiderocol	Imipenem-relebactam [c]	Meropenem-vaborbactam	Ceftazidime-avibactam	Aztreonam-Avibactam	Colistin
	MIC_{50}/MIC_{90} in mg/L (% susceptible by CLSI criteria)[a]						
All (7,995)	≤0.015/0.12 100%	0.12/0.5 99.8%	0.12/1 93.5%	0.03/0.06 98.1%	0.12/0.25 98.6%	≤0.03/0.12 99.9%	0.25/>8 83.1%
MDR (1,995)[d]	≤0.015/0.12 100%	0.5/2 99.2%	0.12/1 90.9%	0.03/0.5 92.3%	0.25/1 94.2%	0.06/0.5 99.6%	0.25/8 88.7%
XDR (241)[d]	0.03/0.25 100%	1/2 96.3%	4/>16 38.2%	16/>32 39.0%	4/>32 55.6%	0.25/0.5 99.2%	0.25/>8 67.2%
Carbapenem-NS (294)[e]	0.03/0.25 100%	0.5/2 96.9%	2/>16 45.2%	8/>32 47.6%	2/>32 61.6%	0.25/0.5 98.0%	0.25/>8 65.5%
Carbapenemase + (227)[f]	0.03/0.25 100%	1/4 96.0%	4/>16 32.6%	16/>32 33.9%	8/>32 50.7%	0.25/0.5 99.1%	0.25/>8 67.4%
KPCs (62)	≤0.015/0.25 100%	0.25/2 98.4%	0.12/0.5 98.4%	0.06/4 91.9%	1/4 98.4%	0.25/0.5 98.4%	0.25/>8 80.6%
MBLs (52)	≤0.015/0.5 100%	1/4 94.2	16/>16 0.0%	>32/>32 13.5%	>32/>32 0.0%	0.12/1 98.1	0.25/>8 80.8%
OXA-48-like (50)	0.03/0.25 100%	0.5/2 100.0	2/8 26.0%	16/>32 22.0%	1/2 100.0%	0.25/0.5 100%	0.25/>8 52.0%
Multiple (63)	0.03/0.12 100%	2/4 92.1%	>16/>16 0.0%	>32/>32 3.2%	>32/>32 6.3%	0.25/0.5 100%	0.25/>8 55.6%
Carbapenemase – (67)[g]	0.06/0.5 100%	0.25/2 100%	0.25/2 88.1%	0.06/2 94.0%	0.25/4 98.5%	0.12/2 94.0%	0.25/>8 59.1%

Abbreviations: MDR, multidrug-resistant; XDR, extensively drug-resistant; NS, non-susceptible; +, positive; -, negative.

[a] MIC results were interpreted according to CLSI criteria for colistin % susceptible value as % intermediate. FDX MICs were read at 100% inhibition tested in CAMHB and FDC MICs were read at significant reduction tested in ID-CAMHB. As breakpoints are not available for FDX, % values represent % of MIC values for FDX ≤ 4 mg/L

[b] Xeruborbactam was tested at fixed 4 mg/L, while the concentration of cefiderocol was varied.

[c] All Enterobacterales species were included in the analysis, but CLSI and EUCAST exclude organisms from the family Morganellaceae.

[d] MDR was defined as resistance to at least one drug in three or more antimicrobial categories. XDR was defined as resistance to all but ≤2 antimicrobial categories

[e] Carbapenem-NS, isolates non-susceptible to imipenem (excluded for *P. mirabilis*, *P. penneri*, and indole-positive Proteeae) and/or meropenem based on CLSI criteria (MIC values ≥2 mg/L).

[f] Includes bla_{KPC-2} (21), bla_{KPC-3} (39), bla_{KPC-18} (1), and $bla_{KPC-166}$ (1) genes; bla_{NDM-1} (28), bla_{NDM-5} (12), bla_{NDM-7} (1), bla_{NDM-71} (1), bla_{VIM-1} (8), and bla_{VIM-4} (2) genes; bla_{OXA-48} (19), $bla_{OXA-181}$ (2), $bla_{OXA-232}$ (26), and $bla_{OXA-244}$ (3) genes; and multiple bla_{NDM-1} + bla_{KPC-2} (3), bla_{NDM-1} + bla_{KPC-3} (2), bla_{NDM-1} + bla_{OXA-48} (20), bla_{NDM-1} + $bla_{OXA-181}$ (2), bla_{NDM-1} + $bla_{OXA-232}$ (2), bla_{NDM-5} + bla_{OXA-48} (26), bla_{NDM-5} + $bla_{OXA-232}$ (4), bla_{VIM-1} + bla_{KPC-2} (3), and bla_{KPC-2} + bla_{OXA-48} (1).

[g] Carbapenemase genes were not detected in the following species: *Citrobacter freundii* species complex (12), *Enterobacter cloacae* species complex (14), *Klebsiella aerogenes* (7), *K. oxytoca* species complex (1), *K. pneumoniae* (15), *Serratia liquefaciens* complex (2), and *S. marcescens* (16).

IDWeek 2026, October 21–24, 2026, Washington, D.C.